Osimertinib treatment increases expression of HER2 and combination treatment with trastuzumab deruxtecan, a HER2-directed antibody-drug conjugate, enhances its efficacy in PDX models of EGFR mutant non-small cell lung cancer

P2.02.27

Matthew Martin[1], Alex Koers[1], Fernando Calero[1], Sara Talbot[1], Adina Hughes[1], Lukasz Magiera[1], Lucy Ireland[1], Benjamin Phillips[2], Ricardo Miragaia[3], and Nicolas Floc'h*[1].

AstraZeneca – 1 Francis Crick Avenue; Cambridge, UK CB2 0AA. 1. Oncology Targeted Discovery, Bioscience 2. Data Sciences & Quantitative Biology, Discovery Sciences 3. Oncology Data Science *presenting author (nicolas.floch@astrazeneca.com)

Introduction

- Osimertinib is a 3rd generation EGFR tyrosine kinase inhibitor that has demonstrated efficacy in the first-line treatment of advanced or metastatic EGFR-mutant NSCLC[1].
- Despite clinical benefit, most patients become refractory to osimertinib[2], highlighting the need for combination strategies to maximize the duration of response or address osimertinib resistance.
- Trastuzumab deruxtecan (T-DXd) is a human epithelial growth factor receptor 2 (HER2)-directed antibody drug conjugate (ADC) approved across multiple indications[3] thus, we wished to understand how T-DXd in combination with osimertinib will perform in EGFRm preclinical models

Results

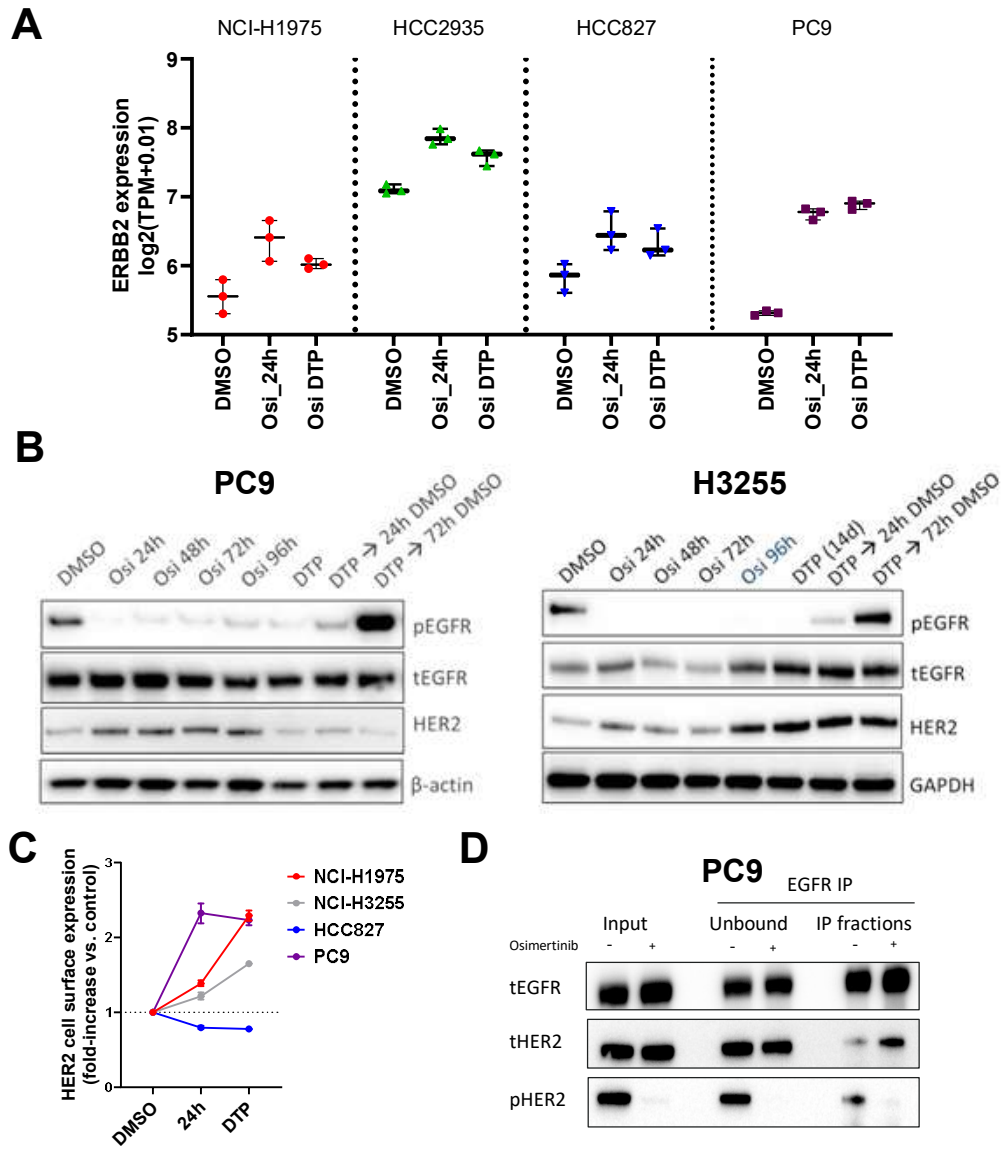
A
NCI-H1975
HCC2935
HCC827
PC9
ERBB2 expression log2(TPM+0.01)
DMSO
Osi_24h
Osi DTP
B
PC9
H3255
pEGFR
tEGFR
HER2
β-actin
GAPDH
C
HER2 cell surface expression (fold-increase vs. control)
NCI-H1975
NCI-H3255
HCC827
PC9
DMSO
24h
DTP
D
PC9
EGFR IP
Input
Unbound
IP fractions
Osimertinib
tEGFR
tHER2
pHER2

Fig. 1 Osimertinib treatment promotes upregulation of HER2 in vitro A Expression of the mRNA for HER2 in EGFRm cell lines treated with osimertinib for 24h or 21 days (DTP) plotted as log2 of transcripts per million (TPM). Expression of HER2 protein, in whole cell lysates (**B**; western blot) or cell surface (**C**; flow cytometry) in EGFRm cell lines treated with osimertinib (DTP = 14 days treatment). Where indicated DTPs were released from drug for 24 or 72h (DMSO). **D** EGFR IP in PC9 treated with osimertinib for 24h

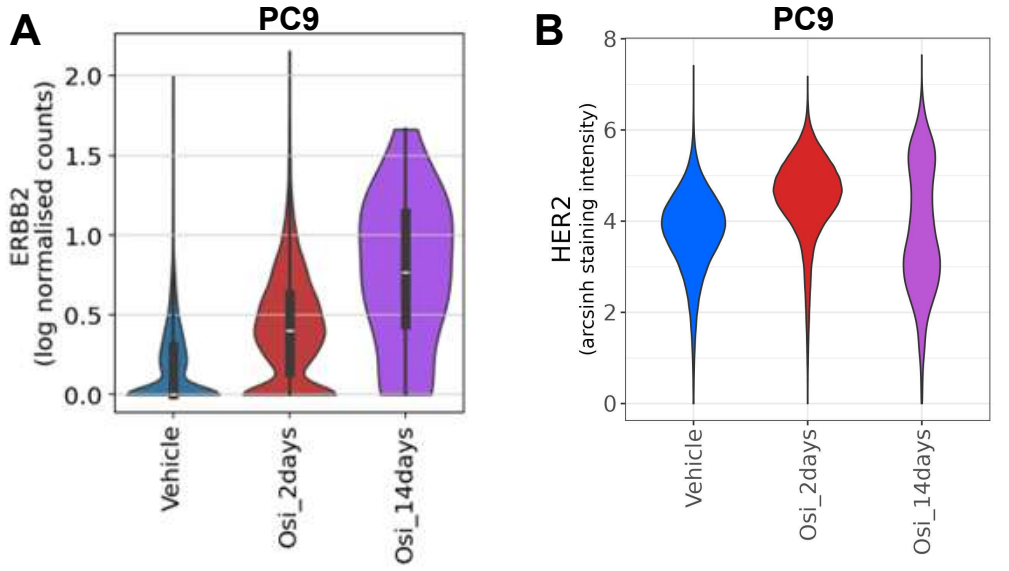
A
PC9
ERBB2 (log normalised counts)
Vehicle
Osi_2days
Osi_14days
B
PC9
HER2 (arcsinh staining intensity)

Figure 2: HER2 transcriptional and proteomic levels are heterogenous in PC9 *in vivo* cell population following osimertinib treatment. Transcriptomic (**A**) and proteomic (**B**) levels of HER2 were measured at single-cell resolution using scRNA-Seq and CyTOF, respectively. Logarithmic normalized counts are shown in A. Arcsinh transformation of staining intensity is shown in B.

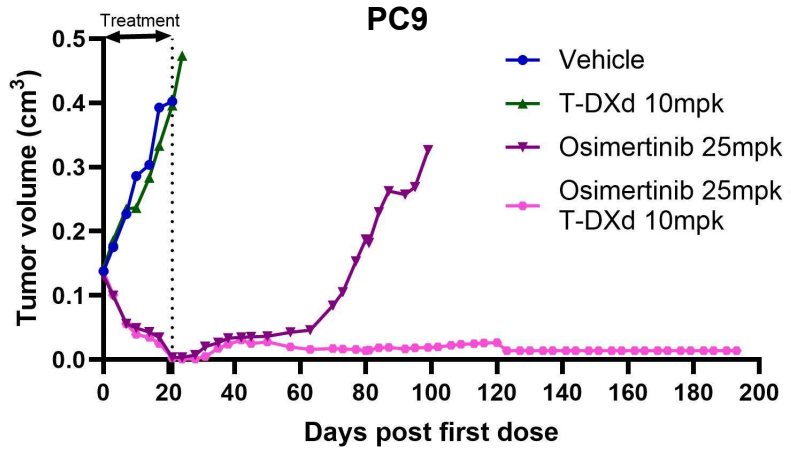
PC9
Treatment
Tumor volume (cm³)
Days post first dose
Vehicle
T-DXd 10mpk
Osimertinib 25mpk
Osimertinib 25mpk + T-DXd 10mpk

Figure 3: Combination of osimertinib and T-DXd results in delayed regrowth over osimertinib monotherapy *in vivo*. Geometric mean of tumour volume is shown. Treatment took place for 21 days and regrowth of tumours was monitored over time.

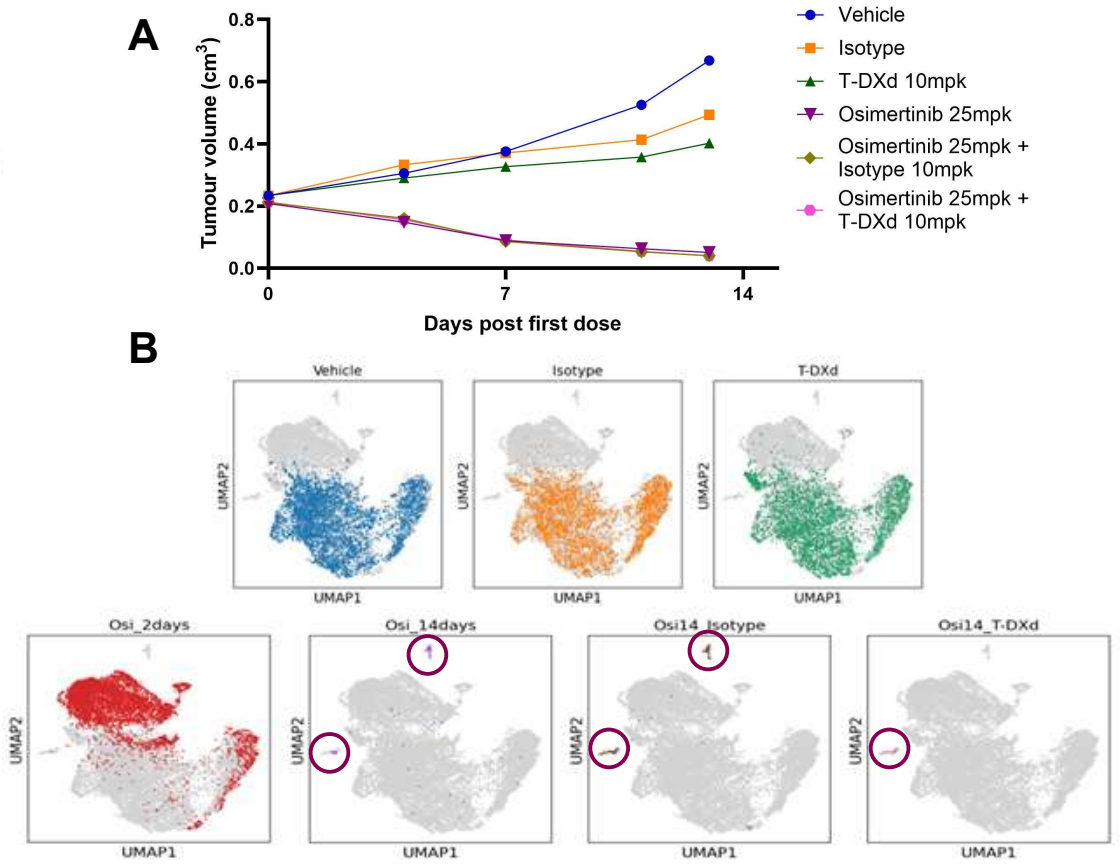
A
Tumour volume (cm³)
Days post first dose
Vehicle
Isotype
T-DXd 10mpk
Osimertinib 25mpk
Osimertinib 25mpk + Isotype 10mpk
Osimertinib 25mpk + T-DXd 10mpk
B
Vehicle
Isotype
T-DXd
Osi_2days
Osi_14days
Osi14_Isotype
Osi14_T-DXd
UMAP1
UMAP2

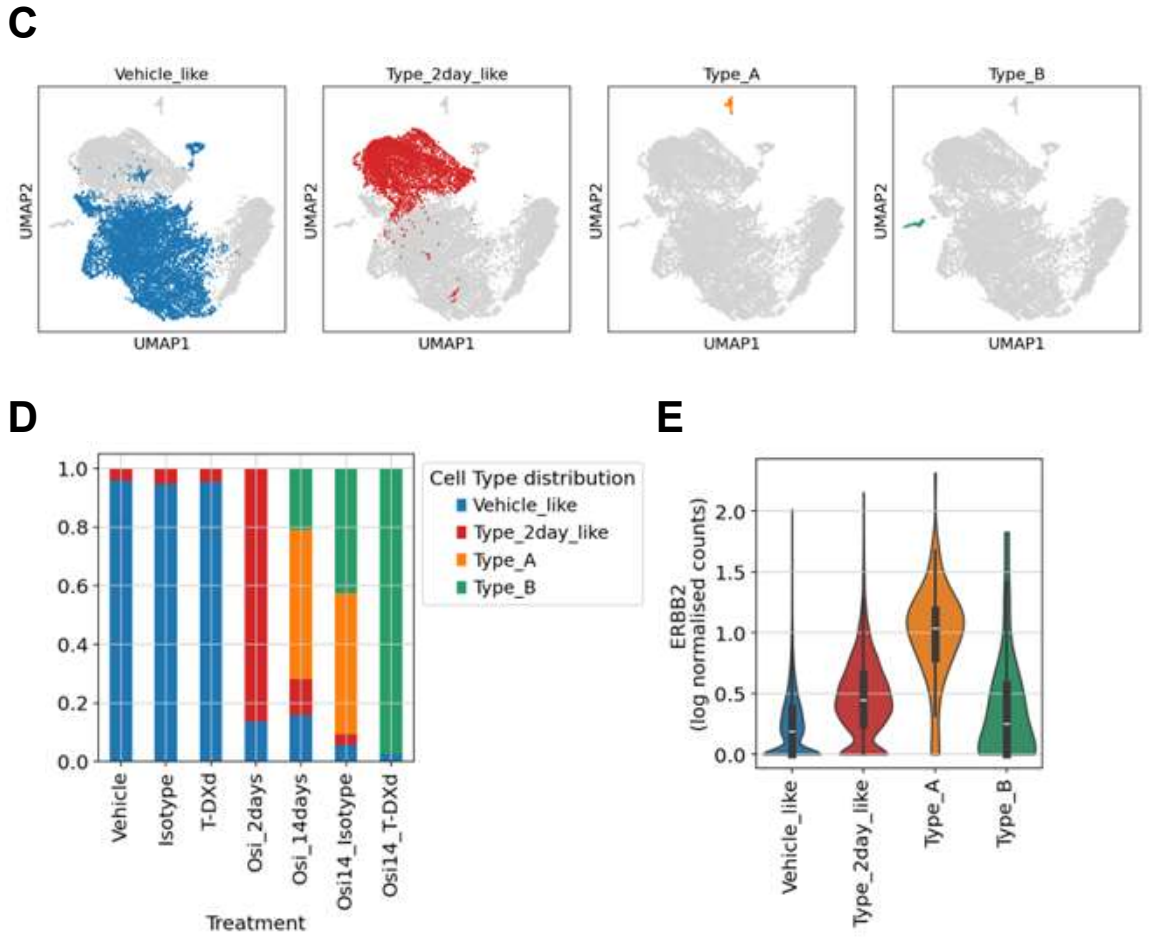
C
Vehicle_like
Type_2day_like
Type_A
Type_B
UMAP1
UMAP2
D
Cell Type distribution
Vehicle_like
Type_2day_like
Type_A
Type_B
Treatment
E
ERBB2 (log normalised counts)

Figure 4.- Combination of osimertinib and T-DXd selectively eliminates a population of osimertinib persister cells with higher levels of HER2 *in vivo*. A. *In vivo* efficacy with indicated treatments in the PC9 model. **B, C.** UMAP visualisation of cells coloured according to treatment (**B**) and cell type (**C**). Osimertinib persister population is transcriptionally heterogenous and different to acute treatment. **D.** Type A cells are specifically ablated by the combination treatment. **E.** Type A depleted cells show higher transcriptional levels of ERBB2.

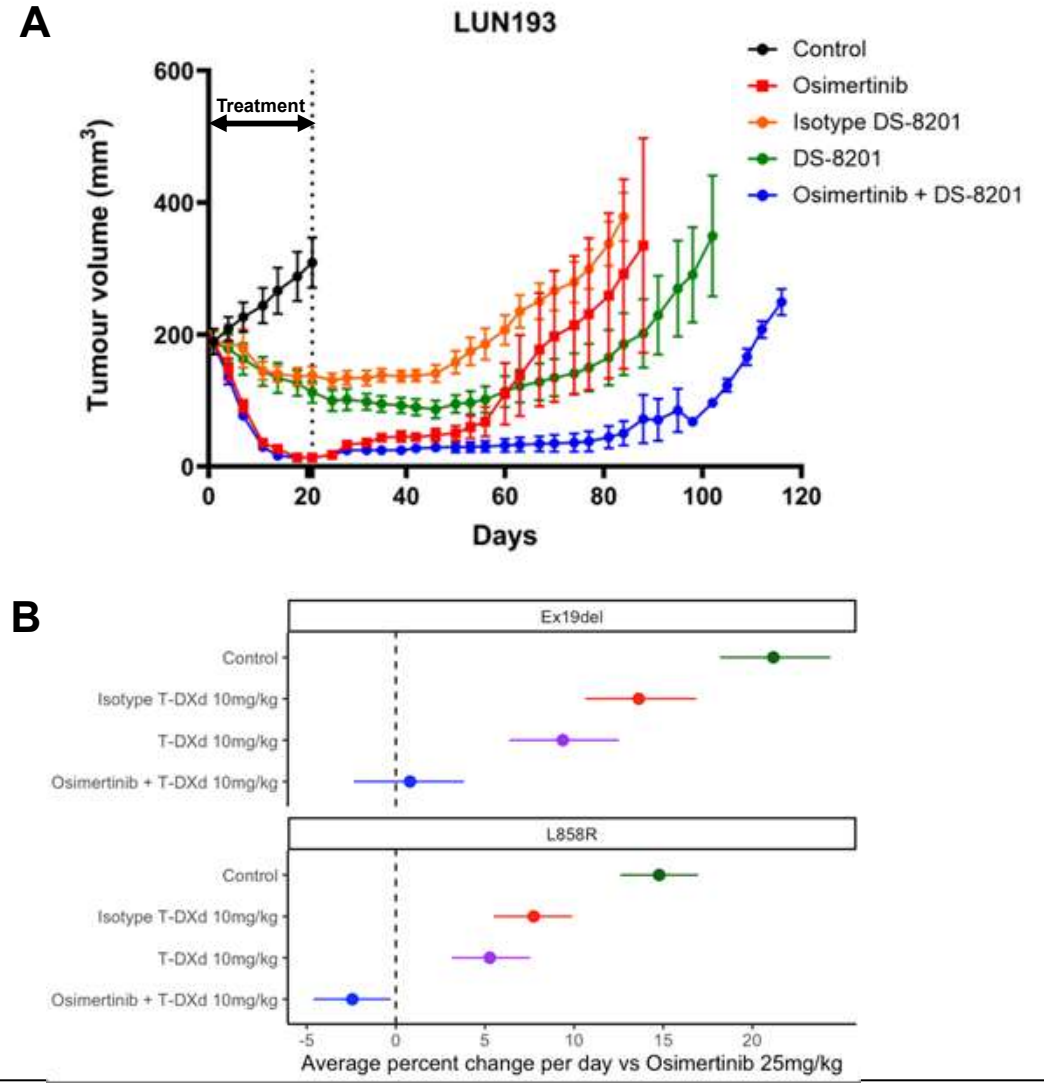
A
LUN193
Treatment
Tumour volume (mm³)
Days
Control
Osimertinib
Isotype DS-8201
DS-8201
Osimertinib + DS-8201
B
Ex19del
L858R
Control
Isotype T-DXd 10mg/kg
T-DXd 10mg/kg
Osimertinib + T-DXd 10mg/kg
Average percent change per day vs Osimertinib 25mg/kg

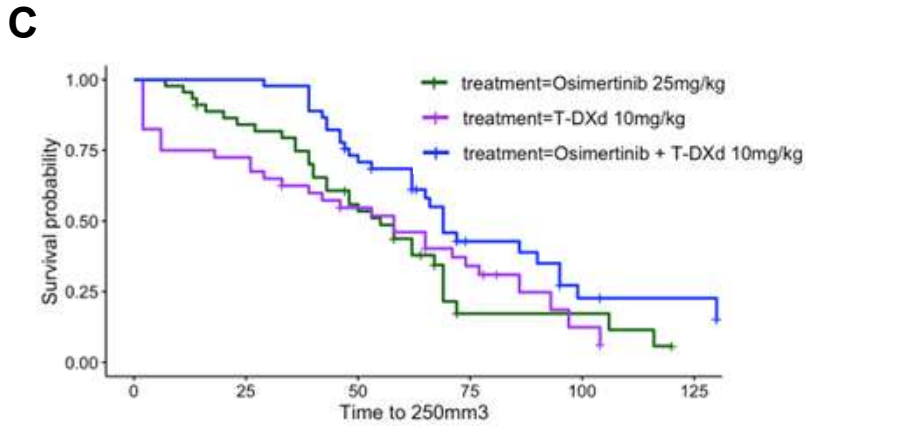
C
treatment=Osimertinib 25mg/kg
treatment=T-DXd 10mg/kg
treatment=Osimertinib + T-DXd 10mg/kg
Survival probability
Time to 250mm3

Fig. 5 Combining Osimertinib and T-DXd gives improved efficacy over Osimertinib monotherapy in a subset of 9 (3Ex19 del/6 L858R) EGFRm PDX models *in vivo*. A *In vivo* efficacy with the indicated treatments in the LUN193 PDX model. Antibody therapy was administered on Day 1, while osimertinib treatment continued daily for 21 days, after which there was a drug-free regrowth phase. **B** Comparison of average percent change per day over day 21 versus osimertinib monotherapy. **C** Survival probability, as determined by time to tumours reaching 250 mm², across 9 EGFRm PDX models for the indicated treatment groups.

Conclusions

- Osimertinib treatment leads to increased expression of HER2 at the mRNA and protein level, translating to enhanced levels at the cell surface.
- scRNAseq of PC9 xenografts shows a subset of osimertinib residual cells have high HER2 expression, and this cell population is eliminated by co-treatment with T-DXd.
- The T-DXd/osimertinib combination leads to improved efficacy over osimertinib monotherapy in 4/9 first-line EGFRm PDX models tested.
- Our preclinical data show the potential of a new combination treatment strategy with T-DXd to be used in the first line setting to enhance osimertinib efficacy.

References

1 - Planchard et al., *N Engl J Med 389*(21): 1935-1948, 2023.
2- Leonetti et al., *Br J Cancer*. 121(9):725-737., 2019.
3- Martin et al., Crit Rev Oncol Hematol., 198:104355. 2024

Acknowledgements

We thank PILs, formulation and cell provision teams for their contributions to this study.
This study is sponsored by AstraZeneca. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201).

Supported by AstraZeneca

Presented at the WCLC 2025 Congress, Barcelona (Spain), September 8, 2025