581P

Trastuzumab deruxtecan in Western patients with HER2-positive advanced gastric or gastroesophageal junction cancer who progressed on or after a trastuzumab-containing regimen: final analysis of DESTINY-Gastric02, a single-arm, phase 2 trial

Geoffrey Ku,[1] Elizabeth Smyth,[2] Filippo Pietrantonio,[3] Ian Chau,[4] Haeseong Park,[5] Katia B. Bencardino,[6] Sara Lonardi,[7] Zev A. Wainberg,[8] Jaffer Ajani,[9] Yelena Janjigian,[1] Fabricio Souza,[10] Lori Jukofsky,[10] Amy Qin,[10] Eddie Hom,[10] Eric Van Cutsem[11]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Oxford National Institute for Health and Care Research Biomedical Research Centre, Churchill Hospital, Oxford, United Kingdom; [3]Fondazione IRCCS Istituto Nazionale dei Tumori, Milan, Italy; [4]The Royal Marsden Hospital, London, United Kingdom; [5]Siteman Cancer Center, Washington University, St. Louis, MO, USA; [6]ASST Grande Ospedale Metropolitano Niguarda, Milan, Italy; [7]Veneto Institute of Oncology IOV-IRCCS, Padua, Italy; [8]UCLA Department of Medicine-Hematology/Oncology, Los Angeles, CA, USA; [9]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [10]Daiichi Sankyo, Basking Ridge, NJ, USA; [11]University Hospitals Gasthuisberg & Leuven & KULeuven, Leuven, Belgium

Objective

- To report final analysis efficacy and safety results with an additional 2.5 years of follow-up from DESTINY-Gastric02, a phase 2 single-arm trial of trastuzumab deruxtecan (T-DXd) in patients from the United States and Europe with human epidermal growth factor receptor 2–positive (HER2+) unresectable/metastatic gastric cancer (GC) or gastroesophageal junction adenocarcinoma (GEJA) who had progressed on or after a first-line trastuzumab-containing regimen

Conclusions

- Final analysis results were consistent with results from the primary and updated analyses, supporting the efficacy of single-agent T-DXd 6.4 mg/kg in Western patients with advanced GC or GEJA in the second-line setting
 - The confirmed objective response rate (cORR) by independent central review (ICR) increased to 43.0%, and the median duration of response (DoR) by ICR increased to 8.3 months
 - Median progression-free survival (PFS; 5.6 months) and overall survival (OS; 12.1 months) remained unchanged
- The safety profile of T-DXd was consistent with previous studies. No new safety signals were observed with longer follow-up

Plain Language Summary

Why did we perform this research?
Human epidermal growth factor receptor 2 (HER2), a protein on the surface of some cancer cells, is found in approximately 5% to 17% of patients with gastric cancer.[1-4] Trastuzumab deruxtecan (T-DXd) is an anticancer therapy that targets HER2 and is approved to treat patients with metastatic gastric or gastroesophageal junction (GEJ) cancer that is HER2-positive (HER2+).[5-7] DESTINY-Gastric02 was a clinical trial that evaluated the treatment benefit and safety of T-DXd in patients from with HER2+ (immunohistochemistry 3+ or 2+/in situ hybridization–positive) metastatic gastric or GEJ cancer. The primary and updated analyses of DESTINY-Gastric02 demonstrated the clinically meaningful activity of T-DXd as second-line therapy in Western patients with HER2+ advanced gastric or GEJ cancer.[7] Efficacy and safety results from the final analysis of the DESTINY-Gastric02 study are reported here.

How did we perform this research?
DESTINY-Gastric02 was a multicenter, open-label, single-arm phase 2 study conducted in the United States and Europe. Patients were enrolled if they had HER2+ gastric or GEJ cancer that had progressed on or after a treatment regimen that included trastuzumab. All enrolled patients received T-DXd 6.4 mg/kg every 3 weeks. The primary endpoint was confirmed objective response rate (cORR) by independent central review (ICR), and secondary endpoints included progression-free survival (PFS) by ICR, overall survival (OS), duration of response (DoR), and safety.

What were the findings of this research?
Results from this study were consistent with earlier efficacy and safety assessments of DESTINY-Gastric02. The cORR by ICR increased to 43.0% and the median DoR by ICR increased to 8.3 months in patients receiving T-DXd; the median PFS (5.6 months) and OS (12.1 months) remained unchanged.

What are the implications of this research?
These findings support the efficacy of T-DXd 6.4 mg/kg as a single agent for Western patients with advanced gastric or GEJ cancer in the second-line setting, with no new safety signals observed with longer follow-up.

Where can I access more information?
To learn more about the DESTINY-Gastric02 study, you can visit: https://clinicaltrials.gov/study/NCT04014075

References

1. Uzunparmak B et al. *Ann Oncol*. 2023;34:1035-1046.
2. Van Cutsem E et al. *Gastric Cancer*. 2015;18:476-484.
3. Subasinghe D et al. *J Int Med Res*. 2023;51:1-16.
4. Yang Y et al. *Acta Biochim Biophys Sin (Shanghai)*. 2021;53(5):547-557.
5. ENHERTU (fam-trastuzumab deruxtecan-nxki) [prescribing information]. Daiichi Sankyo, Inc.; 2026.
6. Shitara K et al. *N Engl J Med*. 2020;382:2419-2430.
7. Van Cutsem E et al. *Lancet Oncol*. 2023;24:744-756.

Poster

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials.
Alternatively, please click on the link below:
https://bit.ly/DSESMOGI2026

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from the European Society of Medical Oncology Gastrointestinal Cancers Congress and the authors of this poster.

This study is sponsored by Daiichi Sankyo. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201).

Poster presented at European Society of Medical Oncology Gastrointestinal Cancers Congress 2026; July 1-4, 2026; Munich, Germany, by Eric Van Cutsem.

Corresponding author email address: Eric.VanCutsem@kuleuven.be

Introduction

- An estimated 5% to 17% of patients with GC have HER2+ (immunohistochemistry [IHC] 3+ or IHC 2+/in situ hybridization–positive [ISH+]) disease[1-4]
- T-DXd, a HER2-directed antibody-drug conjugate, is approved in many countries worldwide for the treatment of adult patients with locally advanced or metastatic HER2+ GC/GEJA who have received a prior trastuzumab-based regimen based on results from the DESTINY-Gastric01, DESTINY-Gastric02, and/or DESTINY-Gastric04 trials[5-11]
- In DESTINY-Gastric02 (NCT04014075), T-DXd showed clinically meaningful activity as a second-line therapy in Western patients with HER2+ (IHC 3+ or IHC 2+/ISH+) advanced GC or GEJA at the primary analysis (data cutoff [DCO], April 9, 2021) and updated analysis (DCO, November 8, 2021)[7,8]
 - At the primary DCO, the cORR by ICR was 38.0% (95% CI, 27.3%-49.6%) and the median DoR by ICR was 8.1 months (95% CI, 4.1 months-not estimable [NE])
 - At the updated DCO, the cORR by ICR was 41.8% (95% CI, 30.8%-53.4%), and the median DoR by ICR was 8.1 months (95% CI, 5.9 months-NE)
- In the phase 3 DESTINY-Gastric04 trial (NCT04704934), second-line treatment with T-DXd resulted in a statistically significant and clinically meaningful improvement in OS compared with ramucirumab + paclitaxel in patients with HER2+ unresectable or metastatic GC/GEJA[9]
- Efficacy and safety results from the final analysis of DESTINY-Gastric02 (DCO, April 25, 2024) are reported here

Methods

- DESTINY-Gastric02 was a multicenter, open-label, single-arm phase 2 trial conducted in the United States and Europe (**Figure 1**)
- Patients with HER2+ unresectable or metastatic GC or GEJA who had progressed on or after a first-line trastuzumab-containing regimen (N = 79) received T-DXd 6.4 mg/kg every 3 weeks (Q3W)
- The primary endpoint was cORR by ICR
- Secondary endpoints included PFS by ICR, OS, DoR, safety, and patient-reported outcomes

Figure 1. DESTINY-Gastric02 study design

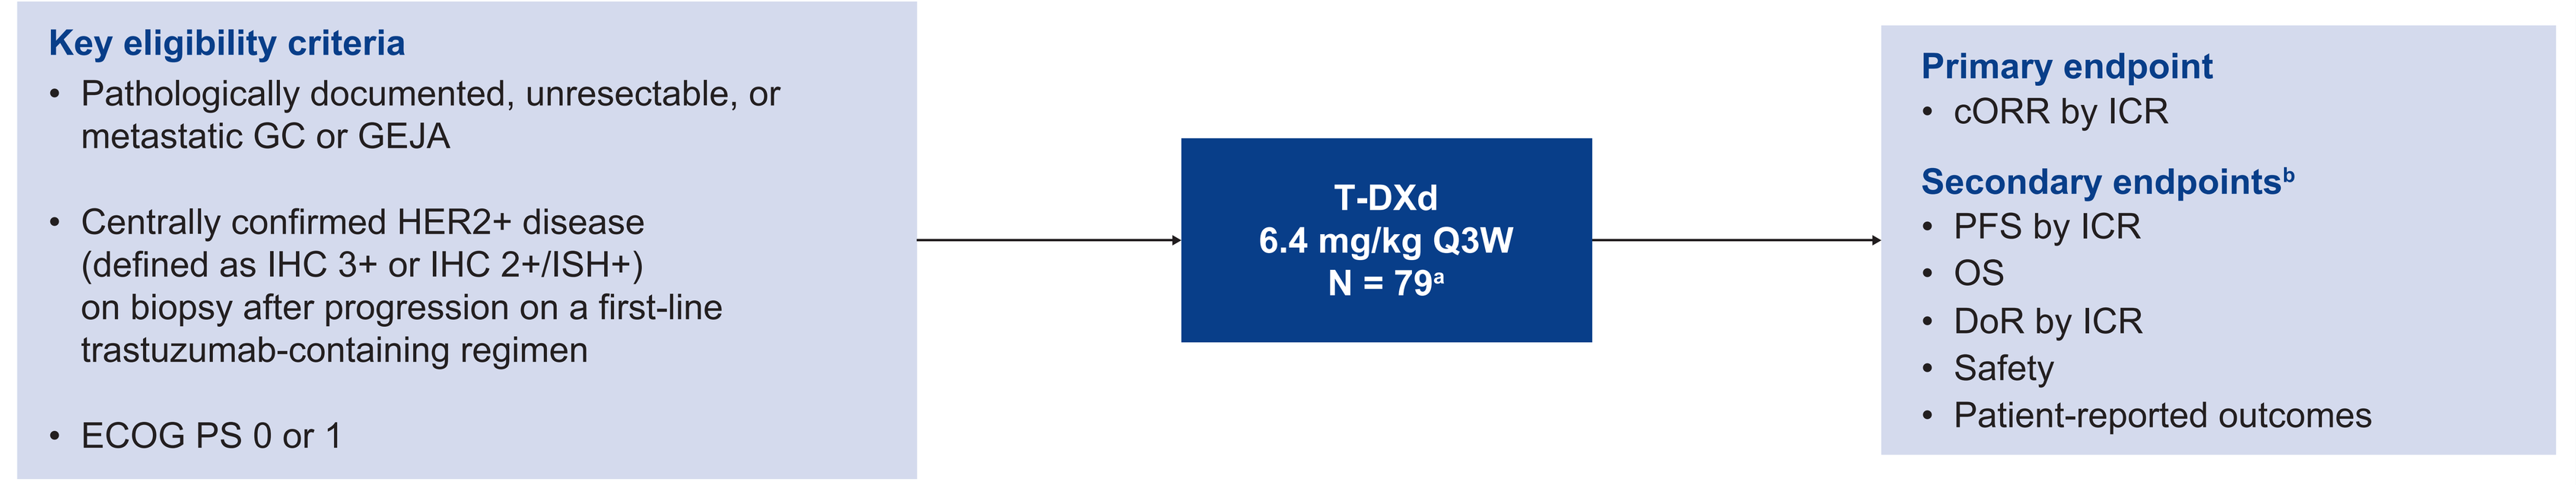

Key eligibility criteria
• Pathologically documented, unresectable, or metastatic GC or GEJA
• Centrally confirmed HER2+ disease (defined as IHC 3+ or IHC 2+/ISH+) on biopsy after progression on a first-line trastuzumab-containing regimen
• ECOG PS 0 or 1
T-DXd
6.4 mg/kg Q3W
N = 79a
Primary endpoint
• cORR by ICR
Secondary endpointsb
• PFS by ICR
• OS
• DoR by ICR
• Safety
• Patient-reported outcomes

[a]Enrollment of 80 patients was planned; actual enrollment was 79 patients. [b]Other secondary endpoints were ORR, PFS, and DoR by investigator assessment; pharmacokinetics; and anti-drug antibodies.

Results

Patients

- Overall, 79 patients received T-DXd
- At the final analysis DCO (April 25, 2024), the median duration of follow-up was 10.2 months (range, 0.7-48.8 months)
- Demographics and baseline characteristics are shown in **Table 1**
- At the DCO, no patients remained on treatment
 - The most common reasons for discontinuation were progressive disease (67.1%) and adverse events (19.0%)

Table 1. Patient demographics

Baseline Characteristics[a]	T-DXd 6.4 mg/kg N = 79
Age, median (range), years	60.0 (20-77)
Male	57 (72.2)
Race	
White	69 (87.3)
Black or African American	1 (1.3)
Asian	4 (5.1)
American Indian or Alaskan Native	0
Native Hawaiian or Pacific Islander	1 (1.3)
Other	3 (3.8)
Missing	1 (1.3)
Country of enrollment	
Belgium	4 (5.1)
Spain	4 (5.1)
Great Britain	17 (21.5)
Italy	20 (25.3)
United States	34 (43.0)
HER2 status by central review	
IHC 3+	68 (86.1)
IHC 2+	10 (12.7)
ISH+	10 (12.7)
ISH–	0
Not evaluable	1 (1.3)

[a]Data are reported as n (%) unless noted otherwise.

Efficacy

- Efficacy outcomes were consistent with the primary and updated analyses
 - cORR by ICR was 43.0% (95% CI, 31.9%-54.7%), with 4 complete responses (5.1%) and 30 partial responses (38.0%) (**Table 2**)
 - Best percentage change of tumor size from baseline by ICR is shown in **Figure 2**
 - Median PFS by ICR was 5.6 months (95% CI, 4.2-8.3 months) (**Figure 3**)
 - Median OS was 12.1 months (95% CI, 9.4-18.5 months) (**Figure 4**)
 - Median DoR by ICR was 8.3 months (95% CI, 5.9 months-NE)

Table 2. Efficacy endpoints

Response Assessment by ICR	T-DXd 6.4 mg/kg N = 79
cORR,[a] % (95% CI)	43.0 (31.9-54.7)
Confirmed best overall response, n (%)	
Complete response	4 (5.1)
Partial response	30 (38.0)
Stable disease	30 (38.0)
Progressive disease	13 (16.5)
Not evaluable	2 (2.5)
DoR,[b] median (95% CI), months	8.3 (5.9-NE)
Time to response, median (95% CI), months	1.4 (1.4-2.7)

[a]Primary endpoint. [b]Secondary endpoint analysis based on responders (n = 34); 18 patients were censored (reasons: initiating new anticancer therapy, event after missing 2 consecutive assessments, and adequate tumor assessment no longer available).

Figure 2. Percentage change of tumor size from baseline

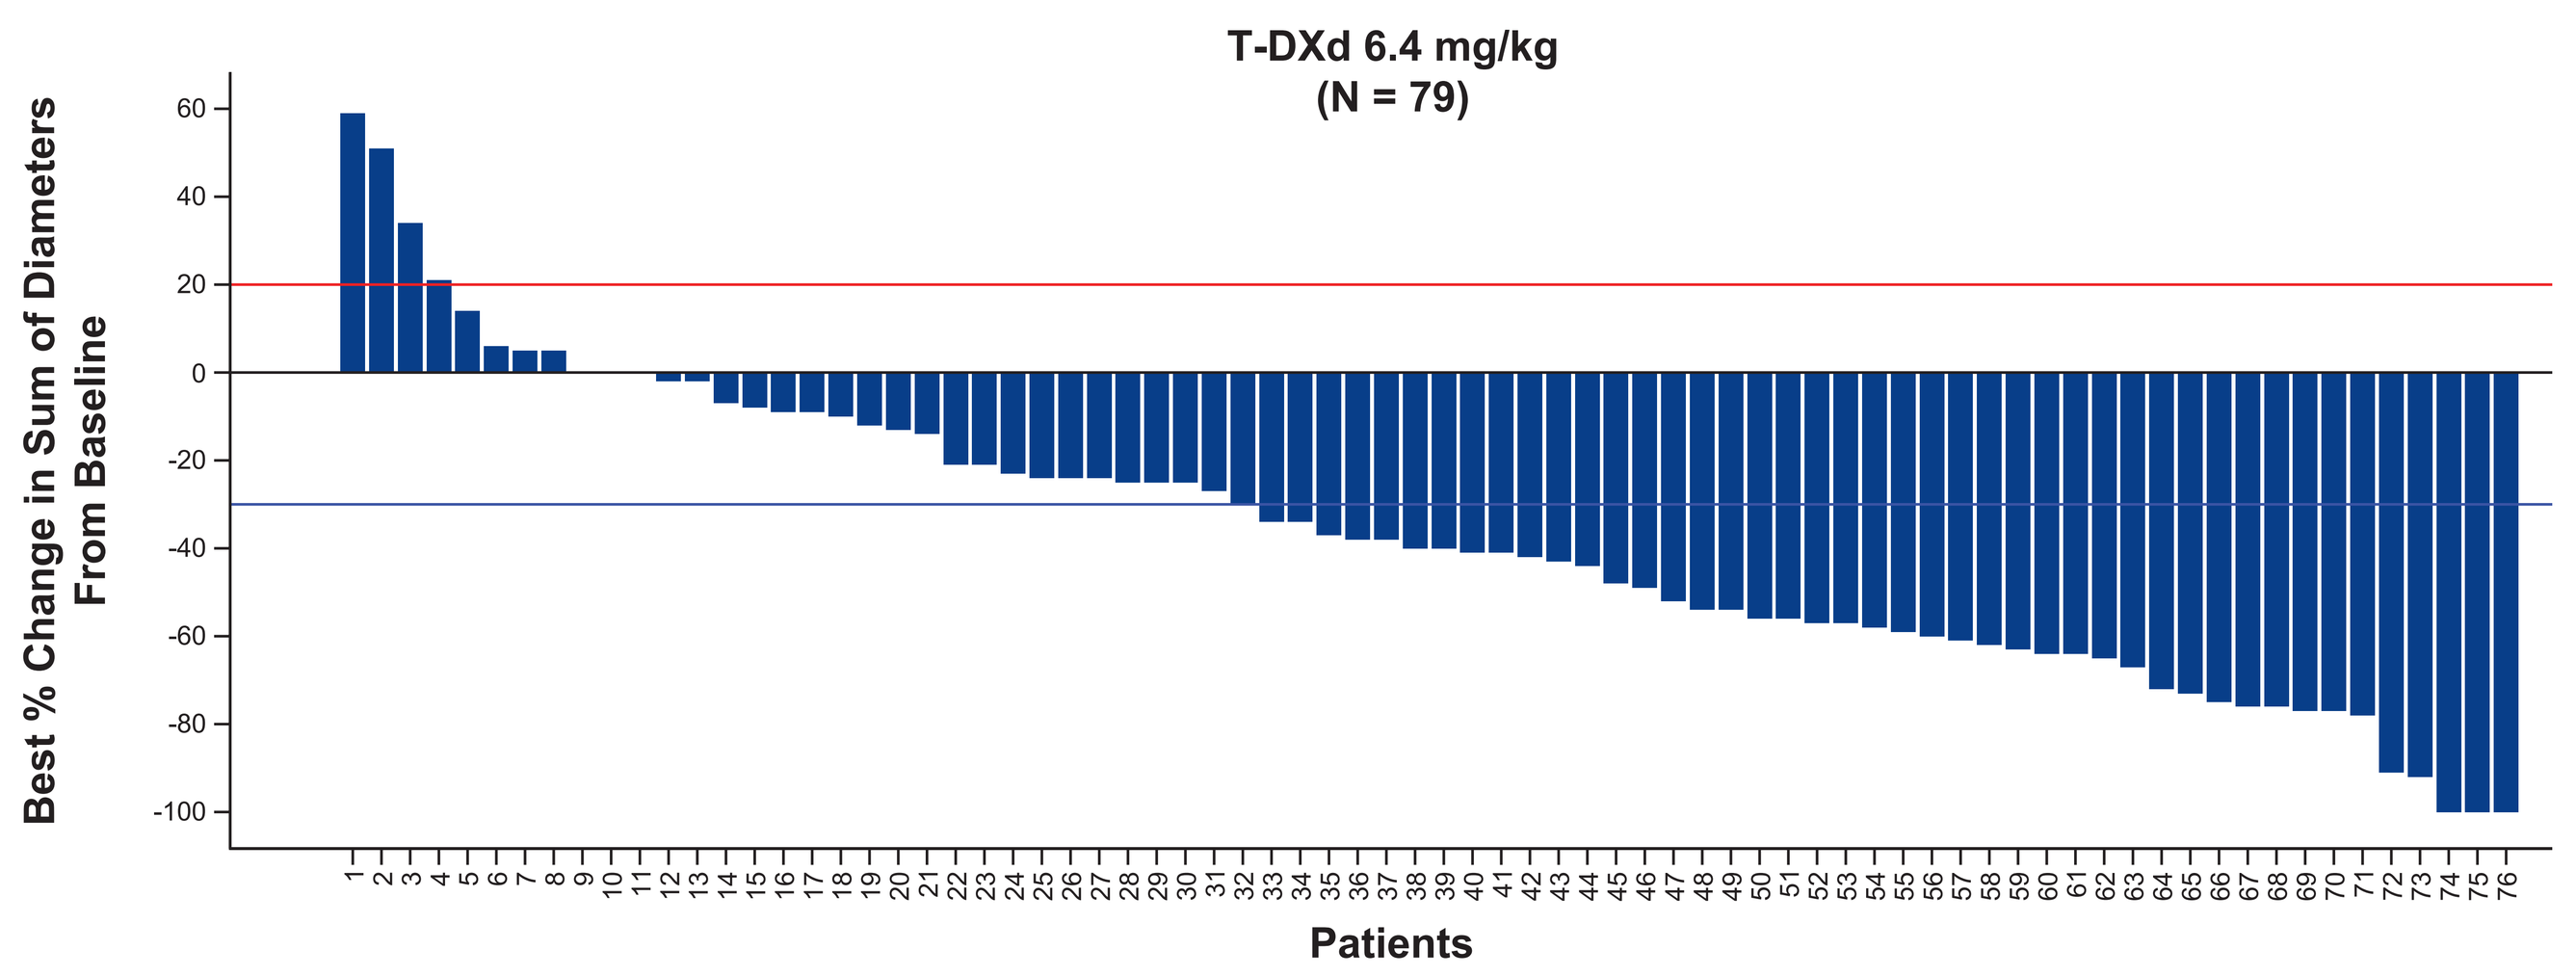

T-DXd 6.4 mg/kg
(N = 79)
Best % Change in Sum of Diameters From Baseline
Patients

Baseline was defined as the last measurement taken before the randomization/registration date. For each patient, the best (minimum) percentage change from baseline in the sum of diameters for all target lesions is represented by a vertical line. Only patients with measurable disease at baseline and at least 1 post-baseline value are included in the waterfall graph.

The red reference line indicates a +20% disease progression and the blue line indicates a -30% of partial response from baseline.

Figure 3. PFS probability by ICR in the full analysis set

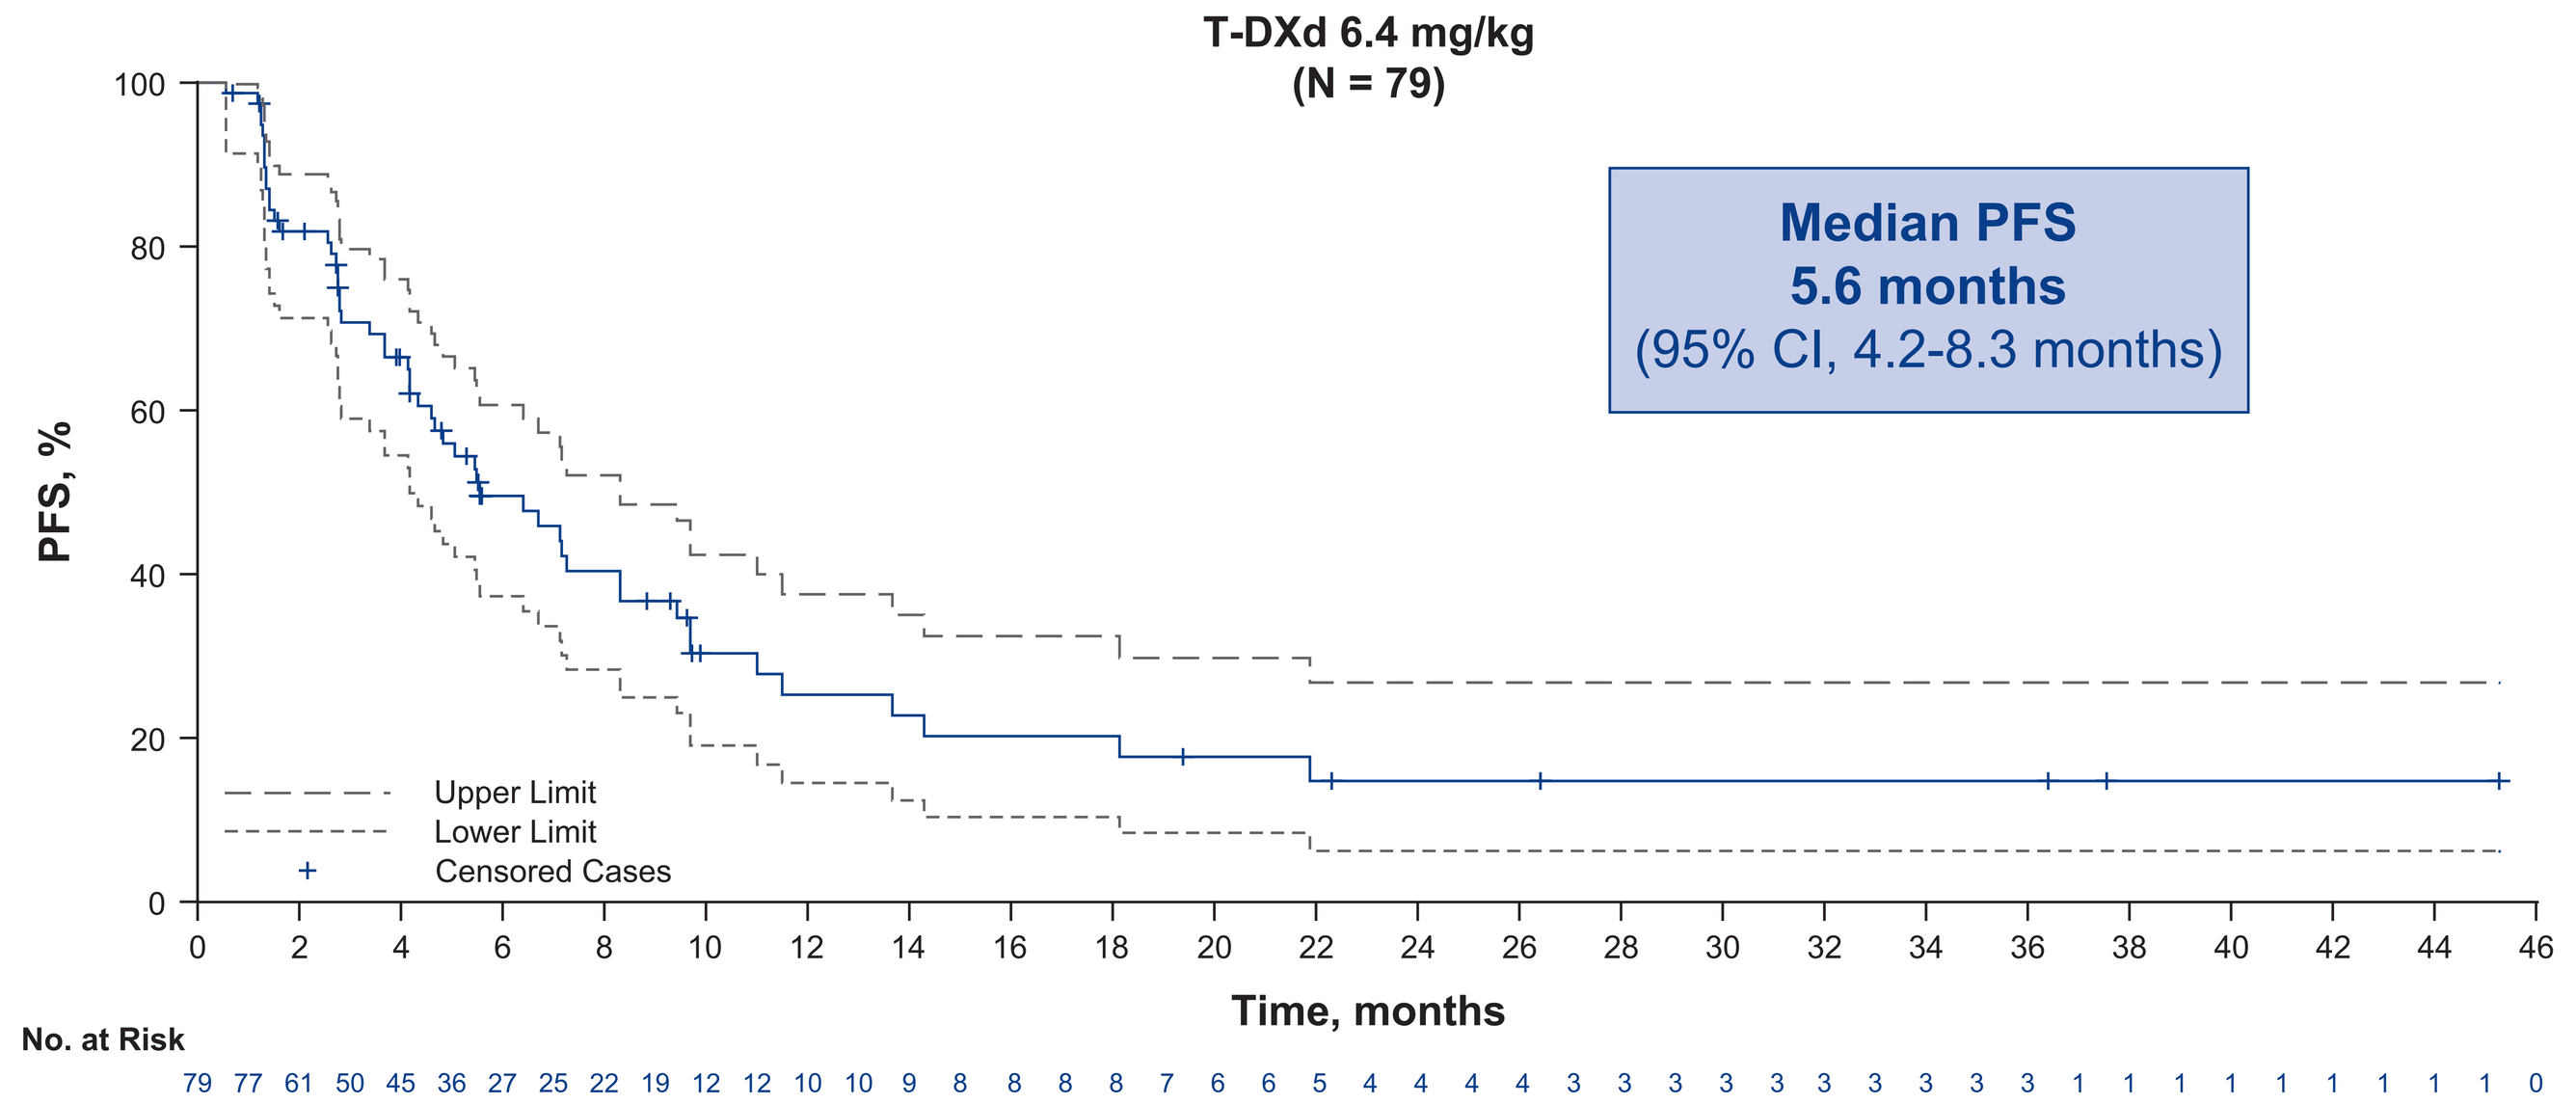

T-DXd 6.4 mg/kg
(N = 79)
Median PFS
5.6 months
(95% CI, 4.2-8.3 months)
PFS, %
Upper Limit
Lower Limit
Censored Cases
Time, months
No. at Risk
79 77 61 50 45 36 27 25 22 19 12 12 10 10 9 8 8 8 8 7 6 6 5 4 4 4 4 3 3 3 3 3 3 3 3 3 3 1 1 1 1 1 1 1 1 1 0

Analysis conducted in the full analysis set based on 52 events (45 progressive disease, 7 deaths).

Figure 4. OS probability in the full analysis set

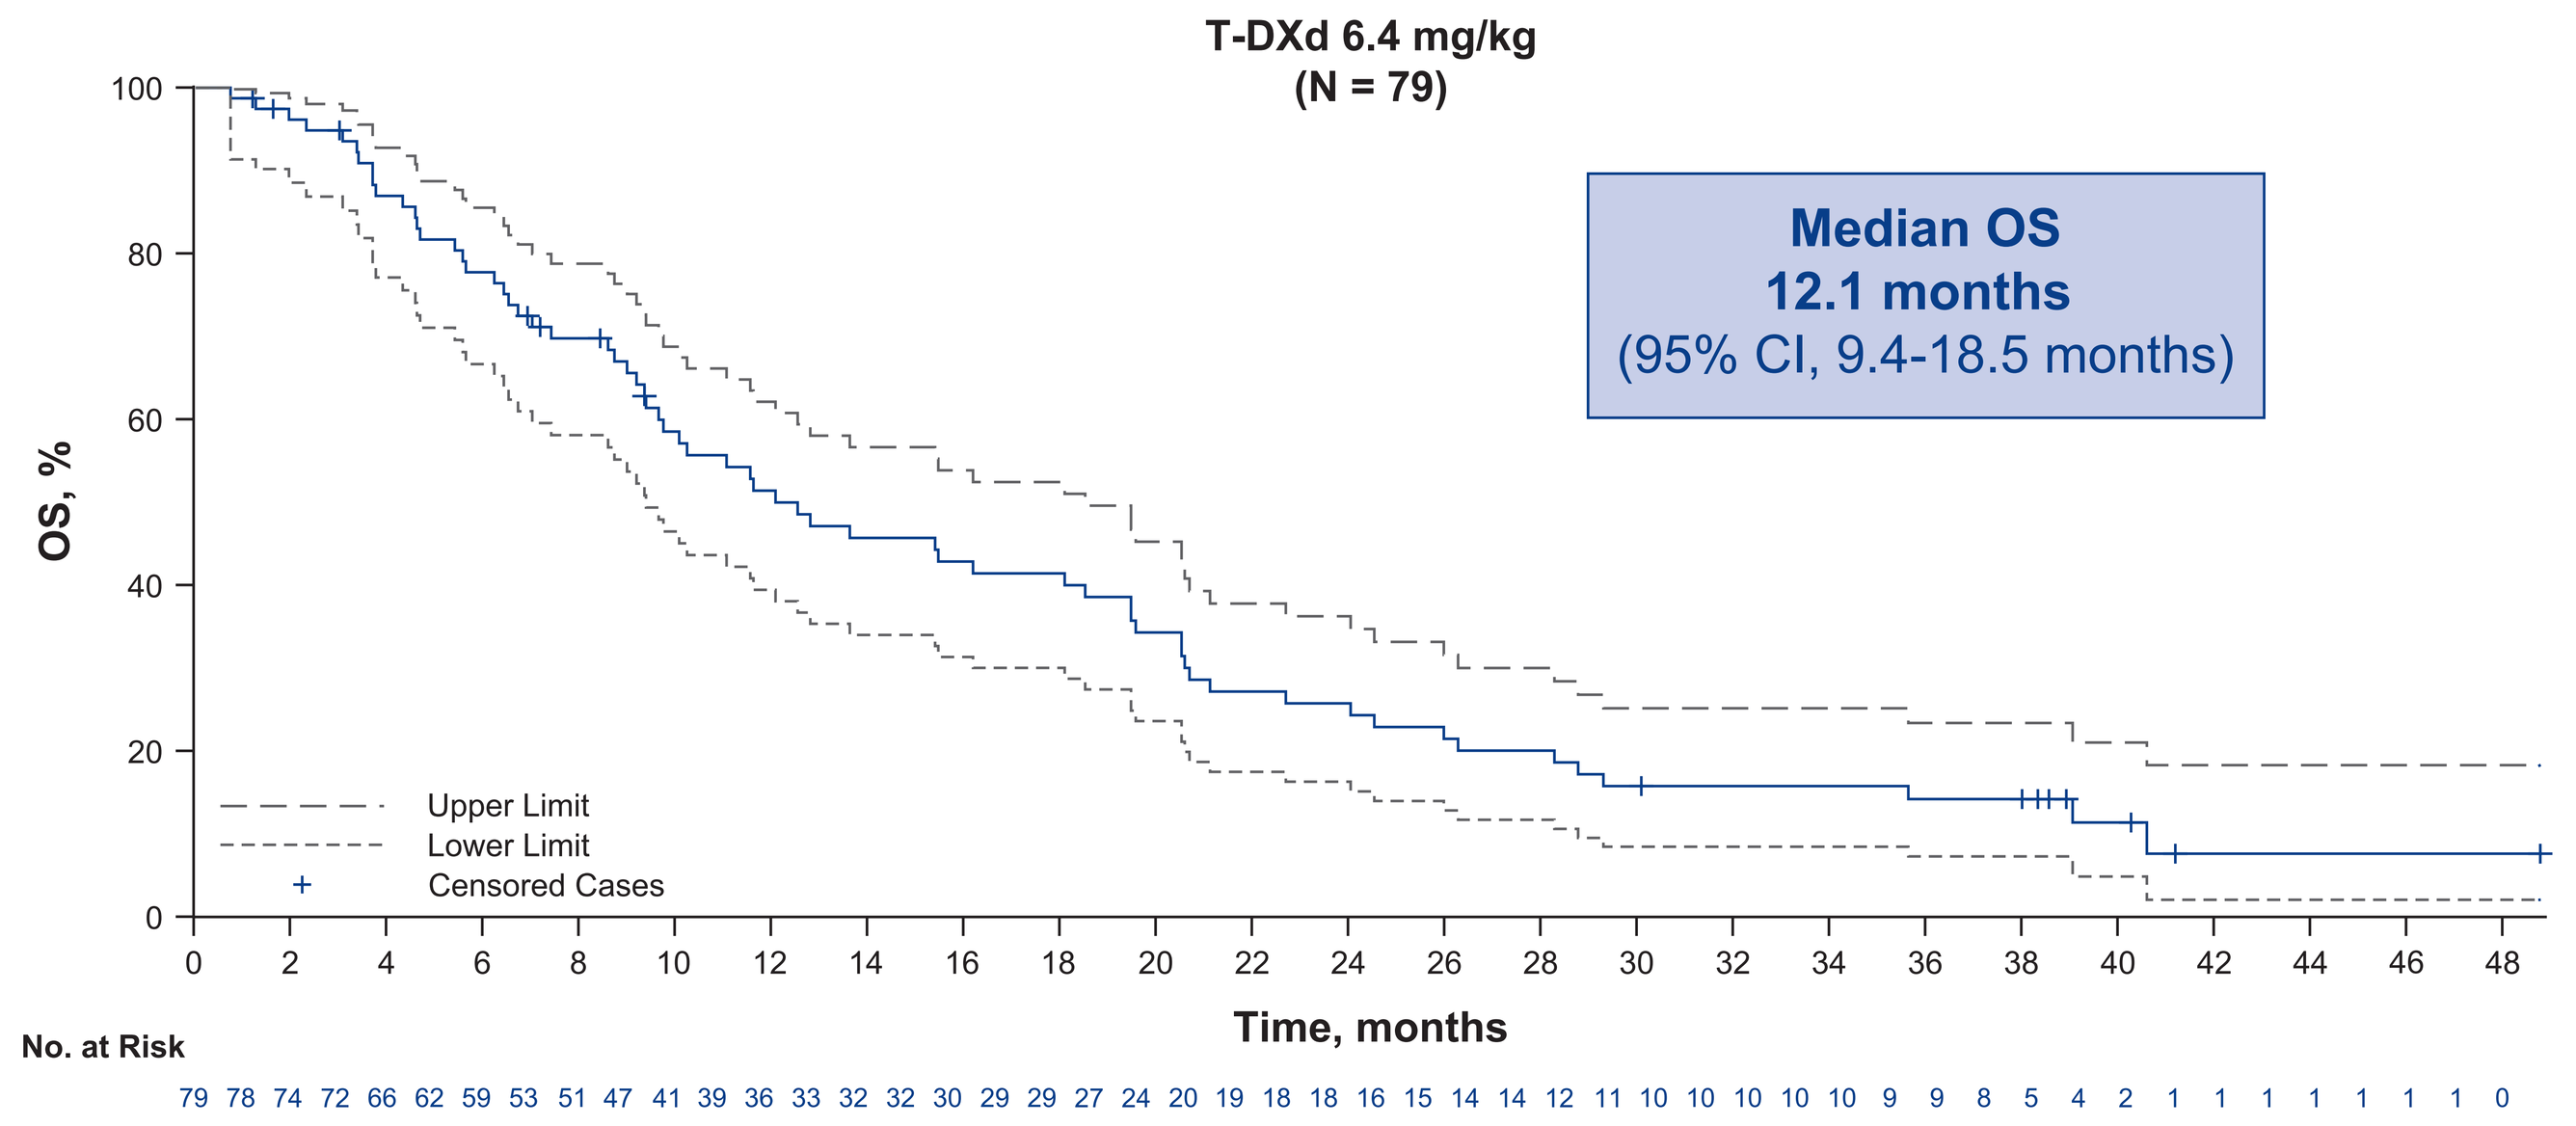

T-DXd 6.4 mg/kg
(N = 79)
Median OS
12.1 months
(95% CI, 9.4-18.5 months)
OS, %
Upper Limit
Lower Limit
Censored Cases
Time, months
No. at Risk
79 78 74 72 66 62 59 53 51 47 41 39 36 33 32 32 30 29 29 27 24 20 19 18 18 16 15 14 14 12 11 10 10 10 10 10 9 9 8 5 4 2 1 1 1 1 1 1 1 0

Analysis conducted in the full analysis set.

Safety

- Median treatment duration was 4.3 months (range, 0.7-39.6 months) and the median relative dose intensity was 96.5% (range, 60.3%-104.1%)
- Rates of treatment-emergent adverse events (TEAEs) were consistent with rates observed in previous analyses[7] (**Table 3**)
- A total of 19 patients (24.1%) had their dose reduced
- Adjudicated, drug-related interstitial lung disease (ILD)/pneumonitis occurred in 11 patients (13.9%; 3 grade 1 events; 6 grade 2 events; and 2 grade 5 events) (**Table 3**)
 - There were 3 additional cases of adjudicated, drug-related ILD/pneumonitis since the updated analysis, with 1 grade 1 event and 2 grade 2 events; 1 event occurred after the updated DCO, and 2 events occurred before the updated DCO and were pending adjudication
- The most common any-grade TEAEs in ≥10% of patients were nausea (67.1%), vomiting (44.3%), and fatigue (41.8%) (**Table 4**)

Table 3. Overall safety summary and adjudicated, drug-related ILD/pneumonitis

n (%)	T-DXd 6.4 mg/kg N = 79
Any TEAE	79 (100)
Drug-related	**75 (94.9)**
TEAE grade ≥3	46 (58.2)
Drug-related	**25 (31.6)**
Serious TEAE	33 (41.8)
Drug-related	**11 (13.9)**
TEAE associated with study drug discontinuation	17 (21.5)
Drug-related	**12 (15.2)**
TEAE associated with dose interruption	25 (31.6)
Drug-related	**11 (13.9)**
TEAE associated with dose reduction	17 (21.5)
Drug-related	**14 (17.7)**
TEAE associated with an outcome of death	11 (13.9)
Drug-related	**2 (2.5)**
Adjudicated, drug-related ILD/pneumonitis	11 (13.9)
Grade ≥3	**2 (2.5)**

Table 4. Most common any-grade TEAEs in ≥10% of patients by preferred term

TEAEs by Preferred Term, n (%)	T-DXd 6.4 mg/kg N = 79	
	Any Grade	Grade ≥3
Nausea	53 (67.1)	6 (7.6)
Vomiting	35 (44.3)	2 (2.5)
Fatigue	33 (41.8)	3 (3.8)
Diarrhea	32 (40.5)	1 (1.3)
Anemia	31 (39.2)	11 (13.9)
Weight decreased	29 (36.7)	4 (5.1)
Decreased appetite	27 (34.2)	4 (5.1)
Constipation	21 (26.6)	0
Alopecia	19 (24.1)	0
Hypokalemia	15 (19.0)	2 (2.5)
Neutrophil count decreased	15 (19.0)	6 (7.6)
Platelet count decreased	15 (19.0)	2 (2.5)
Abdominal pain	14 (17.7)	2 (2.5)
Aspartate aminotransferase increased	14 (17.7)	1 (1.3)
Asthenia	12 (15.2)	1 (1.3)
Blood alkaline phosphatase increased	10 (12.7)	1 (1.3)
Cough	10 (12.7)	1 (1.3)
Neutropenia	10 (12.7)	5 (6.3)
Pyrexia	10 (12.7)	0
COVID-19	9 (11.4)	3 (3.8)
Pneumonitis	9 (11.4)	1 (1.3)
White blood cell count decreased	9 (11.4)	5 (6.3)
Alanine aminotransferase increased	8 (10.1)	1 (1.3)
Back pain	8 (10.1)	0
Dyspnea	8 (10.1)	1 (1.3)
Epistaxis	8 (10.1)	0
Gastroesophageal reflux disease	8 (10.1)	0
Hypoalbuminemia	8 (10.1)	0

Abbreviations

cORR, confirmed ORR; DCO, data cutoff; DoR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; GC, gastric cancer; GEJA, gastroesophageal junction adenocarcinoma; HER2, human epidermal growth factor receptor 2; ICR, independent central review; IHC, immunohistochemistry; ILD, interstitial lung disease; ISH, in situ hybridization; NE, not estimable; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; Q3W, every 3 weeks; T-DXd, trastuzumab deruxtecan; TEAE, treatment-emergent adverse event.

Acknowledgments

Medical writing support, under the direction of the authors, was provided by Laura Halvorson, PhD, and Andre Wang, PharmD, of ApotheCom, and was funded by Daiichi Sankyo in accordance with Good Publication Practice (GPP) guidelines (http://www.ismpp.org/gpp-2022).

Disclosures

Dr. Van Cutsem reports consulting or advisory roles for AbbVie, Agenus, ALX, Amgen, Arcus Biosciences, Astellas, AstraZeneca, Bayer, BeiGene, Bexon Clinical, BioNTech, Boehringer Ingelheim, Bristol Myers Squibb, Cantargia, Daiichi Sankyo, Debiopharm, ElmediX, Eisai, Fosun, Galapagos, GlaxoSmithKline, Ipsen, ITeos, Lilly, Merck Sharp & Dohme, Merck KGaA, Microbial Machines, Mirati, Novartis, Nordic, Novocure, Pierre Fabre, Pfizer, Sanofi, Seattle Genetics, Servier, Simcere, Takeda, Taiho, and Trishula.

References

1. Uzunparmak B et al. *Ann Oncol*. 2023;34:1035-1046.
2. Van Cutsem E et al. *Gastric Cancer*. 2015;18:476-484.
3. Subasinghe D et al. *J Int Med Res*. 2023;51:1-16.
4. Yang Y et al. *Acta Biochim Biophys Sin (Shanghai)*. 2021;53:547-557.
5. ENHERTU (fam-trastuzumab deruxtecan-nxki) [prescribing information]. Daiichi Sankyo, Inc.; 2026.
6. Shitara K et al. *N Engl J Med*. 2020;382:2419-2430.
7. Van Cutsem E et al. *Lancet Oncol*. 2023;24:744-756.
8. Ku G et al. Presented at: ESMO Congress 2022; September 9-13, 2022; Paris, France.
9. Shitara K et al. *N Engl J Med*. 2025;393:336-348.
10. Daiichi Sankyo, Inc. Press release. January 22, 2026. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202601/20260122_E2.pdf. Accessed May 12, 2026.
11. Daiichi Sankyo, Inc. Press release. March 23, 2026. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202603/20260323_E3.pdf. Accessed May 15, 2026.

DESTINY
Gastric02