453P

The value of experiencing deep partial response in the context of long-term outcomes in patients with human epidermal growth factor receptor 2-positive (HER2+) metastatic breast cancer: A DESTINY-Breast03 exploratory analysis

Erika Hamilton,[1] Javier Cortes,[2] Giuseppe Curigliano,[3] Sara Hurvitz,[4] Seock-Ah Im,[5] Waltraud Pasteiner,[6] Shunsuke Nakatani,[7] Deidre Kile,[8] David Leung,[8] Anton Egorov,[9] Hiroji Iwata[10]

[1]Sarah Cannon Research Institute, Nashville, TN, United States; [2]International Breast Cancer Center, Barcelona, Spain; [3]Istituto Europeo di Oncologia, IRCCS and University of Milan, Milan, Italy; [4]University of Washington and Fred Hutchinson Cancer Center, Seattle, WA, United States; [5]Seoul National University Hospital, Seoul, Republic of Korea; [6]Daiichi Sankyo GmbH, Vienna, Austria; [7]Daiichi Sankyo Co, Ltd., Tokyo, Japan; [8]Daiichi Sankyo, Inc., Basking Ridge, NJ, United States; [9]Daiichi Sankyo Oncology France, Rueil-Malmaison, France; [10]Nagoya City University, Graduate School of Medical Sciences, Nagoya, Japan

Objective

- To explore deep partial response (PR) as a potential new response category and to assess efficacy outcomes across response subgroups, with the aim of identifying an additional subset of patients with HER2+ metastatic breast cancer (mBC) previously treated with trastuzumab and a taxane who may derive sustained treatment benefit with trastuzumab deruxtecan (T-DXd) or trastuzumab emtansine (T-DM1)

Conclusions

- Deep PR was defined as either ≥80% to <100% reduction in the sum of target lesion (TL) diameters from baseline with complete response (CR), non-CR/non-progressive disease (PD), or no disease in non-TLs, or as CR in TLs with non-CR/non-PD in non-TLs
 - The ≥70% to <100% and ≥60% to <100% deep PR thresholds were explored for validation; however, the ≥80% to <100% threshold demonstrated the greatest magnitude of benefit compared with non–deep PR
- Patients with deep PR had a longer duration of treatment and improved long-term progression-free survival (PFS) with T-DXd or T-DM1 compared with patients with non–deep PR
 - Moreover, in the T-DXd arm, treatment duration, PFS, and duration of response (DOR) in patients with deep PR were closer to those observed in patients with confirmed CR
- More time was required to reach confirmed CR and deep PR as the best response to treatment compared with patients experiencing non–deep PR (6.4 months and 6.9 months vs 1.7 months, respectively, in the T-DXd arm)
- The observed overall duration of treatment in patients with deep PR (32.1 months) was substantially longer than the time required to achieve the best response (6.9 months), reflecting favorable tolerability and strong patient adherence to treatment
- Deep PR was associated with favorable PFS outcomes, with 77.6% of patients remaining progression-free at 24 months
- These data provide further rationale for sustained T-DXd treatment strategies to achieve maximum tumor reduction and to improve long-term outcomes

Plain Language Summary

Why did we perform this research?
About 20% of breast cancers have high levels of a protein called human epidermal growth factor receptor 2 (HER2).[1,2] Trastuzumab deruxtecan (T-DXd) is a cancer treatment that targets HER2.[3,4] The DESTINY-Breast03 study compared the benefits and safety of T-DXd with another HER2-targeted treatment, trastuzumab emtansine (T-DM1), in patients with HER2-positive breast cancer that could not be removed by surgery (unresectable) and/or had spread to other parts of the body (metastatic).[5,6] Earlier results from DESTINY-Breast03 showed that T-DXd was a more beneficial treatment for patients than T-DM1.[5,6] This analysis aimed to better understand whether a large reduction in tumor size (deep partial response) represents a unique subset of responders and whether it can help identify more patients who may experience long-lasting benefit from treatment.

How did we perform this research?
In the DESTINY-Breast03 study, 524 patients with HER2-positive unresectable or metastatic breast cancer were randomly given either T-DXd 5.4 mg/kg (261 patients) or T-DM1 3.6 mg/kg (263 patients) intravenously.[5,6] For this analysis, a "deep partial response" was defined as a large shrinkage of tumors (at least 60%, 70%, or 80%) or complete disappearance of some tumors while others remained stable and did not grow. We compared how long patients lived without their cancer getting worse between those with a deep partial response and those with a smaller partial response. We also compared deep partial responses with complete responses. Other treatment outcomes were evaluated across response groups, including how quickly tumors shrunk, how long the response lasted, how long patients stayed on treatment, how long cancer remained under control, and why treatment was stopped.

What were the findings of this research?
In the T-DXd treatment group, patients whose tumors showed deep partial response had treatment benefits similar to those whose tumors completely disappeared. Larger reductions in tumor size were linked to longer-lasting benefit, especially in patients treated with T-DXd, and the greatest benefit was seen when tumors shrank by at least 80%.

What are the implications of this research?
These findings suggest that a large reduction in tumor size (deep partial response) may represent a type of treatment response beyond the traditional categories of partial or complete response and may help identify more patients who can experience long-lasting benefit from treatment.

Where can I access more information?
To learn more about the DESTINY-Breast03 study, you can visit https://clinicaltrials.gov/ct2/show/NCT03529110.

References

1. Slamon DJ et al. *Science*. 1989;244(4905):707-712.
2. Iqbal N et al. *Mol Biol Int*. 2014;2014:852748.
3. Nakada T et al. *Chem Pharm Bull*. 2019;67(3):173-185.
4. Ogitani Y et al. *Clin Cancer Res*. 2016;22(20):5097-5108.
5. Cortés J et al. *N Engl J Med*. 2022;386(12):1143-1154.
6. Hurvitz SA et al. *Lancet*. 2023;401(10371):105-117.

Poster

Supplementary material

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials.

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from the European Society for Medical Oncology and the authors of this poster.

This study is sponsored by Daiichi Sankyo. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201).

Presented at the European Society for Medical Oncology Breast Cancer (ESMO BC) Annual Meeting; May 6-8, 2026; Berlin, Germany, by Dr. Erika Hamilton.

Corresponding author email address: erika.hamilton@scri.com

Introduction

- In a pooled analysis of DESTINY-Breast01, -02, and -03, patients who experienced a CR benefited from prolonged PFS compared with patients who experienced a PR or had stable disease (SD)\PD[1]
 - Median PFS by blinded independent central review (BICR) was not reached (95% CI, 37.9 months-not estimable [NE]) among patients with a CR, 21.8 months (95% CI, 18.1-25.0) among patients with a PR, and 8.4 months (95% CI, 6.6-9.7) among patients with SD/PD
 - Median time to best response was 5.7 months (95% CI, 1.2-37.7) for patients with a CR and 1.9 months (95% CI, 1.1-21.0) for patients with a PR
- In DESTINY-Breast03, T-DXd improved PFS and objective response rate (ORR) versus T-DM1 in patients with HER2+ mBC[2-4]
 - Investigator-assessed ORR in the T-DXd arm was 78.9% (95% CI, 73.5-83.7) at the final analysis data cut-off (DCO; June 27, 2025), with 34 patients (13%) experiencing a CR; the median DOR with T-DXd was 31.8 months (95% CI, 23.1-53.4)[4]
- Response Evaluation Criteria in Solid Tumours (RECIST) may have limitations in confirming CR, particularly due to challenges in TL and non-TL selection in certain anatomical sites (eg, lymph nodes or bones) and other location-specific constraints
- Furthermore, because RECIST defines PR as a ≥30% reduction in tumor size, PR represents a heterogeneous group of responses[5]
- Here, we report results from an exploratory analysis of a new response category—deep PR—conducted to identify an additional subset of patients with previously treated HER2+ mBC who may derive benefit from sustained treatment with T-DXd or T-DM1

Methods

- DESTINY-Breast03 was a randomized, multicenter, open-label, phase 3 trial (**Figure 1**)
- Patients were randomly assigned 1:1 to receive T-DXd 5.4 mg/kg or T-DM1 3.6 mg/kg intravenously every 3 weeks
- The response definitions explored in this analysis include the following:
 - Confirmed CR per RECIST 1.1
 - Confirmed PR per RECIST 1.1 (deep PR + non–deep PR)
 - Deep PR: PR with ≥80% to <100% reduction in the sum of TL diameters from baseline with CR, non-CR/non-PD, or no disease in non-TLs (validated at thresholds of ≥70% to <100% and ≥60% to <100%), or as CR in TLs with non-CR/non-PD in non-TLs
 - Non–deep PR: remaining patients with confirmed PR who did not meet the definitions of deep PR
- Optimal deep PR thresholds were examined by assessing PFS with deep PR versus non–deep PR and CR at each deep PR threshold
- Additionally, efficacy outcomes, including treatment duration, time to best response on treatment, DOR, PFS, and reasons for treatment discontinuation, were evaluated by response subgroups

Figure 1. DESTINY-Breast03 Study Design[2,4]

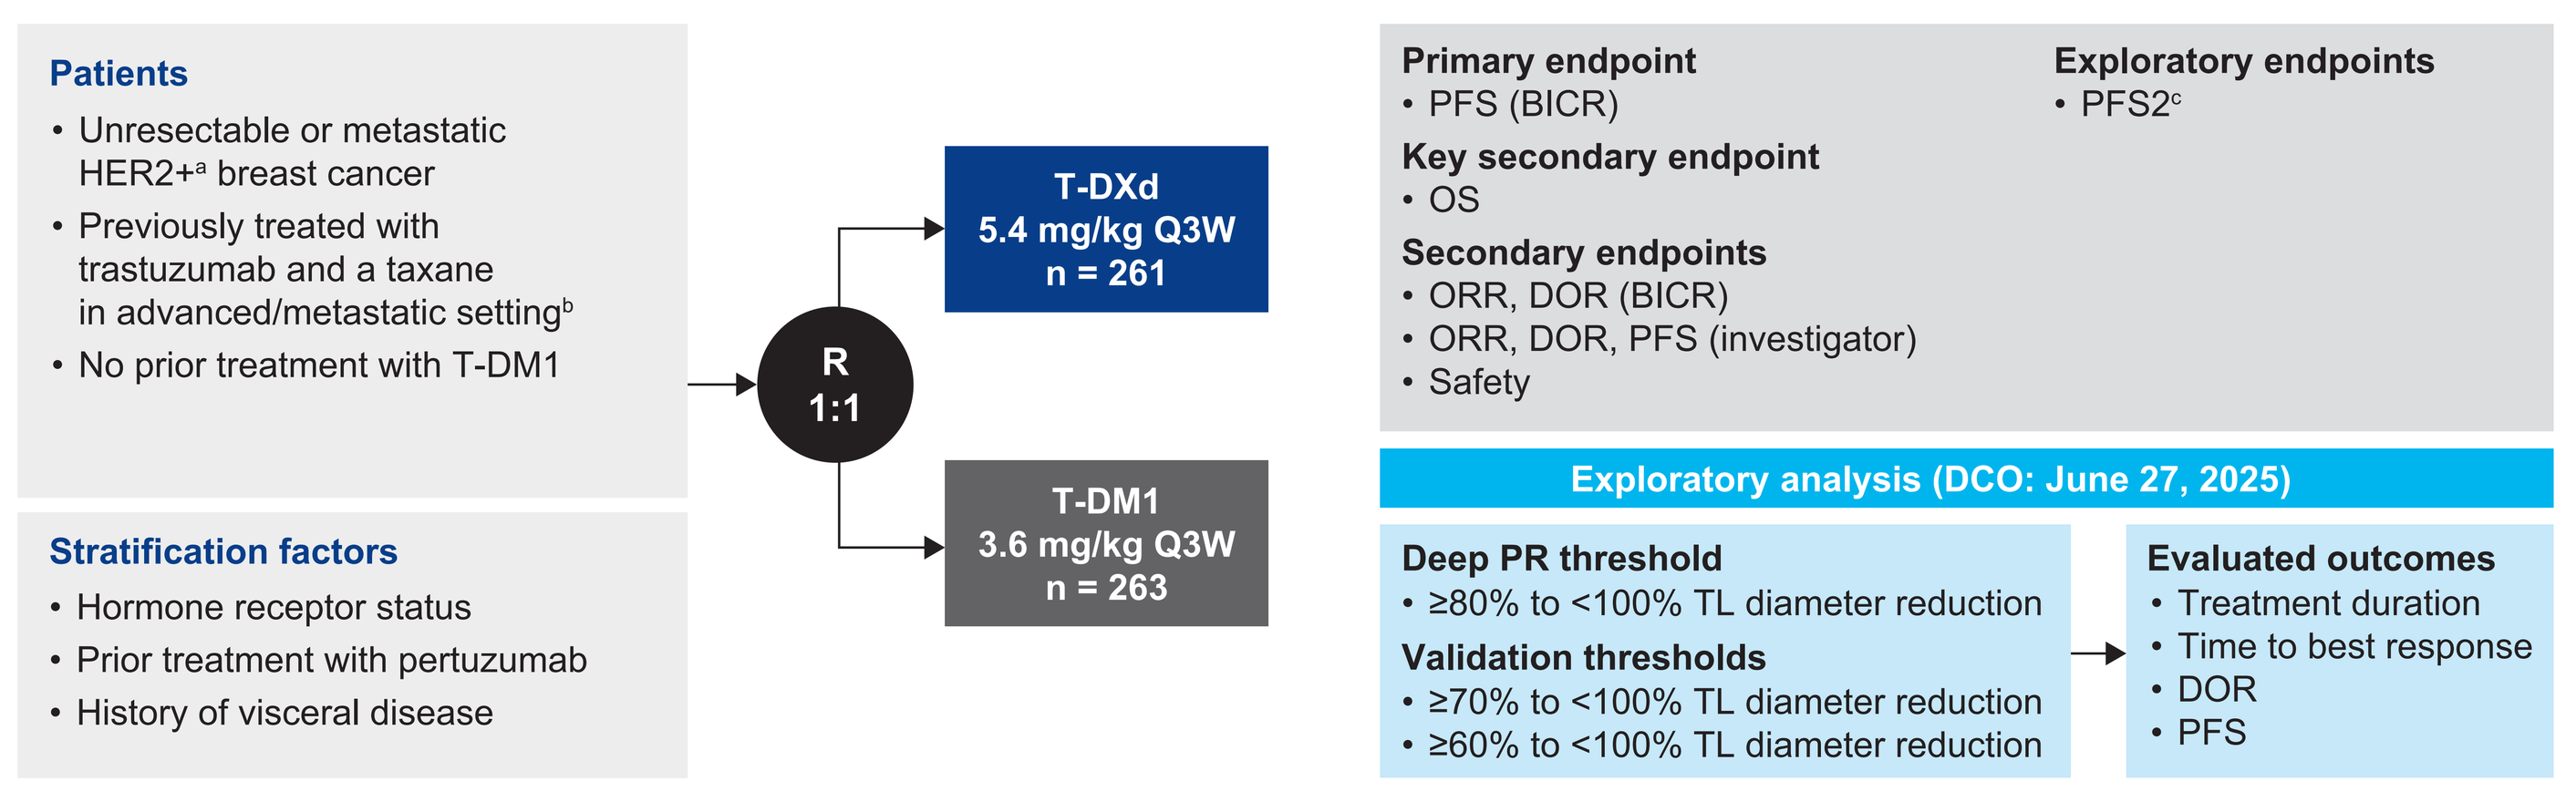

Patients
• Unresectable or metastatic HER2+ᵃ breast cancer
• Previously treated with trastuzumab and a taxane in advanced/metastatic settingᵇ
• No prior treatment with T-DM1
Stratification factors
• Hormone receptor status
• Prior treatment with pertuzumab
• History of visceral disease
R 1:1
T-DXd 5.4 mg/kg Q3W n = 261
T-DM1 3.6 mg/kg Q3W n = 263
Primary endpoint
• PFS (BICR)
Key secondary endpoint
• OS
Secondary endpoints
• ORR, DOR (BICR)
• ORR, DOR, PFS (investigator)
• Safety
Exploratory endpoints
• PFS2ᶜ
Exploratory analysis (DCO: June 27, 2025)
Deep PR threshold
• ≥80% to <100% TL diameter reduction
Validation thresholds
• ≥70% to <100% TL diameter reduction
• ≥60% to <100% TL diameter reduction
Evaluated outcomes
• Treatment duration
• Time to best response
• DOR
• PFS

[a]HER2 IHC 3+ or IHC 2+/ISH+ based on central confirmation.
[b]Progression during or <6 months after completing adjuvant therapy involving trastuzumab and a taxane.
[c]Time from randomization to disease progression on the next line of therapy or death.

Results

Patients

- Overall, 524 patients were randomly assigned to a treatment arm (261 to T-DXd and 263 to T-DM1)
- At final DCO (June 27, 2025), CR as best response to treatment was observed in 34 patients (13.0%) and 13 patients (4.9%) in the T-DXd and T-DM1 arms, respectively.
- PR was observed in 172 (65.9%) and 84 patients (31.9%), respectively
 - Deep PR (≥80% to <100%) was observed in 43 patients (16.5%) in the T-DXd arm and 30 patients (11.4%) in the T-DM1 arm
 - Non–deep PR was observed in 129 (49.4%) and 54 patients (20.5%) in the T-DXd and T-DM1 arms, respectively
- Treatment status of randomly assigned patients is shown in **Table 1**; treatment status at the validation thresholds is reported in **Supplementary Table 1**
- Across response categories and treatment arms, the most common reasons patients discontinued study treatment were PD and adverse events (AEs) (**Table 1; Supplementary Table 2**)
 - Overall, fewer patients in the deep PR group discontinued treatment due to PD compared with the non–deep PR group

Efficacy

- Median treatment duration was longest among patients with CR
- Median treatment duration in patients with deep PR was longer than those with non–deep PR (**Table 2**)
 - In the T-DXd arm, median treatment duration was 32.1 months in the ≥80% to <100% deep PR group and 14.7 months in the non–deep PR group; corresponding treatment durations in the T-DM1 arm were 20.6 months and 9.9 months
 - Treatment durations in the ≥70% to <100% and ≥60% to <100% validation threshold groups are reported in **Supplementary Table 3;** compared to the validation thresholds, the ≥80% to <100% threshold showed the greatest differences in treatment duration between deep PR and non–deep PR response groups
- In the T-DXd arm, response outcomes in patients experiencing deep PR (≥80% to <100%) were generally comparable to those observed in patients with CR (**Table 2**)

PFS

- The ≥80% to <100% deep PR threshold demonstrated the greatest magnitude of benefit compared with non–deep PR (**Table 3; Figure 2**)
 - PFS rates by deep PR threshold (≥80% vs ≥70% vs ≥60% to <100%) are shown in **Figure 3** and **Supplementary Figures 1-3**
- At the ≥80% to <100% threshold, deep PR was associated with sustained responses over time, comparable with CR, particularly in the T-DXd arm

Table 1. Treatment Status and Primary Reasons for Discontinuation From Study Drug by Response Category

n (%)	T-DXd (n = 261[a]) CR (n = 34)	T-DXd Deep PR ≥80% to <100% (n = 43)	T-DXd Non–deep PR (n = 129)	T-DM1 (n = 263[a]) CR (n = 13)	T-DM1 Deep PR ≥80% to <100% (n = 30)	T-DM1 Non–deep PR (n = 54)
Ongoing treatment	10 (29.4)	7 (16.3)	6 (4.7)	1 (7.7)	1 (3.3)	0
Reasons for discontinuation						
PD[b]	6 (25.0)	13 (36.1)	66 (53.7)	1 (8.3)	17 (58.6)	39 (72.2)
AEs	11 (45.8)	15 (41.7)	35 (28.5)	6 (50.0)	3 (10.3)	8 (14.8)
Patient withdrawal	6 (25.0)	5 (13.9)	12 (9.8)	4 (33.3)	3 (10.3)	3 (5.6)
Clinical progression[c]	0	1 (2.8)	3 (2.4)	0	2 (6.9)	4 (7.4)
Death	0	0	3 (2.4)	0	1 (3.4)	0
Physician decision	0	0	1 (0.8)	1 (8.3)	1 (3.4)	0
Lost to follow-up	1 (4.2)	0	0	0	0	0
Protocol deviation	0	0	1 (0.8)	0	0	0
Other	0	2 (5.6)	2 (1.6)	0	2 (6.9)	0

[a]Patients with SD, PD, and those whose responses were not evaluable were excluded from the analysis.
[b]Per RECIST v1.1.
[c]Definitive signs of PD, but a recent radiographic assessment did not meet the criteria for PD per RECIST v1.1.

Table 2. Efficacy Outcomes by Response Category

Median (95% CI), months	T-DXd (n = 261[a]) CR (n = 34)	T-DXd Deep PR ≥80% to <100% (n = 43)	T-DXd Non–deep PR (n = 129)	T-DM1 (n = 263[a]) CR (n = 13)	T-DM1 Deep PR ≥80% to <100% (n = 30)	T-DM1 Non–deep PR (n = 54)
Treatment duration	35.6 (29.3-64.1)	32.1 (23.5-51.5)	14.7 (12.6-17.8)	25.5 (11.2-56.0)	20.6 (12.5-27.2)	9.9 (8.3-16.6)
Time to best response (CR/deep PR/non–deep PR)	6.4 (4.2-8.4)	6.9 (5.6-8.6)	1.7 (1.5-2.7)	4.1 (1.4-19.2)	4.5 (2.8-7.1)	1.6 (1.4-2.7)
Time from best response (CR/deep PR/non–deep PR) to discontinuation	37.4 (26.3-62.5)	18.4 (14.6-25.6)	12.0 (10.6-15.0)	25.3 (9.3-51.8)	13.9 (7.6-21.1)	7.0 (6.1-11.1)
DOR	NE (NE-NE)	NE (27.7-NE)	19.3 (12.7-23.5)	NE (19.0-NE)	15.1 (9.6-30.4)	11.0 (5.6-16.5)

[a]Patients with SD, PD, and those whose responses were not evaluable were excluded from the analysis.

Table 3. PFS Rates

PFS rate (95% CI), %[b]	T-DXd (n = 261[a]) CR (n = 34)	T-DXd Deep PR ≥80% to <100% (n = 43)	T-DXd Non–deep PR (n = 129)	T-DM1 (n = 263[a]) CR (n = 13)	T-DM1 Deep PR ≥80% to <100% (n = 30)	T-DM1 Non–deep PR (n = 54)
12 months	93.9 (77.9-98.4)	95.1 (81.9-98.8)	73.8 (65.0-80.7)	91.7 (53.9-98.8)	82.8 (63.5-92.5)	52.7 (37.6-65.6)
24 months	93.9 (77.9-98.4)	77.6 (61.4-87.7)	45.9 (36.3-54.8)	82.5 (46.1-95.3)	51.8 (31.5-68.7)	27.5 (15.1-41.4)
36 months	77.2 (57.9-88.4)	69.6 (52.7-81.5)	33.6 (24.6-42.8)	82.5 (46.1-95.3)	26.7 (10.5-46.1)	13.7 (5.2-26.4)
48 months	77.2 (57.9-88.4)	66.6 (49.4-79.1)	28.3 (19.6-37.6)	61.9 (27.0-83.9)	26.7 (10.5-46.1)	8.2 (2.2-19.7)
60 months	77.2 (57.9-88.4)	59.7 (41.8-73.7)	25.0 (16.5-34.5)	61.9 (27.0-83.9)	17.8 (4.1-39.4)	8.2 (2.2-19.7)

[a]Patients with SD, PD, and those whose responses were not evaluable were excluded from the analysis.
[b]Estimate and CI for PFS rate at the specified time point are from a Kaplan–Meier analysis.

Figure 2. PFS by Response Category Using the ≥80% to <100% Deep PR Threshold in the A) T-DXd and B) T-DM1 Arms, Including 2- and 5-Year Landmarks

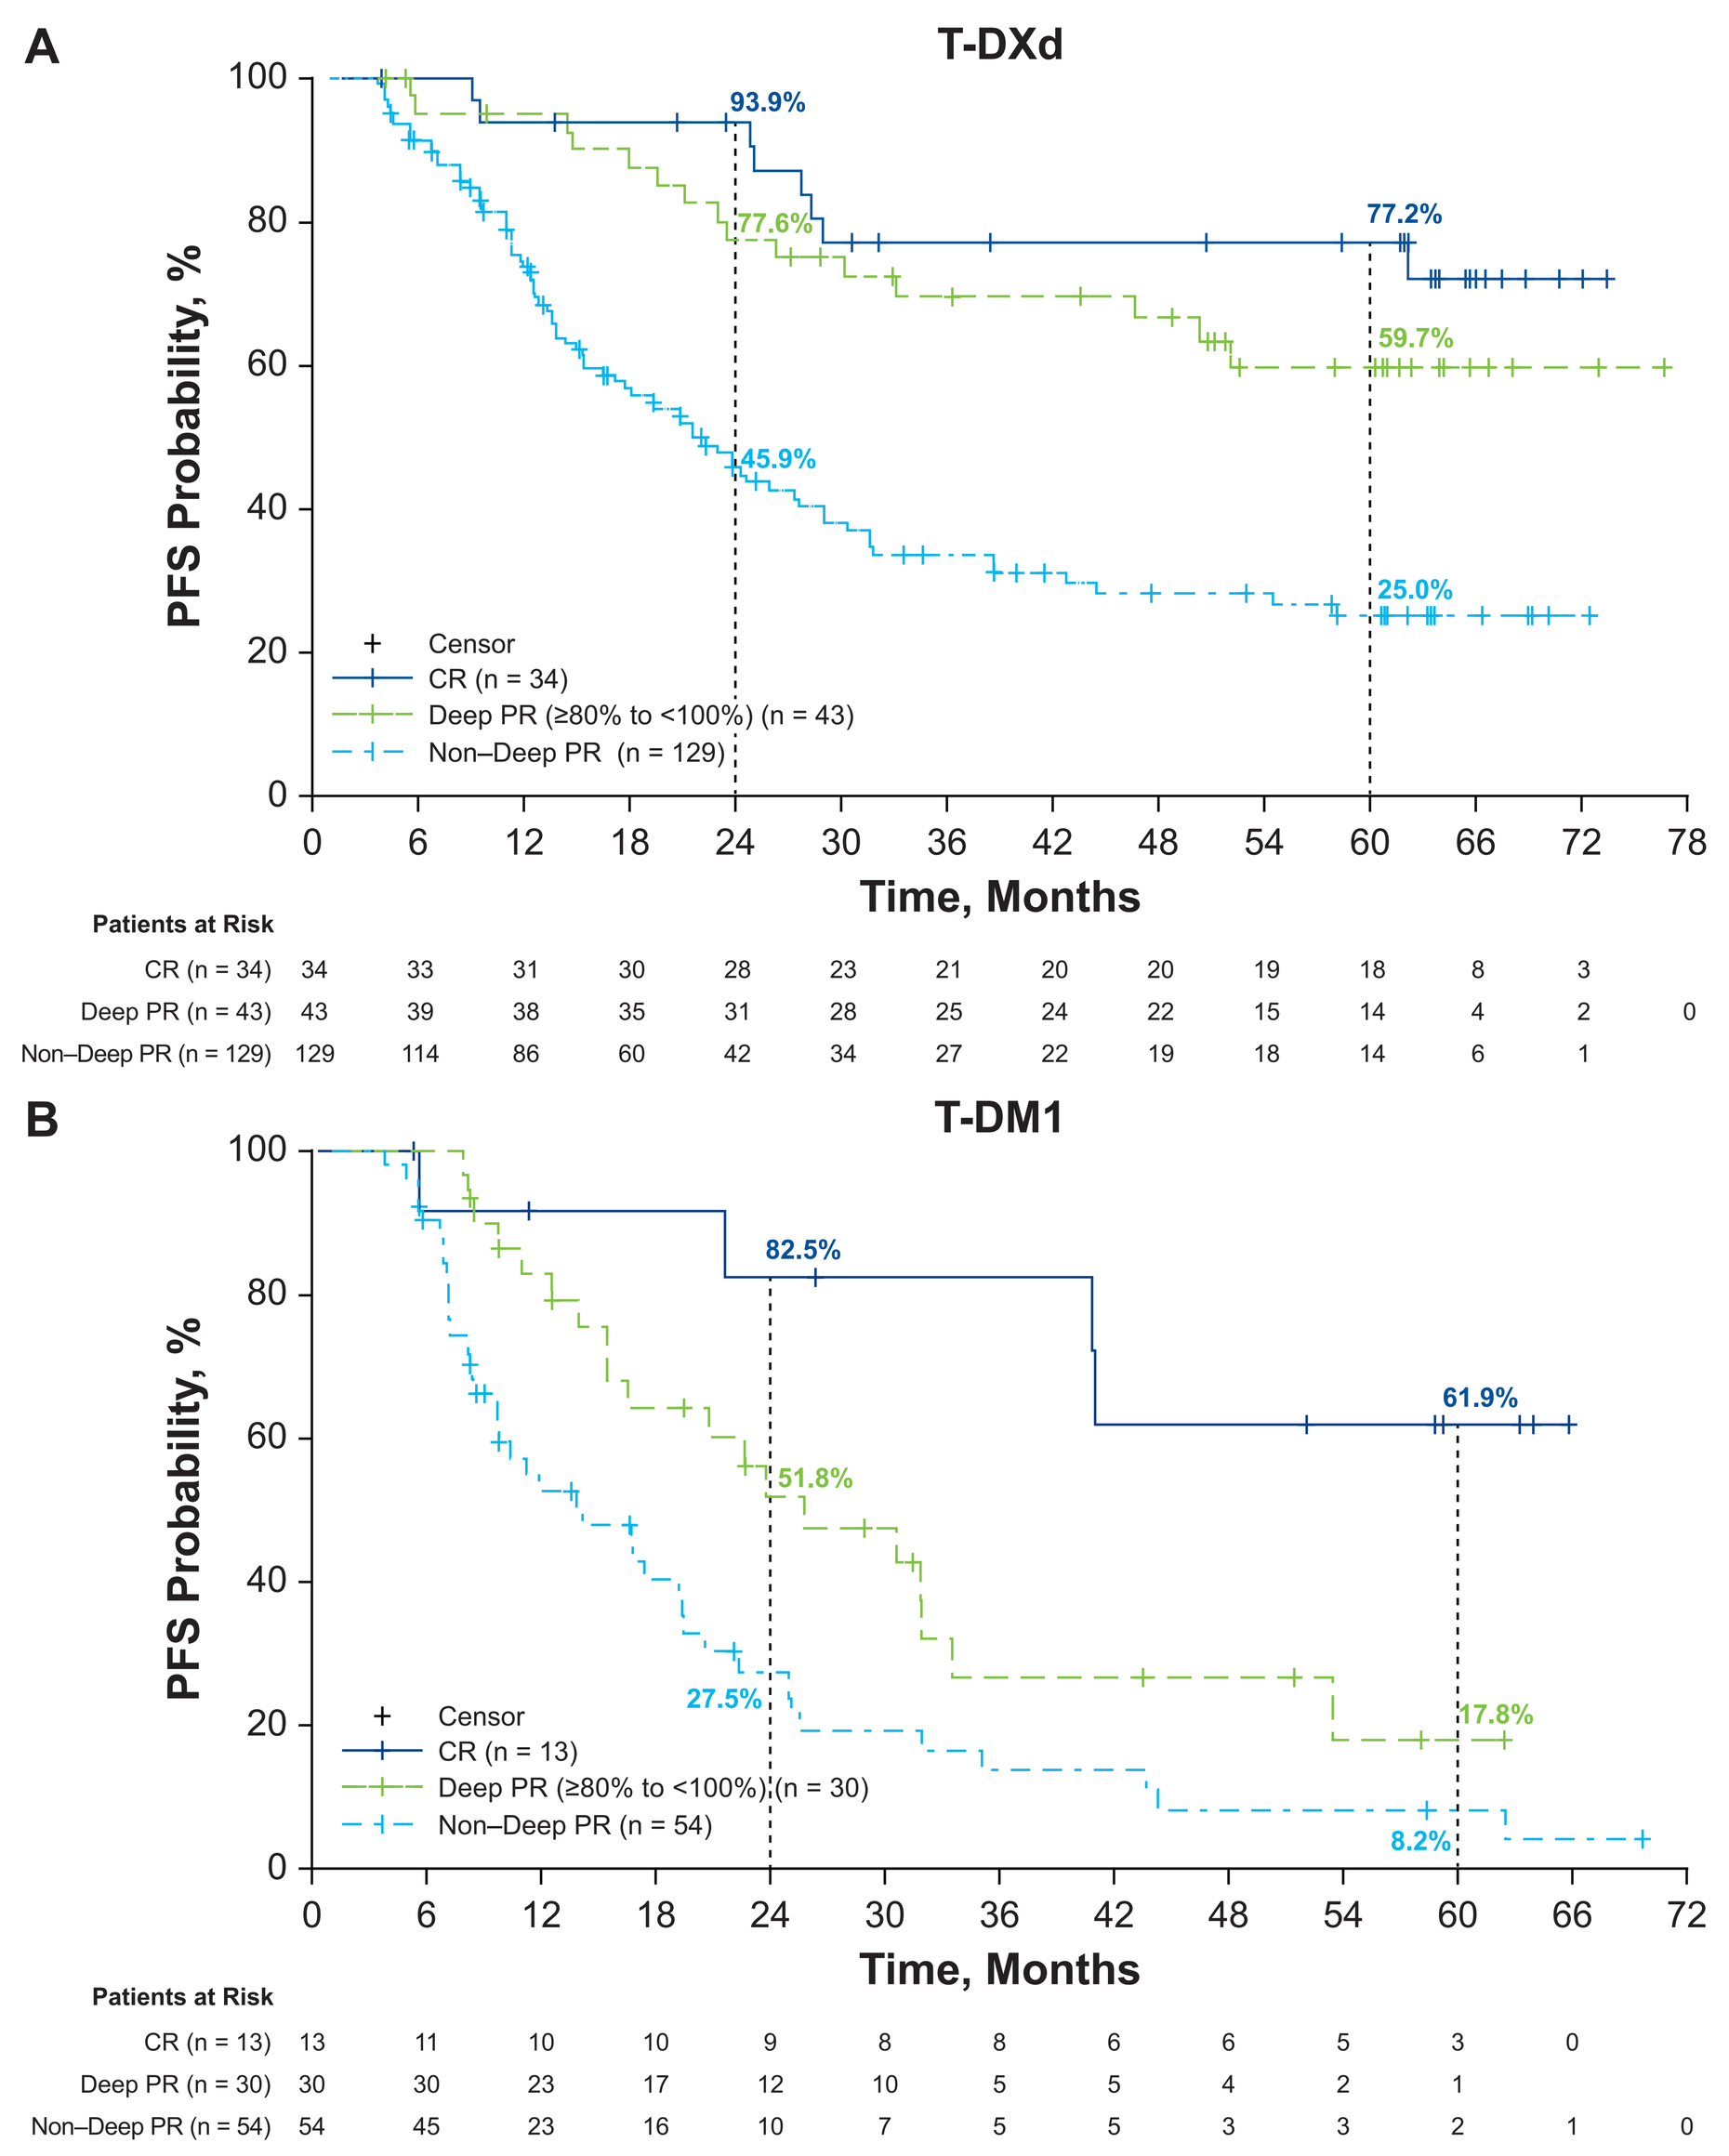

A
T-DXd
PFS Probability, %
Time, Months
93.9%
77.6%
45.9%
77.2%
59.7%
25.0%
Censor
CR (n = 34)
Deep PR (≥80% to <100%) (n = 43)
Non–Deep PR (n = 129)
Patients at Risk
CR (n = 34) 34 33 31 30 28 23 21 20 20 19 18 8 3
Deep PR (n = 43) 43 39 38 35 31 28 25 24 22 15 14 4 2 0
Non–Deep PR (n = 129) 129 114 86 60 42 34 27 22 19 18 14 6 1
B
T-DM1
PFS Probability, %
Time, Months
82.5%
51.8%
27.5%
61.9%
17.8%
8.2%
Censor
CR (n = 13)
Deep PR (≥80% to <100%) (n = 30)
Non–Deep PR (n = 54)
Patients at Risk
CR (n = 13) 13 11 10 10 9 8 8 6 6 5 3 0
Deep PR (n = 30) 30 30 23 17 12 10 5 5 4 2 1
Non–Deep PR (n = 54) 54 45 23 16 10 7 5 5 3 3 2 1 0

Patients with SD, PD, and those whose responses were not evaluable were excluded from the analysis.

Figure 3. 12-Month PFS by Response Category and Deep PR Threshold in the T-DXd Arm

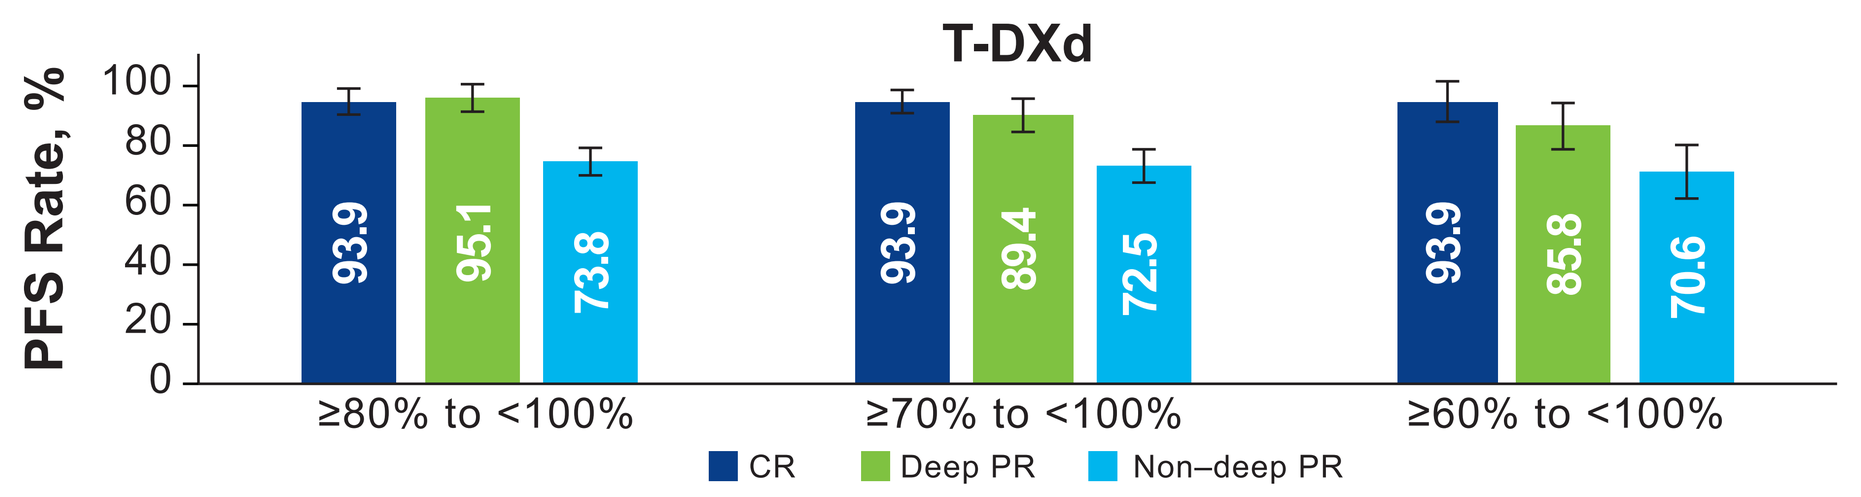

T-DXd
PFS Rate, %
93.9
95.1
73.8
93.9
89.4
72.5
93.9
85.8
70.6
≥80% to <100%
≥70% to <100%
≥60% to <100%
CR
Deep PR
Non–deep PR

Error bars represent 95% CIs.

Sensitivity Analysis

- In the T-DXd arm, median treatment durations were similar between the ≥70% to <100% and ≥60% to <100% deep PR threshold groups (24.4 months and 23.5 months, respectively), and notably shorter than the ≥80% to <100% threshold group (32.1 months) (**Supplementary Table 3**)
- Median time to best response in the T-DXd arm was similar between the ≥70% to <100% and ≥60% to <100% deep PR threshold groups (5.6 months and 5.5 months, respectively), and notably shorter than the ≥80% to <100% threshold group (6.9 months) (**Supplementary Table 4**)
- In both treatment arms, deep PR at the ≥70% to <100% and ≥60% to <100% thresholds was associated with improved PFS outcomes compared with non–deep PR (**Figure 3** and **Supplementary Figures 1-5**)
 - Among patients with CR, the 12-month PFS rate (95% CI) was 93.9% (77.9-98.4%) in the T-DXd arm and 91.7% (53.9-98.8%) in the T-DM1 arm
 - In the ≥70% to <100% deep PR group of the T-DXd arm, the 12-month PFS rate (95% CI) was 89.4% (79.0-94.8%), and it was 72.5% (62.4-80.2%) in patients with non–deep PR; in the T-DM1 arm, rates were 75.1% (57.6-86.2%) and 54.3% (37.9-68.1%), respectively
 - In the ≥60% to <100% deep PR group of the T-DXd arm, the 12-month PFS rate (95% CI) was 85.8% (76.9-91.5%), and it was 70.6% (58.5-79.7%) in patients with non–deep PR; in the T-DM1 arm, rates were 68.2% (53.4-79.2%) and 56.5% (36.0-72.7%), respectively
- At longer follow-up and across treatment arms, the ≥70% to <100% and ≥60% to <100% threshold groups showed a more pronounced decline in PFS rate compared with the ≥80% to <100% threshold group (**Figure 3; Supplementary Figures 2 and 3**)

Abbreviations

AE, adverse event; BICR, blinded independent central review; CR, complete response; DCO, data cutoff; DOR, duration of response; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; ISH, in situ hybridization; mBC, metastatic breast cancer; NE, not estimable; ORR, objective response rate; PD, progressive disease; PFS, progression-free survival; PFS2, progression-free survival from time of randomization to progression on the next line of therapy or death; PR, partial response; RECIST, Response Evaluation Criteria in Solid Tumours; SD, stable disease; T-DM1, trastuzumab emtansine; T-DXd, trastuzumab deruxtecan; TL, target lesion.

Acknowledgments

Medical writing support, under the direction of the authors, was provided by Caylin Bosch, PhD, and Jill Shults, PhD, of ApotheCom, and was funded by Daiichi Sankyo in accordance with Good Publication Practice (GPP) guidelines (http://www.ismpp.org/gpp-2022).

Disclosures

Dr Hamilton reports research funding from AbbVie, Accutar Biotechnology, Artios, Arvinas, AstraZeneca, AtlasMedx, BeiGene, BeOne Medicines, Bicycle Therapeutics, Biohaven Pharmaceuticals, BioNTech, Compugen, Cullinan, Daiichi Sankyo, Dantari, Day One Biopharmaceuticals, Duality Biologics, Ellipses Pharma, Elucida Oncology, Exelixis, FujiFilm, Genmab, Gilead Sciences, H3 Biomedicine, Iambic Therapeutics, Immunogen, Inspirna, InventisBio, Jacobio, Jazz Pharmaceuticals, K-Group Beta, Kind Pharmaceuticals, Lilly, Loxo Oncology, Mabspace Biosciences, Mabwell Bioscience, Marengo Therapeutics, MediLink Therapeutics, Merck, Mersana, Novartis, Olema, Orinove, Orum Therapeutics, Pfizer, Pionyr Immunotherapeutics, Prelude Therapeutics, Profound Bio, Regeneron, Relay Therapeutics, Rgenix, Roche/Genentech, SeaGen, Shattuck Labs, Simcha Therapeutics, Stemline Therapeutics, Sutro, Systimmune, Taiho, Tesaro, TheRas, Treadwell Therapeutics, Verastem, Xadcera Biopharmaceutical, and Zymeworks. Dr Hamilton also reports consulting/advisory roles with Arvinas, AstraZeneca, BeOne Medicines, BioNTech, Boehringer Ingelheim, Boundless Bio, Bristol Myers Squibb, Circle Pharma, Daiichi Sankyo, Gilead Sciences, Halda Therapeutics, Incyclix Bio, IQVIA, Janssen, Jazz Pharmaceuticals, Jefferies LLC, Johnson & Johnson, Lilly, Mersana Therapeutics, Novartis, Pfizer, Precede Biosciences, Pyxis Oncology, Roche/Genentech, Samsung Bioepis, Shorla Pharma, Stemline Therapeutics, and Tempus Labs.

References

1. Saura C et al. *Ann Oncol*. 2026;37(3):353-363.
2. Cortés J et al. *N Engl J Med*. 2022;386(12):1143-1154.
3. Hurvitz SA et al. *Lancet*. 2023;401(10371):105-117.
4. Im SA et al. Presented at SABCS 2025; December 9-12, 2025; San Antonio, TX. Poster #PS5-01-30.
5. Eisenhauer EA et al. *Eur J Cancer*. 2009;45(2):228-247.