Unmet needs and treatment perspectives of patients with previously treated small cell lung cancer: A global, qualitative interview study

#574

Samantha Valliant,[1] Sudhir Unni,[1] Tara Herrmann,[1] Sajid Ahmed,[1] Meredith Venerus,[2] Friso Coerts,[3]* Cecilia Jimenez Moreno,[4] Dorsa Khazaei,[4] Tommi Tervonen,[5] Toshiyuki Kozuki,[6] Jared Weiss,[7] Nicolas Girard[8]

[1]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [2]Daiichi Sankyo, Madrid, Spain; [3]Daiichi Sankyo, Schiphol-Rijk, The Netherlands; [4]Kielo Research, York, UK; [5]Kielo Research, Zug, Switzerland; [6]Kochi University, Kochi, Japan; [7]University of North Carolina, Chapel Hill, NC, USA; [8]Institut Curie, Paris, France.

*At the time of the study.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

OBJECTIVES

- This study aimed to explore the real-world unmet medical needs and treatment perspectives of patients with SCLC receiving treatment in the 2L+ setting. The objectives were to:
 1. Understand the patient experience, disease burden, and unmet medical needs in previously treated SCLC
 2. Identify key drivers of treatment decision-making among patients with SCLC
 3. Determine treatment attributes that influence patients' willingness to initiate or continue treatment

CONCLUSIONS

- This qualitative study in a predominantly ES-SCLC patient population highlights the substantial emotional and physical burden that is placed on patients receiving treatment in the 2L+ setting
- The most bothersome symptoms reported were shortness of breath, fatigue or tiredness, and cough, all of which had a broad negative impact on QOL across social, emotional, and physical domains
- Patients valued concepts of delaying disease worsening and responding to treatment as important treatment-related outcomes

INTRODUCTION

- SCLC is a highly aggressive form of lung cancer, representing about 15% of all lung cancer cases,[1] and over 80% of patients have ES-SCLC at the time of diagnosis[2]
- For patients with ES-SCLC, 1L treatment typically includes platinum-based chemotherapy with anti–PD-(L)1 immunotherapy.[3,4] However, most patients experience disease progression and relapse within 6 months[4–7]
- Until recently, treatment options in 2L have been limited (median OS of 6.0–9.3 months with chemotherapy; 13.6 months with tarlatamab, approved in the USA, Japan, and UK in 2024); they are also associated with high rates of hematologic, gastrointestinal, and neurologic toxicity.[4,8–12] There is no clear global standard of care for 3L+.[3,4,13] As a result, there is a significant unmet need for new treatments that can enhance both survival outcomes and QOL for patients with SCLC

METHODS

Study design

- This was a qualitative interview study in patients with SCLC receiving 2L+ treatment in the USA, Japan, Brazil, China, Italy, Republic of Korea, Singapore, and Taiwan
- Data were collected between November 2024 and March 2025

Participants

- Age ≥18 years with SCLC
 - Patients in the 2L cohort had disease progression on 1 prior SCLC treatment regimen and not progressed on a second regimen, and had to be from the USA, Japan, Brazil, China, Italy, Republic of Korea, Singapore, or Taiwan
 - Patients in the 3L+ cohort had disease progression after ≥2 prior SCLC lines of therapy and had to be from either the USA or Japan

Study procedures

- A brief online survey was used to collect sociodemographic and clinical characteristics
- One-on-one semi-structured interviews were conducted remotely via a secure online platform (Microsoft Teams®). An interview guide was used to structure the discussions. All supporting materials, including the survey, were translated into local languages. The interviews explored the following themes:
 1. Patients' experiences with SCLC symptoms and daily life impacts
 2. Past and current treatment experiences, including benefits, side effects, and treatment burden
 3. Drivers of treatment decision-making, such as expected outcomes and risk tolerance
- For questions relating to symptoms, disease impacts, and treatment expectations (valued treatment outcomes), patients provided spontaneous responses, after which they could provide answers to categories presented to them that they had not already described (probed)

Data preparation and analysis

- Qualitative data preparation involved transcribing interview recordings in the local language, followed by English translation
- Qualitative data were analyzed using interpretative analysis methods and a predefined coding dictionary. Data saturation was assessed iteratively
 - ATLAS.ti version 25 software was used for the qualitative coding
 - R version 4.3 software was used for coding frequency tabulation
- Sociodemographic and clinical data were summarized using descriptive statistics
 - R version 4.2 software was used for all quantitative analyses

RESULTS

Sociodemographic and clinical characteristics

- A total of 30 patients with SCLC (20 [66.7%] with ES-SCLC at time of diagnosis) who were receiving treatment in the 2L+ were included; 21 (70%) received 2L treatment and 9 (30%) received 3L+ treatment (**Table 1**)
- The median age of patients was 59 years (range, 28–81), 50% were female, and most patients had preserved functional status (ECOG PS score of 0–1, 76.7%)

Table 1. Patient-reported baseline sociodemographic and clinical characteristics

	Overall (N=30)	2L cohort (n=21)	3L+ cohort (n=9)
Country, n (%)			
USA	10 (33.3)	5 (23.8)	5 (55.6)
Japan	8 (26.7)	4 (19.0)	4 (44.4)
Brazil	2 (6.7)	2 (9.5)	0
China	2 (6.7)	2 (9.5)	0
Italy	2 (6.7)	2 (9.5)	0
Republic of Korea	2 (6.7)	2 (9.5)	0
Singapore	2 (6.7)	2 (9.5)	0
Taiwan	2 (6.7)	2 (9.5)	0
Age at baseline, median (range), years	59 (28–81)	57 (28–69)	62 (37–81)
Sex, n (%)			
Female	15 (50.0)	9 (42.9)	6 (66.7)
Male	15 (50.0)	12 (57.1)	3 (33.3)
ECOG PS at baseline, n (%)			
0	4 (13.3)	2 (9.5)	2 (22.2)
1	19 (63.3)	13 (61.9)	6 (66.7)
2	2 (6.7)	1 (4.8)	1 (11.1)
3–4	5 (16.7)	5 (23.8)	0

Symptoms

- The most commonly reported symptoms (reported by ≥50%) in the overall population were shortness of breath (90.0%; n=27), fatigue or tiredness (83.3%; n=25), cough (73.3%; n=22), and chest pain (53.3%; n=16; **Figure 1**). These symptoms were consistently reported across both the 2L and 3L+ cohorts
- When asked about the symptoms that had the most significant impact on their lives, patients most frequently reported fatigue or tiredness (63.3%; n=19), followed by shortness of breath (53.3%; n=16) and cough (46.7%; n=14; **Figure 1**)

Impacts of disease on daily life

- Patients described a broad spectrum of emotional, physical, and social impacts from SCLC (**Figure 2**)
- Emotional effects were the most frequently reported (93.3%; n=28), including anxiety or stress, sadness, and fear. These emotions were particularly pronounced in the 3L+ cohort, where 88.9% (n=8) and 77.8% (n=7) of patients experienced anxiety/stress and sadness, respectively, compared with 47.6% (n=10) and 52.4% (n=11) in the 2L cohort
- Physical and social impacts were also common, experienced by 93.3% (n=28) and 86.7% (n=26), respectively, and affected mobility, the ability to perform daily tasks, social engagement, and participation in leisure activities

Treatments received and impacts of the latest treatment

- Self-reported treatments received by line of therapy in the overall, 2L, and 3L+ cohorts are shown in **Table 2**
- Nearly all patients (96.7%, n=29) reported positive impacts from their most recent treatment (**Figure 3**). The most frequently reported positive impacts were treatment effectiveness (56.7%; n=17), improvements in QOL (50.0%; n=15), and symptom relief (36.7%; n=11)
- Negative impacts from the most recent treatment were reported in 86.7% (n=26) of patients (**Figure 3**). The most frequently reported negative impacts included side effects (76.7%; n=23), lack of efficacy (20.0%; n=6), and reduced QOL (20.0%; n=6)

Treatment-related burden

- A burden relating to the mode of treatment administration was reported by two-thirds of patients (66.7%; n=20), with almost all (63.3%; n=19) attributing this burden to time spent in the clinic (**Table 3**)
- Additionally, 23.3% (n=7) reported experiencing a treatment-related financial burden, which was more commonly noted by patients in the 2L cohort than in the 3L+ cohort (**Table 3**)
 - Patients who raised financial concerns were from Brazil, China, Republic of Korea, Taiwan (n=1; 50.0% from each country), and Japan (n=3; 37.5%), suggesting potential country-level patient differences in health insurance coverage, financial status, and how financial strain is perceived or experienced
 - No patients from the USA reported experiencing financial burden, which may be due to the majority (80%) having private medical insurance

Figure 1. Self-reported symptoms (reported by ≥50% of patients in the overall population), including most bothersome

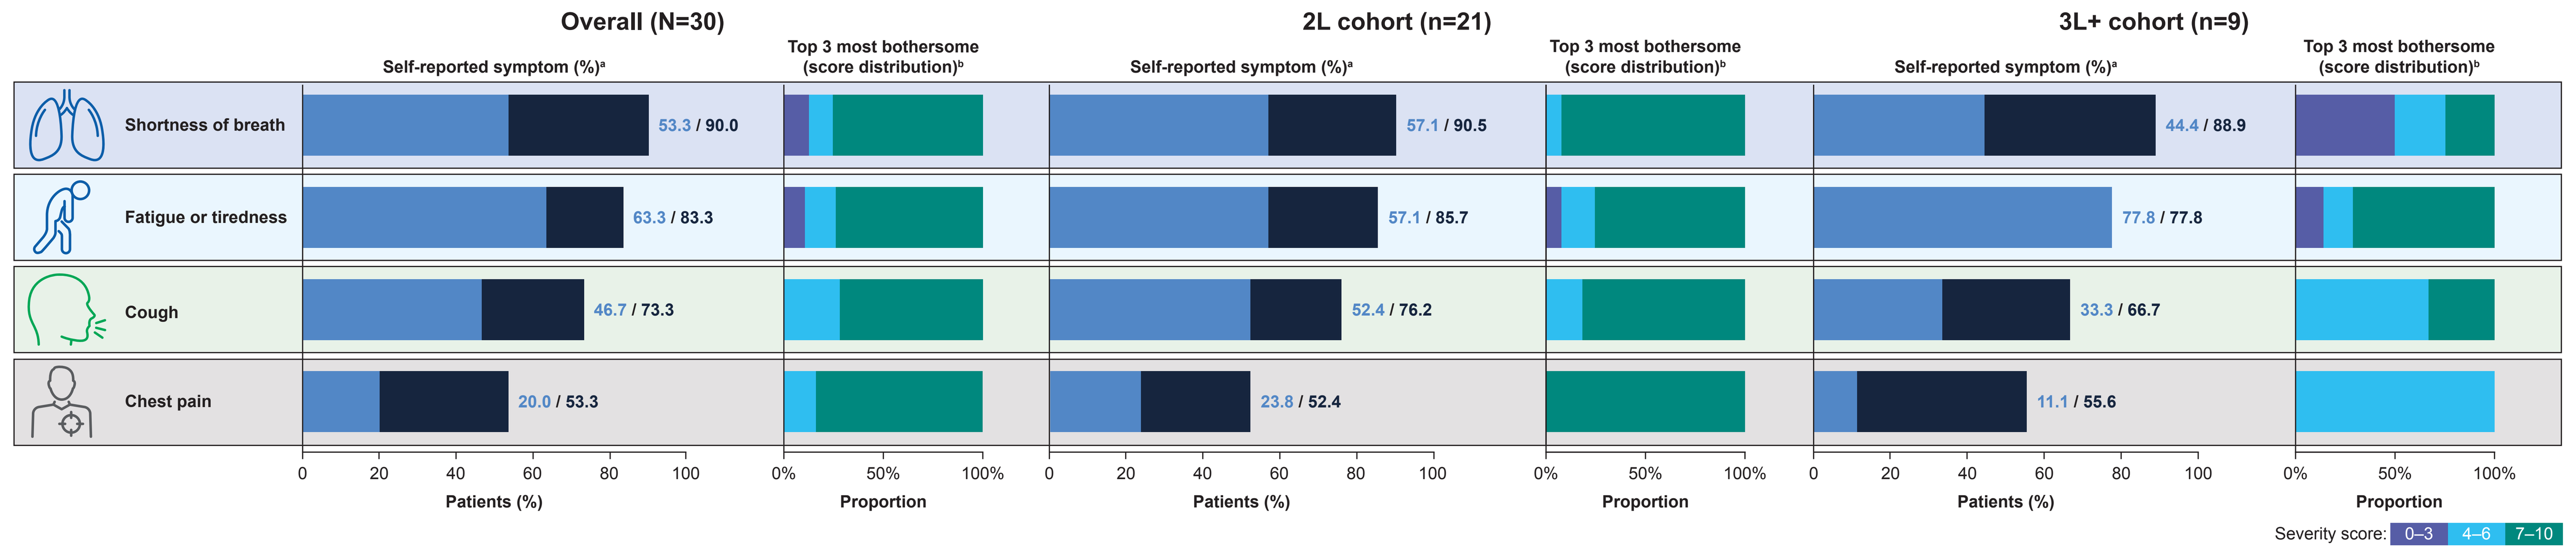
Overall (N=30)
2L cohort (n=21)
3L+ cohort (n=9)
Self-reported symptom (%)a
Top 3 most bothersome (score distribution)b
Shortness of breath
Fatigue or tiredness
Cough
Chest pain
53.3 / 90.0
63.3 / 83.3
46.7 / 73.3
20.0 / 53.3
57.1 / 90.5
57.1 / 85.7
52.4 / 76.2
23.8 / 52.4
44.4 / 88.9
77.8 / 77.8
33.3 / 66.7
11.1 / 55.6
Patients (%)
Proportion
Severity score: 0–3 4–6 7–10

Data labels are: Self-reported as top 3 most bothersome (%) / Total self-reported symptoms (spontaneously or when probed; %).
aBars show total % of patients reporting symptom, with blue sections denoting the % who reported the symptom among the 3 most bothersome. bDistribution of reported top 3 most bothersome symptoms by severity score, where 0=does not bother at all and 10=extremely bothersome.

Figure 2. Self-reported impacts of disease on daily life (reported by ≥50% of patients in the overall population), including most bothersome

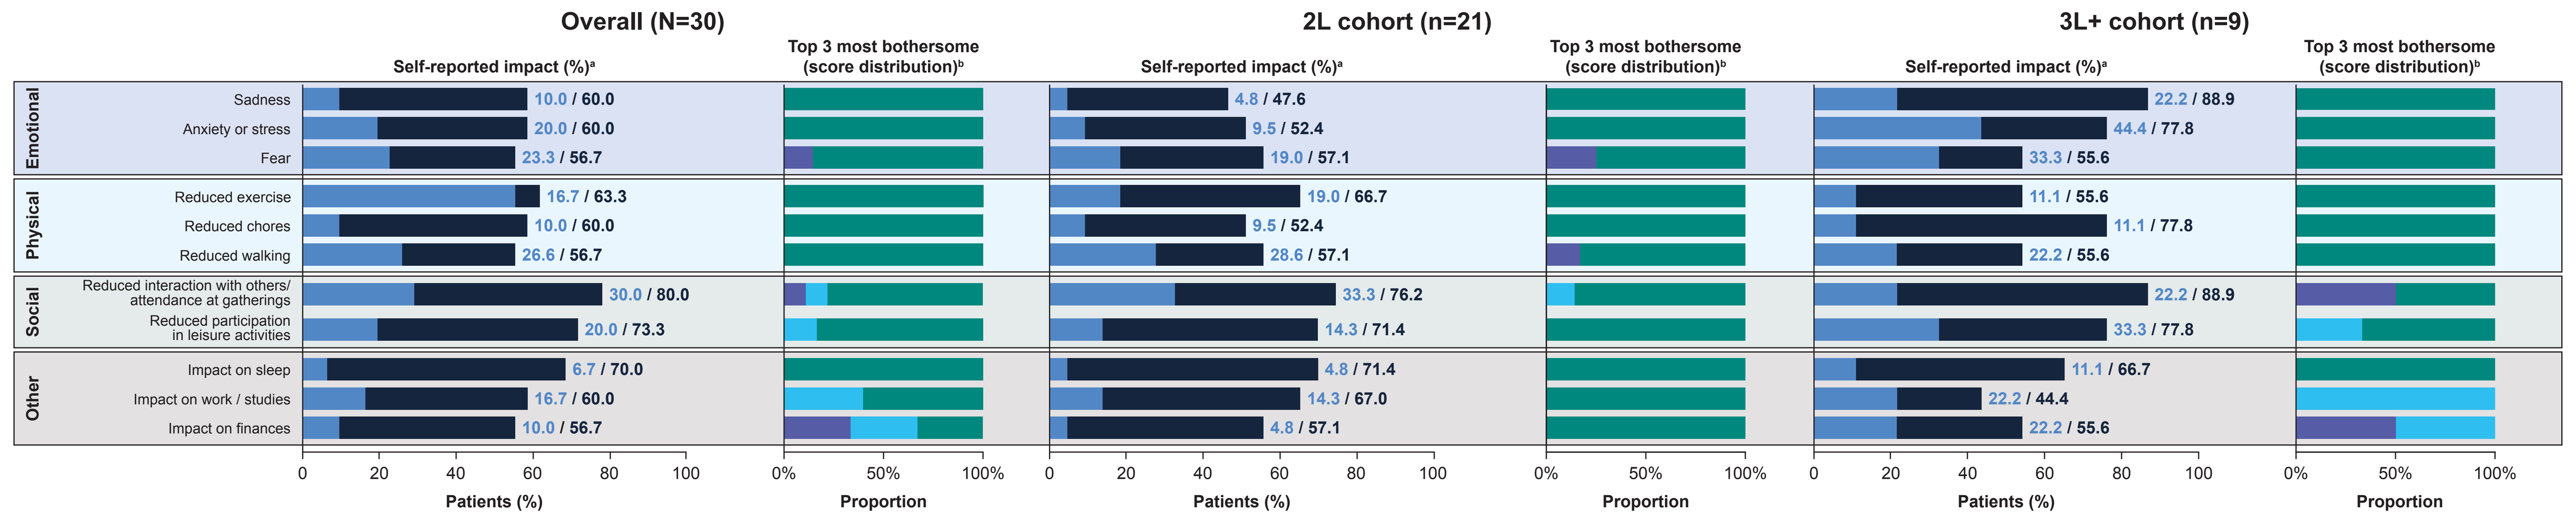
Overall (N=30)
2L cohort (n=21)
3L+ cohort (n=9)
Self-reported impact (%)a
Top 3 most bothersome (score distribution)b
Emotional
Sadness
Anxiety or stress
Fear
Physical
Reduced exercise
Reduced chores
Reduced walking
Social
Reduced interaction with others/ attendance at gatherings
Reduced participation in leisure activities
Other
Impact on sleep
Impact on work / studies
Impact on finances
10.0 / 60.0
20.0 / 60.0
23.3 / 56.7
16.7 / 63.3
10.0 / 60.0
26.6 / 56.7
30.0 / 80.0
20.0 / 73.3
6.7 / 70.0
16.7 / 60.0
10.0 / 56.7
4.8 / 47.6
9.5 / 52.4
19.0 / 57.1
19.0 / 66.7
9.5 / 52.4
28.6 / 57.1
33.3 / 76.2
14.3 / 71.4
4.8 / 71.4
14.3 / 67.0
4.8 / 57.1
22.2 / 88.9
44.4 / 77.8
33.3 / 55.6
11.1 / 55.6
11.1 / 77.8
22.2 / 55.6
22.2 / 88.9
33.3 / 77.8
11.1 / 66.7
22.2 / 44.4
22.2 / 55.6
Patients (%)
Proportion
Severity score: 0–3 4–6 7–10

Data labels are: Self-reported as top 3 most bothersome (%) / Total self-reported impact (spontaneously or when probed; %).
aBars show total % of patients reporting an impact, with blue sections denoting the % who reported the impact among the 3 most bothersome. bDistribution of reported top 3 most bothersome impacts by severity score, where 0=does not bother at all and 10=extremely bothersome.

Table 2. Treatments received by line of therapy[a]

Treatment category, n (%)	Overall (N=30)	2L cohort (n=21)	3L+ cohort (n=9)
1L treatment			
Chemotherapy	17 (56.7)	12 (57.1)	5 (55.6)
Chemotherapy + immunotherapy	5 (16.7)	4 (19.0)	1 (11.1)
Chemotherapy + RT	3 (10.0)	1 (4.8)	2 (22.2)
Chemotherapy + RT + surgery	1 (3.3)	0	1 (11.1)
Immunotherapy	2 (6.7)	2 (9.5)	0
RT	1 (3.3)	1 (4.8)	0
Surgery	1 (3.3)	1 (4.8)	0
2L treatment			
Chemotherapy	20 (66.7)	13 (61.9)	7 (77.8)
Immunotherapy	6 (20.0)	6 (28.6)	0
RT	3 (10.0)	2 (9.5)	1 (11.1)
RT + immunotherapy	1 (3.3)	0	1 (11.1)
3L treatment			
Chemotherapy	2 (6.7)	N/A	2 (22.2)
Chemotherapy + RT	1 (3.3)	N/A	1 (11.1)
Immunotherapy	6 (20.0)	N/A	6 (66.7)
No treatment in this line	21 (70.0)	21 (100)	0
4L+ treatment			
Chemotherapy	1 (3.3)	N/A	1 (11.1)
Clinical trial	1 (3.3)	N/A	1 (11.1)
No treatment in this line	28 (93.3)	21 (100)	7 (77.8)

Figure 3. Impact of the latest treatment

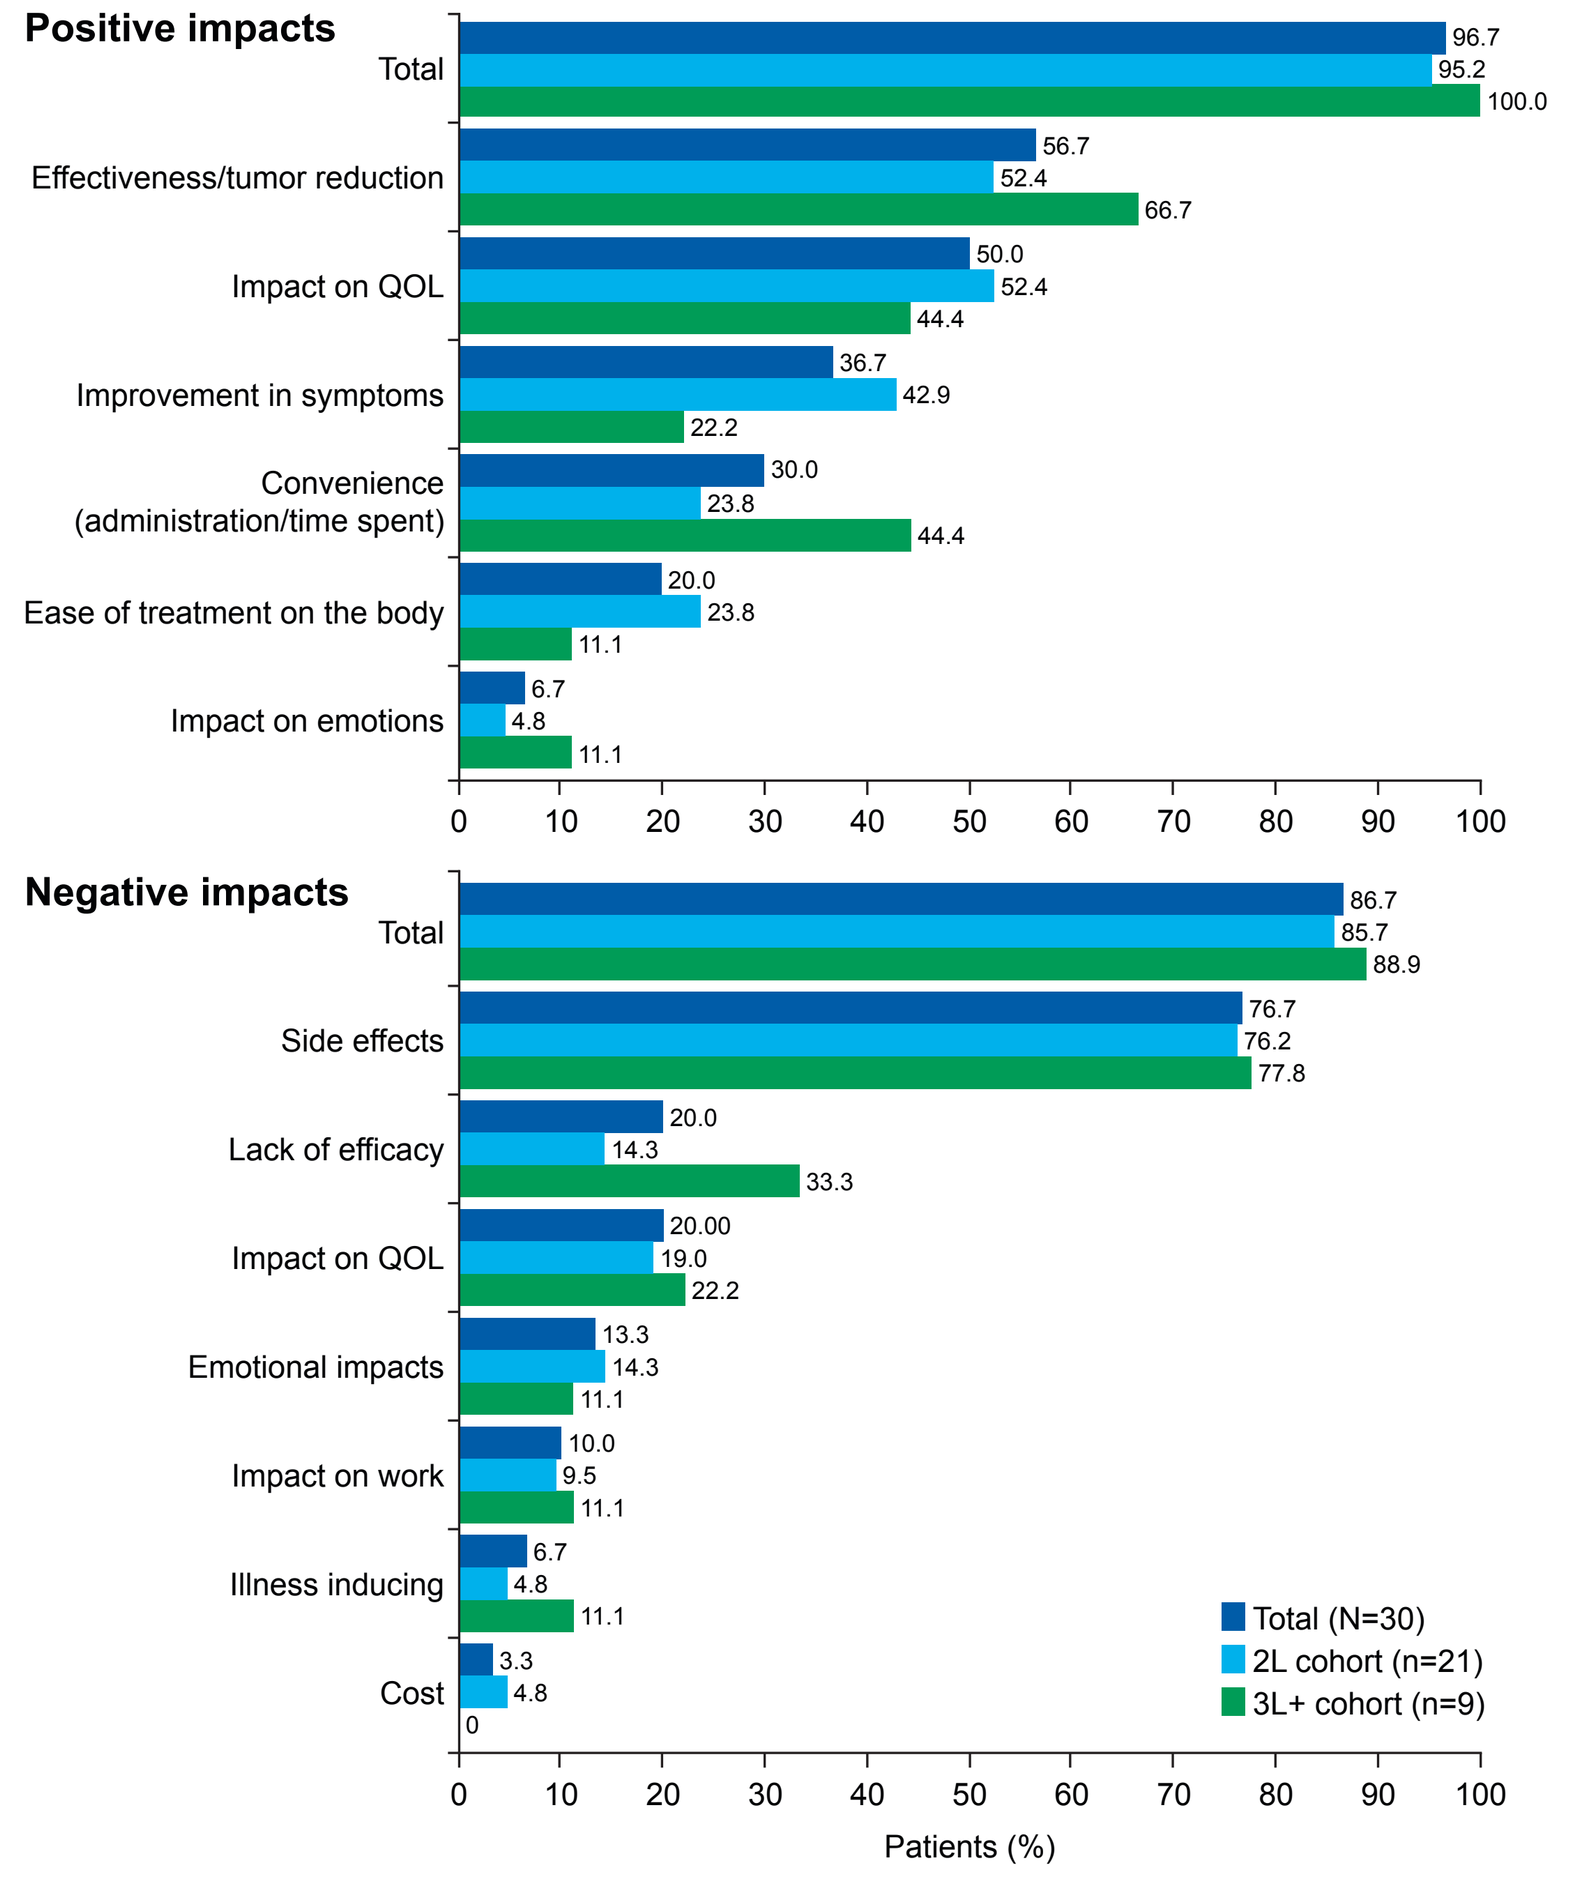
Positive impacts
Total
96.7
95.2
100.0
Effectiveness/tumor reduction
56.7
52.4
66.7
Impact on QOL
50.0
52.4
44.4
Improvement in symptoms
36.7
42.9
22.2
Convenience (administration/time spent)
30.0
23.8
44.4
Ease of treatment on the body
20.0
23.8
11.1
Impact on emotions
6.7
4.8
11.1
Negative impacts
Total
86.7
85.7
88.9
Side effects
76.7
76.2
77.8
Lack of efficacy
20.0
14.3
33.3
Impact on QOL
20.00
19.0
22.2
Emotional impacts
13.3
14.3
11.1
Impact on work
10.0
9.5
11.1
Illness inducing
6.7
4.8
11.1
Cost
3.3
4.8
0
Total (N=30)
2L cohort (n=21)
3L+ cohort (n=9)
Patients (%)

"Nowadays I can do something. I stopped working because I couldn't do it, now I can do something, so I've really improved significantly. Just being able to breathe better and practising some of the things I love to do, like cooking, having a better relationship with my partner, I can't put it into words."

– Patient from Brazil

"About 20 days in, I started losing my hair, which made it obvious to those around me. My family became even more worried because of it. I also experienced some gastrointestinal side effects, which were a bit tough."

– Patient from Japan

Table 3. Treatment-related burden

n (%)	Overall (N=30)	2L cohort (n=21)	3L+ cohort (n=9)
Burden related to mode of treatment administration	24 (80.0)	16 (76.2)	8 (88.9)
Burden description[a]			
Time spent in the clinic	19 (63.3)	14 (66.7)	5 (55.6)
Impact on QOL	6 (20.0)	6 (28.6)	0
Travel time/cost/parking	6 (20.0)	5 (23.8)	1 (11.1)
Impact on work	4 (13.3)	4 (19.0)	0
Impact on veins	4 (13.3)	2 (9.5)	2 (22.2)
Pain at injection site	2 (6.7)	2 (9.5)	0
Impact on family time	1 (3.3)	1 (4.8)	0
Treatment-related financial burden	7 (23.3)	6 (28.6)	1 (11.1)

aPatients could have reported ≥1 burden relating to the mode of treatment administration; counts are per patient, not per code.

Valued treatment outcomes

- Patients identified outcomes relating to delaying disease worsening and responding to treatment as important
 - Longer OS was valued by nearly all patients (96.7%; n=29)
 - PFS was also a priority for nearly all patients (96.7%; n=29), though it was rarely mentioned spontaneously (by 0% in the 2L cohort and 22.2% in the 3L+ cohort), thus was more commonly mentioned once probed (when explained as 'delaying time for your cancer to grow or spread')
 - ORR was valued by 93.3% (n=28) of patients overall, including 90.5% in the 2L cohort and 100% in 3L+ cohort, again mentioned spontaneously rarely (by 4.8% and 0%, respectively), and more commonly once explained as 'higher percentage of patients responding to treatment'
 - Remission or cure ('reduction or disappearance of the signs of cancer') was also commonly reported (86.7%; n=26) and was ranked as the most important outcome most frequently, especially in the 3L+ cohort (ranked first by 46.7%, 28.6%, and 88.9% in the overall, 2L, and 3L+ cohorts, respectively)
- Symptom management, improving QOL, and reducing hospital time were valued (by 76.7% each in the overall population), though ranked lower overall

STUDY LIMITATIONS

- The small overall sample size and limited number of participants per country constrain the generalizability of the findings and prevent country-specific conclusions. Additionally, cultural and geographic differences may introduce subjective variability. However, data saturation was met in this study, providing increased confidence that sufficient key themes were identified within the current sample size of the study to support the conclusions
- Despite varied recruitment methods, selection bias may be present, as participants could differ from the general SCLC population, potentially representing a subgroup who felt relatively well or had the energy and time to participate in an in-depth interview
- Recall bias may have affected data accuracy, eg, in reporting treatments, as responses relied on memory and were not verified by medical records. Similarly, patients' lack of understanding of the difference between 1L maintenance therapy and 2L therapy may have led to misclassification of treatments received in each line

REFERENCES

1. Rudin CM, et al. *Nat Rev Dis Primers.* 2021;7:3.
2. Saltos A, et al. *Front Oncol.* 2020;10:1074.
3. National Comprehensive Cancer Network®. NCCN Guidelines: Small Cell Lung Cancer Version 2.2026. https://www.nccn.org/guidelines/guidelines-detail?category=1&id=1462. Accessed September 26, 2025.
4. Dingemans AMC, et al. *Ann Oncol.* 2021;32:839–853.
5. Slotman BJ, et al. *Lancet.* 2015;385:36–42.
6. Horn L, et al. *N Engl J Med.* 2018;379:2220–2229.
7. Paz-Ares L, et al. *Lancet.* 2019;394:1929–1939.
8. von Pawel J, et al. *J Clin Oncol.* 2014;32:4012–4019.
9. Eckardt JR, et al. *J Clin Oncol.* 2007;25:2086–2092.
10. O'Brien MER, et al. *J Clin Oncol.* 2006;24:5441–5447.
11. Patel S, et al. *Ther Adv Med Oncol.* 2021;13:17588359211020529.
12. Mountzios G, et al. *N Engl J Med.* 2025;393:349–361.
13. The Japan Lung Cancer Society. Lung Cancer Practice Guidelines: including malignant pleural mesothelioma and thymic tumor. 2023 ed. https://www.haigan.gr.jp/guideline/2023/index.html. Accessed September 10, 2025.

ABBREVIATIONS

1L, first line; **2L(+)**, second line (and beyond); **3L(+)**, third line (and beyond); **4L(+)**, fourth line (and beyond); **ECOG PS**, Eastern Cooperative Oncology Group performance status; **(ES-)SCLC**, (extensive-stage) small cell lung cancer; **N/A**, not applicable; **ORR**, overall response rate; **OS**, overall survival; **PD-(L)1**, programmed death-(ligand) 1; **PFS**, progression-free survival; **QOL**, quality of life; **RT**, radiotherapy; **UK**, United Kingdom; **USA**, United States.

AKNOWLEDGMENTS

This study is sponsored by Daiichi Sankyo, Inc. In October 2023, Daiichi Sankyo entered into a global development and commercialization collaboration agreement with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA (MSD) for ifinatamab deruxtecan (I-DXd).

Medical writing support was provided by Sultana Jahan, PhD, and Natalie Dennis of BOLDSCIENCE Inc, and funded by Daiichi Sankyo, Inc.

Editorial support was provided in accordance with Good Publication Practice guidelines (ismpp.org/gpp-2022).

DISCLOSURES

Samantha Valliant is an employee and stockholder of Daiichi Sankyo, Inc.

Presented at ASCO Quality Care Symposium 2025; October 10−11, 2025; Chicago, USA.

Correspondence email address: samantha.valliant@daiichisankyo.com