94

Additional health-related quality of life analysis from DESTINY-Gastric04, a randomized phase 3 study of trastuzumab deruxtecan vs ramucirumab + paclitaxel in patients with human epidermal growth factor receptor 2-positive unresectable/metastatic gastric cancer/ gastroesophageal junction adenocarcinoma

Kohei Shitara,[1] Aziz Zaanan,[2] Sara Lonardi,[3] Christelle de la Fouchardière,[4] Clélia Coutzac,[4] Eric Van Cutsem,[5] Jeroen Dekervel,[5] Lin Shen,[6] Fabio Andre Franke,[7] Yelena Y. Janjigian,[8] Josep Tabernero,[9] Chloe Spalding,[10] Meredith Venerus,[10] Fabricio Souza,[11] Alessandro Ghiretti,[12] Mahmut Gümüş,[13] Filippo Pietrantonio[14]

[1]National Cancer Center Hospital East, Kashiwa, Japan; [2]Department of Gastroenterology and Digestive Oncology, Paris-Cité University, Paris, France; George Pompidou European Hospital, Paris, France; [3]Veneto Institute of Oncology IOV-IRCCS, Padua, Italy; [4]Centre Leon Berard, Lyon, France; [5]University Hospitals Gasthuisberg and University of Leuven, Leuven, Belgium; [6]State Key Laboratory of Holistic Integrative Management of Gastrointestinal Cancers, Department of GI Oncology, Peking University Cancer Hospital & Institute, Beijing, China; [7]Oncosite-Oncoclínicas, Ijuí, Brazil; [8]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [9]Vall d'Hebron Hospital Campus and Institute of Oncology (VHIO), University of Vic-Central University of Catalonia, Barcelona, Spain; [10]Daiichi Sankyo Europe GmbH, Munich, Germany; [11]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [12]Daiichi Sankyo Italia, Rome, Italy; [13]Istanbul Medeniyet University, Prof Dr Suleyman Yalcin City Hospital, Istanbul, Türkiye; [14]Fondazione IRCCS Istituto Nazionale dei Tumori, Milan, Italy

Objective

- To assess the impact of trastuzumab deruxtecan (T-DXd) compared with ramucirumab (RAM) in combination with paclitaxel (PTX) on health-related quality of life (HRQoL) measures in patients with human epidermal growth factor receptor 2-positive (HER2+) unresectable or metastatic gastric cancer (GC)/gastroesophageal junction adenocarcinoma (GEJA) from DESTINYGastric04

Conclusions

- Maintenance of patient-reported HRQoL with T-DXd in patients with HER2+ (immunohistochemistry [IHC] 3+, IHC 2+/in situ hybridization–positive [ISH+]) unresectable or metastatic GC/GEJA was supported by the findings of this analysis
- Time to first deterioration (TTFD) was not negatively impacted with T-DXd
- The mean change from baseline (CFB) in all Functional Assessment of Cancer Therapy–Gastric (FACT-Ga) subscales showed that symptoms and functioning of patients remained stable, with some subscales favoring T-DXd versus RAM + PTX

Plain Language Summary

Why did we perform this research?
Human epidermal growth factor receptor 2 (HER2), a protein on the surface of some cancer cells, is found in approximately 5% to 17% of gastric cancer (GC) cases.[1-4] Trastuzumab deruxtecan (T-DXd) is an anticancer therapy that targets HER2 and is approved to treat patients with metastatic GC or gastroesophageal junction adenocarcinoma (GEJA) that is HER2-positive (HER2+).[5-7] DESTINY-Gastric04 was performed to compare the treatment benefits and safety of T-DXd with ramucirumab plus paclitaxel, as a second-line treatment option, in patients with HER2+ metastatic GC or GEJA.[8] The primary analysis showed that patients who were given T-DXd had survived longer than patients given ramucirumab plus paclitaxel.[8] Patients in the trial reported that their health-related quality of life (HRQoL) was not negatively affected by T-DXd treatment.[8-10] This research examines additional patient-reported HRQoL results from the DESTINY-Gastric04 trial.

How did we perform this research?
Patients who had GC that was confirmed as expressing HER2 from a recent tumor sample were eligible for enrollment into this phase 3 clinical trial. Patients were randomly assigned in a 1:1 ratio to 1 of 2 treatment arms. One arm received T-DXd while the other arm received ramucirumab plus paclitaxel. Patient-reported HRQoL was assessed using standardized questionnaires that measure multiple factors related to changes in emotional, physical, functional, or social well-being, as well as symptom deterioration.

What were the findings of this research?
Results from this study remain consistent with earlier HRQoL assessments in DESTINY-Gastric04. Patients receiving T-DXd did not show deterioration in their symptoms compared with those receiving ramucirumab plus paclitaxel. Additionally, measures of functioning and overall well-being remained stable throughout treatment. Changes in certain measures, such as physical and emotional well-being were more favorable in patients who received T-DXd than those who received ramucirumab plus paclitaxel.

What were the implications of this research?
These findings support that patients with HER2+ GC/GEJA who receive T-DXd do not experience a decline in their quality of life, and further reinforce T-DXd as a therapy choice to treat metastatic HER2+ GC that has progressed on first-line therapy.

Where can I access more information?
To learn more about the DESTINY-Gastric04 study, you can visit: https://clinicaltrials.gov/ct2/show/NCT04704934

References

1. Bartley AN et al. *J Clin Oncol.* 2017;35:446-464.
2. Subasinghe D et al. *J Int Med Res.* 2023;51:1-16.
3. Uzunparmak B et al. *Ann Oncol.* 2023;34:1035-1046.
4. Van Cutsem E et al. *Gastric Cancer.* 2015;18:476-484.
5. ENHERTU (fam-trastuzumab deruxtecan-nxki) [prescribing information]. Daiichi Sankyo, Inc.; 2025.
6. Shitara K et al. *N Engl J Med.* 2020;382:2419-2430.
7. Van Cutsem E et al. *Lancet Oncol.* 2023;24:744-756.
8. Shitara K et al. *N Engl J Med.* 2025;393:336-348.
9. Shitara K et al. Presented at: ASCO Annual Meeting 2025; May 30-June 3, 2025; Chicago, IL, USA.
10. Pietrantonio F et al. Presented at: ESMO Congress 2025; October 17-21, 2025; Berlin, Germany; Poster 2099P.

Poster

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please click on the link below:
https://bit.ly/DS2026ASCO

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from the American Society of Clinical Oncology and the authors of this poster.

This study is sponsored by Daiichi Sankyo. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201).
Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting 2026; May 29-June 2, 2026; Chicago, Illinois, USA, by Dr. Kohei Shitara. Corresponding author email address: kshitara@east.ncc.go.jp

Introduction

- GC is the fifth most prevalent cancer type in terms of incidence and mortality[1]
- Among all patients with GC, approximately 5% to 17% have tumors that are HER2+[2-5]
- T-DXd, a HER2-directed antibody-drug conjugate, is approved in many countries worldwide for the treatment of adult patients with locally advanced or metastatic HER2+ GC/GEJA who have received a prior trastuzumab-based regimen based on results from the DESTINY-Gastric01, DESTINY-Gastric02, and/or DESTINY-Gastric04 trials[6-11]
- In DESTINY-Gastric04 (NCT04704934), second-line treatment with T-DXd resulted in a statistically significant and clinically meaningful improvement in overall survival compared with RAM + PTX in patients with HER2+ (IHC 3+ or IHC 2+/ISH+) unresectable or metastatic GC/GEJA[11]
- Patient-reported HRQoL was also maintained with T-DXd treatment in DESTINYGastric04, with no clinically meaningful changes in patient-reported outcomes[11-13]
- Additional patient-reported HRQoL outcomes were assessed in DESTINY-Gastric04 and are presented here

Methods

- In DESTINY-Gastric04, patients with HER2+ unresectable or metastatic GC/GEJA by local or central testing received T-DXd 6.4 mg/kg or RAM + PTX (**Figure 1**)
- At prespecified time points, patient-reported outcomes were assessed using the FACT-Ga scale and the EuroQol 5-dimension, 5-level (EQ-5D-5L) visual analog scale (VAS)
 - FACT-Ga subscales included emotional well-being (EWB), functional well-being (FWB), physical well-being (PWB), social well-being (SWB), and gastric cancer subscale (GaCS)
 - Subscale scores were combined to obtain the FACT-general (FACT-G; EWB + FWB + PWB + SWB) and FACT-Ga (FACT-G + GaCS) total scores[14]
- TTFD was analyzed for the EQ-5D-5L VAS scale and FACT-Ga total scores and subscales
- TTFD was defined as the time from the date of randomization to the date of the assessment when scales decreased by the following amounts:
 - EQ-5D-5L-VAS: decrease of ≥7 points[14,15]
 - FACT-G: decrease of ≥7.8 points[14,16]
 - FACT-Ga total score: decrease of ≥15.1 points[14,16]
 - EWB, FWB, PWB, and SWB: decrease of ≥3 points[14,16]
 - GaCS: decrease of ≥8.2 points[14,16]
- The mean CFB in the FACT-Ga total score and subscales was assessed, and a mixed model for repeated measures (MMRM) was applied to analyze CFB over time, accounting for within-patient correlations and missing data[14]

Figure 1. DESTINY-Gastric04 Study Design

Patient Population
- HER2+ (IHC 3+ or IHC 2+/ISH+)[a] metastatic GC/GEJA
- HER2 status confirmed locally or centrally[b] on a recent biopsy obtained after progression on trastuzumab
- ECOG PS 0 or 1
- No clinically active CNS metastases[c]

Stratification factors
- HER2 status (IHC 3+ vs IHC 2+/ISH+)
- Geography (Asia [excluding mainland China] vs Western Europe vs mainland China/rest of world)
- Time to progression on 1L therapy (<6 months vs ≥6 months)

R 1:1 → T-DXd 6.4 mg/kg Q3W / RAM + PTX[d]

Primary Endpoint
- OS

Secondary Endpoints
- PFS (INV)[e]
- Confirmed ORR (INV)[e]
- DCR (INV)[e]
- DOR (INV)[e]
- Safety

Exploratory Endpoints
- PROs[f]

Additional Endpoints
- TEAE EAIRs[g]
- Time to first onset of adjudicated drug-related ILD/pneumonitis[h]
- Adjudicated drug-related ILD/pneumonitis incidence by subgroups
- Change from baseline in the FACT-Ga scale
- Time to deterioration

[a]As classified by 2018 ASCO-CAP guidelines. [b]Study protocol originally mandated HER2 status be determined centrally but was later amended to allow local determination. [c]Clinically active CNS metastases were defined as being untreated and symptomatic or requiring therapy with corticosteroids or anticonvulsants. Patients with clinically inactive CNS metastases could be enrolled. [d]RAM administered as 8 mg/kg on days 1 and 15 of each 28-day cycle and PTX administered as 80 mg/m² on days 1, 8, and 15 of each 28-day cycle. [e]Determined by investigator-based assessment per RECIST v1.1. [f]Based on EQ-5D-5L VAS and FACT-Ga subscales. [g]EAIRs were measured to account for differences in treatment duration between T-DXd and RAM + PTX. [h]Time to first onset of adjudicated ILD (days) was defined as the onset date of first ILD event adjudicated as drug-related - first dose date + 1.

Results

Patient-Reported Outcomes

- Median TTFD for each scale is shown in **Figure 2**
- At data cutoff (October 24, 2024), median TTFD was longer and hazard ratios (HRs) favored T-DXd (n = 246) versus RAM + PTX (n = 248) for the following total score and subscales:
 - FACT-G total score (3.5 vs 2.8 months: HR, 0.74; nominal *P* = 0.015)
 - FACT-Ga EWB (5.1 vs 3.7 months: HR, 0.74; nominal *P* = 0.026)
 - FACT-Ga SWB (3.8 vs 2.5 months: HR, 0.71; nominal *P* = 0.006)

Figure 2. TTFD by EQ-5D-5L VAS and FACT-Ga total score and subscales

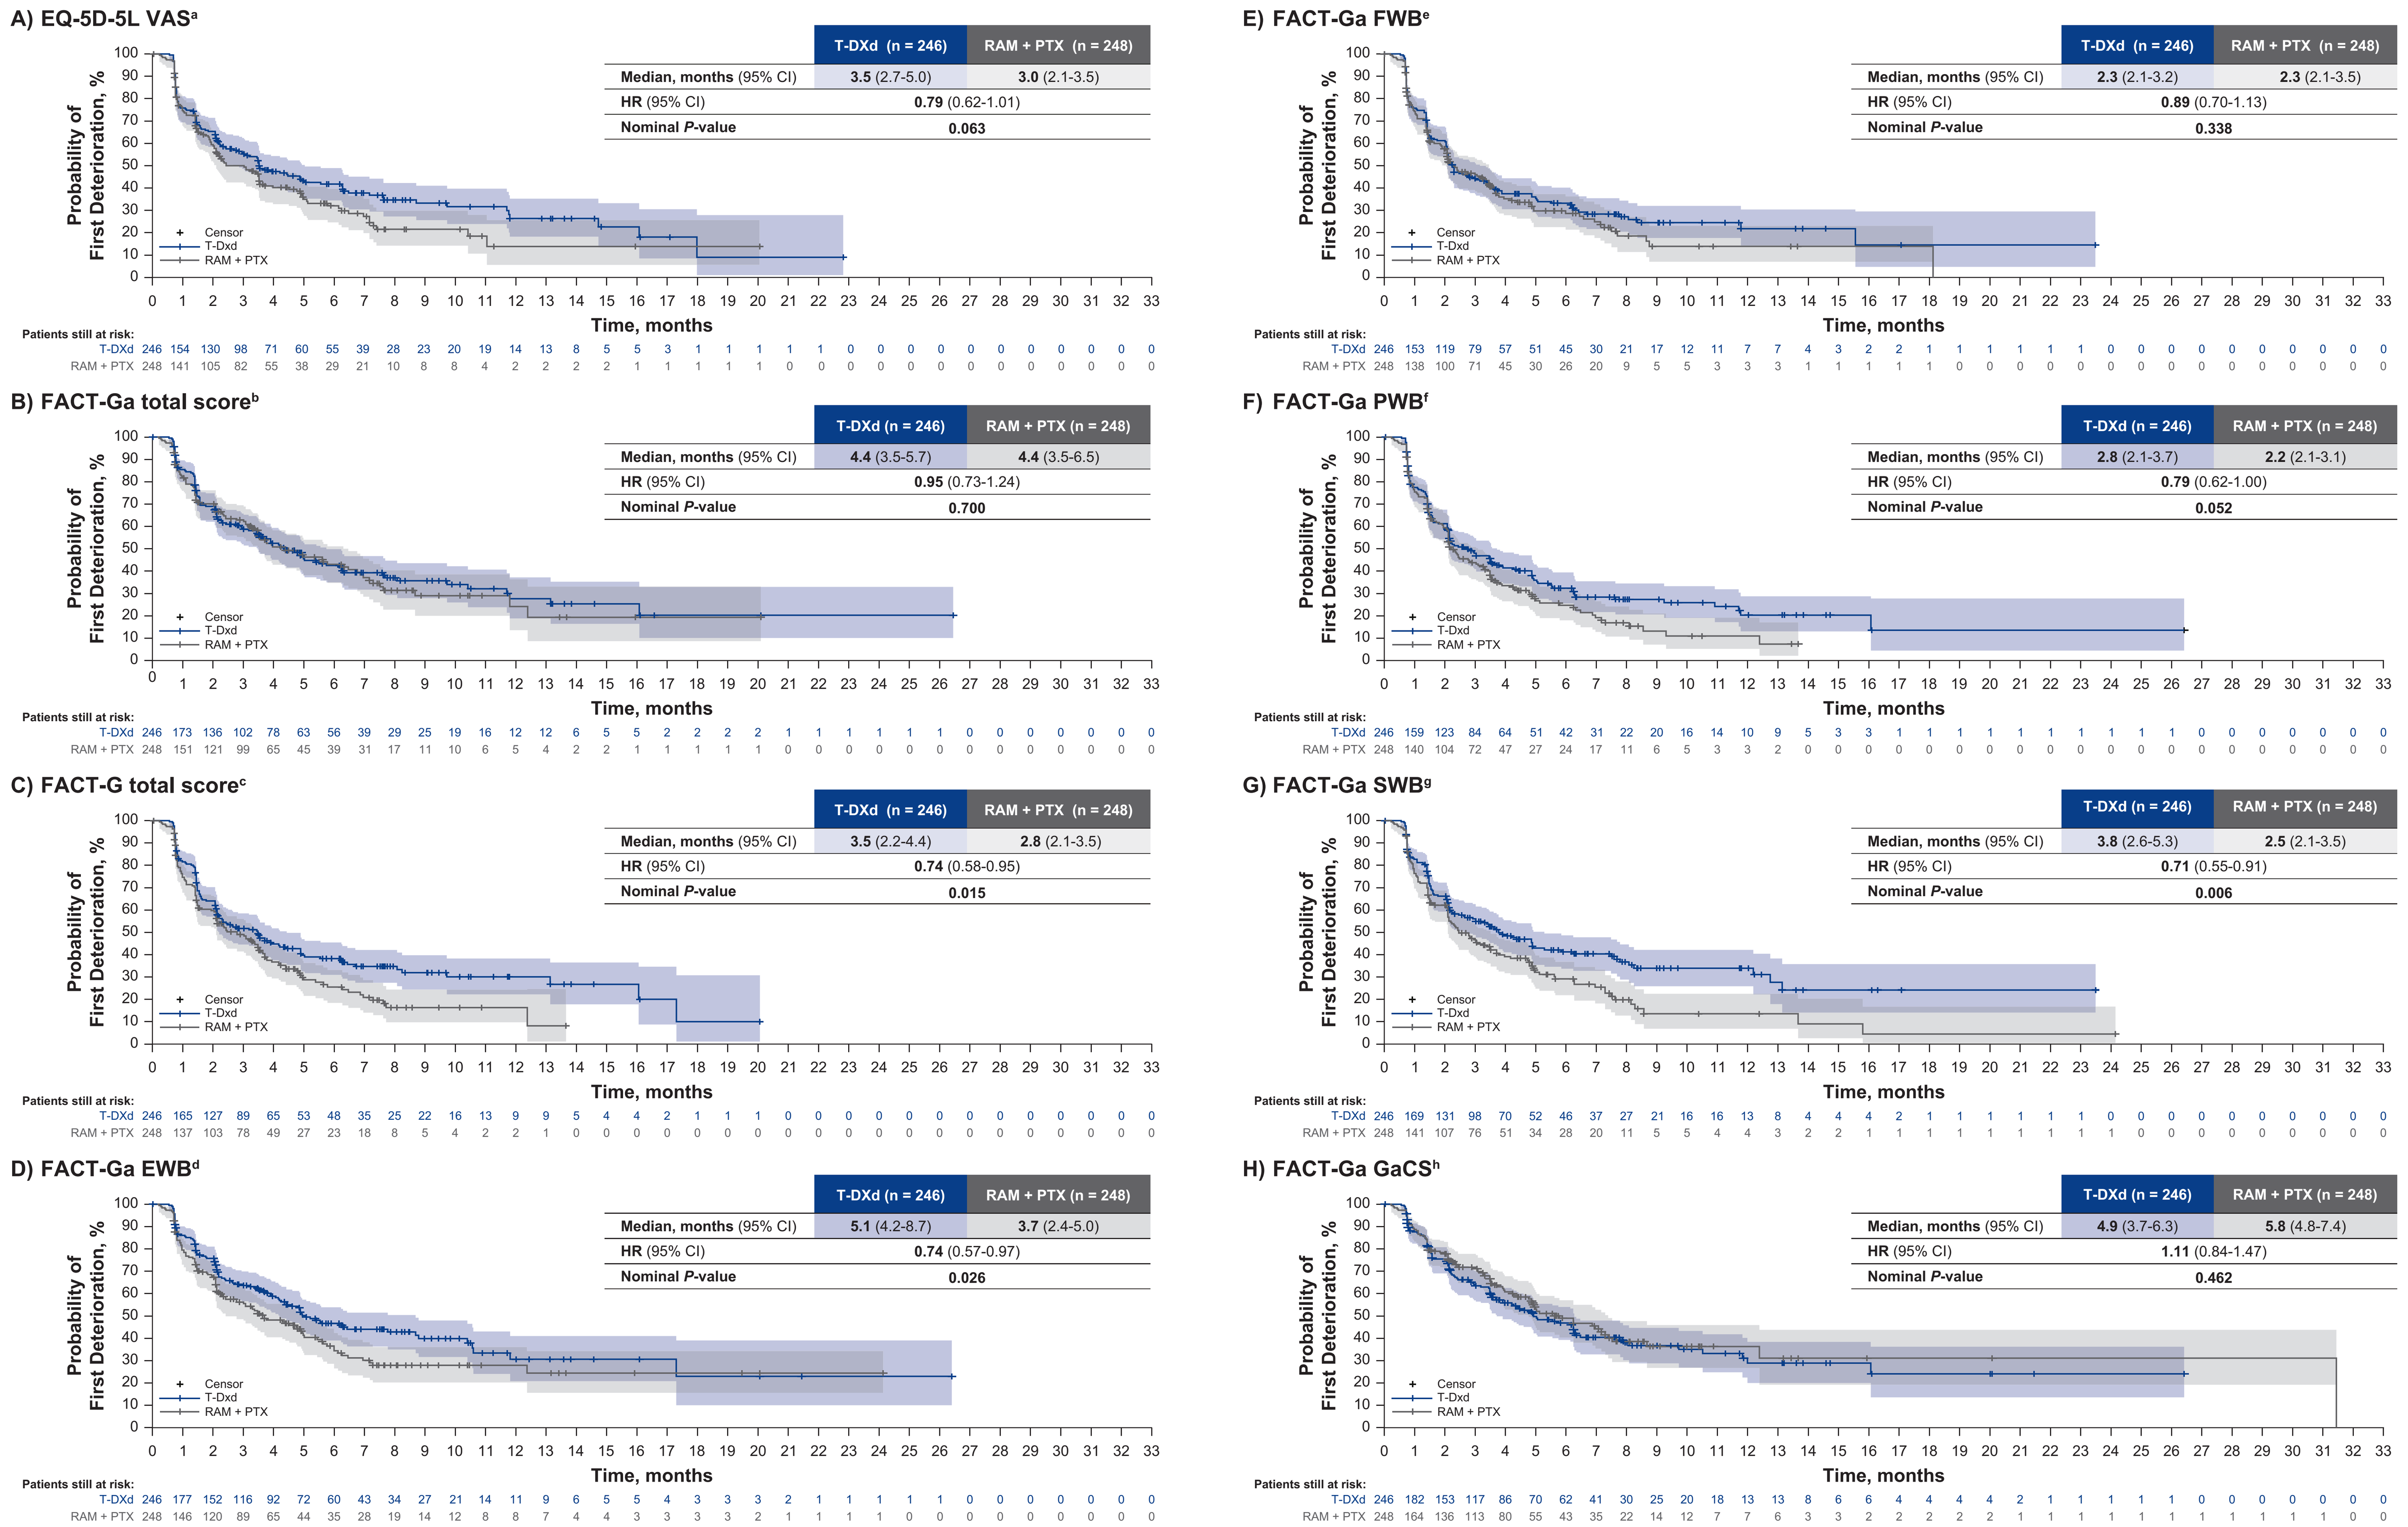
A) EQ-5D-5L VAS[a]
B) FACT-Ga total score[b]
C) FACT-G total score[c]
D) FACT-Ga EWB[d]
E) FACT-Ga FWB[e]
F) FACT-Ga PWB[f]
G) FACT-Ga SWB[g]
H) FACT-Ga GaCS[h]
Probability of First Deterioration, %
Time, months
Censor
T-Dxd
RAM + PTX
Patients still at risk:

Panel	Median, months (95% CI) T-DXd (n = 246)	Median, months (95% CI) RAM + PTX (n = 248)	HR (95% CI)	Nominal *P*-value
A) EQ-5D-5L VAS	3.5 (2.7-5.0)	3.0 (2.1-3.5)	0.79 (0.62-1.01)	0.063
B) FACT-Ga total score	4.4 (3.5-5.7)	4.4 (3.5-6.5)	0.95 (0.73-1.24)	0.700
C) FACT-G total score	3.5 (2.2-4.4)	2.8 (2.1-3.5)	0.74 (0.58-0.95)	0.015
D) FACT-Ga EWB	5.1 (4.2-8.7)	3.7 (2.4-5.0)	0.74 (0.57-0.97)	0.026
E) FACT-Ga FWB	2.3 (2.1-3.2)	2.3 (2.1-3.5)	0.89 (0.70-1.13)	0.338
F) FACT-Ga PWB	2.8 (2.1-3.7)	2.2 (2.1-3.1)	0.79 (0.62-1.00)	0.052
G) FACT-Ga SWB	3.8 (2.6-5.3)	2.5 (2.1-3.5)	0.71 (0.55-0.91)	0.006
H) FACT-Ga GaCS	4.9 (3.7-6.3)	5.8 (4.8-7.4)	1.11 (0.84-1.47)	0.462

[a]TTFD for EQ-5D-5L-VAS was defined as a decrease of ≥7 points. [b]TTFD for FACT-Ga total score was defined as a decrease of ≥15.1 points. [c]TTFD for FACT-G total score was defined as a decrease of ≥7.8 points. [d]TTFD for EWB was defined as a decrease of ≥3 points. [e]TTFD for FWB was defined as a decrease of ≥3 points. [f]TTFD for PWB was defined as a decrease of ≥3 points. [g]TTFD for SWB was defined as a decrease of ≥3 points. [h]TTFD for GaCS was defined as a decrease of ≥8.2 points.
FACT-G total score = EWB + FWB + PWB + SWB; FACT-Ga total score = FACT-G + GaCS. HR was estimated using stratified Cox proportional hazards regression model stratified by the randomization stratification factors (HER2 status), following the pooling strategy described in the study statistical analysis plan. *P*-value is based on stratified log-rank test.

- Mean CFB scores in all FACT-Ga subscales remained stable, with no clinically meaningful changes observed with T-DXd or RAM + PTX (**Figure 3**)

Figure 3. CFB in FACT-Ga total scores and subscales

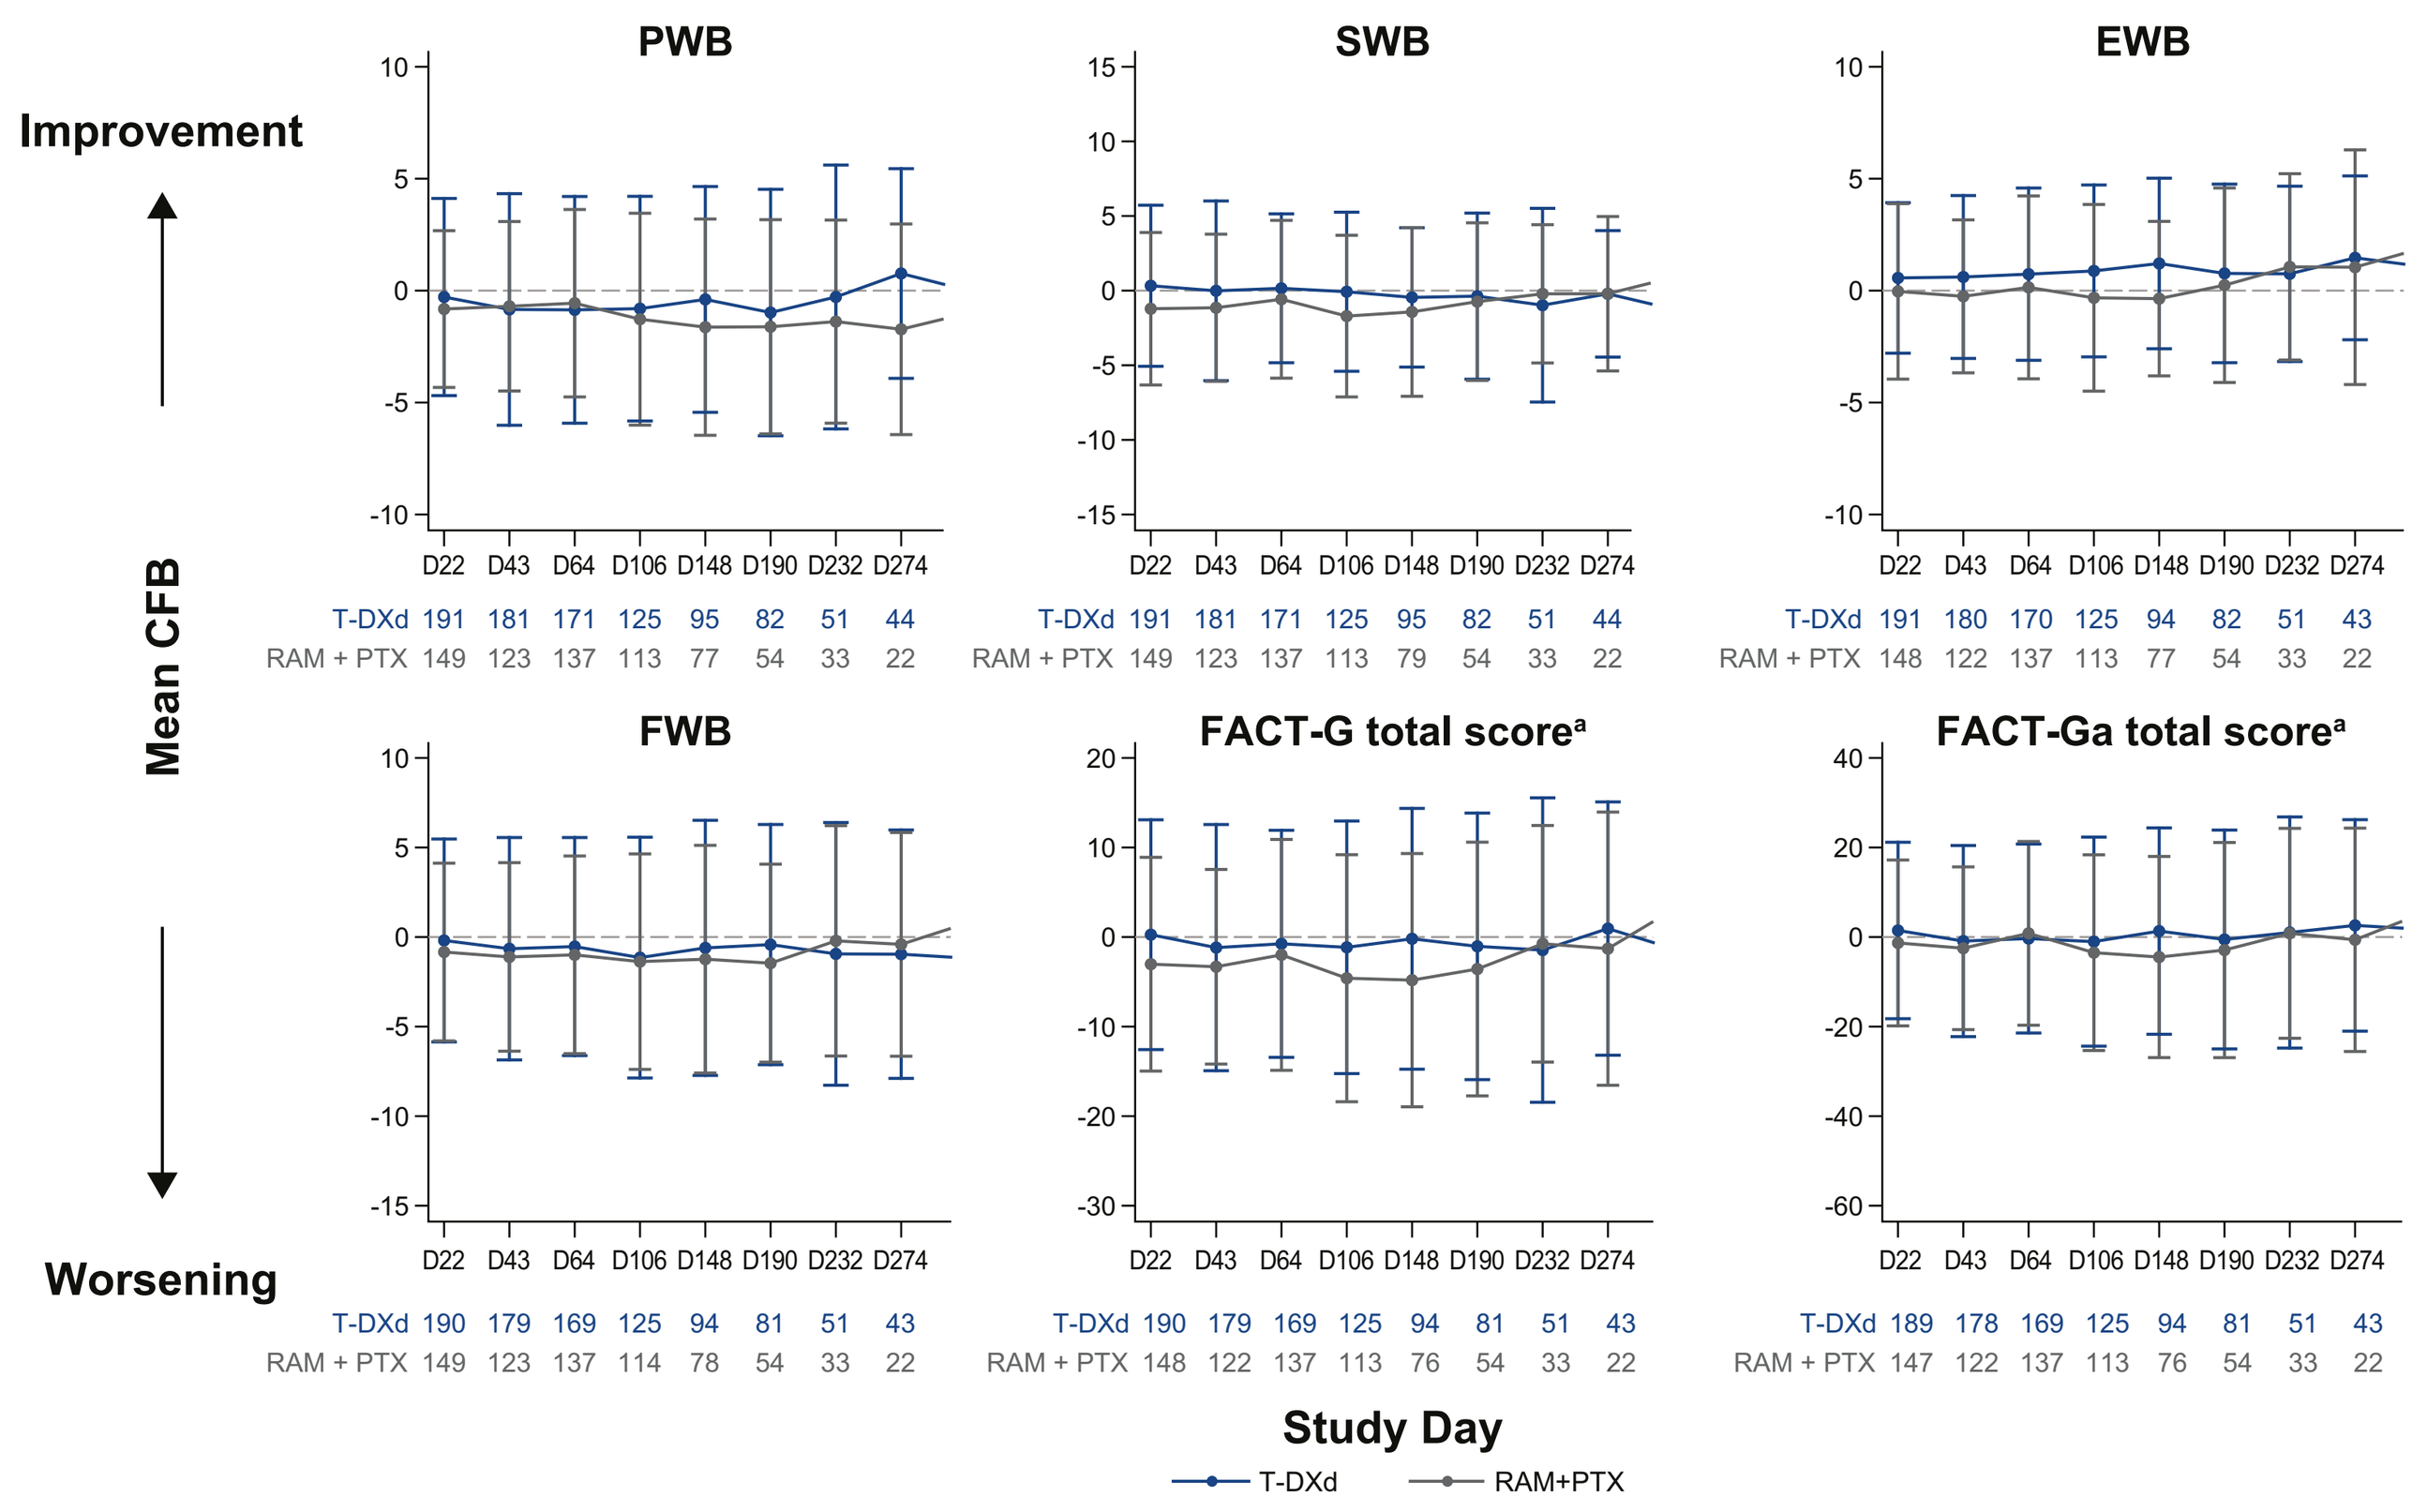
Improvement
Worsening
Mean CFB
PWB
SWB
EWB
FWB
FACT-G total score[a]
FACT-Ga total score[a]
D22 D43 D64 D106 D148 D190 D232 D274
Study Day
T-DXd
RAM+PTX

[a]Previously presented at ESMO Congress 2025, Poster 2099P.[13]
Results for a treatment arm were no longer considered informative once the number of patients who had the specified visit dropped below 10%, which occurred after day 274.

- MMRM analysis of CFB results favored T-DXd versus RAM + PTX for the FACT-G total score (nominal *P* = 0.023), and the FACT-Ga FWB (nominal *P* = 0.045) and SWB (nominal *P* = 0.004) subscales (**Table 1**)

Table 1. MMRM estimated differences in CFB for HRQoL domains between T-DXd and RAM + PTX arms

	Estimate (95% CI)	Nominal *P*-value
FACT-G total score	2.95 (0.40-5.49)	0.023
FACT-Ga total score	2.00 (–1.98-5.98)	0.323
FACT-Ga PWB	0.19 (–0.62-0.99)	0.646
FACT-Ga EWB	0.65 (–0.02-1.32)	0.055
FACT-Ga GaCS	–0.53 (–2.48-1.43)	0.598
FACT-Ga FWB	1.07 (0.02-2.11)	0.045
FACT-Ga SWB	1.45 (0.48-2.42)	0.004

FACT-G total score = EWB + FWB + PWB + SWB; FACT-Ga total score = FACT-G + GaCS. Based on a MMRM including terms for treatment, randomization stratification factors (HER2 status), time of visit, treatment-by-visit interaction and baseline score. The unstructured covariance structure was used to model the correlation within-patient. The analysis only includes assessments up to the time point where there were at least 50 patients remaining in each treatment arm. Difference in CFB (T-DXd - RAM + PTX): positive values indicate higher (better) scores for T-DXd. For the T-DXd and RAM + PTX arms, there were data available for 203-206 and 170-171 patients, respectively.

Abbreviations

CFB, change from baseline; EQ-5D-5L, EuroQol 5-dimension, 5-level; EWB, emotional well-being; FACT, Functional Assessment of Cancer Therapy; FACT-G, Functional Assessment of Cancer Therapy–general; FWB, functional well-being; GaCS, gastric cancer subscale; GC, gastric cancer; GEJA, gastroesophageal junction adenocarcinoma; HER2, human epidermal growth factor receptor 2; HR, hazard ratio; HRQoL, health-related quality of life; IHC, immunohistochemistry; ISH, in situ hybridization; MMRM; mixed model repeated measures; PTX, paclitaxel; PWB, physical well-being; RAM; ramucirumab; SWB, social well-being; T-DXd, trastuzumab deruxtecan; TTFD, time to first deterioration; VAS, visual analog scale.

Acknowledgments

Medical writing support, under the direction of the authors, was provided by Greg Town, BSc, and Andre Wang, PharmD, of ApotheCom, and was funded by Daiichi Sankyo in accordance with Good Publication Practice (GPP) guidelines (http://www.ismpp.org/gpp-2022).

Disclosures

Dr. Shitara reports honoraria from Astellas, AstraZeneca, Bristol Myers Squibb, Eli Lilly, Janssen, and Ono Pharmaceutical; consulting or advisory roles for ALX Oncology Inc, Amgen, Arcus Biosciences Inc., Astellas, AstraZeneca, Bayer, Bristol Myers Squibb, Daiichi Sankyo, GlaxoSmithKline K.K., Guardant Health Japan, HEALIOS K.K., Janssen, Moderna, Inc., MSD, Novartis, Ono Pharmaceutical, Takeda, and Zymeworks Biopharmaceuticals Inc.; and research funding from Amgen, Astellas Pharma, AstraZeneca, Chugai Pharma, Daiichi Sankyo, Eisai, MSD, Ono Pharmaceutical, PPD-SNBL K.K., PRA Health Sciences, Taiho Pharmaceutical, and TORAY.

References

1. Bray F et al. *CA Cancer J Clin.* 2024;74:229-263.
2. Bartley AN et al. *J Clin Oncol.* 2017;35:446-464.
3. Subasinghe D et al. *J Int Med Res.* 2023;51:1-16.
4. Uzunparmak B et al. *Ann Oncol.* 2023;34:1035-1046.
5. Van Cutsem E et al. *Gastric Cancer.* 2015;18:476-484.
6. ENHERTU (fam-trastuzumab deruxtecan-nxki) [prescribing information]. Daiichi Sankyo, Inc.; 2025.
7. Shitara K et al. *N Engl J Med.* 2020;382:2419-2430.
8. Van Cutsem E et al. *Lancet Oncol.* 2023;24:744-756.
9. Daiichi Sankyo, Inc. Press release. January 22, 2026. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202601/20260122_E2.pdf. Accessed May 12, 2026.
10. Daiichi Sankyo, Inc. Press Release. March 23, 2026. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202603/20260323_E3.pdf. Accessed May 15, 2026.
11. Shitara K et al. *N Engl J Med.* 2025;393:336-348.
12. Shitara K et al. Presented at: ASCO Annual Meeting 2025; May 30-June 3, 2025; Chicago, IL, USA.
13. Pietrantonio F et al. Presented at: ESMO Congress 2025; October 17-21, 2025; Berlin, Germany; Poster 2099P.
14. Moehler M et al. *J Clin Oncol.* 2023;41:5388-5399.
15. Pickard AS et al. *Health Qual Life Outcomes.* 2007;5:70.
16. Garland SN et al. *Cancer.* 2011;117:1302-1312.

DESTINY Gastric04