Exposure–response analyses of efficacy and safety with raludotatug deruxtecan (R-DXd), a CDH6-directed antibody–drug conjugate, to inform dose selection for Phase 3 development in platinum-resistant ovarian cancer

236

Felipe K. Hurtado,[1] Emily Schapiro,[2] Elizabeth Lusk,[2] Isabelle Ray-Coquard,[3] Lydia Gaba,[4] Kosei Hasegawa,[5] Alexander Philipovskiy,[6] Izna Ali,[1] YoungJun Yoo,[1] Hiroyuki Inoue,[7] Tsvetomir Mitov,[8] Guillaume Bergthold,[9] Yvonne Lau[1]

[1]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA. [2]A2-Ai LLC, Ann Arbor, MI, USA. [3]Centre Léon Bérard, University Claude Bernard, and GINECO, Lyon, France. [4]Hospital Clinic, Barcelona and GEICO, Barcelona, Spain. [5]Saitama Medical University International Medical Center, Hidaka, Saitama, Japan. [6]Florida Cancer Specialists/Sarah Cannon Research Institute, Lake Mary, FL, USA. [7]Daiichi Sankyo Co., Ltd., Tokyo, Japan. [8]Daiichi Sankyo UK Ltd., Uxbridge, United Kingdom. [9]Daiichi Sankyo France SAS, Rueil-Malmaison, France.

OBJECTIVES

- To characterize the E–R relationships for key efficacy and safety endpoints by pooling data from one Phase 1 and one Phase 2 study of R-DXd
- To investigate the impact of patient-specific covariates on E–R relationships for efficacy and safety
- To inform R-DXd dose selection for Phase 3 clinical investigation in patients with PROC

CONCLUSIONS

- Positive E–R relationships were identified for:
 - Efficacy: ORR (RECIST 1.1), CA-125 response, best tumor response, and PFS, assessed by BICR
 - Safety: Grade ≥3 TEAEs, Grade ≥2 gastrointestinal TEAEs, Grade ≥3 cytopenias (anemia, neutropenia, thrombocytopenia), any-grade ILD/pneumonitis, and TEAEs leading to dose reduction or drug discontinuation
- Covariate analysis demonstrated an association of higher baseline CDH6 expression with improved efficacy, and of Asian race and ECOG PS >0 with increased risk of Grade ≥3 TEAEs
- While higher doses of R-DXd are predicted to maximize efficacy, the 5.6-mg/kg dose provides an overall optimal balance of safety, tolerability, and efficacy compared with 6.4 mg/kg and 4.8 mg/kg
- Integrated E–R analyses, combined with the totality of data from 286 patients, support selection of 5.6 mg/kg Q3W as the optimal R-DXd monotherapy dose for the Phase 2 extension and Phase 3 parts of the REJOICE-Ovarian01 trial,[1] in accordance with the principle of benefit–risk for oncology dose optimization and guidelines from the FDA's Project Optimus

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please visit: https://meetings.asco.org/meetings/2026-asco-annual-meeting/335/17152?presentation=258093

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.

INTRODUCTION

- R-DXd is a novel CDH6-directed ADC comprising a humanized anti-CDH6 IgG1 mAb that is covalently linked to a potent topoisomerase I inhibitor payload (DXd) via a tetrapeptide-based cleavable linker[2]
- CDH6, a mediator of cell–cell adhesion, is expressed in up to 94% of epithelial OC tumors[1,3]
- In a first-in-human Phase 1 trial, R-DXd monotherapy demonstrated encouraging antitumor activity and a manageable safety profile at doses up to 6.4 mg/kg IV Q3W in patients with heavily pretreated OC[4]
- The REJOICE-Ovarian01 trial is evaluating R-DXd in patients with PROC, including a Phase 2 dose-optimization part and a randomized Phase 3 part[1,5]
 - In the primary analysis of the Phase 2 dose-optimization part, R-DXd demonstrated promising efficacy and a manageable safety profile at doses of 4.8, 5.6, and 6.4 mg/kg IV Q3W, with a confirmed ORR of 50.5% across doses[1]
 - The ongoing Phase 3 part is comparing R-DXd 5.6 mg/kg with treatment of physician's choice[1,5]

METHODS

Data source

- These E–R analyses pooled data from patients enrolled in two ongoing clinical studies of R-DXd monotherapy:
 - The first-in-human, dose-escalation and -expansion Phase 1 study (DS6000-A-U101; NCT04707248)[4,6] in patients with advanced OC (N=156) or RCC (N=23) evaluating R-DXd 1.6–9.6 mg/kg IV Q3W (data cutoff: January 10, 2025)
 - The Phase 2 dose-optimization part of the Phase 2/3 REJOICE-Ovarian01 study (NCT06161025)[1] in patients with PROC (N=107), in which patients were randomized 1:1:1 to receive R-DXd 4.8, 5.6, or 6.4 mg/kg IV Q3W and completed ≥18 weeks of follow-up (data cutoff: February 26, 2025)
- The assembled dataset included PK exposure metrics for R-DXd and DXd payload, efficacy and safety outcomes of interest, and individual data on demographics, clinical/disease characteristics, and laboratory and biomarker covariates
 - Neither trial selected patients according to CDH6 expression.[1,4,6] CDH6 expression was assessed in baseline tumor biopsies using an IHC clinical trial assay (SP450; Roche Diagnostics) and quantified as the percentage of viable tumor cells showing CDH6 membrane staining at any intensity (1+/2+/3+)

Analysis approach

- E–R analysis for efficacy was conducted based on pooled data from 241 patients with PROC (**Figure 1**)
 - Endpoints: ORR (percent of patients with PR/CR per RECIST 1.1), CA-125 response (percent of patients with CA-125 objective response per GCIG criteria[7]), best tumor response (best percent change in SLD from baseline), and PFS assessed by BICR per RECIST 1.1
- E–R analysis for safety used pooled data from 286 patients treated with R-DXd across tumor types (OC, RCC)
 - CTCAE-based endpoints: Grade ≥3 TEAEs, Grade ≥2 GI TEAEs (nausea/vomiting), any-grade ILD/pneumonitis, and TEAEs leading to dose reduction or drug discontinuation. The analysis considered only treatment-related TEAEs
 - Laboratory-based endpoints: Grade ≥3 cytopenias (anemia, neutropenia, thrombocytopenia), considering cases of worsening toxicity grade from baseline using the CTCAE grading system
- ORR, CA-125 response, and all safety endpoints, except ILD/pneumonitis, were analyzed by logistic regression. The best tumor response was analyzed using linear regression. The log of R-DXd $C_{avg,0–T}$ best correlated with efficacy endpoints and was chosen as the exposure metric in the exposure–efficacy models
- Time-to-event endpoints (PFS and ILD/pneumonitis) were analyzed using KM curves stratified by exposure quartiles

Figure 1. Analysis dataset

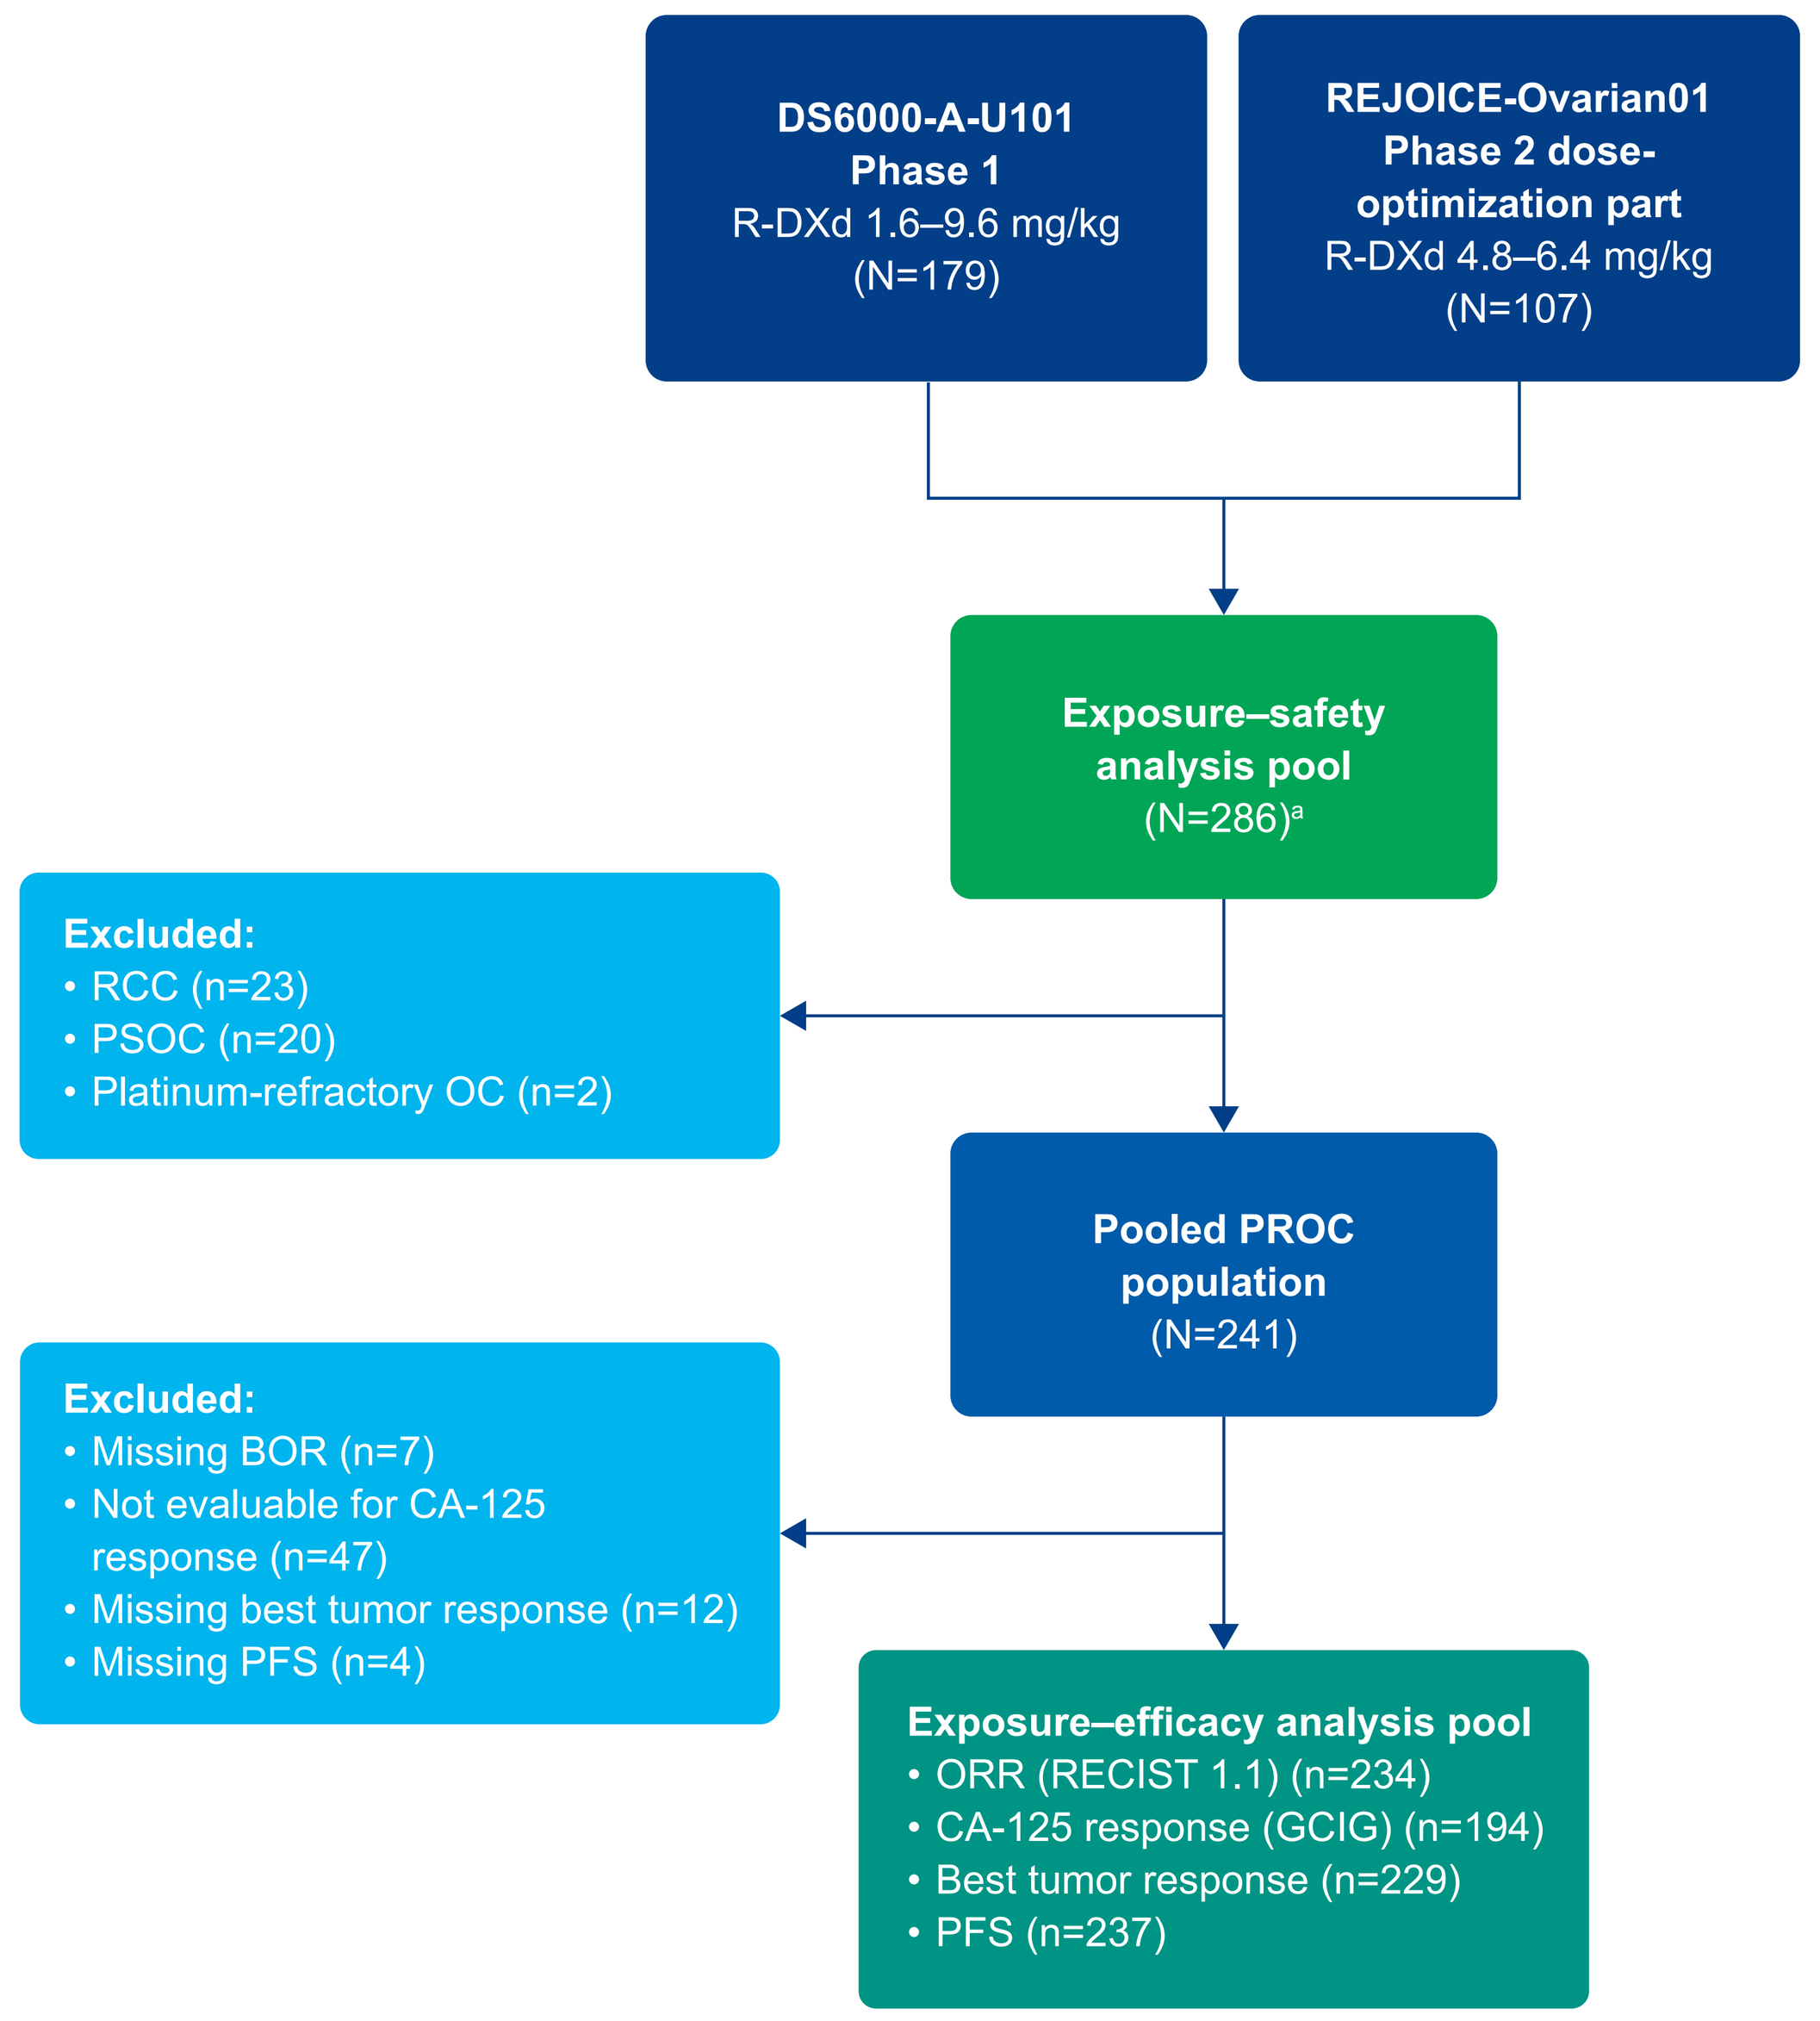
DS6000-A-U101
Phase 1
R-DXd 1.6–9.6 mg/kg
(N=179)
REJOICE-Ovarian01
Phase 2 dose-optimization part
R-DXd 4.8–6.4 mg/kg
(N=107)
Exposure–safety analysis pool
(N=286)a
Excluded:
• RCC (n=23)
• PSOC (n=20)
• Platinum-refractory OC (n=2)
Pooled PROC population
(N=241)
Excluded:
• Missing BOR (n=7)
• Not evaluable for CA-125 response (n=47)
• Missing best tumor response (n=12)
• Missing PFS (n=4)
Exposure–efficacy analysis pool
• ORR (RECIST 1.1) (n=234)
• CA-125 response (GCIG) (n=194)
• Best tumor response (n=229)
• PFS (n=237)

[a]Three patients were excluded from the exposure–safety analysis pool for the evaluation of hematologic AEs due to missing baseline and/or post-baseline laboratory measurements, precluding assessment of worsening toxicity grade.

Exposure metrics

- Exposure metrics for both R-DXd and DXd payload were derived from a joint population PK model (N=286)[8]
 - R-DXd $C_{avg,0–T}$: C_{avg} up to time of event or right-censoring (for PFS), time of first response or best observed outcome (for other efficacy endpoints), time of event (for safety analyses in those with a TEAE), or time of last dose of study drug (for safety analyses in those without a TEAE)
 - R-DXd or DXd C_{max} and C_{avg} at Cycle 1

Covariate analysis

- Numerous prespecified baseline covariates were investigated in the E–R models:
 - Age, sex (safety only), race, ECOG PS, number of prior LOTs, prior treatment (bevacizumab, PARPi; efficacy only), best response to the most recent prior therapy (efficacy only), creatinine clearance (safety only), tumor CDH6 expression (efficacy only), and tumor type (safety only)
- Inclusion of covariates in the final E–R models followed a stepwise forward inclusion/backward elimination procedure to assess statistical significance

Model-based population simulations

- Individual PK exposure was predicted for all patients included in the E–R efficacy analysis population (N=241) for R-DXd doses of clinical interest (4.8, 5.6, and 6.4 mg/kg Q3W). Exposure metrics were simulated using the final population PK model and observed baseline characteristics. The probabilities of key efficacy and safety outcomes were then predicted from the final E–R models, with uncertainty propagated across 500 simulation replicates. The proportion of patients with the event of interest was summarized within each dose group for each replicate, and the median and 95% CI (2.5th and 97.5th percentiles) were calculated across replicates within each dose group

RESULTS

ORR and best tumor response

- Higher exposure to R-DXd ($C_{avg,0–T}$) correlated with higher probability of achieving an objective response (PR/CR per RECIST 1.1; **Figure 2a**), a CA-125 response (per GCIG criteria; data not shown), and a greater reduction in target lesion size from baseline (**Figure 2b**)

PFS

- A trend toward longer PFS with higher exposure to R-DXd ($C_{avg,0–T}$) was observed by KM analysis (**Figure 3**). The relationship was found to be statistically significant (*P*<0.01) based on the log-rank test
 - Higher R-DXd exposure appeared to be associated with longer time to progression or death
 - Time-to-event efficacy endpoints (PFS, DOR) will be reevaluated when more mature data with longer follow-up are available from the REJOICE-Ovarian01 Phase 2 final analysis

Exposure–safety relationships

- R-DXd or DXd payload exposure was positively correlated with all safety endpoints evaluated (**Figure 4**)
 - As PK exposure increased, so did the probability of patients experiencing TEAEs related to R-DXd
- Positive trends were observed between R-DXd C_{max} and risk of any-grade ILD/pneumonitis, suggesting a dose–response relationship (**Figure 5**)
 - Higher exposure appeared to be associated with a shorter time to first ILD/pneumonitis event, with the log-rank test detecting a significant difference between outcomes within each exposure quartile (*P*<0.05). However, the association was not strictly rank-ordered

Covariate analysis

- Covariate analysis showed: (i) baseline CDH6 protein expression was positively associated with ORR and best tumor response and (ii) Asian race and baseline ECOG PS >0 were associated with an increased risk of Grade ≥3 TEAEs (data not shown)

Model-based dose–response simulations

- Simulations demonstrated a dose–response relationship for both efficacy and safety endpoints (**Figure 6**)
- The 5.6-mg/kg Q3W dose of R-DXd is predicted to maximize efficacy while maintaining a superior benefit–risk profile compared with 4.8-mg/kg and 6.4-mg/kg doses

Figure 2. Observed and model-predicted E–R relationships for ORR (a) and best tumor response (b) per RECIST 1.1

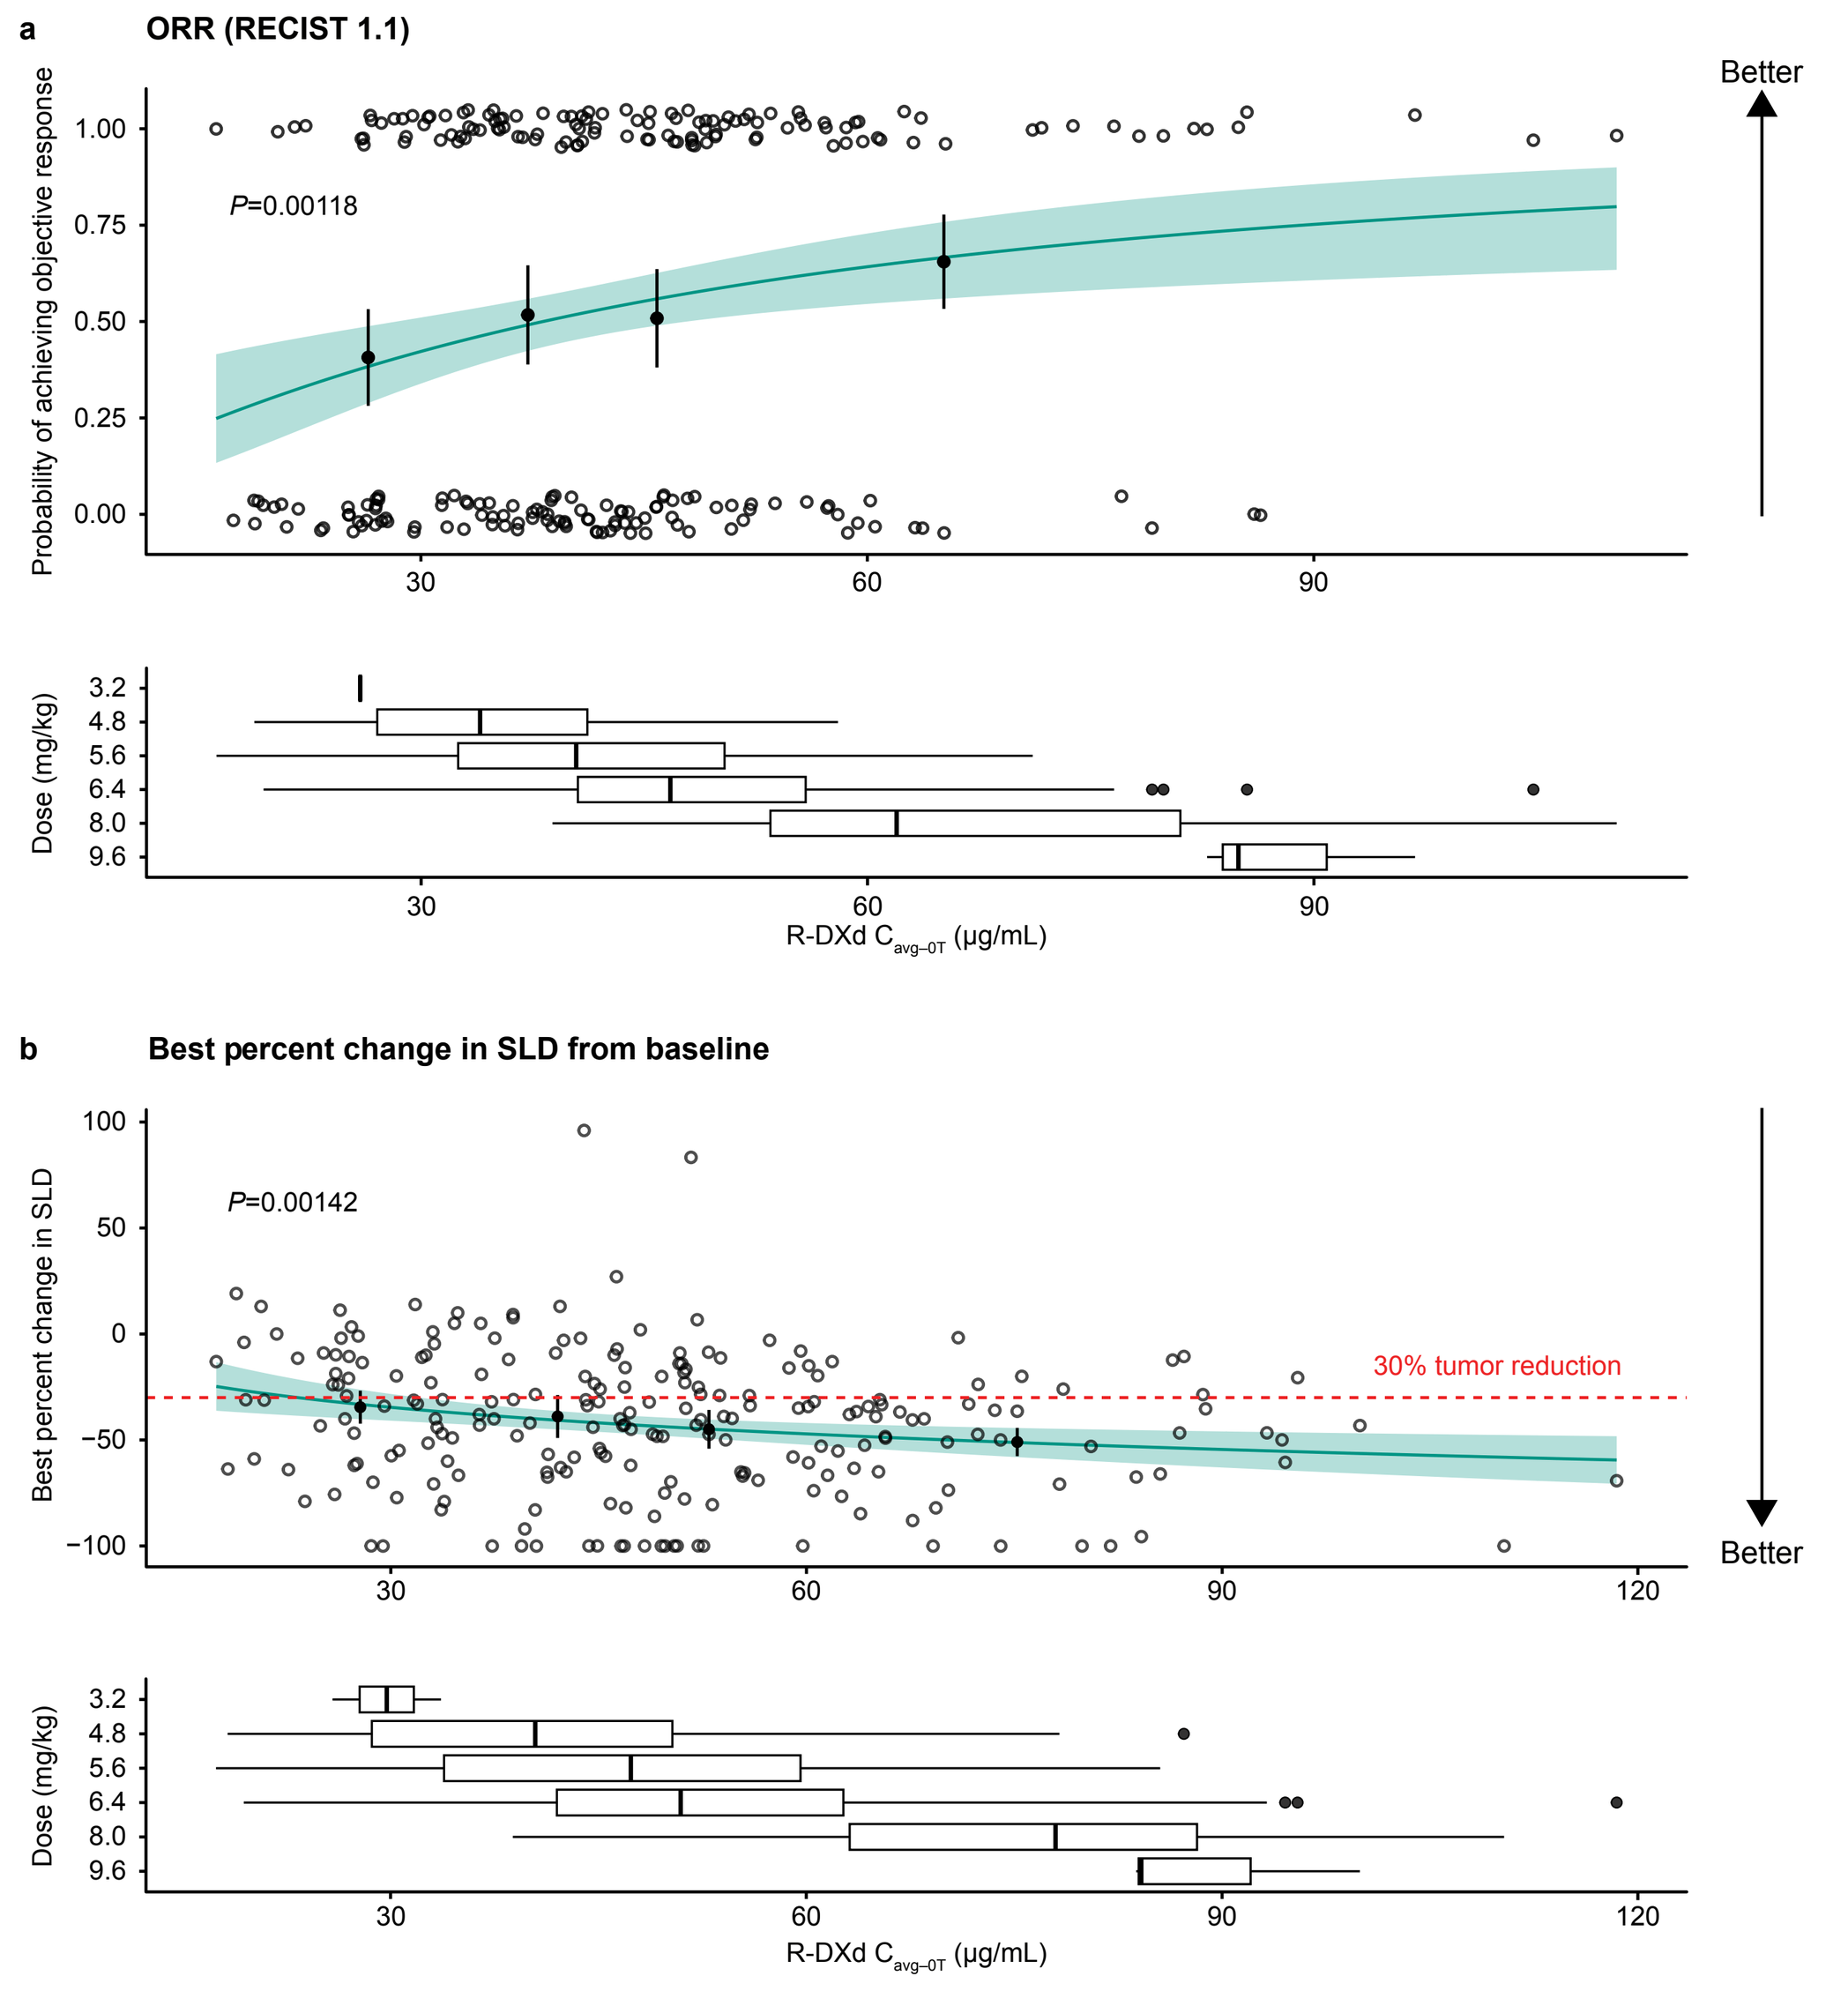
a
ORR (RECIST 1.1)
P=0.00118
Better
Probability of achieving objective response
Dose (mg/kg)
R-DXd Cavg-0T (μg/mL)
b
Best percent change in SLD from baseline
P=0.00142
30% tumor reduction
Better
Best percent change in SLD
Dose (mg/kg)
R-DXd Cavg-0T (μg/mL)

Figure 3. PFS by R-DXd exposure $C_{avg,0–T}$ quartiles

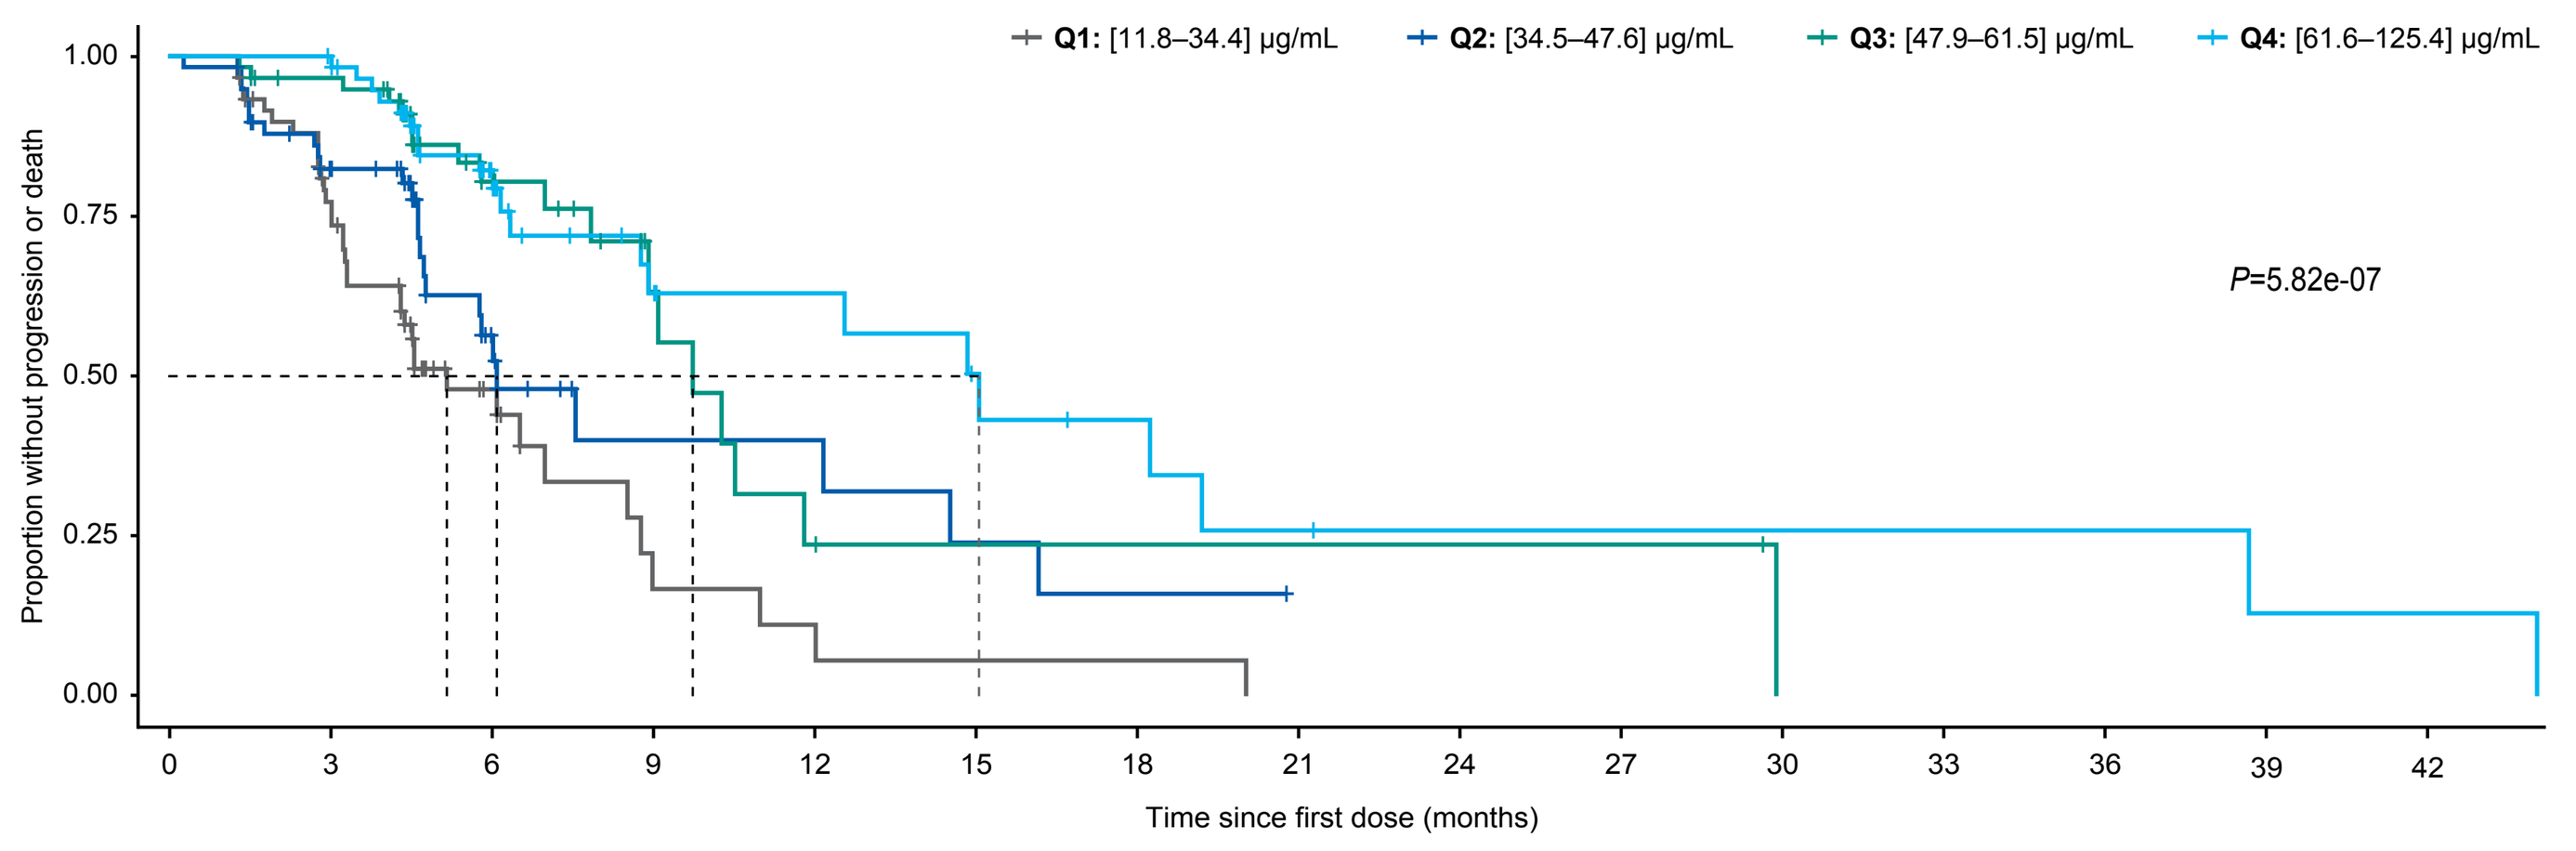
Q1: [11.8–34.4] μg/mL
Q2: [34.5–47.6] μg/mL
Q3: [47.9–61.5] μg/mL
Q4: [61.6–125.4] μg/mL
P=5.82e-07
Proportion without progression or death
Time since first dose (months)

Exposure quartile	Exposure range (μg/mL)	Patients at start (n)	Events (n)	mPFS, months (95% CI)
$C_{avg,0–T}$ Q1	11.8–34.4	60	36	5.2 (4.3–8.8)
$C_{avg,0–T}$ Q2	34.5–47.6	59	25	6.1 (4.8–NE[a])
$C_{avg,0–T}$ Q3	47.9–61.5	59	18	9.8 (8.9–NE[a])
$C_{avg,0–T}$ Q4	61.6–125.4	59	21	15.1 (8.9–NE[a])

[a]The upper bound of the 95% CI could not be estimated due to insufficient information.

P-value comes from log-rank test comparing quartiles of exposure.

Figure 4. E–R relationships for selected safety endpoints

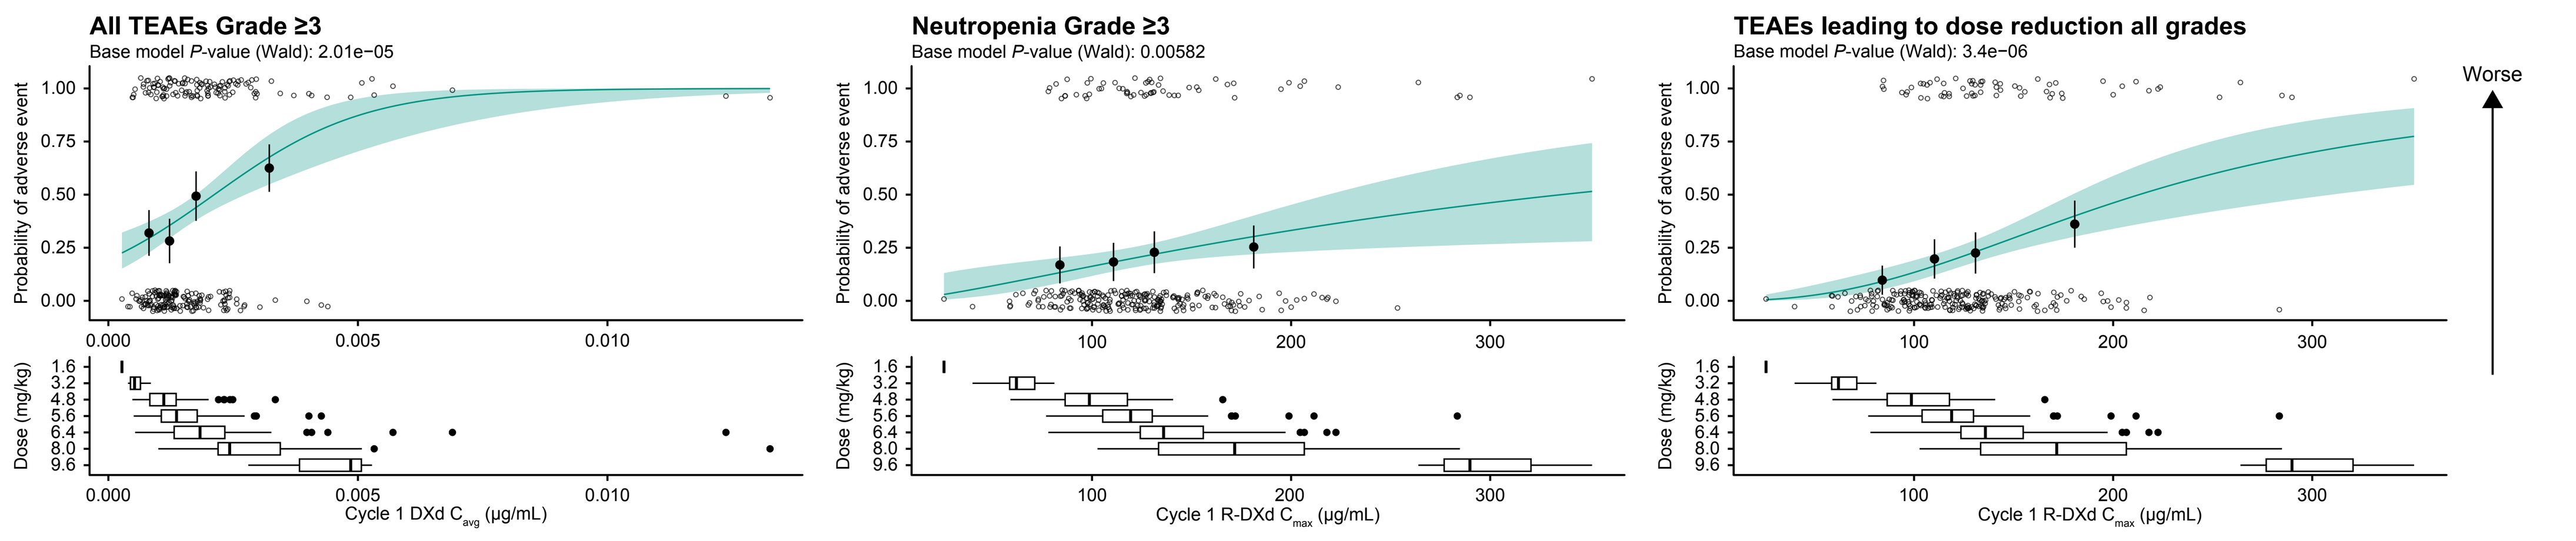
All TEAEs Grade ≥3
Base model P-value (Wald): 2.01e−05
Probability of adverse event
Dose (mg/kg)
Cycle 1 DXd Cavg (μg/mL)
Neutropenia Grade ≥3
Base model P-value (Wald): 0.00582
Cycle 1 R-DXd Cmax (μg/mL)
TEAEs leading to dose reduction all grades
Base model P-value (Wald): 3.4e−06
Worse
Cycle 1 R-DXd Cmax (μg/mL)

P-value calculated using the Wald test.

Figure 5. E–R relationship for any-grade ILD/pneumonitis by Cycle 1 R-DXd C_{max} quartiles

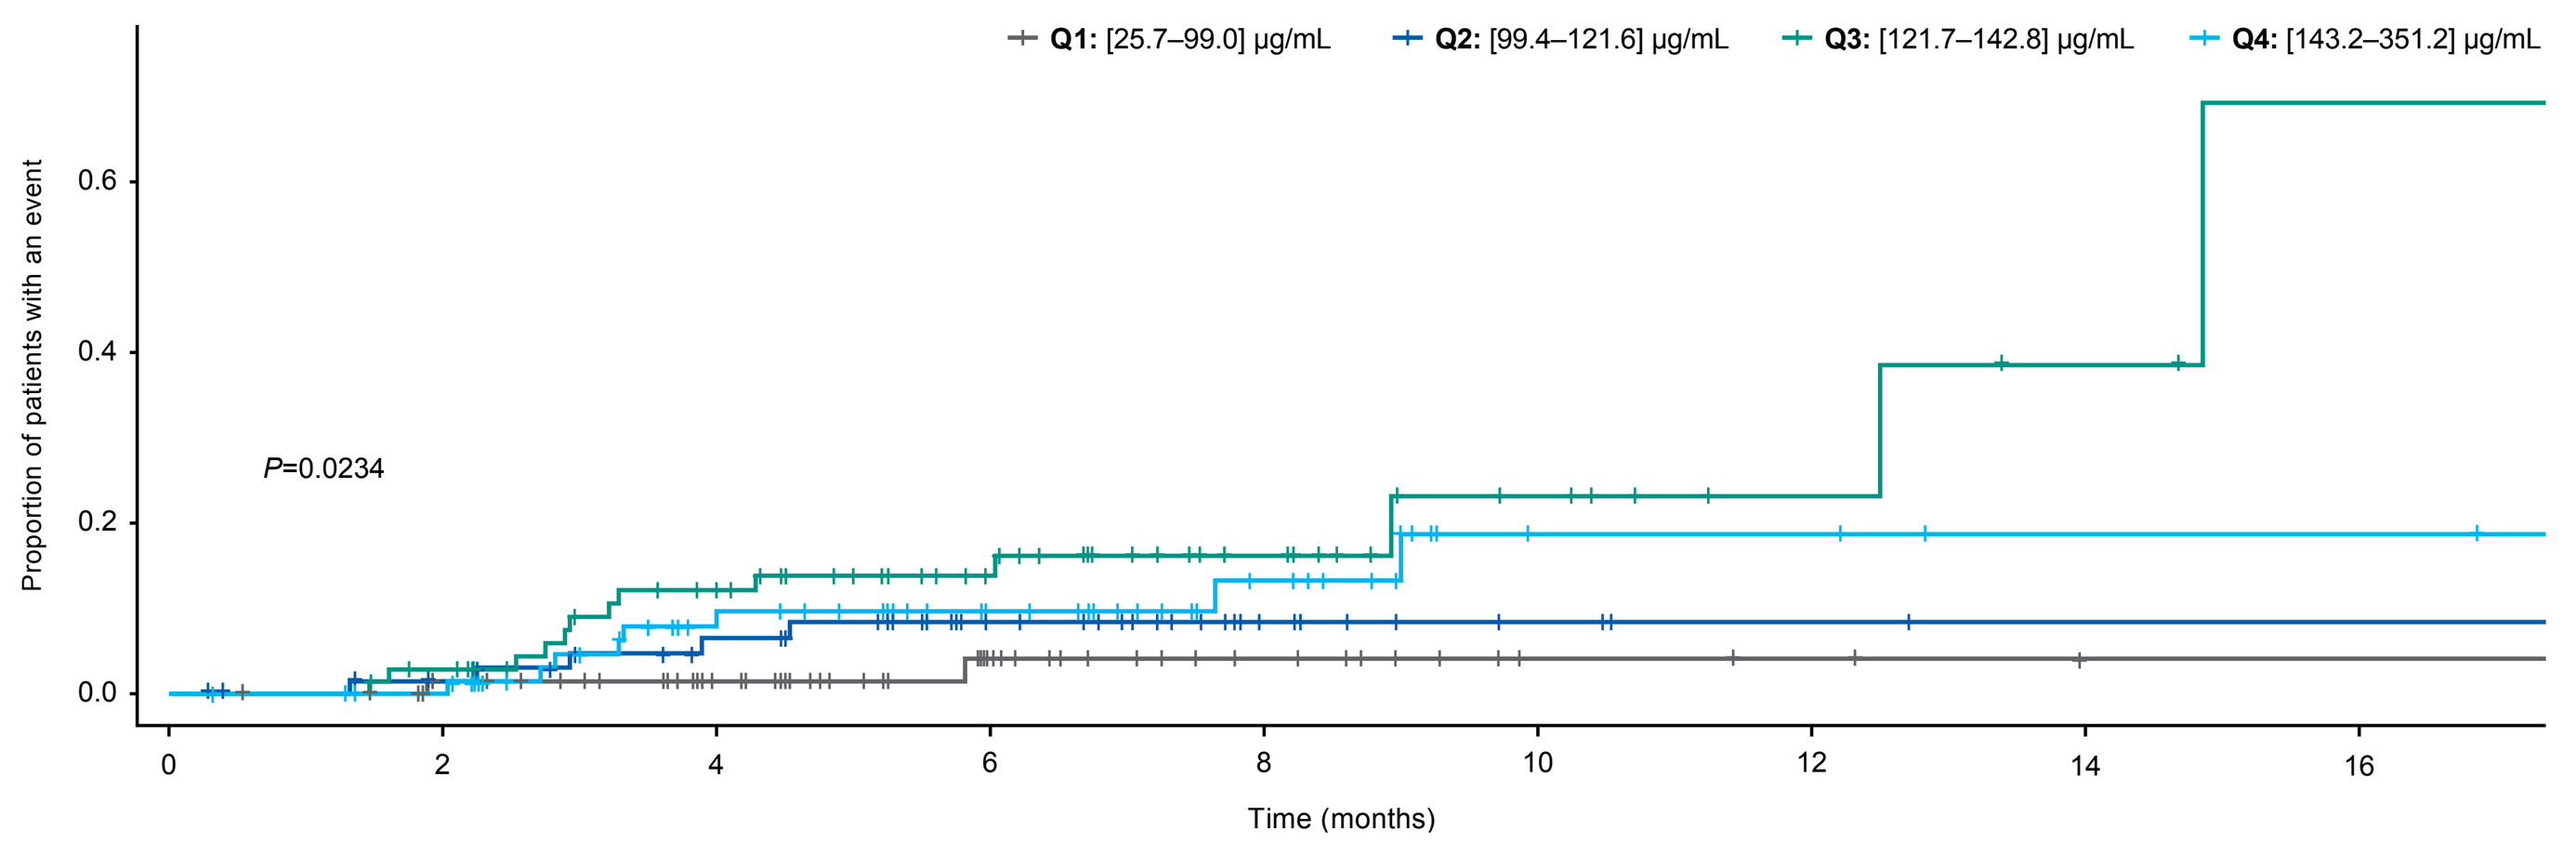
Q1: [25.7–99.0] μg/mL
Q2: [99.4–121.6] μg/mL
Q3: [121.7–142.8] μg/mL
Q4: [143.2–351.2] μg/mL
P=0.0234
Proportion of patients with an event
Time (months)

Exposure quartile	Exposure range (μg/mL)	Patients at start (n)	Events (n)	Patients with any-grade ILD event at 6 months, % (95% CI)
C_{max} Q1	25.7–99.0	72	2	4.1 (0–9.8)
C_{max} Q2	99.4–121.6	71	6	8.4 (1.0–15.2)
C_{max} Q3	121.7–142.8	71	13	13.8 (5.0–21.9)
C_{max} Q4	143.2–351.2	72	11	9.7 (2.0–16.7)

P-value comes from log-rank test comparing quartiles of exposure.

Figure 6. Model-predicted rates of key efficacy and safety outcomes by R-DXd dose

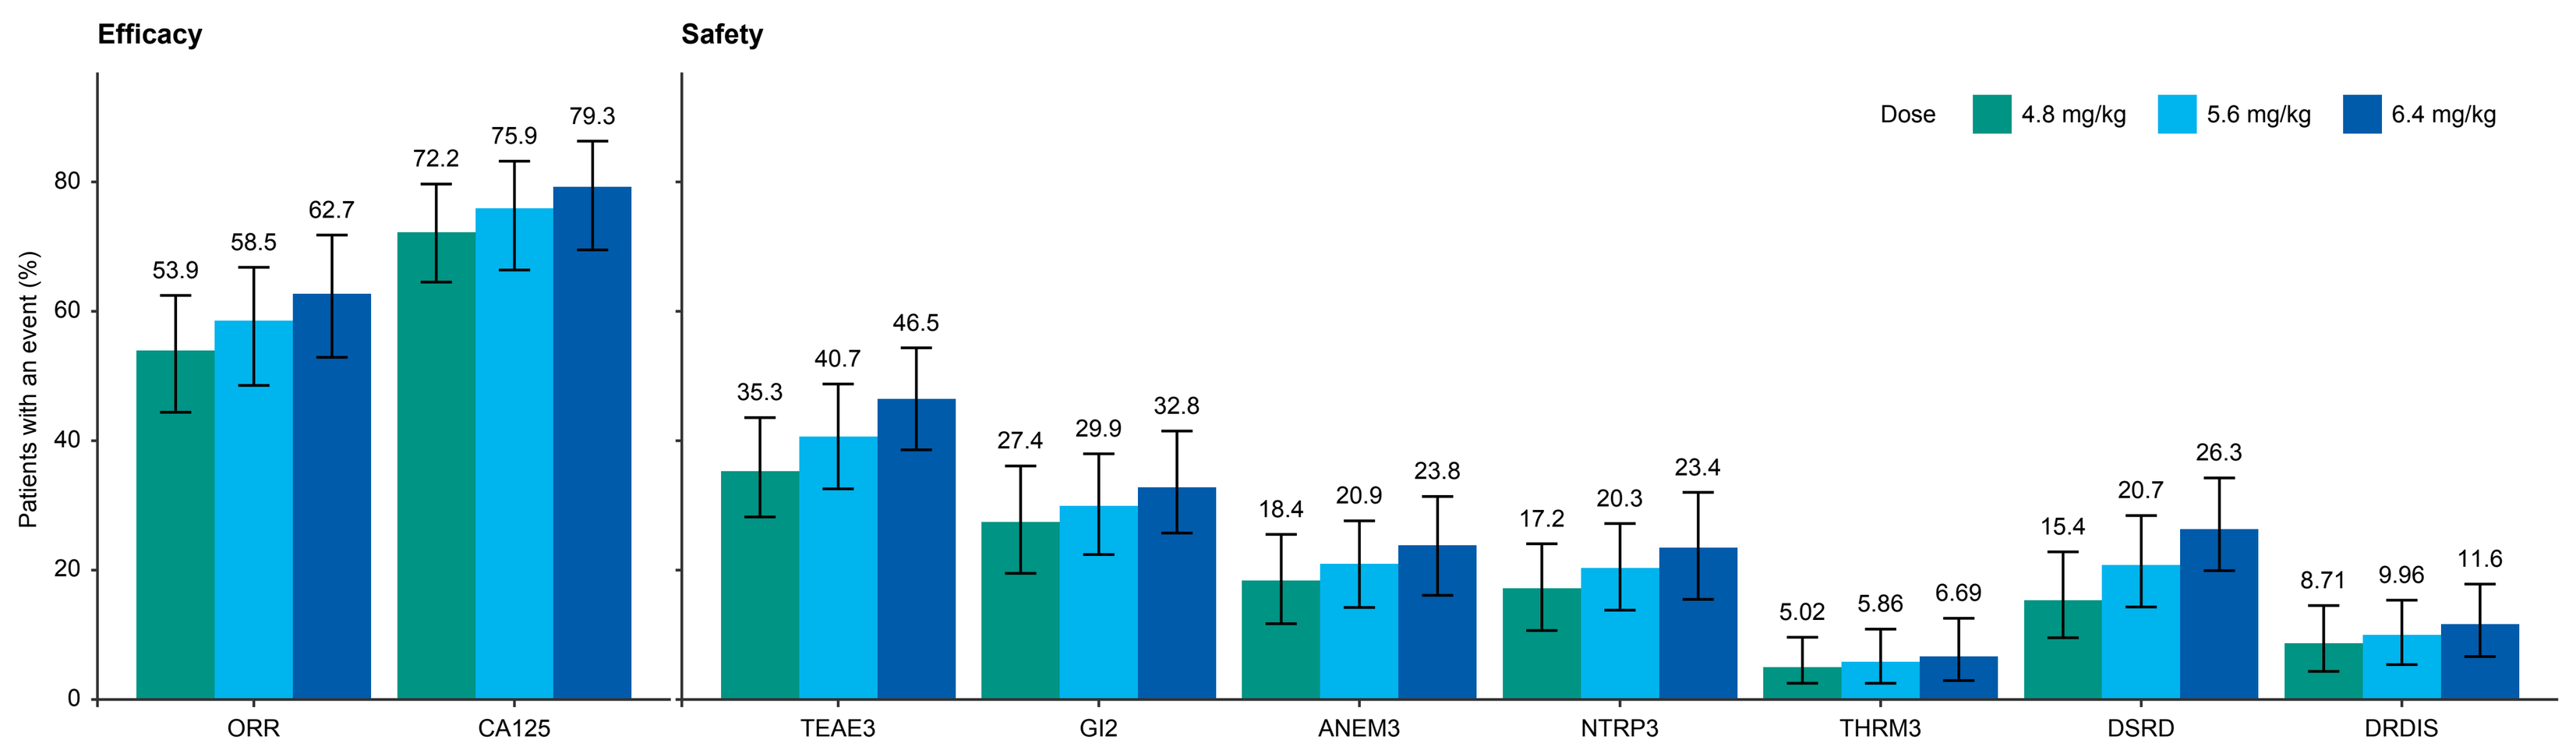
Efficacy
Safety
Dose 4.8 mg/kg 5.6 mg/kg 6.4 mg/kg
Patients with an event (%)
ORR 53.9 58.5 62.7
CA125 72.2 75.9 79.3
TEAE3 35.3 40.7 46.5
GI2 27.4 29.9 32.8
ANEM3 18.4 20.9 23.8
NTRP3 17.2 20.3 23.4
THRM3 5.02 5.86 6.69
DSRD 15.4 20.7 26.3
DRDIS 8.71 9.96 11.6

ORR	ORR (RECIST 1.1)
CA125	CA-125 response
TEAE3	Grade ≥3 TEAEs
GI2	Grade ≥2 GI TEAEs (nausea/vomiting)
ANEM3	Grade ≥3 anemia
NTRP3	Grade ≥3 neutropenia
THRM3	Grade ≥3 thrombocytopenia
DSRD	TEAEs leading to dose reduction
DRDIS	TEAEs leading to drug discontinuation

ACKNOWLEDGMENTS

- We thank the patients, their families, and their caregivers for their participation, and the study staff for their contributions
- This study is sponsored by Daiichi Sankyo, Inc. In October 2023, Daiichi Sankyo entered into a global development and commercialization agreement with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA (MSD) for raludotatug deruxtecan (R-DXd)
- Medical writing support was provided by Anna Atkinson, PhD, of BOLDSCIENCE, Inc., and funded by Daiichi Sankyo, Inc. Editorial support was provided in accordance with Good Publication Practice guidelines (https://www.ismpp.org/gpp-2022)

ABBREVIATIONS

ADC, antibody–drug conjugate; **AE**, adverse event; **BICR**, blinded independent central review; **BOR**, best overall response; **CA-125**, cancer antigen 125; **C_{avg}**, average concentration; **$C_{avg,0–T}$**, average concentration from time 0 to time of event (or record, as applicable); **CDH6**, cadherin 6; **CI**, confidence interval; **C_{max}**, maximum concentration; **CR**, complete response; **CTCAE**, Common Terminology Criteria for Adverse Events; **DOR**, duration of response; **ECOG PS**, Eastern Cooperative Oncology Group performance status; **E–R**, exposure–response; **FDA**, US Food and Drug Administration; **GCIG**, Gynecological Cancer InterGroup; **GI**, gastrointestinal; **IgG1**, immunoglobulin G1; **IHC**, immunohistochemistry; **ILD**, interstitial lung disease; **IV**, intravenous; **KM**, Kaplan–Meier; **LOT**, line of therapy; **mAb**, monoclonal antibody; **m**, median; **NE**, not estimable; **OC**, ovarian cancer; **ORR**, objective response rate; **PARPi**, poly (ADP-ribose) polymerase inhibitor; **PFS**, progression-free survival; **PK**, pharmacokinetics; **PR**, partial response; **PROC**, platinum-resistant ovarian cancer; **PSOC**, platinum-sensitive ovarian cancer; **Q1–Q4**, quartiles of exposure; **Q3W**, every 3 weeks; **RCC**, renal cell carcinoma; **RECIST 1.1**, Response Evaluation Criteria in Solid Tumours version 1.1; **SLD**, sum of longest diameters; **TEAE**, treatment-emergent adverse event.

REFERENCES

1. Ray-Coquard IL, et al. Oral presentation at the European Society for Medical Oncology Congress. October 17–21, 2025; Berlin, Germany. Presentation LBA42.
2. Suzuki H, et al. *Mol Cancer Ther.* 2024;23:257–271.
3. Moore KN, et al. Poster presentation at the Society of Gynecologic Oncology 2026 Annual Meeting on Women's Cancer. April 10–13, 2026; San Juan, Puerto Rico. Presentation 1591.
4. Moore KN, et al. Oral presentation at the Society of Gynecologic Oncology 2024 Annual Meeting on Women's Cancer. March 16–18, 2024; San Diego, CA, USA.
5. Richardson DL, et al. Poster presentation at the American Society of Clinical Oncology Annual Meeting. May 29–June 1, 2026; Chicago, IL, USA. Presentation TPS5637.
6. Hamilton EP, et al. Oral presentation at the American Society of Clinical Oncology Annual Meeting. June 3–7, 2022; Chicago, IL, USA. Abstract 3002.
7. Rustin GJS, et al. *Int J Gynecol Cancer.* 2011;21:419–423.
8. Hurtado FK, et al. Poster presentation at the American Society of Clinical Oncology Annual Meeting. May 29–June 1, 2026; Chicago, IL, USA. Presentation 237.

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting; May 29–June 2, 2026; Chicago, IL, USA.

felipe.kellermannhurtado@daiichisankyo.com