TROPION-Urothelial03: A Phase 2/3 study of datopotamab deruxtecan (Dato-DXd) + platinum chemotherapy vs gemcitabine + platinum chemotherapy in participants with locally advanced or metastatic urothelial carcinoma with progression on or after enfortumab vedotin + pembrolizumab

TPS4642

Matthew D. Galsky,[1] Alexandra Drakaki,[2] Arnab Basu,[3] Bradley McGregor,[4] Jens Bedke,[5] Yohann Loriot,[6] Eiji Kikuchi,[7] Jun Guo,[8] Benjamin Garmezy,[9] Srikala S. Sridhar,[10] Gunnar Klauss,[11] Daisy Lin,[11] Chelsea Liu,[11] Andrea B. Apolo,[12] Thomas Powles[13]

[1]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [2]UCLA Health, Los Angeles, CA, USA; [3]Mayo Clinic, Jacksonville, FL, USA; [4]Dana-Farber/Harvard Cancer Center, Boston, MA, USA; [5]Klinikum Stuttgart Katharinenhospital, Stuttgart, Germany; [6]Gustave Roussy Cancer Institute, Paris, France; [7]St. Marianna University School of Medicine, Kanagawa, Japan; [8]Peking University Cancer Hospital and Research Institute, Beijing, China; [9]Sarah Cannon Research Institute, Nashville, TN, USA; [10]Princess Margaret Cancer Centre, Toronto, ON, Canada; [11]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [12]National Cancer Institute, Bethesda, MD, USA; [13]Barts Cancer Institute, Queen Mary University of London, London, UK

Plain language summary

Why are we performing this research?

- The standard first treatment for patients with locally advanced or metastatic urothelial carcinoma (la/mUC) is enfortumab vedotin (EV) combined with pembrolizumab. However, this therapy stops working for many patients[1,2]
- Chemotherapy using platinum drugs may be recommended as the next treatment option, but only modest benefits are seen, and more effective and tolerable treatments are urgently needed[3–5]
- Datopotamab deruxtecan (Dato-DXd) is an antibody–drug conjugate that consists of an antibody (datopotamab) and an anticancer drug (DXd), joined via a stable cleavable linker[6]
- This study, called TROPION-Urothelial03, will compare Dato-DXd given with platinum-based chemotherapy versus a combination of chemotherapy in patients with la/mUC whose cancer has worsened while or after being treated first with EV and pembrolizumab[7]

How are we performing this research?

- The Phase 2 part of the study will determine the optimal dose of Dato-DXd to be used in the Phase 3 part of the study. In the Phase 3 part of the study, patients will receive either Dato-DXd with platinum-based chemotherapy or gemcitabine with platinum-based chemotherapy[7]
- The main aims of the study are to see how long patients who are given Dato-DXd stay alive overall and remain alive without their cancer growing or spreading[7]

Who will participate in this study?

- We are planning on enrolling approximately 630 patients with la/mUC whose cancer has worsened while receiving or after being treated first with EV and pembrolizumab[7]

Where can I access more information?

- The first results are expected to be available in mid-2029, with the study expected to end in 2030.[7] For more information about TROPION-Urothelial03, please visit https://clinicaltrials.gov/study/NCT07129993. You may also speak to your doctor about clinical studies

1. Powles T, et al. *N Engl J Med.* 2024;390:875–888. 2. Stecca CE, et al. *Can Urol Assoc J.* 2024;18:379–390. 3. Powles T, et al. *Ann Oncol.* 2024;35:485–490. 4. Sternschuss M, et al. *J Clin Oncol.* 2025;43(Suppl 16). Abstract 4573. 5. Kim J, et al. *J Clin Oncol.* 2025;43(Suppl 16). Abstract 785. 6. Okajima D, et al. *Mol Cancer Ther.* 2021;20:2329–2340. 7. ClinicalTrials.gov. https://clinicaltrials.gov/study/NCT07129993. Accessed May 13, 2026.

Background

- Although first-line (1L) therapy with the antibody–drug conjugate (ADC) enfortumab vedotin (EV) plus pembrolizumab has improved outcomes for patients with locally advanced or metastatic urothelial carcinoma (la/mUC),[1–4] approximately half of patients experience disease progression within 2 years of initiating treatment[1,5]
- While there is limited evidence to guide the optimal selection of second-line therapies post–disease progression on or after 1L EV plus pembrolizumab, current guideline recommendations include platinum-based chemotherapy,[3] which has demonstrated modest efficacy in studies to date (complete response, 2.9–10.0%; median duration of response, 3.8 months; median progression-free survival, 3.4–4.4 months; median overall survival, 8.0–12.0 months)[6,7]
- Trophoblast cell surface antigen 2 (TROP2) is a glycoprotein highly expressed in several epithelial tumors, including urothelial carcinoma[8,9]
- Datopotamab deruxtecan (Dato-DXd) is a TROP2-directed ADC comprising an anti-TROP2 monoclonal antibody, a tetrapeptide-based cleavable linker, and a topoisomerase I inhibitor payload (DXd), designed to induce selective tumor-cell death and reduce systemic exposure to the payload[10]
- Dato-DXd has demonstrated durable efficacy and a manageable safety profile in patients with la/mUC both as monotherapy (TROPION-PanTumor01 [NCT03401385]) and in combination with immunotherapy (TROPION-PanTumor03 [NCT05489211])[11,12]
- Here, we describe TROPION-Urothelial03, a Phase 2/3 trial comparing the efficacy and safety of Dato-DXd plus platinum chemotherapy (carboplatin or cisplatin) vs gemcitabine plus platinum chemotherapy in patients with la/mUC with disease progression on or after 1L EV plus pembrolizumab[13]

Enrollment start: September 2025 | Enrollment is ongoing

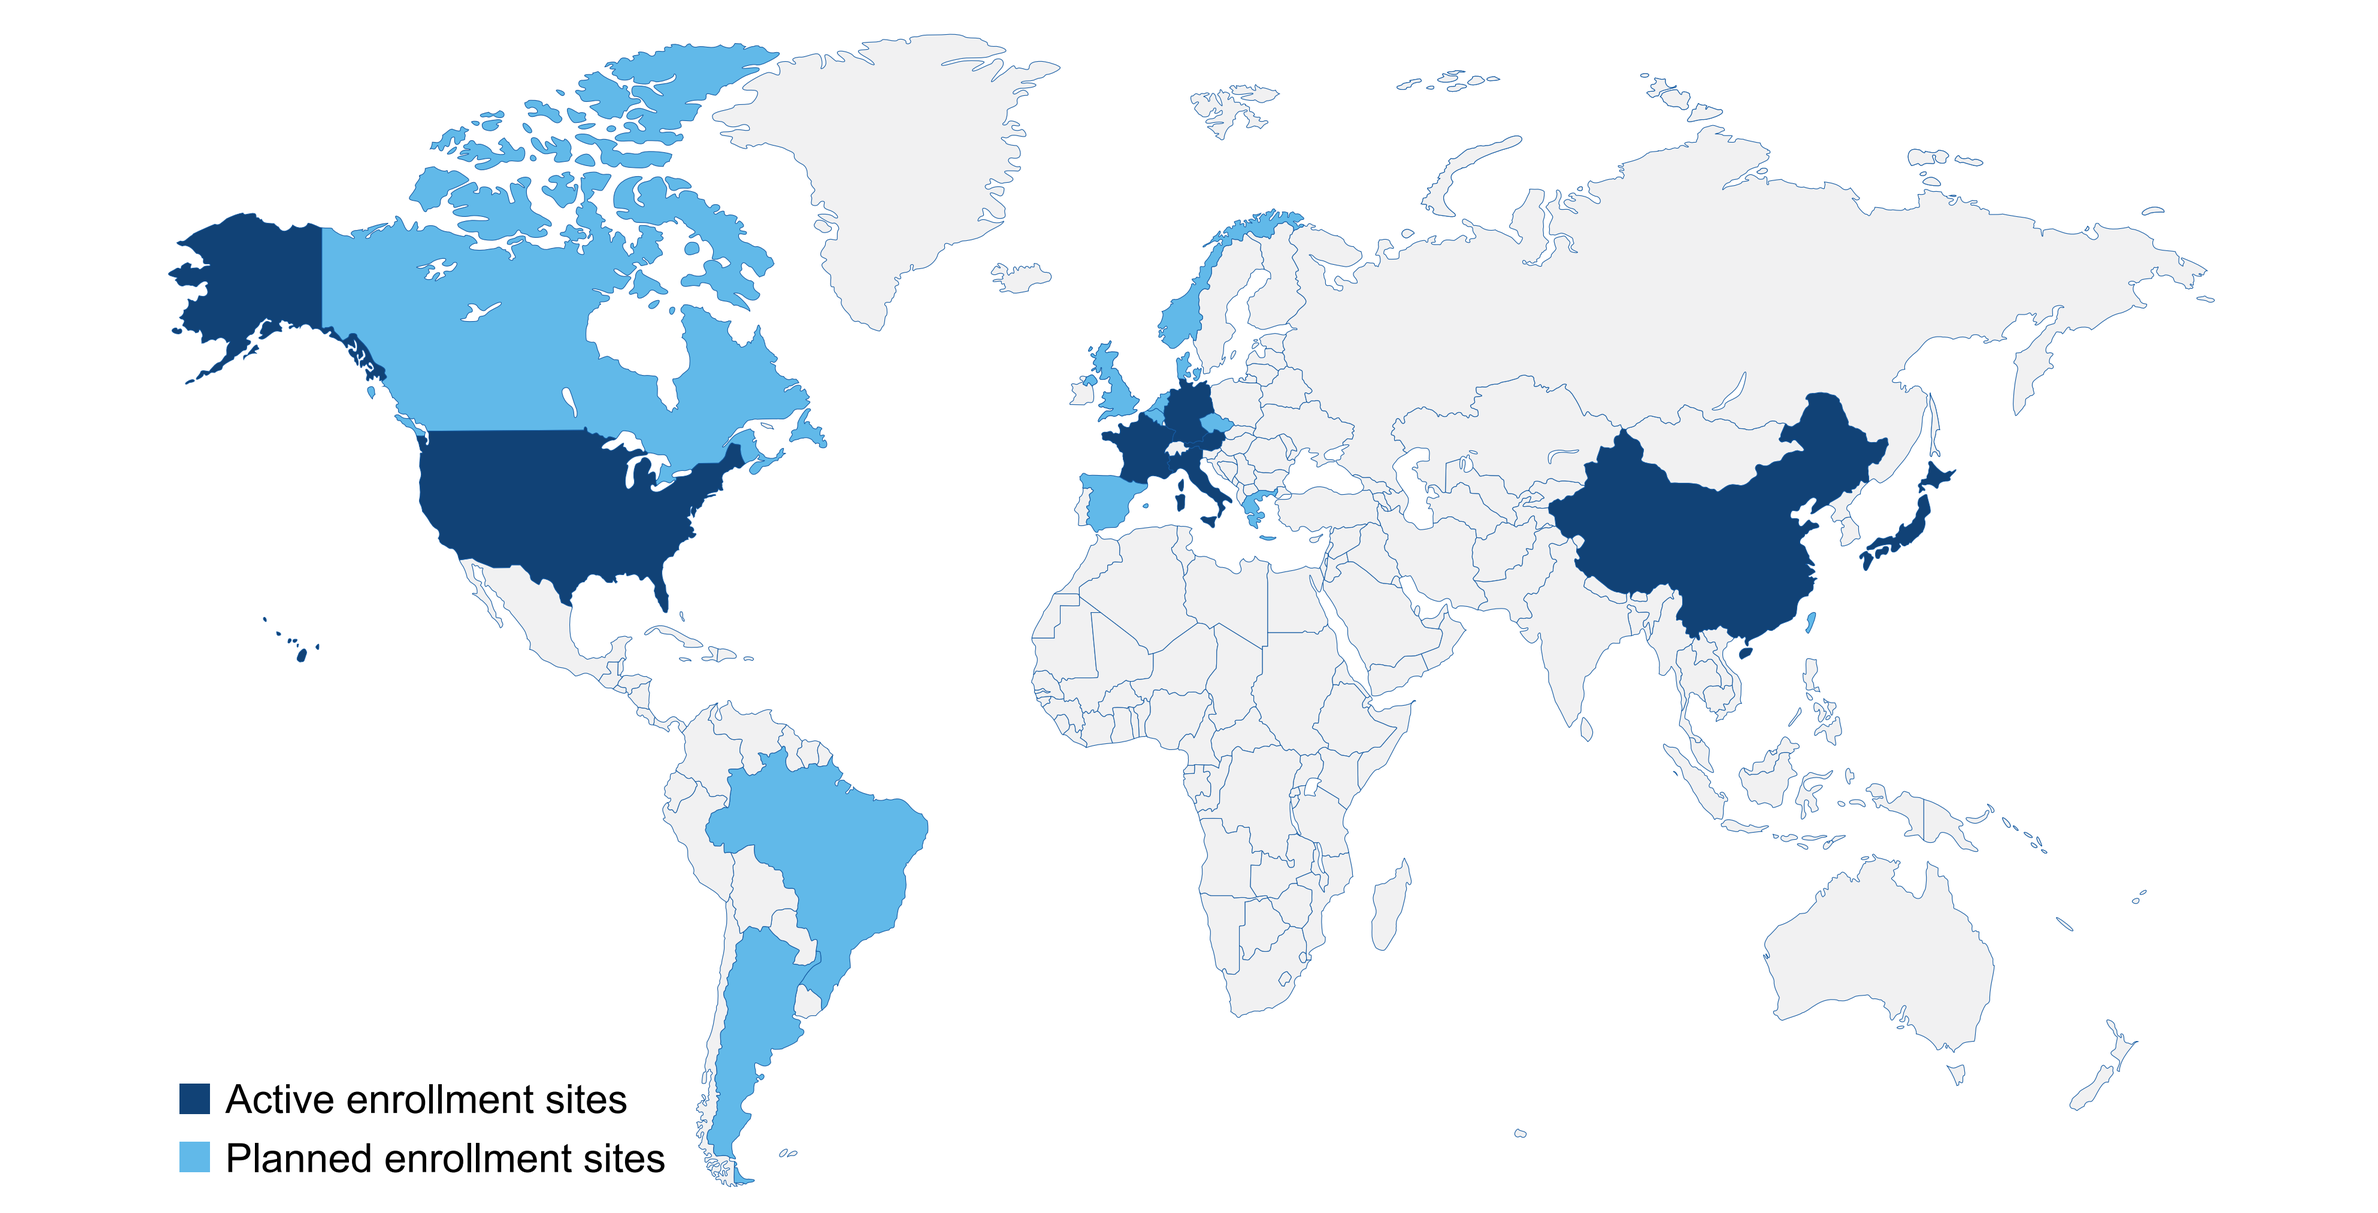

Active enrollment sites
Planned enrollment sites

Countries with active or planned enrollment sites (~276 sites)
Active: Austria, China, France, Germany, Italy, Japan, United States
Planned: Argentina, Belgium, Brazil, Canada, Czech Republic, Denmark, Greece, Netherlands, Norway, Spain, Taiwan, United Kingdom

TROPION-Urothelial03 (NCT07129993): a Phase 2/3 global, multicenter, randomized, open-label study

Key inclusion criteria

- Unresectable la/mUC
- Measurable disease on CT/MRI per RECIST 1.1[a]
- ≥18 years of age or minimum legal adult age
- Radiographic progression or relapse during or after 1L EV plus pembrolizumab
- Tumor tissue sample of sufficient quantity from archival or newly obtained tissue
- Eligible to receive cisplatin- or carboplatin-containing chemotherapy
- Eastern Cooperative Oncology Group performance status of 0 or 1

Phase 2

R 1:1 n≈60

- Dato-DXd 4 mg/kg plus cisplatin or carboplatin Q3W[b] (n≈30)
- Dato-DXd 6 mg/kg plus cisplatin or carboplatin Q3W[b] (n≈30)

Stratification
- Platinum chemotherapy (carboplatin vs cisplatin)

Primary endpoint: ORR by investigator[e]

Phase 3

R 1:1 n≈570

- Dato-DXd at dose determined in Phase 2 plus cisplatin or carboplatin Q3W[b] (n≈285)
- Gemcitabine[c] plus cisplatin or carboplatin Q3W[b,d] (n≈285)

Stratification
- Platinum chemotherapy (carboplatin vs cisplatin)
- Liver metastasis at screening (presence vs absence)
- TTP on 1L EV plus pembrolizumab (<6 vs ≥6 months)

Dual primary endpoints: PFS by BICR[e] and OS

[a]As assessed by investigator. [b]Platinum treatment up to 6 cycles. After completing 6 cycles of Dato-DXd plus platinum chemotherapy, patients will continue Dato-DXd monotherapy Q3W. Before randomization, platinum chemotherapy (carboplatin or cisplatin) will be assigned per the investigator's assessment of cisplatin eligibility. Cisplatin-eligible patients will receive cisplatin. [c]Day 1 and Day 8. [d]Gemcitabine plus cisplatin or gemcitabine plus carboplatin up to 6 cycles. Additional cycles may be allowed in the control group as per local guidelines or local practice. [e]Per RECIST 1.1.

1L, first-line; BICR, blinded independent central review; CT, computed tomography; Dato-DXd, datopotamab deruxtecan; EV, enfortumab vedotin; la/mUC, locally advanced or metastatic urothelial cancer; MRI, magnetic resonance imaging; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; Q3W, every 3 weeks; R, randomization; RECIST 1.1, Response Evaluation Criteria in Solid Tumours, version 1.1; TTP, time to progression.

Key inclusion criteria

- Adults aged ≥18 years[a]
- Histologically or cytologically documented unresectable la/mUC[b]
- Tumor tissue sample of sufficient quantity from archival tissue[c] or newly obtained pretreatment biopsy for exploratory biomarker testing
- Eastern Cooperative Oncology Group performance status of 0 or 1
- Eligible to receive cisplatin- or carboplatin-containing chemotherapy
- Radiographic progression or relapse during or after 1L EV plus pembrolizumab
- Adequate treatment washout period prior to randomization[d]
- Measurable disease on CT/MRI per RECIST 1.1 assessed by investigator
- Adequate organ function based on baseline laboratory testing

[a]If the legal age of consent is >18 years of age, local regulatory requirements will be followed. [b]Patients with la/mUC of the bladder, renal pelvis, ureter, or urethra. Patients with urothelial carcinoma (transitional cell) with squamous differentiation or mixed cell types are eligible if histology is predominantly urothelial. [c]Collected after most recent anticancer treatment and within 12 months before informed consent date is preferred. [d]Defined as major surgery (≥3 weeks [or ≥2 weeks for low-invasive cases]); radiation therapy (≥4 weeks [or ≥2 weeks if palliative single site stereotactic radiation therapy without abdominal radiation]); antibody-based anticancer therapy (≥4 weeks); anticancer chemotherapy (non–antibody-based therapy) and/or retinoid therapy (≥2 weeks or 5 times the half-life of the chemotherapeutic agent, whichever is longer); chloroquine/hydroxychloroquine (>14 days).

1L, first-line; CT, computed tomography; EV, enfortumab vedotin; la/mUC, locally advanced or metastatic urothelial cancer; MRI, magnetic resonance imaging; RECIST 1.1, Response Evaluation Criteria in Solid Tumours, version 1.1.

Key exclusion criteria

- Prior systemic therapy other than the combination of EV plus pembrolizumab for la/mUC
- Prior allogeneic bone marrow or solid organ transplant
- Prior treatment with a TROP2-directed ADC
- Prior brain radiotherapy within 2 weeks or chest radiotherapy within 4 weeks of randomization
- Spinal cord compression or active and untreated CNS metastasis
- History of non-infectious ILD/pneumonitis that required steroids; current or suspected ILD/pneumonitis
- Clinically severe pulmonary compromise resulting from intercurrent pulmonary illnesses
- Uncontrolled or significant cardiovascular disease or cardiovascular events within 6 months prior to randomization

ADC, antibody–drug conjugate; CNS, central nervous system; EV, enfortumab vedotin; ILD, interstitial lung disease; la/mUC, locally advanced or metastatic urothelial cancer; TROP2, trophoblast cell surface antigen 2.

Abbreviations

1L, first-line; **ADC**, antibody–drug conjugate; **BICR**, blinded independent central review; **CNS**, central nervous system; **CT**, computed tomography; **Dato-DXd**, datopotamab deruxtecan; **DCR**, disease control rate; **DOR**, duration of response; **EV**, enfortumab vedotin; **ILD**, interstitial lung disease; **la/mUC**, locally advanced or metastatic urothelial carcinoma; **MRI**, magnetic resonance imaging; **NCI CTCAE v5.0**, National Cancer Institute Common Terminology Criteria for Adverse Events Version 5.0; **ORR**, objective response rate; **OS**, overall survival; **PFS**, progression-free survival; **PRO**, patient-reported outcome; **Q3W**, every 3 weeks; **R**, randomization; **RECIST 1.1**, Response Evaluation Criteria in Solid Tumours, version 1.1; **TROP2**, trophoblast cell surface antigen 2; **TTP**, time to progression; **TTR**, time to response.

Acknowledgments

This study is sponsored by Daiichi Sankyo, Inc. In July 2020, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for datopotamab deruxtecan (Dato-DXd). Medical writing support, under the direction of the authors, was provided by Jessica Gower, PhD, of BOLDSCIENCE, Inc., and was funded by Daiichi Sankyo, Inc. in accordance with Good Publication Practice (GPP) guidelines (http://www.ismpp.org/gpp-2022).

The content in this poster was previously presented at the American Society of Clinical Oncology Genitourinary Cancers Symposium; February 26–28, 2026; San Francisco, CA, USA.

Disclosures

Matthew D. Galsky has served in a consulting or advisory role for AbbVie, Bristol Myers Squibb, EMD Serono, Gilead Sciences, Janssen, Merck, and Pfizer.

Study endpoints

Primary endpoints

Phase 2	Phase 3
ORR by investigator[a]	PFS by BICR[a]
	OS

Secondary endpoints

Phase 2	Phase 3
DOR[b]	PFS by investigator[a]
PFS[b]	ORR[e]
OS	DOR[e]
TTR[b]	TTR[e]
DCR[b,c]	DCR[c,e]
Safety[d]	Safety[d]
Immunogenicity of Dato-DXd (antidrug antibodies)	Immunogenicity of Dato-DXd (antidrug antibodies)
Pharmacokinetics	PROs[f]

[a]Per RECIST 1.1. [b]By investigator per RECIST 1.1. [c]Proportion of patients with a best overall response of confirmed complete response, partial response, or stable disease. [d]Including treatment-emergent adverse events, serious adverse events, and adverse events of special interest (all graded according to the NCI-CTCAE v5.0); deaths; changes from baseline in vital signs; standard clinical laboratory parameters (graded according to the NCI-CTCAE v5.0); electrocardiogram parameters; echocardiogram/multigated acquisition scans; and ophthalmologic findings. [e]By BICR and investigator per RECIST 1.1. [f]European Organization for Research and Treatment of Cancer Quality of Life Questionnaire-muscle invasive bladder cancer 30 (disease symptoms, quality of life, and functioning).

BICR, blinded independent central review; Dato-DXd, datopotamab deruxtecan; DCR, disease control rate; DOR, duration of response; NCI CTCAE v5.0, National Cancer Institute Common Terminology Criteria for Adverse Events Version 5.0; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PRO, patient-reported outcome; RECIST 1.1, Response Evaluation Criteria in Solid Tumours, version 1.1; TTR, time to response.

References

1. Powles T, et al. *N Engl J Med.* 2024;390:875–888.
2. Friedlander TW, et al. *J Clin Oncol.* 2021;39:4528–4528.
3. Flaig TW, et al. *J Natl Compr Canc Netw.* 2024;22:216–225.
4. Stecca CE, et al. *Can Urol Assoc J.* 2024;18:379–390.
5. Jang A and Brown JR. *Explor Target Antitumor Ther.* 2025;6:1002307.
6. Sternschuss M, et al. *J Clin Oncol.* 2025;43(Suppl 16). Abstract 4573.
7. Kim J, et al. *J Clin Oncol.* 2025;43(Suppl 16). Abstract 785.
8. Lombardi P, et al. *Cancers (Basel).* 2023;15:1744.
9. Chou J, et al. *Eur Urol Oncol.* 2022;5:714–718.
10. Okajima D, et al. *Mol Cancer Ther.* 2021;20:2329–2340.
11. Meric-Bernstam F, et al. Oral presentation at the American Society of Clinical Oncology Genitourinary Cancers Symposium. February 13–15, 2025; San Francisco, CA, USA. Presentation 663.
12. Rha SY, et al. Oral presentation at the European Society for Medical Oncology Congress. October 17–21, 2025; Berlin, Germany. Presentation 3072MO.
13. ClinicalTrials.gov. https://clinicaltrials.gov/study/NCT07129993. Accessed May 13, 2026.

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please visit: https://meetings.asco.org/meetings/2026-asco-annual-meeting/335/17145?presentation=266904.

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting; May 29–June 2, 2026; Chicago, IL, USA.

Presenting author email address: matthew.galsky@mssm.edu