TROPION-Lung14: A phase 3 study of osimertinib ± datopotamab deruxtecan as first-line treatment for patients with *EGFR*-mutated locally advanced or metastatic non-small cell lung cancer

Shun Lu,[1] Mariano Provencio Pulla,[2] Aaron Lisberg,[3] Eldsamira Mascarenhas,[4] Junko Tanizaki,[5] Ying Cheng,[6] Hye Ryun Kim,[7] Yinglei Liu,[8] Shengmei Feng,[8] Biao Zhang,[9] Laurence Toms,[10] James Chih-Hsin Yang,[11] Sarah Goldberg[12]

[1]Shanghai Chest Hospital, Medical Oncology Department, Shanghai, China; [2]Hospital Universitario Puerta de Hierro Majadahonda, Department of Medical Oncology, Madrid, Spain; [3]UCLA Health Cancer Care, Santa Monica, Thoracic Medical Oncology, CA, USA; [4]Hospital Sao Rafael, São Paulo, Brazil; [5]Kindai University Faculty of Medicine, Department of Medical Oncology, Osaka, Japan; [6]Jilin Cancer Hospital, Changchun, China; [7]Division of Medical Oncology, Department of Internal Medicine, Yonsei Cancer Center, Yonsei University College of Medicine, Seoul, Republic of Korea; [8]AstraZeneca, Shanghai, China; [9]AstraZeneca, R&D, Gaithersburg, MD, USA; [10]AstraZeneca, R&D, Cambridge, UK; [11]National Taiwan University Hospital, Medical Oncology Department, Taipei City, Taiwan; Yale Cancer Center, New Haven, CT, USA[12]

TPS8647

Plain language summary

Why are we performing this research?

- Epidermal growth factor receptor (EGFR) is a protein that controls cell growth and division; in some cancer cells the gene is mutated and allows cells to grow uncontrollably. Osimertinib is a medicine that blocks the activity of mutated EGFR on cancer cells, stopping their growth[1] and is the recommended treatment for people with *EGFR*-mutated advanced non-small cell lung cancer (NSCLC), for those with *EGFR*-mutated NSCLC where the cancer has been removed by surgery, and for *EGFR*-mutated NSCLC that cannot be removed by surgery and whose disease has not got worse during or after chemoradiotherapy.[2–6] Unfortunately, osimertinib eventually stops working in many people, and the cancer cannot be controlled.[7] Researchers are looking to see if osimertinib would be more effective when it is combined with other medicines. Datopotamab deruxtecan (Dato-DXd) is an antibody-drug conjugate that consists of an antibody (datopotamab) and an anticancer drug (DXd), joined via a stable cleavable linker.[8] Dato-DXd has shown promising antitumour efficacy in previous studies in people with advanced or metastatic NSCLC.
- This study, called TROPION-Lung14, is designed to compare the combination of osimertinib and Dato-DXd with osimertinib alone as a first treatment for people with *EGFR*-mutated advanced NSCLC.

How are we performing this research?

- Eligible patients will be randomly assigned to receive either osimertinib plus Dato-DXd or osimertinib alone. Patients will continue to receive treatment until their disease progresses, side effects become unacceptable, or they choose to leave the study. The main objective of the study is to see how long patients remain alive without their cancer growing or spreading (known as progression-free survival).

Who will participate in this study?

- We are recruiting adults with previously untreated *EGFR*-mutated advanced NSCLC.

Where can I access more information?

The first results are expected to be available in early 2028, with the study expected to end in mid-2032. For more information about TROPION-Lung14, please visit https://clinicaltrials.gov/study/NCT06350097. You may also speak to your doctor about clinical studies.

1. Cross DA, et al. Cancer Discov 2014;4:1046–61; 2. Hendriks LE, et al. Ann Oncol 2023;34:339–57; 3. Jaiyesimi IA, et al. J Clin Oncol 2024;42:e1–e22; 4. Remon J, et al. Ann Oncol 2021;32:1637–42; 5. Pisters K, et al. J Clin Oncol 2022;40:1127–29; 6. Lu S, et al. N Engl J Med 2024;391:585–97; 7. Gomatou G, et al. Cancers (Basel) 2023;15:841; 8. Okajima D, et al. Mol Cancer Ther 2021;20:2329–40.

Background

- Osimertinib, a third-generation, irreversible, CNS-active EGFR-TKI[1–7] is recommended as 1L treatment for *EGFR*-mutated advanced NSCLC,[8,9] as adjuvant treatment for resected stage IB–IIIA *EGFR*-mutated NSCLC[10,11] and for patients with unresectable *EGFR*-mutated stage III NSCLC without progression during or after chemoradiotherapy.[12]
 - The 1L recommendation is based on the results of the phase 3 FLAURA study, which demonstrated significant improvements in both PFS and OS with osimertinib versus first-generation EGFR-TKIs (gefitinib or erlotinib) in patients with previously untreated locally advanced or metastatic NSCLC whose tumors had *EGFR* exon 19 deletions (Ex19del) or exon 21 (L858R) substitution mutations.[3,4]
- Despite the clinical benefit with osimertinib, most patients develop resistance,[13] and treatment options on or after disease progression are limited.
- One potential strategy to extend the efficacy of 1L osimertinib, and delay the onset of resistance, is to use a combination regimen, where osimertinib is combined with treatment(s) with broad antitumour activity.
- Data from the phase 3 FLAURA2 study demonstrated the feasibility of this approach, showing a statistically significant improvement in PFS with 1L osimertinib plus platinum–pemetrexed doublet chemotherapy versus osimertinib alone in patients with *EGFR*-mutated advanced NSCLC.[14]
- Dato-DXd, an ADC composed of a humanized anti-TROP2 monoclonal antibody conjugated to a potent topoisomerase I inhibitor,[15] has demonstrated efficacy and manageable safety in NSCLC as monotherapy, including patients with pretreated (2L+) *EGFR*-mutated locally advanced or metastatic NSCLC in the phase 3 TROPION-Lung01, phase 2 TROPION-Lung05, and phase 1 TROPION-PanTumor01 studies.[16–19]
- Given the data suggesting that Dato-DXd may be clinically active in *EGFR*-mutated NSCLC, it was hypothesized that combining Dato-DXd with 1L osimertinib may enhance its efficacy and potentially overcome treatment resistance.

The phase 3 TROPION-Lung14 study is evaluating the efficacy and safety of osimertinib in combination with Dato-DXd versus osimertinib alone as 1L treatment for patients with *EGFR*-mutated locally advanced or metastatic NSCLC

Key inclusion criteria

- Age ≥18 years (≥20 years in Japan)
- Histologically or cytologically confirmed, non-squamous NSCLC
- Stage IIIB/IIIC or IV metastatic or recurrent NSCLC (based on the 8th edition of the American Joint Committee on Cancer Staging Manual) not amenable to curative surgery or definitive chemoradiation
- No prior EGFR-TKI or other systemic therapy for stage IIIB/IIIC or IV disease
- Ex19del and/or L858R *EGFR* mutation, either alone or in combination with other *EGFR* mutations, which may include T790M
- WHO PS of 0 or 1
- ≥1 measurable lesion per RECIST 1.1, not previously irradiated
- Life expectancy ≥12 weeks
- Adequate bone marrow reserve and organ function
- In randomized period: archival tumor sample (for central confirmation of *EGFR* mutation status) and baseline plasma sample (for retrospective plasma *EGFR* testing)

Key exclusion criteria

- History of another primary malignancy
- Persistent toxicities caused by previous anticancer therapy (excluding alopecia) not yet improved to grade ≤1 or baseline
- Spinal cord compression or unstable brain metastases
- Leptomeningeal carcinomatosis or metastasis
- Significant third-space fluid retention (e.g., ascites or pleural effusion) not amenable for required repeated drainage
- Clinically significant corneal disease
- Any evidence of severe or uncontrolled systemic diseases, including, but not limited to active bleeding diseases, active infection, active ILD/pneumonitis or cardiac disease
- Active or uncontrolled HBV or HCV infection, uncontrolled HIV, uncontrolled infection requiring IV antimicrobials, suspected infection, an inability to rule out infection or active tuberculosis
- Uncontrolled or significant cardiac disease, or resting electrocardiogram with clinically abnormal findings
- History of ILD, drug-induced ILD, radiation pneumonitis requiring steroid treatment or any evidence of clinically active ILD
- Severe pulmonary function compromise
- Any anticancer therapy within the past 12 months
- Prior exposure to any agent including an ADC containing a chemotherapeutic agent targeting topoisomerase I, TROP2-targeted therapy

TROPION-Lung14 (NCT06350097): a phase 3, open-label, multicenter study

Patients

- Age ≥18 years
- Histologically or cytologically confirmed stage IIIB/IIIC or IV non-squamous, *EGFR*-mutated (Ex19del or L858R) NSCLC
- No prior EGFR-TKI or other systemic therapy for stage IIIB/IIIC or IV disease
- ≥1 measurable lesion per RECIST 1.1
- WHO PS of 0 or 1

Non-randomized safety run-in (N=~20)

Osimertinib 80 mg PO QD + Dato-DXd 6 mg/kg IV Q3W*

N=~562 Randomized 1:1

Stratified by:
- *EGFR* mutation type (Ex19del vs L858R)
- WHO PS (0 vs 1)
- CNS metastasis status (yes vs no)

Osimertinib 80 mg PO QD + Dato-DXd 6 mg/kg IV Q3W*

Osimertinib 80 mg PO QD*

Primary Endpoint PFS by BICR

*Treatment will continue until RECIST 1.1-defined progression by investigator, unacceptable toxicity or another discontinuation criterion is met. Following discontinuation, choice of subsequent therapy will be at the discretion of the investigator. Participants will be followed for second progression on a subsequent treatment, defined according to local practice, and for OS.

Enrollment start: April 2024 | Enrollment is ongoing

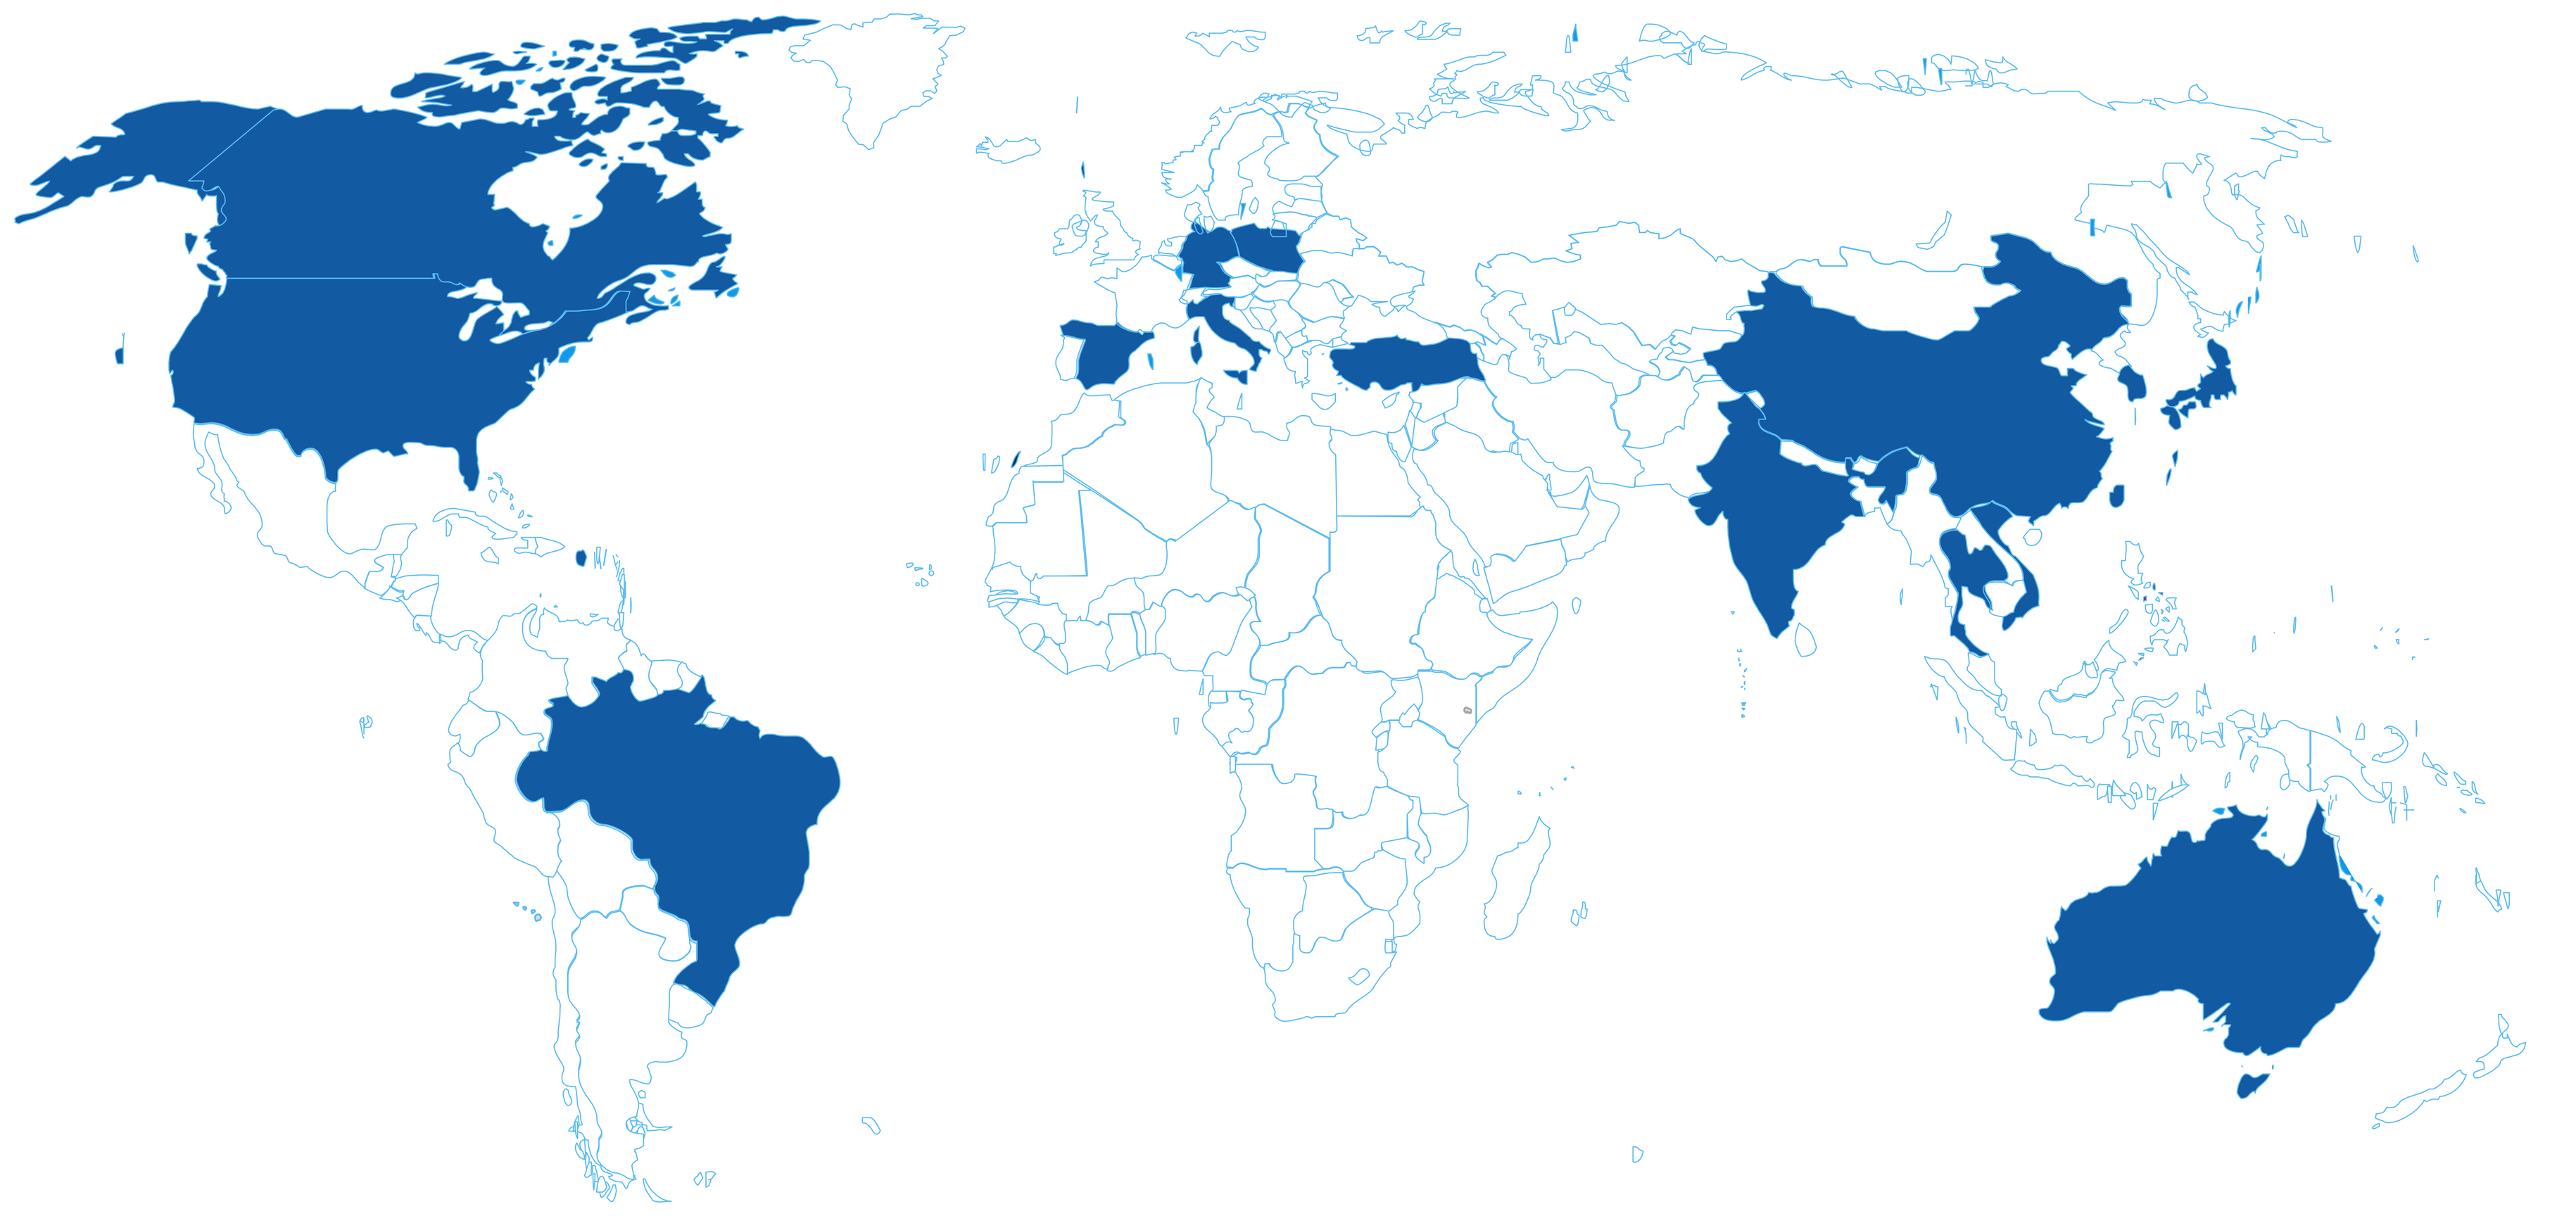

Planned/active recruitment

Countries and regions with participating study sites (~170 sites)
Australia, Brazil, Canada, China, France, Germany, Hong Kong, India, Italy, Japan, Poland, Republic of Korea, Spain, Taiwan, Thailand, Türkiye, USA, Vietnam

Key study endpoints

Primary endpoint

- PFS per RECIST v1.1 by BICR

Secondary endpoints

- OS (key secondary endpoint)
- CNS PFS, per modified RECIST 1.1 by BICR (in patients with ≥1 brain lesion at baseline)
- PFS by investigator
- ORR per RECIST v1.1 by BICR and investigator
- Duration of response per RECIST v1.1 by BICR and investigator
- Determine presence/absence of CNS lesions at progression per modified RECIST 1.1 by BICR (in participants without CNS metastases at baseline)
- PFS2
- Safety and tolerability
- Pharmacokinetics
- Immunogenicity

PFS, defined as time from randomization until progression per RECIST 1.1, or death due to any cause (in absence of progression). OS, defined as time from randomization until date of death due to any cause. CNS PFS, defined as time from randomization until date of objective CNS progression by CNS BICR or death (by any cause in absence of CNS progression). PFS2, defined as time from randomization to earliest progression event (following initial progression) after first subsequent anticancer therapy or death.

Dato-DXd and osimertinib clinical program

Dato-DXd and osimertinib are also being investigated in the ongoing, phase 3 TROPION-Lung15 study (NCT06417814); Dato-DXd with or without osimertinib versus platinum-based doublet chemotherapy for patients with *EGFR*-mutated locally advanced or metastatic NSCLC whose disease has progressed on prior osimertinib

Poster

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please use the link below.

https://bit.ly/4jSOdAo

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Abbreviations

1L/2L, first/second line; ADC, antibody–drug conjugate; BICR, blinded independent central review; CNS, central nervous system; Dato-DXd, datopotamab deruxtecan; EGFR, epidermal growth factor receptor; Ex19del, exon 19 deletion; HBV/HCV, hepatitis B/C virus; HIV, human immunodeficiency virus; ILD, interstitial lung disease; IV, intravenous; NSCLC, non-small cell lung cancer; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PFS2, time to second progression or death; PO, oral; PS, performance status; Q3W, every 3 weeks; QD, once daily; RECIST 1.1, Response Evaluation Criteria in Solid Tumors version 1.1; TKI, tyrosine kinase inhibitor; TROP2, trophoblast cell surface antigen 2; WHO, World Health Organization.

Acknowledgments

This study (NCT06350097) is sponsored by AstraZeneca. In July 2020, Daiichi-Sankyo entered into a global development and commercialization collaboration with AstraZeneca for datopotamab deruxtecan (Dato-DXd). Medical writing support for the development of this poster, under the direction of the authors, was provided by Mark Holland, PhD, of Ashfield MedComms (Manchester, UK), an Inizio Company, and was funded by AstraZeneca in accordance with Good Publication Practice (GPP) guidelines (http://www.ismpp.org/gpp-2022).

Disclosures

Sarah Goldberg reports honoraria from Amgen; a consulting or advisory role for AstraZeneca/MedImmune, Daiichi Sankyo, Genzyme, Janssen, Merck, Regeneron and Summit Therapeutics; and research funding from Amgen (Inst), AstraZeneca, Boehringer Ingelheim, Bristol Myers Sqiubb (Inst), Genentech (Inst), Merck (Inst), Mirati Therapeutics, ORIC Pharmaceuticals (Inst), Pfizer (Inst) and Spectrum Pharmaceuticals (Inst). For co-author disclosures, please refer to the abstract.

References

1. Cross DA, et al. Cancer Discov 2014;4:1046–61.
2. Mok TS, et al. N Engl J Med 2017;376:629–40.
3. Soria J-C, et al. N Engl J Med 2018;378:113–25.
4. Ramalingam SS, et al. N Engl J Med. 2020;382:41–50.
5. Wu Y-L, et al. N Engl J Med 2020;383:1711–23.
6. Wu Y-L, et al. J Clin Oncol 2018;36:2702–9.
7. Reungwetwattana T, et al. J Clin Oncol 2018;36:3290–7.
8. Hendriks LE, et al. Ann Oncol 2023;34:339–57.
9. Jaiyesimi IA, et al. J Clin Oncol 2024;42:e1–e22.
10. Remon J, et al. Ann Oncol 2021;32:1637–42.
11. Pisters K, et al. J Clin Oncol 2022;40:1127–29.
12. Lu S, et al. N Engl J Med 2024;391:585–97.
13. Gomatou G, et al. Cancers (Basel) 2023;15:841.
14. Planchard D, et al. N Engl J Med 2023;389:1935–48.
15. Okajima D, et al. Mol Cancer Ther 2021;20:2329–40.
16. Ahn MJ, et al. J Clin Oncol 2025;43:260–72.
17. Shimizu T, et al. J Clin Oncol 2023;41:4678–87.
18. Garon EB, et al. Ann Oncol 2021;32(S5):S1326–7 [LBA49].
19. Sands J, et al. J Clin Oncol 2025:JCO2401349.

Poster presented at the American Society of Clinical Oncology (ASCO) 2025; Chicago, IL, USA; May 30–June 3, 2025 by Shun Lu. Corresponding author email address: shunlu@sjtu.edu.cn