P1820

Pharmacokinetic–tumor growth inhibition modeling of raludotatug deruxtecan (R-DXd) to support phase 3 dose selection in platinum-resistant ovarian cancer

YoungJun Yoo,[1] Kevin Koloskoff,[2] Sandra Re,[1] Izna Ali,[1] Felipe K. Hurtado,[1] Raouf El Cheikh[2]

[1]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [2]Daiichi Sankyo France, SAS, Rueil-Malmaison, France

OBJECTIVES

- To investigate the effect of R-DXd exposure on tumor size in patients with PROC using a PK–TGI model
- To predict the change from baseline in tumor size with R-DXd doses of 4.8, 5.6, and 6.4 mg/kg Q3W, to support selection of the optimal dose for the Phase 3 part of the REJOICE-Ovarian01 trial[1]
- To identify covariates affecting the PK–TGI relationship. The effect of covariates, including but not limited to baseline tumor CDH6 expression, prior bevacizumab treatment, and the number of prior lines of therapy, on tumor response was investigated

CONCLUSIONS

- The PK–TGI model characterized changes in tumor size as a function of R-DXd concentration and quantified the effect of baseline tumor CDH6 expression on tumor size reduction in patients with PROC
- Simulations showed a positive exposure–response relationship, with greater reductions in tumor size at higher doses
- There was a positive association between baseline CDH6 expression and tumor size decrease; however, the effect tended to plateau at higher expression, and clinically meaningful tumor responses were predicted across a range of CDH6 expression levels
- Together with the totality of the clinical data, exposure–response analyses of key efficacy and safety endpoints, and an overall assessment of the benefit–risk profile, these analyses supported the selection of 5.6 mg/kg Q3W as the optimal R-DXd monotherapy dose for the randomized Phase 3 part of the REJOICE-Ovarian01 trial[1]

Copies of this poster obtained through this Quick Response (QR) code and/or https://bit.ly/DSAACR26 are for personal use only and may not be reproduced without written permission of the authors.

INTRODUCTION

- R-DXd is a novel CDH6-directed ADC comprising a humanized anti-CDH6 IgG1 mAb that is covalently linked to a potent topoisomerase I inhibitor payload (DXd) via a tetrapeptide-based cleavable linker[2]
- In a first-in-human Phase 1 trial, R-DXd showed encouraging activity at doses of 4.8–6.4 mg/kg Q3W in patients with heavily pretreated OC[3]
- The REJOICE-Ovarian01 trial is evaluating R-DXd in patients with PROC, and it includes a Phase 2 dose-optimization part and a randomized Phase 3 part[1]
 - In the dose-optimization primary analysis of the Phase 2 part, R-DXd demonstrated promising efficacy and manageable safety at doses of 4.8, 5.6, and 6.4 mg/kg Q3W, with a confirmed ORR of 50.5% across doses[1]
 - The ongoing Phase 3 part is comparing R-DXd 5.6 mg/kg with treatment of physician's choice in patients with PROC[1]

METHODS

Data source

- This PK–TGI modeling analysis pooled data from patients with PROC enrolled in two ongoing clinical trials of R-DXd monotherapy:
 - The first-in-human, dose-escalation/dose-expansion Phase 1 trial (DS6000-A-U101; NCT04707248), in which patients with OC or RCC received R-DXd at 1.6–9.6 mg/kg Q3W (data cut-off: August 21, 2025)[3,4]
 - The dose-optimization primary analysis of the Phase 2 part of the Phase 2/3 REJOICE-Ovarian01 trial (NCT06161025), in which patients with PROC were randomized to receive R-DXd at 4.8, 5.6, or 6.4 mg/kg Q3W (data cut-off: February 26, 2025)[1]
- The assembled dataset included plasma concentration data for R-DXd and DXd payload, tumor size measurements (defined as SLD by BICR per RECIST 1.1), and data on demographic, clinical, laboratory, and biomarker covariates
 - Tumor size measurements ≤5 mm were considered BLQ and modeled as censored observations (0–5 mm) using the M4 method
 - Neither trial selected patients according to CDH6 expression.[1,3] CDH6 expression was assessed in baseline tumor biopsies using a clinical trial IHC assay (SP450; Roche Diagnostics, Indianapolis, IN, USA) and quantified as the percentage of viable tumor cells positive for CDH6 membrane staining at any intensity (1+/2+/3+)

Model development

- The PK–TGI model incorporated a joint ADC–payload PK model and a longitudinal TGI model (**Figure 1**)
- The TGI model was adapted from previously published modeling frameworks[6] and incorporated three components:
 - A drug-induced tumor killing rate (k_{kill}) that is driven by R-DXd concentration and attenuated over time by a resistance term ($e^{-\lambda \cdot t}$) **(Eq. 1)**
 - A logistic growth model that describes tumor growth **(Eq. 2)**
 - Covariate effects on k_{kill} **(Eq. 3)**
- Nonlinear mixed-effects modeling was performed using MonolixSuite version 2021R1 (Simulations Plus, Lancaster, CA, USA)
- Covariates for the TGI model were examined graphically and selected using the COSSAC method, which relies on trends in empirical Bayes estimates versus covariates; in addition, physiologic or mechanistic plausibility, and prior knowledge of covariate relationships were considered

Simulations

- Simulations were performed to characterize the longitudinal change from baseline in tumor size up to 48 weeks
- Simulations incorporated parameter uncertainty by sampling from the uncertainty distribution of the fixed effects
- Predictions were generated for the 4.8-, 5.6-, and 6.4-mg/kg doses for a typical patient, as defined by estimated typical parameters and reference covariate values (i.e. baseline body weight, 60 kg; tumor CDH6 expression, 75%; and no prior bevacizumab)
 - Effects of significant covariates on the tumor response were also simulated for a typical patient at 5.6 mg/kg

Figure 1. Schematic diagram of the R-DXd PK–TGI model

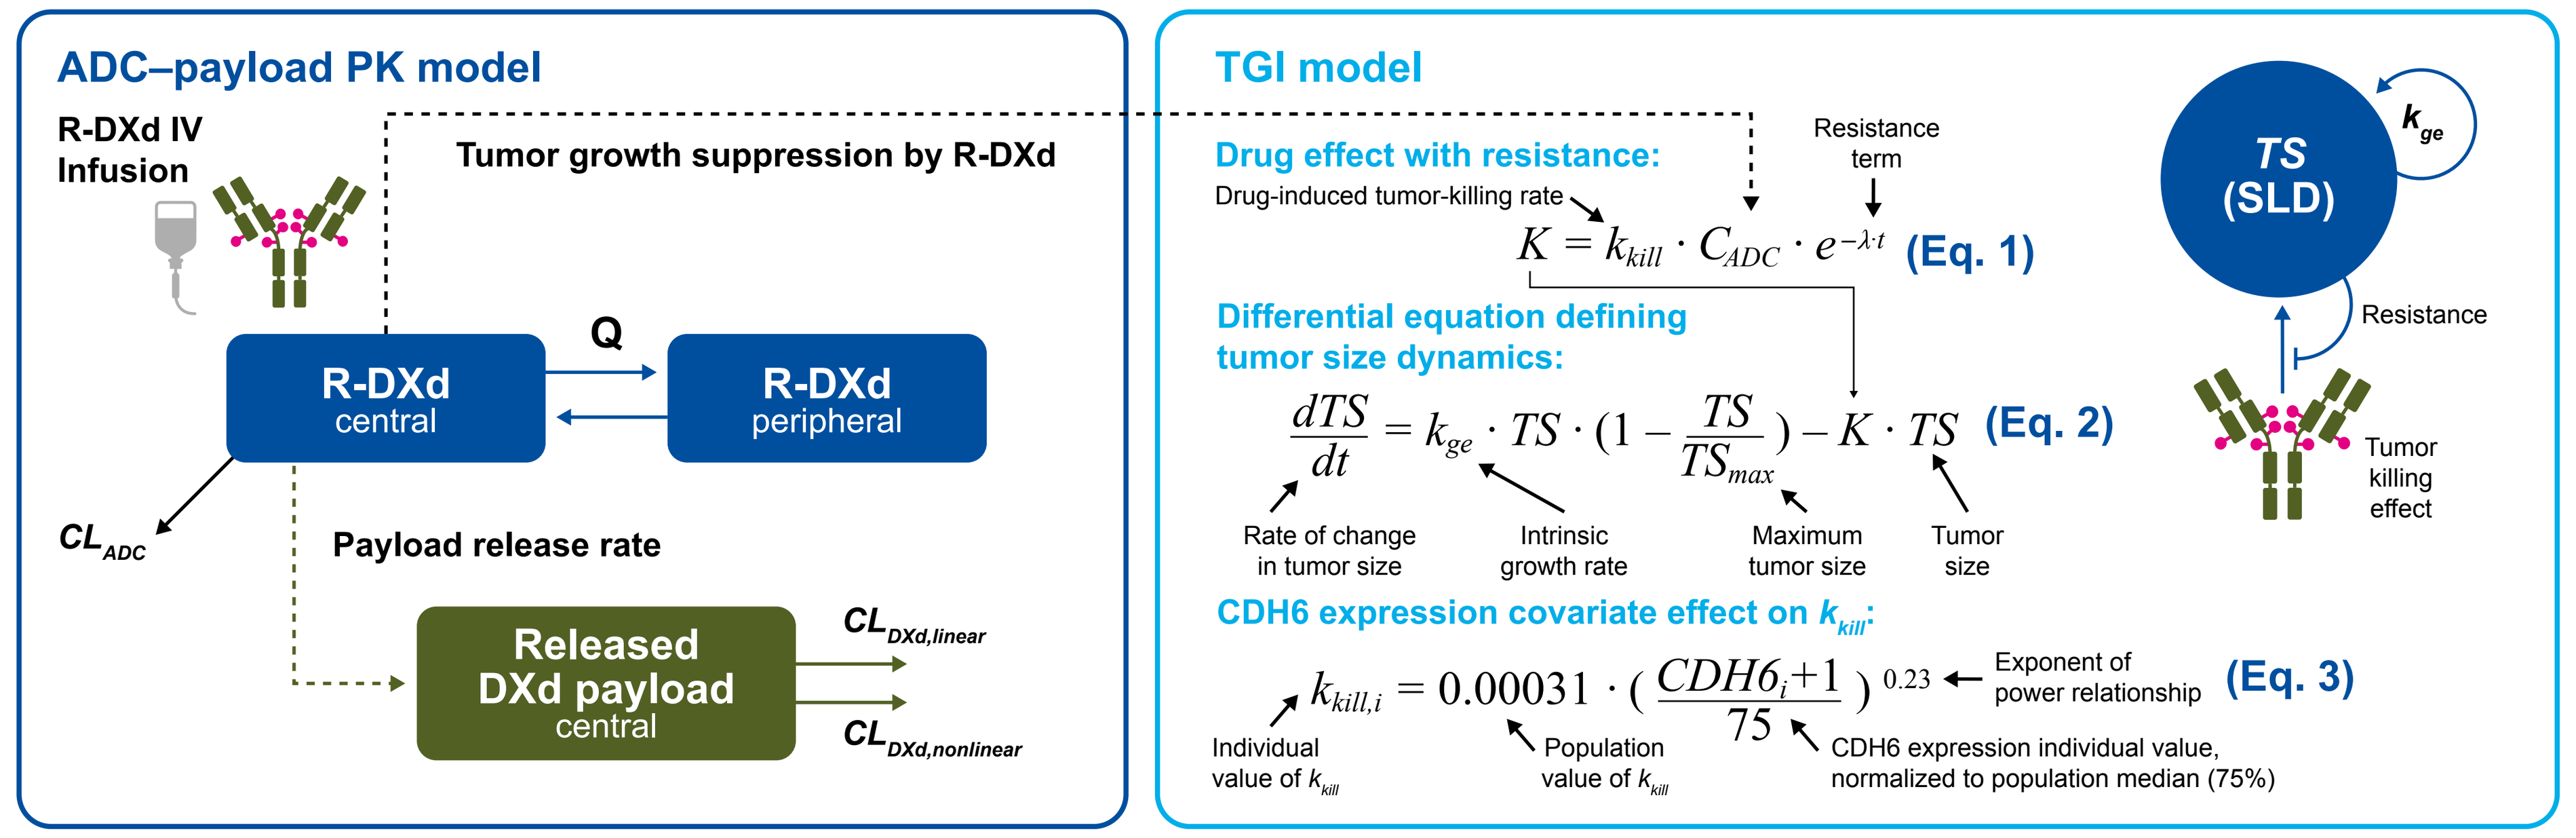
ADC–payload PK model
R-DXd IV Infusion
Tumor growth suppression by R-DXd
Q
R-DXd central
R-DXd peripheral
CL_{ADC}
Payload release rate
Released DXd payload central
$CL_{DXd,linear}$
$CL_{DXd,nonlinear}$
TGI model
Drug effect with resistance:
Drug-induced tumor-killing rate
Resistance term
$K = k_{kill} \cdot C_{ADC} \cdot e^{-\lambda \cdot t}$ (Eq. 1)
Differential equation defining tumor size dynamics:
$\frac{dTS}{dt} = k_{ge} \cdot TS \cdot (1 - \frac{TS}{TS_{max}}) - K \cdot TS$ (Eq. 2)
Rate of change in tumor size
Intrinsic growth rate
Maximum tumor size
Tumor size
CDH6 expression covariate effect on k_{kill}:
$k_{kill,i} = 0.00031 \cdot (\frac{CDH6_i+1}{75})^{0.23}$ (Eq. 3)
Exponent of power relationship
Individual value of k_{kill}
Population value of k_{kill}
CDH6 expression individual value, normalized to population median (75%)
TS (SLD)
k_{ge}
Resistance
Tumor killing effect

RESULTS

Patient population

- The analysis included data from 229 patients with PROC who received ≥1 dose of R-DXd, had ≥1 PK measurement, and had a baseline and ≥1 post-baseline tumor size measurement, with ≥18 weeks of follow-up (tumor assessments every 6 weeks for 24 weeks, then every 12 weeks) (**Figure 2; Table 1**)

Model development and evaluation

- Model parameters were estimated with adequate precision (**Table 2**)
- TS_{max} was fixed at 500 mm, as the maximum observed tumor size measurement in the analysis dataset was 308 mm
- Baseline CDH6 expression and prior bevacizumab treatment were retained as significant covariates in the final model
 - Higher CDH6 expression was associated with a modest, less-than-proportional increase in tumor killing effect (eg, doubling CDH6 increased k_{kill} by 17%)
 - Prior bevacizumab treatment was associated with a 20.6% decrease in tumor killing effect
- Goodness-of-fit plots showed that model appropriately described the observed tumor size data (**Figure 3**)

Figure 2. Analysis dataset

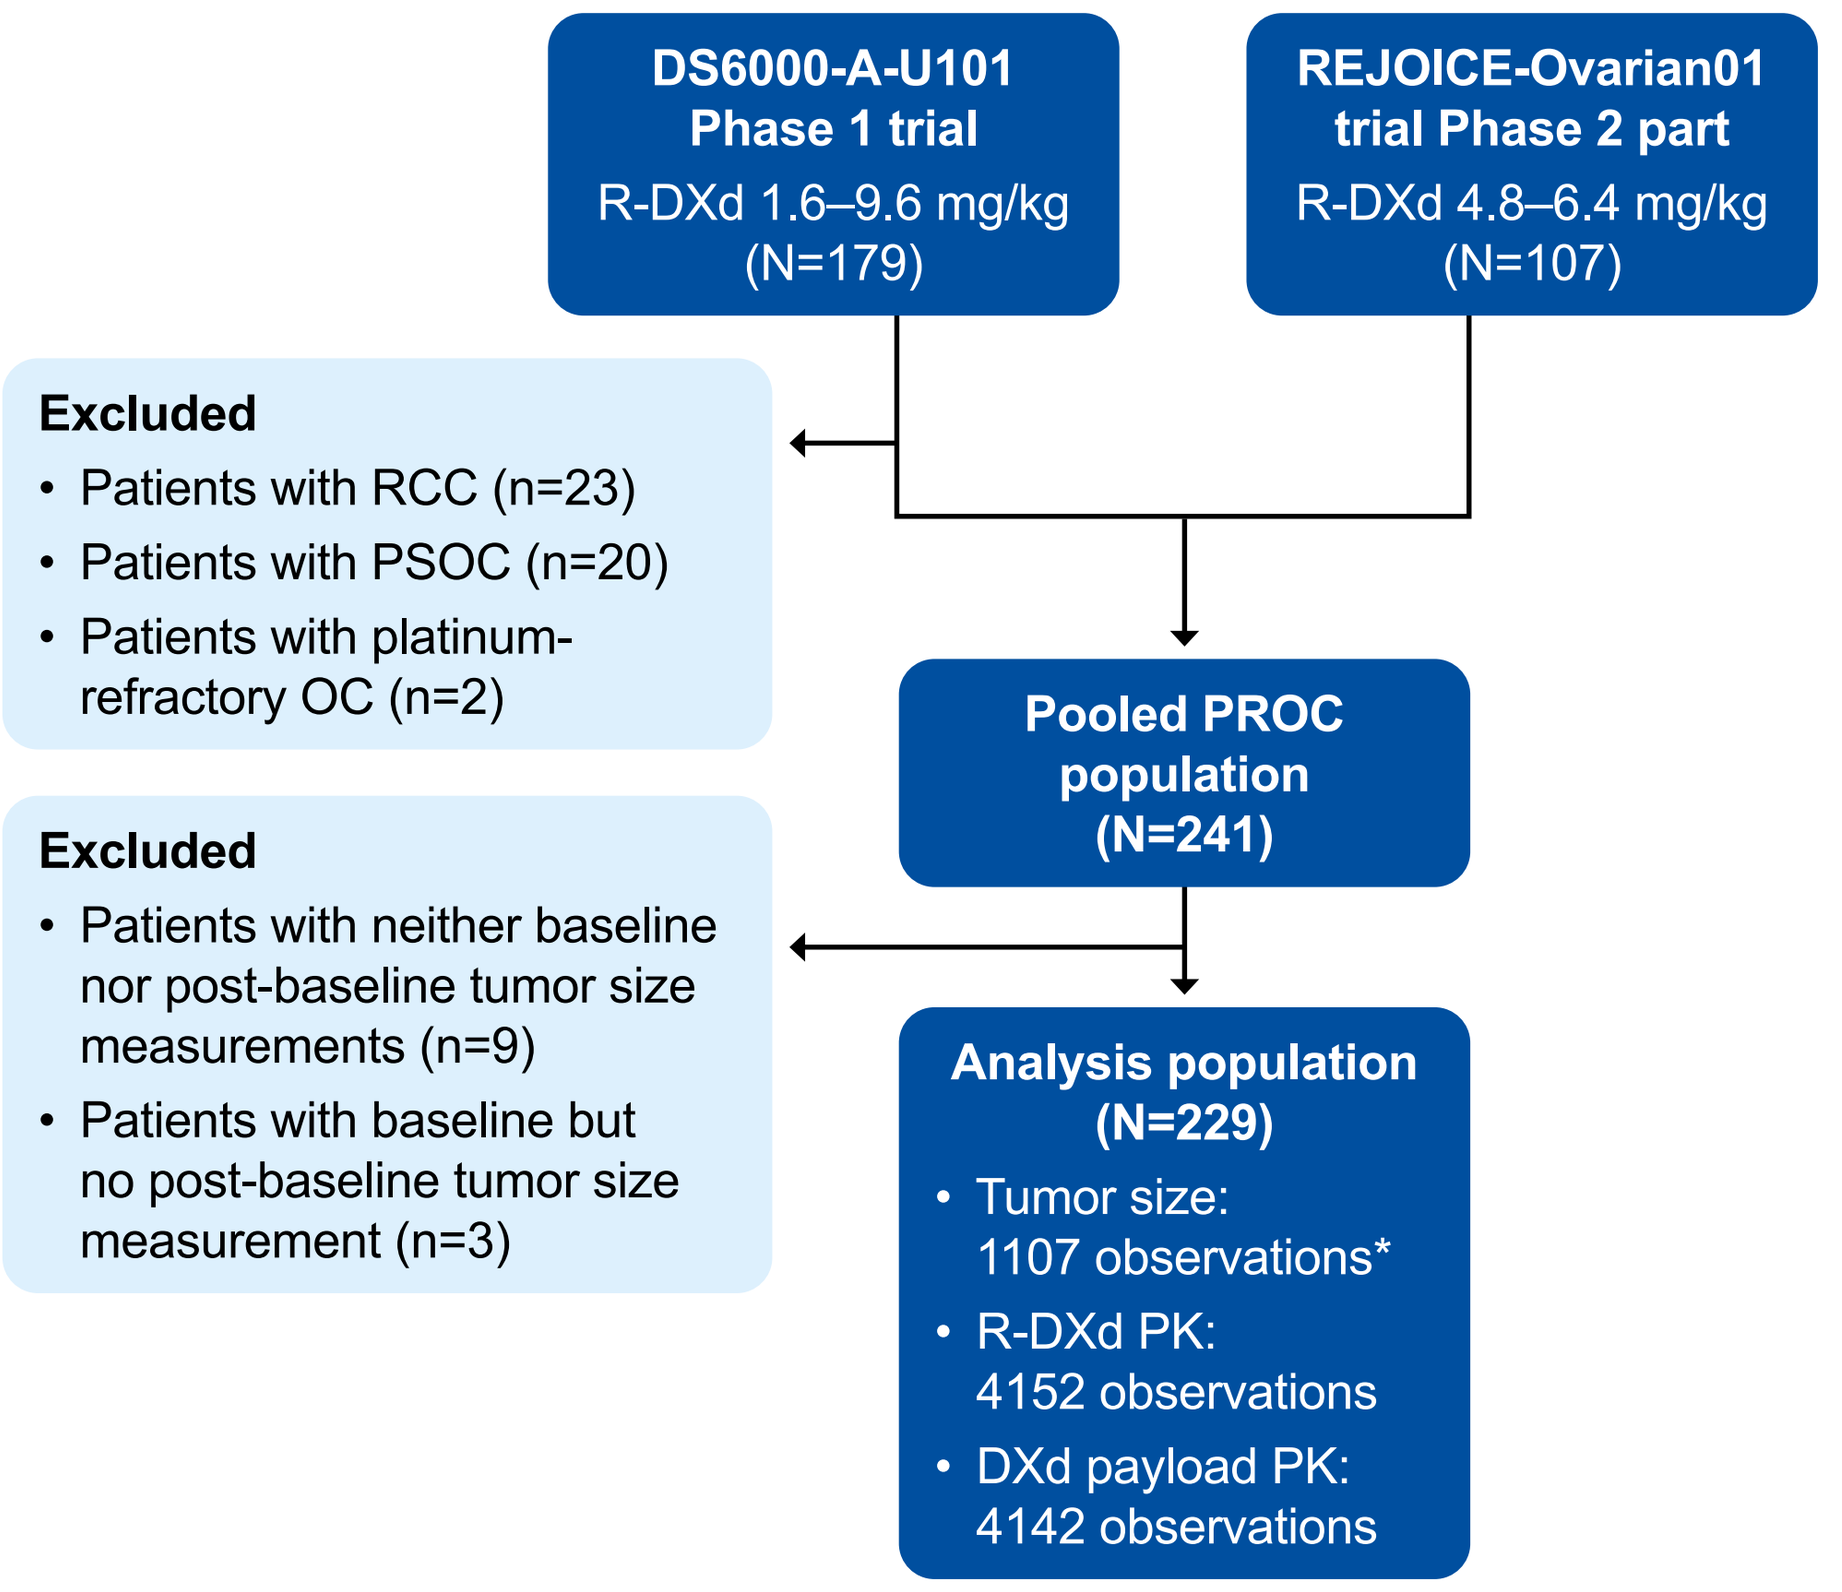
DS6000-A-U101 Phase 1 trial
R-DXd 1.6–9.6 mg/kg
(N=179)
REJOICE-Ovarian01 trial Phase 2 part
R-DXd 4.8–6.4 mg/kg
(N=107)
Excluded
• Patients with RCC (n=23)
• Patients with PSOC (n=20)
• Patients with platinum-refractory OC (n=2)
Pooled PROC population
(N=241)
Excluded
• Patients with neither baseline nor post-baseline tumor size measurements (n=9)
• Patients with baseline but no post-baseline tumor size measurement (n=3)
Analysis population
(N=229)
• Tumor size: 1107 observations*
• R-DXd PK: 4152 observations
• DXd payload PK: 4142 observations

*Of the 1107 observations, 62 tumor size measurements included in the analysis were tagged as BLQ.

Table 1. Baseline characteristics

	DS6000-A-U101 Phase 1 trial (n=129)	REJOICE-Ovarian01 Phase 2 part (n=100)	Total (N=229)
Starting dose of R-DXd, n (%)*			
3.2 mg/kg	2 (1.6)	–	2 (0.9)
4.8 mg/kg	41 (31.8)	31 (31.0)	72 (31.4)
5.6 mg/kg	30 (23.3)	35 (35.0)	65 (28.4)
6.4 mg/kg	39 (30.2)	34 (34.0)	73 (31.9)
8.0 mg/kg	14 (10.9)	–	14 (6.1)
9.6 mg/kg	3 (2.3)	–	3 (1.3)
Age, median (range), years	64 (35–84)	60 (34–81)	62 (34–84)
Body weight, median (range), kg	59 (32–178)	60 (36–101)	60 (32–178)
Race, n (%)			
White	67 (51.9)	41 (41.0)	108 (47.2)
Asian	54 (41.9)	38 (38.0)	92 (40.2)
Black/African American	4 (3.1)	0	4 (1.7)
Other or unknown	4 (3.1)	21 (21.0)	25 (10.9)
Tumor size, median (range), mm	59 (10–285)	55.5 (10–274)	57 (10–285)
Tumor CDH6 expression, median (range), %	70 (0–100)	80 (0–100)	75 (0–100)
Prior bevacizumab treatment, n (%)			
Yes	88 (68.2)	85 (85.0)	173 (75.5)
No	41 (31.8)	15 (15.0)	56 (24.5)

*One patient in the Phase 1 trial received a starting dose of R-DXd of 1.6 mg/kg. This patient had PSOC and was therefore excluded from the present analysis.

Table 2. Final TGI model parameter estimates

Parameter	Unit	Estimate (%RSE)	Inter-individual variability, %CV* (%RSE)
Intrinsic tumor growth rate (k_{ge})	1/day	0.00055 (22.2)	188 (9.46)
Maximum tumor size (TS_{max})	mm	500 (fixed)	–
Drug-induced tumor-killing rate (k_{kill})	1/day	0.00031 (12.2)	109 (6.59)
Tumor resistance parameter (λ)	1/day	0.015 (8.97)	122 (9.94)
Covariate effect of baseline CDH6 expression on k_{kill}	–	0.23 (20.8)	–
Covariate effect of prior bevacizumab on k_{kill}	–	–0.23 (59.1)	–

*%CV was calculated as $\sqrt{\exp(w^2)-1}\times 100$.

Figure 3. Diagnostic plot of observed versus predicted tumor size

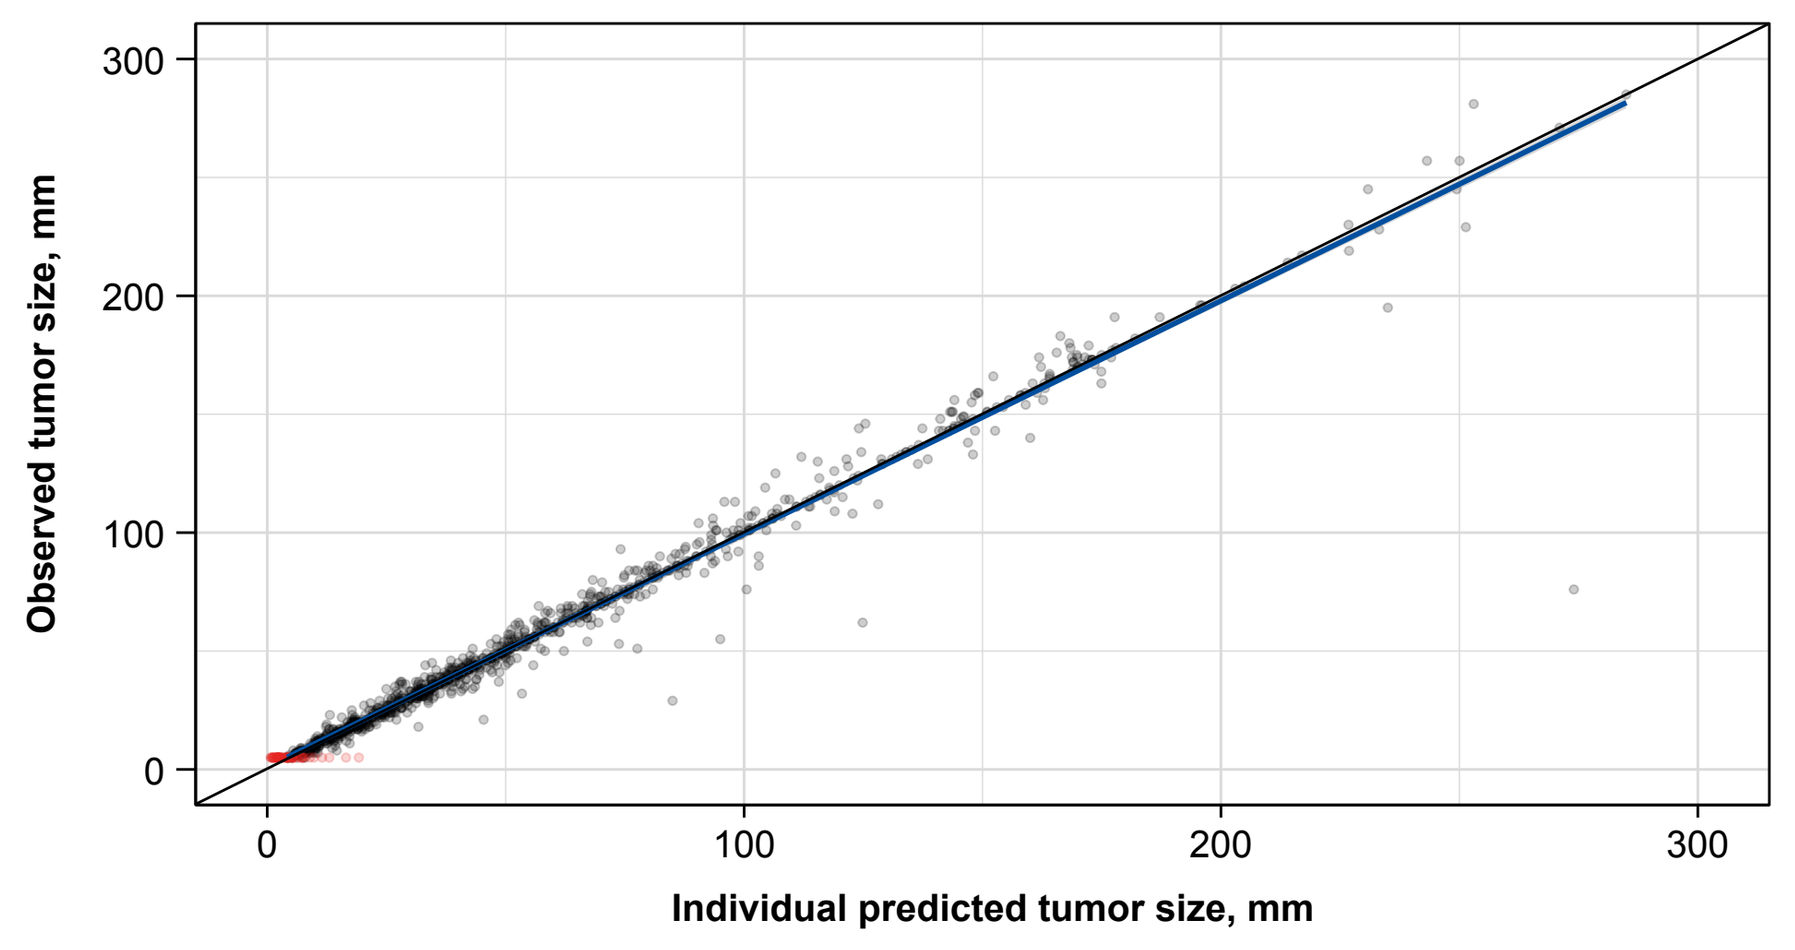
Observed tumor size, mm
Individual predicted tumor size, mm
0
100
200
300

Circles show observed versus predicted values (with values tagged as BLQ shown in red). Blue lines depict the linear interpolation with the 95% CI in gray.

Simulations

- The simulations demonstrated a positive exposure–response relationship, with greater tumor shrinkage at higher doses of R-DXd (**Figure 4**)
- Predicted median tumor shrinkage (90% CI) at Week 24 was −51% (−60 to −43), −57% (−66 to −49), and −62% (−71 to −54) for 4.8, 5.6, and 6.4 mg/kg, respectively
- There was a positive association between baseline CDH6 expression and predicted decreases in tumor size. Tumor response tended to saturate at higher CDH6 expression levels (**Figure 5**)
- Clinically meaningful tumor responses (i.e. ≥30% tumor size reduction for PR per RECIST 1.1) were predicted across a range of CDH6 expression levels at an R-DXd dose of 5.6 mg/kg (**Figure 6**)
- In addition, prior bevacizumab treatment was associated with reduced tumor response in the covariate analysis

Figure 4. Predicted median change from baseline in tumor size at given time points, by dose level

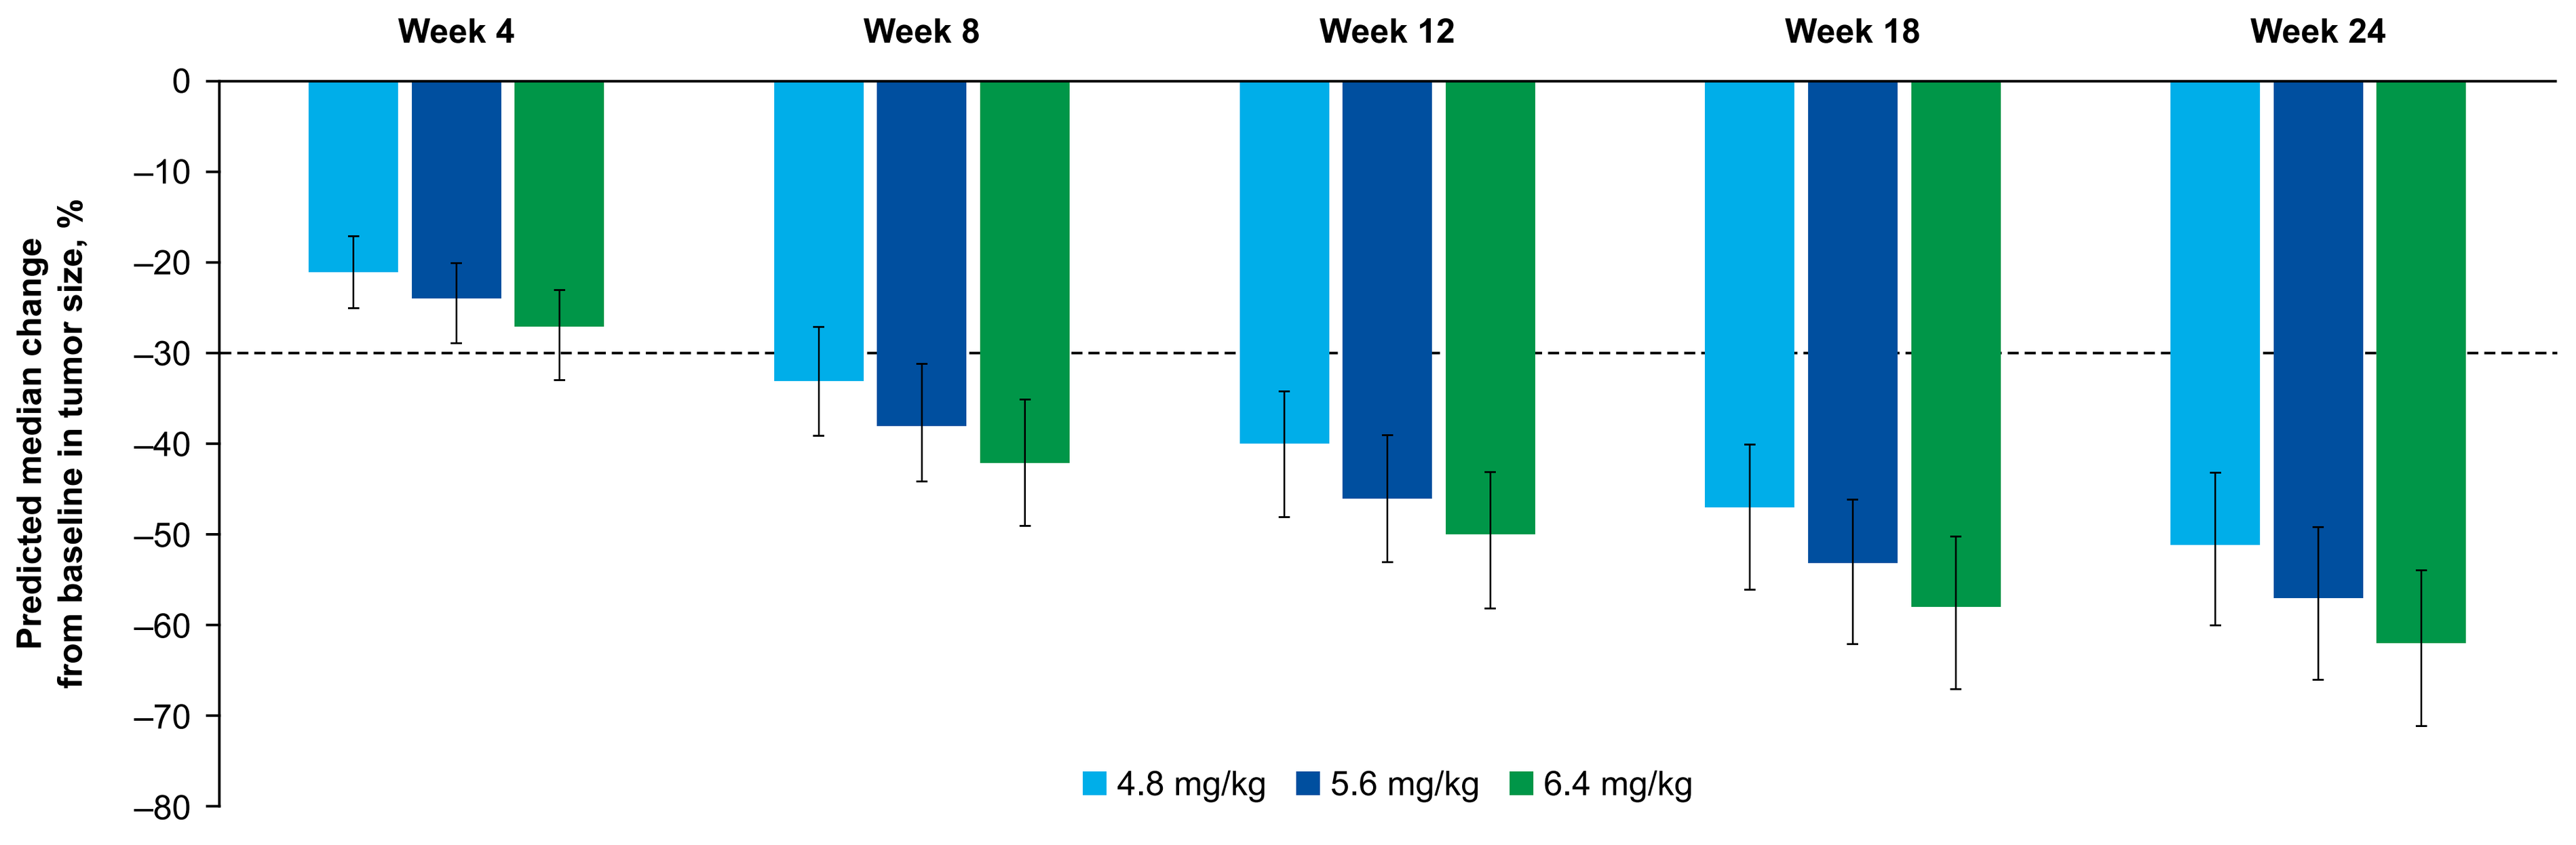
Week 4
Week 8
Week 12
Week 18
Week 24
Predicted median change from baseline in tumor size, %
0
−10
−20
−30
−40
−50
−60
−70
−80
4.8 mg/kg
5.6 mg/kg
6.4 mg/kg

Error bars indicate 90% CIs. The dashed line indicates the tumor size reduction required to meet the threshold for PR per RECIST 1.1.

Figure 5. Predicted median change from baseline in tumor size at Week 24 for the 5.6-mg/kg dose, by baseline CDH6 expression

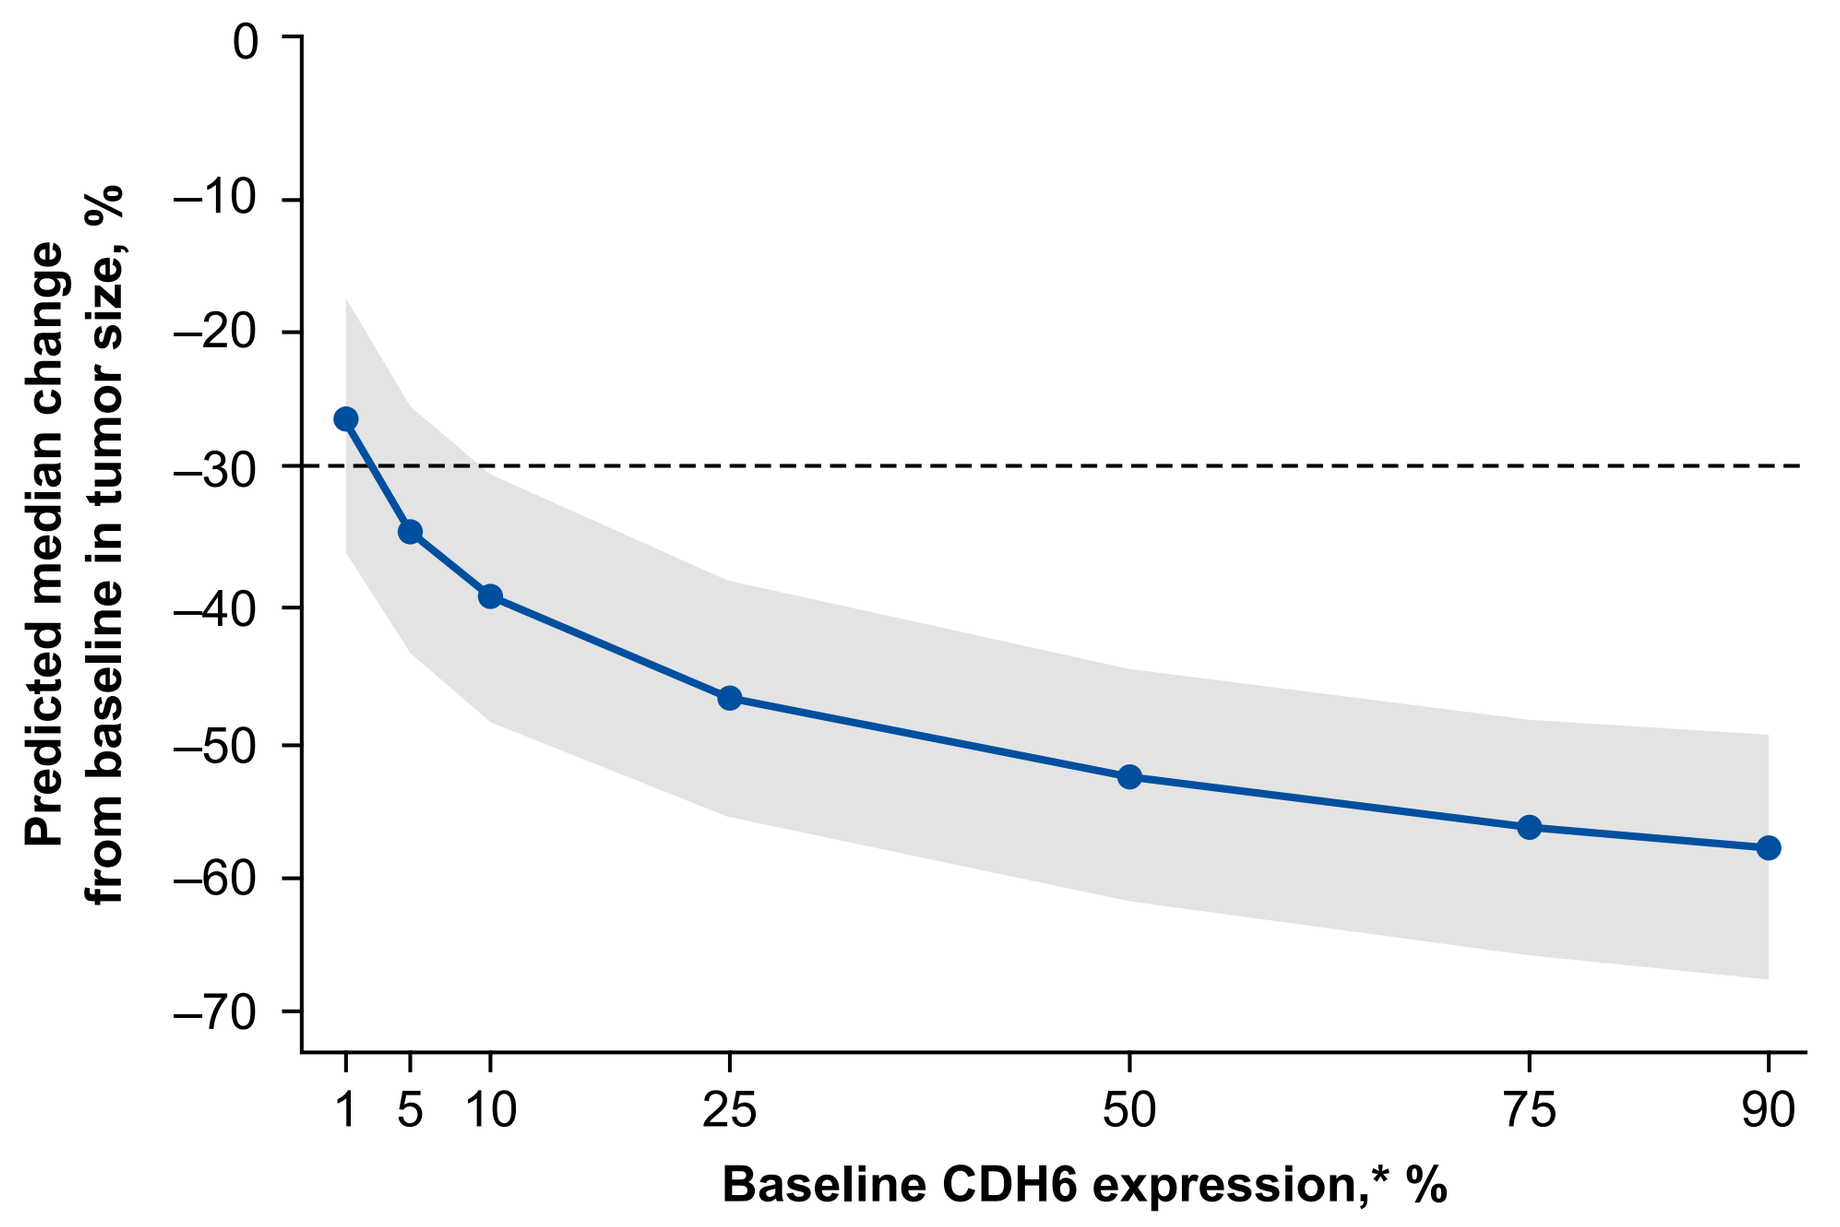
Predicted median change from baseline in tumor size, %
0
−10
−20
−30
−40
−50
−60
−70
1
5
10
25
50
75
90
Baseline CDH6 expression,* %

*Expressed as the percentage of viable tumor cells positive for CDH6 membrane staining at any intensity. The solid line indicates the median, the shaded area indicates the 90% CI, and the dashed line indicates the tumor size reduction required to meet the threshold for PR per RECIST 1.1.

Figure 6. Predicted time-course of tumor size for the 5.6-mg/kg dose, by baseline CDH6 expression

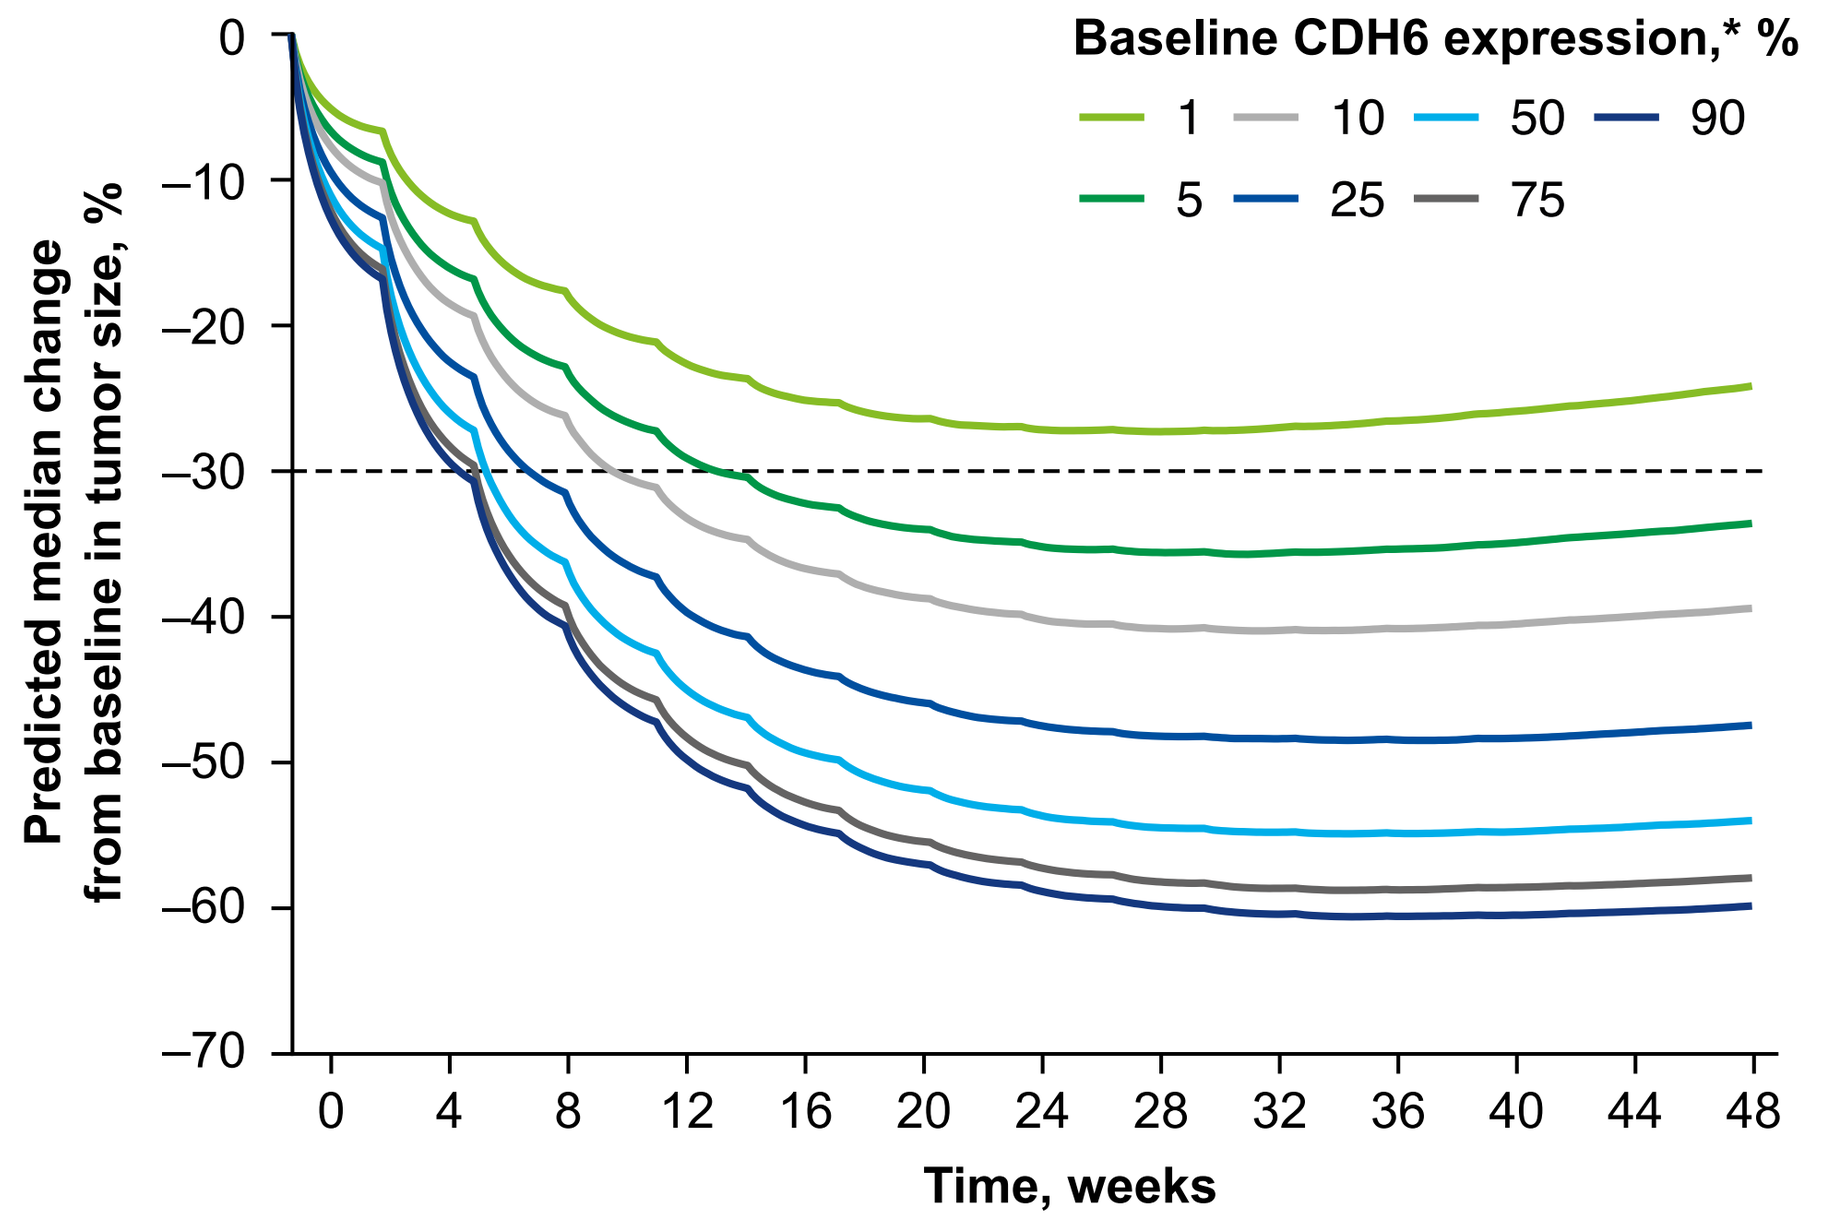
Baseline CDH6 expression,* %
1
10
50
90
5
25
75
Predicted median change from baseline in tumor size, %
0
−10
−20
−30
−40
−50
−60
−70
0
4
8
12
16
20
24
28
32
36
40
44
48
Time, weeks

*Expressed as the percentage of viable tumor cells positive for CDH6 membrane staining at any intensity. Solid lines indicate the median at selected CDH6 expression levels, and the dashed line indicates the tumor size reduction required to meet the threshold for PR per RECIST 1.1.

DISCLOSURES

YoungJun Yoo is a full-time employee of Daiichi Sankyo, Inc.

ABBREVIATIONS

ADC, antibody–drug conjugate; **BICR**, blinded independent central review; **BLQ**, below the limit of quantification; C_{ADC}, ADC plasma concentration; **CDH6**, cadherin-6; **CI**, confidence interval; $CL_{ADC/DXd}$, clearance of ADC/DXd; **COSSAC**, conditional sampling for stepwise approach based on correlation tests; **CV**, coefficient of variation; ***dTS/dt***, rate of change in tumor size; **Eq.**, equation; **IgG1**, immunoglobulin G1; **IHC**, immunohistochemistry; **IV**, intravenous; ***K***, drug effect; k_{ge}, intrinsic tumor growth rate; k_{kill}, drug-induced tumor-killing rate; k_{out}, relative change in payload release rate; k_{rel}, payload release rate constant; $k_{rel(initial/ss)}$, payload release rate constant (initial/at steady state); **mAb**, monoclonal antibody; **OC**, ovarian cancer; **ORR**, objective response rate; **PK**, pharmacokinetics; **PR**, partial response; **PROC**, platinum-resistant ovarian cancer; **PSOC**, platinum-sensitive ovarian cancer; **Q**, intercompartmental clearance; **Q3W**, every 3 weeks; **RCC**, renal cell carcinoma; **RECIST 1.1**, Response Evaluation Criteria in Solid Tumours version 1.1; **RSE**, relative standard error; **SLD**, sum of lesion diameters; ***t***, time; **TGI**, tumor growth inhibition; ***TS***, tumor size; TS_{max}, maximum tumor size; λ, tumor resistance parameter.

ACKNOWLEDGMENTS

Funding for this study was provided by Daiichi Sankyo Company, Limited (Daiichi Sankyo, Co., Ltd.) and Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc. Rahway, NJ, USA. (MSD) for raludotatug deruxtecan (R-DXd).

We thank the patients, their families, and their caregivers for their participation, and the study staff for their contributions.

We also thank Samarth Patel, MSc, and colleagues, of Daiichi Sankyo, Inc., for their work on dataset programming.

Medical writing support was provided by Mark Dyson, DPhil on behalf of BOLDSCIENCE®, Inc., and was funded by Daiichi Sankyo, Inc. Editorial support was provided in accordance with Good Publication Practice guidelines (https://ismpp.org/gpp-2022).

REFERENCES

1. Ray-Coquard I, et al. *Ann Oncol.* 2025;36(Suppl 2):Abstract LBA42.
2. Suzuki H, et al. *Mol Cancer Ther.* 2024;23:257–271.
3. Moore K, et al. *Gynecol Oncol.* 2024;190(Suppl. 1):Abstract 04 [LBA].
4. Hamilton EP, et al. *J Clin Oncol.* 2022;40(16 Suppl.):3002.
5. Ahn JE, et al. *J Pharmacokinet Pharmacodyn.* 2008;35:401–421.
6. Claret L, et al. *J Clin Oncol.* 2009;27:4103–4108.

Presented at the American Association for Cancer Research (AACR) Annual Meeting; April 17–22, 2026; San Diego, CA, USA

youngjun.yoo@daiichisankyo.com