HERTHENA-PanTumor01 (NCT06172478): Expansion of a Global Phase 2 Trial of HER3-DXd in Patients With Advanced/Metastatic HR+/HER2− Breast Cancer

Poster 113

Funda Meric-Bernstam,[1] Christos Fountzilas,[2] Izuma Nakayama,[3] Takahiro Kogawa,[4] Tomohiro Nishina,[5] Thomas Powles,[6] Jin Won Kim,[7] Yen-Yang Chen,[8] François Ghiringhelli,[9] Valentina Gambardella,[10] James W. Smithy,[11] Jérome Fayette,[12] Fan Jin,[13] Y. K. Chang,[14] Kendall Sullivan,[14] Sue Yueh,[14] Ajlan Atasoy,[14] Andrea Sporchia,[14] Ragini Kudchadkar,[14] Hidetoshi Hayashi[15]

[1]Investigational Cancer Therapeutics Department, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Department of Medicine, Roswell Park Comprehensive Cancer Center, Buffalo, NY, USA; [3]Department of Gastrointestinal Oncology, National Cancer Center Hospital East, Kashiwa, Japan; [4]Early Clinical Development Division, The Cancer Institute Hospital of JFCR, Tokyo, Japan; [5]Gastrointestinal Medical Oncology, National Hospital Organization Shikoku Cancer Center, Ehime, Japan; [6]Oncology Department, Barts Cancer Institute, Queen Mary University of London, London, UK; [7]Seoul National University Bundang Hospital, Seongnam, South Korea; [8]Kaohsiung Chang Gung Memorial Hospital, Kaohsiung, Taiwan; [9]Centre Georges-François Leclerc, Dijon, France; [10]INCLIVA Health Research Institute, Valencia, Spain; [11]Department of Medicine, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [12]Medicine Department, Centre Léon Bérard, Lyon, France; [13]Merck & Co., Inc., North Wales, PA, USA; [14]Daiichi Sankyo, Inc, Basking Ridge, NJ, USA; [15]Medical Oncology, Kindai University Hospital, Osaka, Japan

BACKGROUND

- **Increased tumor expression of human epidermal growth factor receptor 3 (HER3)**, a key member of the HER/EGFR family of receptor tyrosine kinases, is associated with **poor clinical outcomes across multiple types of solid tumors**, including **HR+/HER2− breast cancer**[1]
- **Patritumab deruxtecan (HER3-DXd)** is an **antibody-drug conjugate** composed of a human anti-HER3 monoclonal antibody linked to a topoisomerase I inhibitor payload via a stable tetrapeptide-based, tumor-selective, cleavable linker (Figure 1)[2-4]

Figure 1. Structure of HER3-DXd

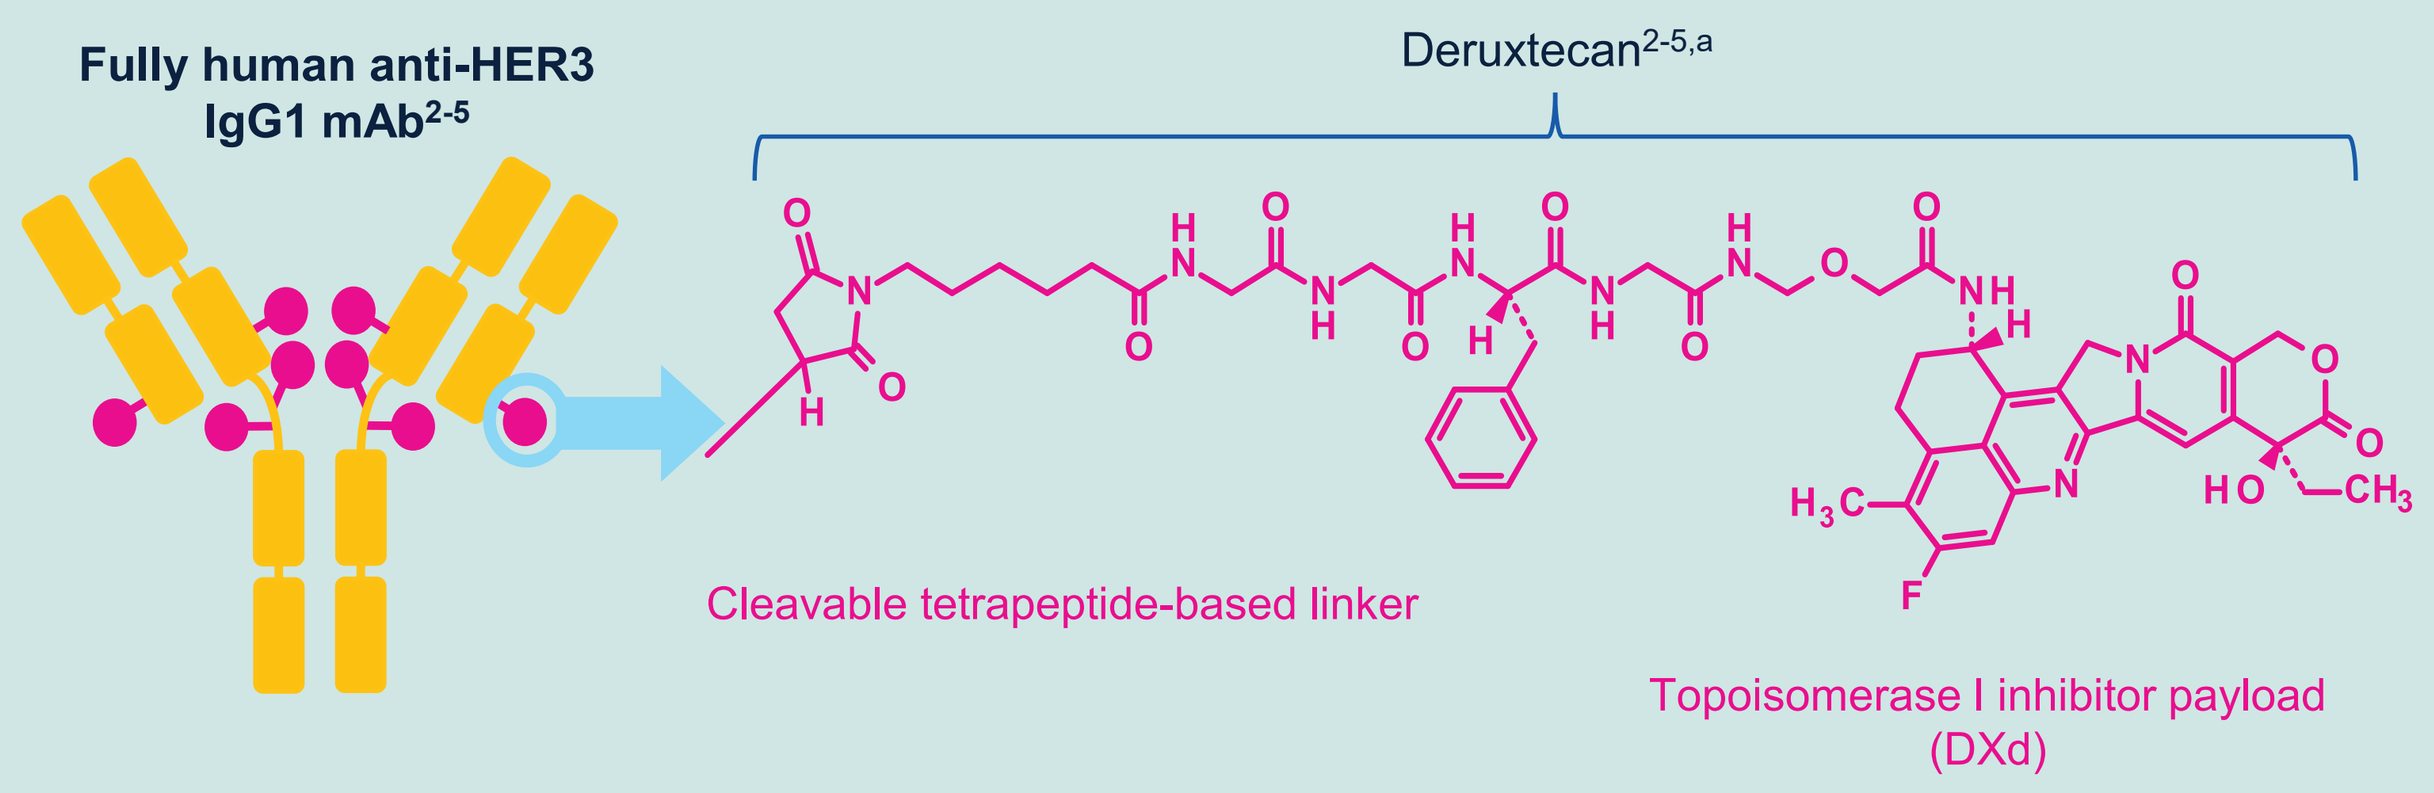
Fully human anti-HER3 IgG1 mAb[2-5]
Deruxtecan[2-5,a]
Cleavable tetrapeptide-based linker
Topoisomerase I inhibitor payload (DXd)

The 7 Key Attributes of HER3-DXd

The mAb directs the DXd ADC to the tumor cell
1. High drug-to-antibody ratio of ≈8[2,5,b]

The linker binds the mAb to the payload
2. Plasma-stable linker-payload[2-4,b]
3. Tumor-selective cleavable linker[2-6,b]

The payload induces cell death when delivered to the tumor
4. Topoisomerase I inhibitor[2-5,b]
5. High potency[2-5,b]
6. Short systemic half-life[2,3,b,c]
7. Bystander antitumor effect[2,7,b]

[a] Refers to the linker and payload. [b] The clinical relevance of these features is under investigation. [c] Based on animal data.

HER3-DXd has shown **dose-dependent antitumor activity** in various xenograft models of breast cancer,[8] with **activity seen across baseline HER3 levels**,[9,10] both in the presence and the absence of HER2 expression[11]

HER3-DXd has demonstrated **clinically meaningful activity and a manageable safety profile** in patients with advanced breast cancer across baseline tumor HER3 expression levels[5,12-14]

Antitumor activity was observed in HR+/HER2– metastatic breast cancer following progression on CDK4/6 inhibitors in the **ICARUS-BREAST01 study**, irrespective of baseline HER3 expression levels (ORR, 54%; 90% CI, 45%-62%),[12] as well as in the 3L setting for patients with **HER3-expressing tumors**, whose disease was refractory to current standard of care therapies from the **U31402-A-J101 study** (ORR, 30%; 95% CI, 22%-39%)[13]

Figure 2. Select Efficacy Data From Trials of HER3-DXd in Metastatic Breast Cancer[12,13]

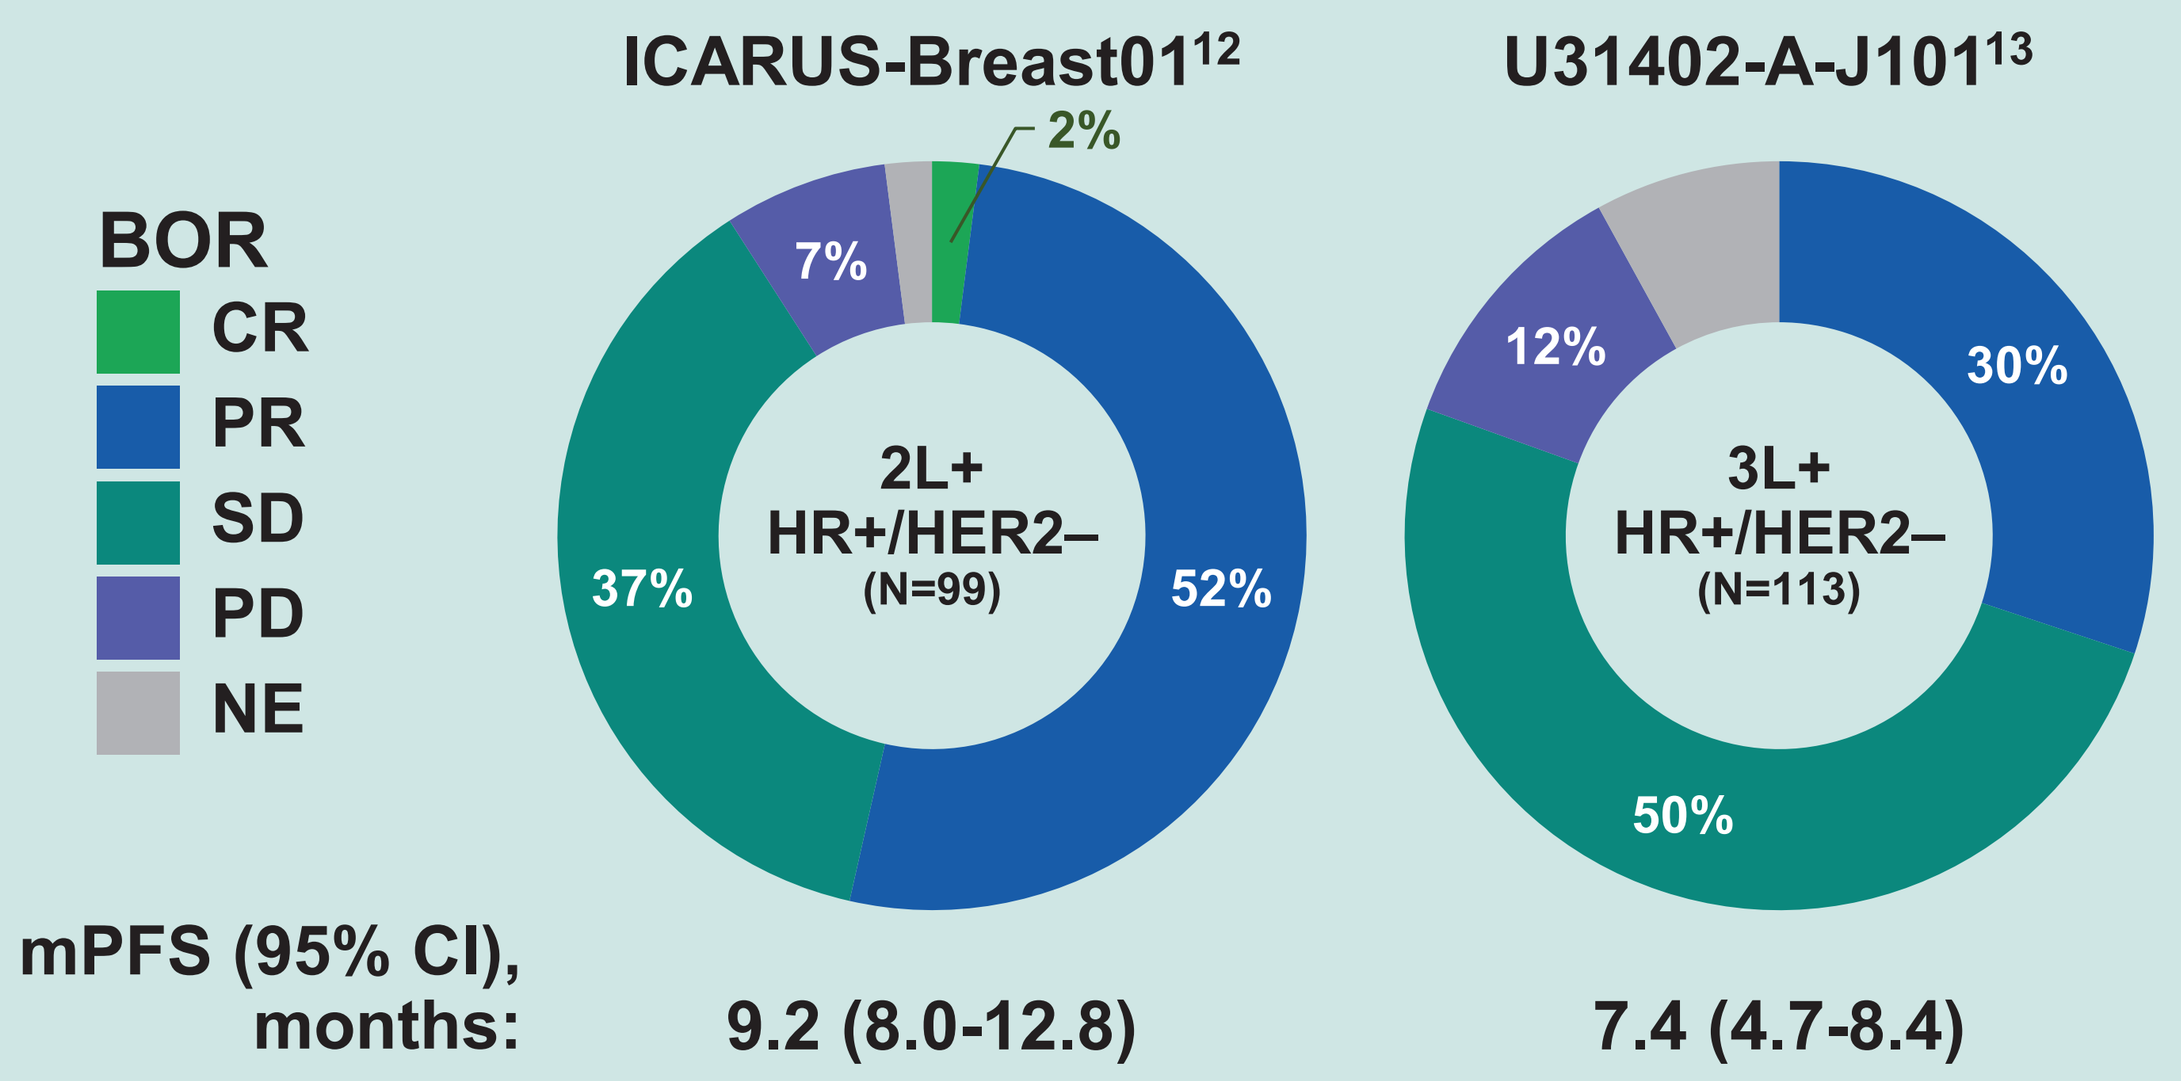
ICARUS-Breast01[12]
U31402-A-J101[13]
BOR
CR
PR
SD
PD
NE
2%
7%
37%
52%
2L+
HR+/HER2–
(N=99)
12%
30%
50%
3L+
HR+/HER2–
(N=113)
mPFS (95% CI), months:
9.2 (8.0-12.8)
7.4 (4.7-8.4)

HERTHENA-PanTumor01 (NCT06172478) is a global, multicohort, open-label, single-arm, phase 2 trial evaluating the efficacy and safety of HER3-DXd in patients with refractory, unresectable locally advanced or metastatic tumors[15,16]

This study includes 13 cohorts in total; 3 recently added cohorts (2L HER2– gastric cancer, non-AGA NSCLC, and metastatic breast cancer) are currently open for enrollment.
Here we present details of the newly opened HR+/HER2− metastatic breast cancer cohort

Copies of this poster obtained through this QR code and/or https://bit.ly/DSMBCC26 are for personal use only and may not be reproduced without written permission of the authors.

Corresponding author: Funda Meric-Bernstam
Contact: fmeric@mdanderson.org

STUDY DESIGN

Key Eligibility Criteria for All Cohorts

- Previously treated, unresectable, stage III or IV metastatic disease
- Pretreatment biopsy
- No preselection based on HER3 expression

- **The HERTHENA-PanTumor01 study includes 10 previously reported cohorts in multiple indications,[15,16] with 3 recently added expansion cohorts**
- **This poster presents the newly opened HR+/HER2– metastatic breast cohort**

HR+/HER2− breast cancer expansion cohort (n=90)

- **Pathologically documented HER2-negative (IHC 0/1+ or IHC 2+/ISH negative) and HR-positive expression as classified by ASCO-CAP guidelines**
- **Prior treatment with only 1 line of chemotherapy for metastatic disease, with clinically or radiologically documented evidence of tumor progression on or after a CDK4/6 inhibitor combined with endocrine therapy[a]**

→ **HER3-DXd 5.6 mg/kg IV Q3W** →

Treatment continues until:
- Disease progression
- TEAE leading to study drug discontinuation
- Withdrawal
- Death
- Pregnancy
- Loss to follow-up

- Tumor assessments will occur Q6W (±7 days) for the first 48 weeks, then Q12W (±14 days)
- Blood samples will be collected for biomarker, pharmacokinetic, and antidrug antibody analyses during treatment and at study discontinuation

[a] Prior treatment with phosphoinositide 3-kinase inhibitors, mechanistic target of rapamycin inhibitors, protein kinase B inhibitors also known as AKT-inhibitors and poly ADP ribose polymerase inhibitors are allowed.

KEY ELIGIBILITY CRITERIA

Inclusion Criteria

All cohorts

- Age ≥18 years
- Locally advanced unresectable or metastatic disease (not curable by surgery or radiation)
- ≥1 measurable lesion[a] (per RECIST version 1.1)
- Pretreatment tumor tissue sample of adequate quality[b]
- ECOG PS of 0 or 1
- Adequate bone marrow reserve and organ function

HR+/HER2− breast cancer expansion cohort

- **Locally advanced or metastatic HR+/HER2− disease (IHC 0/1+ or IHC 2+/ISH negative)[c]**
- Only 1 prior line of chemotherapy for metastatic disease
- Disease progression during or after combined CDK4/6 inhibitor and endocrine therapy

Exclusion Criteria

- **Prior treatment with an anti-HER3 antibody or a topoisomerase I inhibitor** (alone or as part of ADC therapy)
- A history of ILD/pneumonitis requiring corticosteroid therapy or current or suspected ILD/pneumonitis
- Clinically severe respiratory compromise resulting from intercurrent pulmonary illnesses
- Clinically active brain metastases or spinal cord compression,[d] or any history or current evidence of LMD

[a] On CT or MRI by investigator assessment. [b] From ≥1 lesion not previously irradiated and either performed since progression per RECIST 1.1 with the most recent systemic cancer therapy regimen, or freshly collected. Tumor tissue samples must contain ≥100 viable tumor cells. [c] As classified by ASCO-CAP guidelines. [d] Defined as being symptomatic or untreated or requiring therapy with corticosteroids or anticonvulsants.

STUDY ENDPOINTS

Primary Endpoint

- ORR as assessed by investigator per RECIST 1.1

Secondary Endpoints

- Safety and tolerability
- CBR,[a] DOR, DCR, TTR, and PFS as assessed by investigator per RECIST 1.1
- OS
- PK
- Correlation between HER3 protein expression by IHC and efficacy

[a] Defined as the proportion of patients with a best overall response of confirmed complete response, confirmed partial response, or stable disease lasting ≥183 days.

ENROLLMENT STATUS

- **Recruitment for the breast cancer cohort is underway**, with a planned enrollment of **90 patients** in sites spanning **North America**, **Europe**, **East Asia**, **and Australasia**

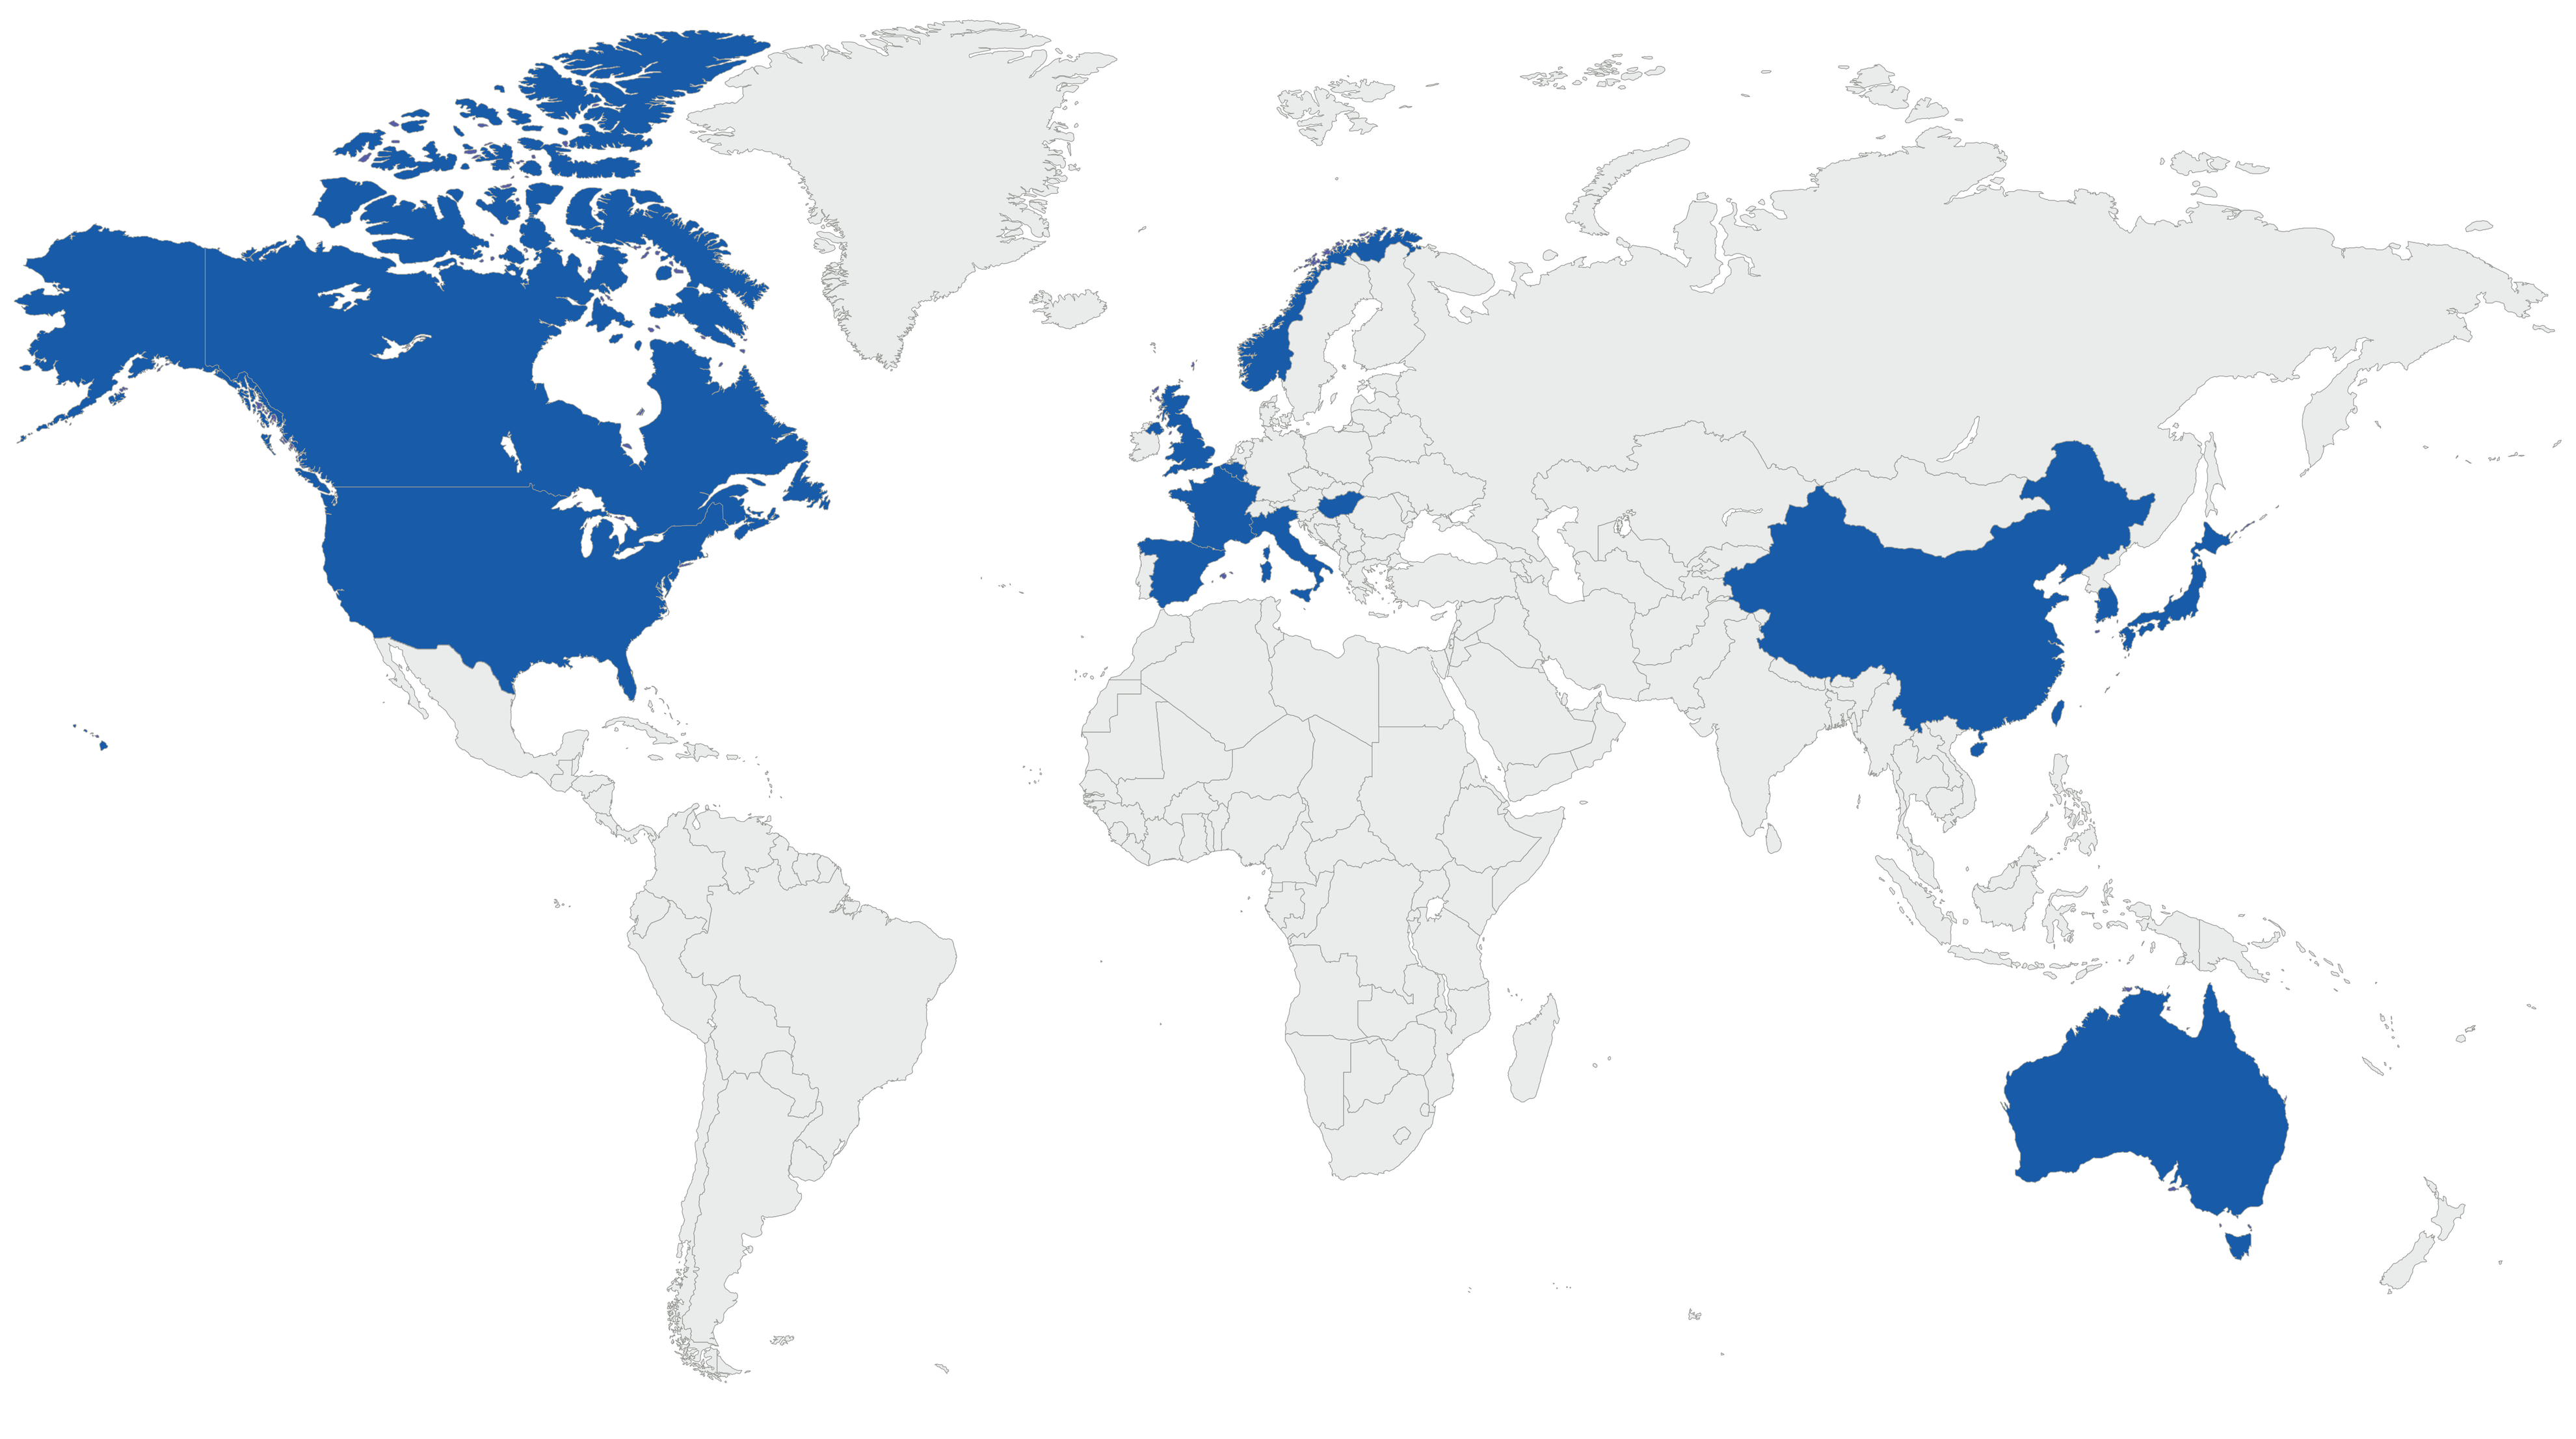

ABBREVIATIONS

2L, second line; 3L, third line; ADC, antibody-drug conjugate; AGA, actionable genomic alteration; ASCO-CAP, American Society of Clinical Oncology–College of American Pathologists; BOR, best overall response; CBR, clinical benefit rate; CDK4/6, cyclin-dependent kinase; CR, complete response; CT, computed tomography; DCR, disease control rate; DOR, duration of response; DXd, topoisomerase I inhibitor payload (an exatecan derivative); ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; HER, human epidermal growth factor receptor; HR, hormone receptor; Ig, immunoglobulin; IHC, immunohistochemistry; ILD, interstitial lung disease; ISH, in situ hybridization; IV, intravenous; LMD, leptomeningeal disease; mAb, monoclonal antibody; mPFS, median PFS; MRI, magnetic resonance imaging; NE, not estimable; NSCLC, non-small cell lung cancer; ORR, objective response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PK, pharmacokinetics; PR, partial response; Q3W, every 3 weeks; Q6W, every 6 weeks; Q12W, every 12 weeks; RECIST, Response Evaluation Criteria in Solid Tumors; SD, stable disease; TEAE, treatment-emergent adverse event; TTR, time to response.

ACKNOWLEDGMENTS

Funding for this study was provided by Daiichi Sankyo Company, Limited, and Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA
Medical writing support was provided by Simon Foale, PhD, and Ellie Angioni, BSc (Nucleus Global, an Inizio company), and was funded by Daiichi Sankyo, Inc. Editorial support was provided in accordance with Good Publication Practice guidelines (https://ismpp.org/gpp-2022).

REFERENCES

1. Inaki K, et al. *PLoS One*. 2022;17(9):e0274140.
2. Nakada T, et al. *Chem Pharm Bull (Tokyo)*. 2019;67(3):173-185.
3. Ogitani Y, et al. *Clin Cancer Res*. 2016;22(20):5097-5108.
4. Koganemaru S, et al. *Mol Cancer Ther*. 2019;18(11):2043-2050.
5. Hashimoto Y, et al. *Clin Cancer Res*. 2019;25(23):7151-7161.
6. Haratani K, et al. *J Clin Invest*. 2020;130(1):374-388.
7. Ogitani Y, et al. *Cancer Sci*. 2016;107(7):1039-1046.
8. Hashimoto Y, et al. AACR 2020. Abstract 5200.
9. Òdena A, et al. *Clin Cancer Res*. 2025;31(12 suppl). Abstract P2-03-25.
10. Òdena A, et al. EACR 2022. Poster P2-013.
11. Koyama K, et al. *Cancer Res*. 2020;80(16 suppl). Abstract 5201.
12. Pistilli B, et al. *Nat Med*. 2025;31(10):3492-3503.
13. Krop IE, et al. *J Clin Oncol*. 2023;41(36):5550-5560.
14. Brasó-Maristany F, et al. *Nat Commun*. 2024;15(1):5826.
15. Bhatia AK, et al. *J Clin Oncol*. 2024;42(16 suppl). Abstract TPS3164.
16. Powles TB, et al. *Ann Oncol*. 2024;35(suppl 2):S534-S535. Abstract 690TiP.

Presented at: 43rd Annual Miami Breast Cancer Conference; March 5-8, 2026; Miami, FL.