IDeate-Lung02: a Phase 3, randomized, open-label study of ifinatamab deruxtecan (I-DXd) vs treatment of Physician's choice in relapsed small cell lung cancer

TPS8126

Taofeek K. Owonikoko,[1] Lauren Byers,[2] Ying Cheng,[3] Hidetoshi Hayashi,[4] Luis Paz-Ares,[5] Maurice Pérol,[6] Jennifer Turner,[7] Meng Qian,[7] Cecilio Roi Garcia,[7] Juliette Godard,[8] Charles M. Rudin[9]

[1]University of Maryland Marlene and Stewart Greenebaum Comprehensive Cancer Center, Baltimore, MD, USA; [2]Department of Thoracic/Head and Neck Medical Oncology, The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [3]Department of Oncology, Jilin Cancer Hospital, Changchun, China; [4]Department of Medical Oncology, Kindai University, Osaka, Japan; [5]Hospital Universitario 12 de Octubre, Madrid, Spain; [6]Léon Bérard Center, Lyon, France; [7]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [8]Daiichi Sankyo, SAS, Rueil-Malmaison, France; [9]Memorial Sloan Kettering Cancer Center, New York, NY, USA.

PURPOSE

- IDeate-Lung02 (NCT06203210) is a Phase 3 trial of I-DXd in adult patients with relapsed SCLC and only 1 prior line of platinum-based therapy[1]
- The study will compare I-DXd with TPC chemotherapy (topotecan, amrubicin, or lurbinectedin) to evaluate the efficacy and safety profile of I-DXd

INTRODUCTION

- Patients with ES-SCLC have few effective treatment options beyond 1L therapy, and there is a substantial unmet need for therapies that provide durable clinical benefit
 - Despite high initial response rates to combination platinum-based chemotherapy plus immunotherapy in 1L ES-SCLC, median OS is 12.3−13.0 months[2,3]
 - Patients with brain metastases have worse survival outcomes (median OS of 8.5 months)[2] and remain difficult to treat[4]
 - Treatment options beyond 1L have limited efficacy (median OS of 6.0–8.1 months with topotecan, 7.5 months with amrubicin, and 9.3 months with lurbinectedin in 2L) and are associated with high rates of hematologic toxicity,[5–8] highlighting the need for novel therapies
- B7-H3 (CD276), a type 1 transmembrane protein belonging to the B7 family, is expressed in many solid tumors but has low or no expression in normal tissue[9–11]
- B7-H3 exhibits moderate-to-high expression in 65% of patients with SCLC, which is associated with a shorter OS (7.4 months vs 23.8 months in patients with low or absent B7-H3)[12]
- I-DXd is a B7-H3–directed ADC comprising a B7-H3 mAb linked to a topoisomerase I inhibitor payload (DXd) via a stable cleavable linker, designed to enhance selective tumor-cell death and reduce systemic exposure[13] (**Figure 1**)
- In the Phase 1/2 IDeate-Pantumor01 study (NCT04145622), I-DXd demonstrated durable efficacy (ORR, 52.4%; median DOR, 5.9 months; median OS, 12.2 months) irrespective of B7-H3 status in 21 patients with pretreated SCLC[14]
- I-DXd is being investigated in a dose-optimization Phase 2 study in patients with pretreated ES-SCLC (IDeate-Lung01 [NCT05280470])[15]
- Here, we describe IDeate-Lung02 (NCT06203210), a Phase 3 trial comparing I-DXd with TPC (topotecan, amrubicin, or lurbinectedin) in patients with relapsed SCLC who have received only 1 prior line of platinum-based therapy[1]

Figure 1. I-DXd was designed with 7 key attributes

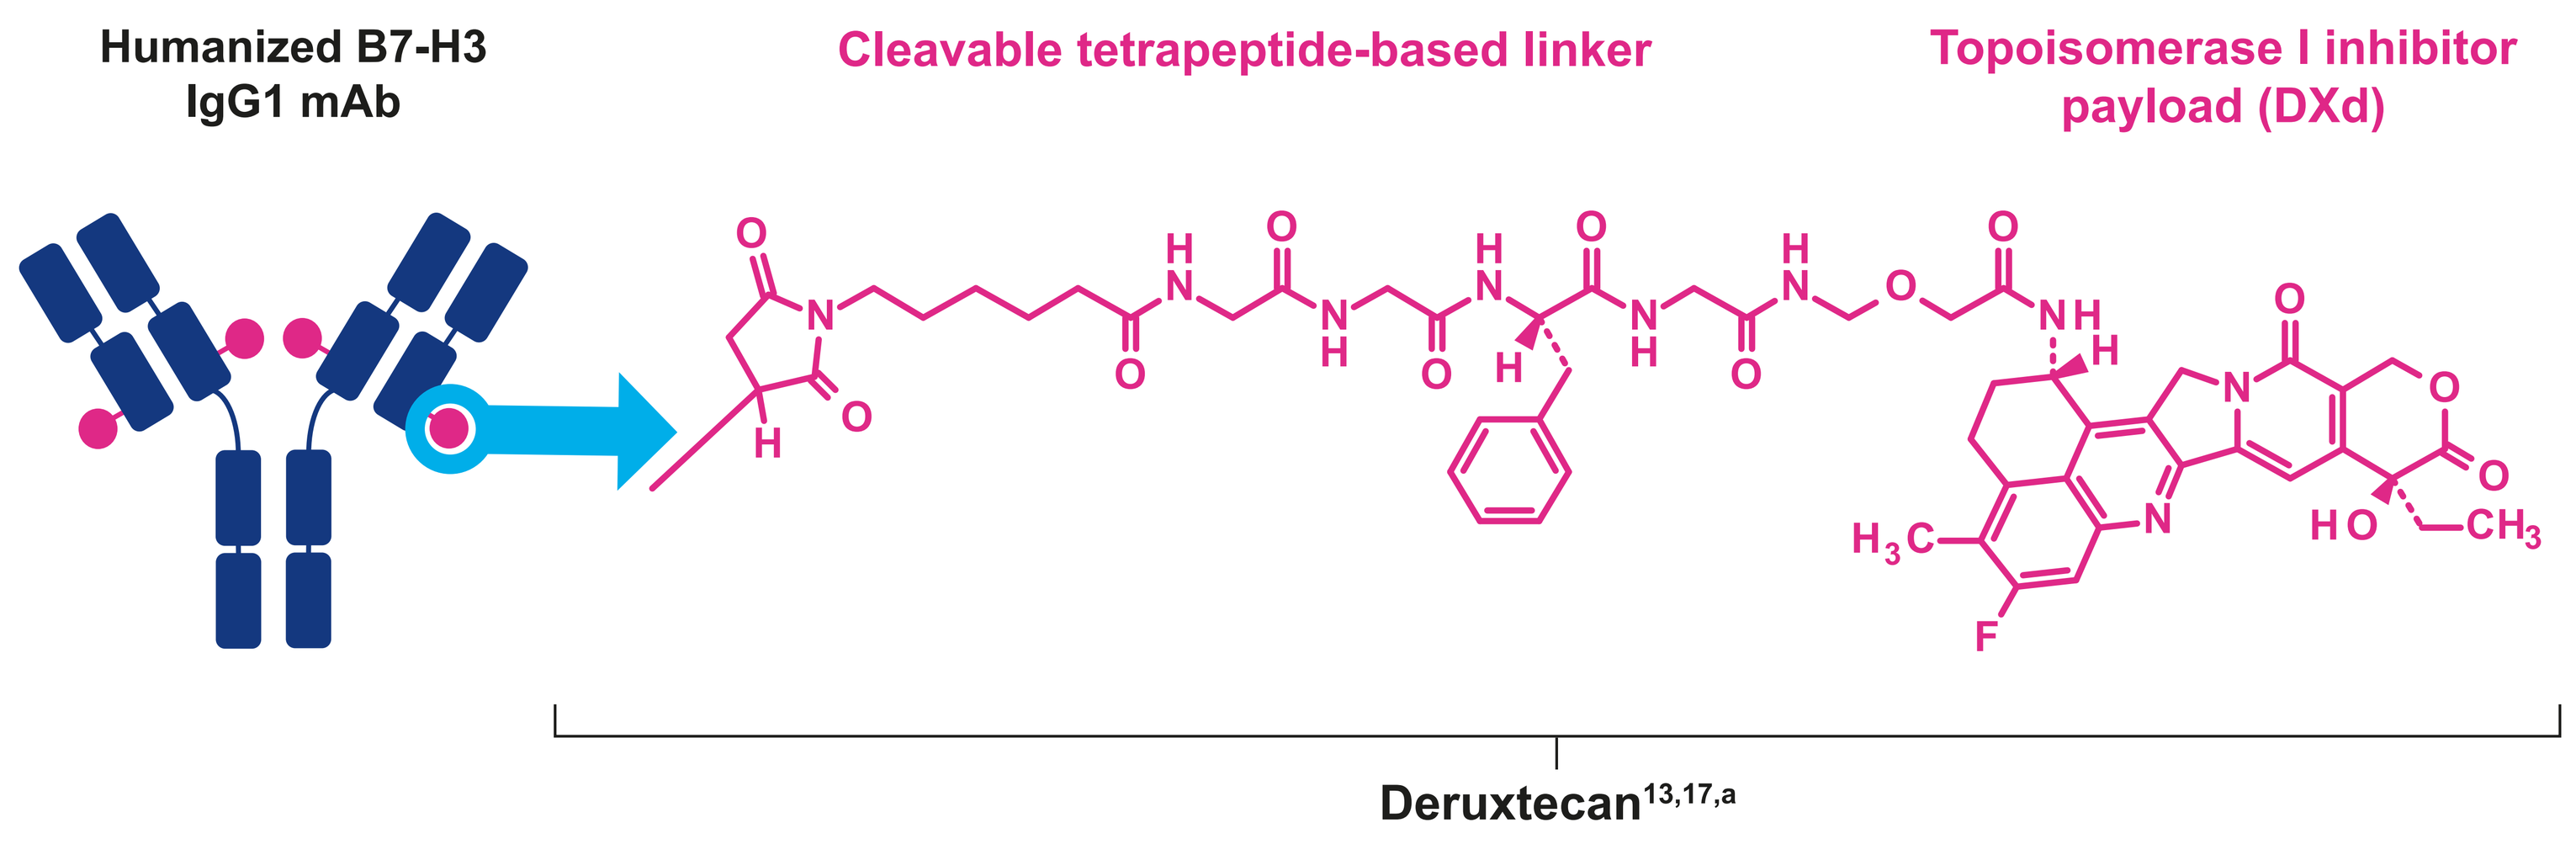
Humanized B7-H3
IgG1 mAb
Cleavable tetrapeptide-based linker
Topoisomerase I inhibitor
payload (DXd)
Deruxtecan[13,17,a]

The **mAb** directs the DXd ADC to the tumor cell

1. Optimized drug-to-antibody ratio ~4[13,16,b,c]

The **linker** binds the mAb to the payload

2. Plasma-stable linker–payload[13,16,b]
3. Tumor-selective cleavable linker[13,16,b]

The **payload** induces cell death when delivered to the tumor

4. Topoisomerase I inhibitor[13,16,17,b]
5. High potency[13,16,b]
6. Short systemic half-life[13,16,b,c]
7. Bystander antitumor effect[16,17,18,b]

[a]Refers to the linker and payload. [b]The clinical relevance of these features is under investigation. [c]Based on animal data.

METHODS

- IDeate-Lung02 (NCT06203210) is a global, multicenter, randomized, open-label, Phase 3 trial comparing I-DXd with TPC in adult patients with relapsed SCLC who had received only 1 prior line of platinum-based therapy
- A total of 468 patients (~234 in each arm) will be randomized 1:1 to receive either I-DXd 12 mg/kg IV Q3W or TPC (topotecan, amrubicin, or lurbinectedin; **Figure 2**)
- The dual primary endpoints are ORR assessed by BICR per RECIST 1.1 and OS; study endpoints are presented in **Table 1**
- Patients are being enrolled across sites in Asia, Australia, Europe, North America, and South America (**Figure 3**)

Figure 2. Study design

Key inclusion criteria	Key exclusion criteria
Histologically or cytologically documented SCLC	Prior treatment with orlotamab, enoblituzumab, or other B7-H3–targeted agents, including I-DXd
Age ≥18 years or minimal legal adult age (whichever is greater)	Prior discontinuation of an ADC that consists of an exatecan derivative (eg, trastuzumab deruxtecan) due to treatment-related toxicities
Received only 1 prior line of platinum-based therapy	Prior treatment with any of the comparators or a topoisomerase I inhibitor
≥1 measurable lesion per RECIST 1.1	Clinically active brain metastasis, spinal cord compression, or leptomeningeal carcinomatosis
Radiologically documented PD on or after platinum-based therapy	Clinically severe pulmonary compromise resulting from intercurrent pulmonary illnesses
ECOG PS 0–1	History of ILD/pneumonitis that required corticosteroids; current or suspected ILD/pneumonitis
Must provide adequate baseline tumor samples of sufficient quantity and quality	Uncontrolled or significant cardiovascular disease
Patients with asymptomatic brain metastases (untreated or previously treated) are eligible	Known, uncontrolled HIV infection; active or uncontrolled HBV or HCV infection; uncontrolled systemic bacterial, fungal, or viral infection; or active, known, or suspected autoimmune disease

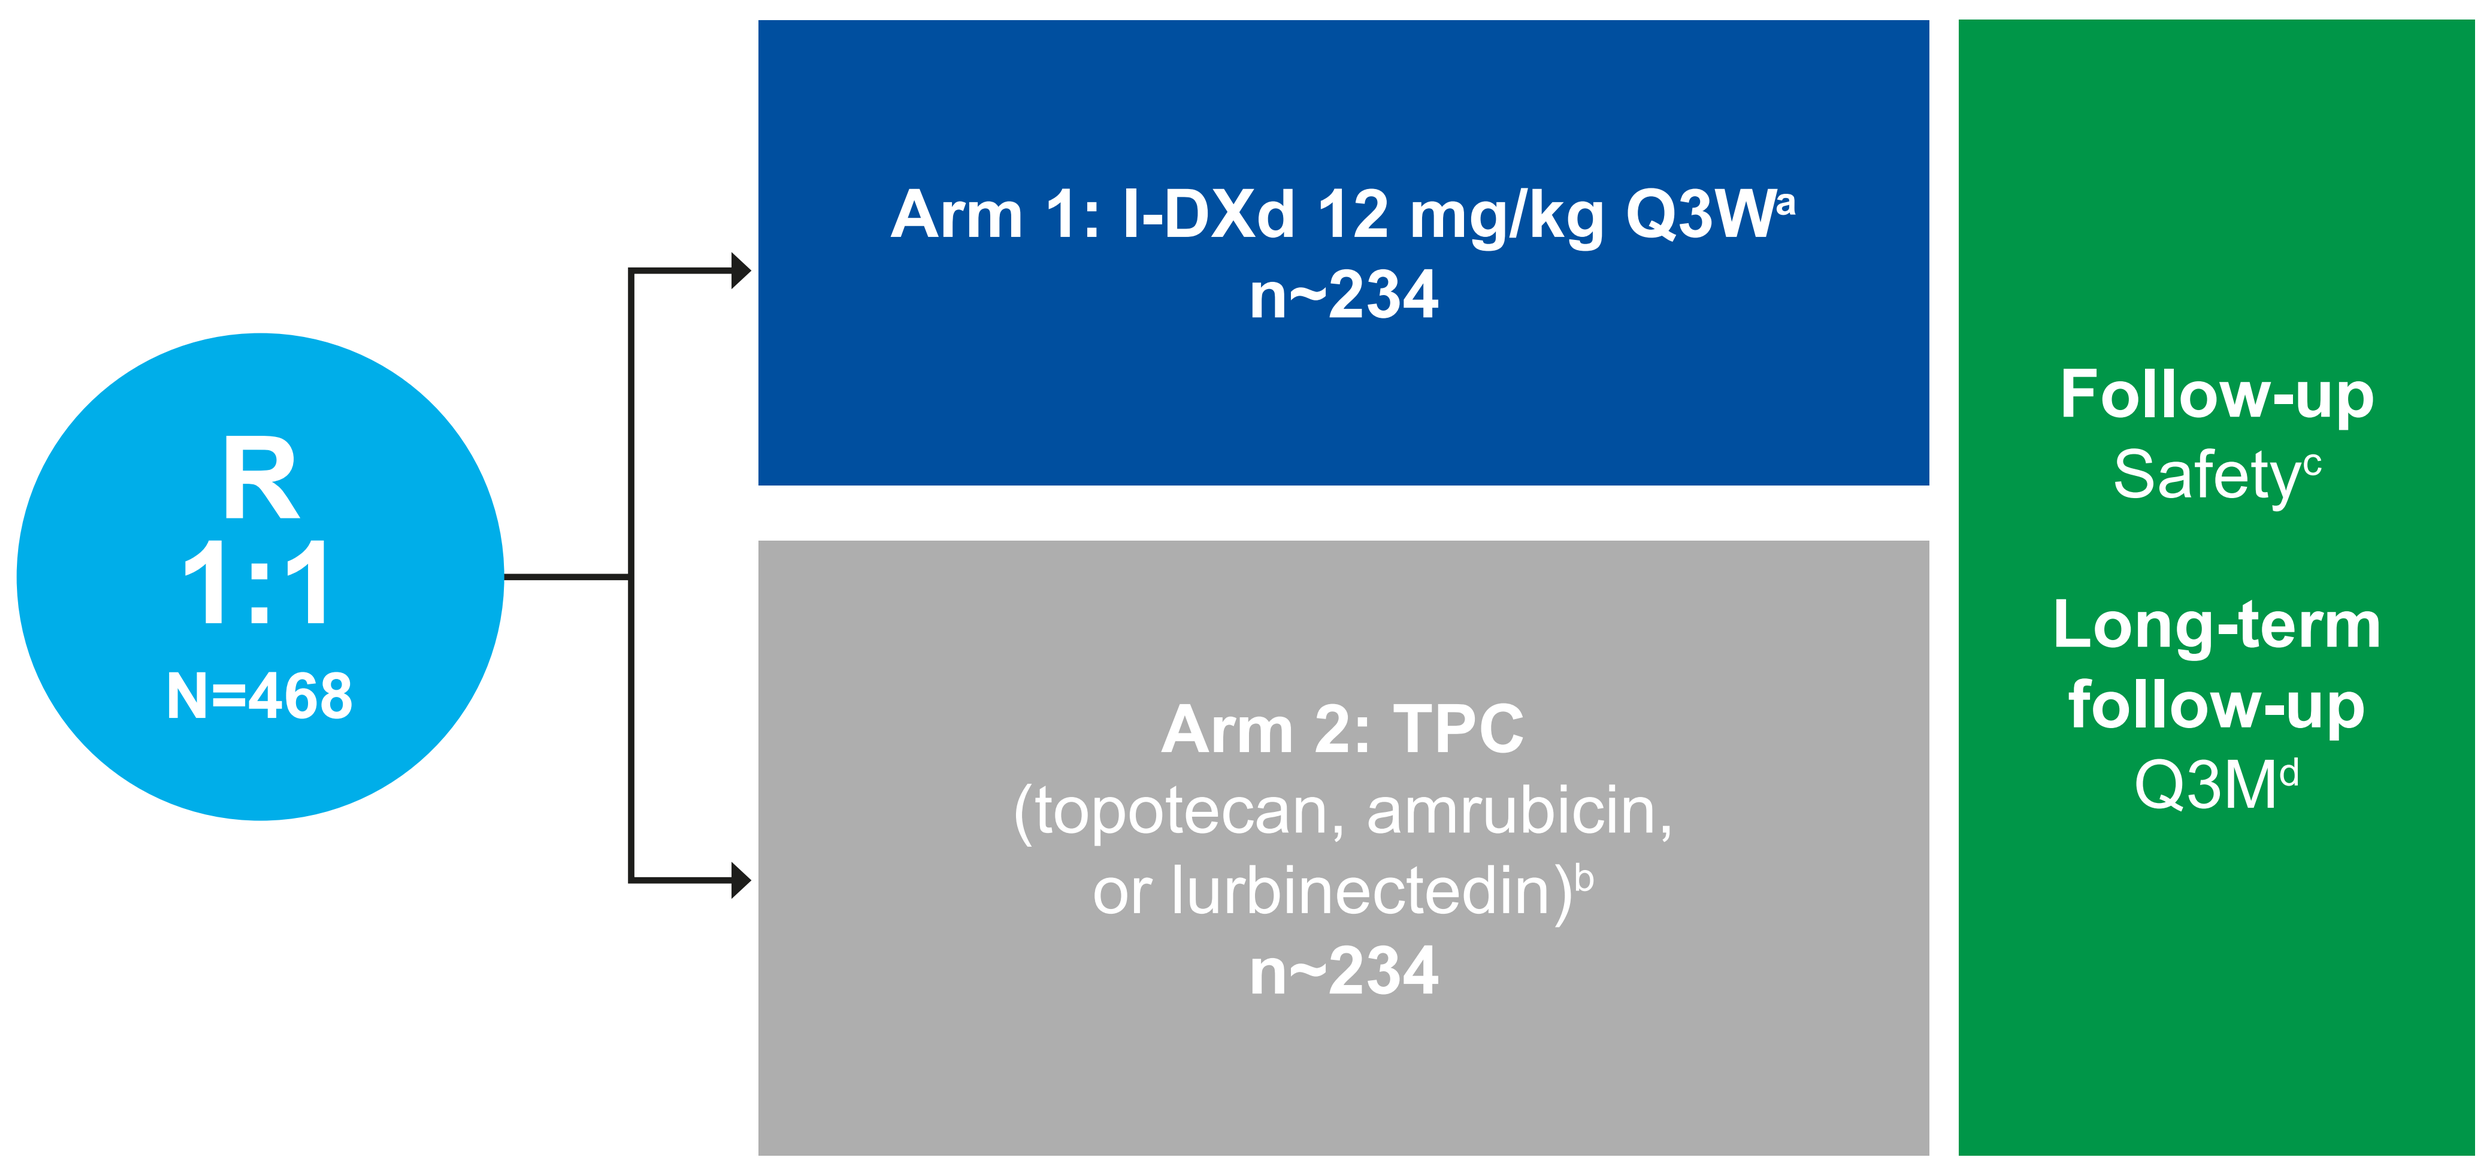
R
1:1
N=468
Arm 1: I-DXd 12 mg/kg Q3W[a]
n~234
Arm 2: TPC
(topotecan, amrubicin,
or lurbinectedin)[b]
n~234
Follow-up
Safety[c]
Long-term
follow-up
Q3M[d]

Stratification

Chemotherapy-free interval following 1L therapy (<90 vs ≥90 days)
TPC (topotecan vs amrubicin vs lurbinectedin)
Treatment with prior PD-(L)1 inhibitors (yes vs no)
Presence or history of asymptomatic brain metastases (yes vs no)

[a]Until PD, unacceptable toxicity, loss to follow-up, consent withdrawal, death, or other reason. [b]Comparator treatments will only be utilized in countries where they are approved in second and subsequent LoTs for patients with SCLC who progressed on or after platinum-based therapy; ≥50% of patients in the comparator group will receive topotecan. [c]Safety follow-up visit will occur 40 days (+7 days) after the last dose. [d]Long-term follow-up will occur to assess survival; assess tumor progression until PD for patients discontinuing for reasons other than PD; and to collect information on further anticancer treatments, Q3M (90 ±14 days) from study-drug discontinuation (end of treatment), withdrawal of consent, or from when a study-termination criterion is met.

Key statistical considerations:

- BICR-assessed ORR will be analyzed using a Cochran–Mantel–Haenszel test at a 2-sided 1% alpha level
- OS will be analyzed using a log-rank test at a 2-sided 4% significance level, under a 2-look group sequential design
- An estimated 356 OS events will be needed to detect a hazard ratio of 0.70 with 90% power

Table 1. Study endpoints

Primary endpoints
ORR assessed by BICR[a]
OS
Secondary endpoints
ORR assessed by Investigator[a]
PFS assessed by BICR and Investigator[a]
DOR assessed by BICR and Investigator[a]
DCR assessed by BICR and Investigator[a]
TTR assessed by BICR and Investigator[a]
Patient-reported outcomes (EORTC questionnaires)
Safety[b]
Immunogenicity
Pharmacokinetics
Relationship between B7-H3 expression and outcomes

[a]Per RECIST 1.1. [b]TEAEs, deaths, serious TEAEs, adverse events of special interest, and TEAEs leading to dose modification or discontinuation.

Figure 3. Study site locations

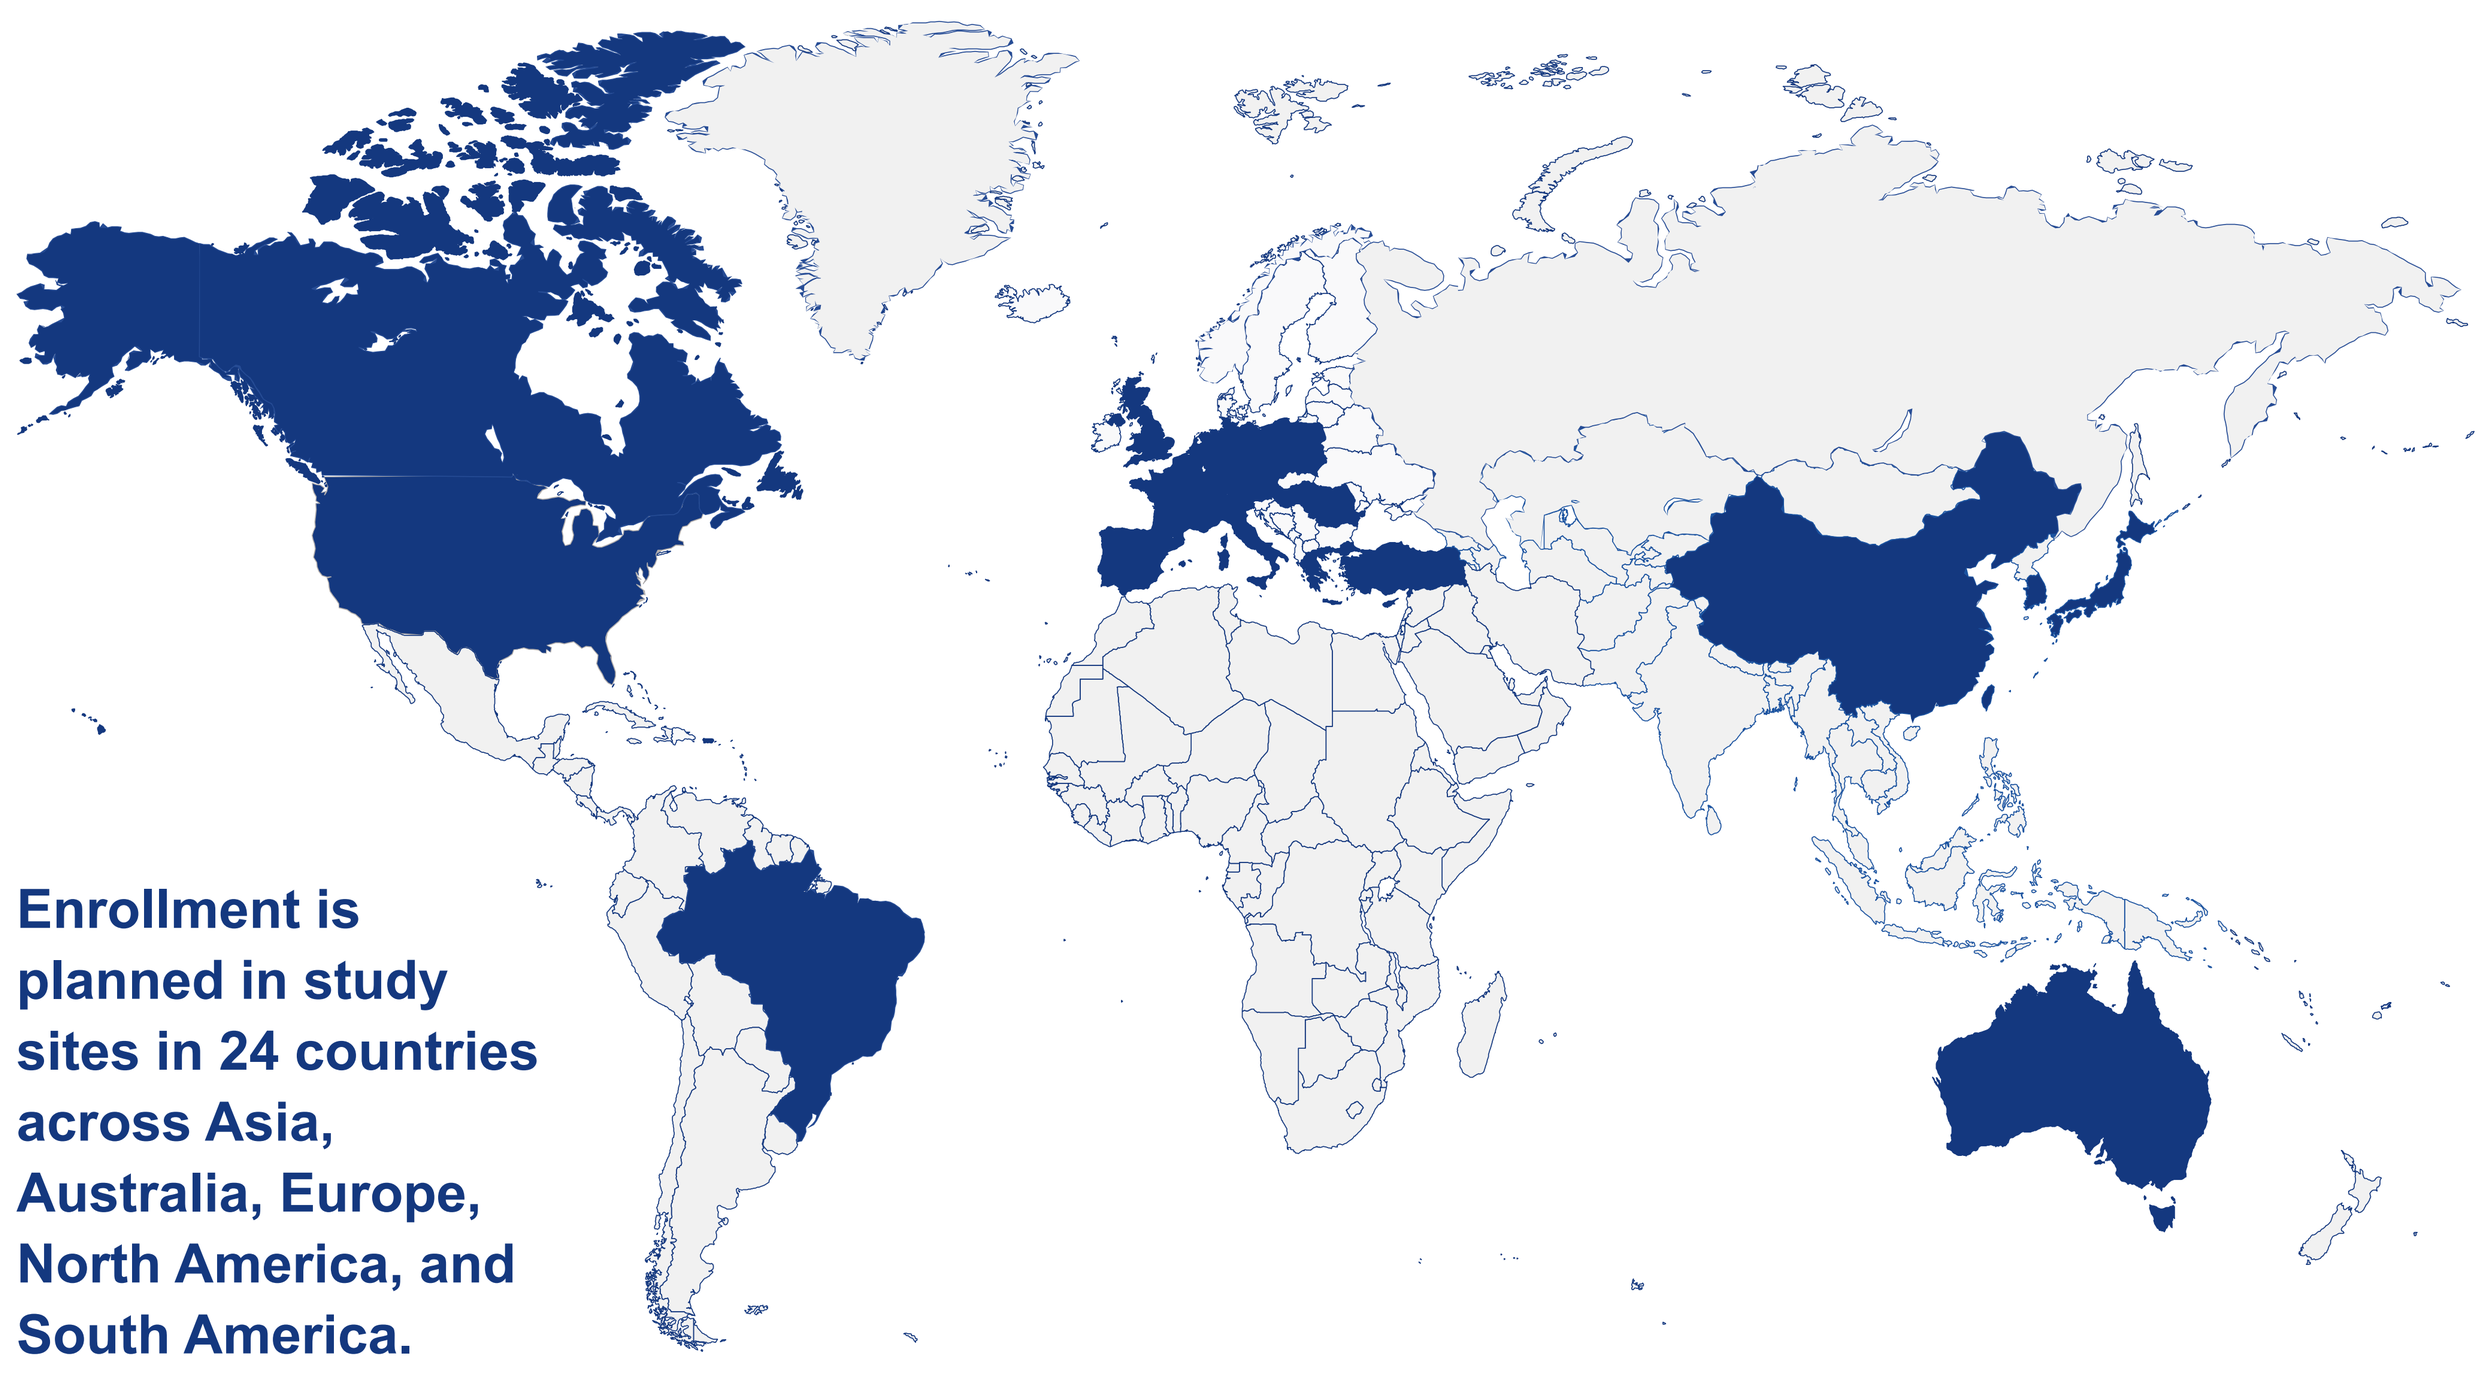
Enrollment is planned in study sites in 24 countries across Asia, Australia, Europe, North America, and South America.

REFERENCES

1. ClinicalTrials.gov. https://clinicaltrials.gov/study/NCT06203210. Accessed April 22, 2024.
2. Horn L, et al. *N Engl J Med.* 2018;379:2220–2229.
3. Paz-Ares L, et al. *Lancet.* 2019;394:1929–1939.
4. Althoff FC, et al. *Front Oncol.* 2023;13:1273478.
5. Eckardt JR, et al. *J Clin Oncol.* 2007;25:2086–2092.
6. O'Brien ME, et al. *J Clin Oncol.* 2006;24:5441–5447.
7. Patel S, et al. *Ther Adv Med Oncol.* 2021;13:17588359211020529.
8. von Pawel J, et al. *J Clin Oncol.* 2014;32:4012–4019.
9. Getu AA, et al. *Mol Cancer.* 2023;22:43.
10. Dong P, et al. *Front Oncol.* 2018;8:264.
11. Wang L, et al. *Tumour Biol.* 2016;37:2961–2971.
12. Qiu MJ, et al. *Front Oncol.* 2021;11:600238.
13. Yamato M, et al. *Mol Cancer Ther.* 2022;21:635–646.
14. Patel MR, et al. Presented at the European Society for Medical Oncology Congress 2023. October 20–24, 2023; Madrid, Spain. Abstract 690P.
15. ClinicalTrials.gov. https://clinicaltrials.gov/study/NCT05280470. Accessed February 29, 2024.
16. Daiichi Sankyo. Data on file.
17. Nakada T, et al. *Chem Pharm Bull (Tokyo).* 2019;67:173–185.
18. Ogitani Y, et al. *Cancer Sci.* 2016;107:1039–1046.

ABBREVIATIONS

1L, first-line; **2L**, second-line; **ADC**, antibody–drug conjugate; **B7-H3**, B7 homolog 3; **BICR**, blinded independent central review; **CD276**, cluster of differentiation 276; **DCR**, disease control rate; **DOR**, duration of response; **ECOG PS**, Eastern Cooperative Oncology Group performance status; **EORTC**, European Organisation for Research and Treatment of Cancer; **ES**, extensive-stage; **HBV**, hepatitis B virus; **HCV**, hepatitis C virus; **HIV**, human immunodeficiency virus; **I-DXd**, ifinatamab deruxtecan; **IgG1**, immunoglobulin G1; **ILD**, interstitial lung disease; **IV**, intravenously; **LoT**, line of therapy; **mAb**, monoclonal antibody; **ORR**, objective response rate; **OS**, overall survival; **PD**, progressive disease; **PD-1**, programmed cell death protein 1; **PD-L1**, programmed death ligand 1; **PFS**, progression-free survival; **Q3M**, every 3 months; **Q3W**, every 3 weeks; **R**, randomization; **RECIST 1.1**, Response Evaluation Criteria in Solid Tumors, version 1.1; **SCLC**, small cell lung cancer; **TEAE**, treatment-emergent adverse event; **TPC**, treatment of Physician's choice; **TTR**, time to response.

ACKNOWLEDGMENTS

Funding for this study was provided by Daiichi Sankyo Company, Limited (Daiichi Sankyo, Co., Ltd.) and Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc. Rahway, NJ, USA.

Medical writing support was provided by Dave Buist, PhD, of BOLDSCIENCE®, Inc., and was funded by Daiichi Sankyo, Inc. and Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.

Editorial support was provided in accordance with Good Publication Practice guidelines https://www.ismpp.org/gpp-2022.

Copies of this presentation obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

382b

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting; May 31–June 4, 2024; Chicago, IL, USA.