San Antonio Breast Cancer Symposium, December 10–13, 2024

P5-07-24

HER2+ early breast cancer treatment and outcomes by risk of recurrence: a retrospective US electronic health records study

Reshma Mahtani,[1] Gregory Vidal,[2] Sam Hillman,[3]* Ellie John,[4] Simon M Collin,[5] Gráinne Long[5]

[1]Miami Cancer Institute, FL, US; [2]West Cancer Center & Research Institute, Germantown, TN, US; [3]RWE Data Science, Global Data and Analytics, Oncology Business Unit, AstraZeneca, Cambridge, UK; [4]GMA Payer Biometrics, Statistics, Oncology Business Unit, AstraZeneca, Cambridge, UK; [5]Oncology Outcomes Research (O2R), Evidence Generation, Publications, and Partnerships, Global Medical Affairs, Oncology Business Unit, AstraZeneca, Cambridge, UK

*Has moved organization since publication initiated; current affiliation: Daiichi Sankyo, Uxbridge, UK

Objective

- To describe use of neoadjuvant and (post-neo)adjuvant therapies* and clinical outcomes by risk of recurrence in a real-world population of patients with human epidermal growth factor receptor 2–positive (HER2+) early breast cancer (eBC) from the US

Conclusions

- Within 5 years of diagnosis, 28% of patients with high-risk HER2+ eBC had invasive disease or died compared with 19% of patients with non-high-risk HER2+ eBC
- Use of neoadjuvant and (post-neo)adjuvant therapy increased over time in patients with HER2+ eBC who were at high risk of recurrence
- Although recent guidelines recommend the use of neoadjuvant therapy,[1] one-third of patients with high-risk HER2+ eBC did not receive neoadjuvant therapy between 2018 and 2021
- There remains a need for more effective therapies to optimize patient outcomes in HER2+ eBC, particularly for patients at high risk of recurrence

*The term (post-neo)adjuvant is used in this study to describe treatment received after surgery, post-neoadjuvant or adjuvant

Plain language summary

Why did we perform this research?

In roughly 20% of breast cancers, tumors have higher than normal levels of a protein called human epidermal growth factor receptor 2 (HER2-positive), which helps cancer cells to grow.[1,2] Some people with HER2-positive (HER2+) early breast cancer (cancer that has not spread beyond the breast or the lymph nodes in the armpits) are at higher risk of the cancer coming back after receiving treatment.[1] We performed this study to understand how people with high-risk and non-high-risk HER2+ early breast cancer are treated and how well the currently available treatments work.

How did we perform this research?

We used a database of electronic health records to collect information about the treatments and survival rates of people from the US with high-risk and non-high-risk early breast cancer (people at high risk were those with more severe disease based on tumor size and cancer stage). We focused on people diagnosed with HER2+ early breast cancer between 2011 and 2021.

What were the findings of this research?

We saw increases in the use of drug treatments given before and after surgery to reduce cancer in recent years (2018–2021) compared with an earlier period (2011–2013) in people with high-risk and non-high-risk HER2+ early breast cancer. Despite this increase, many people at high risk still did not receive neoadjuvant therapy. After 5 years of treatment for HER2+ early breast cancer, people at high risk were more likely to experience the return of cancer or death compared with those at non-high risk. People at high risk, compared with non-high risk, also experienced a faster progression from early breast cancer to metastatic breast cancer (cancer that has spread beyond the breast).

What are the implications of this research?

Despite the increase in the use of neoadjuvant and (post-neo)adjuvant treatments in recent years, there is a need for treatments that improve survival and reduce the likelihood of cancer returning in people with HER2+ early breast cancer.

1. Morales S, et al. *Cancers (Basel)*. 2021;13:5771; 2. Wolff AC, et al. *J Clin Oncol*. 2013;31:3997–4013

Poster

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of the virtual poster. Alternatively, please click on the link: https://bit.ly/3zK3q4V

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.

This study was sponsored by AstraZeneca. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201).
Poster presented at SABCS 2024, December 10–13, San Antonio, TX, US, by Reshma Mahtani. Corresponding author email address: Reshma.Mahtani@baptisthealth.net

Introduction

- Breast cancer (BC) is the most common cancer in women worldwide, and the second most common overall, with 2.31 million new cases in 2022[2]
- It is estimated that HER2+ BC accounts for up to 20% of all BC cases,[3,4] and is associated with high recurrence and mortality[5]
- In the US, around two-thirds of BC cases are diagnosed as eBC[6]
- The current recommended treatment for HER2+ eBC in the neoadjuvant setting consists of a multi-agent regimen of dual human epidermal growth factor receptor-directed therapy with chemotherapy[1,7]
- Patients with HER2+ eBC remain at risk of recurrence despite the use of standard-of-care treatments[8,9]
- There is limited information on real-world treatment patterns and how treatment outcomes might differ by risk of recurrence in patients with HER2+ eBC

Methods

- This was a retrospective observational study in patients with HER2+ eBC who received neoadjuvant and/or (post-neo)adjuvant treatment* **(Figure 1)**
- De-identified patient-level data were obtained from the US nationwide Flatiron Health electronic health record-derived database†
- Analyses were categorized by date of eBC diagnosis: 2011–2013, 2014–2017, or 2018–2021
- Patient data were stratified by high-risk or non-high-risk status, and treatment data included receipt of neoadjuvant therapy and therapy after surgery ([post-neo]adjuvant)

Figure 1. Study design

Cohort inclusion criteria and patient flow

Starting population (N=14312)
↓
Patients who were diagnosed with eBC between January 2011 and December 2021 and who were ≥18 years of age at the time of eBC diagnosis (n=13127)
↓
Record of group Stage I, II, or IIIa at eBC diagnosis (either clinical or pathologic) (n=11247)
↓
Patients did not receive treatment as part of a clinical trial during the study period (n=11073)
↓
Patients with no other prior primary cancers or other malignancies (except non-melanoma skin cancers) (n=10774)

→ **Patients with HER2+ eBC (N=1290)**

High risk of relapse (n=366)	Non-high risk of relapse (n=924)

Patients were defined as **'high risk'** if they met either of the following criteria:‡
A) **Pathologic stage (n=261)**
- Any node-positive tumor or large tumor size (T3/T4) if node negative

B) **Clinical stage in patients who received neoadjuvant therapy (n=133)**
- IIb, IIc, IIIa, IIIb, or IIIc

↓

Patients were followed from diagnosis to death, last known activity date, or end of study period (December 31, 2022), whichever occurred first, to analyze:
- Patient and tumor characteristics
- **Neoadjuvant** and **(post-neo)adjuvant** treatments received

*The term (post-neo)adjuvant is used in this study to describe treatment received after surgery, post-neoadjuvant or adjuvant; †the US nationwide Flatiron Health electronic health record-derived database consists of processed longitudinal patient-level medical record data (structured and unstructured) obtained from ~280 US cancer clinics (~800 sites of care; primarily community and academic cancer clinics). Data are curated via technology-enabled abstraction including demographics, diagnosis (eg staging, histopathology, and biomarkers), treatment, and outcomes (eg mortality);[10,11] ‡n=28 patients met criteria for both clinical and pathologic high risk
eBC, early breast cancer; HER2+, human epidermal growth factor receptor 2–positive

Results and interpretation

- A total of 1290 patients with HER2+ eBC were eligible within the diagnosis dates (2011–2013, n=351; 2014–2017, n=515; 2018–2021, n=424). Patient demographic and clinical characteristics are shown in **Table 1**

Table 1. Patient demographic and clinical characteristics

	High-risk HER2+ eBC (n=366)	Non-high-risk HER2+ eBC (n=924)
Age, median years (min–max)	56.0 (23.0–84.0)	59.0 (23.0–85.0)
Female, n (%)	363 (99.2)	918 (99.4)
HR status, n (%)		
HR+	265 (72.4)	675 (73.1)
HR−	100 (27.3)	249 (26.9)
Unknown	1 (0.3)	0
Histology, n (%)		
IDC	345 (94.3)	853 (92.3)
ILC	12 (3.3)	34 (3.7)
Other	5 (1.4)	30 (3.2)
Unknown	4 (1.1)	7 (0.8)
Treatment pathway, n (%)*		
No treatment	1 (0.3)	34 (3.7)
Neoadjuvant treatment followed by surgery	3 (0.8)	1 (0.1)
Surgery only	41 (11.2)	159 (17.2)
Surgery followed by adjuvant treatment	161 (44.0)	547 (59.2)
Neoadjuvant treatment followed by surgery and post-neoadjuvant treatment	160 (43.7)	183 (19.8)
pCR after neoadjuvant therapy, n/number for which pCR was recorded (%)†	70/161 (43.5)	106/182 (58.2)

*No patients received neoadjuvant treatment only; †pCR was not recorded for two high-risk and two non-high-risk patients
eBC, early breast cancer; IDC, invasive ductal carcinoma; ILC, invasive lobular carcinoma; HER2+, human epidermal growth factor receptor 2–positive; HR, hormone receptor; HR+, HR-positive; HR−, HR-negative; pCR, pathologic complete response

- A total of 1258 patients with HER2+ eBC received treatment, 347 (27.6%) patients received neoadjuvant therapy, and 1050 (83.5%) patients received (post-neo)adjuvant therapy. Breakdown of neoadjuvant and (post-neo)adjuvant therapy over time by hormone receptor status and drug class is shown in **Table 2**

Table 2. Neoadjuvant and (post-neo)adjuvant therapy use in patients with high-risk and non-high-risk HER2+ eBC, according to HR status and drug class

	High-risk HER2+ eBC*						Non-high-risk HER2+ eBC†					
	2011–2013		2014–2017		2018–2021		2011–2013		2014–2017		2018–2021	
	HR+ (n=80)	HR− (n=29)	HR+ (n=113)	HR− (n=33)	HR+ (n=72)	HR− (n=38)	HR+ (n=182)	HR− (n=59)	HR+ (n=271)	HR− (n-98)	HR+ (n=222)	HR− (n=92)
Neoadjuvant therapy												
CT, n (%)	14 (17.5)	6 (20.7)	47 (41.6)	21 (63.6)	45 (62.5)	27 (71.1)	8 (4.4)	1 (1.7)	42 (15.5)	22 (22.4)	77 (34.7)	29 (31.5)
ET, n (%)	0	0	3 (2.7)	1 (3.0)	8 (11.1)	3 (7.9)	1 (0.5)	0	2 (0.7)	0	5 (2.3)	2 (2.2)
HER2-directed, n (%)‡	12 (15.0)	4 (13.8)	48 (42.5)	21 (63.6)	45 (62.5)	26 (68.4)	7 (3.8)	1 (1.7)	41 (15.1)	21 (21.4)	77 (34.7)	29 (31.5)
(Post-neo)adjuvant therapy												
CT, n (%)	29 (36.3)	7 (24.1)	40 (35.4)	9 (27.3)	19 (26.4)	6 (15.8)	60 (33.0)	30 (50.8)	104 (38.4)	36 (36.7)	78 (35.1)	30 (32.6)
ET, n (%)	55 (68.8)	2 (6.9)	76 (67.3)	4 (12.1)	45 (62.5)	1 (2.6)	120 (65.9)	2 (3.4)	172 (63.5)	4 (4.1)	137 (61.7)	1 (1.1)
HER2-directed, n (%)‡	45 (56.3)	15 (51.7)	85 (75.2)	28 (84.8)	61 (84.7)	34 (89.5)	77 (42.3)	33 (55.9)	157 (57.9)	54 (55.1)	153 (68.9)	63 (68.5)

*160 (43.7%) patients with high-risk HER2+ eBC received both neoadjuvant and (post-neo)adjuvant therapies; †183 (19.8%) patients with non-high-risk HER2+ eBC received both neoadjuvant and (post-neo)adjuvant therapies; ‡other treatments comprising CDK4/6 inhibitors, immunotherapies, non-HER2 ADC, anti-angiogenesis drugs, PARP inhibitors, PI3Ks, TKIs, and other targeted therapies are not shown and may include some HER2-directed therapies
ADC, antibody-drug conjugate; CDK4/6, cyclin-dependent kinase 4/6; CT, chemotherapy; eBC, early breast cancer; ET, endocrine therapy; HER2, human epidermal growth factor receptor 2; HER2+, HER2-positive; HR, hormone receptor; HR+, HR-positive; HR−, HR-negative; PARP, poly-ADP ribose polymerase; PI3K, phosphoinositide 3-kinase; TKI, tyrosine kinase inhibitor

- For patients with high-risk HER2+ eBC defined by pathologic stage (n=261), 49.3% (34/69) received neoadjuvant therapy between 2018 and 2021, an increase from 4.3% (4/94) between 2011 and 2013 and 23.5% (23/98) between 2014 and 2017
- For patients with high-risk HER2+ eBC defined by pathologic and clinical stage, there was an increase in use of both neoadjuvant and (post-neo)adjuvant therapies over time; an increase in use of neoadjuvant therapy was also observed in patients with non-high-risk HER2+ eBC (**Figure 2**)

Figure 2. Use of neoadjuvant and (post-neo)adjuvant therapies in patients with high-risk and non-high-risk HER2+ eBC

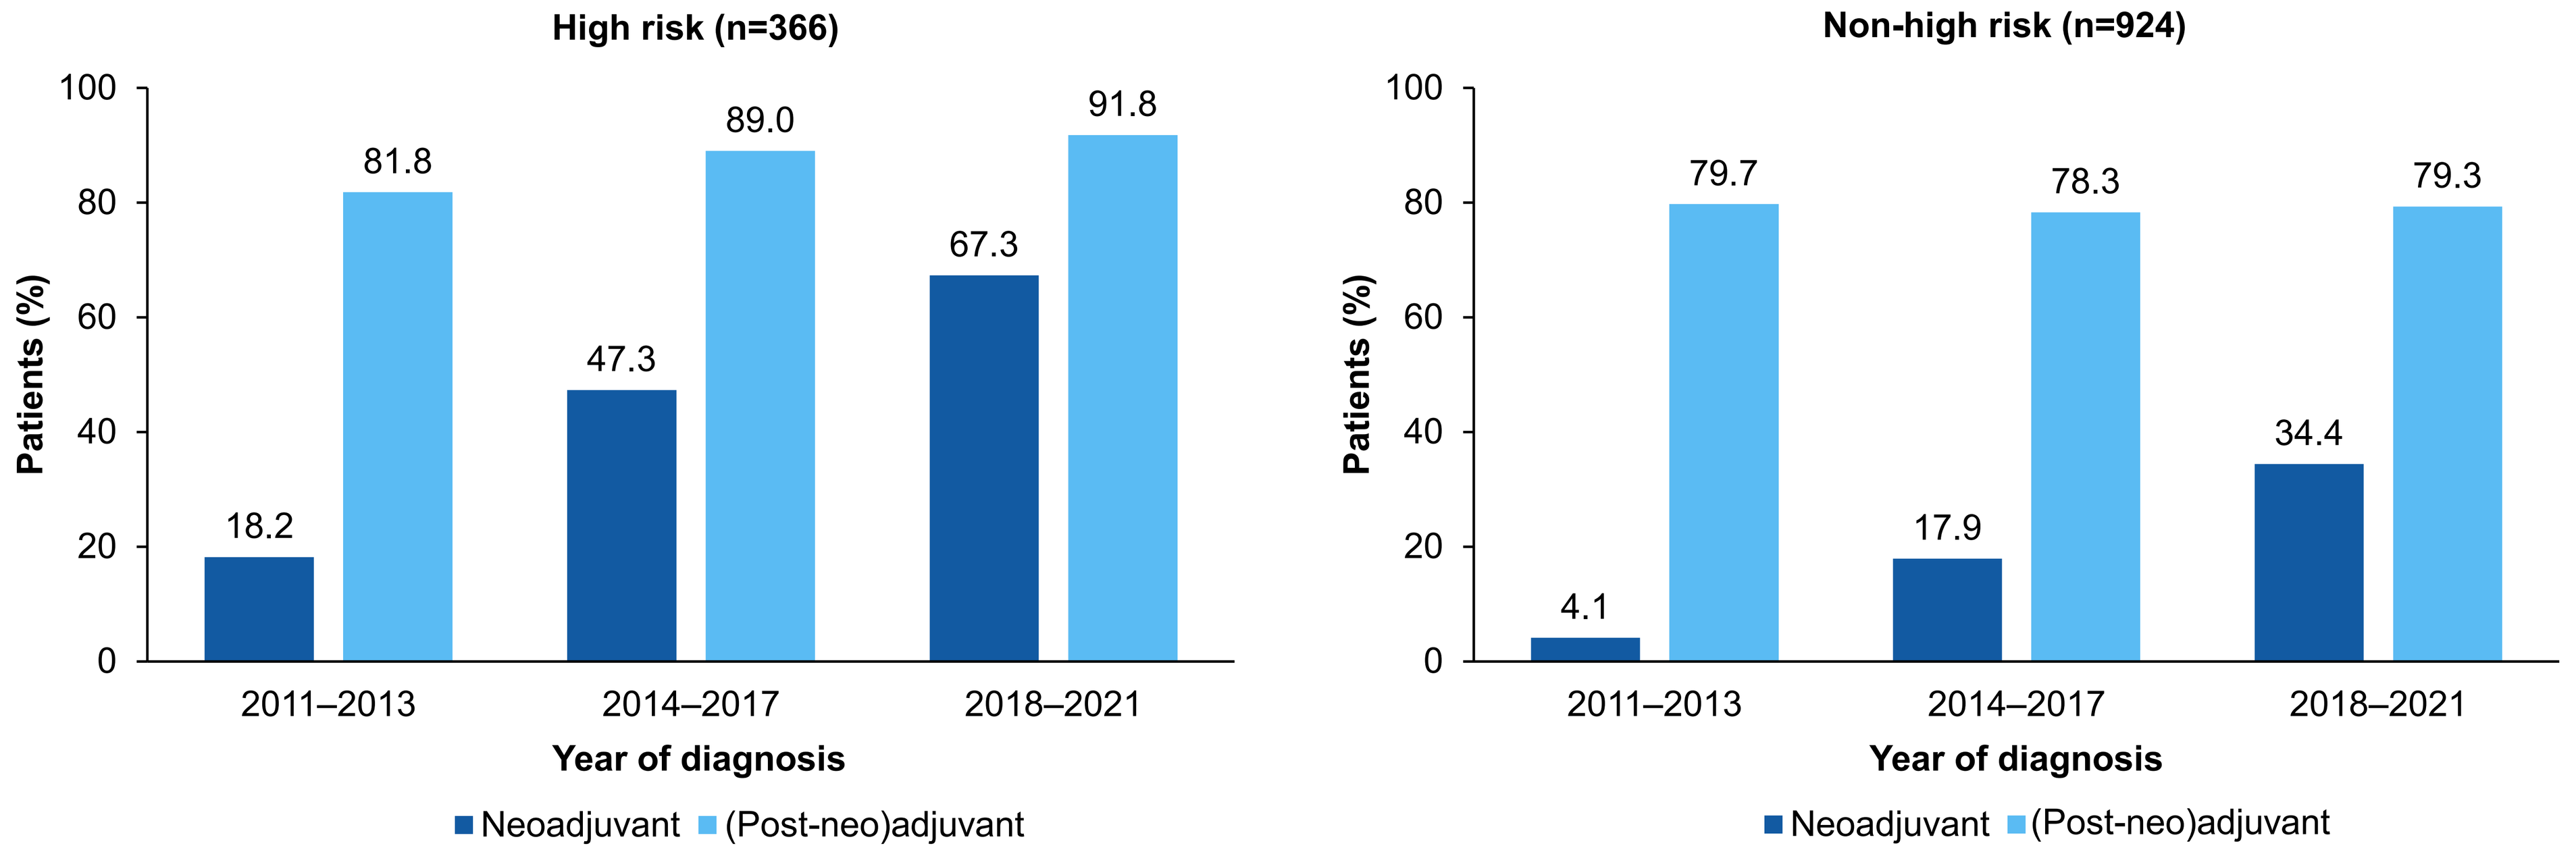
High risk (n=366)
Non-high risk (n=924)
Patients (%)
18.2
81.8
47.3
89.0
67.3
91.8
4.1
79.7
17.9
78.3
34.4
79.3
2011–2013
2014–2017
2018–2021
Year of diagnosis
Neoadjuvant
(Post-neo)adjuvant

eBC, early breast cancer; HER2+, human epidermal growth factor receptor 2–positive

- The 5-year invasive disease-free survival probability was 72.3% (95% confidence interval [CI] 66.8, 77.1) for patients with high-risk HER2+ eBC and 80.7% (95% CI 77.6, 83.5) for patients with non-high-risk HER2+ eBC (**Figure 3**)

Figure 3. Invasive disease-free survival* in patients with high-risk and non-high-risk HER2+ eBC

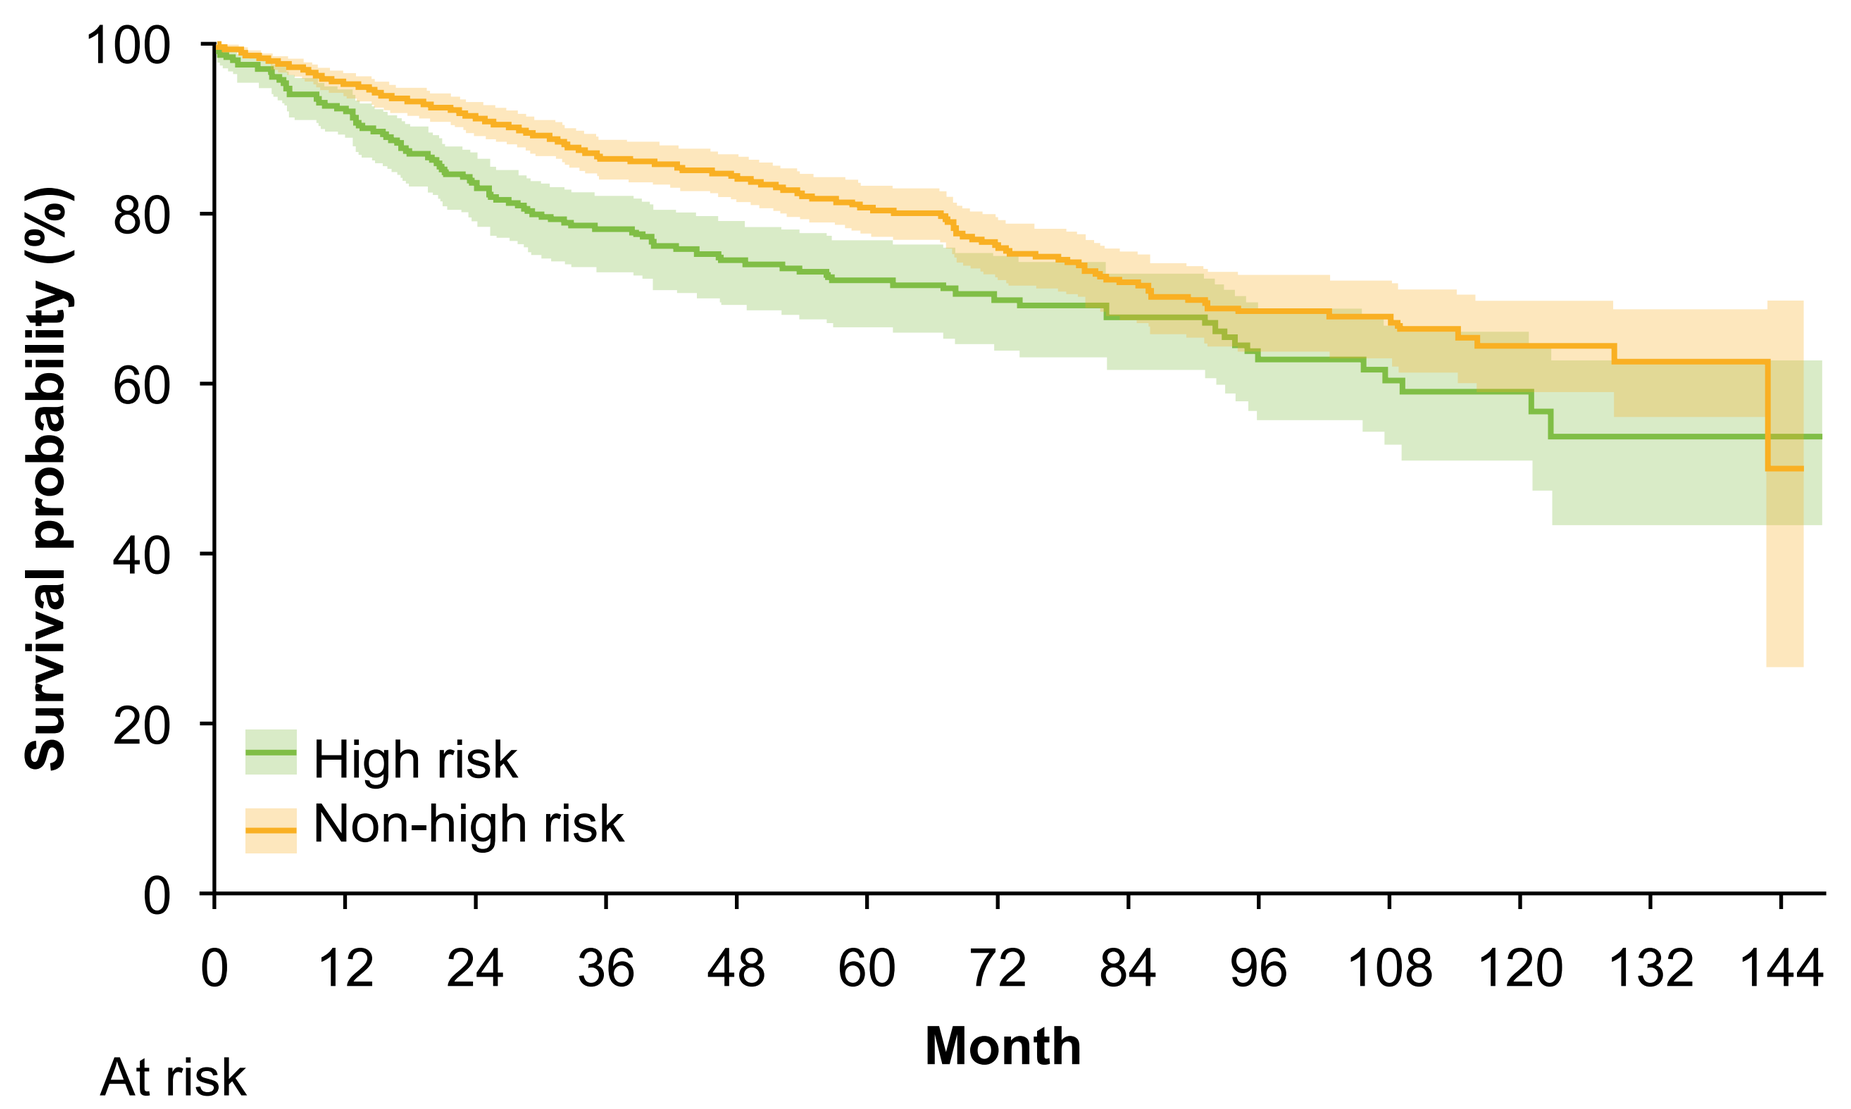
Survival probability (%)
High risk
Non-high risk
Month

At risk

	0	12	24	36	48	60	72	84	96	108	120	132	144
High risk	366	318	254	207	172	140	117	91	71	47	25	9	1
Non-high risk	924	807	687	576	491	364	250	192	133	95	60	22	1

*Defined as time from date of initial diagnosis (if no surgery) or date of definitive breast cancer surgery until the earliest date of invasive locoregional recurrence, invasive distant recurrence, second primary malignancy, or death. If no event of interest was observed, patients were censored at the date of last contact or study end date, whichever occurred first
eBC, early breast cancer; HER2+, human epidermal growth factor receptor 2–positive

- The 5-year overall survival probability was 86.9% (95% CI 82.3, 90.4) for patients with high-risk HER2+ eBC and 91.8% (95% CI 89.4, 93.7) for patients with non-high-risk HER2+ eBC (**Figure 4**)

Figure 4. Overall survival in patients with high-risk and non-high-risk HER2+ eBC*

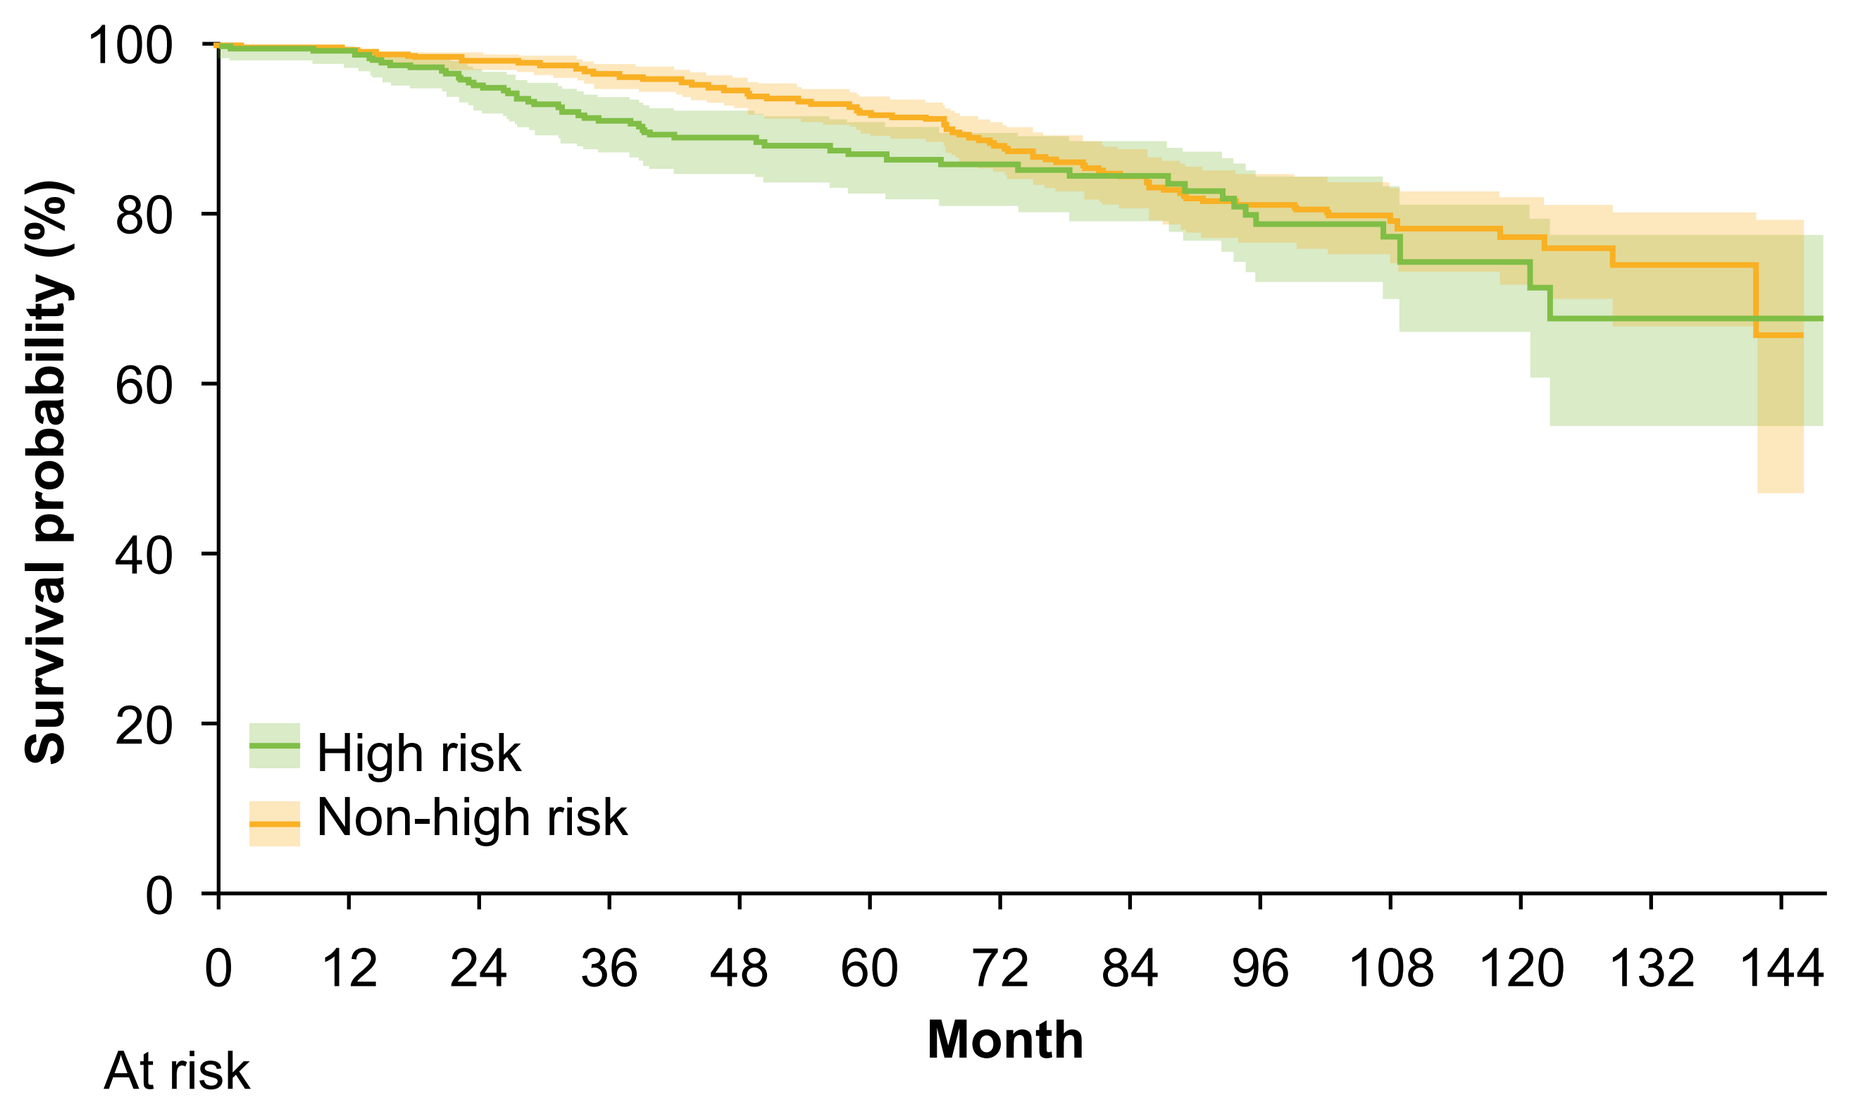
Survival probability (%)
High risk
Non-high risk
Month

At risk

	0	12	24	36	48	60	72	84	96	108	120	132	144
High risk	366	340	289	239	201	158	130	101	77	53	25	9	1
Non-high risk	924	840	739	637	544	413	289	226	159	109	68	26	2

*Data have not been adjusted for potential confounders
eBC, early breast cancer; HER2+, human epidermal growth factor receptor 2–positive

Acknowledgments

Under the guidance of the authors and in accordance with Good Publication Practice, medical writing support was provided by Hope Price, MSc, of Helios Medical Communications, part of Helios Global Group, Cheshire, UK, and was funded by AstraZeneca.

Disclosures

Reshma Mahtani reports research funding from Gilead Sciences; consulting fees from Agendia, Daiichi Sankyo / AstraZeneca, Eisai, Genentech / Roche, Gilead Sciences, Hologic / Biotheranostics, Lilly, Merck, Novartis, Pfizer, Puma Biotechnology, Sanofi, Sermonix Pharmaceuticals, Stemline Therapeutics; and meeting/travel support from AstraZeneca, Biotheranostics, Daiichi Sankyo / Lilly, Eisai, Genentech/Roche, Novartis, Pfizer, Puma Biotechnology, Sanofi, and Seagen.

References

1. Korde LA, et al. *J Clin Oncol.* 2021;39:1485–1505
2. International Agency for Research on Cancer. Breast cancer. Available from: https://www.iarc.who.int/cancer-type/breast-cancer (Accessed October 10, 2024)
3. Morales S, et al. *Cancers (Basel)*. 2021;13:5771
4. Wolff AC, et al. *J Clin Oncol.* 2013;31:3997–4013
5. Patel A, et al. *Cancers (Basel)*. 2020;12:2081
6. American Cancer Society. Breast cancer facts & figures 2022–2024. Available from: https://www.cancer.org/content/dam/cancer-org/research/cancer-facts-and-statistics/breast-cancer-facts-and-figures/2022-2024-breast-cancer-fact-figures-acs.pdf (Accessed October 11, 2024)
7. Harbeck N. *Breast*. 2022;62(Suppl. 1):S12–S16
8. Perez EA, et al. *J Clin Oncol.* 2014;32:3744–3752
9. Cameron D, et al. *Lancet*. 2017;389:1195–1205
10. Birnbaum B, et al. Model-assisted cohort selection with bias analysis for generating large-scale cohorts from the EHR for oncology research. January 13, 2020. Available from: http://arxiv.org/abs/2001.09765 (Accessed October 10, 2024)
11. Ma X, et al. Comparison of population characteristics in real-world clinical oncology databases in the US: Flatiron Health, SEER, and NPCR. June 7, 2023. Available from: https://www.medrxiv.org/content/10.1101/2020.03.16.20037143v3 (Accessed October 10, 2024)