ICARUS-BREAST02: A phase Ib/II, Multicenter, Open-label, Modular Study to Explore the Safety, Tolerability, and Activity of Patritumab Deruxtecan (HER3- DXd) Monotherapy and Combinations in Patients with Advanced Breast Cancer (ABC) after Progression on T-DXd

Barbara Pistilli[1,2], Stefan Michiels[3], Rasha Cheikh-Hussin[1,2], Magali Lacroix-Triki[4], Pierre Guyader[3], Lambros Tselikas[5], Souad Cosse[6], Tiffany Malleck[1], Jacqueline Deneuve[1], Ghada Nachabeh[6], Françoise Farace[7,8], Marianne Oulhen[7,8], Patricia Kannouche[9], Andrea Sporchia[10], Esther Pang[10], David W. Sternberg[10], Dalila Sellami[10], Noemie Corcos[2], Fabrice André[1,7,11], Fernanda Mosele[1,7], Guillaume Montagnac[2]

(1) Department of Medical Oncology, Gustave Roussy, Villejuif, France, (2) INSERM U1279, Gustave Roussy, Villejuif, France, (3) Department of Biostatistics and Epidemiology, Gustave Roussy, Oncostat INSERM U1018, (4) Department of Pathology, Gustave Roussy, Villejuif, France, (5) Surgery and Interventional Department, Gustave Roussy, Villejuif, France, (6) Projects and Promotion Division, Gustave Roussy, Villejuif, France, (7) INSERM U981, Gustave Roussy, Villejuif, France, (8) Gustave Roussy, University Paris-Saclay, "Rare Circulating Cells" Translational Platform, 32 CNRS UMS3655 – INSERM US23 AMMICA, F-94805, Villejuif, France, (9) UMR9019-CNRS, University Paris-Saclay, Gustave Roussy, 94805 Villejuif France, (10) Daiichi Sankyo Inc, NJ, USA, (11) CentraleSupélec, Inria, University Paris-Saclay, Gif-Sur-Yvette, France

PLAIN LANGUAGE SUMMARY

Why is this study being conducted ?

HER3 is a protein overexpressed in 50%-70% of breast cancers, playing a significant role in cancer growth, spread, and treatment resistance. Patritumab Deruxtecan (HER3-DXd) is a new antibody-drug conjugate (ADC) combining a HER3-directed antibody with a chemotherapy agent. It has shown promising effects in patients with ABC particularly those who have already been treated with multiple therapies. Since resistance to HER2-directed therapies can be sometimes mediated by HER3, our study aims to evaluate whether HER3-DXd works and is tolerated in patients whose cancers have become resistant to Trastuzumab Deruxtecan (T-DXd).

How is this study being conducted and who are the patients that can participate ?

The ICARUS-BREAST02 study is designed to test HER3-DXd as a single drug or in combination with other cancer treatments in patients with advanced HER2-positive or HER2-low breast cancer who have already been treated with T-DXd. The study currently has two parts:

- ***Module 0:*** *HER3-DXd alone in HER2-low ABC*
- ***Module 1:*** *HER3-DXd in combination with olaparib (a cancer drug shown to enhance HER3-DXd activity in lab studies) in HER2-positive ABC initially, and then both HER2-low and HER2-positive ABC in the later part.*

Patients will receive treatment until their cancer worsens or unacceptable side effects occur. HER3-DXd is given through an IV every three weeks, while olaparib is an oral treatment. This study started in May 2024, with the first patients receiving HER3-DXd alone to assess safety after prior T-DXd exposure, before adding olaparib in another part of the study.
The study main goal is to determine safety and efficacy of HER3-DXd as single agent or in combination after progression on T-DXd. To better understand how HER3-DXd works, blood and tumor samples will be collected at the beginning, during, and after stopping the treatment. Additional treatment combinations may be tested in the future.

STUDY RATIONALE

- HER3, part of the ErbB family, is overexpressed in 50%-70% of breast cancers and contributes to cancer progression and therapy resistance[1-10].
- HER3-DXd is an antibody-drug conjugate containing a human anti- HER3 immunoglobulin G1 monoclonal antibody conjugated via a cleavable peptide linker to an exatecan derivative (topoisomerase-I inhibitor) payload, with a drug to antibody ratio of 8:1[11-14].
- HER3-DXd has shown promising activity in patients with heavily pretreated ABC, across different subtypes and a wide range of HER3 expression[15].
- Recently, in ICARUS-BREAST01 phase-2 study, we observed a clinically meaningful activity and manageable safety profile of HER3-DXd in patients with HR+/HER2- ABC progressing after 2 or more lines of therapy, including CDK4/6inh: ORR 53.5% [95%CI, 43.2; 63.6]; mDoR 8.7 months [8.1; 12.5]; mPFS 9.4 months [95%CI 8.1; 13.4][16].
- Despite T-DXd's efficacy in HER2-positive and HER2-low ABC, strategies post-T-DXd remain undefined.
- **As HER3 expression has been associated with resistance to HER2-directed therapies and that topoisomerase-I inhibitors and PARP-inhibitors showed synergistic efficacy in preclinical models[17,18], ICARUS-BREAST02 study aims to assess the efficacy and safety of HER3-DXd, either as single agent or in combination with olaparib (or other agents) following progression on T-DXd.**

References

1. Papa F et al, Cancer Treat Rev 2024; 2.Jura N, et al, Proc Natl Acad Sci USA 2009; 3.Hynes NE et al, Nat Rev Cancer 2005; 4.Ocana et al, J Natl Cancer Inst 2013; 5.Chandarlapaty S et al, Cancer Cell, 2011; 6. Rodrik-Outmezguine VS et al, Cancer Discov 2011; 7.Chakrabarty A et al, Proc Natl Acad Sci U S A 2012; 8. Huang X et al, Cancer Res 2010; 9.Liu B et al, Int J Cancer 2007; 10. Morrison MM et al, Oncogene 2016; 11.Hashimoto Y, et al. Clin Cancer Res. 2019; 12. Nakada T, et al. Chem Pharm Bull (Tokyo). 2019; 13. OgitaniY, et al. Clin Cancer Res. 2016; 14. KoganemaruS, et al. Mol Cancer Ther. 2019; 15. Krop I et al, JCO 2023, 16. Pistilli B et al, ESMO 2024; 17. Boerner et al, PLoS One 2015; 18. Murai et al, J Pharmacol Exp Ther. 2014

STUDY DESIGN

ICARUS-BREAST02 (NCT06298084)

- **Phase Ib/II Multicenter, Open-label, Platform Study, currently composed of 2 modules:**
- **Module 0: HER3-DXd as single agent in patients with HER2-low ABC (cohort 2) in the safety run-in (part 1a) and dose-expansion part (part 2)**
- **Module 1: HER3-DXd in combination with olaparib in patients with HER2-positive ABC (cohort 1) in the dose-finding part (part 1b) and in patients with HER2-positive or HER2-low ABC (cohort 1 and cohort 2) in the dose-expansion part (part 2)**

KEY ELIGIBILITY CRITERIA

MAIN INCLUSION CRITERIA:

- ❑ All patients must have disease progression while on T-DXd or within 2 months from discontinuation
- ❑ Patients are included in **2 cohorts:**
 - **Cohort 1:** patients with HER2-positive (IHC 3+ or IHC2+/ISH positive), HR+/HR- ABC, must have received prior therapy with trastuzumab and taxanes
 - **Cohort 2:** patients with HER2-low (IHC 2+/ISH negative or IHC 1+), HR+/HR-ABC; patients with HR-positive ABC must have received ET and CDK4/6 inhibitors; may have received prior treatment with Sacituzumab Govitecan
- ❑ Female or male patient aged ≥18 years
- ❑ ECOG PS ≤1
- ❑ Patients must have a tumor site easily accessible to biopsy and must have accepted to perform pre-treatment, on-treatment, and end-of-treatment biopsies
- ❑ Patients must have at least one radiologically measurable lesion (different from the biopsy site) according to RECIST V1.1 criteria

MAIN EXCLUSION CRITERIA:

- ❑ Patients with any history of ILD (including pulmonary fibrosis or radiation pneumonitis), current ILD, or suspected to have ILD as assessed by imaging during screening
- ❑ Spinal cord compression or active brain metastases, patients with clinically inactive or treated brain metastases who are asymptomatic are eligible
- ❑ Patients with leptomeningeal disease
- ❑ Patients with clinically significant corneal disease

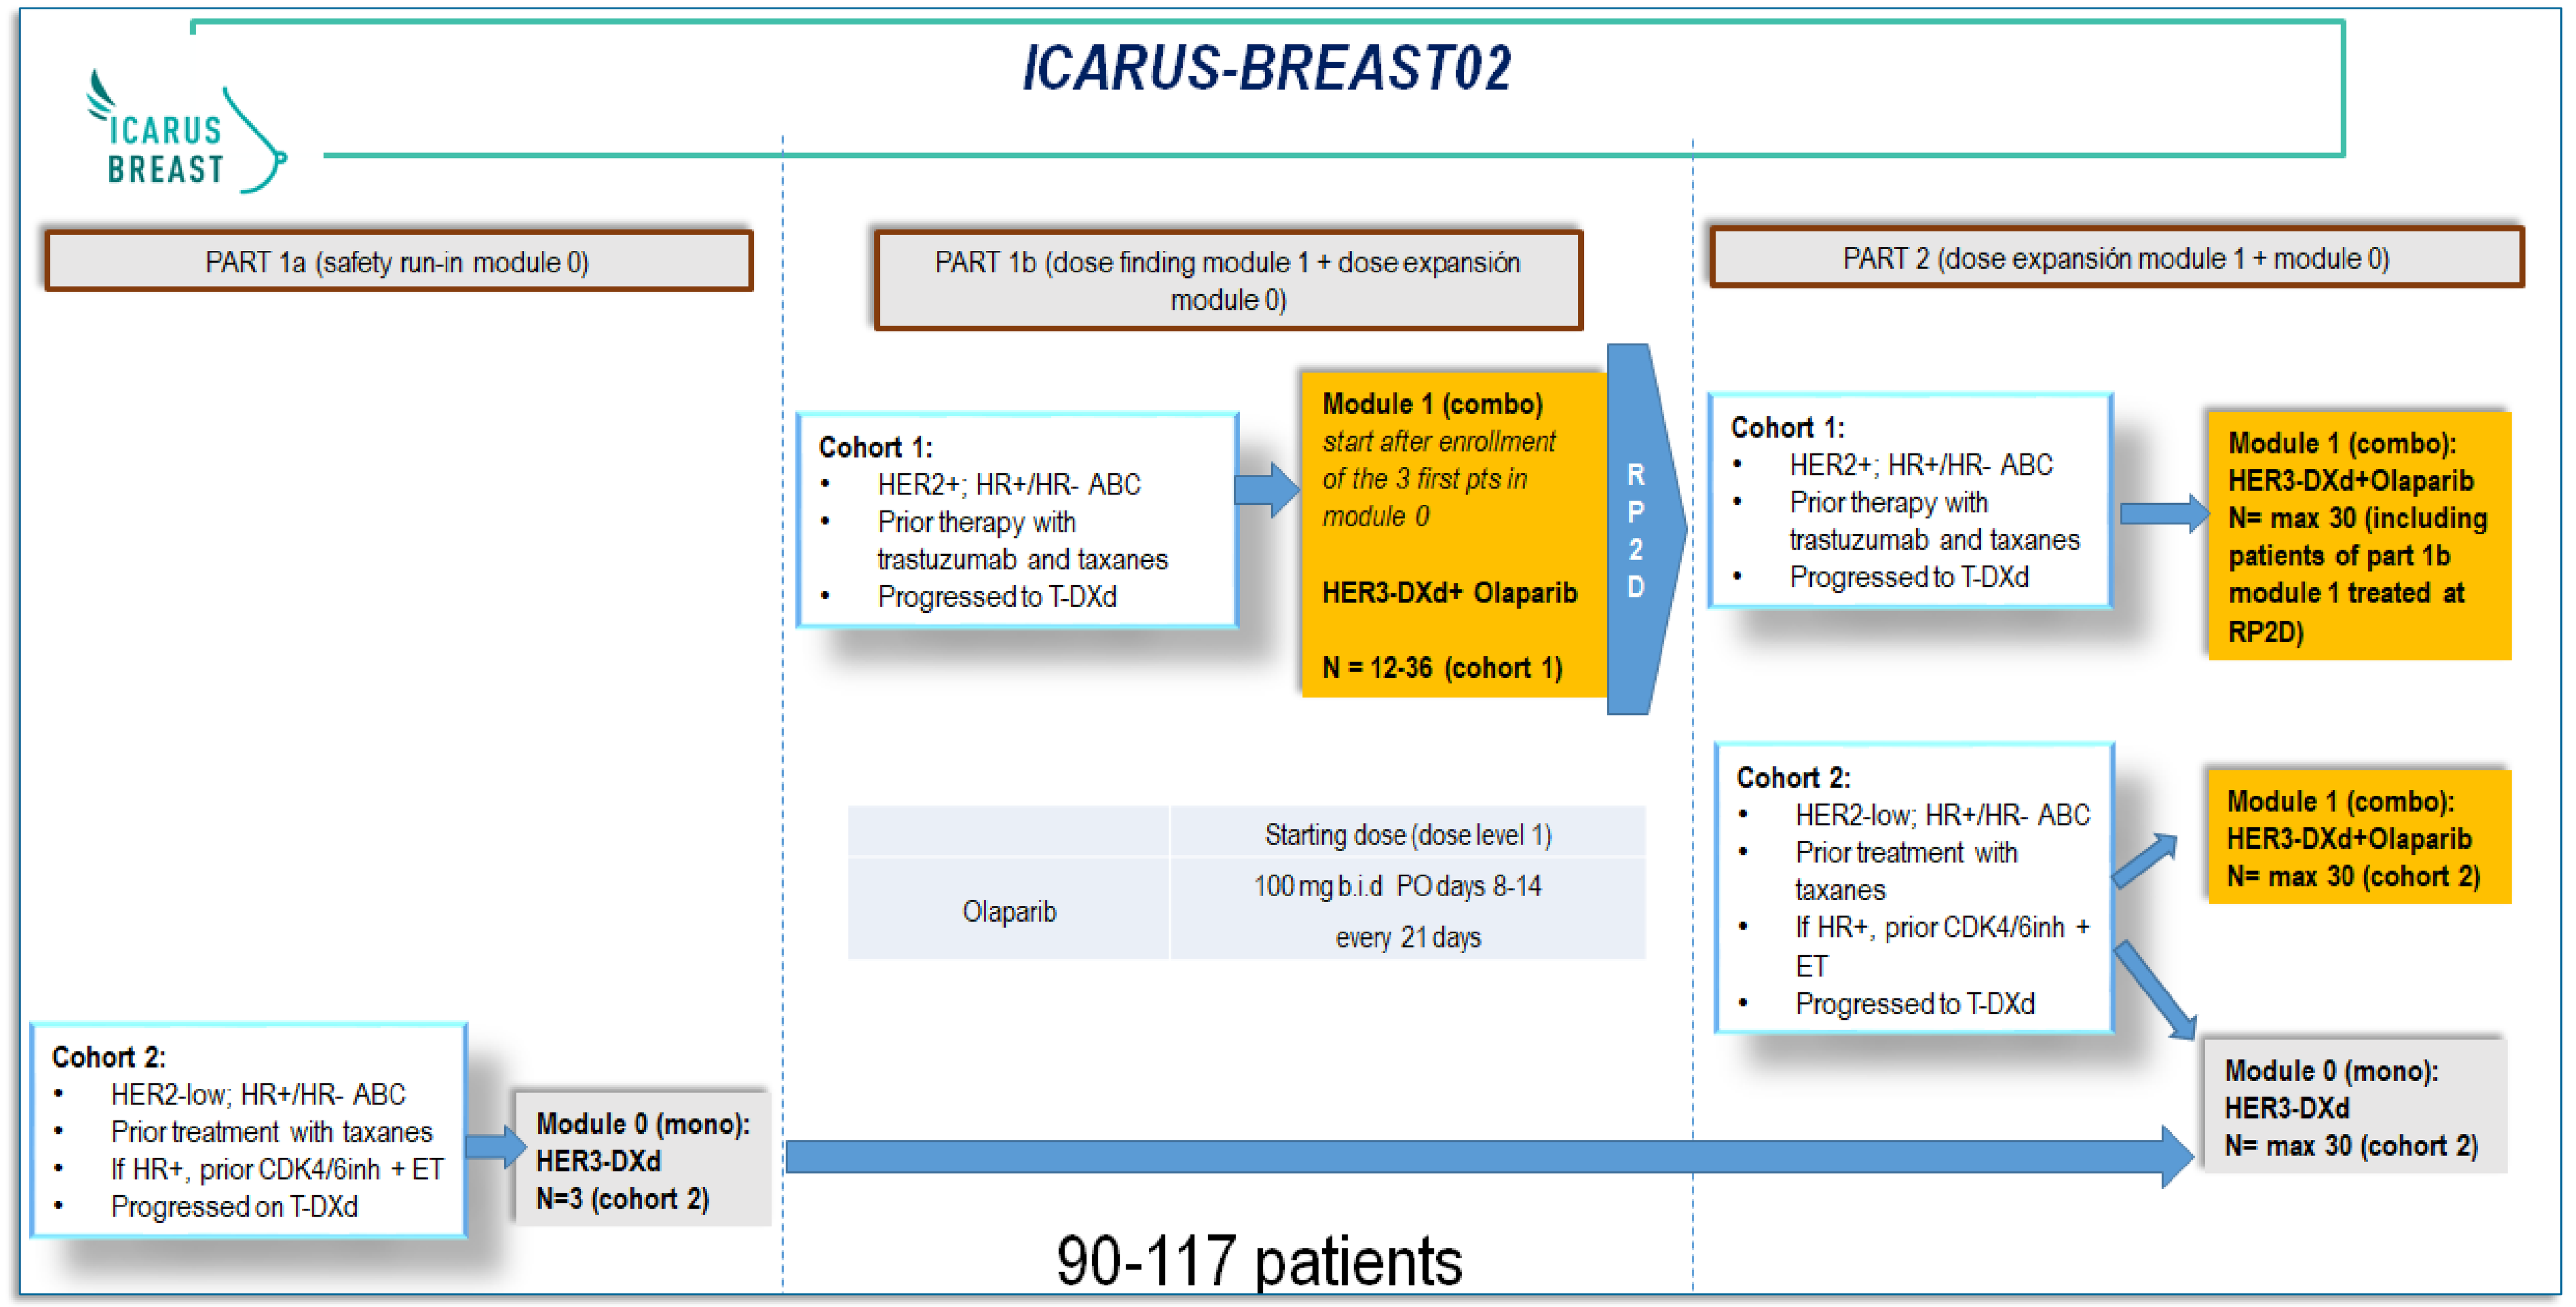
ICARUS BREAST
ICARUS-BREAST02
PART 1a (safety run-in module 0)
PART 1b (dose finding module 1 + dose expansión module 0)
PART 2 (dose expansión module 1 + module 0)
Cohort 1:
HER2+; HR+/HR- ABC
Prior therapy with trastuzumab and taxanes
Progressed to T-DXd
Module 1 (combo)
start after enrollment of the 3 first pts in module 0
HER3-DXd+ Olaparib
N = 12-36 (cohort 1)
RP2D
Cohort 1:
HER2+; HR+/HR- ABC
Prior therapy with trastuzumab and taxanes
Progressed to T-DXd
Module 1 (combo):
HER3-DXd+Olaparib
N= max 30 (including patients of part 1b module 1 treated at RP2D)
Olaparib
Starting dose (dose level 1)
100 mg b.i.d PO days 8-14
every 21 days
Cohort 2:
HER2-low; HR+/HR- ABC
Prior treatment with taxanes
If HR+, prior CDK4/6inh + ET
Progressed to T-DXd
Module 1 (combo):
HER3-DXd+Olaparib
N= max 30 (cohort 2)
Cohort 2:
HER2-low; HR+/HR- ABC
Prior treatment with taxanes
If HR+, prior CDK4/6inh + ET
Progressed on T-DXd
Module 0 (mono):
HER3-DXd
N=3 (cohort 2)
Module 0 (mono):
HER3-DXd
N= max 30 (cohort 2)
90-117 patients

TUMOR AND BLOOD SAMPLE COLLECTION: BASELINE → On-treatment (C1D3/C1D7 for tumor) → EOT

CURRENT STUDY STATUS

- Enrollment is terminated in the Safety Run-In part (part 1a) with no dose limiting toxicities (DLT) detected
- The study is currently enrolling in Dose-finding part (part 1b) for module 1 and Dose-expansion part (part 2) for module 0
- **New combination treatment modules could be added, via protocol amendment**

KEY STUDY ENDPOINTS

PRIMARY ENDPOINTS:

- **Part 1a and 1b:** Safety as measured by DLTs, frequency and severity of any AEs, TEAEs, SAEs, AESIs graded by NCI-CTCAE v5.0;
- **Part 2:** Anti-tumor activity of HER3-DXd single agent and combinations as measured by investigator assessed ORR per RECIST 1.1

SECONDARY ENDPOINTS:

- **Part 1a and 1b:** Anti-tumor activity of HER3-DXd single agent and combinations as measured by investigator assessed ORR per RECIST 1.1
- **Part 2:** Safety as measured by DLTs, frequency and severity of any AEs, TEAEs, SAEs, AESIs graded by NCI-CTCAE v5.0

EXPLORATORY ENDPOINTS:

- Mechanism of action of HER3-DXd monotherapy and in combination with olaparib or other agents
- DXd uptake and target antigens modulation
- Modulation of immune tumor microenvironment
- Genomic alterations and gene expression modulation associated with response/resistance

Acknowledgements

This study is sponsored by Gustave Roussy Cancer Center and supported by Daiichi-Sankyo

CONTACT: barbara.pistilli@gustaveroussy.fr