American Society of Clinical Oncology Quality Care Symposium (ASCO QCS), September 27–28, 2024

Abstract 398

Treatment patterns and clinical outcomes in HER2-low metastatic breast cancer, 2018–2023: a retrospective observational study

Erica L Mayer,[1] Simon M Collin,[2] Sam Hillman,[3]* Luis C Berrocal-Almanza,[2] Joseph Sparano,[4] Clara Lam[5]

[1]Dana-Farber Cancer Institute, Boston, MA, US; [2]Oncology Outcomes Research, Evidence Generation, Publications & Partnerships (EGP2), Oncology Business Unit Medical, AstraZeneca, Cambridge, UK; [3]Center of Oncology Data Excellence (CODE), Evidence Generation, Publications & Partnerships (EGP2), Oncology Business Unit Medical, AstraZeneca, Cambridge, UK; [4]Icahn School of Medicine at Mount Sinai, Tisch Cancer Institute, New York, NY, US; [5]US Medical Affairs, Oncology Business Unit, AstraZeneca, Gaithersburg, MD, US

*Has moved organization since publication initiated; current affiliation: Hillman Analytics

Objective

- To assess recent real-world treatment patterns and clinical outcomes in patients from the US with hormone receptor–positive (HR+), human epidermal growth factor receptor 2–low (HER2-low) metastatic breast cancer (mBC)

Conclusions

- Treatment of HR+, HER2-low mBC was heterogeneous from first line (1L) onwards, characterized by use of endocrine therapy (ET) with or without cyclin-dependent kinase 4/6 inhibitors (CDK4/6is), replaced in later lines by chemotherapy (CT)
- Despite receiving ET- and CT-based regimens, there is potential for improvement in clinical outcomes in patients with HR+, HER2-low mBC, highlighting the need for new treatment options in this patient population
- Future analyses of real-world progression-free survival (rwPFS) could focus on evaluating patients in key subgroups, such as early progressors, to understand differences in their treatment patterns and outcomes

Plain language summary

Why did we perform this research?

Some people with metastatic breast cancer (mBC) (cancer that has spread from its original site) have low levels of a protein called human epidermal growth factor receptor 2 (HER2); these tumors are classified as HER2-low. Often, mBC tumors also have proteins that can attach to hormones (hormone receptor–positive [HR+]), which stimulate the tumor to grow and spread.[1,2] The most common first (or first-line [1L]) treatments for people with HR+, HER2-low mBC are endocrine therapies (ETs) (also called hormone therapies, these are cancer treatments that add, remove, or block hormones); however, people who do not respond to ETs have been limited to receiving chemotherapy (CT).[3] We performed this study to understand how people with HR+, HER2-low mBC are treated in regular clinical practice (outside of controlled clinical trials), and how well the currently available treatments work.

How did we perform this research?

We used an existing database of anonymous medical records to collect information about the treatments and outcomes of people with HR+, HER2-low mBC from the US. We focused on people diagnosed in 2018 through 2022.

What were the findings of this research?

We saw that ET was the most common 1L treatment received. A greater variety of therapies was given to people who went on to receive further rounds of treatment, including treatment only with CT. Patient survival and the time until disease worsened (progression) became shorter with each line of therapy.

What are the implications of this research?

This study highlights the need for rapid adoption of new treatment options that can help to slow progression and improve survival in people with HR+, HER2-low mBC.

1. Schettini F, et al. *NPJ Breast Cancer*. 2021;7:1; 2. Viale G, et al. Poster presented at ASCO 2022 (Abstract 1087); 3. Roy AM, et al. *Cancer*. 2023;129:2773–2788

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please visit the link below:

Poster https://bit.ly/47dxL8a

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from ASCO® or the authors of this poster.

This study was sponsored by AstraZeneca. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201).
Poster presented at ASCO QCS 2024 by Aaron Kershner on behalf of the authors. Corresponding author: Erica L Mayer; email address: Erica_Mayer@dfci.harvard.edu

Introduction

- It is estimated that approximately 50% of patients with primary or mBC have HER2-low tumors (immunohistochemistry [IHC] 1+ or IHC 2+ / in situ hybridization−)[1]
- The prevalence of HER2-low tumors is reported to be higher among patients who have tumors that are HR+ (approximately 65%) compared with patients whose tumors are hormone receptor–negative (37% to 55%)[2,3]
- ET with or without targeted therapy, including CDK4/6is or inhibitors of the PIK3CA/AKT/mTOR pathway, is the early-line standard of care for patients with HR+, HER2-low tumors[4]
- Current treatment options for patients who progress after ET-based therapy include conventional CT followed by antibody-drug conjugates[4]

Methods

- This was a retrospective, observational study in patients diagnosed with de novo or recurrent HR+, HER2-low mBC between January 2018 and February 2023 who received at least 1 line of therapy (LOT) post mBC diagnosis (**Figure 1**)
- De-identified data were obtained from the US nationwide Flatiron Health electronic health record-derived database, comprising patient-level data originating from ~280 US cancer clinics (~800 sites of care; primarily community oncology settings), and curated via technology-enabled abstraction[5,6]
- Real-world overall survival (rwOS) and rwPFS after mBC diagnosis and from the start of each LOT were calculated using the Kaplan-Meier method (data censored at the last activity date during the study period)
 - rwOS: time from index date to date of death
 - rwPFS: time from index date to date of the earliest progression event or death due to any cause

Figure 1. Study flow

Inclusion criteria and patient flow

Potential patients in the database (N=35546)
↓
Patients who were HER2− with no prior evidence of ever being HER2+ (test/treatment) (N=11216)
↓
Patients aged ≥18 years who were HER2-low and HR+ with metastatic diagnosis date between January 2018 and February 2023 (N=3954)
↓
At least 1 line of treatment post mBC diagnosis (N=3511)
↓
No evidence of use of clinical study drugs (January 2018 to May 2023) (N=3405)
↓
No prior cancer (excluding non-melanoma skin cancer) (N=2692)

Eligible population (N=2692)

Patients were classified as HER2-low if the closest result to the date of mBC diagnosis was an IHC score of 1+ or IHC 2+ if they had a negative ISH test at any timepoint; patients were not included if they had ever had a positive HER2 test

Follow up

Patients were followed from index date to death, last known activity date, or end of the study period (September 30, 2023)

HER2, human epidermal growth factor receptor 2; HER2−, HER2-negative; HER2+, HER2-positive; HR+, hormone receptor–positive; IHC, immunohistochemistry; ISH, in situ hybridization; mBC, metastatic breast cancer

Results and interpretation

- A total of 2692 patients with HR+, HER2-low mBC were eligible for the study. Median follow up from mBC diagnosis was 38.3 months (interquartile range [IQR] 24.4, 53.4)
- Patient demographics and clinical characteristics are shown in **Table 1**

Table 1. Patient demographic and clinical characteristics*

HR+, HER2-low mBC (N=2692)	
Median age at mBC diagnosis, years (IQR)	65 (56, 73)
Female sex, n (%)	2636 (97.9)
De novo mBC, n (%)†	1106 (41.1)
Recurrent mBC, n (%)†	1336 (49.6)
Disease stage at initial mBC diagnosis, n (%)	
I/II/III	1336 (49.6)
IV	1106 (41.1)
Unknown	250 (9.3)

*Demographic and clinical characteristics were captured from data entered prior to and at the index date (date of mBC diagnosis); †status was unknown for 250 patients (9.3%)
HER2, human epidermal growth factor receptor 2; HR+, hormone receptor–positive; IQR, interquartile range; mBC, metastatic breast cancer

- Overall, the median number of LOTs was 2 (IQR 1, 3) for patients who received treatment in the metastatic setting and up to the end of the study
- A total of 1484 (55.1%) patients received second-line (2L), 778 (28.9%) third-line (3L), 408 (15.2%) fourth-line (4L), and 195 (7.2%) fifth-line (5L) therapy (**Figure 2**)
 - Serial ET cycles accounted for 65.7% of 2L, 45.2% of 3L, 27.9% of 4L, and 18.5% of 5L therapy
 - Most patients received ET-based therapy in 1L; of those patients who went on to receive later lines of therapy, the proportion receiving ET decreased with each line compared with other treatments, eg single-agent CT

Figure 2. Treatment patterns over LOT

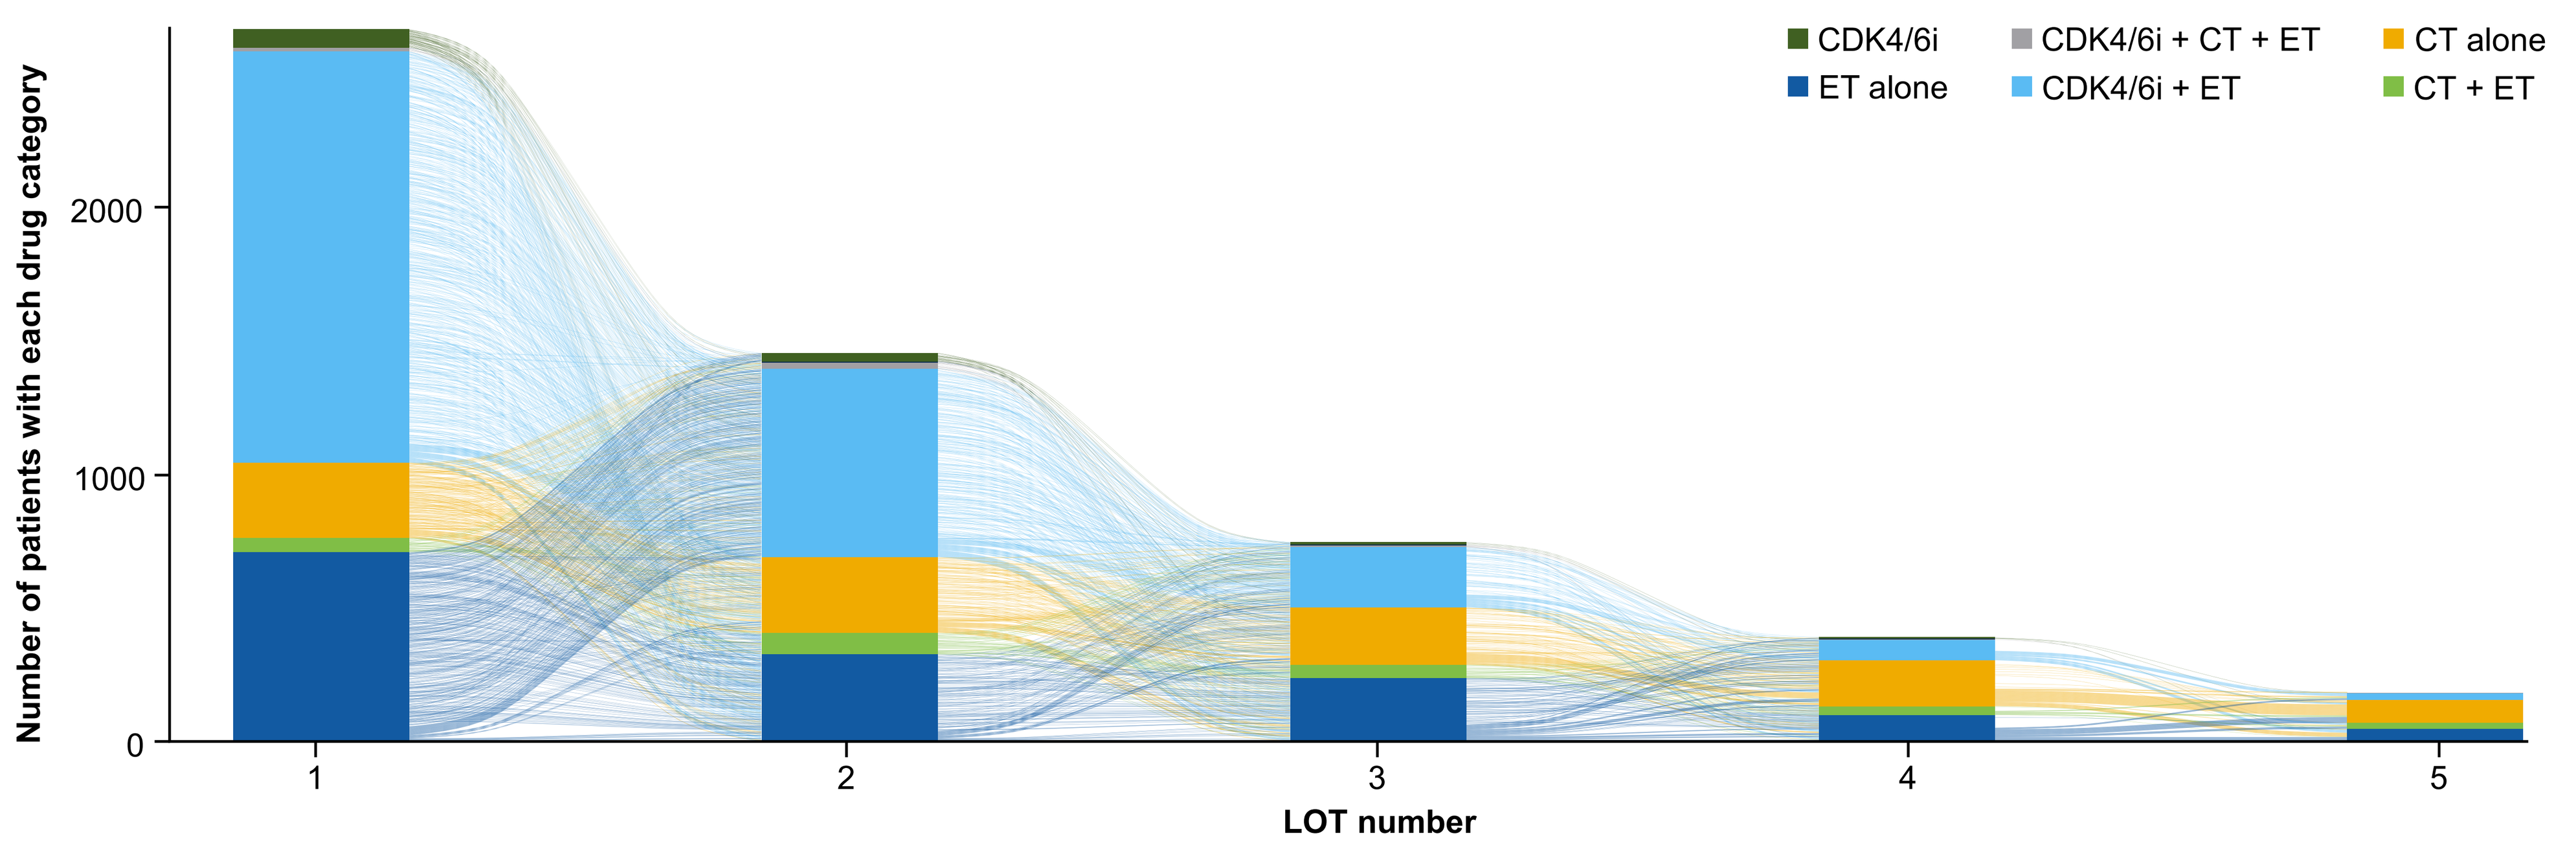
CDK4/6i
ET alone
CDK4/6i + CT + ET
CDK4/6i + ET
CT alone
CT + ET
Number of patients with each drug category
0
1000
2000
1
2
3
4
5
LOT number

	1L (N=2692)	2L (n=1484)	3L (n=778)	4L (n=408)	5L (n=195)
Cumulative frequency of patients who received ET in this and previous LOT,* n (%)†	2315 (86.0)	975 (36.2)	352 (13.1)	114 (4.2)	36 (1.3)
Cumulative frequency of patients who had CT in this LOT,* n (%)†	350 (13.0)	140 (5.2)	26 (1.0)	4 (0.1)	1 (<0.1)
Patients who had not received any further treatment at each LOT, n (%)‡	NA	1208 (44.9)	706 (47.6)	370 (47.6)	213 (52.2)

*Only includes patients who received ET/CT in all previous lines of therapy (other drugs given in the LOT are ignored); †as cumulative data are given, denominator is 2692. Categories are not mutually exclusive (some patients may have received CT combined with ET, either CT or ET monotherapy, or CT/ET combined with other classes of drugs; ‡including patients who did not go on to the next LOT because they were still on a previous LOT at end of study: 1L 139/2692 (5.2%), 2L 80/1484 (5.4%), 3L 33/778 (4.2%), 4L 19/408 (4.7%), 5L 12/195 (6.2%)
1L, first line; 2L, second line; 3L, third line; 4L, fourth line; 5L, fifth line; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; CT, chemotherapy; ET, endocrine therapy; LOT, line of therapy; NA, not applicable

- In 1L, 84.7% of patients received an ET-based therapy, and only 10.0% received CT monotherapy; by 4/5L, use of single-agent CT had increased to approximately 40%, a similar level to that of ET-based therapy in those lines (**Table 2**)
- Of the 2692 patients in the study population, 2133 (79.2%) received CDK4/6i in any LOT and 509 (18.9%) received CDK4/6i in at least 2 LOTs

Table 2. Top five drug regimens by LOT

	1L (N=2692)	2L (n=1484)	3L (n=778)	4L (n=408)	5L (n=195)
ET + CDK4/6i, n (%)	1537 (57.1)	694 (46.8)	217 (27.9)	76 (18.6)	25 (12.8)
Endocrine monotherapy, n (%)	684 (25.4)	202 (13.6)	126 (16.2)	52 (12.8)	21 (10.8)
ET + mTORi, n (%)	9 (0.3)*	61 (4.1)	55 (7.1)	25 (6.1)	17 (8.7)
ET + CT, n (%)	50 (1.9)	74 (5.0)	42 (5.4)	29 (7.1)	22 (11.3)
CT alone, n (%)	269 (10.0)	270 (18.2)	210 (27.0)	161 (39.5)	82 (42.1)
CDK4/6i alone, n (%)	69 (2.6)	29 (2.0)*	8 (1.0)*	3 (0.7)*	3 (1.5)*

Drug regimens are mutually exclusive. *Not a top five drug category in the specified LOT
1L, first line; 2L, second line; 3L, third line; 4L, fourth line; 5L, fifth line; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; CT, chemotherapy; ET, endocrine therapy; LOT, line of therapy; mTORi, mammalian target of rapamycin inhibitor

- Overall median rwOS was 42.4 months, decreasing from 39.9 months in 1L to 8.9 months in 5L (**Figure 3**)
- Overall median rwPFS was 17.1 months, decreasing from 16.3 months in 1L to 3.9 months in 5L (**Figure 4**)

Figure 3. rwOS from mBC diagnosis

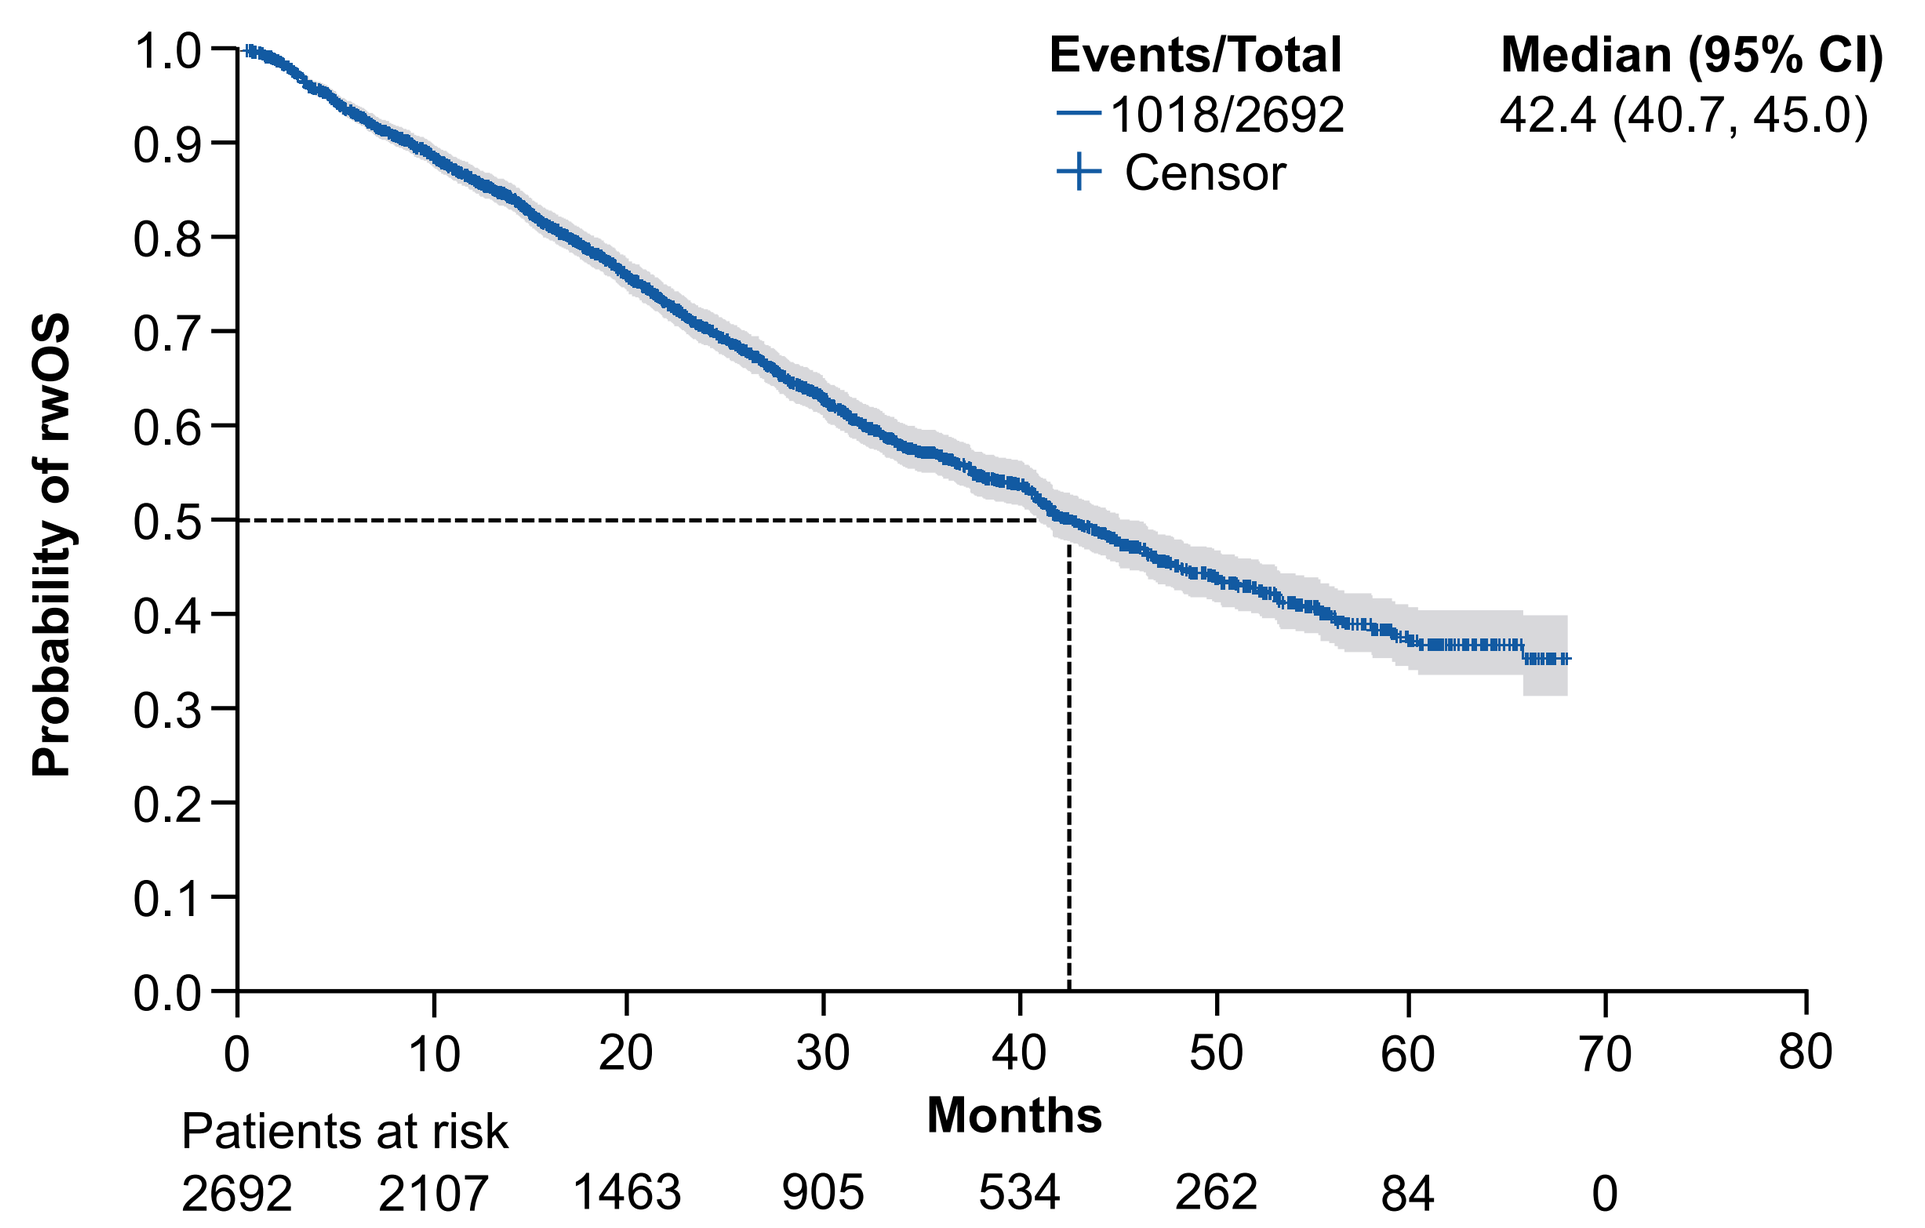
Events/Total
1018/2692
Censor
Median (95% CI)
42.4 (40.7, 45.0)
Probability of rwOS
Months
Patients at risk
2692 2107 1463 905 534 262 84 0

Median rwOS by LOT, months (95% CI)	
1L (N=2692)	39.9 (37.1, 42.3)
2L (n=1484)	26.1 (23.5, 28.4)
3L (n=778)	16.8 (14.9, 19.2)
4L (n=408)	11.8 (10.3, 14.5)
5L (n=195)	8.9 (7.4, 12.3)

1L, first line; 2L, second line; 3L, third line; 4L, fourth line; 5L, fifth line; CI, confidence interval; LOT, line of therapy; rwOS, real-world overall survival

Figure 4. rwPFS from mBC diagnosis

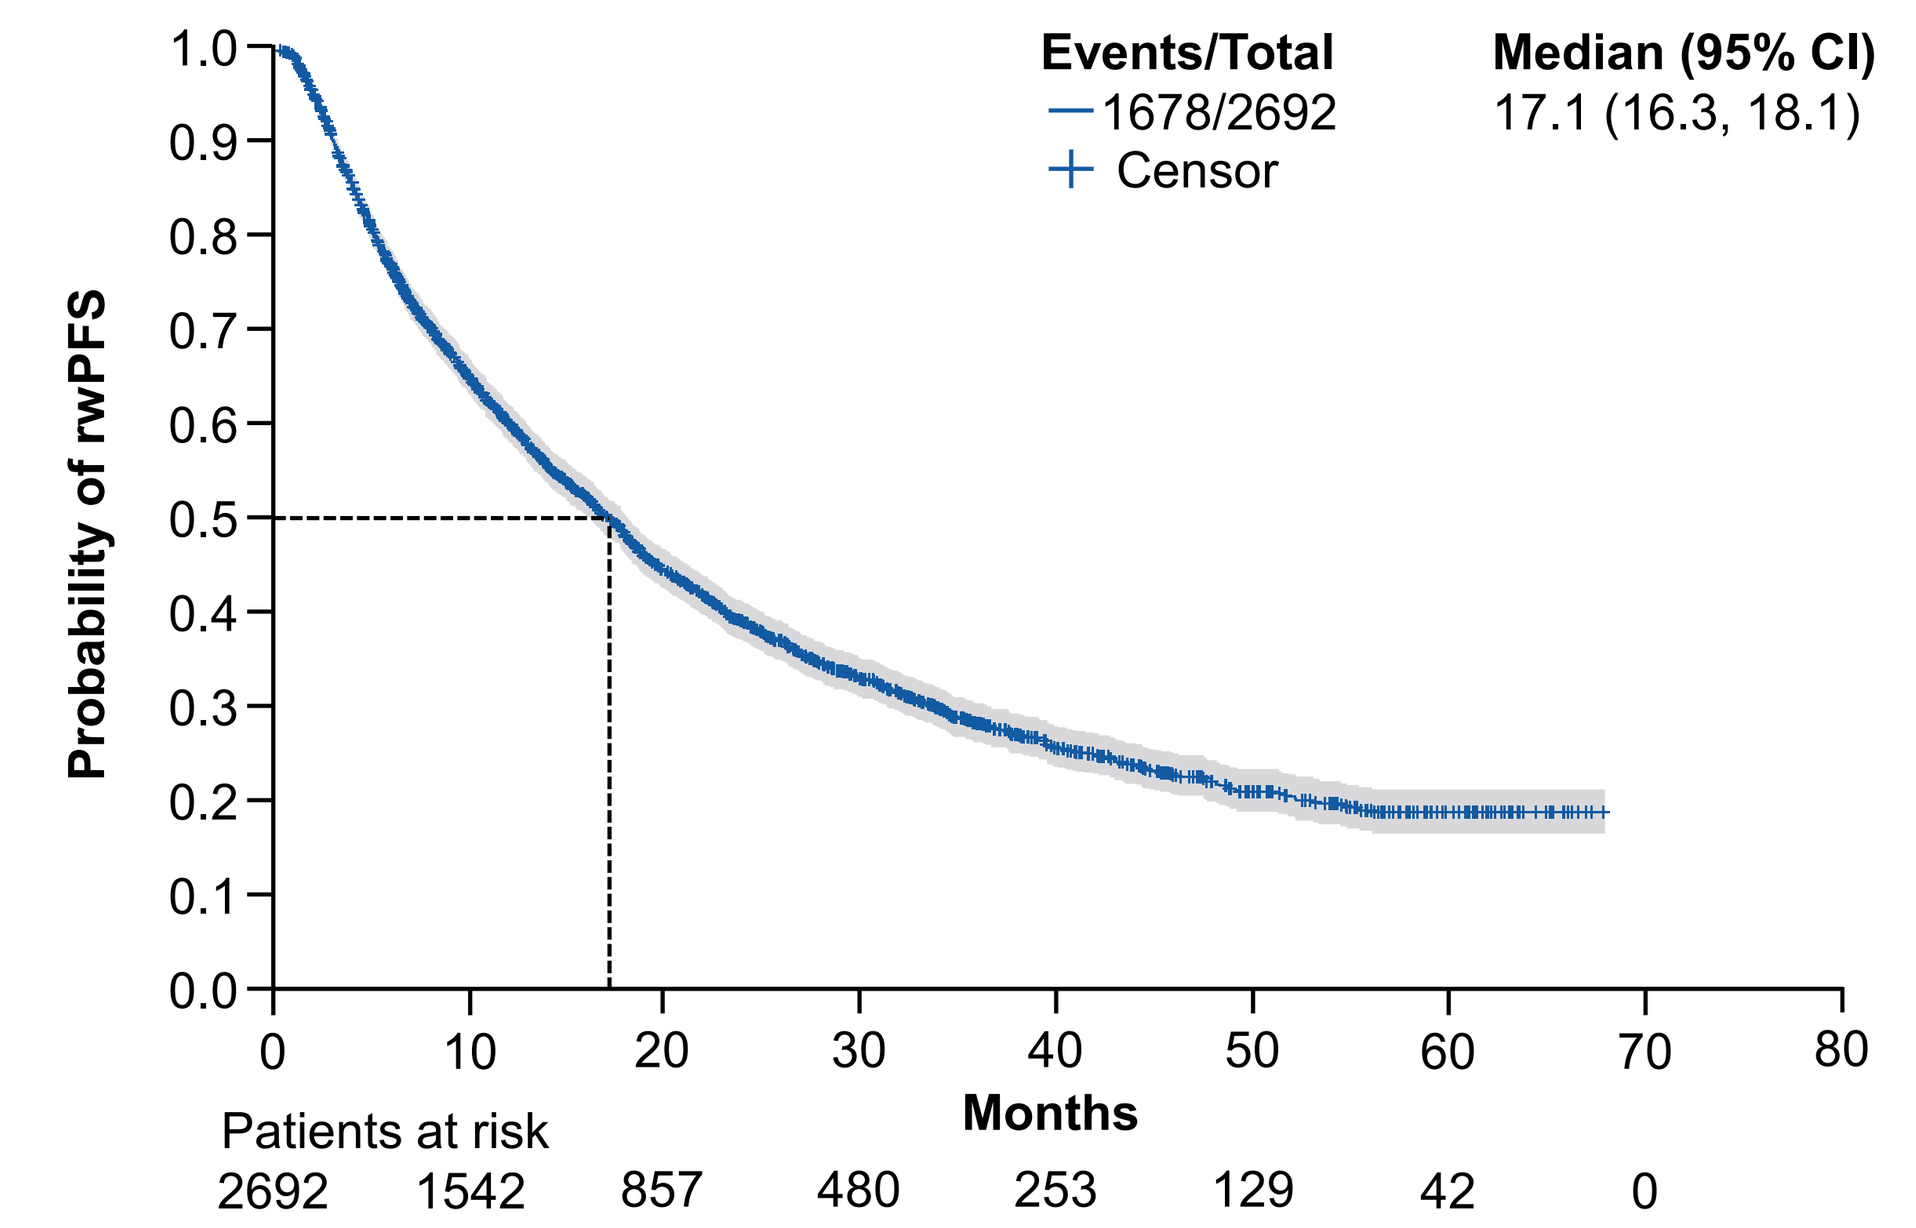
Events/Total
1678/2692
Censor
Median (95% CI)
17.1 (16.3, 18.1)
Probability of rwPFS
Months
Patients at risk
2692 1542 857 480 253 129 42 0

Median rwPFS by LOT, months (95% CI)	
1L (N=2692)	16.3 (15.2, 17.3)
2L (n=1484)	9.1 (8.2, 10.0)
3L (n=778)	6.2 (5.6, 7.2)
4L (n=408)	5.1 (4.7, 6.2)
5L (n=195)	3.9 (3.3, 5.0)

1L, first line; 2L, second line; 3L, third line; 4L, fourth line; 5L, fifth line; CI, confidence interval; LOT, line of therapy; rwPFS, real-world progression-free survival

- **Study limitations:** The study period captured only a small number of patients who received HER2-directed therapy approved recently for HER2-low mBC. Additionally, the database does not capture reasons for treatment discontinuation in a LOT other than progression or death

Acknowledgments

Under guidance of the authors and in accordance with Good Publication Practice, medical writing support was provided by Carmen Grimaldos, PhD, Georgina Stooke-Vaughan, PhD, and Stephen Purver, MChem, of Helios Medical Communications, part of Helios Global Group, and was funded by AstraZeneca.

Disclosures

Erica L Mayer reports research consulting or advisory role for Lilly, Novartis, and AstraZeneca.

References

1. Prat A, et al. *JAMA Oncol*. 2022;8:1676–1687
2. Schettini F, et al. *NPJ Breast Cancer*. 2021;7:1
3. Viale G, et al. Poster presented at ASCO 2022 (Abstract 1087)
4. Roy AM, et al. *Cancer*. 2023;129:2773–2788
5. Birnbaum B, et al. aiXiv 2020. Preprint published online January 13, 2020. Available from: https://arxiv.org/abs/2001.09765
6. Ma X, et al. MedRxiv 2023. Preprint published online June 7, 2023. Available from: https://www.medrxiv.org/content/10.1101/2020.03.16.20037143v3