Pharmacokinetics and exposure–safety analyses across tumor types for ifinatamab deruxtecan (I-DXd) in the first-in-human IDeate-PanTumor01 study

P042

Brittany P. Tran[1], Soniya S. Vaidya[1], Narasimha M. Midde[1], Naoko Okamoto[1], Jasmeet Singh[1], Meng Qian[1], Naoto Yoshizuka[1], Pengcheng Lu[2], Yvonne Lau[1]

[1]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [2]Sarah Cannon Research Institute, Nashville, TN, USA

Copies of this poster obtained through this QR code and/or https://bit.ly/DSACCP2024 are for personal use only and may not be reproduced without permission of the authors.

PURPOSE

- To characterize the pharmacokinetics of I-DXd and DXd in patients with SCLC, ESCC, mCRPC and sqNSCLC based on the dose-escalation and dose-expansion parts of the IDeate-PanTumor01 study (NCT04145622)
- To perform tumor-agnostic ES analyses to support dose-selection considerations in tumor types beyond SCLC for I-DXd

CONCLUSIONS

- Of the 17 safety endpoints evaluated in ES analyses, 7 occurred with a frequency of >10% and had a statistically significant relationship with exposure: any-grade GI TEAE; Grade ≥3 TEAE; dose reduction associated with an AE; any treatment-emergent serious AE; anemia (any grade and Grade ≥3); and any AE leading to dose interruption
- Along with previously published safety and efficacy data from the IDeate-PanTumor01 study,[1] the PK and exploratory ES analyses suggest comparable PK and ES relationships at the 12 mg/kg Q3W dose of I-DXd across SCLC, ESCC, mCRPC and sqNSCLC in IDeate-PanTumor01
- The favorable benefit–risk profile across tumor types supports the continued development of I-DXd in SCLC and in other tumor types

INTRODUCTION

- I-DXd is a B7-H3–directed ADC comprising an anti–B7-H3 IgG1 mAb linked to a potent topoisomerase I inhibitor payload (DXd) via a stable cleavable linker, designed to enhance selective tumor-cell death and reduce systemic exposure[2]
- I-DXd has demonstrated a manageable safety profile and promising antitumor activity in patients with advanced solid tumors, including SCLC, ESCC, mCRPC and sqNSCLC in the Phase 1/2 IDeate-PanTumor01 study (NCT04145622)[1] (**Figure 1**)
- In the ongoing Phase 2 IDeate-Lung01 study (NCT05280470) in patients with extensive-stage SCLC, I-DXd 12 mg/kg IV Q3W was determined to be the recommended dose for further clinical investigation[3]

Figure 1: Ongoing IDeate-PanTumor01 study

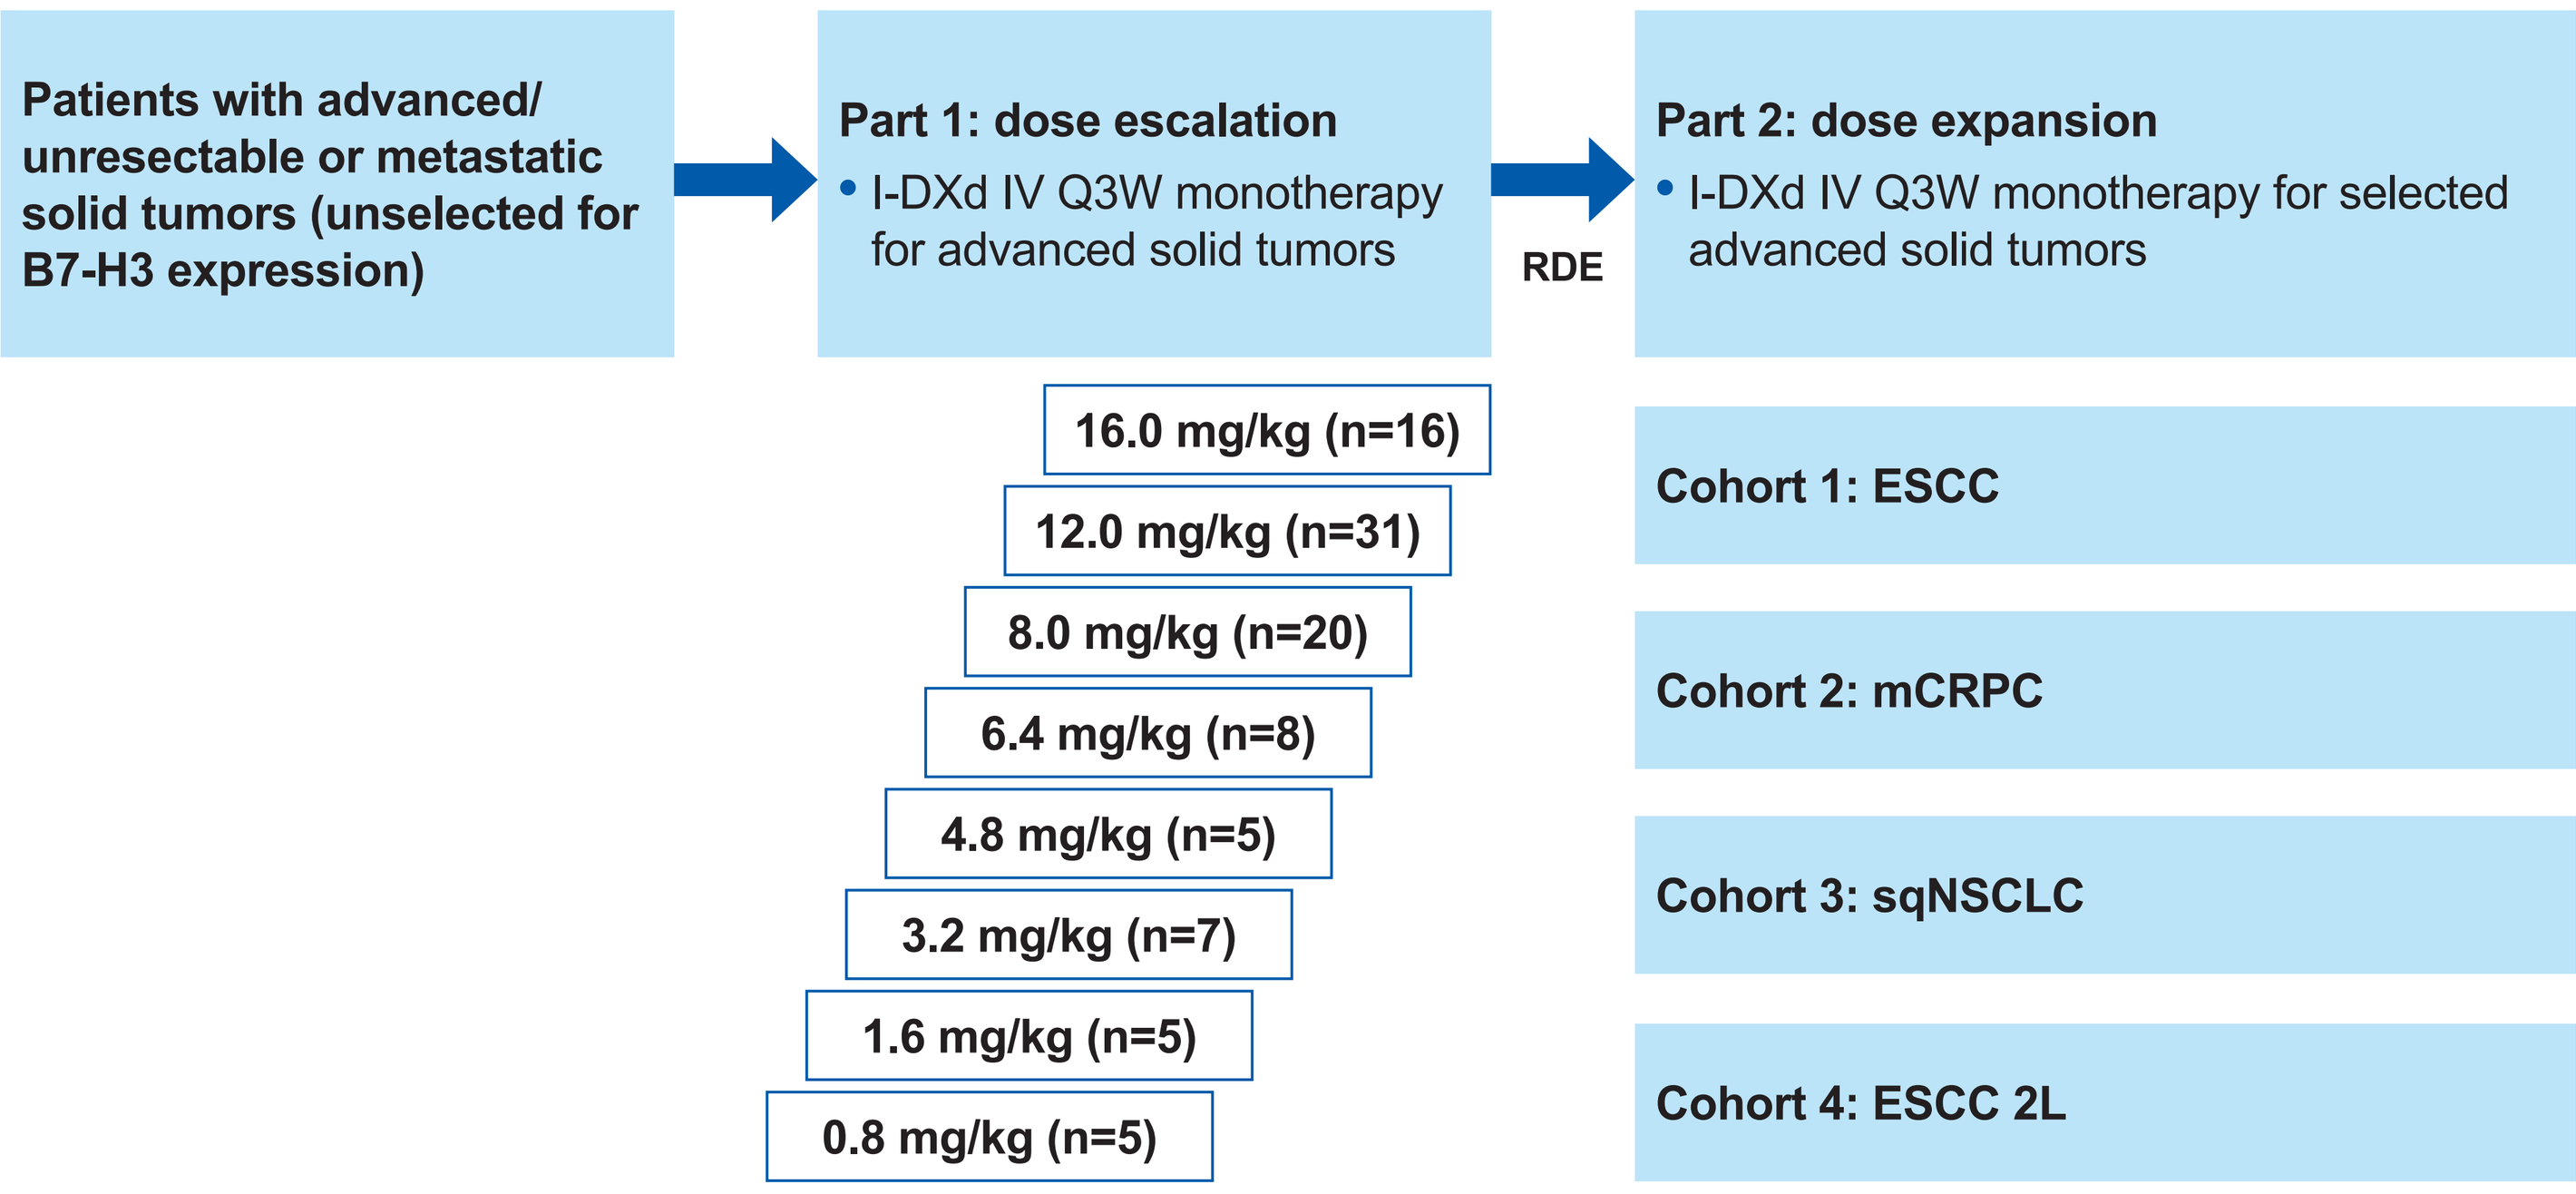
Patients with advanced/ unresectable or metastatic solid tumors (unselected for B7-H3 expression)
Part 1: dose escalation
• I-DXd IV Q3W monotherapy for advanced solid tumors
RDE
Part 2: dose expansion
• I-DXd IV Q3W monotherapy for selected advanced solid tumors
16.0 mg/kg (n=16)
12.0 mg/kg (n=31)
8.0 mg/kg (n=20)
6.4 mg/kg (n=8)
4.8 mg/kg (n=5)
3.2 mg/kg (n=7)
1.6 mg/kg (n=5)
0.8 mg/kg (n=5)
Cohort 1: ESCC
Cohort 2: mCRPC
Cohort 3: sqNSCLC
Cohort 4: ESCC 2L

168 patients from Part 1 and Part 2 of the ongoing IDeate-PanTumor01 study were included in this analysis, out of a planned total of approximately 250 patients in this study. All patients in Part 1 were analyzed. In addition, 71 patients dosed at I-DXd 12 mg/kg in Part 2 were analyzed (25 in Cohort 1, 40 in Cohort 2, and 6 in Cohort 3).

METHODS

- PK and safety data from all doses of I-DXd (0.8–16 mg/kg IV Q3W) in the IDeate-PanTumor01 study (data cutoff: January 31, 2023) were analyzed
- PK parameters, calculated by non-compartmental analysis, were evaluated for I-DXd (ADC) and DXd (released payload)
- ES analyses were completed as follows:
 - 17 safety endpoints were included in the ES analyses: Grade ≥3 TEAEs; anemia (any grade and Grade ≥3); drug discontinuation; dose interruption; dose reduction; GI TEAE (any grade and Grade ≥3); ILD (any grade and Grade ≥3); left ventricular ejection fraction decrease (any grade and Grade ≥3); neutropenia (any grade and Grade ≥3); any treatment-emergent serious AE; and thrombocytopenia (any grade and Grade ≥3)
 - Univariate logistic regression was used to explore relationships between exposure and safety endpoints that had a frequency of >10%
 - Observed exposure metrics of I-DXd and DXd (AUC_{21d} and C_{max}) were used in the ES analyses

RESULTS

Pharmacokinetics

- PK and safety results available from 168 patients were evaluated
- The observed Cycle 1 PK profiles for 97 patients with SCLC, ESCC, mCRPC, and sqNSCLC treated with I-DXd 12 mg/kg IV Q3W across both the escalation and expansion parts of the study were comparable (**Figure 2**)
- Observed PK parameters at 12 mg/kg in patients with SCLC (n=8, dose escalation) and in the dose-expansion cohorts (ESCC [n=25], mCRPC [n=38], and sqNSCLC [n=6]) were comparable across tumor types for both I-DXd and DXd (**Table 1**)

Figure 2. Arithmetic mean (± SD) Cycle 1 PK profiles for patients with SCLC, ESCC, mCRPC, and sqNSCLC who received I-DXd 12 mg/kg[a]

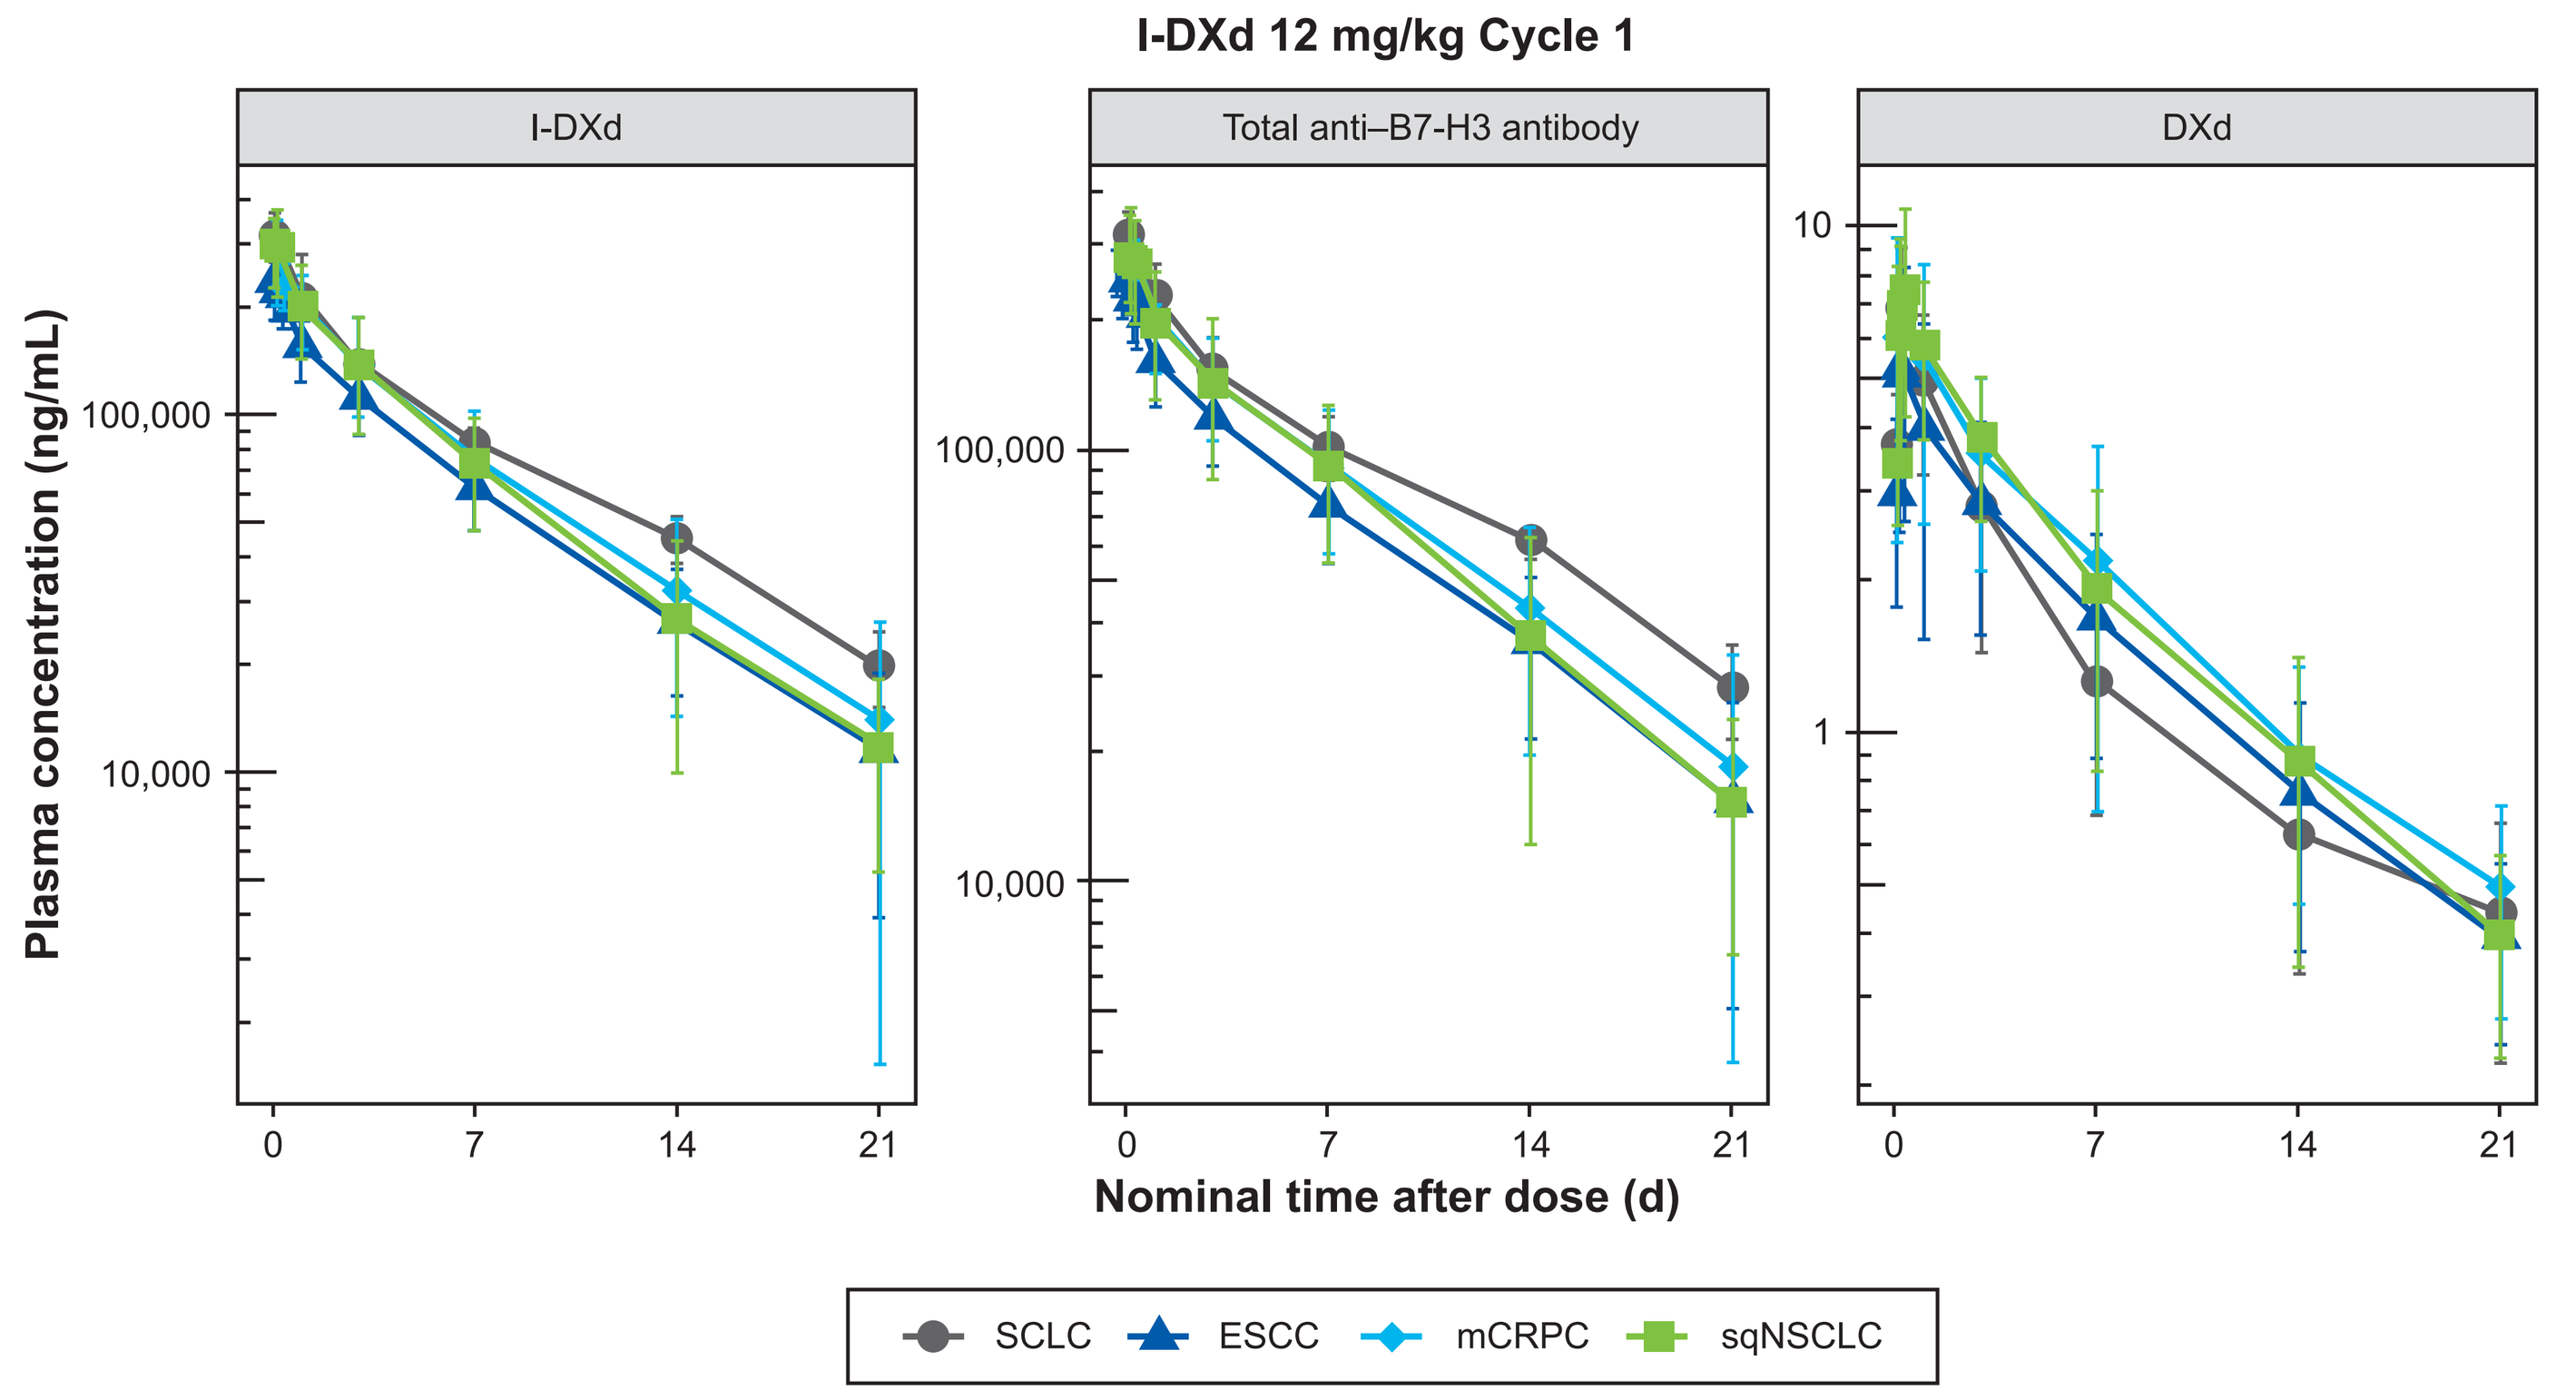
I-DXd 12 mg/kg Cycle 1
I-DXd
Total anti–B7-H3 antibody
DXd
Plasma concentration (ng/mL)
100,000
10,000
10
1
0
7
14
21
Nominal time after dose (d)
SCLC
ESCC
mCRPC
sqNSCLC

[a]Plasma concentration samples taken outside of the protocol-defined sampling window were excluded from the calculation of the mean. The mean plasma concentration is represented at nominal timepoints where there are ≥2 observations. Plasma concentration samples from cycles after a dose reduction in a given patient were excluded.

Table 1. Cycle 1 PK parameters of I-DXd and DXd at 12 mg/kg across tumor types in the dose-expansion cohorts (ESCC, mCRPC, sqNSCLC) and in the SCLC population (dose-escalation part)

Cohort	n	Median (min, max)	n	Arithmetic mean (CV%)	n	Arithmetic mean (CV%)	n	Arithmetic mean (CV%)
I-DXd		**T_{max} (h)**		**C_{max} (µg/mL)**		**AUC_{21d} (d*µg/mL)**		**$t_{½}$ (d)**
SCLC (12 mg/kg, dose escalation)	8	2.4 (1.6, 7.9)	8	319 (14.6)	7	1700 (14.6)	7	6.94 (21.7)
ESCC (12 mg/kg, dose expansion)	25	2.9 (1.5, 45.6)	25	253 (18.4)	23	1230 (21.4)	22	5.27 (30.5)
mCRPC (12 mg/kg, dose expansion)	38	2.95 (1.4, 23.6)	38	298 (17.5)	34	1490 (32.3)	33	5.25 (38.7)
SqNSCLC (12 mg/kg, dose expansion)	6	1.75 (1.5, 3)	6	314 (21.8)	5	1420 (36.3)	5	4.56 (23.3)
DXd		**T_{max} (h)**		**C_{max} (ng/mL)**		**AUC_{21d} (d*ng/mL)**		**$t_{½}$ (d)**
SCLC (12 mg/kg, dose escalation)	8	5.4 (2.9, 8.2)	8	6.48 (40.7)	7	32.0 (40.1)	7	7.35 (27.6)
ESCC (12 mg/kg, dose expansion)	25	5.1 (2.7, 47.2)	25	6.14 (50.4)	23	33.0 (52.1)	21	7.32 (23.2)
mCRPC (12 mg/kg, dose expansion)	38	5 (2.9, 25.3)	38	7.31 (43.5)	33	40.9 (55.7)	30	7.35 (26.7)
SqNSCLC (12 mg/kg, dose expansion)	6	6.6 (2.9, 24.6)	6	7.96 (40.2)	5	42.2 (38.7)	5	8.64 (79.7)

Summary statistics only include values for $t_{½}$ where adjusted r^2 ≥0.75.

Figure 3. Exposure–safety logistic regression: Grade ≥3 TEAEs[a]

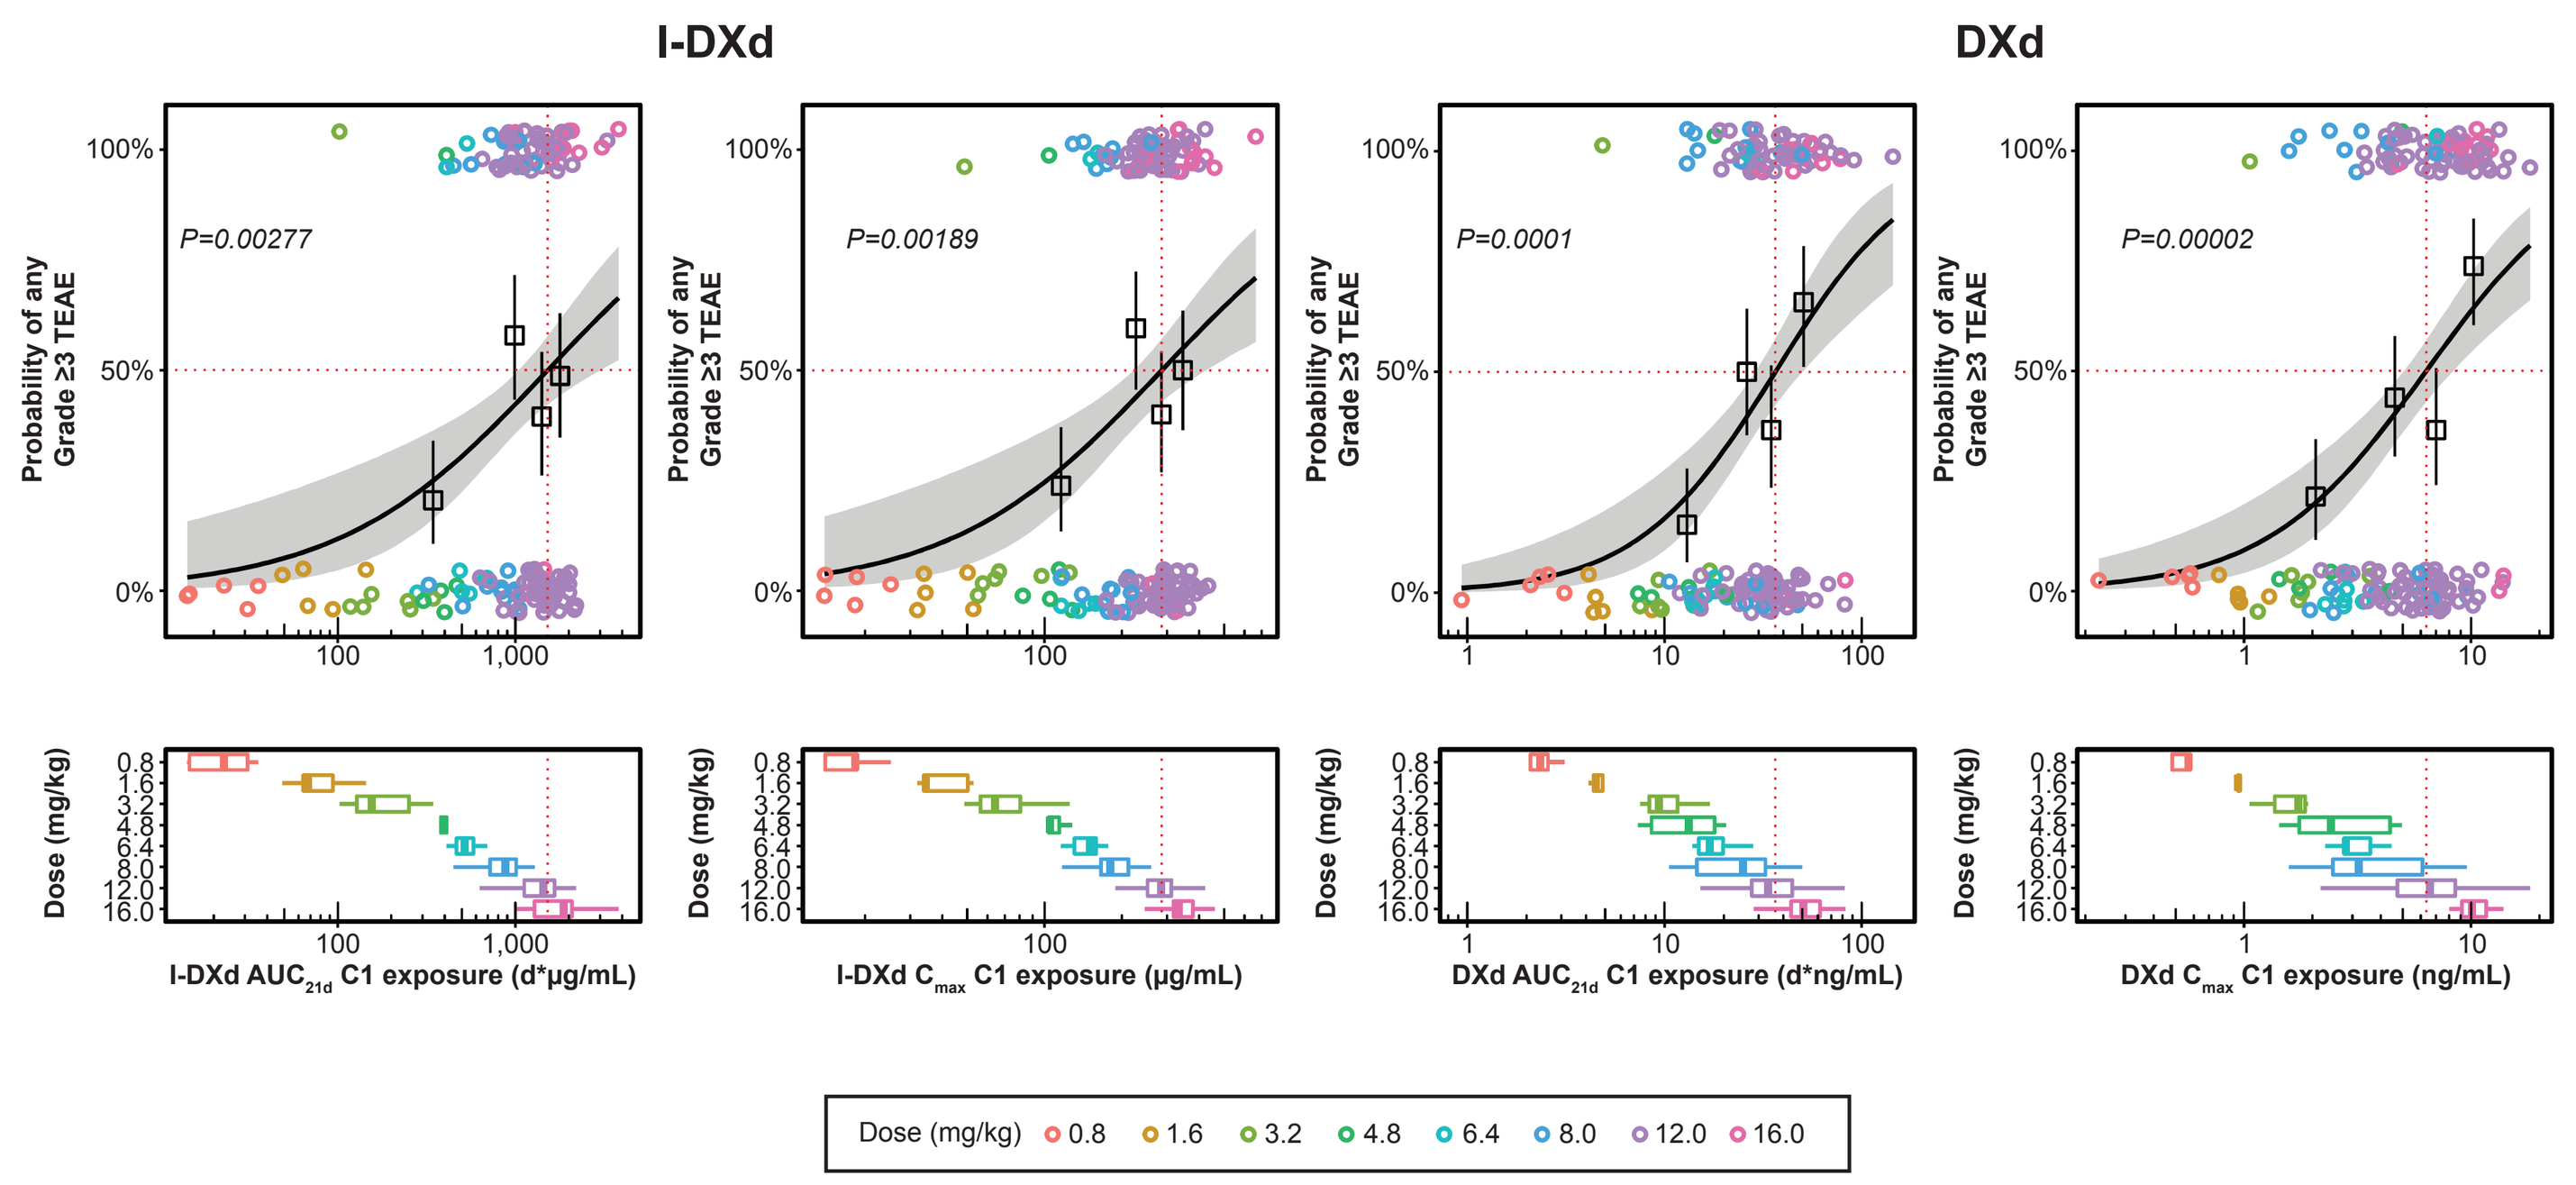
I-DXd
DXd
Probability of any Grade ≥3 TEAE
P=0.00277
P=0.00189
P=0.0001
P=0.00002
Dose (mg/kg)
I-DXd AUC21d C1 exposure (d*µg/mL)
I-DXd Cmax C1 exposure (µg/mL)
DXd AUC21d C1 exposure (d*ng/mL)
DXd Cmax C1 exposure (ng/mL)
Dose (mg/kg) 0.8 1.6 3.2 4.8 6.4 8.0 12.0 16.0

Figure 4. Exposure–safety logistic regression: Grade ≥3 anemia[a]

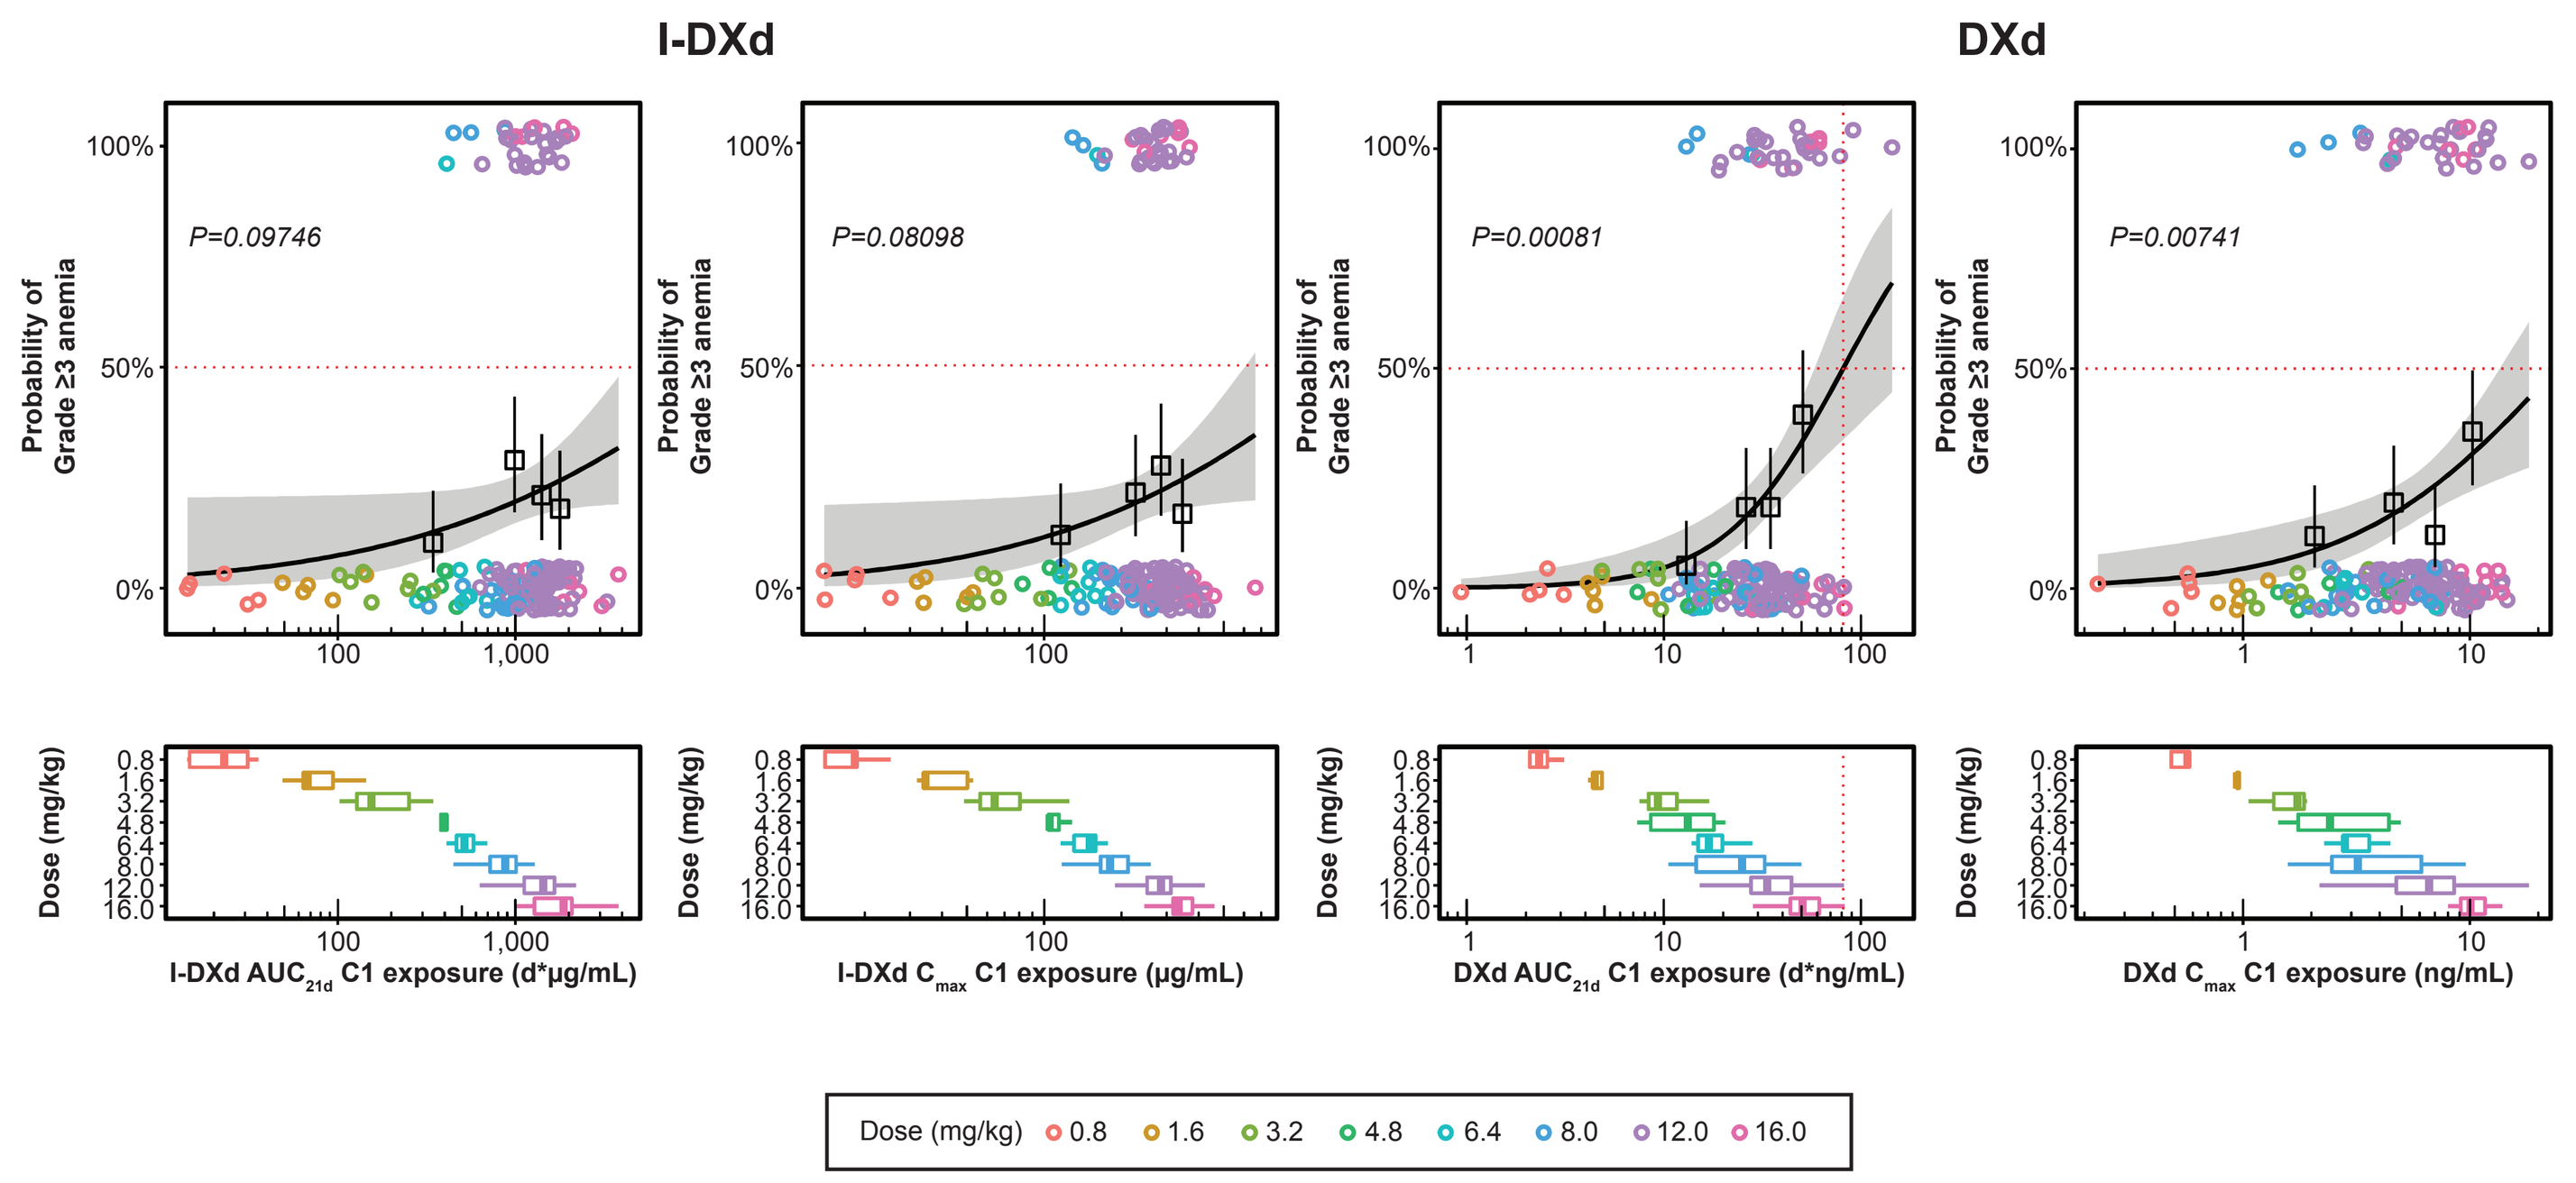
I-DXd
DXd
Probability of Grade ≥3 anemia
P=0.09746
P=0.08098
P=0.00081
P=0.00741
Dose (mg/kg)
I-DXd AUC21d C1 exposure (d*µg/mL)
I-DXd Cmax C1 exposure (µg/mL)
DXd AUC21d C1 exposure (d*ng/mL)
DXd Cmax C1 exposure (ng/mL)
Dose (mg/kg) 0.8 1.6 3.2 4.8 6.4 8.0 12.0 16.0

[a]Solid black line: logistic regression fit; shaded area: 90% CI, box/whiskers: predicted probability of having an AE (median/90% CI); circles: observed AE data (AE = No ~0%, AE = Yes ~100%); dashed red lines: horizontal and vertical intersection of 50% probability of an AE; dose level box plots: median/IQR.

Exposure–safety analysis

- Endpoints with >10% incidence (9 out of 17): Grade ≥3 TEAE, anemia (any-grade and Grade ≥3), dose interruption, dose reduction, any-grade GI TEAE, any-grade neutropenia, any treatment-emergent serious AE, and any-grade thrombocytopenia
- Endpoints with >10% incidence and statistically significant (*P*<0.05) relationship with exposure (7 out of 17):
 - Significant with both I-DXd and DXd Cycle 1 exposure (AUC_{21d} and C_{max}): any-grade GI TEAE, Grade ≥3 TEAE (**Figure 3**), and dose reduction (Cycle 1 AUC_{21d} only)
 - Significant with DXd Cycle 1 exposure (AUC_{21d} and C_{max}): any treatment-emergent serious AE, anemia (any-grade and Grade ≥3 [**Figure 4**]), and dose interruption (DXd Cycle 1 AUC_{21d} only)
- The safety profile of I-DXd was generally consistent across patients with SCLC, ESCC, and mCRPC; incidence of Grade ≥3 TEAEs was <50%. The ES relationship was explored by tumor type, and trends were consistent for the limited available data. The sqNSCLC and ESCC 2L expansion cohorts are ongoing, and further analysis will be conducted

ACKNOWLEDGMENTS

We thank the patients, their families, and their caregivers for their participation and study staff for their contributions. Thank you to Felipe K. Hurtado of Daiichi Sankyo, Inc. for contributions to the analysis.

DISCLOSURES

This study is sponsored by Daiichi Sankyo, Inc. In October 2023, Daiichi Sankyo entered into a global development and commercialization collaboration agreement with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA for ifinatamab deruxtecan (I-DXd).

REFERENCES

1. Patel MR, et al. Presented at the European Society for Medical Oncology Congress 2023. October 20–24, 2023; Madrid, Spain. Abstract 690P.
2. Yamato M, et al. *Mol Cancer Ther.* 2022;21:635–646.
3. Rudin CM, et al. Presented at the IASLC World Conference on Lung Cancer. September 7–10, 2024. San Diego, CA. Abstract OA04.03 and presentation.

ABBREVIATIONS

2L, second-line; ACCP, American College of Clinical Pharmacology; ADC, antibody–drug conjugate; AE, adverse event; AUC_{21d}, area under the plasma concentration-time curve for the 21-day dosing interval; B7-H3, B7 homolog 3; C, cycle; CI, confidence interval; C_{max}, maximum plasma concentration; CV%, coefficient of variation; d, day; DXd, deruxtecan; ES, exposure–safety; ESCC, esophageal squamous cell carcinoma; GI, gastrointestinal; h, hour; I-DXd, ifinatamab deruxtecan; ILD, interstitial lung disease; IQR, interquartile range; IV, intravenous; mCRPC, metastatic castration-resistant prostate cancer; PK, pharmacokinetics; Q3W, every 3 weeks; RDE, recommended dose for expansion; SCLC, small cell lung cancer; SD, standard deviation; sqNSCLC, squamous non-small cell lung cancer; $t_{½}$, half-life; TEAE, treatment-emergent adverse event; T_{max}, time to reach maximum plasma concentration.

Presented at the ACCP annual meeting. September 8–10, 2024; Bethesda, MD, USA.

brittany.tran@daiichisankyo.com