Poster 432P

Exploratory biomarker analysis of trastuzumab deruxtecan versus treatment of physician's choice in HER2-low, hormone receptor–positive metastatic breast cancer in DESTINY-Breast04

Naoto Ueno,[1] Naoki Niikura,[2] Toshinari Yamashita,[3] William Jacot,[4] David Cameron,[5] Junji Tsurutani,[6] Joo-Hyuk Sohn,[7] Eriko Tokunaga,[8] Maria Vidal,[9] Yeon Hee Park,[10] Keun Seok Lee,[11] Yee Soo Chae,[12] Aleix Prat,[13] Fumitaka Suto,[14] Yusuke Kuwahara,[14] Aislyn Boran,[14] Maki Kobayashi,[15] Vinit Kumar,[14] Cecilia Orbegoso Aguilar,[14] Shanu Modi[16]

[1]University of Hawai'i Cancer Center, Honolulu, HI, USA; [2]Tokai University School of Medicine, Isehara, Japan; [3]Kanagawa Cancer Center, Yokohama, Japan; [4]Institut du Cancer de Montpellier, Université de Montpellier, Montpellier, France; [5]Edinburgh University Cancer Centre, University of Edinburgh, Edinburgh, United Kingdom; [6]Advanced Cancer Translational Research Institute, Showa University, Tokyo, Japan; [7]Severance Hospital, Yonsei University College of Medicine, Seoul, Republic of Korea; [8]NHO Kyushu Cancer Center, Fukuoka, Japan; [9]Hospital Clinic de Barcelona, Barcelona, Spain; [10]Samsung Medical Center, Seoul, Republic of Korea; [11]Center for Breast Cancer, National Cancer Center, Goyang-Si, Republic of Korea; [12]School of Medicine, Kyungpook National University Chilgok Hospital, Daegu, Republic of Korea; [13]Translational Genomics and Targeted Therapies Group, August Pi i Sunyer Biomedical Research Institute (IDIBAPS), Barcelona, Spain; [14]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [15]Daiichi Sankyo Co., Ltd, Tokyo, Japan; [16]Memorial Sloan Kettering Cancer Center, New York, NY, USA

Objective

- As part of the exploratory objectives of DESTINY-Breast04 (NCT03734029), relationships of baseline biomarkers for human epidermal growth factor receptor 2 (HER2) expression and mutation, DNA damage response (DDR) or cell proliferation, *BRCA1/2* or homologous recombination repair (HRR) gene alteration, and the tumor microenvironment with clinical outcomes were explored in patients with HER2-low, hormone receptor–positive (HR+) metastatic breast cancer (mBC) treated with trastuzumab deruxtecan (T-DXd) versus treatment of physician's choice (TPC)

Conclusions

- Treatment with T-DXd demonstrated clinically meaningful improvement in progression-free survival (PFS) and confirmed objective response rate (cORR) compared with TPC across biomarker-defined subgroups, including *HER2* gene expression or mutation, *BRCA1/2* or HRR gene alteration status, DDR/cell proliferation gene expression signature status, and tumor microenvironment status
- Based on these results, there does not appear to be a singular mechanism of sensitivity or resistance to T-DXd compared with TPC in DESTINY-Breast04, which is consistent with another study[1]
- Limitations of this analysis include the low patient numbers in some biomarker subgroups and limitations of circulating tumor DNA panel sequencing for detection of structural variants (*BRCA1/2* and HRR alteration status)

Plain language summary

Why did we perform this research?
Trastuzumab deruxtecan (T-DXd) is an antibody-drug conjugate that can direct its chemotherapy payload specifically to tumor cells that express a protein called human epidermal growth factor receptor 2 (HER2) and affect neighboring cells that may not express HER2.[1] Breast cancer can be categorized by measuring the amount of HER2 protein expressed on the surface of tumor cells. In DESTINY-Breast04, a clinical trial in patients with metastatic breast cancer (mBC) whose tumors express low levels of HER2, T-DXd resulted in better treatment outcomes versus chemotherapy of physician's choice (TPC),[2] which led to the approval of T-DXd for the treatment of patients with HER2-low metastatic breast cancer.[3,4] The benefit of T-DXd over TPC in patients with HER2-low mBC in DESTINY-Breast04 has been shown regardless of the level of HER2 protein expression, the molecular subtype of the tumor, the presence or absence of certain tumor mutations, or markers of resistance to other therapies.[5] In the current analysis, we investigated the effect of T-DXd according to *HER2* gene expression or mutation, biomarkers of DNA damage or cell proliferation to assess the effect of the topoisomerase I chemotherapy agent of T-DXd, and biomarkers of tumor microenvironment immune status.

How did we perform this research?
Blood or tumor tissue samples collected from patients in DESTINY-Breast04 before they started the trial treatment (T-DXd or TPC) were measured to determine the levels of biomarkers, gene expression, or gene alterations. Biomarkers were categorized as having high or low levels based on the median value or as the presence or absence of a specific gene alteration. Clinical outcomes were assessed to investigate whether biomarker levels or gene alterations were associated with the effect of treatment with T-DXd versus TPC.

What were the findings of this research?
Treatment with T-DXd consistently demonstrated clinically meaningful improvement in outcomes compared with TPC across all the biomarker groups analyzed, including *HER2* gene expression or mutation, DNA damage and cell proliferation biomarkers, and tumor microenvironment immune status.

What were the implications of this research?
These results demonstrated that the clinical outcomes of T-DXd do not appear to be influenced by the biomarkers assessed in this analysis.

Where can I access more information?
DESTINY-Breast04: ClinicalTrial.gov. Trastuzumab Deruxtecan (DS-8201a) Versus Investigator's Choice for HER2-Low Breast Cancer That Has Spread or Cannot Be Surgically Removed. https://www.clinicaltrials.gov/ct2/show/NCT03734029

1. Ogitani Y et al. *Cancer Sci.* 2016;107:1039-1046.
2. Modi S et al. *N Engl J Med.* 2022;387:9-20.
3. Daiichi Sankyo Inc. ENHERTU® (fam-trastuzumab deruxtecan-nxki) for injection, for intravenous use. 2024.
4. Daiichi Sankyo Europe GmbH. Enhertu 100 mg powder for concentrate for solution infusion (SPC). 2024.
5. Modi S et al. *J Clin Oncol.* 2023;41:1020-1020.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

This study was sponsored by Daiichi Sankyo. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201).

This information is the intellectual property of the authors/presenters. Please contact Dr Naoto Ueno at nueno@cc.hawaii.edu for permission to reprint and/or redistribute.

Poster presented at the ESMO Congress 2024; September 13-17, 2024; Barcelona, Spain

Introduction

- T-DXd is a HER2-directed antibody-drug conjugate that can target tumor cells with low levels of HER2 expression and deliver the DXd payload to neighboring tumor cells, regardless of HER2 protein expression, through the bystander antitumor effect[2,3]
- Based on results of the DESTINY-Breast04 trial, T-DXd is approved for the treatment of patients with unresectable or metastatic HER2-low (immunohistochemistry [IHC] score 1+ or IHC 2+ and in situ hybridization–negative [ISH-]) breast cancer who have received chemotherapy in the metastatic setting or developed disease recurrence during or within 6 months of completing adjuvant chemotherapy[4,5]
- DESTINY-Breast04 demonstrated significantly longer PFS and overall survival for T-DXd versus TPC, and clinical benefit was observed regardless of HER2 IHC status or previous treatment with cyclin-dependent kinase 4/6 inhibitors[6]
- Previous exploratory biomarker analysis in DESTINY-Breast04 demonstrated that the clinical benefit of T-DXd versus TPC was observed regardless of intrinsic subtype (HER2-enriched, luminal A, or luminal B), *ESR1* or *PIK3CA* gene mutation status, or the presence or absence of known markers of cyclin-dependent kinase 4/6 inhibitor resistance[7]
- The cytotoxic payload of T-DXd is a topoisomerase I inhibitor[8]; therefore, biomarkers of DDR and cell proliferation pathways were hypothesized to be potentially predictive or prognostic of response.[9] Preclinical research suggests that T-DXd increases antitumoral immune cells in the tumor microenvironment[9]; therefore, biomarkers of the tumor microenvironment immune status, such as total lymphocyte levels, were hypothesized to be associated with antitumor activity[10]

Methods

- We performed this exploratory biomarker analysis in patients with HER2-low HR+ mBC enrolled in DESTINY-Breast04 (data cutoff, January 11, 2022)
- Samples for biomarker analysis were collected at baseline before study treatment, after the last line of prior systemic therapy
 - Gene expression levels were derived from RNA sequencing, performed on tumor tissue samples. Gene expression signature scores were calculated using the single sample gene set enrichment analysis method[11] with the Molecular Signature Database hallmark gene set[12]
 - Circulating tumor DNA (ctDNA) plasma samples were analyzed using the Guardant OMNI panel (approximately 500 genes) to detect *HER2*-activating mutations (defined as *HER2* gain of function mutation according to the OncoKB database), *BRCA1/2* inactivating alterations (nonsense mutations, frameshift insertions/deletions, splice site alterations, homozygous deletions, loss of heterozygosity deletions), per the Guardant pipeline, and HRR gene alterations (per published methods[13])
 - Multicolor IHC analysis was performed on baseline (pretreatment) tumor tissue samples to determine the density of immune cells. Levels of total lymphocytes (including T cells, B cells, and natural killer [NK] cells), M1 macrophages, CD8+ T cells, CD56+ NK cells, CD4+ T cells, CD20+ B cells, and CD8+Ki67+ T cells were defined as immune active tumor microenvironment markers and levels of regulatory T cells (Tregs), PD1+ T cells, and M2 macrophages were defined as immune suppressive tumor microenvironment markers
- High versus low biomarker levels were defined, respectively, as greater than or equal to versus lower than the median value across the entire biomarker dataset
- We investigated relationships between biomarker status and clinical outcomes of cORR or PFS. Median PFS (mPFS) was estimated using the Kaplan-Meier method, and hazard ratios were calculated by comparing T-DXd with TPC. *P* values for biomarker interaction were assessed for T-DXd. The threshold for significance was *P* <0.05

Results

- Overall, cORR was higher and mPFS was longer with T-DXd versus TPC in the biomarker-defined populations, which was consistent with the intention-to-treat (ITT) population, and clinical outcomes were comparable between the biomarker-evaluable populations (**QR code Supplementary Table 1**)

HER2 status

- The clinical benefit of T-DXd over TPC was observed regardless of HER2 IHC status, *HER2* gene expression level, or *HER2* mutation status, and there was no statistically significant interaction based on these HER2 biomarkers (**Figures 1, 2, QR code Supplementary Figure 1**)
 - A non–significant trend for improved cORR (**Figure 1A**) and longer PFS (**Figure 2B**) was apparent in patients with higher versus lower *HER2* gene expression levels, particularly in the T-DXd arm

DDR and cell proliferation signatures

- The clinical benefit of T-DXd over TPC was observed regardless of the level of DDR or cell proliferation gene expression signatures, and there was no statistically significant biomarker interaction (**Figure 3, QR code Supplementary Figure 2**)
 - cORR was similar between patients with high and low levels of DDR or cell proliferation gene expression signatures (**QR code Supplementary Figure 2A**). Although the interaction *P* value was <0.05 for the G2/M checkpoint gene expression signature, the range of signatures examined, representing cell proliferation and DDR transcriptional programs, did not show any significant association. Overall, a consistent, but non–statistically significant, trend for shorter PFS in both treatment arms was observed in patients with higher levels of DDR or cell proliferation gene expression signatures (**QR code Supplementary Figure 2B**)

BRCA1/2 and HRR alteration status

- The clinical benefit of T-DXd over TPC was maintained regardless of *BRCA1/2* or HRR gene alteration status, and there was no statistically significant biomarker interaction (**Figure 4, Supplementary Figure 3A**)
 - A trend for shorter PFS was observed in both treatment arms for patients with *BRCA1/2* or HRR gene alterations compared with those with no alterations detected

Tumor microenvironment status

- The clinical benefit of T-DXd over TPC was observed for the majority of immune features measured by multicolor IHC analysis (**Figures 5, 6**)
 - For patients with higher levels of total lymphocytes in the stroma, a trend towards longer PFS was observed compared with those with lower levels in both treatment arms (**Figure 6**), which was consistent with trends observed across immune active subgroups (**Figure 5B**)
 - A consistent trend towards longer PFS was observed with a majority of tumoral and stromal immune markers (**Figure 5**)

Figure 1. Association of baseline HER2-related biomarkers with (A) cORR and (B) hazard ratio for PFS

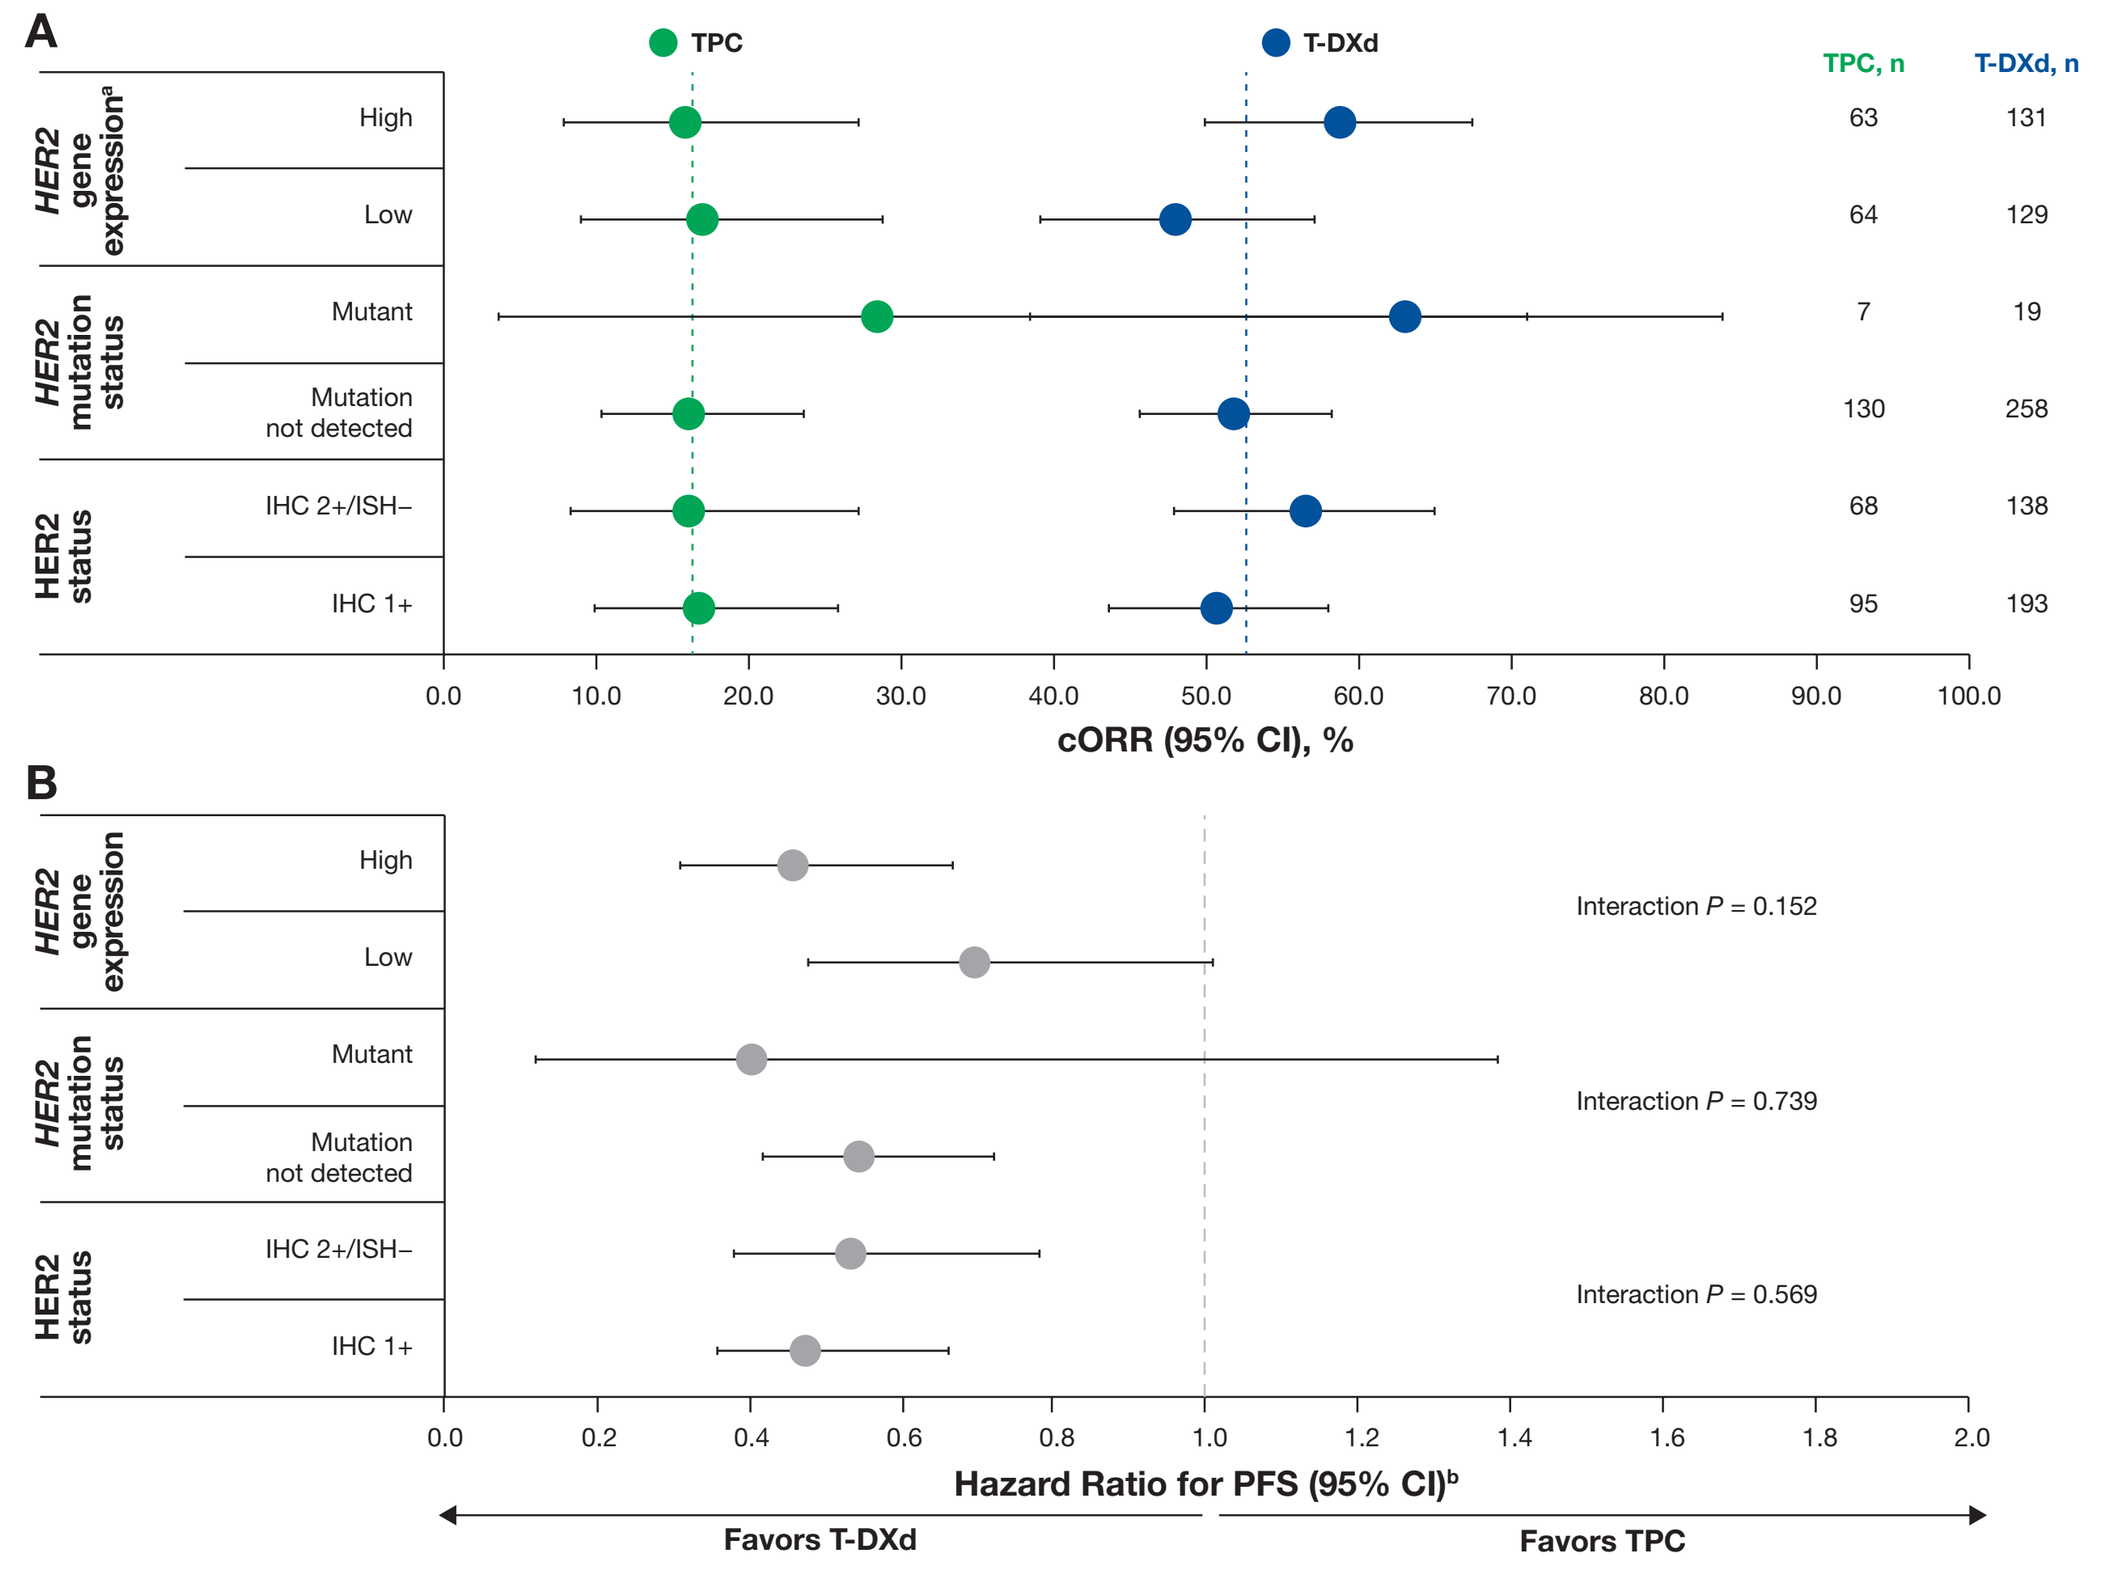

A
TPC
T-DXd
TPC, n
T-DXd, n
HER2 gene expression[a]
High 63 131
Low 64 129
HER2 mutation status
Mutant 7 19
Mutation not detected 130 258
HER2 status
IHC 2+/ISH– 68 138
IHC 1+ 95 193
cORR (95% CI), %
B
Interaction P = 0.152
Interaction P = 0.739
Interaction P = 0.569
Hazard Ratio for PFS (95% CI)[b]
Favors T-DXd
Favors TPC

Dashed vertical lines in panel A show cORR in the HR+ ITT population.
[a]High and low were defined according to the median value.
[b]Hazard ratio for T-DXd vs TPC.

Figure 2. Probability of PFS according to (A) HER2 IHC status and (B) *HER2* gene expression at baseline

A

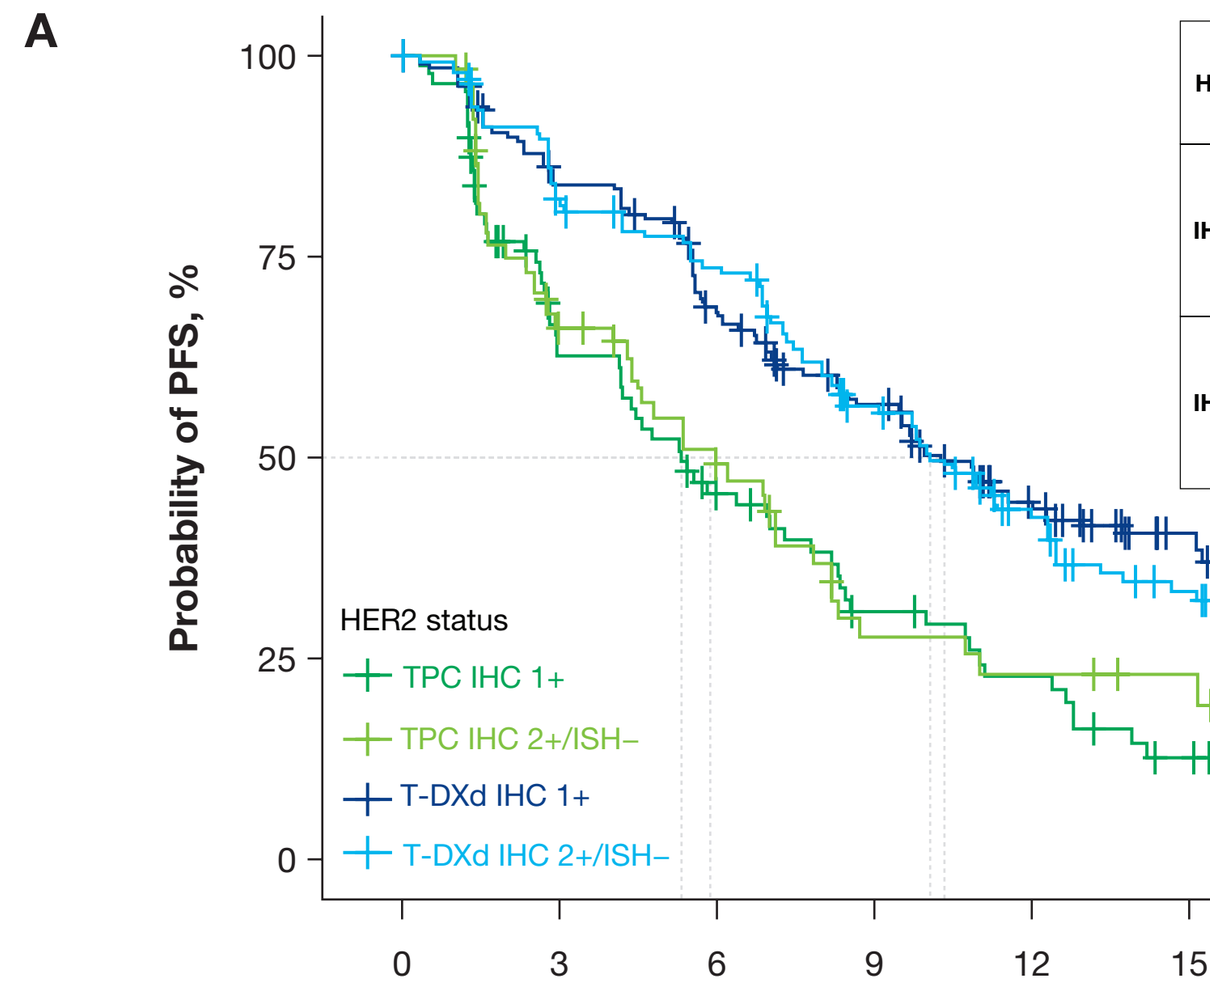

Probability of PFS, %
HER2 status
TPC IHC 1+
TPC IHC 2+/ISH–
T-DXd IHC 1+
T-DXd IHC 2+/ISH–
Time, months

HER2 status	Treatment arm (n)	mPFS (95% CI), months	Hazard ratio (95% CI)	Interaction *P* value
IHC 1+	T-DXd (193)	10.32 (8.52-12.30)	0.48 (0.36-0.66)	0.569
	TPC (95)	5.33 (4.14-7.79)		
IHC 2+/ISH–	T-DXd (138)	10.06 (8.19-12.23)	0.54 (0.38-0.78)	
	TPC (68)	5.88 (4.31-7.86)		

Number at risk	0	3	6	9	12	15	18	21	24	27	30
TPC IHC 1+	95	48	32	20	14	6	2	1	1	0	0
TPC IHC 2+/ISH–	68	37	25	12	10	6	1	0	0	0	0
T-DXd IHC 1+	193	156	123	97	64	44	23	13	4	0	0
T-DXd IHC 2+/ISH–	138	109	95	68	45	29	14	4	3	1	0

B

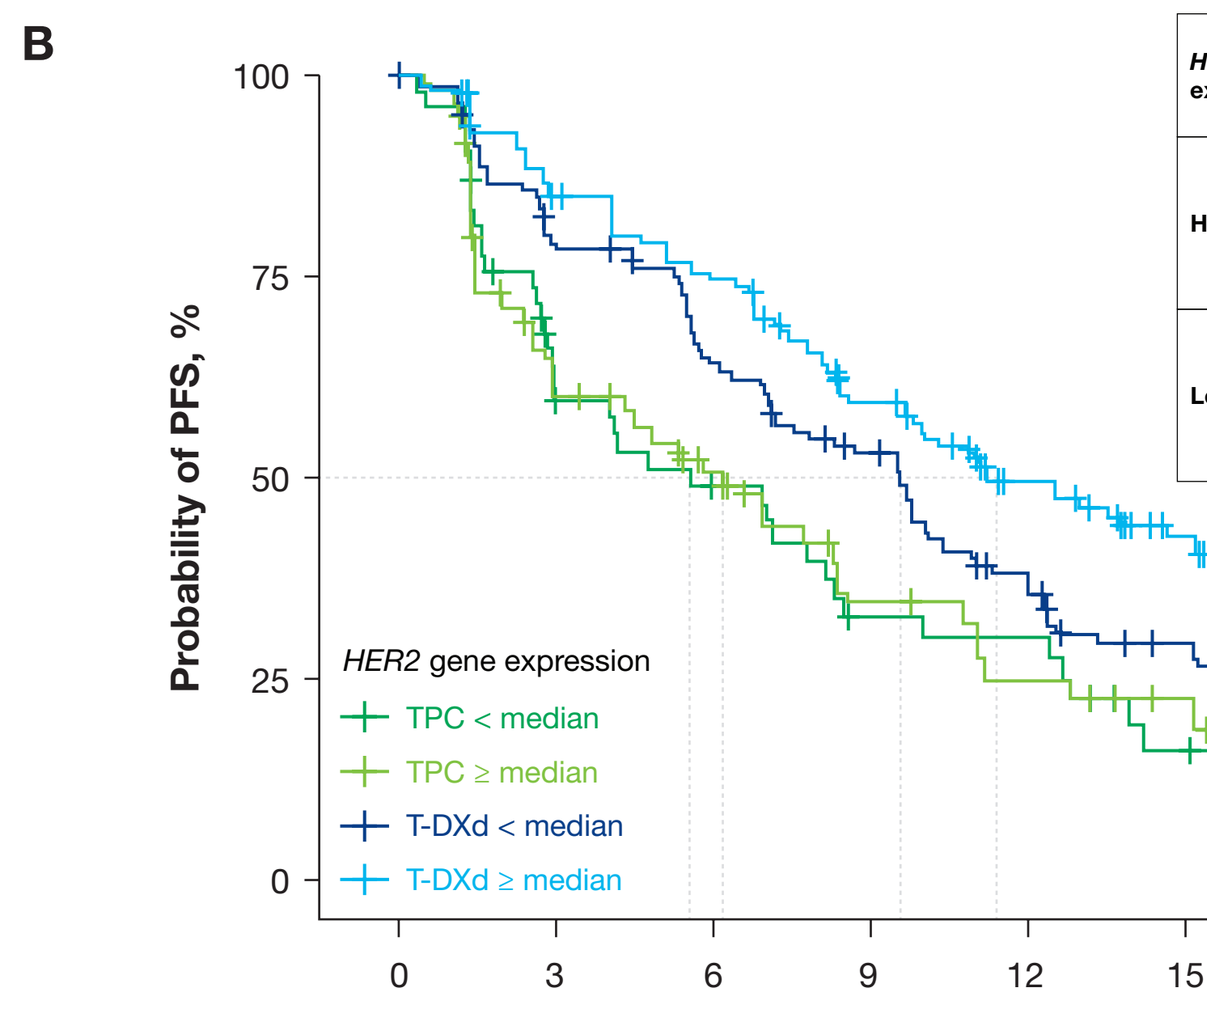

Probability of PFS, %
HER2 gene expression
TPC < median
TPC ≥ median
T-DXd < median
T-DXd ≥ median
Time, months

HER2 gene expression level	Treatment arm (n)	mPFS (95% CI), months	Hazard ratio (95% CI)	Interaction *P* value
High (≥ median)	T-DXd (131)	11.41 (9.53-15.12)	0.46 (0.31-0.67)	0.152
	TPC (63)	6.21 (2.83-8.55)		
Low (< median)	T-DXd (129)	9.57 (7.04-10.32)	0.70 (0.48-1.01)	
	TPC (64)	5.56 (2.93-8.32)		

Number at risk	0	3	6	9	12	15	18	21	24	27	30
TPC < median	64	28	21	13	12	5	2	1	1	0	0
TPC ≥ median	63	33	24	15	10	6	1	0	0	0	0
T-DXd < median	129	98	79	62	40	24	11	6	3	0	0
T-DXd ≥ median	131	106	92	69	48	34	18	8	3	1	0

Median PFS was estimated by the Kaplan-Meier method, and the hazard ratio was calculated comparing the T-DXd and TPC arms in each biomarker-derived subgroup.

Figure 3. Probability of PFS according to (A) DNA repair and (B) G2/M checkpoint gene expression signature levels at baseline

A

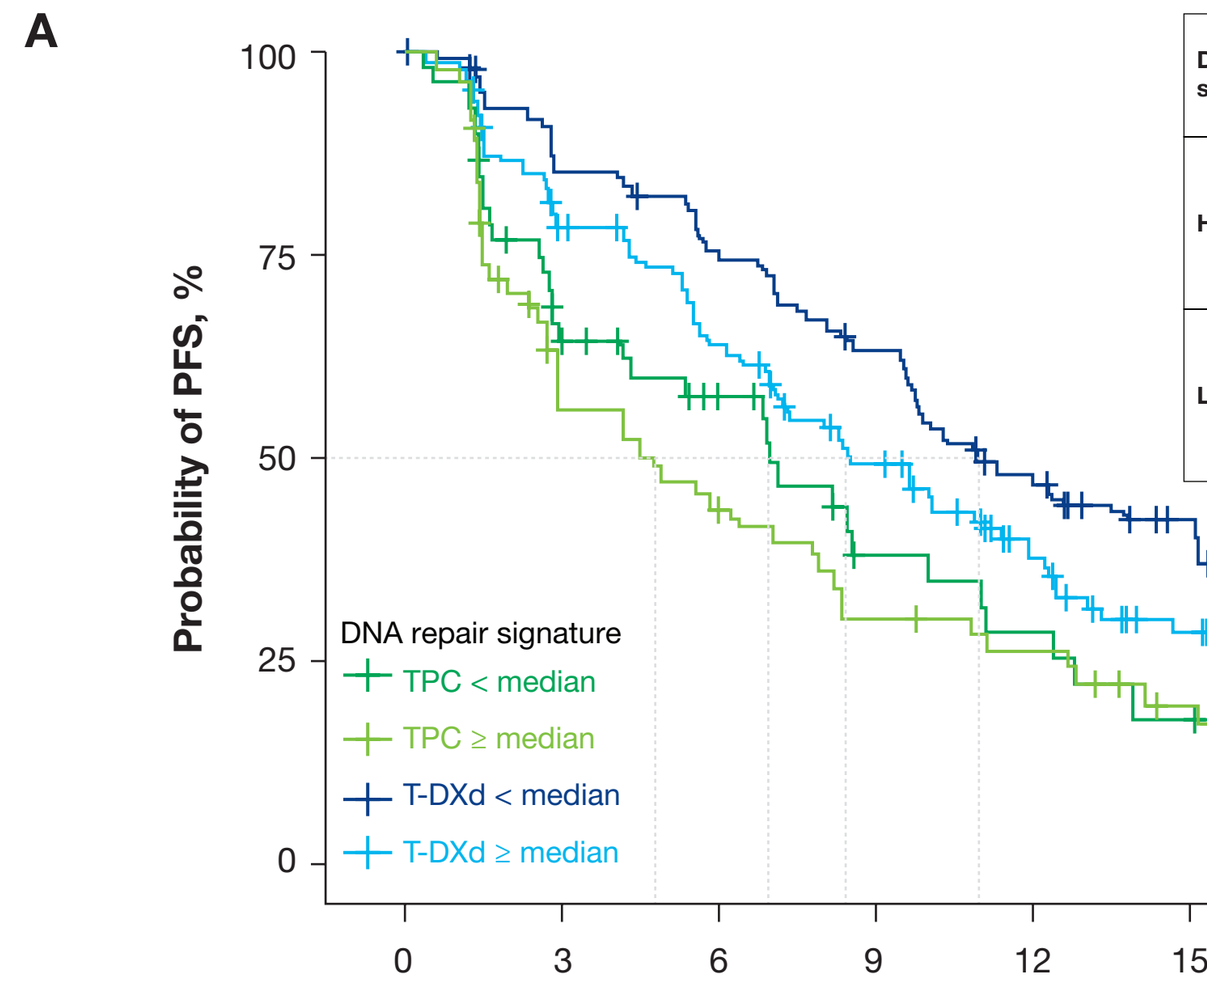

Probability of PFS, %
DNA repair signature
TPC < median
TPC ≥ median
T-DXd < median
T-DXd ≥ median
Time, months

DNA repair signature	Treatment arm (n)	mPFS (95% CI), months	Hazard ratio (95% CI)	Interaction *P* value
High (≥ median)	T-DXd (126)	8.45 (6.97-11.41)	0.62 (0.43-0.89)	0.634
	TPC (68)	4.77 (2.73-7.86)		
Low (< median)	T-DXd (134)	10.98 (9.67-15.12)	0.52 (0.35-0.78)	
	TPC (59)	6.94 (2.93-9.99)		

Number at risk	0	3	6	9	12	15	18	21	24	27	30
TPC < median	59	30	22	12	9	4	2	1	1	0	0
TPC ≥ median	68	31	23	16	13	7	1	0	0	0	0
T-DXd < median	134	109	95	78	56	40	22	13	5	1	0
T-DXd ≥ median	126	95	76	53	32	18	7	1	1	0	0

B

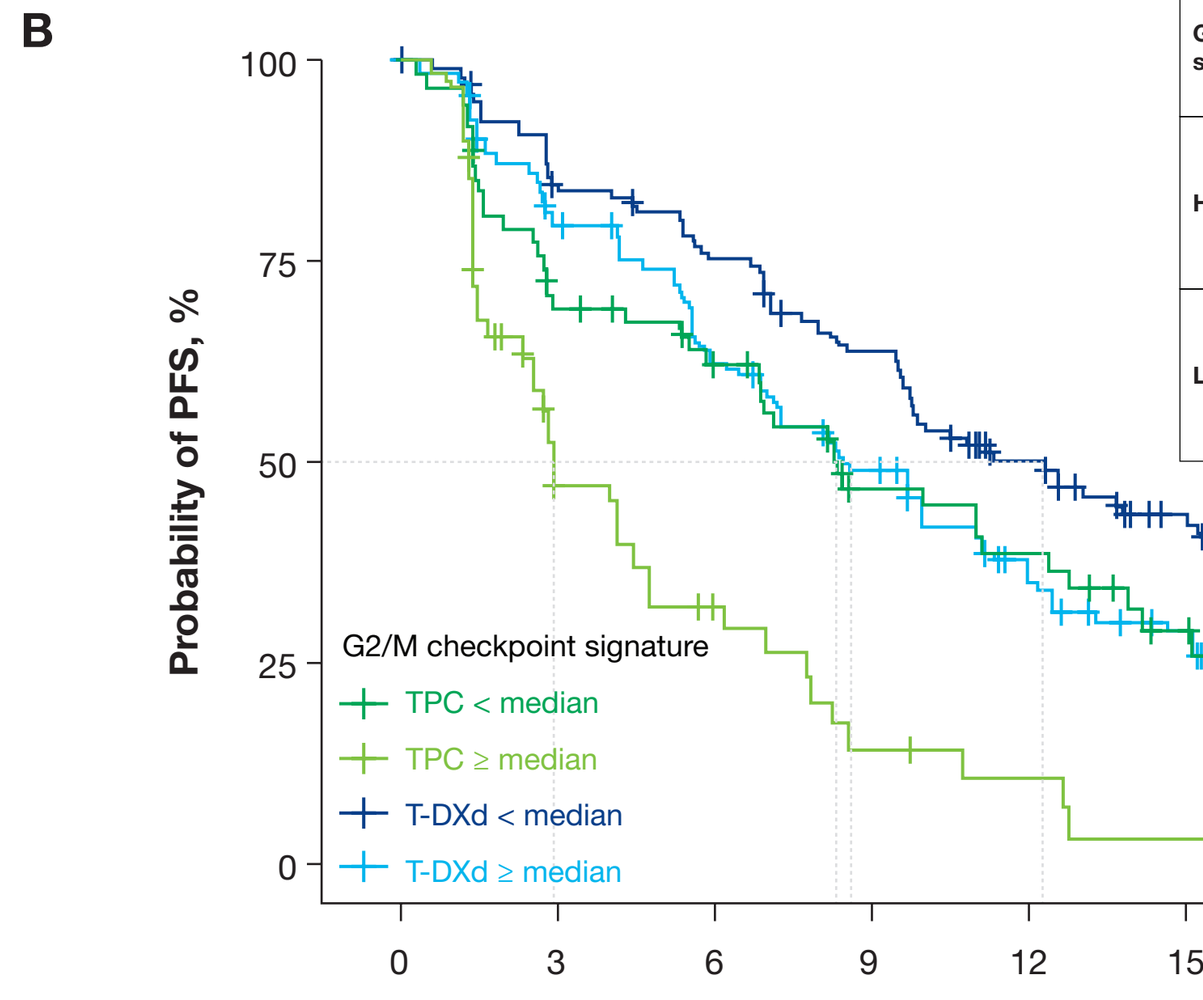

Probability of PFS, %
G2/M checkpoint signature
TPC < median
TPC ≥ median
T-DXd < median
T-DXd ≥ median
Time, months

G2/M checkpoint signature	Treatment arm (n)	mPFS (95% CI), months	Hazard ratio (95% CI)	Interaction *P* value
High (≥ median)	T-DXd (135)	8.65 (7.04-10.88)	0.37 (0.25-0.54)	0.039
	TPC (59)	2.96 (2.37-4.77)		
Low (< median)	T-DXd (125)	12.30 (9.73-16.44)	0.66 (0.45-0.98)	
	TPC (68)	8.35 (5.82-12.39)		

Number at risk	0	3	6	9	12	15	18	21	24	27	30
TPC < median	68	42	34	23	19	10	3	1	1	0	0
TPC ≥ median	59	19	11	5	3	1	0	0	0	0	0
T-DXd < median	125	99	88	71	48	32	17	11	6	1	0
T-DXd ≥ median	135	105	83	60	40	26	12	3	0	0	0

Median PFS was estimated by the Kaplan-Meier method, and the hazard ratio was calculated comparing the T-DXd and TPC arms in each biomarker-derived subgroup.

Figure 4. Probability of PFS according to (A) *BRCA1/2* and (B) HRR gene alteration status at baseline

A

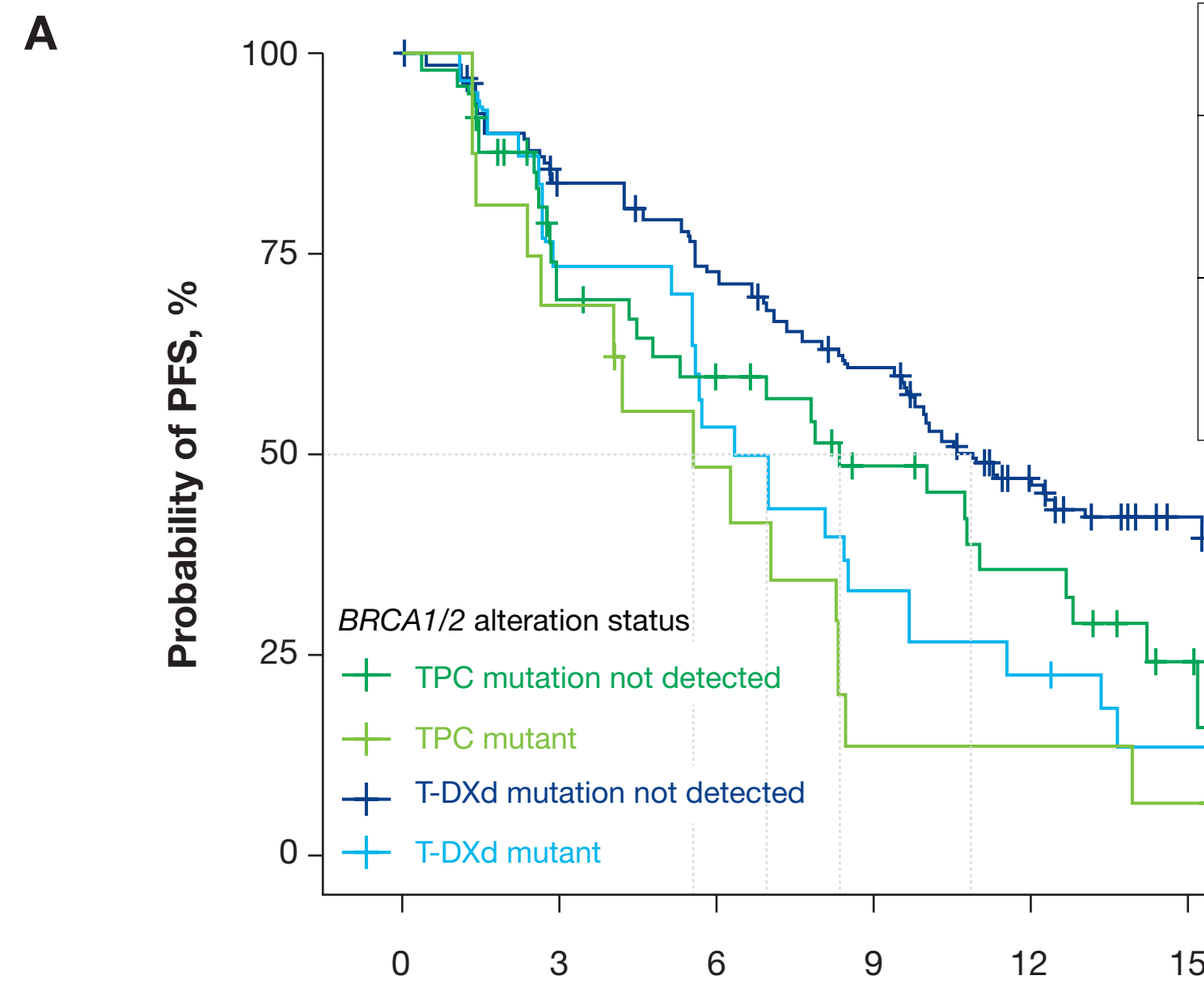

Probability of PFS, %
BRCA1/2 alteration status
TPC mutation not detected
TPC mutant
T-DXd mutation not detected
T-DXd mutant
Time, months

BRCA1/2 alteration status	Treatment arm (n)	mPFS (95% CI), months	Hazard ratio (95% CI)	Interaction *P* value
BRCA1/2 mutant	T-DXd (31)	6.94 (5.39-9.53)	0.71 (0.37-1.37)	0.882
	TPC (16)	5.56 (2.37-8.32)		
BRCA1/2 mutation not detected	T-DXd (135)	10.85 (9.57-15.16)	0.62 (0.41-0.95)	
	TPC (53)	8.35 (4.47-12.66)		

Number at risk	0	3	6	9	12	15	18	21	24	27	30
TPC mutation not detected	53	30	23	16	11	4	0	0	0	0	0
TPC mutant	16	11	7	2	2	1	0	0	0	0	0
T-DXd mutation not detected	135	107	91	74	49	34	15	8	2	0	0
T-DXd mutant	31	22	16	10	6	3	2	1	1	0	0

B

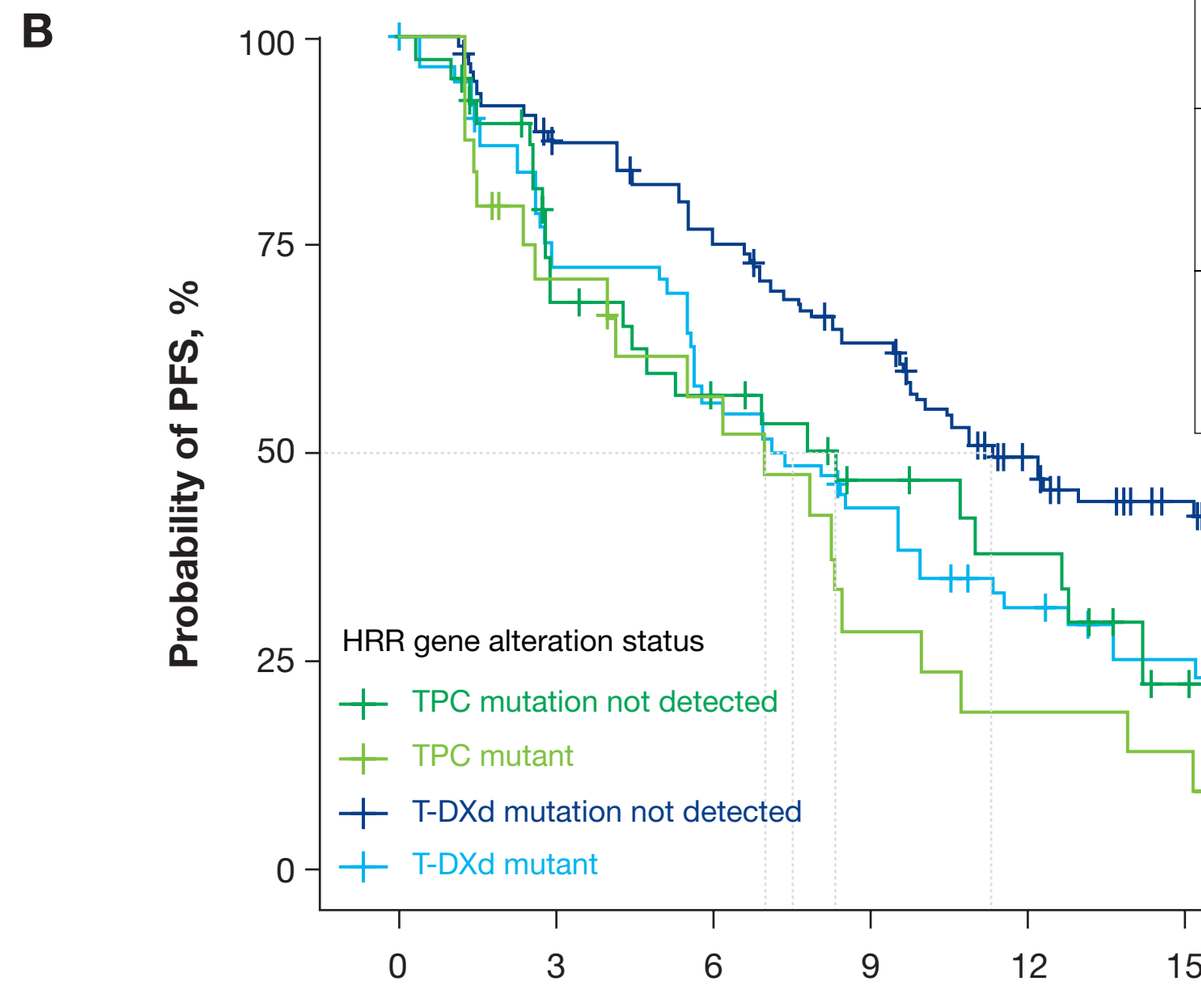

Probability of PFS, %
HRR gene alteration status
TPC mutation not detected
TPC mutant
T-DXd mutation not detected
T-DXd mutant
Time, months

HRR gene alteration status	Treatment arm (n)	mPFS (95% CI), months	Hazard ratio (95% CI)	Interaction *P* value
HRR gene mutant	T-DXd (64)	7.53 (5.59-9.96)	0.72 (0.42-1.21)	0.616
	TPC (26)	7.0 (2.63-8.45)		
HRR gene mutation not detected	T-DXd (102)	11.31 (9.67-20.09)	0.57 (0.35-0.92)	
	TPC (43)	8.35 (4.31-12.79)		

Number at risk	0	3	6	9	12	15	18	21	24	27	30
TPC mutation not detected	43	25	18	12	9	2	0	0	0	0	0
TPC mutant	26	16	12	6	4	3	0	0	0	0	0
T-DXd mutation not detected	102	84	72	58	38	25	9	5	2	0	0
T-DXd mutant	64	45	35	26	17	12	8	4	1	0	0

Median PFS was estimated by the Kaplan-Meier method, and the hazard ratio was calculated comparing the T-DXd and TPC arms in each biomarker-derived subgroup.

Figure 5. Association of tumor microenvironment immune features with (A) cORR and (B) hazard ratio for PFS

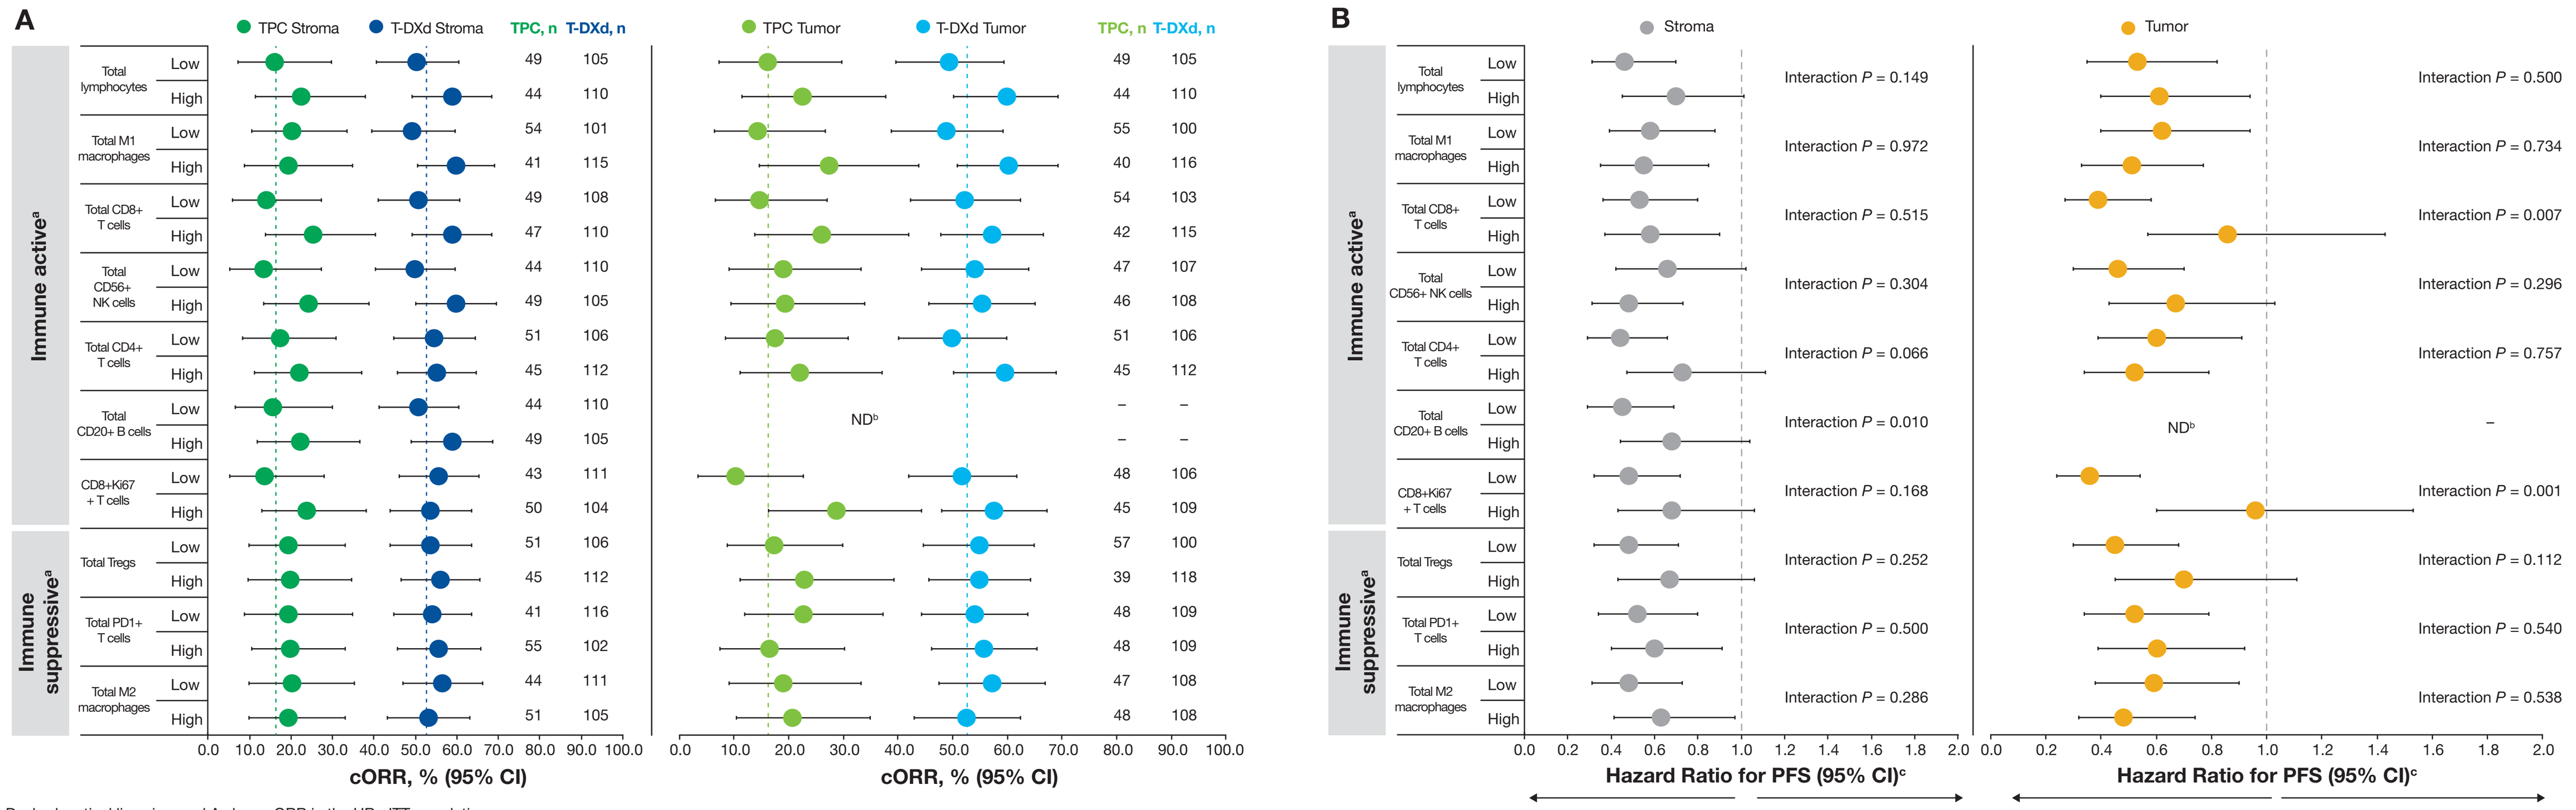

A
TPC Stroma
T-DXd Stroma
TPC, n
T-DXd, n
TPC Tumor
T-DXd Tumor
Immune active[a]
Total lymphocytes Low 49 105 / 49 105
Total lymphocytes High 44 110 / 44 110
Total M1 macrophages Low 54 101 / 55 100
Total M1 macrophages High 41 115 / 40 116
Total CD8+ T cells Low 49 108 / 54 103
Total CD8+ T cells High 47 110 / 42 115
Total CD56+ NK cells Low 44 110 / 47 107
Total CD56+ NK cells High 49 105 / 46 108
Total CD4+ T cells Low 51 106 / 51 106
Total CD4+ T cells High 45 112 / 45 112
Total CD20+ B cells Low 44 110 / – –
Total CD20+ B cells High 49 105 / – –
CD8+Ki67 + T cells Low 43 111 / 48 106
CD8+Ki67 + T cells High 50 104 / 45 109
Immune suppressive[a]
Total Tregs Low 51 106 / 57 100
Total Tregs High 45 112 / 39 118
Total PD1+ T cells Low 41 116 / 48 109
Total PD1+ T cells High 55 102 / 48 109
Total M2 macrophages Low 44 111 / 47 108
Total M2 macrophages High 51 105 / 48 108
ND[b]
cORR, % (95% CI)
B
Stroma
Tumor
Interaction P = 0.149 / Interaction P = 0.500
Interaction P = 0.972 / Interaction P = 0.734
Interaction P = 0.515 / Interaction P = 0.007
Interaction P = 0.304 / Interaction P = 0.296
Interaction P = 0.066 / Interaction P = 0.757
Interaction P = 0.010 / –
Interaction P = 0.168 / Interaction P = 0.001
Interaction P = 0.252 / Interaction P = 0.112
Interaction P = 0.500 / Interaction P = 0.540
Interaction P = 0.286 / Interaction P = 0.538
Hazard Ratio for PFS (95% CI)[c]
Favors T-DXd
Favors TPC

Dashed vertical lines in panel A show cORR in the HR+ ITT population.
[a]High and low were defined according to the median value.
[b]Not evaluated because cell density was zero in more than half of the patients.
[c]Hazard ratio for T-DXd vs TPC.

Figure 6. Probability of PFS according to levels of stromal total lymphocytes at baseline

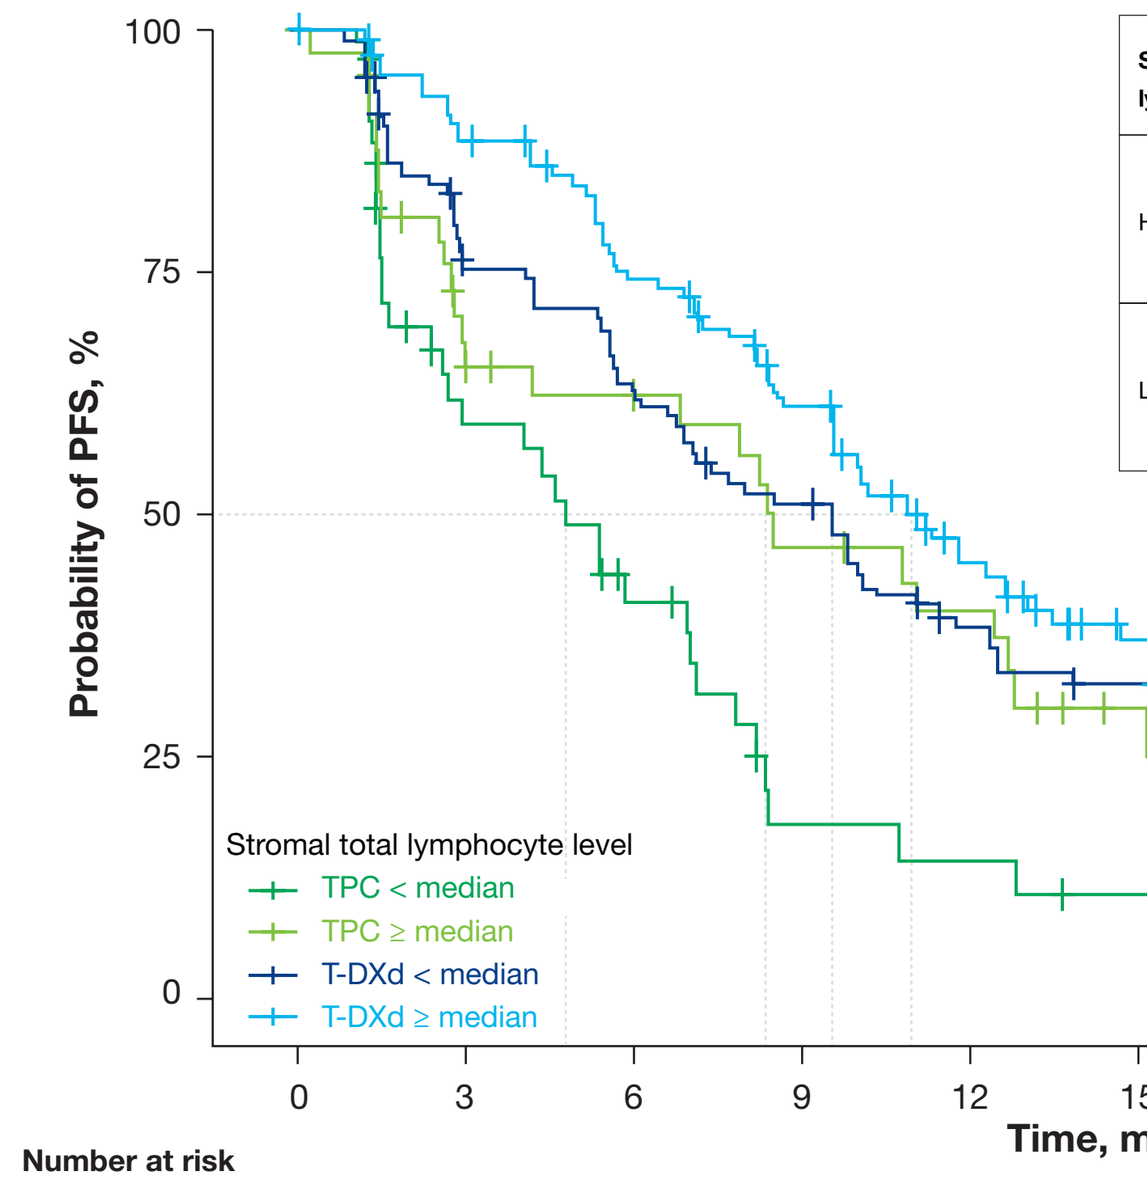

Probability of PFS, %
Stromal total lymphocyte level
TPC < median
TPC ≥ median
T-DXd < median
T-DXd ≥ median
Time, months

Stromal total lymphocyte level	Treatment arm (n)	mPFS (95% CI), months	Hazard ratio (95% CI)	Interaction *P* value
High (≥ median)	T-DXd (110)	10.95 (9.57-12.99)	0.70 (0.45-1.10)	0.149
	TPC (44)	8.32 (2.96-12.66)		
Low (< median)	T-DXd (105)	9.53 (6.74-11.41)	0.46 (0.31-0.70)	
	TPC (49)	4.77 (2.60-7.00)		

Number at risk	0	3	6	9	12	15	18	21	24	27	30
TPC < median	49	23	14	5	4	2	1	0	0	0	0
TPC ≥ median	44	24	20	15	12	6	1	0	0	0	0
T-DXd < median	105	75	62	50	33	27	17	8	5	0	0
T-DXd ≥ median	110	95	78	60	39	23	8	4	2	1	0

Median PFS was estimated by the Kaplan-Meier method, and the hazard ratio was calculated comparing the T-DXd and TPC arms in each biomarker-derived subgroup.

Abbreviations

cORR, confirmed objective response rate; ctDNA, circulating tumor DNA; DDR, DNA damage repair; HER2, human epidermal growth factor receptor 2; HRR, homologous recombination repair; HR+, hormone receptor–positive; IHC, immunohistochemistry; ISH, in situ hybridization; ITT, intention-to-treat; mPFS, median PFS; NA, not available; ND, not detected; NK, natural killer; PFS, progression-free survival; RNA seq, RNA sequencing; TPC, treatment of physician's choice; T-DXd, trastuzumab deruxtecan.

Acknowledgments

Under the guidance of authors and in accordance with Good Publication Practice, medical writing and editorial support was provided by Sara Duggan, PhD, of ApotheCom, funded by Daiichi Sankyo, Inc.

Disclosures

Dr Ueno holds consulting roles, speaker or preceptorship roles, and research agreements with AstraZeneca plc, Bayer AG, Pfizer Inc., Gilead Sciences, Inc., Chugai Pharmaceutical Co., CytoDyn Inc., Daiichi Sankyo, Inc., DynaMed, LLC, Eisai Co., Ltd., KeChow Pharma, Inc., Lavender Health Ltd., OBI Pharma Inc., OncoCyte Co., Ourotech, Inc., DBA Pear Bio, Kirilys Therapeutics, Inc., Peptilogics, Inc., Phoenix Molecular Designs, Preferred Medicine, Inc., Puma Biotechnology, Inc., Sumitomo Dainippon Pharma, Inc., Sysmex Co. Ltd., Takeda Pharmaceuticals, Inc., Unitech Medical, Inc., CARNA Biosciences, Inc., ChemDiv, Inc., DualityBio, LARVOL, Oncolys BioPharma Inc., Rakuten Medical, Inc., Merck Co., AnHeart Therapeutics Inc., Carisma Therapeutics, Inc., Lilly, Inc. and Therimunex.

References

1. Mosele MF et al. *Ann Oncol.* 2022;Vol 33:S1440.
2. Ogitani Y et al. *Clin Cancer Res.* 2016;22:5097-5108.
3. Ogitani Y et al. *Cancer Sci.* 2016;107:1039-1046.
4. Daiichi Sankyo Inc. ENHERTU® (fam-trastuzumab deruxtecan-nxki) for injection, for intravenous use. 2024.
5. Daiichi Sankyo Europe GmbH. Enhertu 100 mg powder for concentrate for solution infusion (SPC). 2024.
6. Modi S et al. *N Engl J Med.* 2022;387:9-20.
7. Modi S et al. *J Clin Oncol.* 2023;41:1020.
8. Nakada T et al. *Chem Pharm Bull.* 2019;67:173-185.
9. Thomas A, Pommier Y. *Clin Cancer Res.* 2019;25:6581-6589.
10. Ríos-Hoyo A et al. *J Clin Oncol.* 2024. DOI: 10.1200/jco.23.02614.
11. Barbie DA et al. *Nature.* 2009;462:108-112.
12. Liberzon A et al. *Cell Systems.* 2015;1:417-425.
13. Carr TH et al. *Cancers (Basel).* 2021;13:5830.

DESTINY-Breast04