Impact of differences in body mass index, body surface area and lean body mass on clinical outcomes in patients with atrial fibrillation receiving edoxaban: 4-year follow-up data from ETNA-AF-Europe

Giuseppe Boriani[1], Liliana Zaremba-Pechman[2], Richa Chhabra[2], Raffaele De Caterina[3] on behalf of the ETNA-AF-Europe investigators

[1]Cardiology Division, Department of Biomedical, Metabolic and Neural Sciences, University of Modena and Reggio Emilia, Modena, Italy; [2]Daiichi Sankyo Europe GmbH, Munich, Germany;
[3]University of Pisa, and Cardiology Division, Pisa University Hospital, 56124 Pisa, Italy.

BACKGROUND

- In patients with atrial fibrillation (AF) treated with DOACs (direct oral anticoagulants), extremes in body weight and its anthropometric correlates ─ body mass index (BMI), body surface area (BSA) and lean body mass (LBM) ─ may affect drug exposure and the occurrence of thromboembolic or haemorrhagic events.[1]
- While there have been suggestions to avoid DOAC use in patients with obesity >120 kg or with a BMI >40 kg/m^2 over concerns of reduced drug exposure and risk of under-dosing; thus far, there have been no recommendations related to the use of DOACs in patients with low body weight.[1,2]
- Studying the impact of body weight, anthropometric correlates and sex differences on clinical outcomes after treatment with a DOAC is crucial for optimising AF management and improving patient care.[3]

PURPOSE

- To analyse differences in clinical outcomes according to BMI, BSA and LBM categories, and sex, in edoxaban-treated patients with AF using 4-year data from ETNA-AF-Europe (NCT02944019).

METHODS

- ETNA-AF-Europe was a post-authorisation, prospective observational study conducted across 776 sites from 10 European countries, which assessed the risks and benefits of edoxaban use in patients with AF.[4]
- In this *post hoc* analysis, patients were categorised into tertiles (low, middle and high) for BMI, BSA and LBM (calculated using the Boer formula[5]) and stratified by sex.
- Patient demographics and characteristics were collected at baseline.
- Incidence of any thromboembolic and haemorrhagic events per 100 patient-years are reported using 4-year follow-up data.

RESULTS

Baseline characteristics

- Of 13,164 patients analysed, 7461 were male, and 5703 were female.
- **Table 1** shows the ranges of BMI, BSA and LBM, defining the low, middle and high tertiles, and their corresponding baseline characteristics.
- Irrespective of sex, patients in the low tertile for BMI, BSA and LBM were older and more likely to have renal impairment, history of any stroke (**Table 1**) and to receive recommended edoxaban 30 mg or non-recommended edoxaban 60 mg than other tertiles.
- Patients in the high tertile were more likely to have diabetes and hypertension (**Table 1**).

Clinical outcomes by BMI, BSA and LBM tertiles and by sex

- Overall, the rate of any thromboembolic event was slightly lower, whereas the corresponding rate of any haemorrhagic event was slightly higher in males versus females (**Figure 1**).
- For BMI, BSA and LBM in males, rates of any thromboembolic event were similar across tertiles (**Figure 1**).
 - — For BMI, rates of any haemorrhagic event were similar across tertiles in males, but highest in the low tertile for BSA and LBM
- In females, rates of any thromboembolic event were similar for all tertiles of BMI, but highest in the low tertile for BSA and LBM (**Figure 1**).
 - — For BMI, BSA and LBM, rates of any haemorrhagic event were highest in the low tertile in females

Table 1. Baseline characteristics according to BMI, BSA and LBM tertiles and stratified by sex

Baseline characteristics	Males				Females			
	Total	Low tertile	Middle tertile	High tertile	Total	Low tertile	Middle tertile	High tertile
BMI, kg/m^2, tertile range (n)	n.a (7461)	13.8, <25.9 (2332)	≥25.9, ≤29.4 (2525)	>29.4, 59.2 (2280)	n.a (5703)	14.7, <25.0 (1778)	≥25.0, ≤29.3 (1879)	>29.3, 68.6 (1813)
Age, median (IQR)	74 (67, 79)	76 (69, 81)	74 (67, 79)	71 (64, 77)	76 (70, 81)	78 (72, 83)	77 (71, 82)	74 (68, 79)
Any stroke, n (%)	455 (6.1)	152 (6.5)	165 (6.5)	110 (4.8)	357 (6.3)	121 (6.8)	119 (6.3)	86 (4.7)
Comorbidities, n (%)								
Diabetes mellitus	1735 (23.3)	397 (17.0)	540 (21.4)	732 (32.1)	1146 (20.1)	244 (13.7)	344 (18.3)	519 (28.6)
Hypertension	5649 (75.7)	1591 (68.2)	1949 (77.2)	1911 (83.8)	4506 (79.0)	1278 (71.9)	1510 (80.4)	1557 (85.9)
Renal impairment*	3751 (50.3)	1625 (69.7)	1294 (51.2)	674 (29.6)	3825 (67.1)	1465 (82.4)	1339 (71.3)	886 (48.9)
BSA, m^2, tertile range (n)	n.a (7461)	1.28, <1.93 (2358)	≥1.93, ≤2.09 (2403)	>2.09, ≤2.94 (2375)	n.a (5703)	1.24, <1.69 (1818)	≥1.69, ≤1.84 (1831)	>1.84, 2.84 (1821)
Age, median (IQR)	74 (67, 79)	77 (71, 82)	74 (68, 79)	69 (62, 75)	76 (70, 81)	79 (73, 84)	76 (71, 81)	73 (66, 78)
Any stroke, n (%)	455 (6.1)	166 (7.0)	152 (6.3)	109 (4.6)	357 (6.3)	132 (7.3)	104 (5.7)	90 (4.9)
Comorbidities, n (%)								
Diabetes mellitus	1735 (23.3)	473 (20.1)	550 (22.9)	645 (27.2)	1146 (20.1)	293 (16.1)	354 (19.3)	460 (25.3)
Hypertension	5649 (75.7)	1722 (73.0)	1839 (76.5)	1890 (79.6)	4506 (79.0)	1376 (75.7)	1473 (80.4)	1496 (82.2)
Renal impairment*	3751 (50.3)	1795 (76.1)	1254 (52.2)	543 (22.9)	3825 (67.1)	1559 (85.8)	1316 (71.9)	815 (44.8)
LBM, kg, tertile range (n)	n.a (7461)	36.1, <59.3 (2360)	≥59.3, ≤65.2 (2398)	>65.2, 107.5 (2378)	n.a (5703)	27.9, <44.2 (1817)	≥44.2, ≤49.0 (1836)	>49.0, 83.1 (1817)
Age, median (IQR)	74 (67, 79)	77 (71, 82)	74 (68, 79)	69 (62, 75)	76 (70, 81)	79 (74, 84)	76 (71, 81)	73 (66, 78)
Any stroke, n (%)	455 (6.1)	164 (6.9)	157 (6.5)	106 (4.5)	357 (6.3)	132 (7.3)	111 (6.0)	83 (4.6)
Comorbidities, n (%)								
Diabetes mellitus	1735 (23.3)	463 (19.6)	550 (22.9)	655 (27.5)	1146 (20.1)	323 (17.8)	359 (19.6)	425 (23.4)
Hypertension	5649 (75.7)	1715 (72.7)	1837 (76.6)	1899 (79.9)	4506 (79.0)	1417 (78.0)	1467 (79.9)	1461 (80.4)
Renal impairment*	3751 (50.3)	1796 (76.1)	1255 (52.3)	541 (22.8)	3825 (67.1)	1530 (84.2)	1318 (71.8)	842 (46.3)

*Renal impairment was defined as: Cockcroft-Gault formula ≤80 mL/min or CrCl ≤80 mL/min or investigator-reported renal disease (excluding Stage 1 chronic kidney disease) if these data or measurement of serum creatinine were not available.
BMI, body mass index; **BSA**, body surface area; **CrCl**, creatine clearance; **IQR**, interquartile range; **LBM**, lean body mass; **n.a**, not available

CONCLUSIONS

At the 4-year follow-up in this real-life registry, low rates of any thromboembolic event were observed regardless of patients' BMI, BSA, LBM or sex

BSA and LBM, rather than BMI, should be considered when analysing bleeding outcomes in AF patients treated with edoxaban as higher bleeding rates were seen in the low versus middle and high tertiles for BSA and LBM

Figure 1. Clinical outcomes according to BMI, BSA and LBM tertiles and stratified by sex

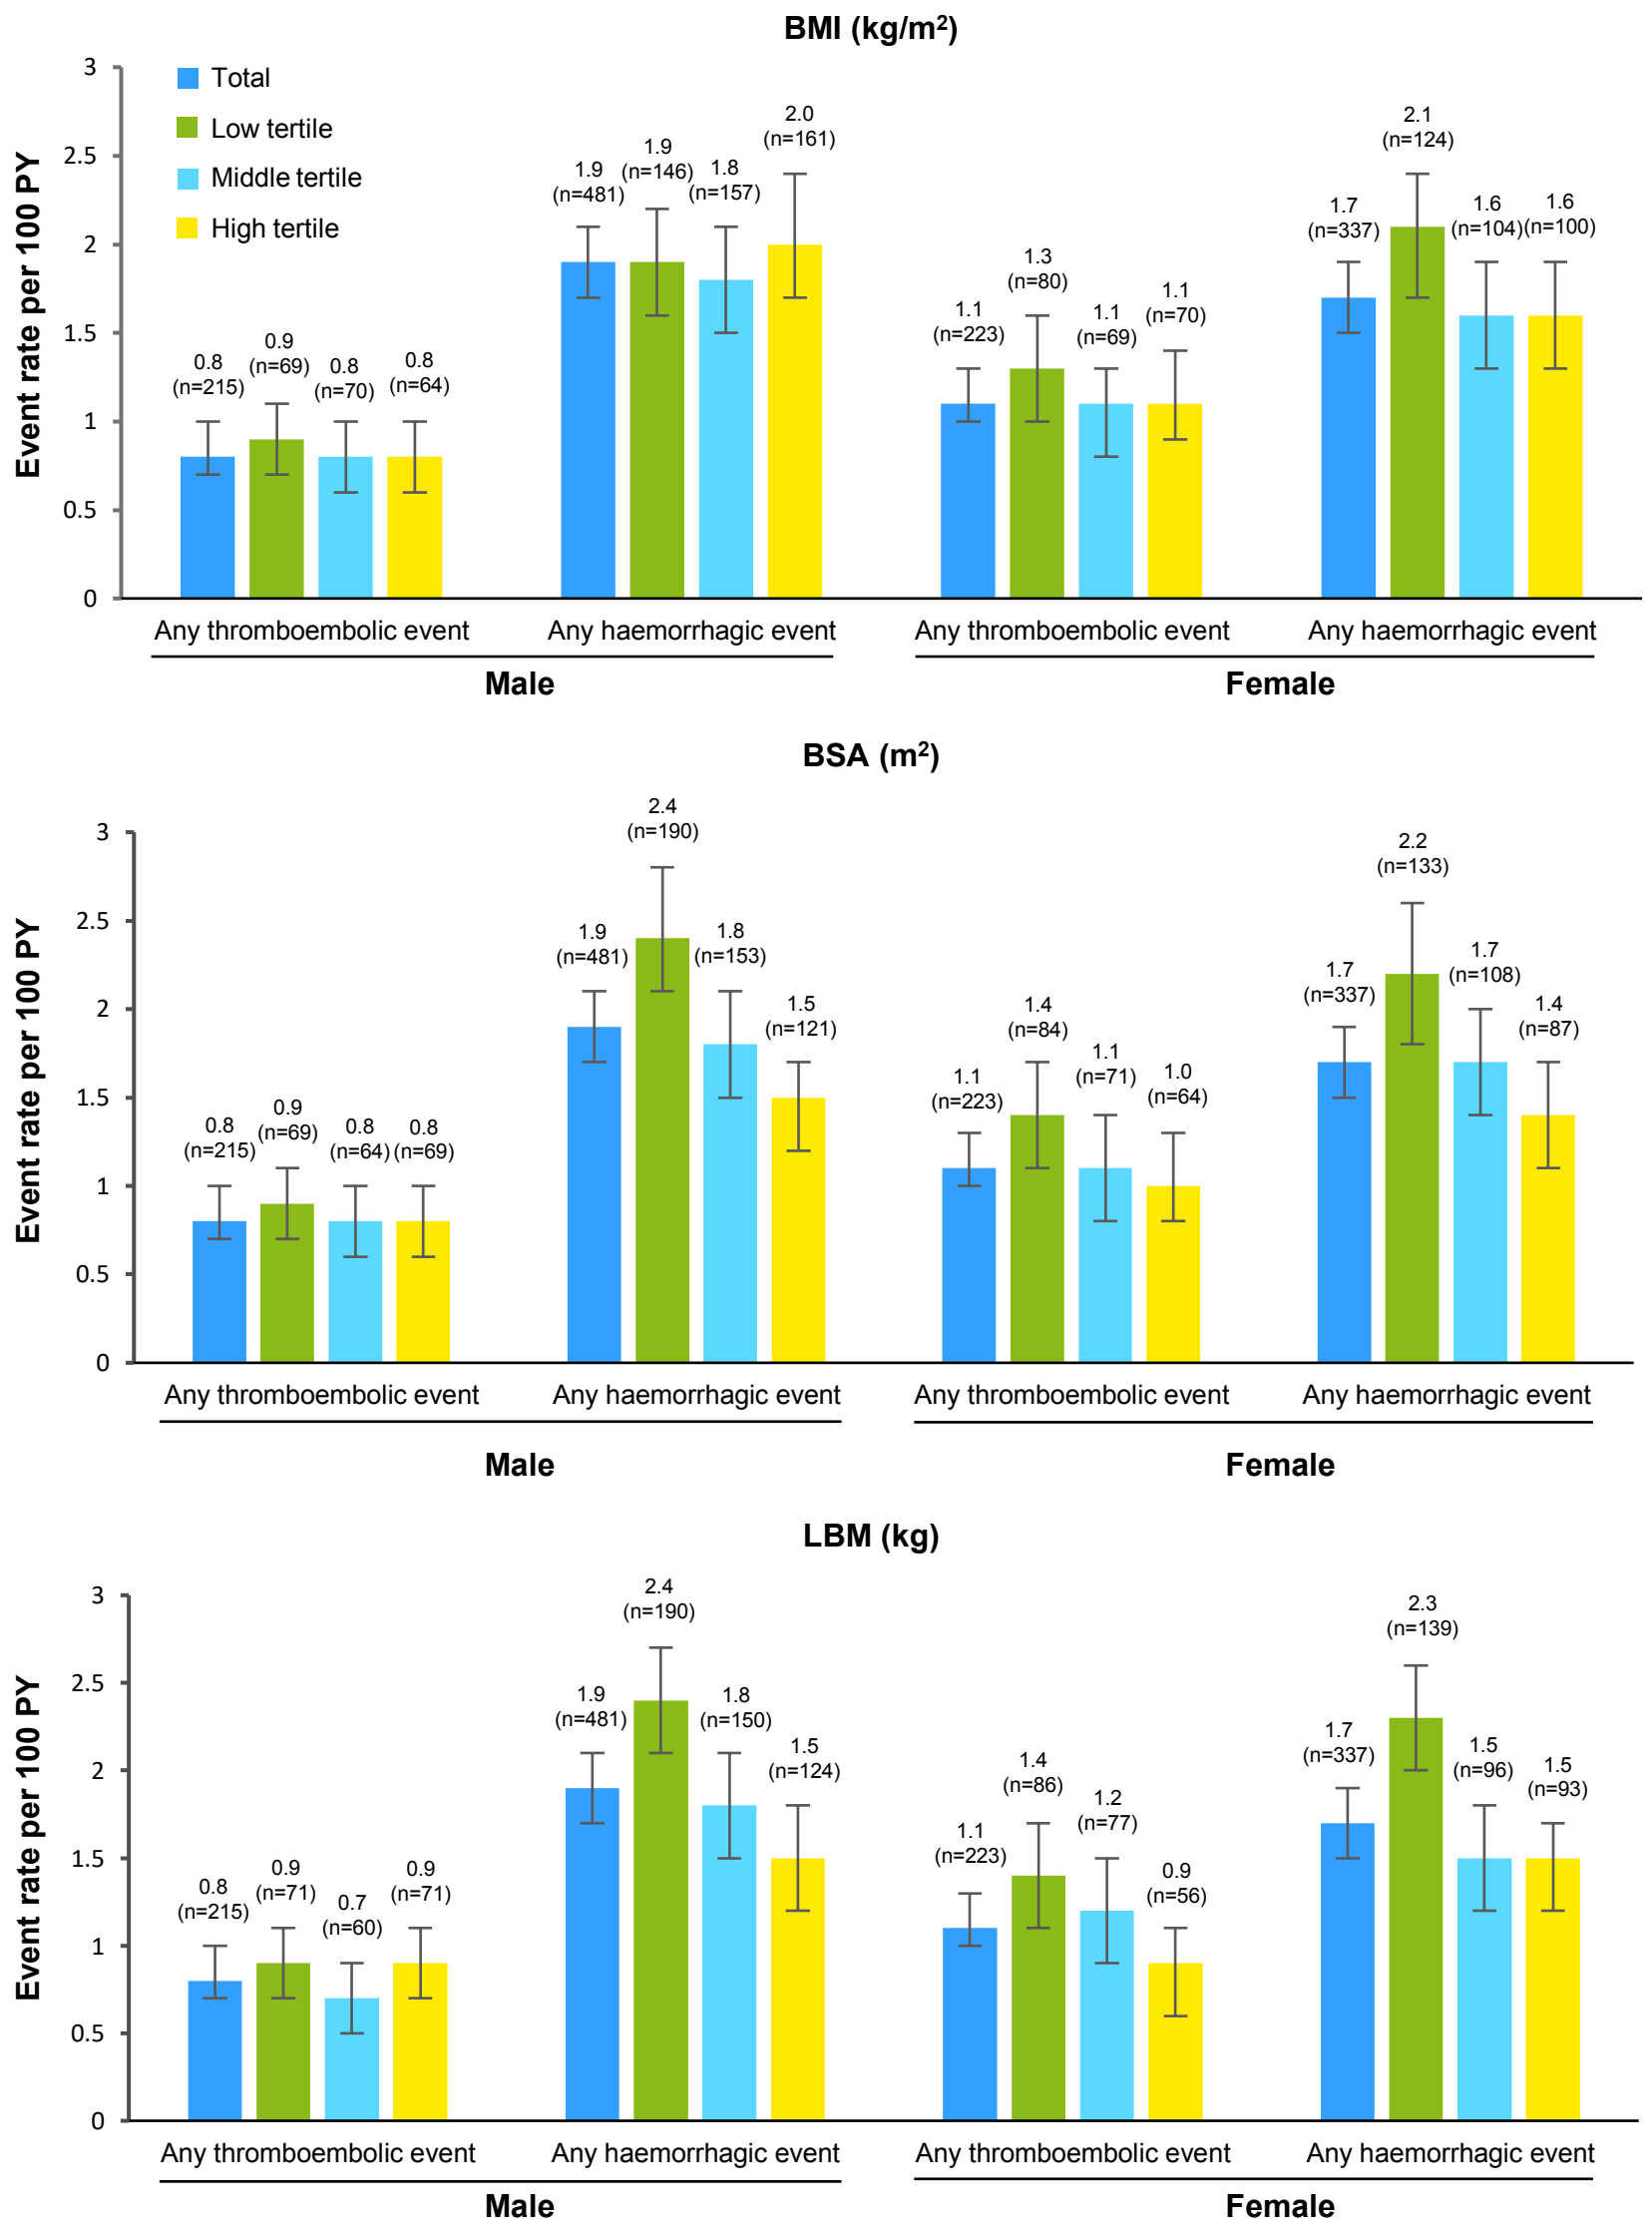
BMI (kg/m^2)
Total
Low tertile
Middle tertile
High tertile
Event rate per 100 PY
0.8 (n=215)
0.9 (n=69)
0.8 (n=70)
0.8 (n=64)
1.9 (n=481)
1.9 (n=146)
1.8 (n=157)
2.0 (n=161)
1.1 (n=223)
1.3 (n=80)
1.1 (n=69)
1.1 (n=70)
1.7 (n=337)
2.1 (n=124)
1.6 (n=104)
1.6 (n=100)
Any thromboembolic event
Any haemorrhagic event
Male
Female
BSA (m^2)
0.8 (n=215)
0.9 (n=69)
0.8 (n=64)
0.8 (n=69)
1.9 (n=481)
2.4 (n=190)
1.8 (n=153)
1.5 (n=121)
1.1 (n=223)
1.4 (n=84)
1.1 (n=71)
1.0 (n=64)
1.7 (n=337)
2.2 (n=133)
1.7 (n=108)
1.4 (n=87)
LBM (kg)
0.8 (n=215)
0.9 (n=71)
0.7 (n=60)
0.9 (n=71)
1.9 (n=481)
2.4 (n=190)
1.8 (n=150)
1.5 (n=124)
1.1 (n=223)
1.4 (n=86)
1.2 (n=77)
0.9 (n=56)
1.7 (n=337)
2.3 (n=139)
1.5 (n=96)
1.5 (n=93)

n is the number of events. 95% CIs for event rates were calculated using a normal approximation method. A thromboembolic event was defined as an adjudicated ischaemic stroke, venous thromboembolism, adjudicated systemic embolic event or transient ischaemic attack. A haemorrhagic event was defined as adjudicated intracranial bleeding, adjudicated major bleeding or adjudicated clinically relevant non-major bleeding
BMI, body mass index; **BSA**, body surface area; **CI**, confidence interval; **LBM**, lean body mass; **PY**, patient years

Presented at: European Society of Cardiology (ESC) 2024, London, UK, 30 August ─ 2 September 2024

REFERENCES

1. Rocca B, et al. Eur Heart J. 2018;39:1672–86
2. Martin K, et al. *Thromb Haemost.* 2016;14:1308–13
3. Nwanosike EM, et al. Eur J Clin Pharmacol. 2024;163–73
4. Kirchhof P, et al. *Int J Cardiol.* 2024;132118
5. Boer P. *Am J Physiol-Renal Physiol.* 1984;247:F632

ACKNOWLEDGEMENTS

This ePoster was sponsored by Daiichi Sankyo Europe GmbH, Munich, Germany.
Writing and editorial support were provided by Kathryn Ball from inScience Communications, Springer Healthcare Ltd, UK, and funded by Daiichi Sankyo Europe GmbH, Munich, Germany