Edoxaban dose, frailty, and outcomes in patients with atrial fibrillation: the ETNA-AF-Europe 4-year follow-up

Stefano Fumagalli[1], Igor Diemberger[2], Joris J Komen[3], Eva-Maria Fronk[3], Paulus Kirchhof[4], Raffaele De Caterina[5] on behalf of the ETNA-AF-Europe investigators

[1]Department of Experimental and Clinical Medicine, University of Florence, Florence, Italy; [2]Department of Medical and Surgical Sciences, University of Bologna, Bologna, Italy; [3]Daiichi Sankyo Europe GmbH, Munich, Germany; [4]Vascular Center Hamburg, Department of Cardiology, Hamburg, Germany; [5]University of Pisa, and Cardiology Division, Pisa University Hospital, 56124 Pisa, Italy

BACKGROUND

- Older people with atrial fibrillation (AF) are often also frail, which is associated with an increased stroke and bleeding risk.[1,2]
- Frailty is a common reason to choose non-recommended doses of direct oral anticoagulants (DOACs) in AF patients.[3]
- However, how this impacts stroke and bleeding outcomes is currently unknown.

PURPOSE

- To assess clinical outcomes in frail patients with AF receiving non-recommended versus recommended doses of edoxaban using 4-year follow-up data from ETNA-AF-Europe (NCT02944019).

METHODS

- The prospective, observational ETNA-AF-Europe study enrolled patients from 776 sites across 10 European countries, which assessed the risks and benefits of edoxaban use in patients with AF.[4]
- In this subanalysis, data for patients with perceived or objective frailty were combined.
 — **Perceived frailty** was based on investigators' own clinical binary judgement for each patient
 — **Objective frailty** was determined using a simplified adaptation of the Rockwood's Frailty Index. Patients with a missing index were categorised as non-frail
- Baseline characteristics and hazard ratios (HRs) with 95% confidence intervals (CIs) are reported that assessed risk of outcomes in frail patients prescribed non-recommended versus recommended edoxaban doses.
- Data were adjusted for age, sex and derived versions of the CHA_2DS_2-VASc and HAS-BLED scores.
- Net clinical benefit was defined as any stroke/systemic embolic event (SEE), transient ischaemic attack, venous thromboembolic event, major bleeding or cardiovascular death, whichever came first.

RESULTS

Patient demographics and baseline characteristics

- Of 13,164 patients, 1786 were frail (13.6%):
 — Patients with perceived (n=1410) versus objective (n=540) frailty were older (median age [interquartile range]: 82.0 years [78.0–86.0]) years versus 77.0 years (71.0–82.0) and more often female (57.9% versus 31.9%)
 — Overall, 164 patients had both perceived and objective frailty
- Frail patient's baseline characteristics, stratified according to recommended or non-recommended doses of edoxaban received, are reported in **Table 1**.
- Age categories within each recommendation group are presented in **Figure 1**.
 — With the exception of the recommended edoxaban 60 mg group, the proportion of frail patients was higher in the older (>80 years) versus younger (≤74 years or 75–80 years) age categories (**Figure 1**)

Clinical outcomes in frail patients according to non-recommended versus recommended edoxaban doses

- Clinical outcomes in frail patients receiving non-recommended edoxaban 30 mg versus recommended 60 mg and non-recommended edoxaban 60 mg versus recommended 30 mg are summarised in **Figure 2**.
- Risk of all-cause death was higher in frail patients treated with non-recommended 30 mg (n=169) versus recommended 60 mg (n=622) (**Figure 2a**).
 — The annualised rate of any stroke/SEE was not significantly higher with non-recommended 30 mg versus recommended 60 mg doses
 — There was no association between treatment dose received (non-recommended 30 mg versus recommended 60 mg) and risk of major bleeding or net clinical benefit
- Patients who received non-recommended 60 mg (n=183) versus recommended 30 mg (n=695; **Figure 2b**) doses had no significant association with major bleeding, net clinical benefit or all-cause death, and they demonstrated a higher risk of any stroke or SEE.

Table 1. Patient demographics and baseline characteristics

	Frail patients (objective or perceived frailty)			
	Recommended edoxaban 60 mg (n=622)	Non-recommended edoxaban 60 mg (n=183)	Recommended edoxaban 30 mg (n=695)	Non-recommended edoxaban 30 mg (n=169)
Male	404 (65.0)	68 (37.2)	236 (34.0)	92 (54.4)
Age, years, median (IQR)	78.0 (72.0, 82.0)	81.0 (76.0, 86.0)	84.0 (80.0, 88.0)	80.0 (75.0, 83.0)
Weight, kg, median (IQR)	81.0 (74.0, 93.0)	65.0 (59.0, 75.0)	64.0 (55.0, 75.0)	80.0 (73.0, 87.0)
Derived CrCl, mL/min, median (IQR)	72.5 (61.0, 87.5)	45.4 (39.4, 52.9)	40.2 (33.6, 45.6)	59.7 (54.5, 71.6)
CHA_2DS_2-VASc score, median (IQR)	4.0 (3.0, 5.0)	4.0 (4.0, 5.0)	4.0 (4.0, 6.0)	4.0 (3.0, 5.0)
HAS-BLED score, median (IQR)	3.0 (2.0, 3.0)	3.0 (3.0, 4.0)	3.0 (3.0, 4.0)	3.0 (3.0, 3.0)
Diabetes mellitus	231 (37.1)	60 (32.8)	200 (28.8)	47 (27.8)
Hypertension	543 (87.3)	156 (85.2)	582 (83.7)	142 (84.0)
Congestive heart failure	216 (34.7)	57 (31.1)	240 (34.5)	59 (34.9)
Chronic obstructive pulmonary disease	130 (20.9)	36 (19.7)	107 (15.4)	30 (17.8)
History of stroke/TIA/SEE	185 (29.7)	45 (24.6)	142 (20.4)	22 (13.0)
History of major bleeding	8 (1.3)	4 (2.2)	23 (3.3)	6 (3.6)

Data are n (%) unless otherwise stated
CHA_2DS_2-VASc, heart failure, hypertension, age (≥65 years =1 point, ≥75 years =2 points), diabetes, prior stroke/transient ischaemic attack/SEE (2 points), vascular disease, and female sex category; **CrCl**, creatinine clearance; **HAS-BLED,** hypertension, abnormal renal/liver function, stroke, bleeding history or predisposition, elderly (aged >65 years), drugs/alcohol concomitantly; **IQR**, interquartile range; **SD**, standard deviation; **SEE**, systemic embolic event; **TIA**, transient ischaemic attack

CONCLUSIONS

In this large European AF registry, the use of the non-recommended 30 mg (reduced) dose of edoxaban was associated with higher all-cause death and no benefits regarding the risk of major bleeding in frail patients

According to our results, the presence of frailty should not drive changes from the dosing recommendations for edoxaban

Figure 1. Age category of frail patients receiving non-recommended and recommended 60 mg or 30 mg edoxaban doses

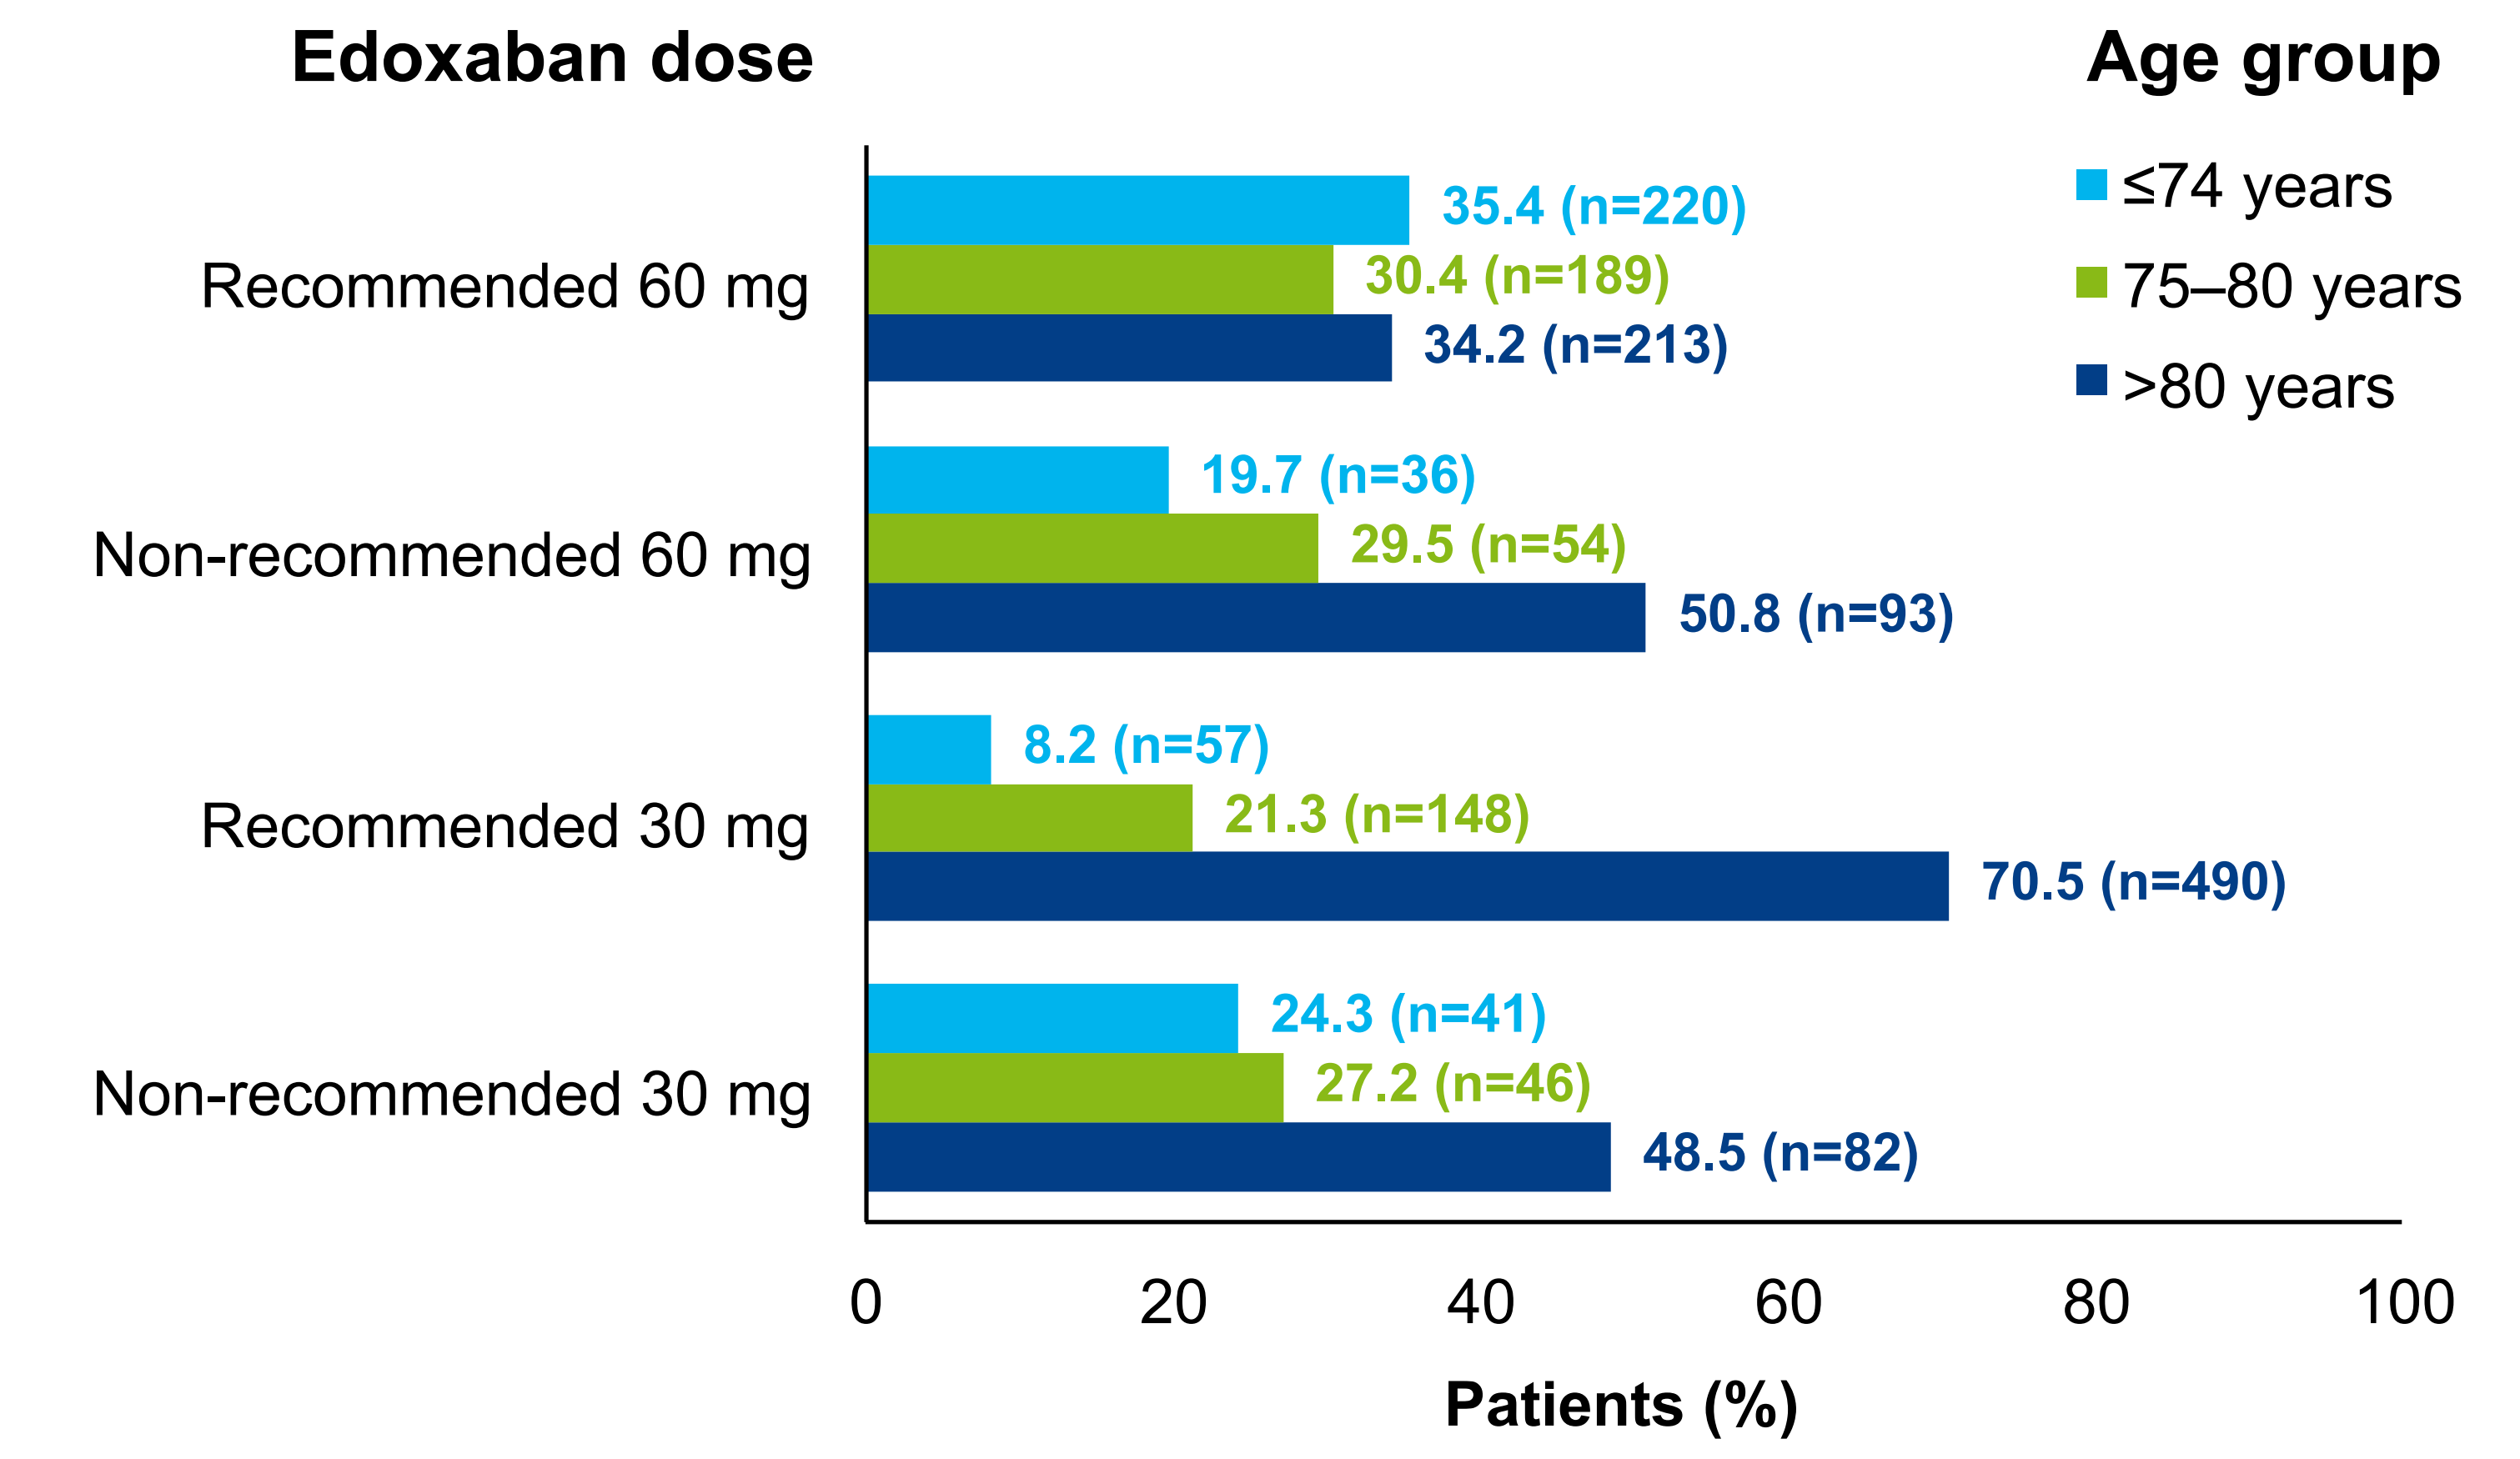
Edoxaban dose
Age group
≤74 years
75–80 years
>80 years
Recommended 60 mg
35.4 (n=220)
30.4 (n=189)
34.2 (n=213)
Non-recommended 60 mg
19.7 (n=36)
29.5 (n=54)
50.8 (n=93)
Recommended 30 mg
8.2 (n=57)
21.3 (n=148)
70.5 (n=490)
Non-recommended 30 mg
24.3 (n=41)
27.2 (n=46)
48.5 (n=82)
0
20
40
60
80
100
Patients (%)

Figure 2. Clinical outcomes in frail patients receiving a) non-recommended edoxaban 30 mg versus recommended 60 mg or b) non-recommended edoxaban 60 mg versus recommended 30 mg

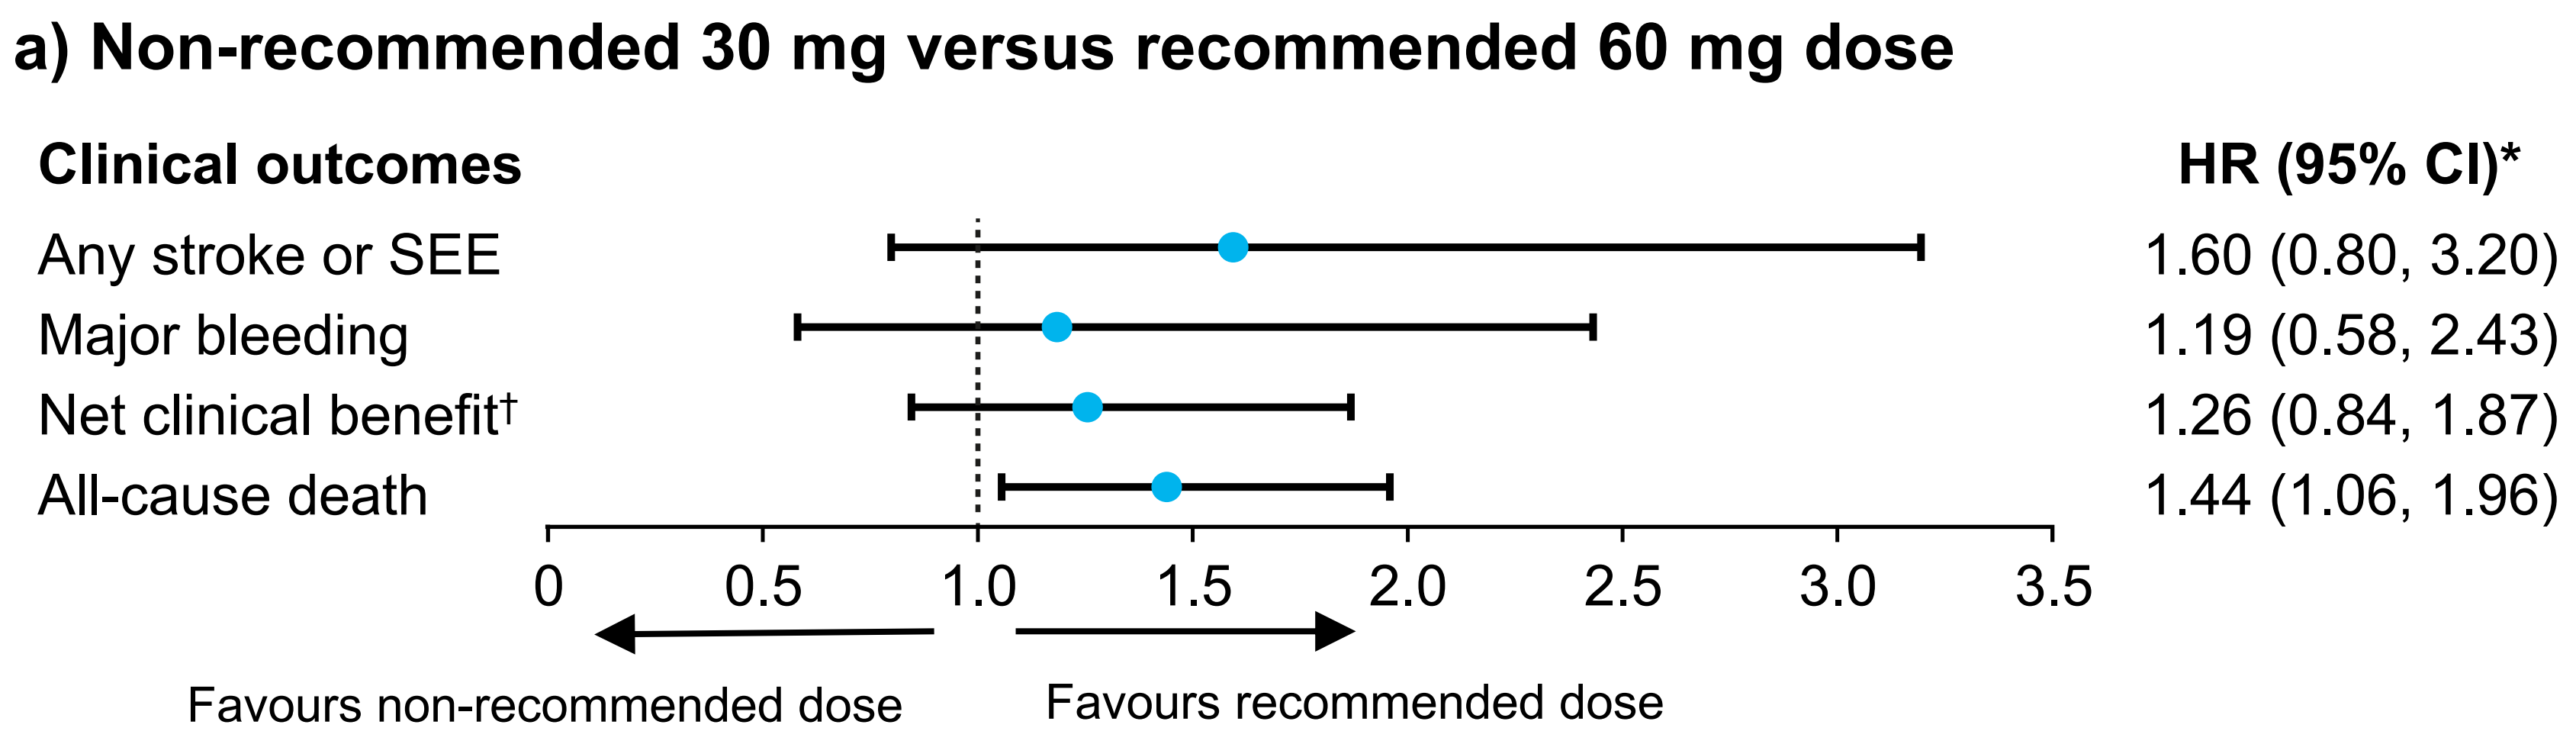
a) Non-recommended 30 mg versus recommended 60 mg dose
Clinical outcomes
HR (95% CI)*
Any stroke or SEE
1.60 (0.80, 3.20)
Major bleeding
1.19 (0.58, 2.43)
Net clinical benefit†
1.26 (0.84, 1.87)
All-cause death
1.44 (1.06, 1.96)
0
0.5
1.0
1.5
2.0
2.5
3.0
3.5
Favours non-recommended dose
Favours recommended dose

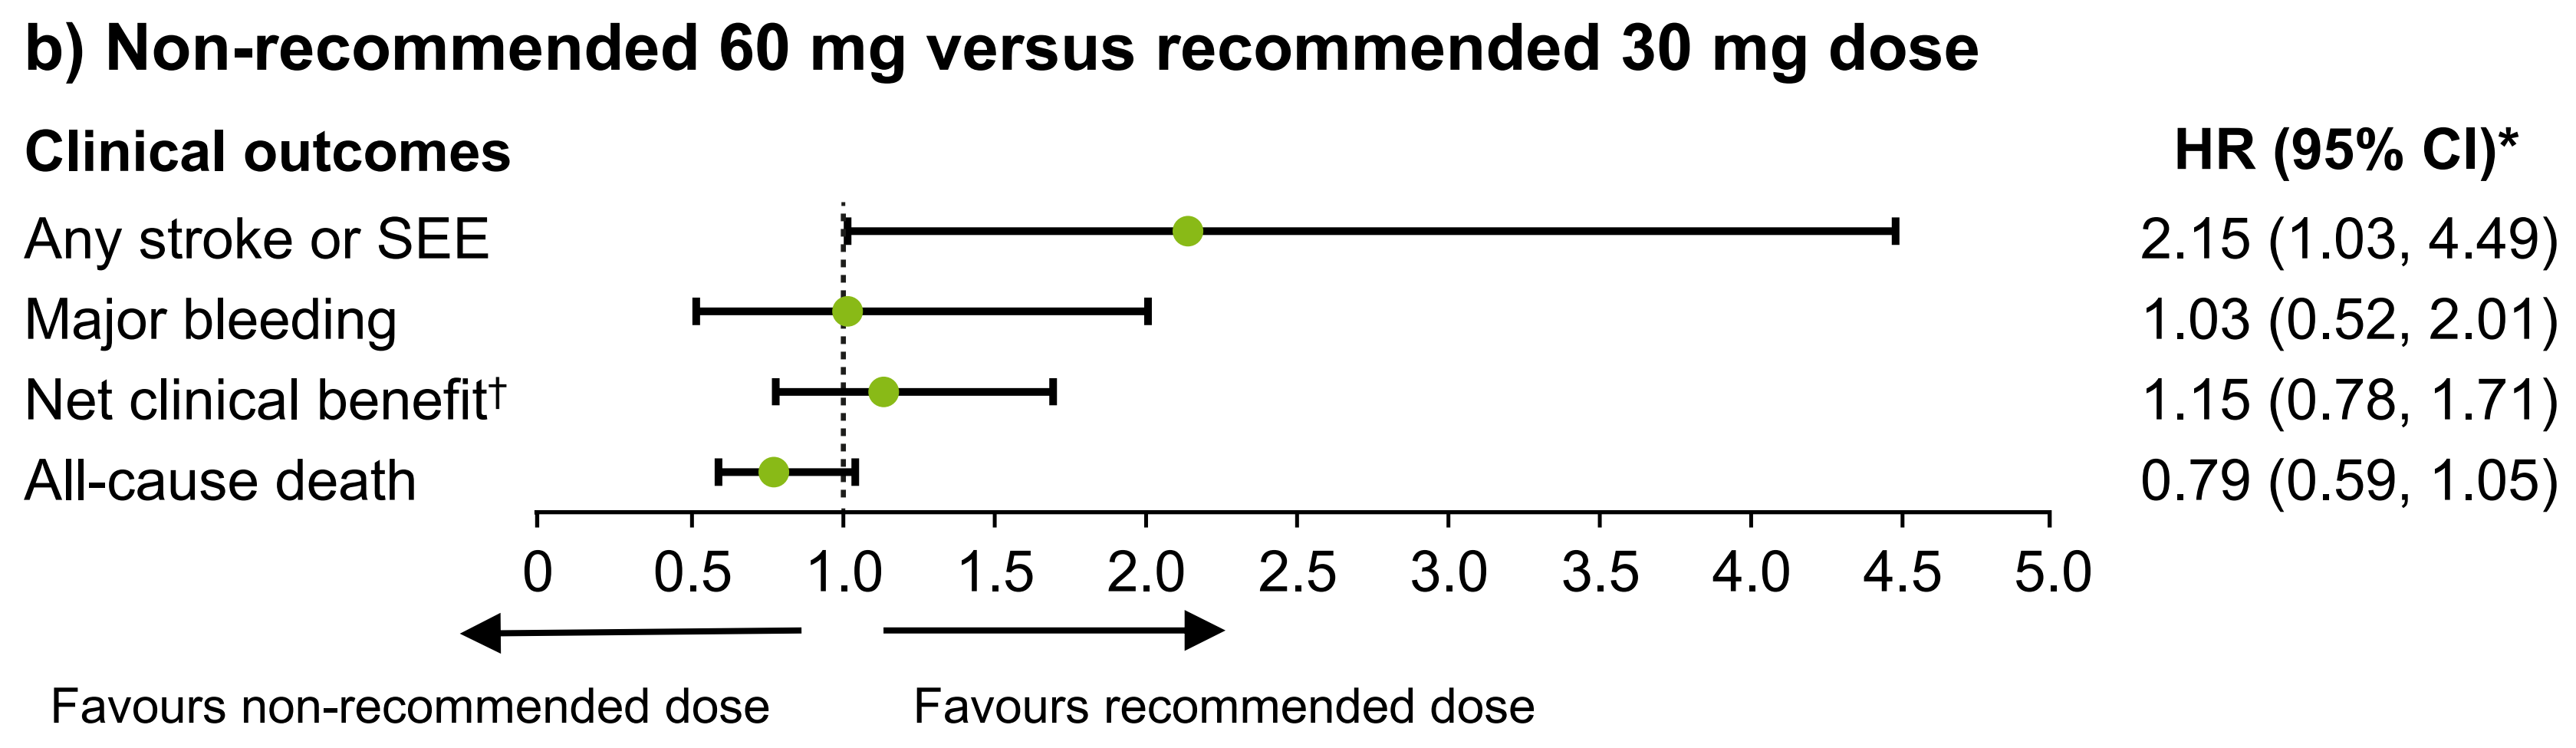
b) Non-recommended 60 mg versus recommended 30 mg dose
Clinical outcomes
HR (95% CI)*
Any stroke or SEE
2.15 (1.03, 4.49)
Major bleeding
1.03 (0.52, 2.01)
Net clinical benefit†
1.15 (0.78, 1.71)
All-cause death
0.79 (0.59, 1.05)
0
0.5
1.0
1.5
2.0
2.5
3.0
3.5
4.0
4.5
5.0
Favours non-recommended dose
Favours recommended dose

*HRs (95% CIs) calculated from adjusted Cox-regression model. The model includes dose recommendation, frailty and the corresponding interaction term, as well as age, sex and derived versions of the CHA2DS2-VASc and HAS-BLED scores (**Table 1**) as additional covariates; †defined as any stroke/SEE, transient ischaemic attack, venous thromboembolic event, major bleeding or cardiovascular death, whichever came first.
CI, confidence interval; **HR**, hazard ratio; **SEE**, systemic embolic event

Presented at: European Society of Cardiology (ESC) 2024, London, UK, 30 August — 2 September 2024

REFERENCES

1. Wilkinson C, et al. *Age Ageing*. 2019;48:196–203
2. Steffel J, et al. *Europace*. 2021;23:1612–76
3. Diemberger I, et al. *Europace*. 2022;24:1404–11
4. Kirchhof P, et al. *Int J Cardiol.* 2024;408:132118

ACKNOWLEDGEMENTS

This ePoster was sponsored by Daiichi Sankyo Europe GmbH, Munich, Germany.
Writing and editorial support were provided by Aoife Egan from inScience Communications, Springer Healthcare Ltd, UK, and funded by Daiichi Sankyo Europe GmbH, Munich, Germany

ETNA-AF Europe