Valemetostat Monotherapy in Patients With Relapsed/Refractory Non-Hodgkin Lymphomas: Primary Results From a Phase 1 Trial

TCL-391

Dai Maruyama[1], Pierluigi Porcu[2], Kensei Tobinai[1], Pamela B. Allen[3], Kenji Ishitsuka[4], Kunihiro Tsukasaki[5], Shigeru Kusumoto[6], Tomoko Narita[6], Francine Foss[7], Nobuhiko Yamauchi[8], Junichiro Yuda[8], Satoko Morishima[9], Yoshitaka Imaizumi[10,11], Koji Izutsu[1], Tatyana Feldman[12], Toyotaka Kawamata[13,14], Yasuyuki Kakurai[15], Hideyuki Yamauchi[15], Noha Biserna[16], Ai Inoue[16], Masaya Tachibana[15], Yoshiyuki Hizukuri[16], Steven M. Horwitz[17], Eric Jacobsen[18]

[1]National Cancer Center Hospital, Tokyo, Japan; [2]Department of Medical Oncology, Thomas Jefferson University, Philadelphia, PA, US; [3]Emory University, Atlanta, GA, US; [4]Department of Hematology and Rheumatology, Kagoshima University Hospital, Kagoshima, Japan; [5]Department of Hematology, International Medical Center, Saitama Medical University, Saitama, Japan; [6]Nagoya City University Graduate School of Medical Sciences, Nagoya, Japan; [7]Yale University School of Medicine, New Haven, CT, US; [8]National Cancer Center Hospital East, Chiba, Japan; [9]University of the Ryukyus Hospital, Okinawa, Japan; [10]Department of Hematology, Nagasaki University Hospital, Nagasaki, Japan; [11]Department of Hematology, National Hospital Organization Nagasaki Medical Center, Nagasaki, Japan; [12]John Theurer Cancer Center at Hackensack Meridian Health School of Medicine, Hackensack, NJ, US; [13]Department of Hematology/Oncology, The Institute of Medical Science, University of Tokyo, Tokyo, Japan; [14]Department of Hematology, Tokyo Metropolitan Bokutoh Hospital, Tokyo, Japan; [15]Daiichi Sankyo Co., Ltd., Tokyo, Japan; [16]Daiichi Sankyo Inc., Basking Ridge, NJ, US; [17]Memorial Sloan Kettering Cancer Center, New York, NY, US; [18]Dana-Farber Cancer Institute, Boston, MA, US

PURPOSE

- To describe the safety, tolerability, and preliminary efficacy of valemetostat tosylate (valemetostat) in patients with relapsed/refractory (R/R) non-Hodgkin lymphomas (NHLs) enrolled in the DS3201-A-J101 ("J101"; NCT02732275) trial

CONCLUSIONS

- Valemetostat monotherapy showed promising clinical activity in patients with R/R peripheral T-cell lymphoma (PTCL), R/R adult T-cell leukemia/lymphoma (ATLL), and R/R B-cell NHL (B-NHL)
 - Valemetostat induced durable responses, with median durations of response (DORs) of > 1.5 years (y) in all three NHL subtypes
- Valemetostat demonstrated acceptable safety and tolerability
 - Treatment-emergent adverse events (TEAEs) were generally manageable; cytopenias were common but did not require treatment discontinuation
- Ongoing trials of valemetostat in patients with R/R PTCLs include the phase 2 monotherapy VALENTINE-PTCL01 trial (NCT04703192)

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without written permission of the authors.

BACKGROUND

- Treatment options are limited and prognosis is often poor for patients with R/R NHLs
- Enhancer of zeste homolog (EZH)2 and its close homolog EZH1 catalyze the trimethylation of histone H3 at lysine 27 (H3K27me3), leading to transcriptional repression[1,2]
- Valemetostat is a novel, potent, and selective dual inhibitor of EZH2 and EZH1 that suppresses aberrant H3K27me3, thereby promoting antitumorigenic processes[2–4]
 - Valemetostat shows broad-spectrum antitumor activity in preclinical models of NHL[3,5,6]
- Valemetostat monotherapy is approved in Japan for the treatment of R/R ATLL[7,8]
- Here, we report primary outcomes for patients with R/R NHLs treated with valemetostat monotherapy in the J101 trial

METHODS

Study design

- Adult patients eligible for participation in the J101 trial had confirmed B-NHLs or T-cell NHLs (T-NHLs) as defined by the World Health Organization 2016 classification criteria and had relapsed from, were refractory to, or ineligible for standard therapies
- The trial included a dose-escalation part followed by a dose-expansion part (**Figure 1**)
- Patients with B- or T-NHLs were enrolled in the dose-escalation part and received valemetostat once daily at doses of 150–300 mg in continuous 28-day treatment cycles; in the dose-expansion part, patients with T-NHLs received valemetostat at the recommended dose for expansion (RDE) of 200 mg

Efficacy analysis

- Efficacy assessments included objective response rate (ORR), complete response (CR) rate, and DOR based on the International Working Group 2007 response criteria

H3K27me3 analysis

- H3K27me3 levels were assessed in normal granulocytes via flow cytometry
 - Whole blood was collected before dosing on day 1 of cycle 1 (C1D1), on C2D1 ± 5 days (Japan), or on or after C2D1 (US), and at end of treatment in patients in Japan who discontinued before completion of cycle 1

Figure 1. Study design, key inclusion criteria, and endpoints of the J101 trial

DS3201-A-J101 ("J101"; NCT02732275): Multicenter, single-arm, phase 1, dose-escalation and -expansion trial of valemetostat in R/R B- or T-NHLs

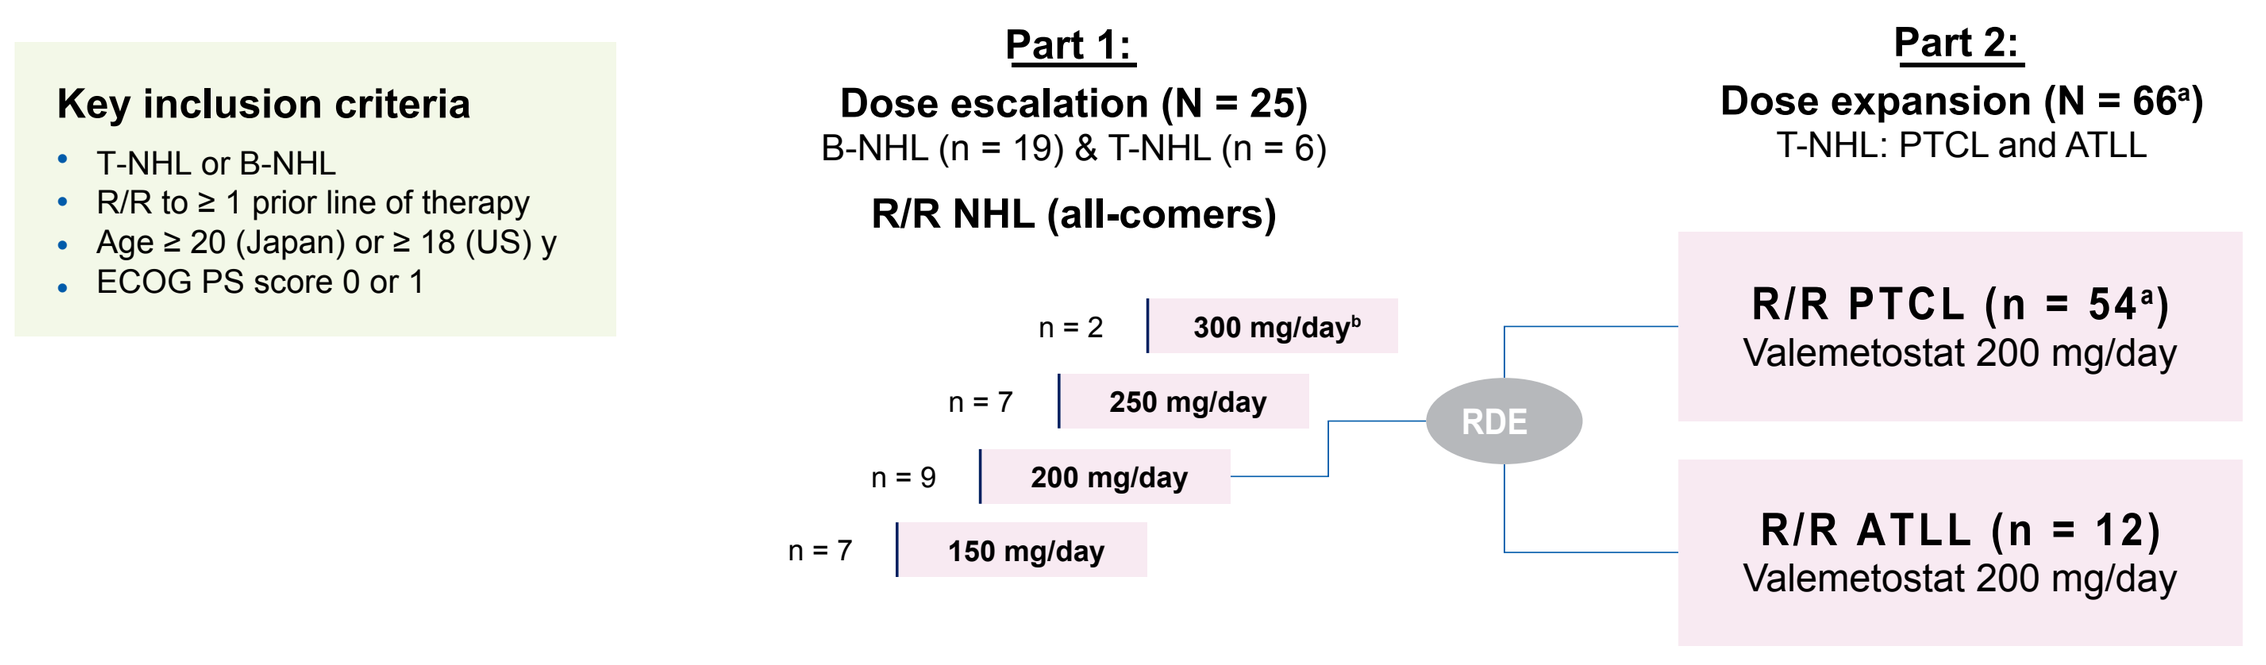
Key inclusion criteria
T-NHL or B-NHL
R/R to ≥ 1 prior line of therapy
Age ≥ 20 (Japan) or ≥ 18 (US) y
ECOG PS score 0 or 1
Part 1:
Dose escalation (N = 25)
B-NHL (n = 19) & T-NHL (n = 6)
R/R NHL (all-comers)
n = 2
300 mg/day[b]
n = 7
250 mg/day
n = 9
200 mg/day
n = 7
150 mg/day
RDE
Part 2:
Dose expansion (N = 66[a])
T-NHL: PTCL and ATLL
R/R PTCL (n = 54[a])
Valemetostat 200 mg/day
R/R ATLL (n = 12)
Valemetostat 200 mg/day

Primary endpoints: Safety (including DLTs, TEAEs), RP2D, and PK parameters

Secondary endpoints: MTD, efficacy

[a]Number includes 1 patient excluded from all analyses due to a GCP violation; [b]Two of 2 patients in the 300 mg/day cohort experienced DLTs: 1 patient had Grade 3 anemia requiring transfusion and Grade 4 platelet count decreased, and another patient had Grade 4 platelet count decreased.
DLT, dose-limiting toxicity; ECOG PS, Eastern Cooperative Oncology Group performance status; GCP, good clinical practice; MTD, maximum tolerated dose; PK, pharmacokinetics; RP2D, recommended phase 2 dose.

RESULTS

Patient enrollment and disposition

- Patients were enrolled from April 2016 through June 2021
- Median treatment duration at data cutoff (Dec 31, 2022) was 130 days (range, 1–2112)
- Eight (8/71; 11%) patients with T-NHL and 2/19 (11%) patients with B-NHL were receiving ongoing treatment

Demographics and baseline characteristics

- The most common PTCL subtypes were PTCL, not otherwise specified (PTCL, NOS; n = 26, 46%) and angioimmunoblastic T-cell lymphoma (AITL; n = 23, 40%); the most common B-NHL subtypes were diffuse large B-cell lymphoma (DLBCL; n = 7, 37%) and follicular lymphoma (FL; n = 7, 37%; **Table 1**)

Table 1. Baseline demographic and disease characteristics

Characteristic	Total (N = 90)	PTCL (n = 57)	ATLL (n = 14)	B-NHL (n = 19)
Age, y, median (range)	67.5 (26–88)	68 (26–83)	66.5 (37–78)	66 (44–88)
Sex, n (%)				
Male	53 (59)	35 (61)	8 (57)	10 (53)
Female	37 (41)	22 (39)	6 (43)	9 (47)
Country of enrollment, n (%)				
US	44 (49)	39 (68)	5 (36)	0
Japan	46 (51)	18 (32)	9 (64)	19 (100)
ECOG PS, n (%)				
0	44 (49)	21 (37)	8 (57)	15 (79)
1	45 (50)	36 (63)	5 (36)	4 (21)
≥ 2	1 (1)[a]	0	1 (7)[a]	0
Cancer type, n (%)				
B-NHL	19 (21)	0	0	19 (100)
DLBCL	7 (8)	0	0	7 (37)
FL	7 (8)	0	0	7 (37)
Indolent BCL excluding FL	3 (3)	0	0	3 (16)
MCL	1 (1)	0	0	1 (5)
Other BCL	1 (1)	0	0	1 (5)
T-NHL	71 (79)	57 (100)	14 (100)	0
ATLL	14 (16)	0	14 (100)	0
PTCL	57 (63)	57 (100)	0	0
ALCL	2 (2)	2 (4)	0	0
AITL	23 (26)	23 (40)	0	0
PTCL, NOS	26 (29)	26 (46)	0	0
Other TCL[b]	6 (7)	6 (11)	0	0
Prior lines of therapy, median (range)	2 (0–8)	2 (1–8)	2.5 (1–8)	2 (0–6)
***EZH2* GOF mutation**	1 (2)[c]	0[d]	0[e]	1 (11)[f]
Prior HCT, n (%)	18 (20)	16 (28)	2 (14)	0
Allogeneic	4 (4)	2 (4)	2 (14)	0
Autologous	14 (16)	14 (25)	0	0

Data are n (%) or median (range). [a]One patient with an ECOG PS score of 4 was eligible for the study at the time of screening (with a performance score of 0); this ECOG PS score worsened to 4 on C1D1, which was the time point when the data presented here were collected; [b]Includes patients with SPTCL (n = 2), ENKTCL (nasal type; n = 1), hepatosplenic T-cell lymphoma (n = 1), nodal PTCL with TFH phenotype (n = 1), and primary cutaneous gamma-delta T-cell lymphoma (n = 1); [c]*EZH2* mutation status assessed in a total of 49 patients; [d]*EZH2* mutation status assessed in 35 patients with PTCL; [e]*EZH2* mutation status assessed in 5 patients with ATLL; [f]*EZH2* mutation status assessed in 9 patients with B-NHL. ALCL, anaplastic large-cell lymphoma; BCL, B-cell lymphoma; ENKTCL, extranodal NK/T-cell lymphoma; GOF, gain of function; HCT, hematopoietic cell transplantation; MCL, mantle cell lymphoma; NK, natural killer; SPTCL, subcutaneous panniculitis-like T-cell lymphoma; TCL, T-cell lymphoma; TFH, T follicular helper.

Preliminary efficacy

- The ORRs were 55% (30/55; 95% confidence interval [CI], 40.6–68.0) in patients with PTCL, 64% (9/14; 95% CI, 35.1–87.2) in patients with ATLL, and 47% (9/19; 95% CI, 24.4–71.1) in patients with B-NHL (**Table 2**)
- Median progression-free survival (PFS) was 7.7 months (95% CI, 3.6–19.8) in patients with PTCL, 4.1 months (95% CI, 1.9–32.2) in patients with ATLL, and 22.1 months (95% CI, 7.2–not reached [NR]) in patients with B-NHL
- Thirty-two (67%) patients with PTCL (**Figure 2A**), 6 (60%) with ATLL (**Figure 2B**), and 9 (47%) with B-NHL (**Figure 2C**) experienced a maximum reduction of > 50% from baseline in measurable target lesions
- Exposure duration and clinical responses for individual patients with PTCL, ATLL, and B-NHL are shown in **Figure 3A–C**

Table 2. Clinical response

Response	PTCL (n = 55[a]) AITL (n = 22)	PTCL (n = 55[a]) PTCL, NOS (n = 26)	PTCL (n = 55[a]) All PTCL (n = 55[a])	ATLL (n = 14)	B-NHL (n = 19)
Best overall response, n (%)					
CR	10 (45)	7 (27)	17 (31)	4 (29)	2 (11)
PR	4 (18)	6 (23)	13 (24)	5 (36)	7 (37)
SD	1 (5)	3 (12)	4 (7)	1 (7)	8 (42)
PD	5 (23)	8 (31)	15 (27)	3 (21)	2 (11)
NE	1 (5)	0	1 (2)	0	0
Not done	1 (5)	2 (8)	5 (9)	1 (7)	0
ORR, n (%)[b]	14 (64)	13 (50)	30 (55)	9 (64)	9 (47)
95% CI[c]	40.7, 82.8	29.9, 70.1	40.6, 68.0	35.1, 87.2	24.4, 71.1
TTR, median (range), months	1.9 (1.0–3.75)	1.8 (1.6–5.6)	1.8 (1.0–5.6)	1.9 (1.7–19.4)	3.7 (1.9–10.1)
DOR, median (range), months[d]	21.9 (0.03+ to 22.0)	NR (0.03+ to 32.7+)	21.9 (0.03+ to 32.7+)	21.2 (1.4–38.7)	18.4 (0.03+ to 64.0+)
95% CI[e]	1.9, 22.0	4.0, NR	10.2, NR	1.4, 38.7	5.3, NR
Patients censored, n (%)[f]	9 (64)	9 (69)	20 (67)	3 (33)	5 (56)

[a]Two patients without measurable lesions were excluded from response analyses; [b]Responders for ORR were patients whose best response was assessed as CR or PR (including CRu for ATLL); [c]95% CI based on Clopper-Pearson method; [d]Median is from Kaplan-Meier estimate; [e]CI for median is computed using Brookmeyer-Crowley; [f]Percentage is calculated using the number of patients with response as denominator. CRu, uncertified CR; NE, not evaluable; PD, progressive disease; PR, partial response; TTR, time to response; SD, stable disease.

Figure 2. Change in target lesions from baseline in patients with (A) PTCL, (B) ATLL, and (C) B-NHL

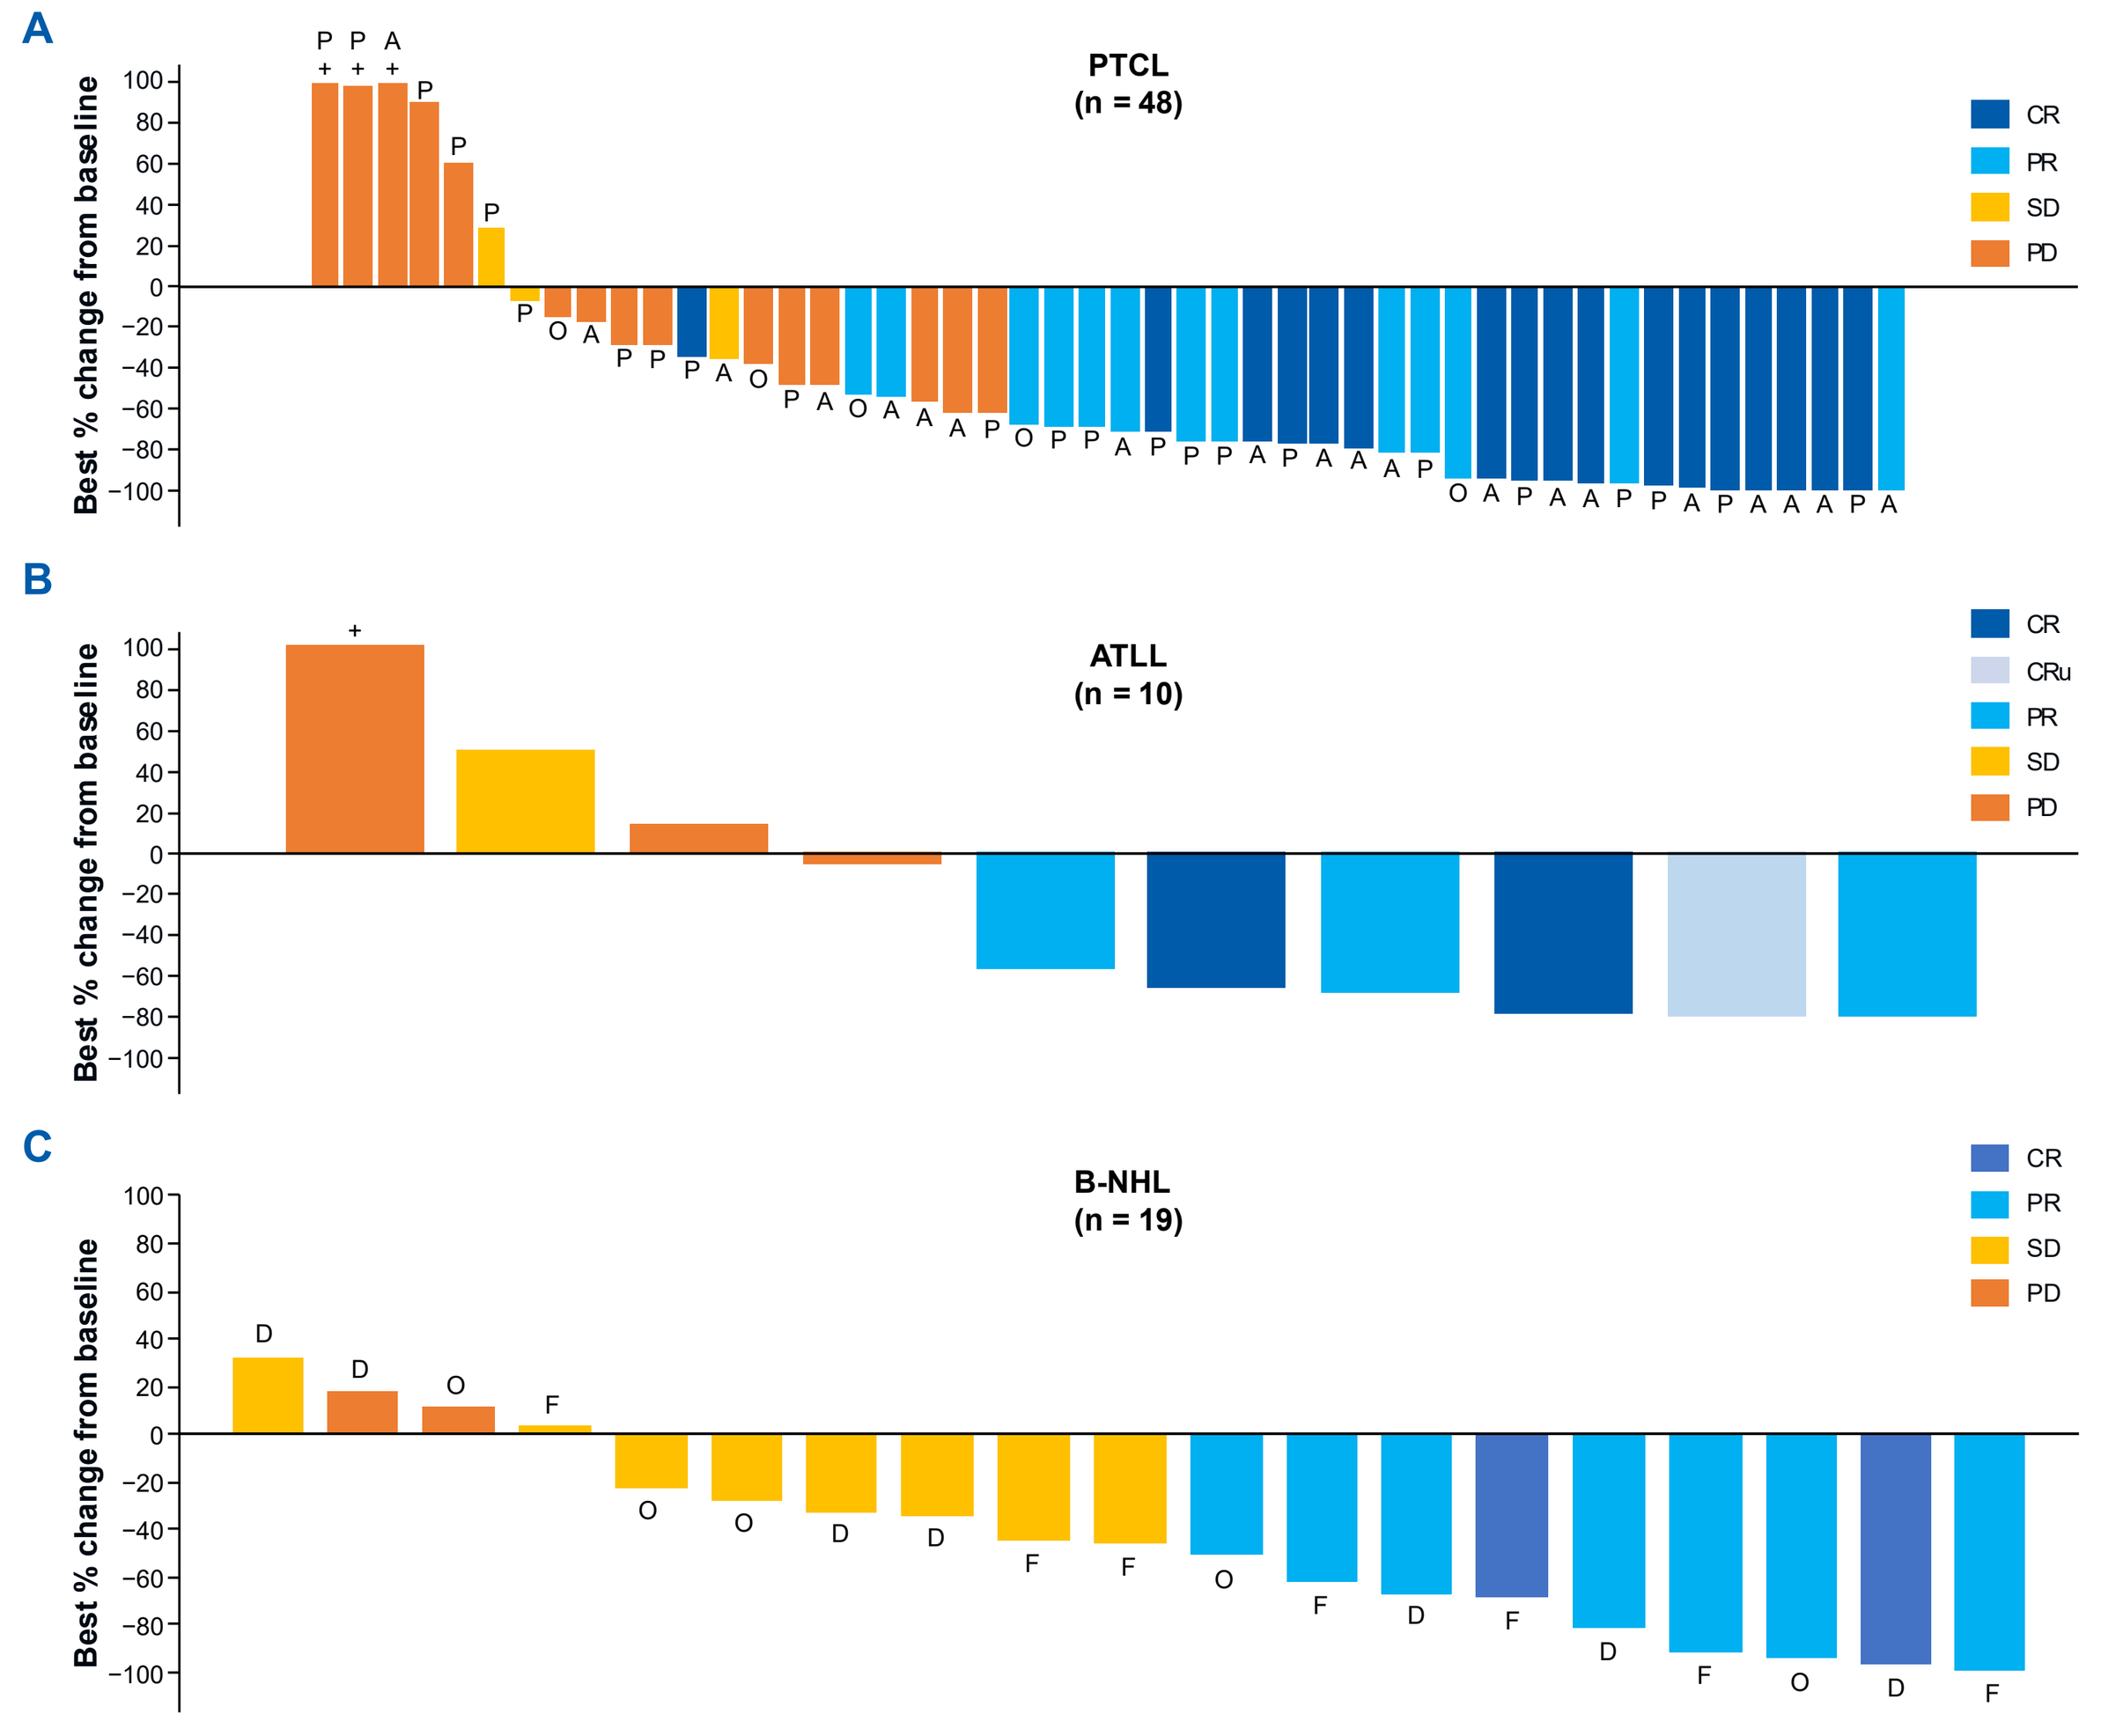
A
PTCL
(n = 48)
B
ATLL
(n = 10)
C
B-NHL
(n = 19)
Best % change from baseline
CR
CRu
PR
SD
PD

Analysis includes all patients with measurable lesions at baseline and at least 1 valid post-baseline assessment. Baseline was defined as the last measurement taken before the first dose of study drug. For each patient, the best percent change from baseline in the sum of area for all target lesions was represented by a vertical bar. Some patients were assessed as having PD because of a new lesion, even though the sum of area in target lesions was reduced by more than 50%. Disease subtypes are shown in panel A as "A" for AITL, "D" for DLBCL, "F" for FL, "P" for PTCL, NOS, and "O" for other lymphomas.

Figure 3. Exposure duration and clinical responses in patients with (A) PTCL, (B) ATLL, and (C) B-NHL

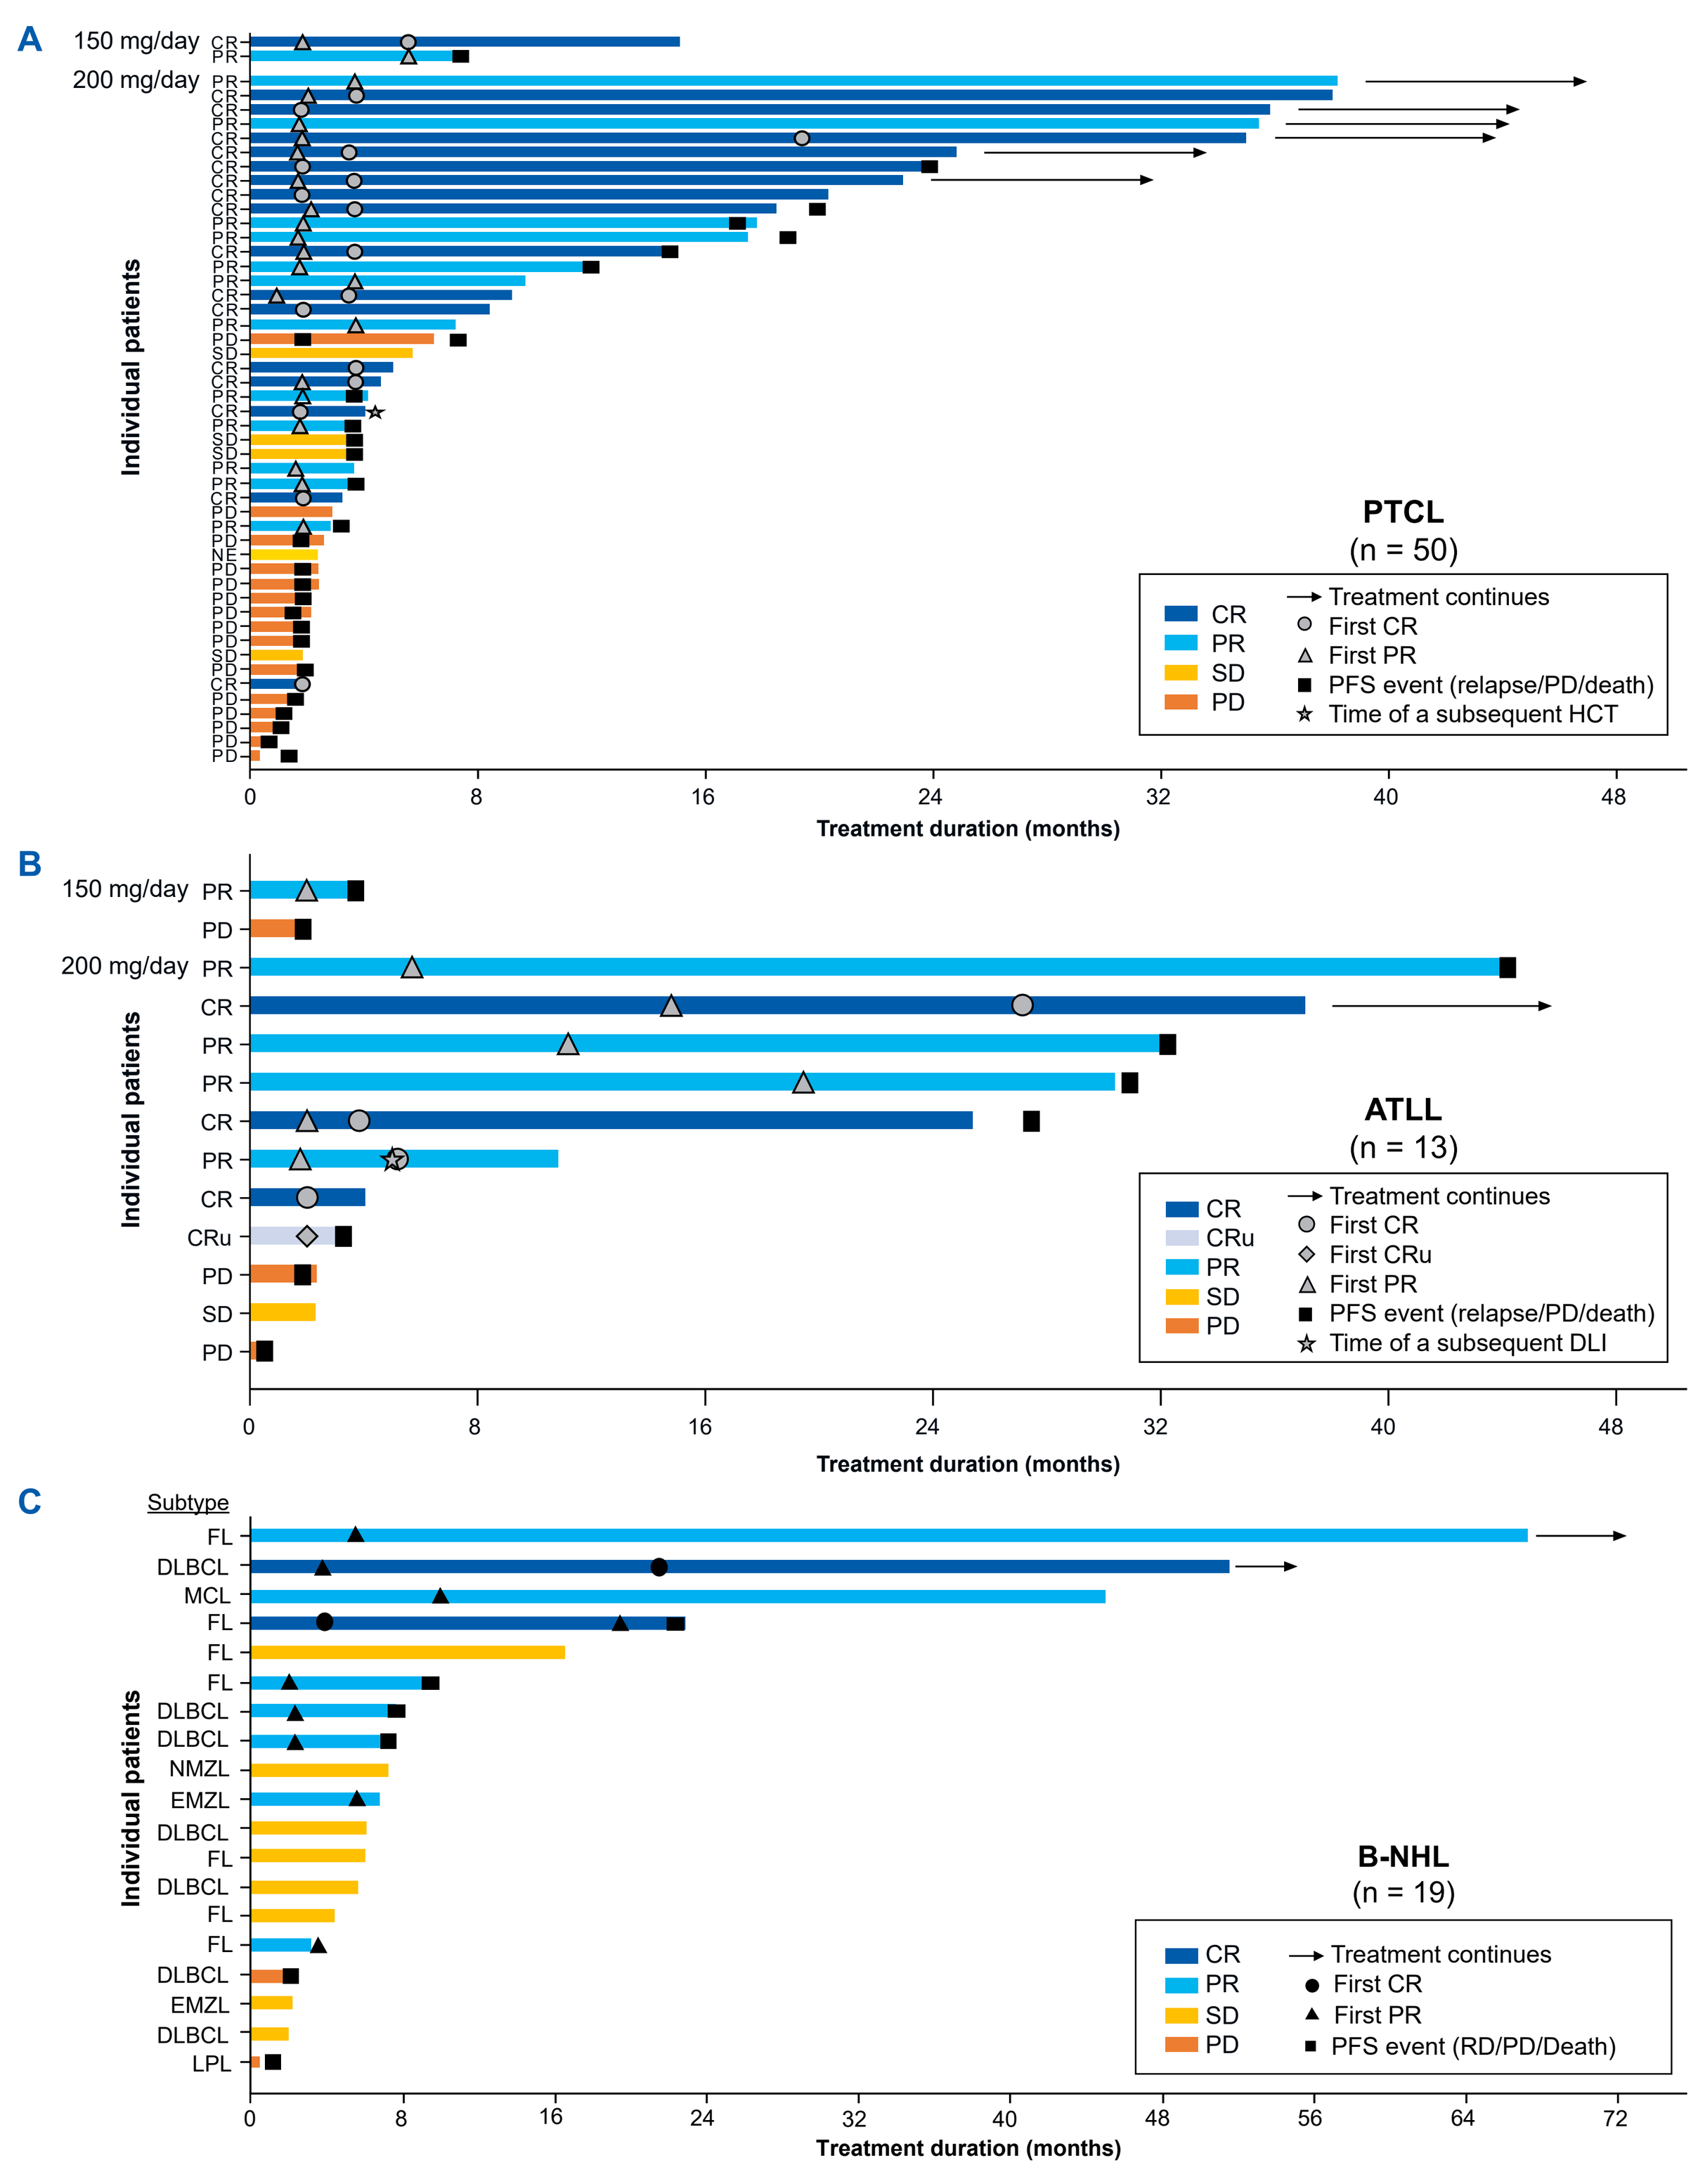
A
150 mg/day
200 mg/day
Individual patients
PTCL
(n = 50)
CR
PR
SD
PD
Treatment continues
First CR
First PR
PFS event (relapse/PD/death)
Time of a subsequent HCT
Treatment duration (months)
B
ATLL
(n = 13)
CR
CRu
PR
SD
PD
Treatment continues
First CR
First CRu
First PR
PFS event (relapse/PD/death)
Time of a subsequent DLI
Treatment duration (months)
C
Subtype
B-NHL
(n = 19)
CR
PR
SD
PD
Treatment continues
First CR
First PR
PFS event (RD/PD/Death)
Treatment duration (months)

Swimmer's plot excludes patients without available response assessments. DLI, donor lymphocyte infusion; EMZL, extranodal marginal zone lymphoma; LPL, lymphoplasmacytic lymphoma; NMZL, nodal marginal zone lymphoma; RD, relapsed disease.

Safety and tolerability

- All 90 patients with T-NHLs and B-NHLs experienced at least 1 TEAE, including 78 (87%) who had treatment-related TEAEs
- The most common TEAEs (any grade) were platelet count decreased (58%), dysgeusia (50%), anemia (42%), neutrophil count decreased (33%), and alopecia (33%; **Table 3**); the most common Grade ≥ 3 TEAEs were cytopenias, including decreased neutrophil count (23%), platelet count (20%), and lymphocyte count (19%; **Table 4**)
- TEAEs required treatment discontinuation for 7 (8%) patients, dose reduction for 9 (10%) patients, and treatment interruption for 42 (47%) patients
- MTD was not reached between 150 and 300 mg/day based on the modified continual reassessment method (mCRM); RP2D/RDE was determined to be 200 mg/day based on the mCRM, safety findings, preliminarily efficacy, and PK/pharmacodynamic analyses

Gene mutation analysis

- In patients with PTCL, Ras homolog family member A (*RHOA*) was identified as the most commonly mutated gene (29% [10/35]; **Figure 4**)
- A GOF *EZH2* mutation was identified in 1 patient with R/R FL; this patient achieved a PR
 - PRs were also observed in 3 patients with B-NHL that did not have *EZH2* mutations

H3K27me3 analysis

- Biomarker analyses demonstrated on-target pharmacodynamic activity of valemetostat in reducing H3K27me3 levels in evaluated tissue specimens
 - Higher area under the plasma concentration-time curve during the dosing interval (AUC_{tau}) of valemetostat was associated with marginal H3K27me3 reductions in normal granulocytes (**Figure 5**)

Table 3. TEAEs (all grades) that occurred in ≥ 15% of patients with NHLs

Preferred term	Valemetostat dose 150 mg/day (n = 7)	200 mg/day (n = 74)	250 mg/day (n = 7)	300 mg/day (n = 2)	All dose levels (N = 90)
Platelet count decreased	5 (71)	38 (51)	7 (100)	2 (100)	52 (58)
Dysgeusia	4 (57)	34 (46)	6 (86)	1 (50)	45 (50)
Anemia	3 (43)	30 (41)	3 (43)	2 (100)	38 (42)
Neutrophil count decreased	2 (29)	23 (31)	3 (43)	2 (100)	30 (33)
Alopecia	3 (43)	23 (31)	4 (57)	0	30 (33)
White blood cell count decreased	1 (14)	20 (27)	4 (57)	2 (100)	27 (30)
Diarrhea	2 (29)	20 (27)	4 (57)	0	26 (29)
Lymphocyte count decreased	3 (43)	12 (16)	6 (86)	2 (100)	23 (26)
Nausea	2 (29)	18 (24)	1 (14)	0	21 (23)
Fatigue	2 (29)	13 (18)	2 (29)	0	17 (19)
Alanine aminotransferase increased	2 (29)	13 (18)	2 (29)	0	17 (19)
Aspartate aminotransferase increased	1 (14)	13 (18)	1 (14)	0	15 (17)
Pyrexia	0	13 (18)	2 (29)	0	15 (17)
Decreased appetite	2 (29)	13 (18)	0	0	15 (17)
Cough	0	14 (19)	0	0	14 (16)

Table 4. TEAEs (Grade ≥ 3) that occurred in ≥ 3 of patients with NHLs

Preferred term	Valemetostat dose 150 mg/day (n = 7)	200 mg/day (n = 74)	250 mg/day (n = 7)	300 mg/day (n = 2)	All dose levels (N = 90)
Neutrophil count decreased	0	17 (23)	2 (29)	2 (100)	21 (23)
Platelet count decreased	2 (29)	13 (18)	1 (14)	2 (100)	18 (20)
Lymphocyte count decreased	2 (29)	10 (14)	5 (71)	0	17 (19)
Anemia	2 (29)	10 (14)	1 (14)	1 (50)	14 (16)
White blood cell count decreased	1 (14)	10 (14)	1 (14)	1 (50)	13 (14)
Hypertension	0	5 (7)	0	0	5 (6)
Gamma-glutamyltransferase increased	1 (14)	4 (5)	1 (14)	0	6 (7)
Aspartate aminotransferase increased	0	4 (5)	0	0	4 (4)
Febrile neutropenia	0	3 (4)	0	0	3 (3)
Hypokalemia	0	3 (4)	0	0	3 (3)
Hypophosphatemia	0	3 (4)	0	0	3 (3)
Blood alkaline phosphatase increased	0	3 (4)	0	0	3 (3)
Pneumocystis jirovecii pneumonia	1 (14)	2 (3)	0	0	3 (3)

Figure 4. Gene mutation profiles of patients with (A) PTCL, (B) ATLL, and (C) B-NHL

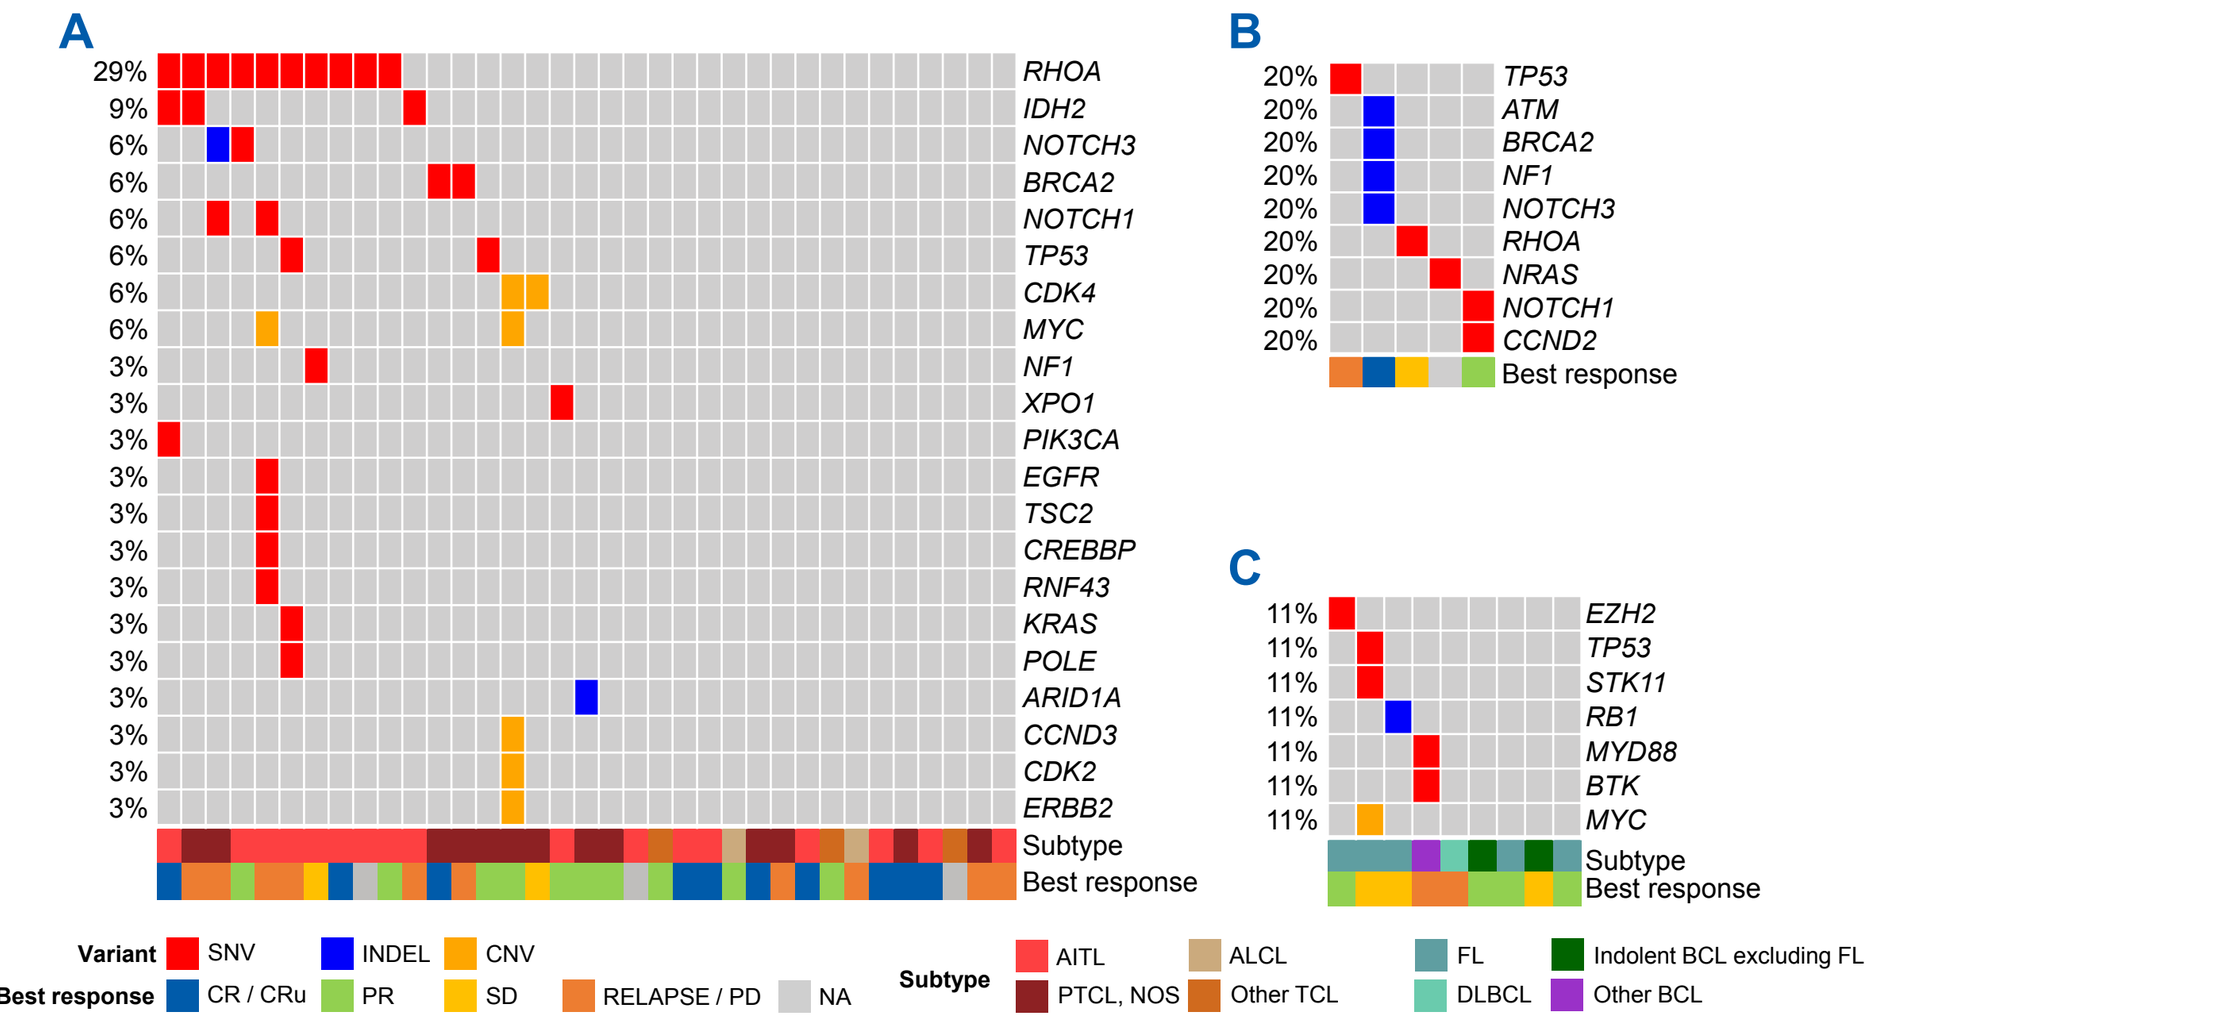
A
RHOA
IDH2
NOTCH3
BRCA2
NOTCH1
TP53
CDK4
MYC
NF1
XPO1
PIK3CA
EGFR
TSC2
CREBBP
RNF43
KRAS
POLE
ARID1A
CCND3
CDK2
ERBB2
Subtype
Best response
B
TP53
ATM
BRCA2
NF1
NOTCH3
RHOA
NRAS
NOTCH1
CCND2
Best response
C
EZH2
TP53
STK11
RB1
MYD88
BTK
MYC
Subtype
Best response
Variant
SNV
INDEL
CNV
Best response
CR / CRu
PR
SD
RELAPSE / PD
NA
Subtype
AITL
PTCL, NOS
ALCL
Other TCL
FL
DLBCL
Indolent BCL excluding FL
Other BCL

Gene mutation profiles at baseline were evaluated for (A) PTCL (n = 35), (B) ATLL (n = 5), and (C) B-NHL (n = 9). The most highly mutated gene was *RHOA*, which was found in 10 (28.6%) patients with PTCL, including 8 (22.9%) patients with AITL. In our study cohort, p.Gly17Val was the most common mutation, and was found in all patients with AITL, which is in line with previous studies.[9,10] There was 1 patient carrying an SNV in *EZH2* representing p.Tyr646Ser from the B-NHL cohort (FL), which has been reported as a GOF mutation in past studies.[11] The percentage on the y-axis indicates frequency of patient(s) harboring a mutation in a corresponding gene among each cohort.
CNV, copy number variant; INDEL, insertion-deletion; NA, not applicable; SNV, single nucleotide variant.

Figure 5. Change in H3K27me3 across AUC_{tau} levels by dose cohort in the biomarker analysis set

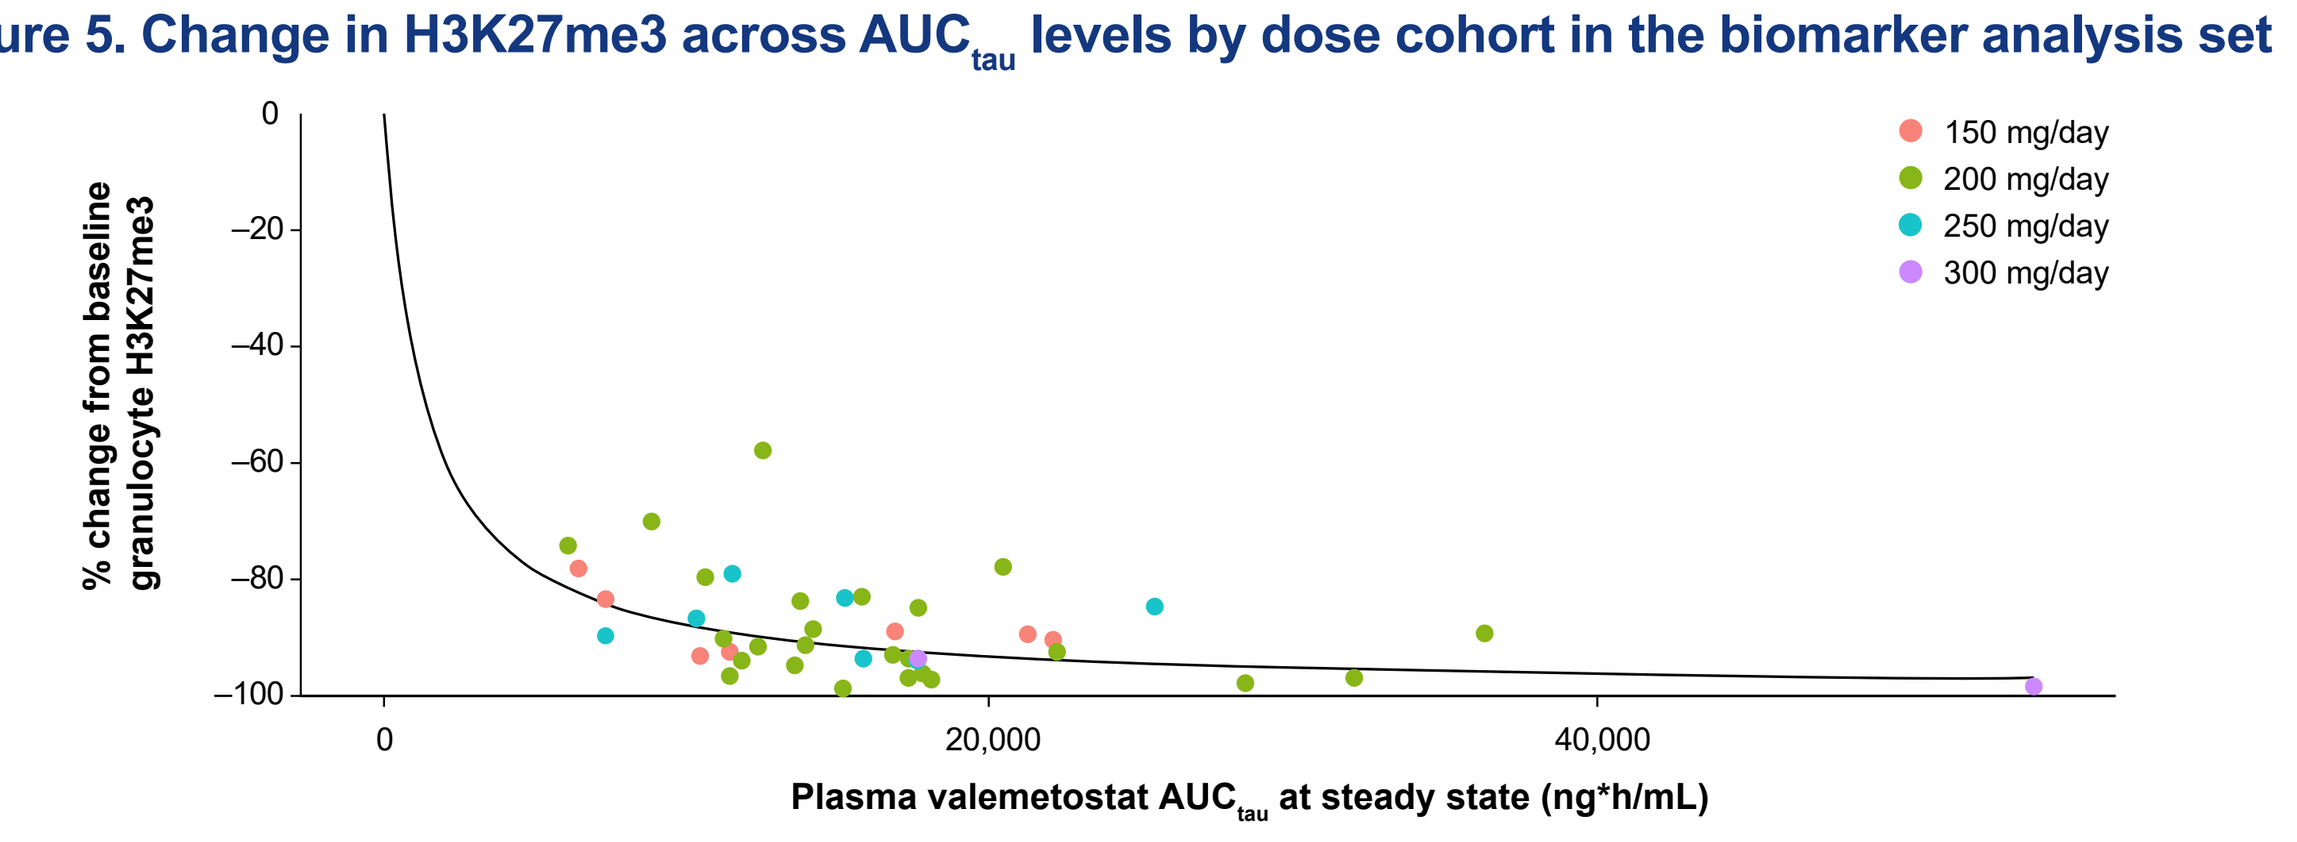
% change from baseline granulocyte H3K27me3
150 mg/day
200 mg/day
250 mg/day
300 mg/day
Plasma valemetostat AUCtau at steady state (ng*h/mL)

REFERENCES

1. Herviou L, et al. *Oncotarget* 2016;7:2284–2296.
2. Nakagawa M, Kitabayashi I. *Cancer Sci* 2018;109:2342–2348.
3. Yamagishi M, et al. *Cell Rep* 2019;29:2321–2337e7.
4. Fujikawa D, et al. *Blood* 2016;127:1790–1802.
5. Hama Y, et al. *Blood* 2019;134:4642.
6. Honma D, et al. *Cancer Sci* 2017;108:2069–2078.
7. Izutsu K, et al. *Blood* 2023;141:1159–1168.
8. EZHARMIA® (valemetostat tosilate). [package insert]. Tokyo, Japan: Daiichi Sankyo; 2022.
9. Manso R, et al. *Blood* 2014;123:2893–2894.
10. Yoo HY, et al. *Nat Genet* 2014;46:371–375.
11. Bödör C, et al. *Blood* 2013;122:3165–3168.

ACKNOWLEDGMENTS

- The authors thank the patients, families, and caregivers for their participation, and the study staff for their contributions
- The authors thank the following investigators[a] for contributing to the enrollment of patients and study design: Jonathan Brammer, Kisato Nosaka, Jia Ruan, Atae Usunomiya, Jie Wang, and Jasmine Zain
- This study is sponsored by Daiichi Sankyo Co., Ltd.
- Editorial support was provided by Luca Scrivano, PhD, and Korin Albert, PhD, of Excerpta Medica, funded by Daiichi Sankyo Co., Ltd., and in accordance with Good Publication Practice guidelines

[a]Investigators are listed in alphabetical order of surname.

Presented at the 12th Annual Meeting of the Society of Hematologic Oncology (SOHO); September 4–7; Houston, TX, US.