Valemetostat for Patients With Relapsed/Refractory Peripheral T-Cell Lymphomas: The Phase 2 VALENTINE-PTCL01 Trial

TCL-394

Steven M. Horwitz, MD[1], Koji Izutsu, MD, PhD[2], Neha Mehta-Shah, MD[3], Raul Córdoba, MD, PhD[4], **Stefan K. Barta, MD, MRCP, MS[5]**, Emmanuel Bachy, MD, PhD[6], Giuseppe Gritti, MD, PhD[7], Eric Jacobsen, MD[8], Shigeru Kusumoto, MD, PhD[9], Yann Guillermin, MD[10], Anca Prica, MD[11], Dok Hyun Yoon, MD, PhD[12], Eva Domingo Domenech, MD[13], Jie Wang, MD, MS[14], Jin Seok Kim, MD, PhD[15], Kate Cwynarski, MBBS, PhD, FRCP, FRCPath[16], Marjolein van der Poel, MD, PhD[17], Ai Inoue, MD, PhD[18], Jin Jin, PhD[18], Sutan Wu, PhD[18], Keiko Nakajima, MD, MS[18], Pier Luigi Zinzani, MD[19]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, US; [2]National Cancer Center Hospital, Tokyo, Japan; [3]School of Medicine, Washington University, St. Louis, MO, US; [4]Health Research Institute IIS-FJD, Fundación Jiménez Díaz University Hospital, Madrid, Spain; [5]Perelman Center for Advanced Medicine – University of Pennsylvania, Philadelphia, PA, US; [6]Lyon-Sud Hospital Center, Pierre-Bénite, France; [7]ASST Papa Giovanni XXIII, Bergamo, Italy; [8]Dana-Farber Cancer Institute, Boston, MA, US; [9]Nagoya City University Graduate School of Medical Sciences, Nagoya, Japan; [10]Léon Bérard Center, Lyon, France; [11]University Health Network, Toronto, ON, Canada; [12]Asan Medical Center, University of Ulsan College of Medicine, Seoul, Korea; [13]Institut Catalá d'Oncologia L'Hospitalet, Barcelona, Spain; [14]Duke Cancer Center, Durham, NC, US; [15]Severance Hospital, Yonsei University Health System, Seoul, Korea; [16]University College London Hospital, London, UK; [17]Maastricht University Medical Center, Maastricht, Netherlands; [18]Daiichi Sankyo Inc., Basking Ridge, NJ, US; [19]IRCCS Azienda Ospedaliero-Universitaria di Bologna, Istituto di Ematologia "Seràgnoli", and Dipartimento di Scienze Mediche e Chirurgiche, Università di Bologna, Bologna, Italy

PURPOSE

- To describe the safety, tolerability, and efficacy of valemetostat in patients with relapsed/refractory (R/R) peripheral T-cell lymphoma (PTCL) enrolled in the open-label, single-arm, global, phase 2 VALENTINE-PTCL01 study (DS3201-A-U202; NCT04703192)

CONCLUSIONS

- Valemetostat demonstrated a high response rate and durable responses in patients with R/R PTCL, who have limited treatment options
 - Responses were observed across all PTCL subtypes
 - Clinical responses were observed in patients who relapsed or were refractory to their last line of therapy (LOT), and regardless of the number of prior treatments, including those heavily pretreated
 - Ten (8.4%) patients treated with valemetostat proceeded to allogeneic hematopoietic cell transplantation (allo-HCT)
- Valemetostat demonstrated an acceptable safety profile
 - The most frequently observed treatment-emergent adverse events (TEAEs) were cytopenias
 - TEAEs were generally manageable and rarely led to treatment discontinuation

Mobile-friendly Poster

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without written permission of the authors.

INTRODUCTION

- PTCLs account for 10% of non-Hodgkin lymphomas in Western countries and 20% of all lymphomas in Asia[1,2]
- Patients with PTCL typically have poor prognosis, as evidenced by low overall survival (OS) and progression-free survival (PFS) rates[3]
- Over expression of enhancer of zeste homolog (EZH)2, the catalytic subunit of polycomb repressive complex 2, drives the development and progression of many types of cancers, including PTCL[4–7]
- Valemetostat tosylate (valemetostat) is a novel, potent, and selective dual inhibitor of EZH2 and EZH1 approved in Japan at a dose of 200 mg/day for the treatment of R/R PTCL and adult T-cell leukemia/lymphoma[8]

METHODS

Study design

- Eligible patients were ≥ 18 years of age, had confirmed PTCL as defined by World Health Organization (WHO) 2016 classification criteria,[9] and had R/R disease after ≥ 1 prior line of systemic therapy
- Patients received valemetostat 200 mg/day in continuous 28-day cycles until disease progression or unacceptable toxicity (**Figure 1**)

Efficacy analysis

- The primary endpoint was objective response rate (ORR) by computed tomography (CT)-based blinded independent central review (BICR) assessment according to Lugano 2014 criteria[10]
- Positron emission tomography (PET)-CT–based clinical response assessment (BICR) was an exploratory endpoint

Safety analysis

- The safety and tolerability of valemetostat was a secondary endpoint

Figure 1. Study design, key inclusion criteria, and endpoints

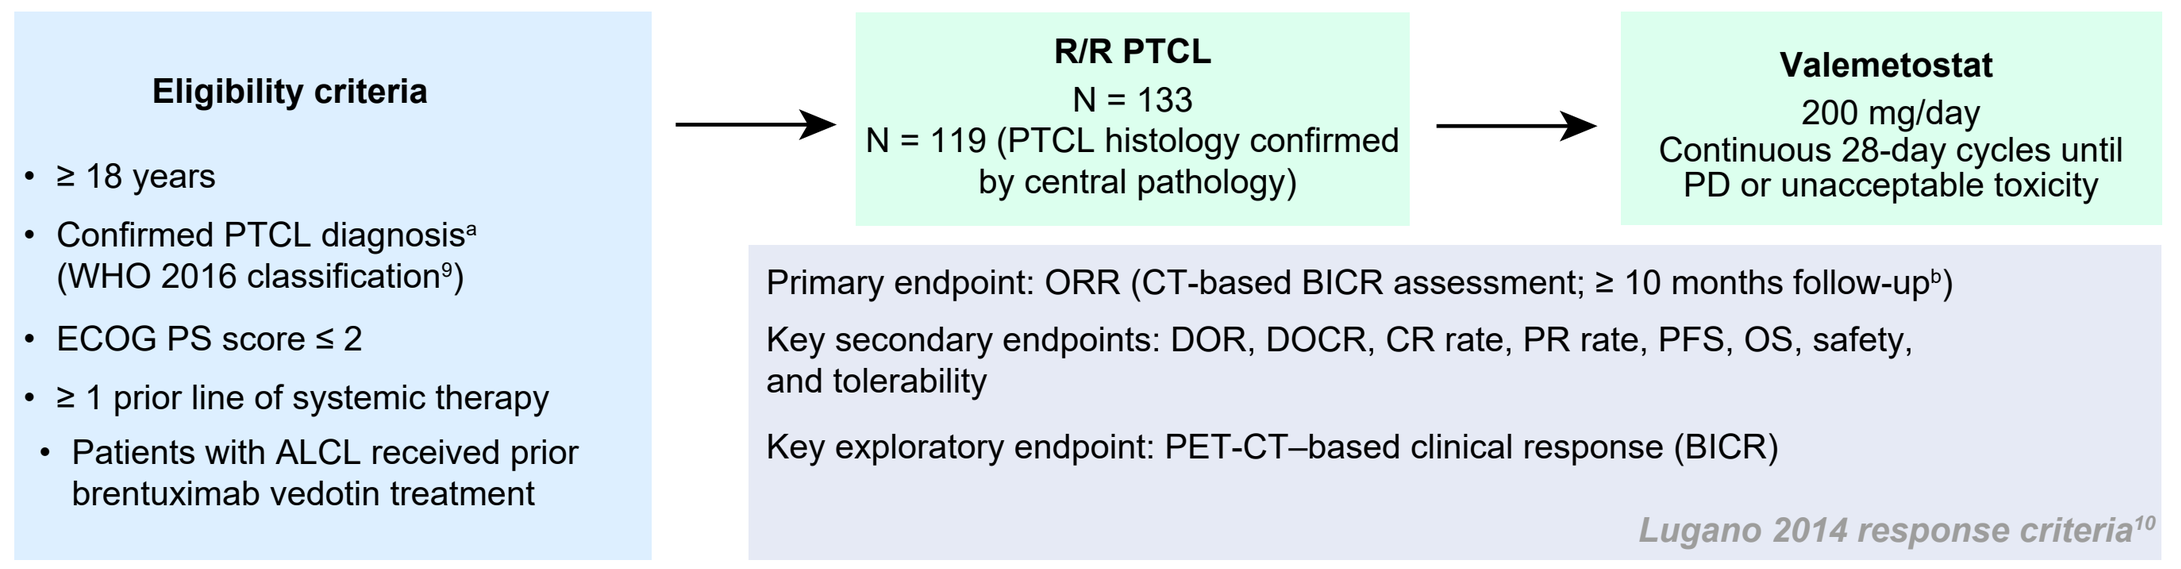
Eligibility criteria
≥ 18 years
Confirmed PTCL diagnosis[a] (WHO 2016 classification[9])
ECOG PS score ≤ 2
≥ 1 prior line of systemic therapy
Patients with ALCL received prior brentuximab vedotin treatment
R/R PTCL
N = 133
N = 119 (PTCL histology confirmed by central pathology)
Valemetostat
200 mg/day
Continuous 28-day cycles until PD or unacceptable toxicity
Primary endpoint: ORR (CT-based BICR assessment; ≥ 10 months follow-up[b])
Key secondary endpoints: DOR, DOCR, CR rate, PR rate, PFS, OS, safety, and tolerability
Key exploratory endpoint: PET-CT–based clinical response (BICR)
Lugano 2014 response criteria[10]

[a]PTCL subtypes included AITL, FTL, PTCL-TFH, PTCL-NOS, ALCL (ALK[+/−]), EATL, MEITL, HSTL, PCGTL, or CD8[+] PCAECTCL; subtypes were determined prior to the initiation of study drug according to 2016 WHO classification. [b]Primary analysis was planned at least 10 months after the first dose of the last enrolled patient.
AITL, angioimmunoblastic T-cell lymphoma; ALCL, anaplastic large cell lymphoma; ALK, anaplastic lymphoma kinase; CD, cluster of differentiation; CD8[+] PCAECTCL, primary cutaneous CD8[+] aggressive epidermotropic cytotoxic T-cell lymphoma; CR, complete response; DOCR, duration of complete response; DOR, duration of response; EATL, enteropathy-associated T-cell lymphoma; ECOG PS, Eastern Cooperative Oncology Group performance status; FTL, follicular T-cell lymphoma; HSTL, hepatosplenic T-cell lymphoma; MEITL, monomorphic epitheliotropic intestinal T-cell lymphoma; NOS, not otherwise specified; PCGTL, primary cutaneous gamma delta T-cell lymphoma; PD, progressive disease; PR, partial response; TCL, T-cell lymphoma; TFH, T follicular helper.

RESULTS

Patient characteristics

- Overall, 133 patients with R/R PTCL were included in the study and received ≥ 1 valemetostat dose (**Figure 2**)
- Median age was 69 (range 22–85) years; most patients (68.4%) were male; the median number of the prior LOTs was 2 (range 1–2); and 26.3% of patients had previously undergone hematopoietic cell transplantation (HCT) (**Table 1**)
- In total, 119 patients had a confirmed PTCL subtype and were eligible for efficacy analysis
 - The most common PTCL subtypes were AITL (31.6%) and PTCL, NOS (30.8%)

Figure 2. Enrollment and disposition

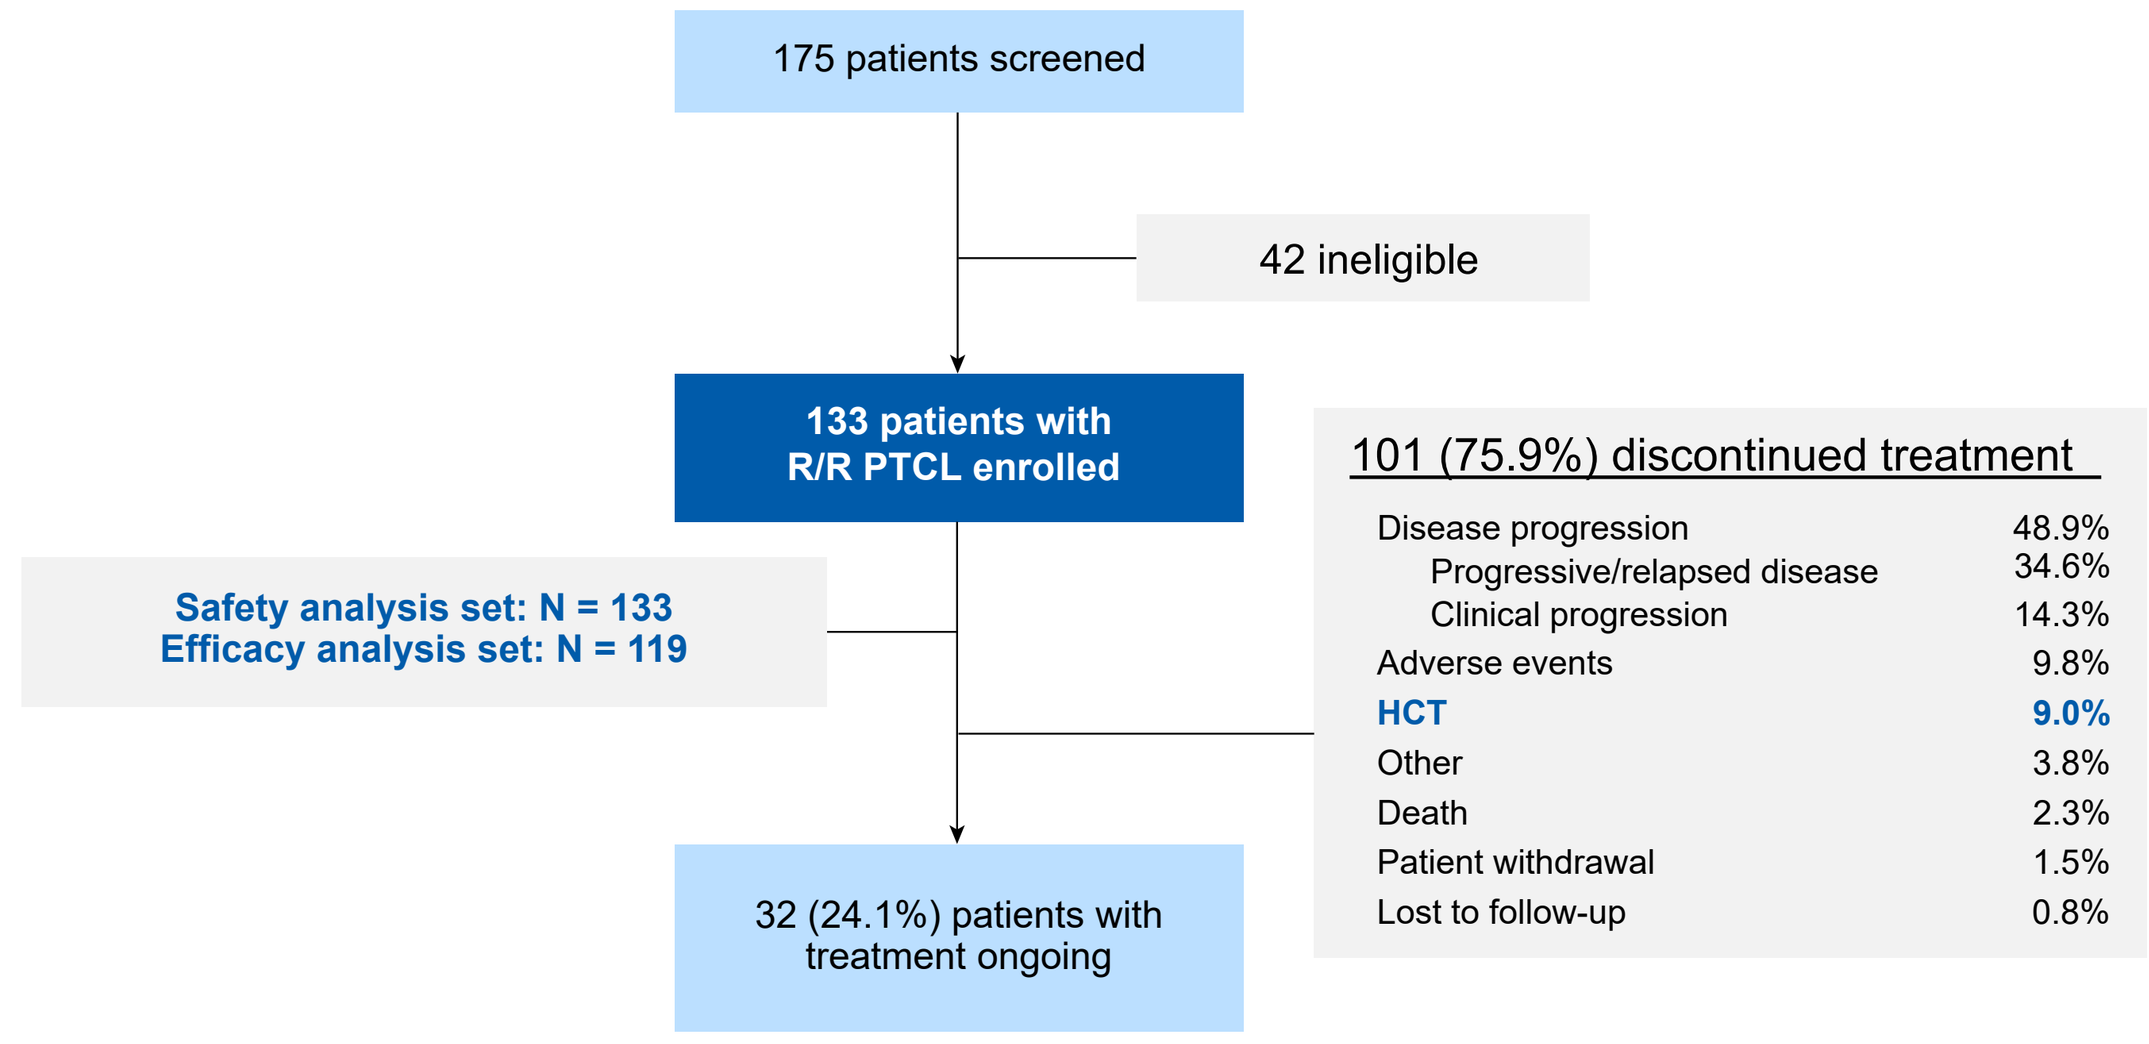
175 patients screened
42 ineligible
133 patients with R/R PTCL enrolled
Safety analysis set: N = 133
Efficacy analysis set: N = 119
101 (75.9%) discontinued treatment
Disease progression 48.9%
Progressive/relapsed disease 34.6%
Clinical progression 14.3%
Adverse events 9.8%
HCT 9.0%
Other 3.8%
Death 2.3%
Patient withdrawal 1.5%
Lost to follow-up 0.8%
32 (24.1%) patients with treatment ongoing

Table 1. Baseline demographics and disease characteristics

Characteristic	PTCL (N = 133)
Median age, years (range)	69.0 (22–85)
Sex, n (%)	
Male	91 (68.4)
Female	42 (31.6)
ECOG PS score, n (%)	
0	58 (43.6)
1	65 (48.9)
≥ 2	10 (7.6)
Median prior LOT (range)	2.0 (1–12)
1	36 (27.1)
2	36 (27.1)
≥ 3	61 (45.9)
Prior HCT, n (%)	35 (26.3)
Autologous	32 (24.1)
Allogeneic	5 (3.8)
PTCL subtypes, n (%)	
TFH phenotype	
AITL	42 (31.6)
Nodal PTCL with TFH phenotype	8 (6.0)
FTL	3 (2.3)
PTCL-NOS	41 (30.8)
ALCL	
ALK[+]	7 (5.3)
ALK[−]	2 (1.5)
MEITL	1 (0.8)
CD8[+] PCAECTCL	1 (0.8)
PCGTL	1 (0.8)
Other TCL[a]	13 (9.8)
Non-TCL or undetermined[b]	6 (4.5)
Missing[c]	8 (6.0)

[a]Includes patients with eligible but undetermined subtypes. [b]Includes patients with undetermined eligibility due to the limited sample tumor tissue. [c]Includes patients with either no sample or a sample insufficient for review.

Disposition

- At the data cut-off date of May 5, 2023, 24.1% (32/133) of patients were still receiving treatment
 - Median treatment duration was 18.0 (range 0.3–93.4) weeks

Efficacy

- In the R/R PTCL efficacy analysis set (N = 119), CT-based ORR was 43.7% (52/119; 95% confidence interval [CI], 34.6–53.1; **Figure 3**)
- Median time to response was 8.1 (range, 5–37) weeks, and median DOR was 11.9 months (95% CI, 7.8–not evaluable [NE]; **Figure 4**)
- ORR per PTCL subtype ranged from 31.7% (PTCL-NOS) to 54.8% (AITL; **Figure 5**)

Figure 3. Clinical response (BICR assessment)

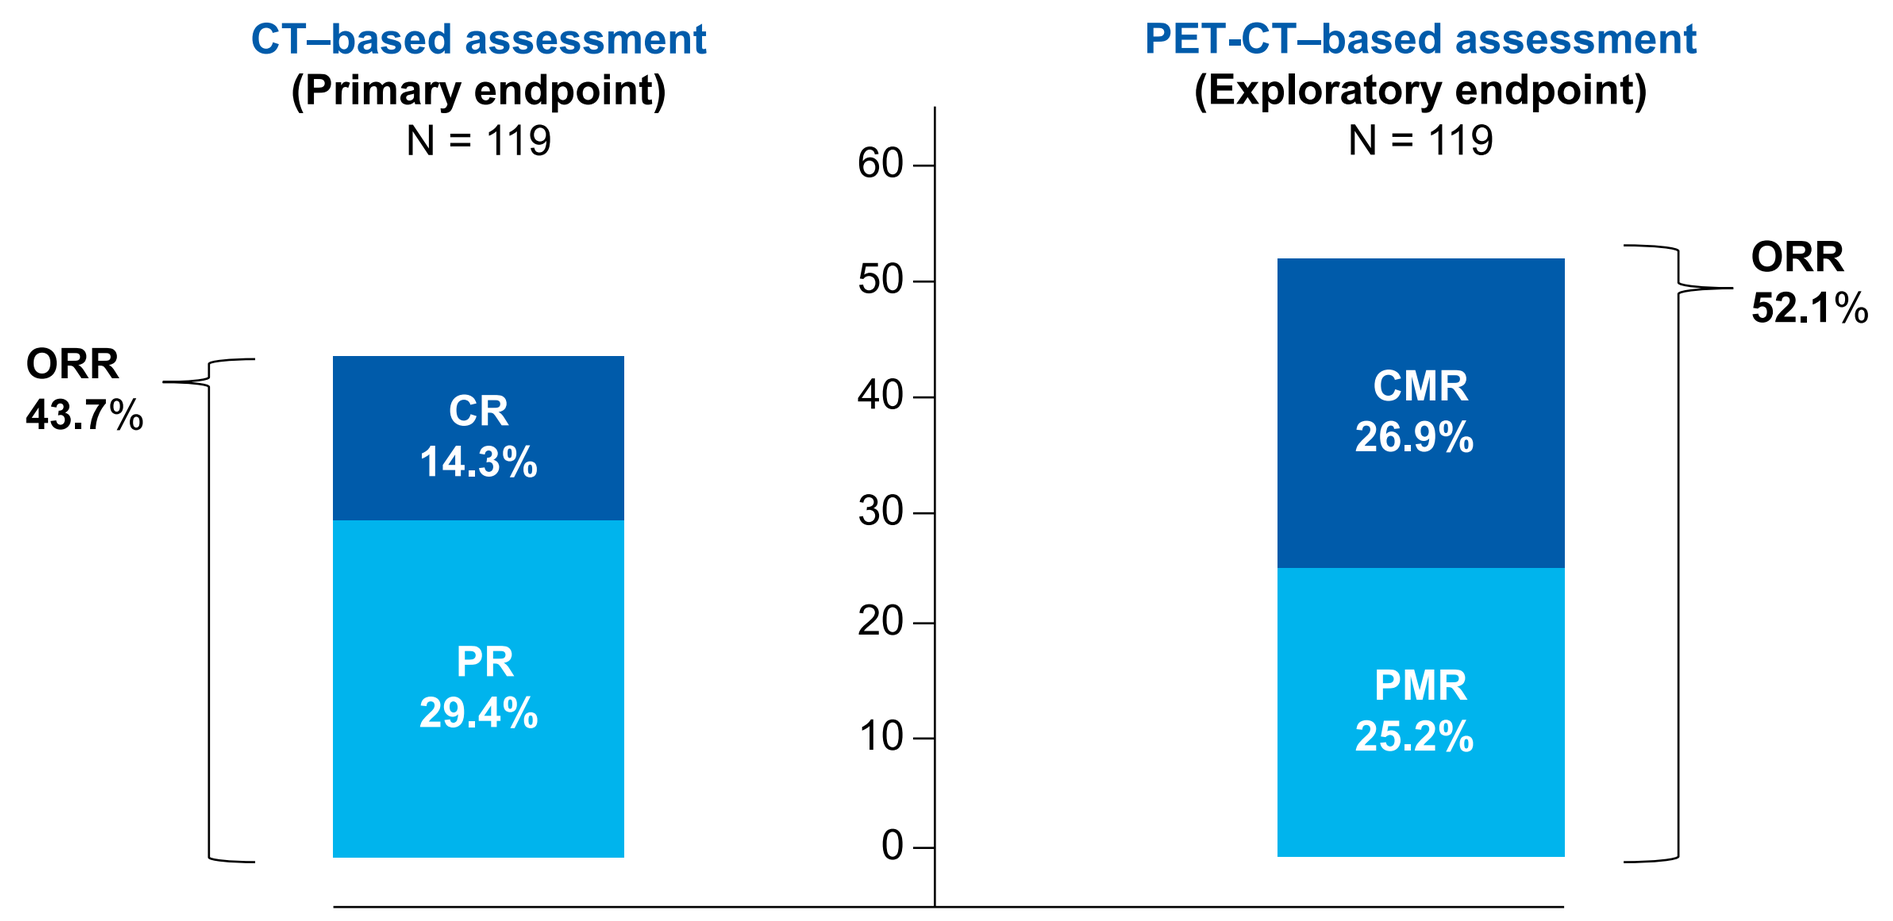
CT–based assessment
(Primary endpoint)
N = 119
ORR 43.7%
CR 14.3%
PR 29.4%
PET-CT–based assessment
(Exploratory endpoint)
N = 119
ORR 52.1%
CMR 26.9%
PMR 25.2%

CMR, complete metabolic response; PMR, partial metabolic response.

Figure 4. Duration of response (CT-based BICR assessed)

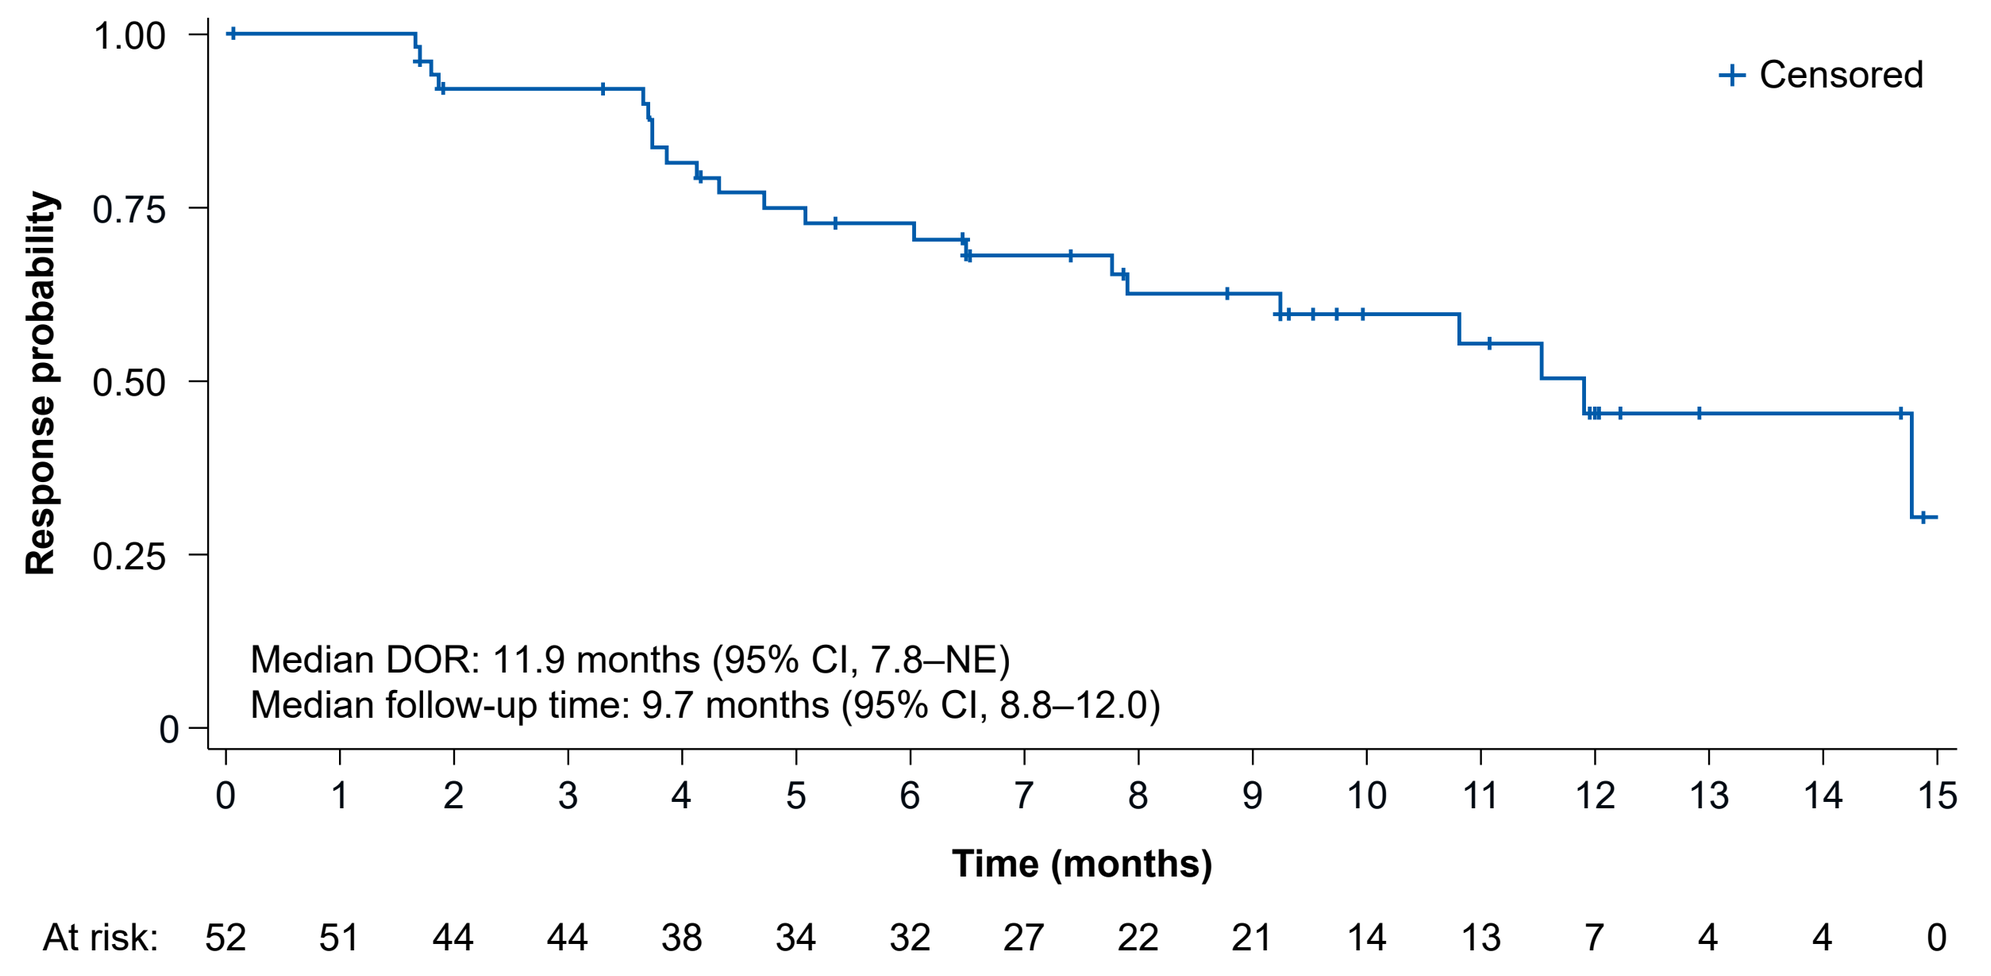
Response probability
Censored
Median DOR: 11.9 months (95% CI, 7.8–NE)
Median follow-up time: 9.7 months (95% CI, 8.8–12.0)
Time (months)
At risk: 52 51 44 44 38 34 32 27 22 21 14 13 7 4 4 0

Figure 5. CT-based (A) and PET-CT-based (B) responses by PTCL subtype (BICR assessment)

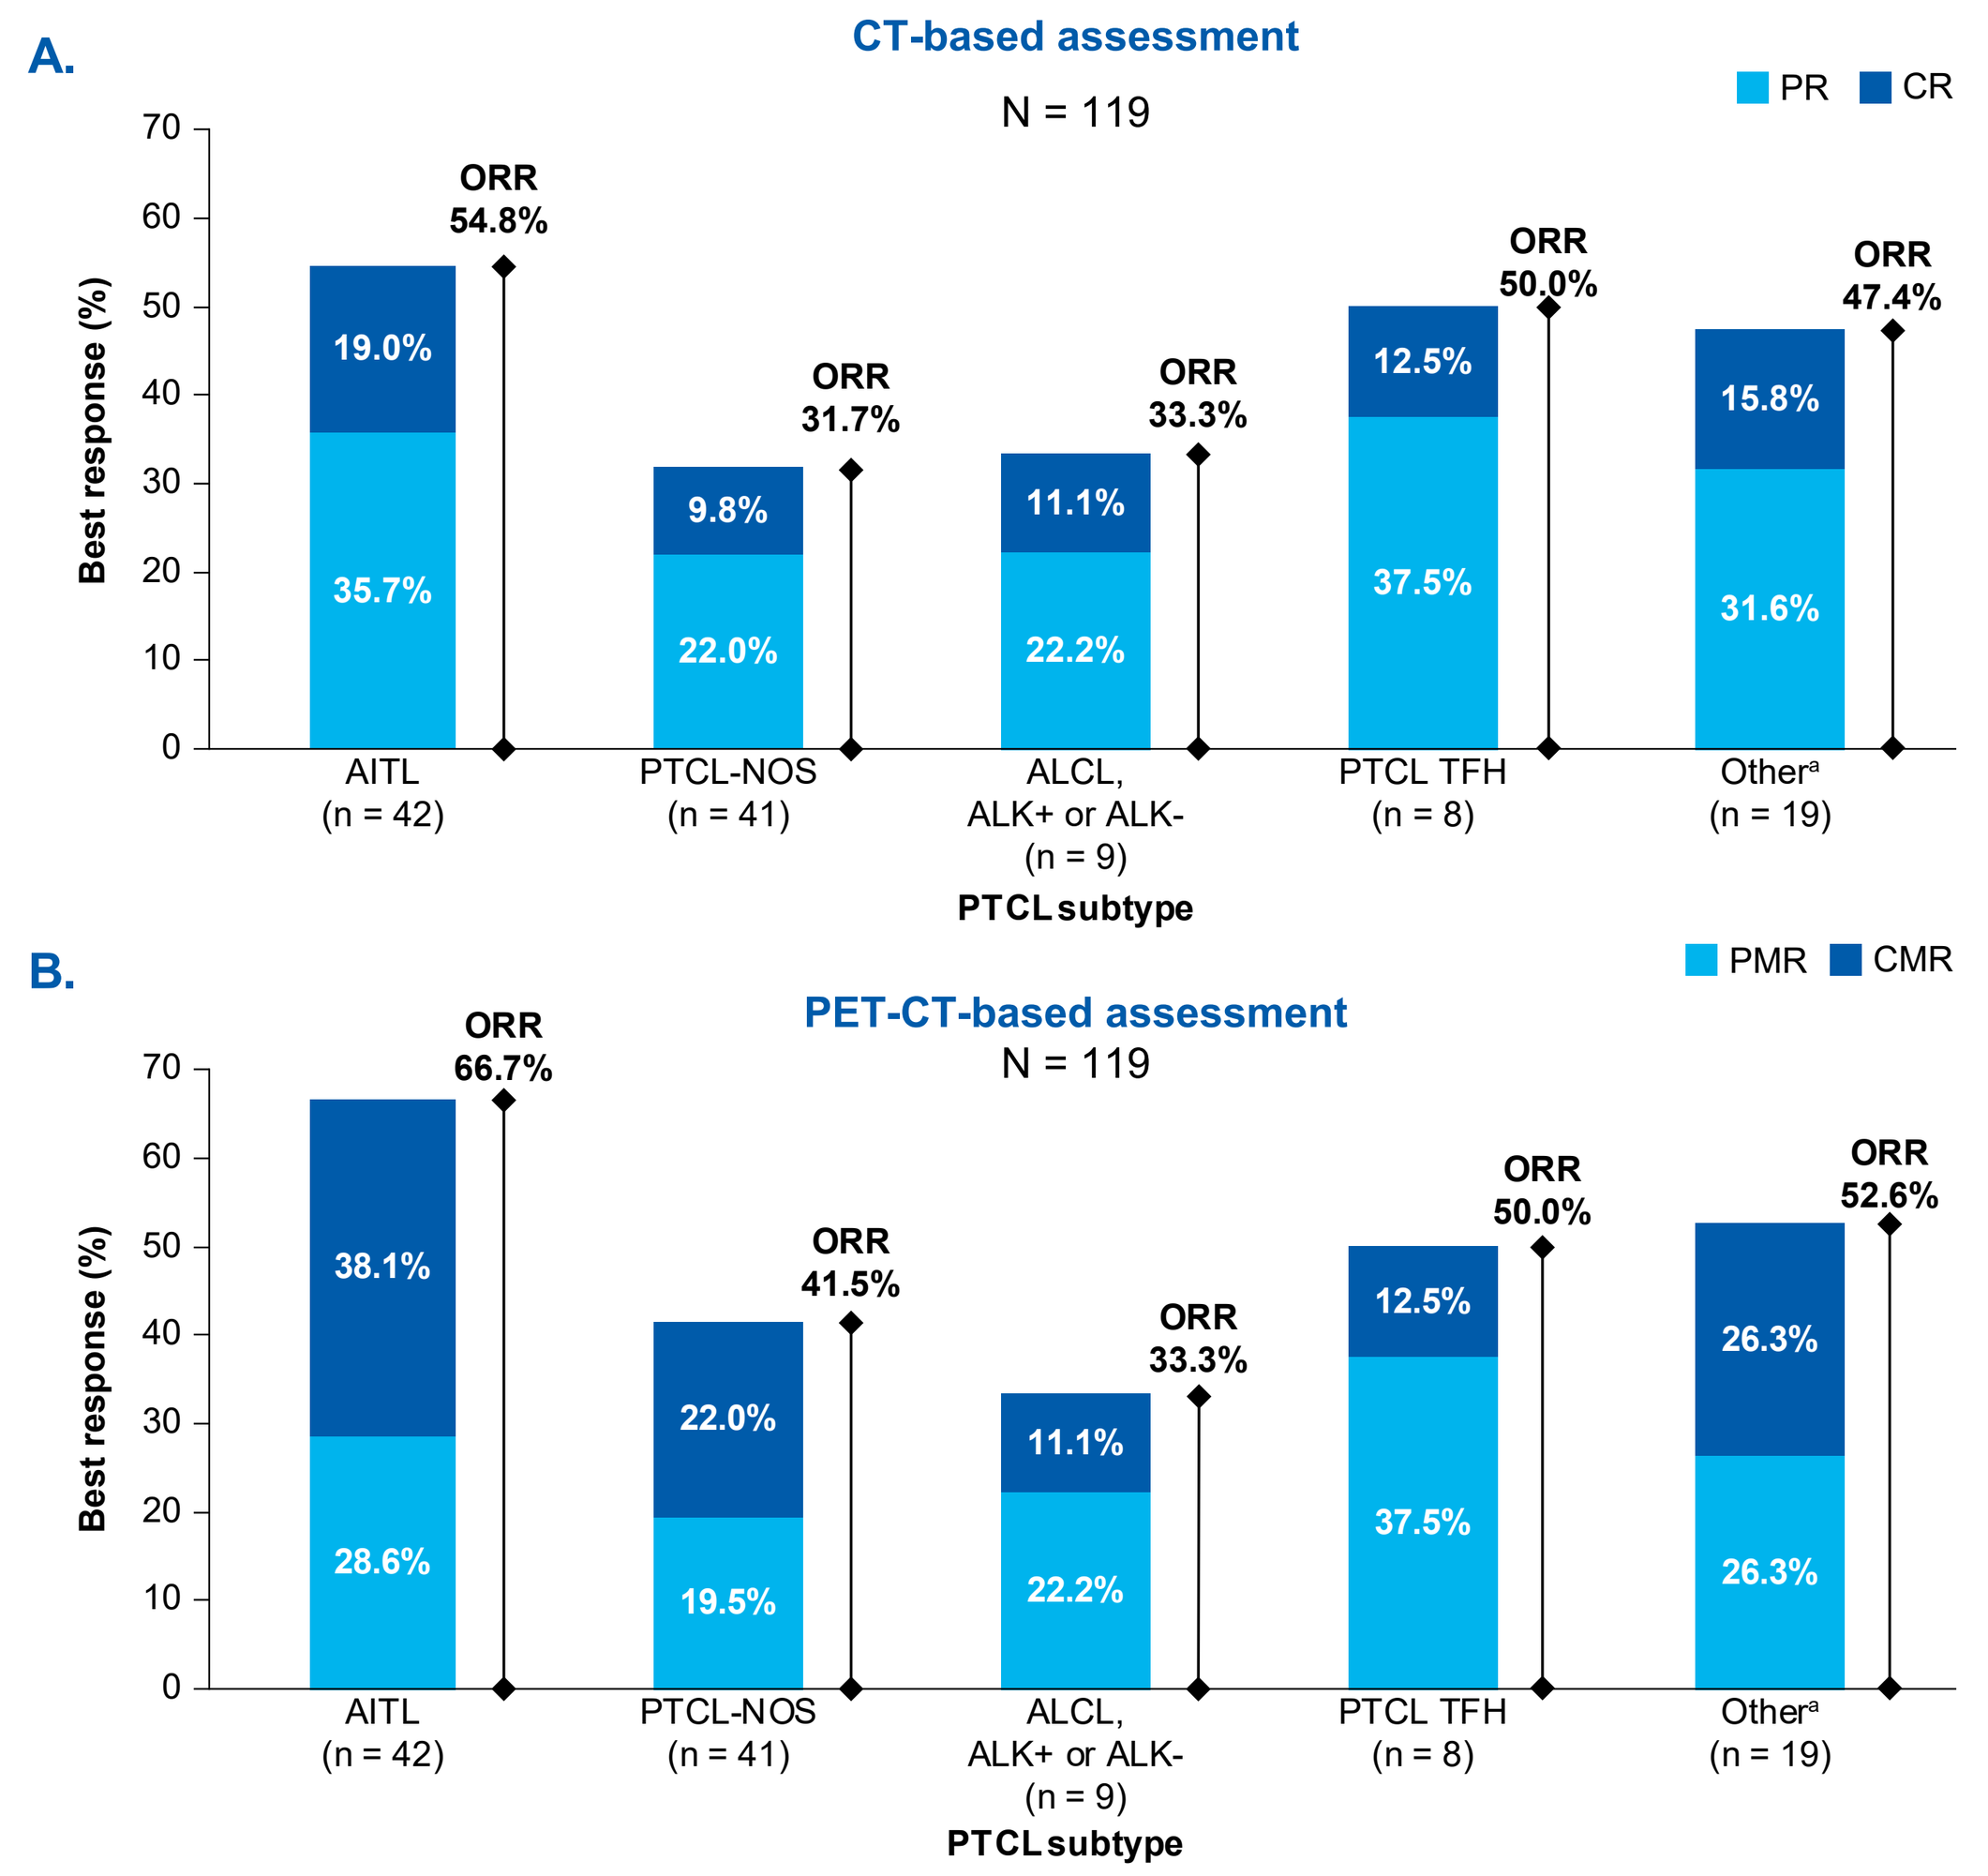
A.
CT-based assessment
N = 119
PR CR
Best response (%)
AITL (n = 42): 35.7%, 19.0%, ORR 54.8%
PTCL-NOS (n = 41): 22.0%, 9.8%, ORR 31.7%
ALCL, ALK+ or ALK- (n = 9): 22.2%, 11.1%, ORR 33.3%
PTCL TFH (n = 8): 37.5%, 12.5%, ORR 50.0%
Other[a] (n = 19): 31.6%, 15.8%, ORR 47.4%
PTCL subtype
B.
PET-CT-based assessment
N = 119
PMR CMR
AITL (n = 42): 28.6%, 38.1%, ORR 66.7%
PTCL-NOS (n = 41): 19.5%, 22.0%, ORR 41.5%
ALCL, ALK+ or ALK- (n = 9): 22.2%, 11.1%, ORR 33.3%
PTCL TFH (n = 8): 37.5%, 12.5%, ORR 50.0%
Other[a] (n = 19): 26.3%, 26.3%, ORR 52.6%
PTCL subtype

[a]Other TCLs include 3 patients with FTL, 1 with PCGTL, 1 with CD8[+] PCAECTCL, 1 with MEITL, and 13 with other eligible, but undetermined PTCL subtypes.

- PET-CT-based ORR was 52.1% (62/119; 95% CI, 42.8–61.3; **Figure 3**)
 - In total, 26.9% (32/119) achieved CMR
 - In total, 25.2% (30/119) achieved PMR
- ORR was nominally better for patients with fewer prior treatment lines (1 vs 2 vs ≥ 3) and for those with relapsed disease (vs refractory) (**Figure 6**)
- Median CT-based PFS was 5.5 months (95% CI, 3.5–8.3) and median OS was 17.0 months (95% CI, 13.5–NE; **Figure 7**)
- Ten (8.4%) patients treated with valemetostat proceeded to allo-HCT, including 6 patients (5.0%) with CR; median time from first valemetostat dose to subsequent allo-HCT was 6.7 months

Figure 6. CT-based ORR by number of prior LOTs and last treatment outcome

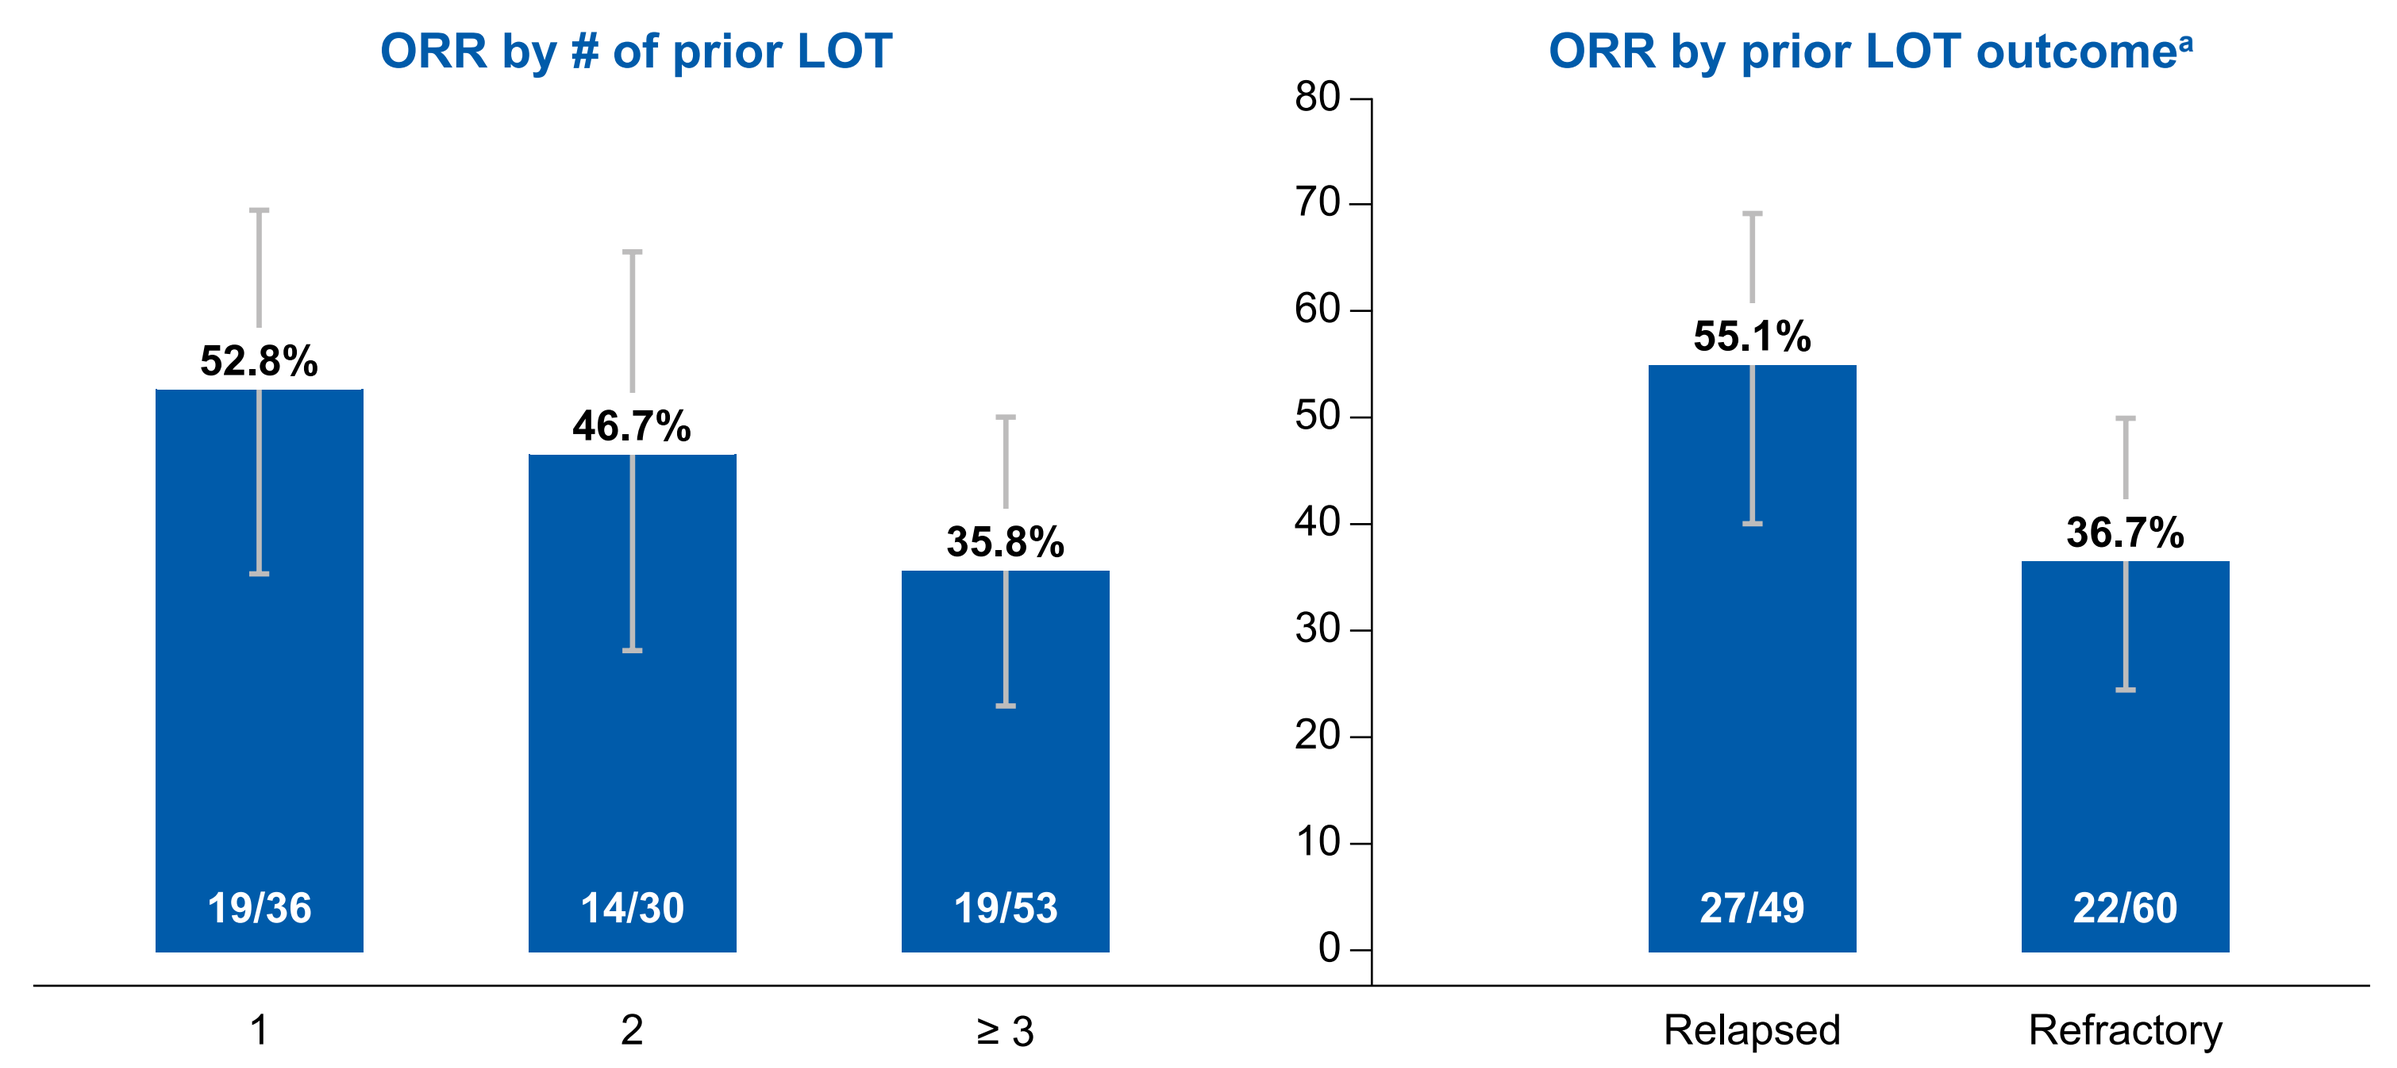
ORR by # of prior LOT
1: 52.8% (19/36)
2: 46.7% (14/30)
≥ 3: 35.8% (19/53)
ORR by prior LOT outcome[a]
Relapsed: 55.1% (27/49)
Refractory: 36.7% (22/60)

Data cutoff: May 5, 2023.
[a]If the best response of the last prior LOT was 'not assessable' or 'unknown', the patient was excluded. Refractory includes patients who had only one prior LOT and did not achieve CR, and patients who had ≥ one prior LOT and did not achieve CR or PR in their latest LOT. Relapsed (including progressed) includes patients that had CR as best response in their latest LOT and relapsed, and patients who had ≥ one prior LOT and had PR as best response in their latest LOT and subsequently progressed.
Error bars are 95% CI. Numbers in the bars are the number of patients.

Figure 7. Kaplan-Meier estimated PFS (A) and OS (B)

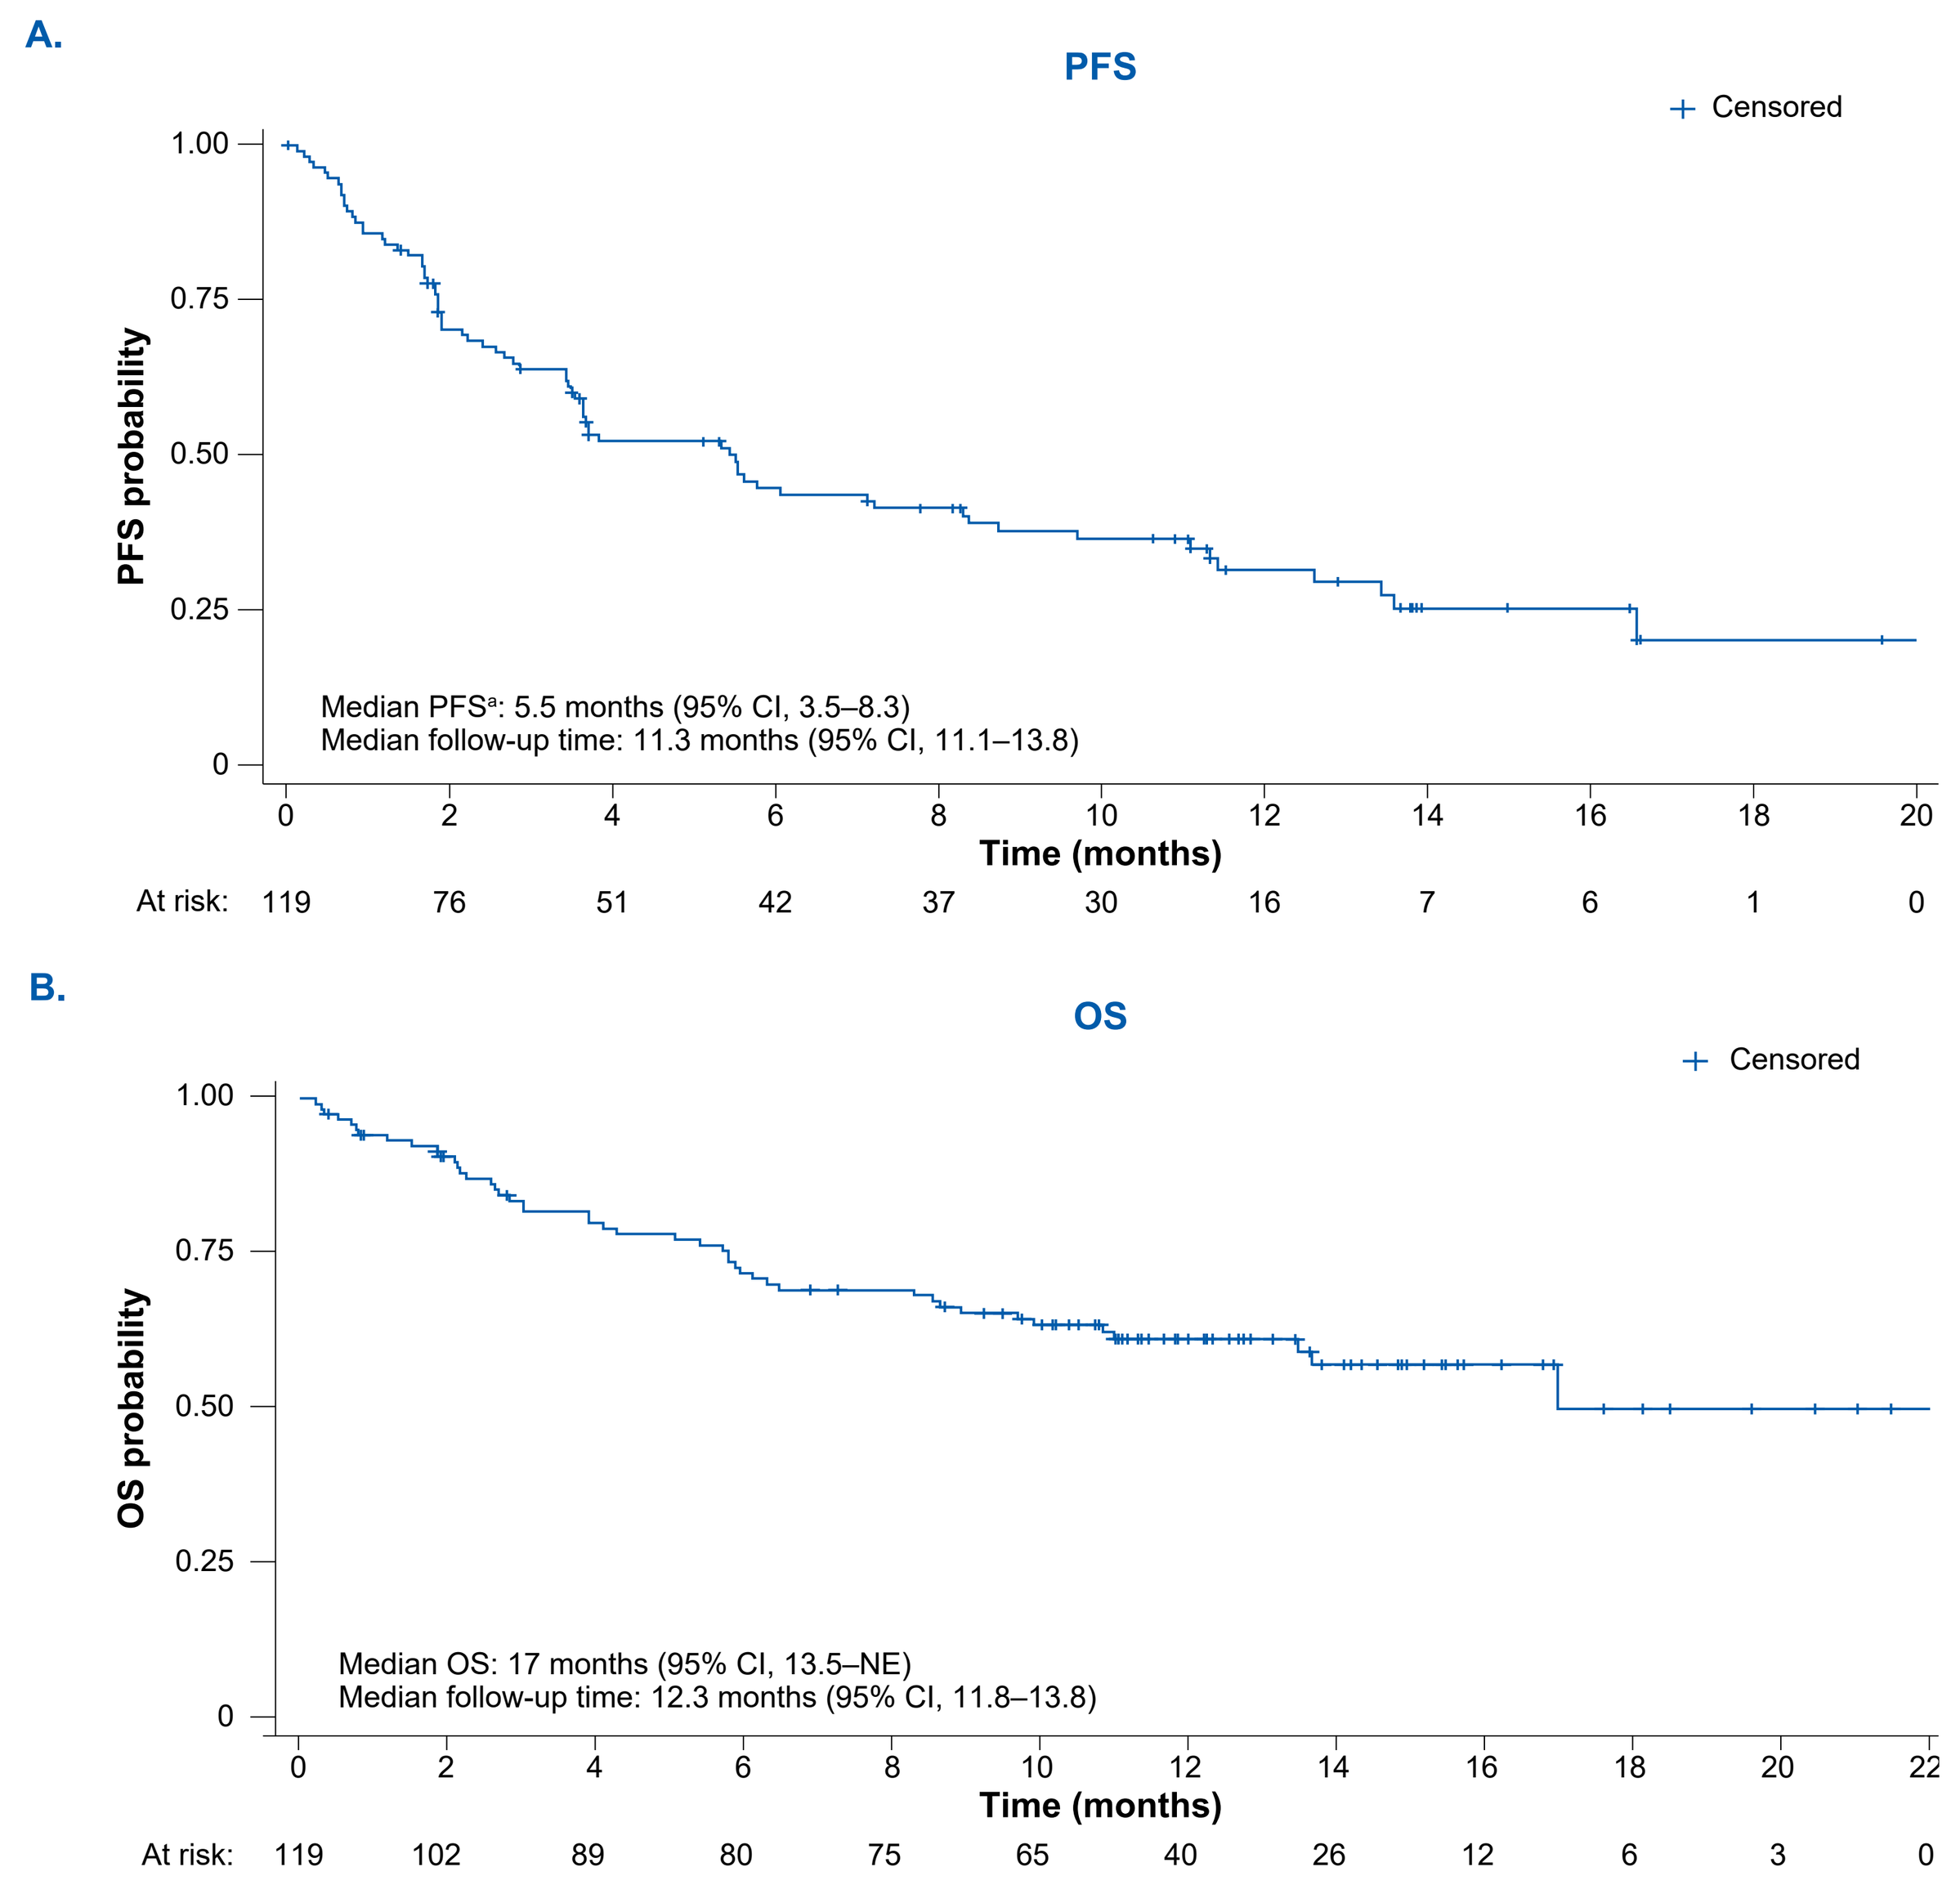
A.
PFS
Censored
PFS probability
Median PFS[a]: 5.5 months (95% CI, 3.5–8.3)
Median follow-up time: 11.3 months (95% CI, 11.1–13.8)
Time (months)
At risk: 119 76 51 42 37 30 16 7 6 1 0
B.
OS
Censored
OS probability
Median OS: 17 months (95% CI, 13.5–NE)
Median follow-up time: 12.3 months (95% CI, 11.8–13.8)
Time (months)
At risk: 119 102 89 80 75 65 40 26 12 6 3 0

[a]PFS evaluated by BICR CT-based assessment.

Safety and tolerability

- Almost all patients (128/133; 96.2%) experienced ≥ 1 any grade TEAE (**Table 2**)
- Grade ≥ 3 TEAEs occurred in 57.9% (77/133) of patients
- TEAEs led to treatment discontinuation, dose reduction, or dose interruption in 9.8% (13/133), 15.8% (21/133), and 49.6% (66/133) of patients, respectively (**Table 3**)
- Cytopenias were common (**Figure 8**), and were manageable with dose modifications and/or supportive therapies such as transfusions and granulocyte colony-stimulating factor
 - Thrombocytopenia was the most frequent any grade (49.6%; 66/133) and Grade ≥ 3 (23.3%; 31/133) TEAE
 - Median time to first onset of Grade ≥ 3 thrombocytopenia (platelet count < 50×10^9/L) was 18 days from first dose
 - Median time to platelet count recovery was 12 days
- Two patients developed secondary acute myeloid leukemia and discontinued treatment

Table 2. TEAE and TRAE summary

Event, %	PTCL (N = 133)
Any TEAE	96.2
Any TRAE	79.7
Grade ≥ 3 TEAEs	57.9
Grade ≥ 3 TRAEs	36.8
Serious TEAEs	39.8
Serious TRAEs	6.8
TEAEs leading to death	11.3
TRAEs leading to death	0
TEAEs leading to treatment discontinuation	9.8
TRAEs leading to treatment discontinuation	6.8
TEAEs leading to dose reduction	15.8
TRAEs leading to dose reduction	12.0
TEAEs leading to dose interruption	49.6
TRAEs leading to dose interruption	31.6

TRAE, treatment-related adverse event.

Table 3. TEAEs that led to dose modifications

	TEAEs that led to dose modifications[a] (N = 133)		
Preferred term	Treatment discontinuation %	Dose reduction %	Dose interruption %
Any TEAE	**9.8**	**15.8**	**49.6**
Thrombocytopenia[b]	2.3	5.3	16.5
Anemia[c]	0	3.8	9.8
COVID-19	0	1.5	8.3
Neutropenia[d]	0	2.3	5.3

[a]The table includes TEAEs that led to treatment interruption in ≥ 5% of patients. [b]Thrombocytopenia includes platelet count decrease. [c]Anemia includes hemoglobin decrease, and red blood cell count decrease. [d]Neutropenia includes neutrophil count decrease.

Figure 8. TEAEs that occurred in ≥ 15% of patients

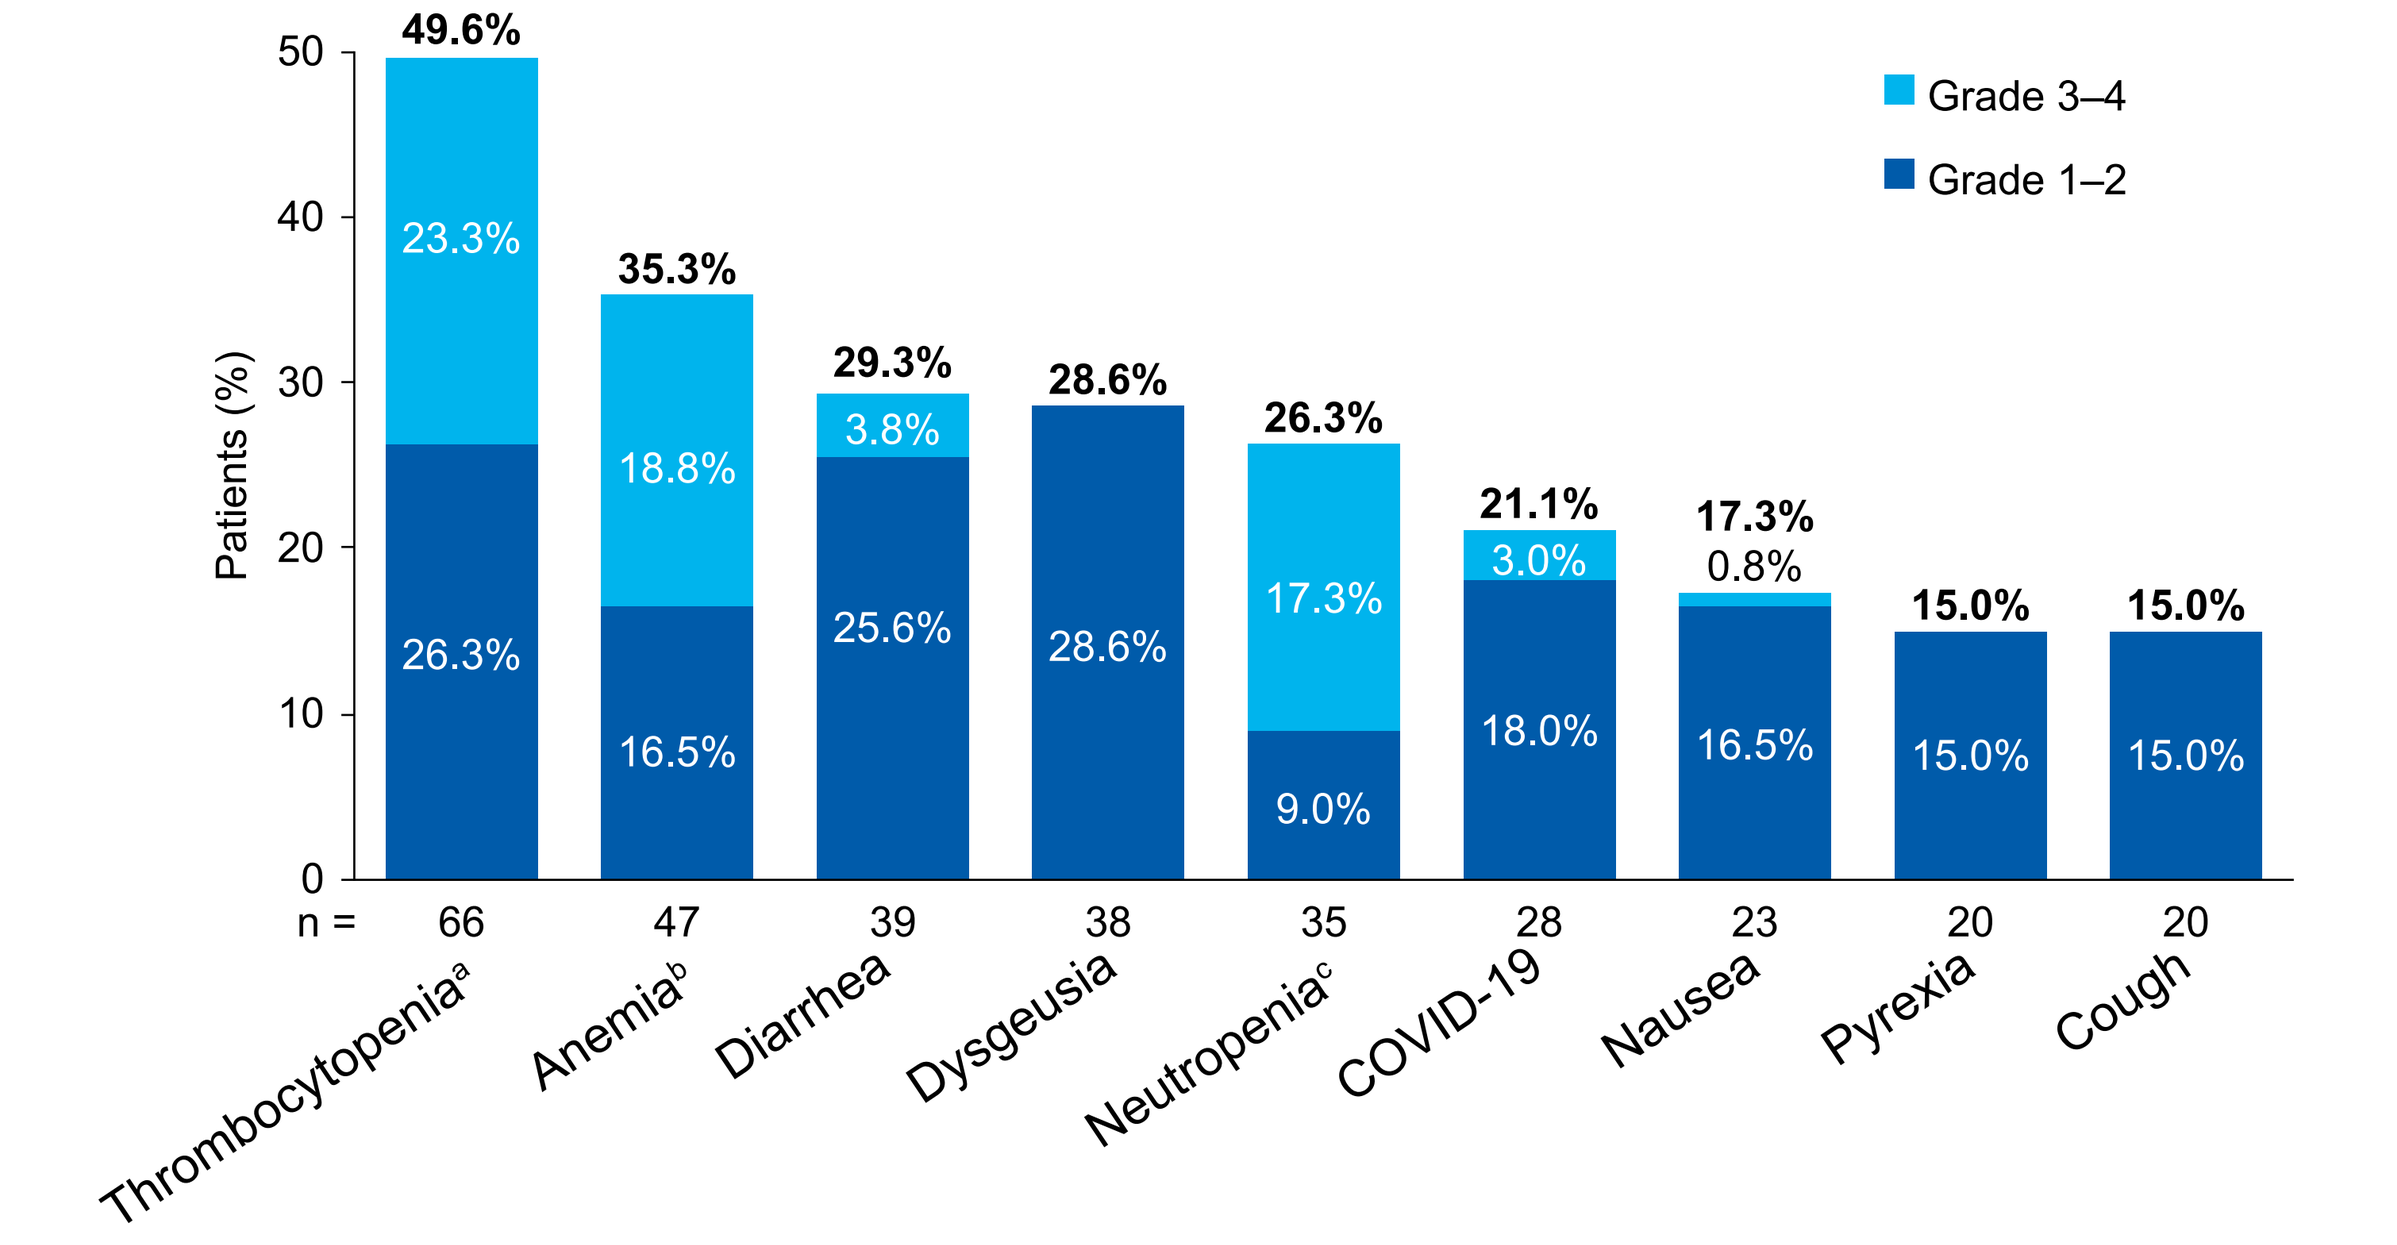
Grade 3–4
Grade 1–2
Patients (%)
Thrombocytopenia[a] (n = 66): 49.6%; 23.3%; 26.3%
Anemia[b] (n = 47): 35.3%; 18.8%; 16.5%
Diarrhea (n = 39): 29.3%; 3.8%; 25.6%
Dysgeusia (n = 38): 28.6%; 28.6%
Neutropenia[c] (n = 35): 26.3%; 17.3%; 9.0%
COVID-19 (n = 28): 21.1%; 3.0%; 18.0%
Nausea (n = 23): 17.3%; 0.8%; 16.5%
Pyrexia (n = 20): 15.0%; 15.0%
Cough (n = 20): 15.0%; 15.0%

[a]Thrombocytopenia includes platelet count decrease. [b]Anemia includes hemoglobin decrease, and red blood cell count decrease. [c]Neutropenia includes neutrophil count decrease.

REFERENCES

1. Vose J, et al. *J Clin Oncol* 2008;26:4124–4130.
2. Ling L, et al. *Br J Haematol* 2017;178:772–780.
3. Sibon D, et al. *Cancers* 2022;14:2332.
4. Herviou L, et al. *Oncotarget* 2016;7:2284–2296.
5. Yamagishi M, et al. *Cell Rep* 2019;29:2321–2337.
6. Shen X, et al. *Mol Cell* 2008;32:491–502.
7. Nakagawa M, Kitabayashi I. *Cancer Sci* 2018;109:2342–2348.
8. EZHARMIA® (valemetostat tosilate). [package insert]. Tokyo, Japan: Daiichi Sankyo; 2024.
9. Swerdlow SH, et al. *Blood* 2016;127:2375–2390.
10. Cheson BD et al. *J Clin Oncol* 2014;32:3059–30.

ACKNOWLEDGMENTS

- The authors thank the patients, families, and caregivers for their participation, and the study staff for their contributions
- This study is sponsored by Daiichi Sankyo Co., Ltd.
- Editorial support was provided by Declan Grewcock, PhD, of Excerpta Medica, funded by Daiichi Sankyo Co., Ltd., and in accordance with Good Publication Practice guidelines

Presented at the 12th Annual Meeting of the Society of Hematologic Oncology (SOHO); September 4–7; Houston, TX, US.