A Phase 1 Trial of Valemetostat in Patients With Relapsed or Refractory (R/R) Acute Myeloid Leukemia (AML) or Acute Lymphocytic Leukemia (ALL)

AML-385

Harry P. Erba,[1] Olalekan O. Oluwole,[2] Dale Bixby,[3] Raajit K. Rampal,[4] Amir T. Fathi,[5] Prashni Paliwal,[6] Jin Jin,[6] Ai Inoue,[6]* Naval Daver[7]

[1]Duke Cancer Institute, Durham, NC; [2]Vanderbilt-Ingram Cancer Center, Nashville, TN; [3]University of Michigan Rogel Cancer Center, Ann Arbor, MI; [4]Memorial Sloan-Kettering Cancer Center, New York, NY; [5]Harvard Medical School and Massachusetts General Hospital Cancer Center, Boston, MA; [6]Daiichi Sankyo Inc., Basking Ridge, NJ; [7]The University of Texas MD Anderson Cancer Center, Houston, TX.

*Former employee at the time of the study.

PURPOSE

- To determine the safety, tolerability, and maximum tolerated dose (MTD) and recommended dose for expansion (RDE) of valemetostat tosylate (valemetostat) in patients with relapsed or refractory (R/R) acute myeloid leukemia (AML) or acute lymphocytic leukemia (ALL) enrolled in a phase 1 clinical trial

CONCLUSIONS

- Valemetostat monotherapy demonstrated acceptable and manageable safety in patients with R/R AML and R/R ALL
 - The most common treatment-emergent adverse events (TEAEs) were mainly gastrointestinal events (diarrhea, nausea, decreased appetite, and vomiting)
 - The most common Grade ≥ 3 TEAEs and most common serious adverse events (SAEs) were mainly infections (febrile neutropenia, pneumonia, sepsis, and cellulitis)
- Orally administered valemetostat was rapidly absorbed; plasma concentration was dose-dependent, and the time to maximum plasma concentration (T_{max}) was reached in 2–4 hours
- Preliminary efficacy analyses showed 2 responders with AML (1 complete remission with incomplete blood count recovery [CRi] and 1 partial response [PR]; n = 22), and 1 responder with ALL (complete response [CR]; n = 6)

Copies of this presentation obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this material.

BACKGROUND

- Despite relatively high rates of initial response with induction therapies, up to 80% of patients with AML eventually relapse or develop refractory disease[1,2]
- Alternative treatment options are needed to improve outcomes for patients with R/R AML[3–5]
- Enhancer of zeste homolog (EZH)2 and its close homolog EZH1 catalyze the trimethylation of histone H3 at lysine 27 (H3K27me3) (**Figure 1**); overexpression of *EZH2* and global H3K27me3 accumulation result in abnormal epigenetic regulation and altered transcriptome in various cancers[6–8]
 - *EZH2* plays a dual role as a tumor suppressor in normal hematopoiesis and as an oncogene in lymphoma and AML[6,7]

Figure 1. Mechanism of action of valemetostat

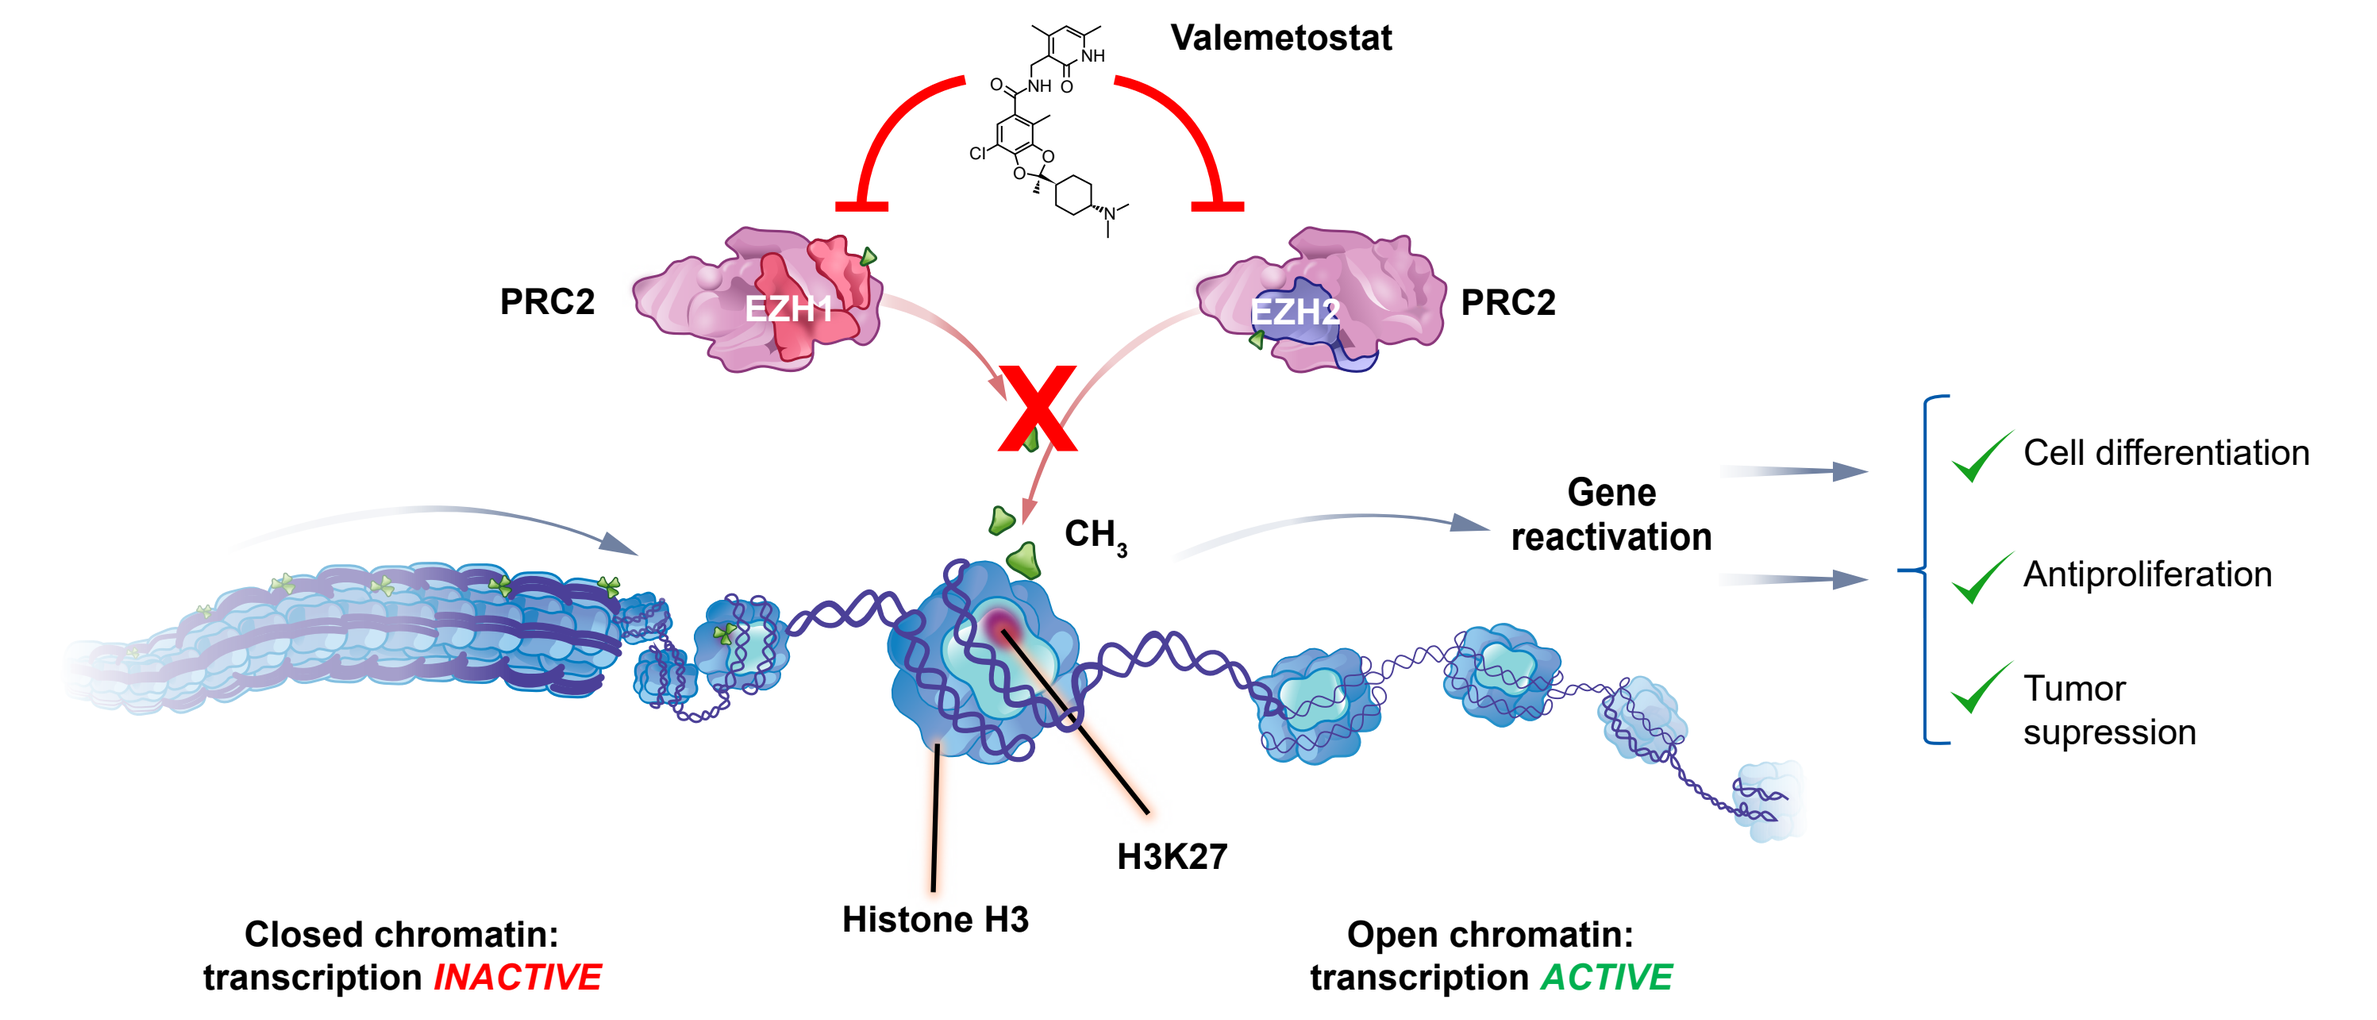
Valemetostat
PRC2
EZH1
EZH2
PRC2
CH3
Gene reactivation
Cell differentiation
Antiproliferation
Tumor supression
H3K27
Histone H3
Closed chromatin: transcription INACTIVE
Open chromatin: transcription ACTIVE

CH3, trimethyl group; PRC2, polycomb repressive complex 2.

- Valemetostat is a novel, potent, and selective dual inhibitor of EZH2 and EZH1 that suppresses aberrant H3K27me3, thereby promoting antitumorigenic processes[9,10]
 - Valemetostat monotherapy is approved in Japan for the treatment of R/R adult T-cell leukemia/lymphoma and peripheral T-cell lymphoma[11]
- In preclinical models of AML, valemetostat has been shown to reduce leukemic stem cells (LSCs) and delay disease progression
 - Quiescent LSCs are highly dependent on EZH1 and EZH2; valemetostat has a synergistic effect against LSCs[12]
- The phase 1, non-randomized, multicenter, open-label, dose-escalation and -expansion DS3201-A-U102 study (NCT03110354) assessed valemetostat monotherapy in patients with R/R AML or ALL

METHODS

Study design

- Patients aged ≥ 18 years with confirmed AML or ALL (World Health Organization 2008 criteria[13]) that had failed any prior induction therapy regimen or had relapsed after prior therapy were enrolled at 6 sites in the United States
- The trial prospectively included a dose-escalation part followed by a dose-expansion part (**Figure 2**)
 - The dose-escalation part assessed valemetostat at escalating doses of 100–700 mg/day in continuous 28-day treatment cycles; dose-escalation was guided by a Bayesian logistic regression model
 - The dose-expansion part would further assess valemetostat at RDE

Endpoints

- Primary endpoints:
 - Dose-escalation: safety and tolerability assessment of valemetostat in patients with R/R AML or ALL, and determination of MTD and RDE
 - Dose-expansion: confirmation of safety and tolerability of valemetostat at the RDE in patients with R/R AML or ALL

Figure 2. Study design, key inclusion criteria, and endpoints of the U102 trial

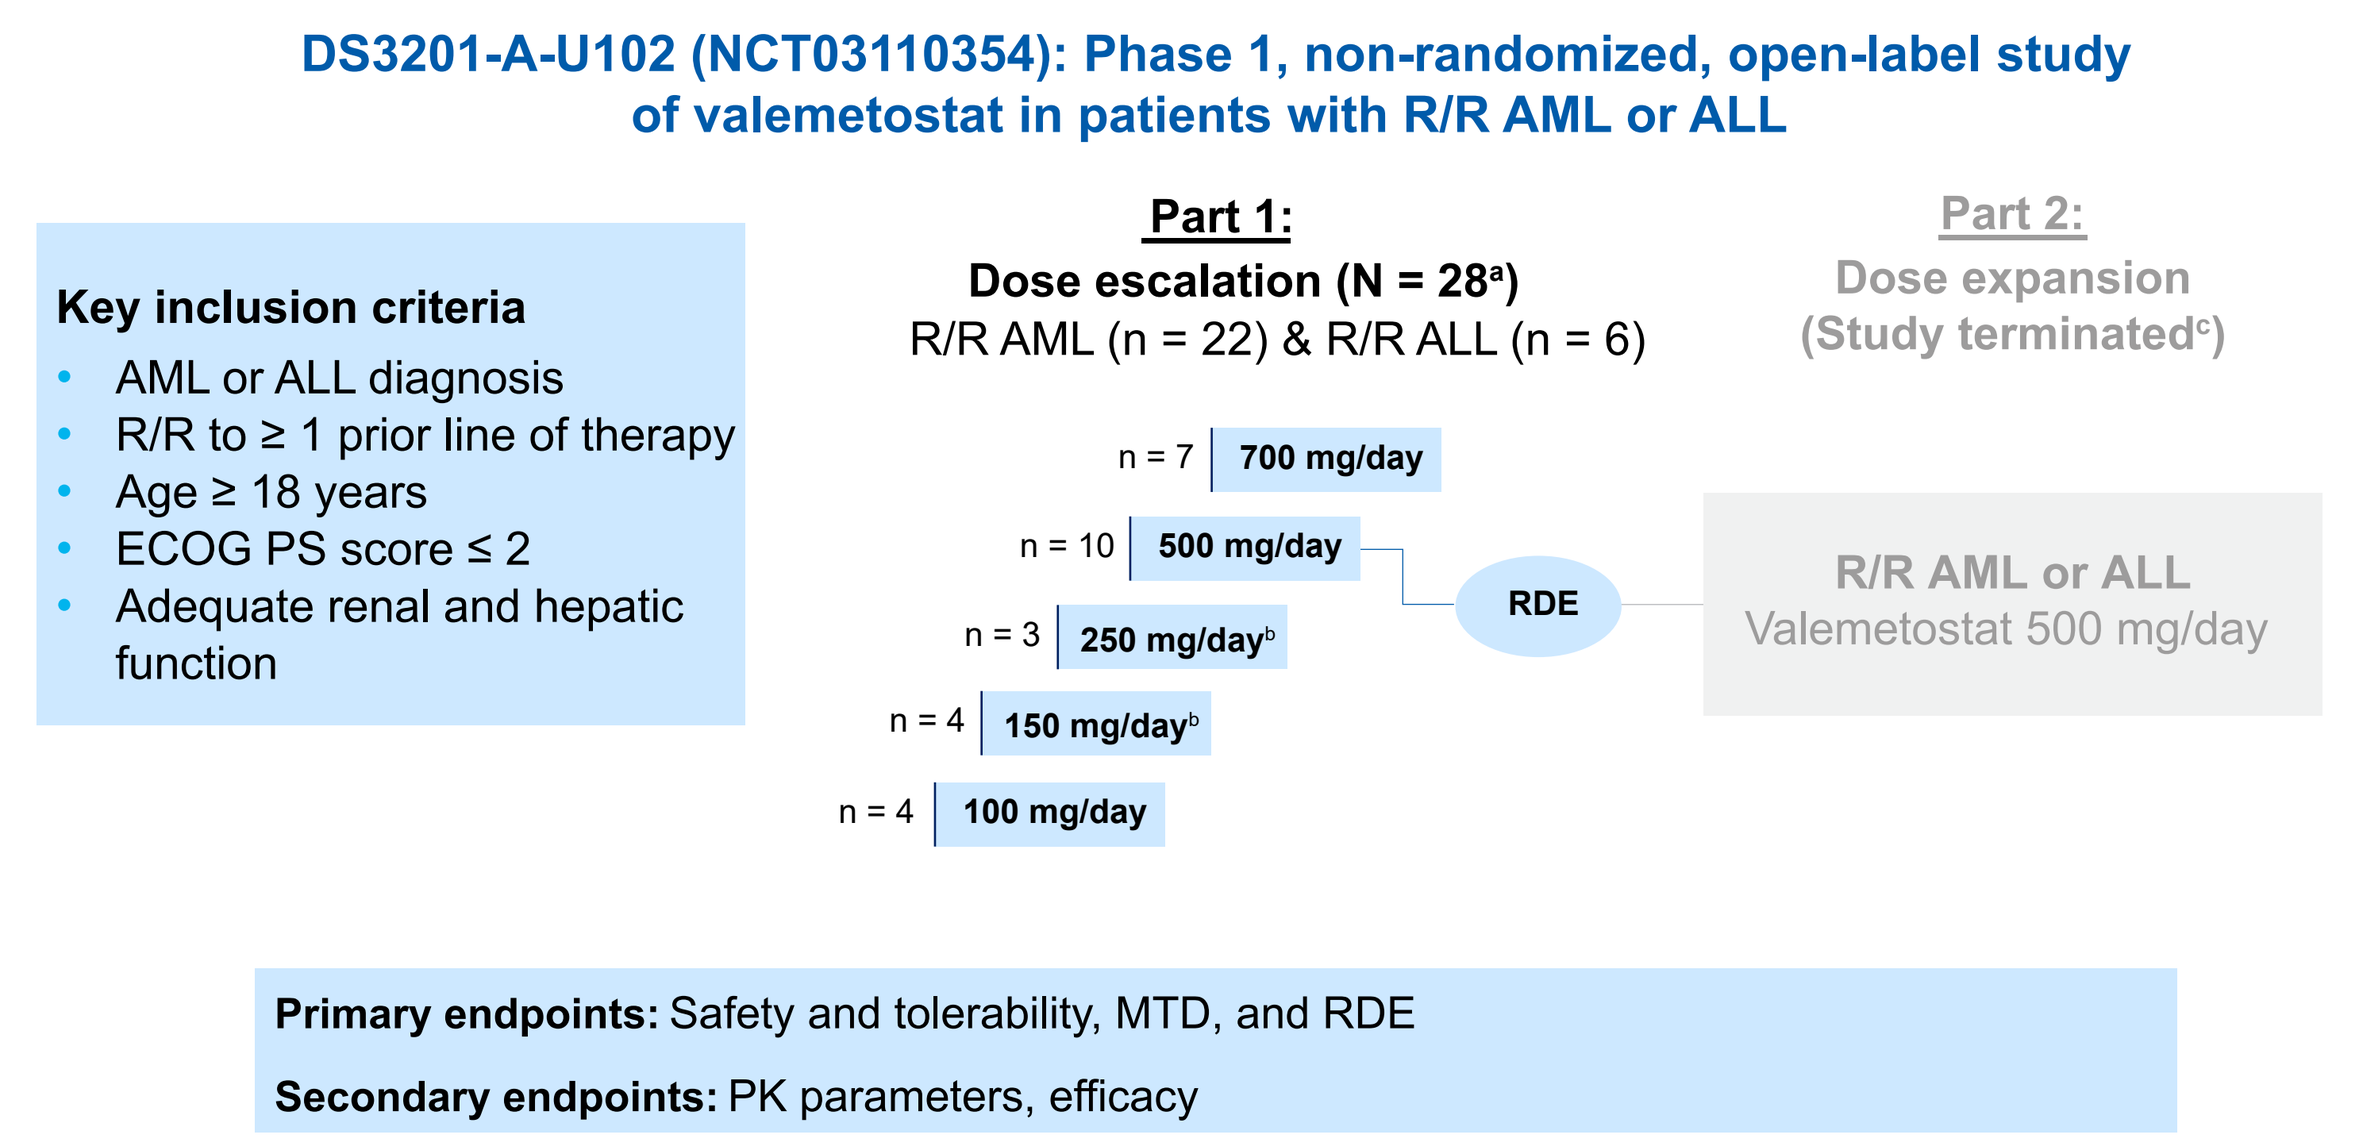
DS3201-A-U102 (NCT03110354): Phase 1, non-randomized, open-label study of valemetostat in patients with R/R AML or ALL
Key inclusion criteria
AML or ALL diagnosis
R/R to ≥ 1 prior line of therapy
Age ≥ 18 years
ECOG PS score ≤ 2
Adequate renal and hepatic function
Part 1:
Dose escalation (N = 28a)
R/R AML (n = 22) & R/R ALL (n = 6)
Part 2:
Dose expansion
(Study terminatedc)
n = 7 700 mg/day
n = 10 500 mg/day
n = 3 250 mg/dayb
n = 4 150 mg/dayb
n = 4 100 mg/day
RDE
R/R AML or ALL
Valemetostat 500 mg/day
Primary endpoints: Safety and tolerability, MTD, and RDE
Secondary endpoints: PK parameters, efficacy

[a]All the enrolled patients were included in the safety and PK data sets; 1 patient in the 500 mg cohort and 1 patient in the 700 mg cohort were excluded from the full analysis set; [b]All patients in the 150 mg and 250 mg cohorts had AML; [c]This study was terminated at the end of the dose-escalation part due to slow accrual.
ALL, acute lymphocytic leukemia; AML, acute myeloid leukemia; CTCAE, Common Terminology Criteria for Adverse Events; ECOG PS, Eastern Cooperative Oncology Group performance status; MTD, maximum tolerated dose; PK, pharmacokinetics; RDE, recommended dose for expansion; R/R, relapsed/refractory.

- Secondary endpoints included pharmacokinetics (PK), overall response rate (ORR), duration of response (DOR), and overall survival (OS)
- Safety was assessed by investigator reporting of TEAEs from first dose through 30 days after last dose of study drug
 - In the dose-escalation part, safety assessment for dose-limiting toxicities (DLTs) was performed after cycle 1, day 28
- Clinical response was assessed in bone marrow aspirates collected at screening, on day 8, and at the start of each subsequent treatment cycle
 - Clinical responses were defined using revised International Working Group (IWG) 2003 response criteria for AML[14] or National Comprehensive Cancer Network (NCCN) 2016 response criteria for ALL[15]
 - ORR was the proportion of patients achieving CR, CRi/complete remission with incomplete platelet recovery (CRp), or PR
 - DOR was defined as the time from first objective response until confirmed disease progression
- OS was defined as the time from enrollment until death from any cause

RESULTS

Patient enrollment and disposition

- Overall, 28 patients with R/R AML (n = 22) and ALL (n = 6) were enrolled from April 2017 through March 2021, all in the dose-escalation portion
- Median treatment duration at data cutoff (March 9, 2021) was 34 days (range, 4–331)
- At data cutoff, all 28 patients had discontinued study treatment; the primary reasons for discontinuation were progressive disease (n = 11, 39%), failure to achieve response (n = 8, 29%), TEAEs (n = 4, 14%), death (n = 2, 7%), start of new therapy (n = 1, 4%), and other (n = 2, 7%)

Demographics and baseline characteristics

- Median age was 61.5 years (range 20–85; **Table 1**)
- Median prior lines of therapy was 3 (range 1–10) for patients with AML and was 2 (range 2–3) for patients with ALL
- Most patients (n = 21, 75%) had an Eastern Cooperative Oncology Group performance status (ECOG PS) score of 1 at baseline

Safety and tolerability

- All 28 patients experienced at least 1 TEAE (**Table 2**)
 - Grade ≥ 3 TEAEs were reported in 24 (86%) patients, including treatment-related Grade ≥ 3 events in 7 (25%) patients
- TEAEs required treatment discontinuation for 7 (25%) patients and dose reduction for 3 (11%) patients

Table 1. Baseline demographics

Characteristic	Total (N = 28)
Age, years, median (range)	61.5 (20–85)
Sex, n (%)	
Male	10 (36)
Female	18 (64)
Race, n (%)	
White	22 (79)
Black or African American	2 (7)
Asian	2 (7)
Other	2 (7)
ECOG PS score, n (%)	
0	3 (11)
1	21 (75)
2	4 (14)
Prior lines of therapy, median (range)	2 (1–10)
Time since diagnosis, days, median (range)	536 (40–1548)

ECOG PS, Eastern Cooperative Oncology Group performance status.

Table 2. Overview of TEAEs in the valemetostat dose-escalation trial

Parameters	Valemetostat dose					All dose levels
	100 mg/day (n = 4)	150 mg/day (n = 4)	250 mg/day (n = 3)	500 mg/day (n = 10)	700 mg/day (n = 7)	(N = 28)
Any TEAE, n (%)	4 (100)	4 (100)	3 (100)	10 (100)	7 (100)	28 (100)
Grade ≥ 3 TEAE	3 (75)	3 (75)	3 (100)	9 (90)	6 (86)	24 (86)
TEAEs leading to dose reduction	0	0	0	1 (10)	2 (29)	3 (11)
TEAEs leading to treatment discontinuation	2 (50)	0	0	3 (30)	2 (29)	7 (25)
Any TRAE, n (%)	2 (50)	2 (50)	1 (33)	5 (50)	6 (86)	16 (57)
Grade ≥ 3 TRAE	0	1 (25)	0	3 (30)	3 (43)	7 (25)
TRAEs leading to dose reduction	0	0	0	1 (10)	2 (29)	3 (11)
TRAEs leading to treatment discontinuation	0	0	0	2 (20)	1 (14)	3 (11)
Any SAE, n (%)	3 (75)	3 (75)	3 (100)	7 (70)	6 (86)	22 (79)
Grade ≥ 3 SAE	3 (75)	3 (75)	3 (100)	7 (70)	6 (86)	22 (79)
Treatment-related SAE, n (%)	0	1 (25)	0	2 (20)	2 (29)	5 (18)
Grade ≥ 3 treatment-related SAE	0	1 (25)	0	2 (20)	2 (29)	5 (18)

SAE, serious adverse event; TEAE, treatment-emergent adverse event; TRAE, treatment-related TEAE.

- Fatal (Grade 5) TEAEs were reported in 3 patients, including (n = 1 each) respiratory failure and sepsis, intracranial hemorrhage, and sepsis; these were not considered to be related to study drug
- The most common TEAEs were diarrhea and nausea (43% each), followed by vomiting and decreased appetite (36% each) (**Table 3**)
- SAEs occurred in 22 (79%) patients (**Table 4**)
- One patient experienced 2 events of differentiation syndrome as confirmed by Montesinos criteria[16]

Table 3. TEAEs (all grades) that occurred in ≥ 20% of patients

Preferred term	Valemetostat dose, n (%)					All dose levels
	100 mg/day (n = 4)	150 mg/day (n = 4)	250 mg/day (n = 3)	500 mg/day (n = 10)	700 mg/day (n = 7)	(N = 28)
Diarrhea	0	2 (50)	2 (67)	4 (40)	4 (57)	12 (43)
Nausea	1 (25)	2 (50)	1 (33)	4 (40)	4 (57)	12 (43)
Vomiting	0	1 (25)	0	6 (60)	3 (43)	10 (36)
Decreased appetite	2 (50)	0	1 (33)	2 (20)	5 (71)	10 (36)
Febrile neutropenia	1 (25)	3 (75)	1 (33)	2 (20)	2 (29)	9 (32)
Hypomagnesemia	0	1 (25)	1 (33)	4 (40)	2 (29)	8 (29)
Dysgeusia	0	0	1 (33)	4 (40)	2 (29)	7 (25)
Headache	2 (50)	1 (25)	0	1 (10)	3 (43)	7 (25)
Hypokalemia	0	2 (50)	0	2 (20)	2 (29)	6 (21)
Hypotension	0	0	1 (33)	2 (20)	3 (43)	6 (21)
Pneumonia	3 (75)	2 (50)	0	1 (10)	0	6 (21)

TEAE, treatment-emergent adverse event.

Table 4. Summary of SAEs

Preferred term	Valemetostat dose					All dose levels
	100 mg/day (n = 4)	150 mg/day (n = 4)	250 mg/day (n = 3)	500 mg/day (n = 10)	700 mg/day (n = 7)	(N = 28)
Any treatment-emergent SAE, n (%)	3 (75)	3 (75)	3 (100)	7 (70)	6 (86)	**22 (79)**[c]
SAEs reported in ≥ 3 patients, n (%)						
Febrile neutropenia	1 (25)	2 (50)	1 (33)	2 (20)	2 (29)	8 (29)
Pneumonia	3 (75)	2 (50)	0	1 (10)	0	6 (21)
Sepsis	1 (25)	1 (25)	0	1 (10)	1 (14)	4 (14)
Cellulitis	0	1 (25)	0	0	2 (29)	3 (11)

SAE, serious adverse event.

Maximum Tolerated Dose

- The 500 mg/day dose level was determined to be the MTD and selected as the RDE, but the study was closed following completion of the dose-escalation due to low enrollment

PK parameters

- Valemetostat was rapidly absorbed, and plasma concentration increased proportionately to the administered dose (**Figure 3**)
- Median T_{max} ranged from 2 to 4 hours

Figure 3. Linear and semi-log mean valemetostat plasma concentrations by dose level on cycle 1 day 1

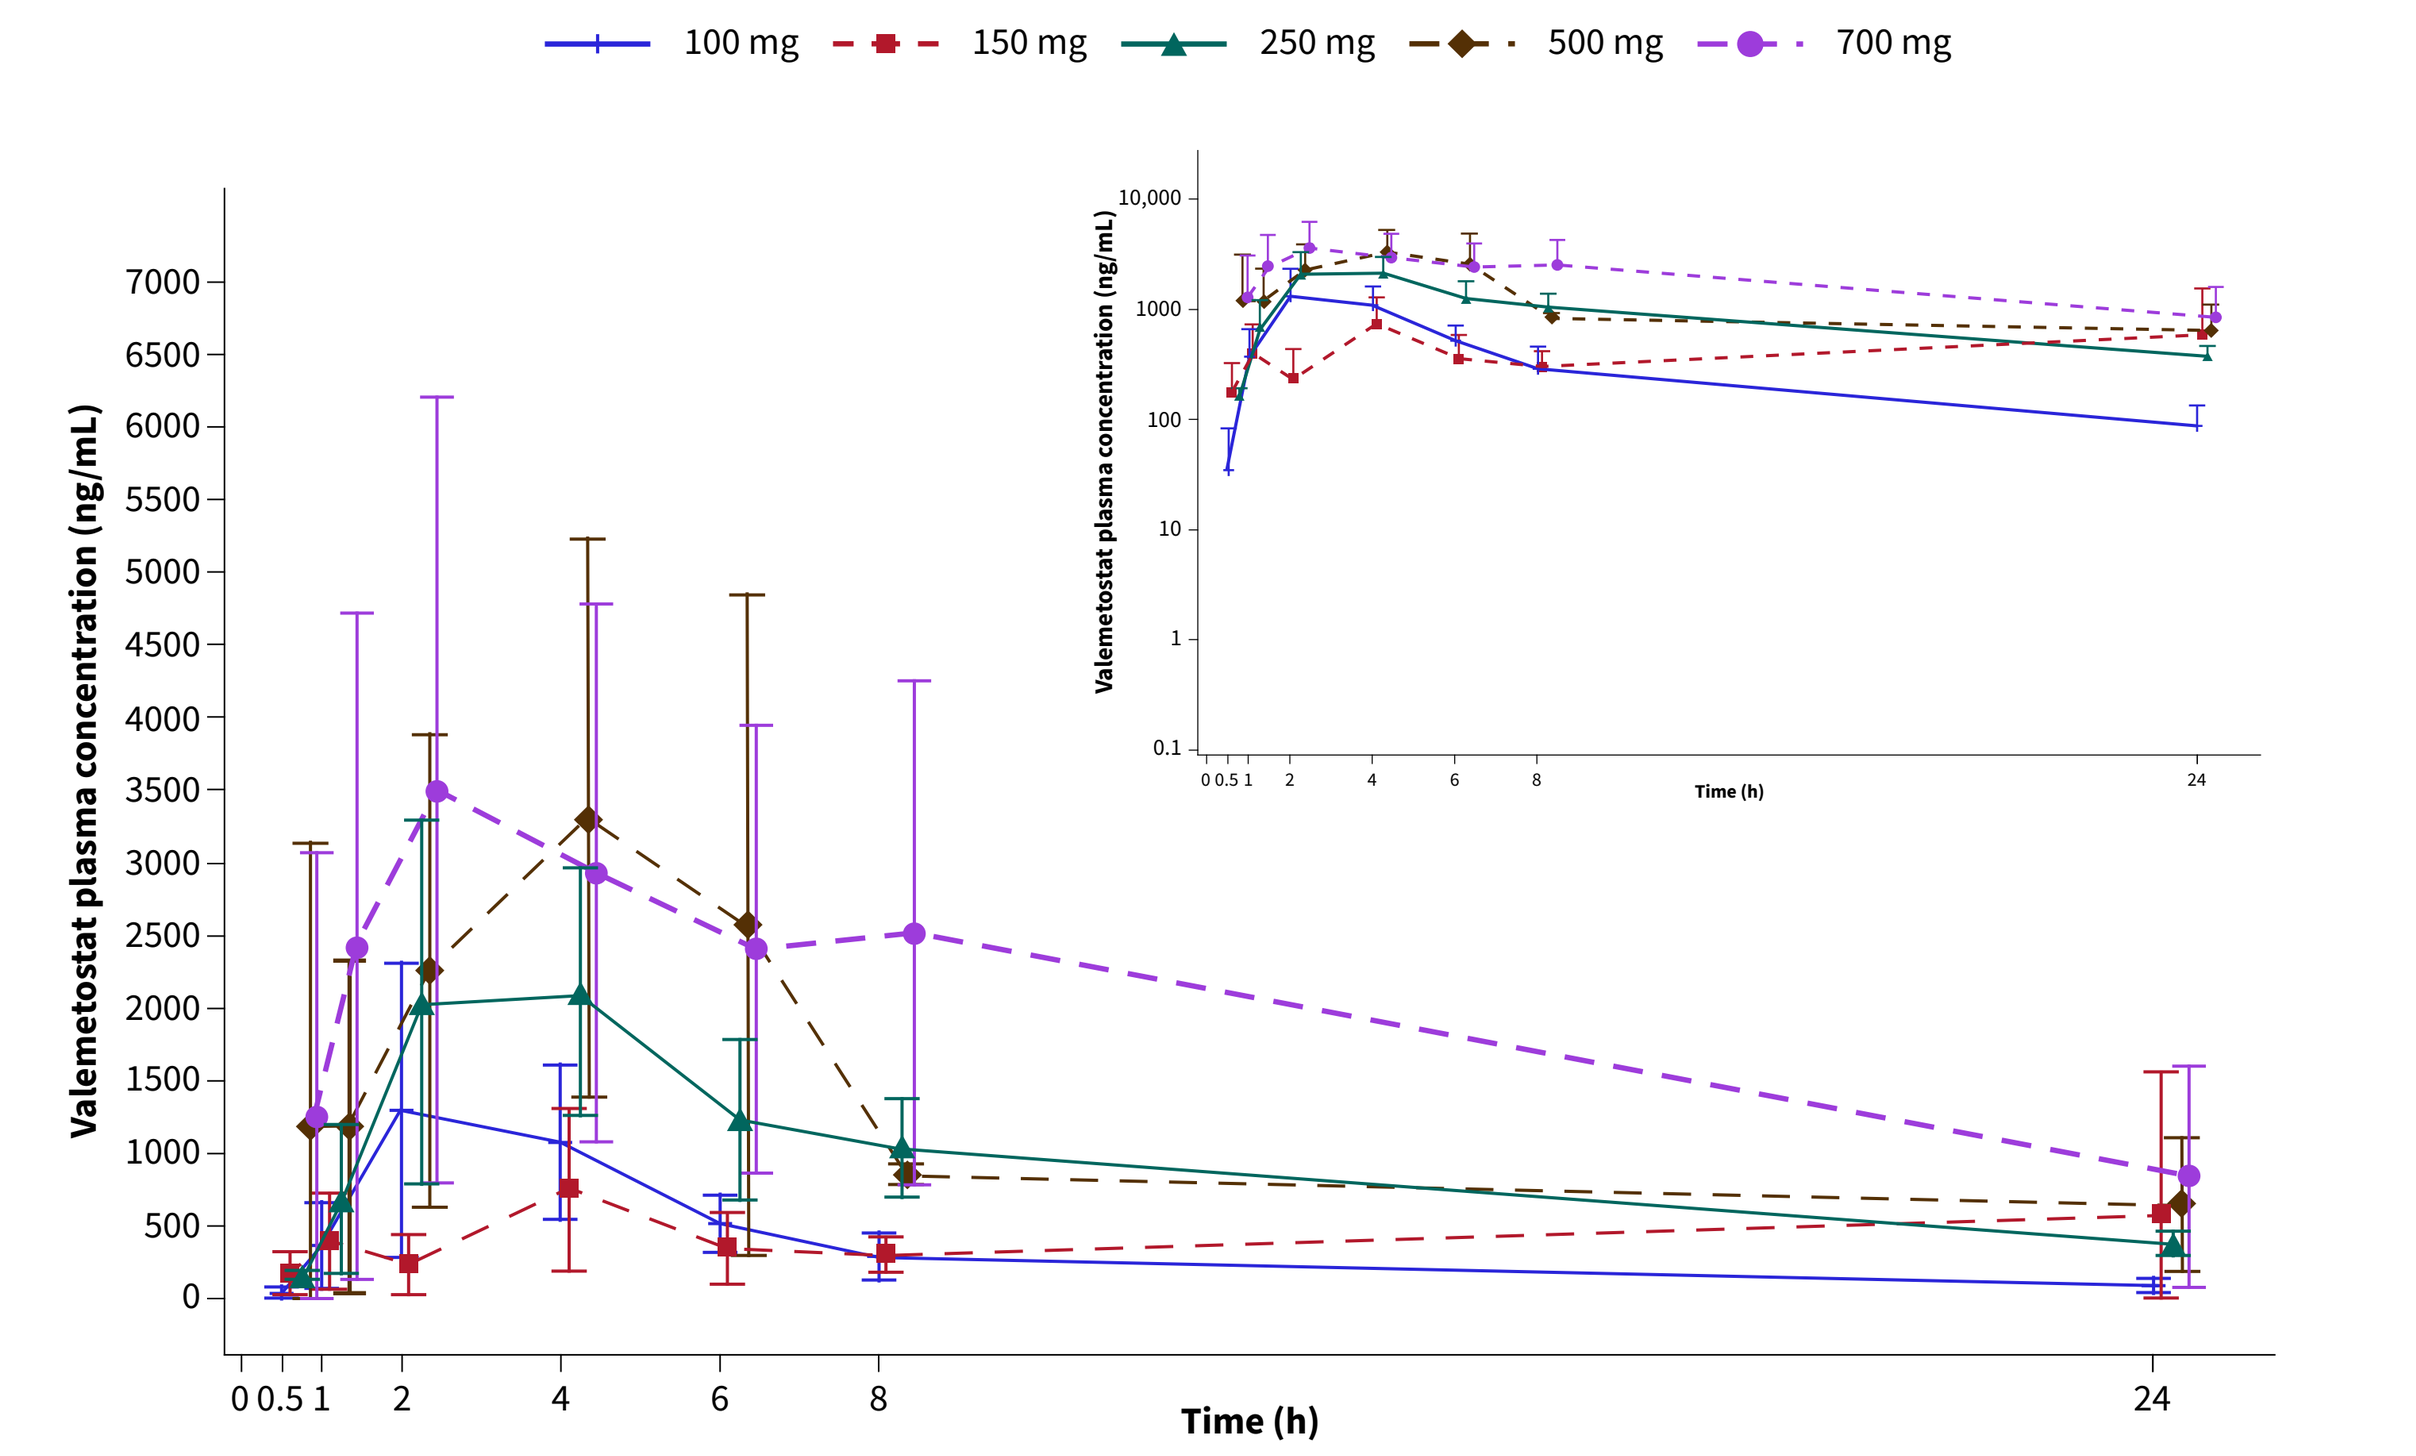
100 mg
150 mg
250 mg
500 mg
700 mg
Valemetostat plasma concentration (ng/mL)
Time (h)
0 0.5 1 2 4 6 8 24
10,000
1000
100
10
1
0.1

Efficacy

- The efficacy-evaluable set included 26 (93%) patients
 - Two patients (1 each in the 500 mg and 700 mg cohorts) were excluded from the efficacy analyses due to missing post-baseline bone marrow assessments
- In the AML cohort (n = 20), 2 patients achieved CRi (n = 1) and PR (n = 1); both responding patients were in the 500 mg dose cohort and had core binding factor AML (CBF-AML)
- In the ALL cohort (n = 6), 1 patient in the 700 mg cohort achieved CR

REFERENCES

1. Patel A, et al. *Oncol Res* 2021;28(7):811–814.
2. Medeiros BC, et al. *Am J Hematol* 2019;94(7):803–811.
3. Roman Diaz JL, et al. *Cancers* 2024;16(3):677.
4. Wysota M, et al. *Curr Oncol Rep* 2024;26(4):409–420.
5. Thol F, Heuser M. *HemaSphere* 2021;5(6):e572.
6. Nakagawa M, Kitabayashi I *Cancer Sci* 2018;109(8):2342–2348.
7. Herviou L, et al. *Oncotarget* 2016;7(3):2284–2296.
8. Yamagishi M, et al. *Cell Rep* 2019;29(8):2321-2337.e7.
9. Izutsu K, et al. *Blood* 2023;141(10):1159–1168.
10. Keam SJ. *Drugs* 2022;82(16):1621–1627.
11. EZHARMIA® (valemetostat tosilate). [package insert]. Tokyo, Japan: Daiichi Sankyo; 2022.
12. Fujita S, et al. *Leukemia* 2018;32(4):855–864.
13. Vardinam JW, et al. *Blood* 2009;114(5):937–951.
14. Cheson BD, et al. *J Clin Oncol* 2003;21(24):4642–4649.
15. Referenced with permission from the NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for Acute Lymphoblastic Leukemia V.1.2016. © National Comprehensive Cancer Network, Inc. 2024. All rights reserved. To view the most recent and complete version of the guideline, go online to NCCN.org
16. Norsworthy KJ, et al. *Clin Cancer Res* 2020;26(16):4280–4288.

ACKNOWLEDGMENTS

- The authors thank the patients, families, and caregivers for their participation, and the study staff for their contributions
- This study is sponsored by Daiichi Sankyo Co., Ltd.
- Editorial support was provided by Luca Scrivano, PhD, and Brian Kaiser, PhD, of Excerpta Medica, funded by Daiichi Sankyo Co., Ltd., and in accordance with Good Publication Practice guidelines

Presented at the 12th Annual Meeting of the Society of Hematologic Oncology (SOHO); September 4–7; Houston, TX, US.