QuANTUM-First Trial: *FMS*-Like Tyrosine Kinase 3-Internal Tandem Duplication (*FLT3*-ITD)–Specific Measurable Residual Disease (MRD) Clearance Assessed Through Induction and Consolidation Is Associated with Improved Overall Survival in Newly Diagnosed *FLT3*-ITD+ AML Patients

Alexander E. Perl,[1] Harry P. Erba,[2] Pau Montesinos,[3] Radovan Vrhovac,[4] Elżbieta Patkowska,[5] Hee-Je Kim,[6] Pavel Zak,[7] Po-Nan Wang,[8] Jaime E. Connolly Rohrbach,[9] Ken C.N. Chang,[9] Li Liu,[9] Yasser Mostafa Kamel,[9] Karima Imadalou,[9] Jorge Cortes,[10] Mikkael A. Sekeres,[11] Hervé Dombret,[12] Sergio Amadori,[13] Jianxiang Wang,[14] Richard F. Schlenk,[15] Mark J. Levis[16]

[1]University of Pennsylvania, Philadelphia, PA, USA; [2]Duke Cancer Institute, Durham, NC, USA; [3]La Fe University and Polytechnic Hospital, Valencia, Spain; [4]University Hospital Centre Zagreb, Zagreb, Croatia; [5]Institute of Hematology and Blood Transfusion, Warsaw, Poland; [6]Catholic Hematology Hospital, Seoul St. Mary's Hospital, College of Medicine, The Catholic University of Korea, Seoul, South Korea; [7]University Hospital Hradec Kralove, Hradec Kralove, Czechia; [8]Chang Gung Medical Foundation, Linkou, Taiwan; [9]Daiichi Sankyo, Inc, Basking Ridge, NJ, USA; [10]Georgia Cancer Center at Augusta University, Augusta, GA, USA; [11]Sylvester Cancer Center, University of Miami Health System, Miami, FL, USA; [12]Saint Louis Hospital, University of Paris, Paris, France; [13]Tor Vergata Polyclinic Hospital Rome, Rome, Italy; [14]Institute of Hematology and Blood Diseases Hospital, Tianjin, China; [15]Heidelberg University Hospital and German Cancer Research Center, Heidelberg, Germany; [16]Johns Hopkins University, Baltimore, MD, USA

AML-197

Copies of materials obtained through the QR code or /bit.ly link are for personal use only and may not be reproduced without written permission of the authors.

BACKGROUND

- FMS-like tyrosine kinase 3–internal tandem duplication (*FLT3*-ITD) mutations:
 – Common in acute myeloid leukemia (AML) and are a negative prognostic marker[1–3]
- 3 United States Food and Drug Administration- and/or European Medicines Agency-approved FLT3 inhibitors: midostaurin,[4] gilteritinib,[5] and quizartinib[6]
- Quizartinib:
 – Type II inhibitor[1,6] active against *FLT3*-ITD mutations[1,3]
 – More potent and selective than either midostaurin or gilteritinib[1,3]
 – Improved survival when added to induction, consolidation, and continuation therapy of newly diagnosed adults with *FLT3*-ITD+ AML[6]

Figure 1. Addition of Quizartinib to Intensive Induction, Consolidation, and Continuation Therapy Improved Overall Survival (OS) in QuANTUM-First Phase 3 Trial

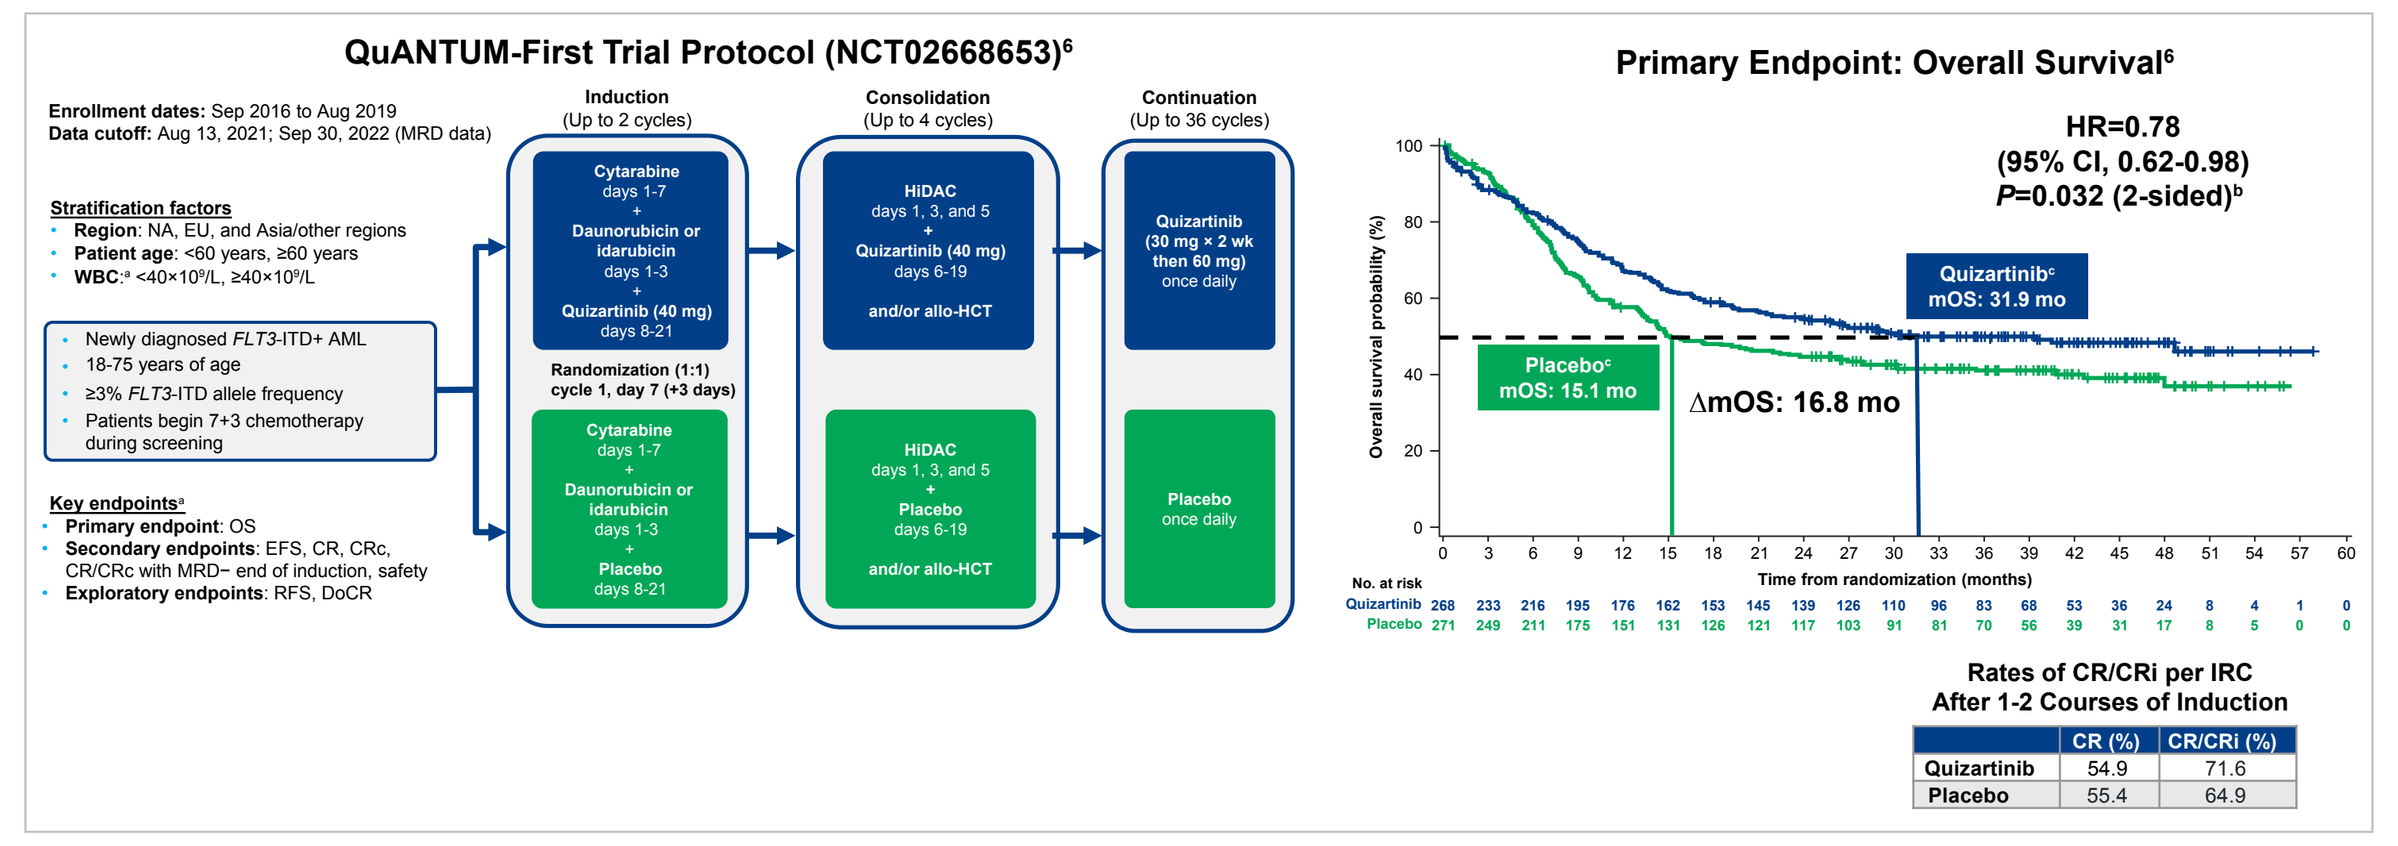
QuANTUM-First Trial Protocol (NCT02668653)[6]
Enrollment dates: Sep 2016 to Aug 2019
Data cutoff: Aug 13, 2021; Sep 30, 2022 (MRD data)
Induction (Up to 2 cycles)
Consolidation (Up to 4 cycles)
Continuation (Up to 36 cycles)
Randomization (1:1) cycle 1, day 7 (+3 days)
Primary Endpoint: Overall Survival[6]
HR=0.78
(95% CI, 0.62-0.98)
P=0.032 (2-sided)[b]
Quizartinib[c] mOS: 31.9 mo
Placebo[c] mOS: 15.1 mo
ΔmOS: 16.8 mo
Rates of CR/CRi per IRC After 1-2 Courses of Induction
CR (%) CR/CRi (%)
Quizartinib 54.9 71.6
Placebo 55.4 64.9

[a]A hierarchical testing procedure was used to test the primary endpoint of OS, followed by EFS, CR, CRc, CR with *FLT3*-ITD MRD negativity, and CRc with *FLT3*-ITD MRD negativity. [b]*P* value was calculated using a stratified log-rank test. [c]Median follow-up time for both arms was 39.2 months. Allo-HCT, allogeneic hematopoietic cell transplantation; AML, acute myeloid leukemia; CI, confidence interval; CR, complete remission; CRc, composite complete remission; CRi, complete remission with incomplete neutrophil or platelet recovery; DoCR, duration of complete remission; EFS, event-free survival; EU, European Union; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; HiDAC, high-dose cytarabine; HR, hazard ratio; IRC, independent review committee; mo, months; mOS, median overall survival; MRD, measurable residual disease; NA, North America; OS, overall survival; RFS, relapse-free survival; WBC, white blood cell; wk, weeks.

- Measurable residual disease (MRD):
 – Key prognostic factor in AML[7–9]
 – Conventional Polymerase chain reaction (PCR) for *FLT3*-ITD less useful due to insensitivity (~1%)[8]
- PCR-next–generation sequencing (NGS) is sensitive and specific for *FLT3*-ITD MRD (targeting exons 14-15)[8,10]:
 – PCR amplification step[8]
 – Amplicons analyzed by NGS[8]
 – Developed specifically for this trial[8,10]
 – Lower limit of quantification (LLOQ)=10^{-4}
 – Lower limit of detection=2×10^{-6}
 – Often identifies multiple internal tandem duplication (ITD) sequences

Figure 2. MRD and QuANTUM-First

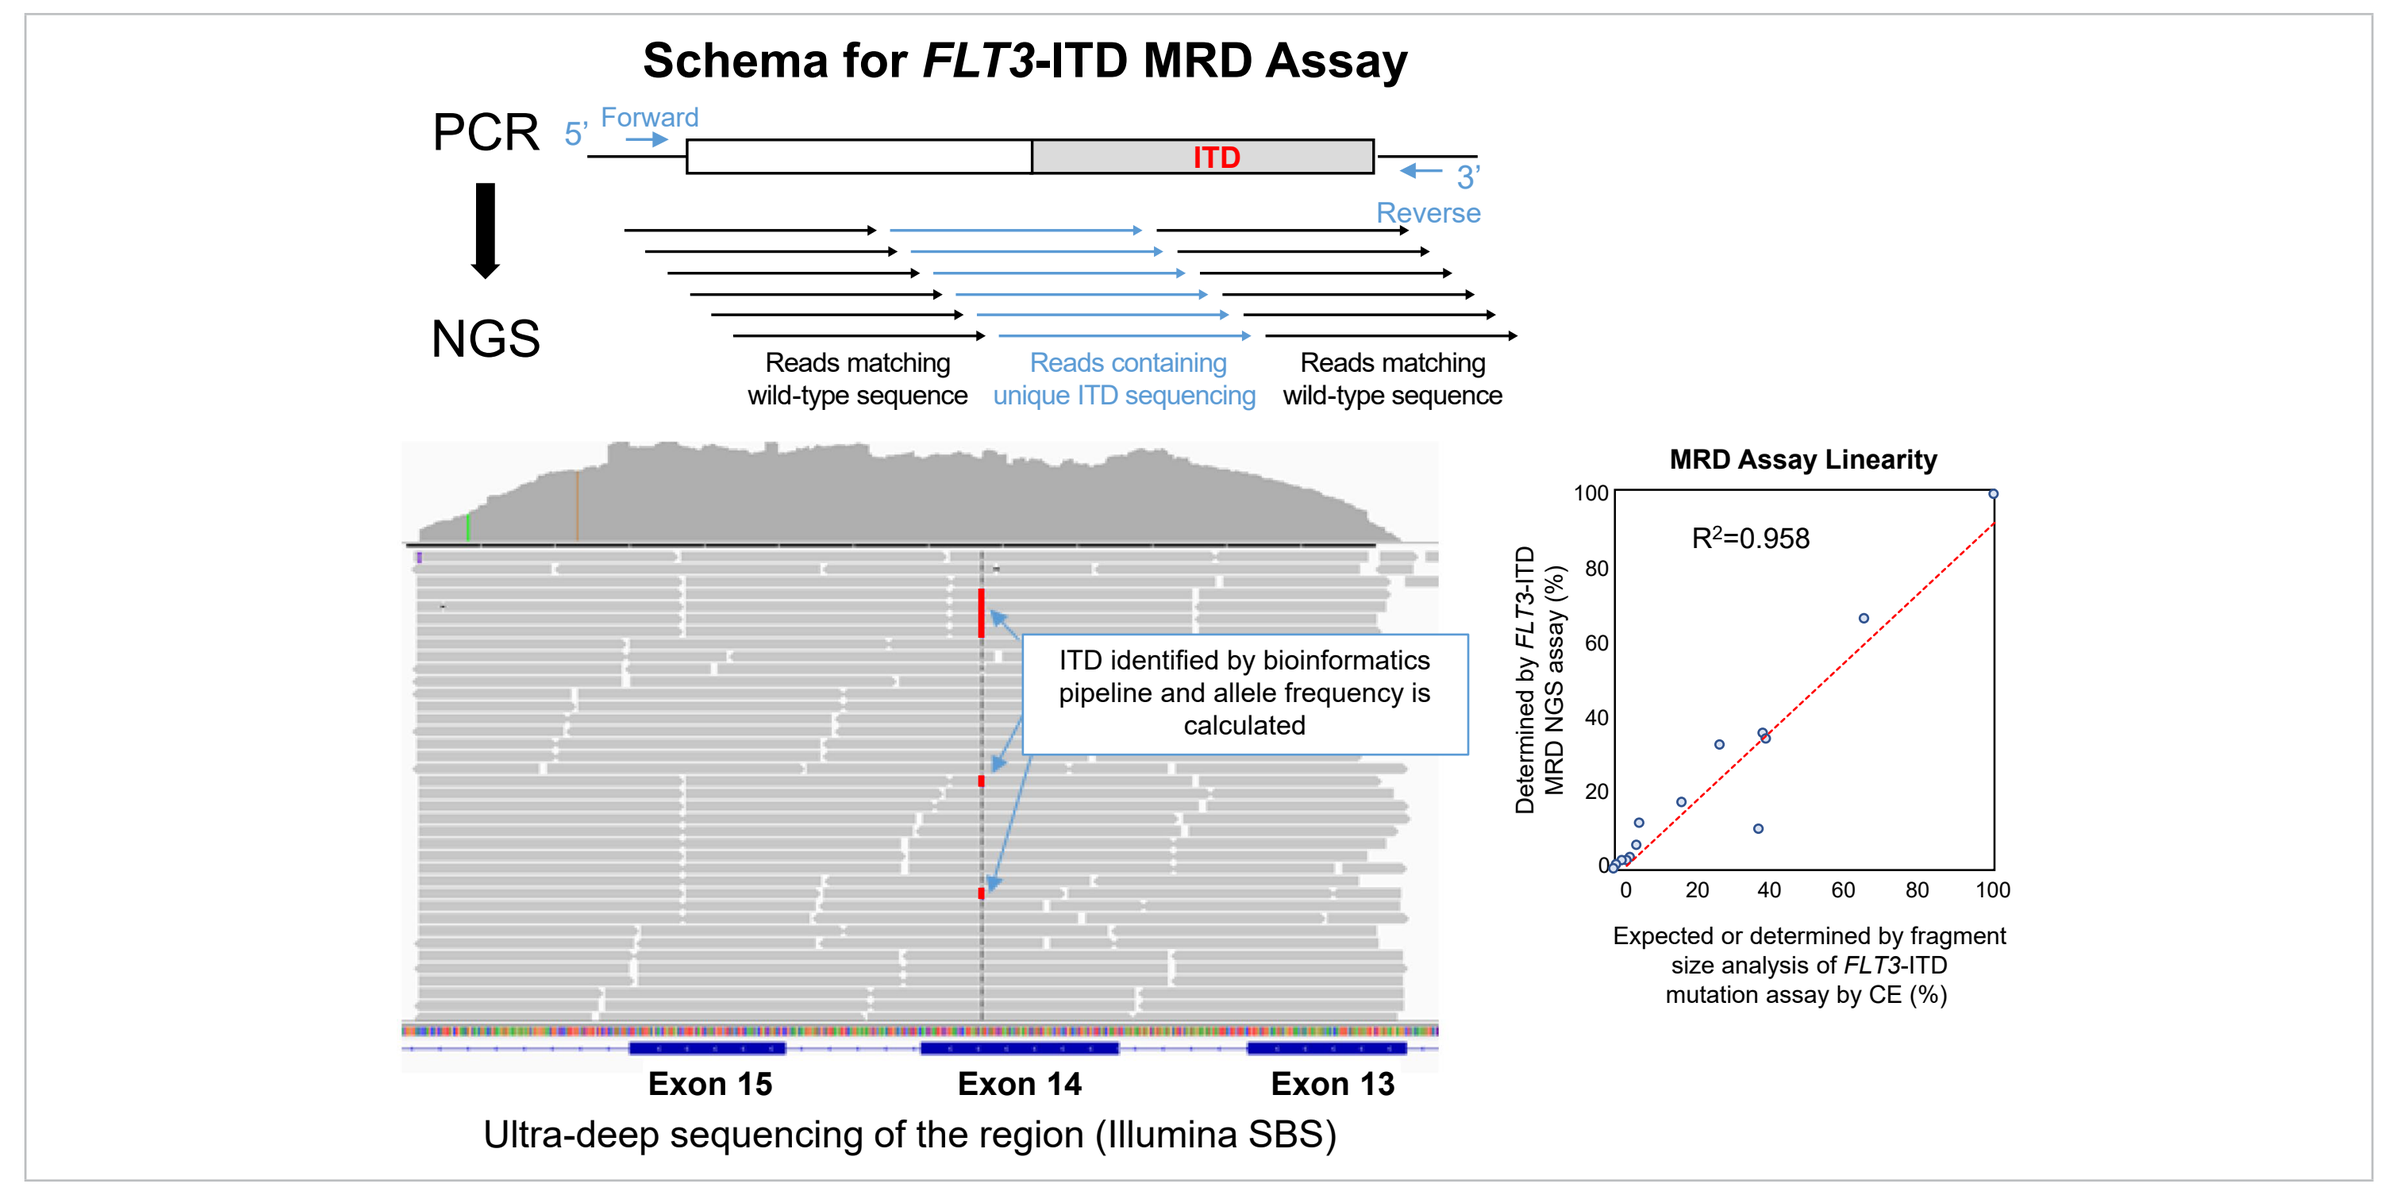
Schema for FLT3-ITD MRD Assay
PCR
NGS
Forward
Reverse
ITD
Reads matching wild-type sequence
Reads containing unique ITD sequencing
Reads matching wild-type sequence
ITD identified by bioinformatics pipeline and allele frequency is calculated
MRD Assay Linearity
R²=0.958
Determined by FLT3-ITD MRD NGS assay (%)
Expected or determined by fragment size analysis of FLT3-ITD mutation assay by CE (%)
Exon 15
Exon 14
Exon 13
Ultra-deep sequencing of the region (Illumina SBS)

CE, capillary electrophoresis; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; ITD, internal tandem duplication; MRD, measurable residual disease; NGS, next-generation sequencing; PCR, polymerase chain reaction.

Table 1. MRD, Long ITD Inserts, and Possibly Multiple ITDs Negatively Impact Survival

	FLT3-ITD MRD+ (n=47)	*FLT3*-ITD MRD– (n=114)	HR (95% CI)	*P* value
4-year CIR[11]	75%	33%	3.70 (2.31-5.94)	*P*<0.001
4-year OS[11]	31%	57%	2.47 (1.59-3.84)	*P*<0.001

	TKD1 sole (n=84)	JMD sole (n=251)	*P* value
4-year OS[13]	29%	44%	*P*=0.032

	TKD1 sole (n=84)		JMD sole (n=251)	
	Midostaurin (n=55)	Placebo (n=29)	Midostaurin (n=119)	Placebo (n=132)
4-year OS[13]	32%	26%	48%	40%
P value	*P*=0.256		*P*=0.047	

CI, confidence interval; CIR, cumulative incidence of relapse; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; HR, hazard ratio; ITD, internal tandem duplication; JMD, juxtamembrane domain; MRD, measurable residual disease; OS, overall survival; TKD1, tyrosine kinase domain-1.

- A retrospective *FLT3*-ITD PCR-NGS MRD analysis of 161 newly diagnosed *FLT3*-ITD+ patients with AML enrolled in phase 3 HOVON-SAKK clinical trials[11]
 – MRD after 2 cycles of intensive chemotherapy was associated with increased relapse risk and reduced OS[11]
- Distal ITD insertion sites are associated with long ITD insert size.[12] In the RATIFY trial, patients with the most distal ITD insertion site (TKD1) had a significantly inferior OS compared with patients with more proximal insertion sites (JMD)[13]
 – The negative impact conferred by the most distal ITD insertion sites was not improved by midostaurin treatment[13]
- Retrospective United Kingdom cooperative group data suggested multiple *FLT3*-ITDs worsen survival, but follow-up studies did not confirm this.[14,15] A limitation of these studies was the low-sensitivity PCR assay used, which cannot detect low-level variant allele frequency (VAF) *FLT3*-ITDs easily seen by PCR-NGS

AIMS

- Using samples from QuANTUM-First analyzed for *FLT3*-ITD MRD by PCR-NGS, we sought to answer the following:
 – Do deeper remissions, defined as having lower *FLT3*-ITD MRD at defined therapy time points, correlate with survival?
 – Does the addition of quizartinib to intensive chemotherapy result in lower levels of *FLT3*-ITD MRD (eg, deeper remissions)?
 – Does ITD length at diagnosis impact the outcome and if so, what is the impact of quizartinib on these outcomes?
 – Does the presence of multiple ITD clones at diagnosis impact the outcome, and if so, what is the impact of quizartinib on these outcomes?

METHODS

- Assay used for MRD analysis:
 – *FLT3*-ITD mutations were obtained from 800 ng to 1100 ng of genomic DNA and cross-validated against enrollment ITD sequences
 – VAF (*FLT3*-ITD/total *FLT3*) calculated
 – Two cutoffs for MRD were used:
 - 10^{-4} leukemia cells (predefined/per protocol, based on the assay LLOQ)
 - Zero/undetectable (post hoc analysis)
- Statistical methods:
 – Comparison of the *FLT3*-ITD MRD VAF between treatment arms across time points was made using a Wilcoxon rank-sum test
 – Comparisons of OS by ITD length and number of ITD inserts were made using unstratified Cox regression analysis
 – All *P* values were not adjusted for multiplicity

Figure 3. Sample Acquisition/Collection

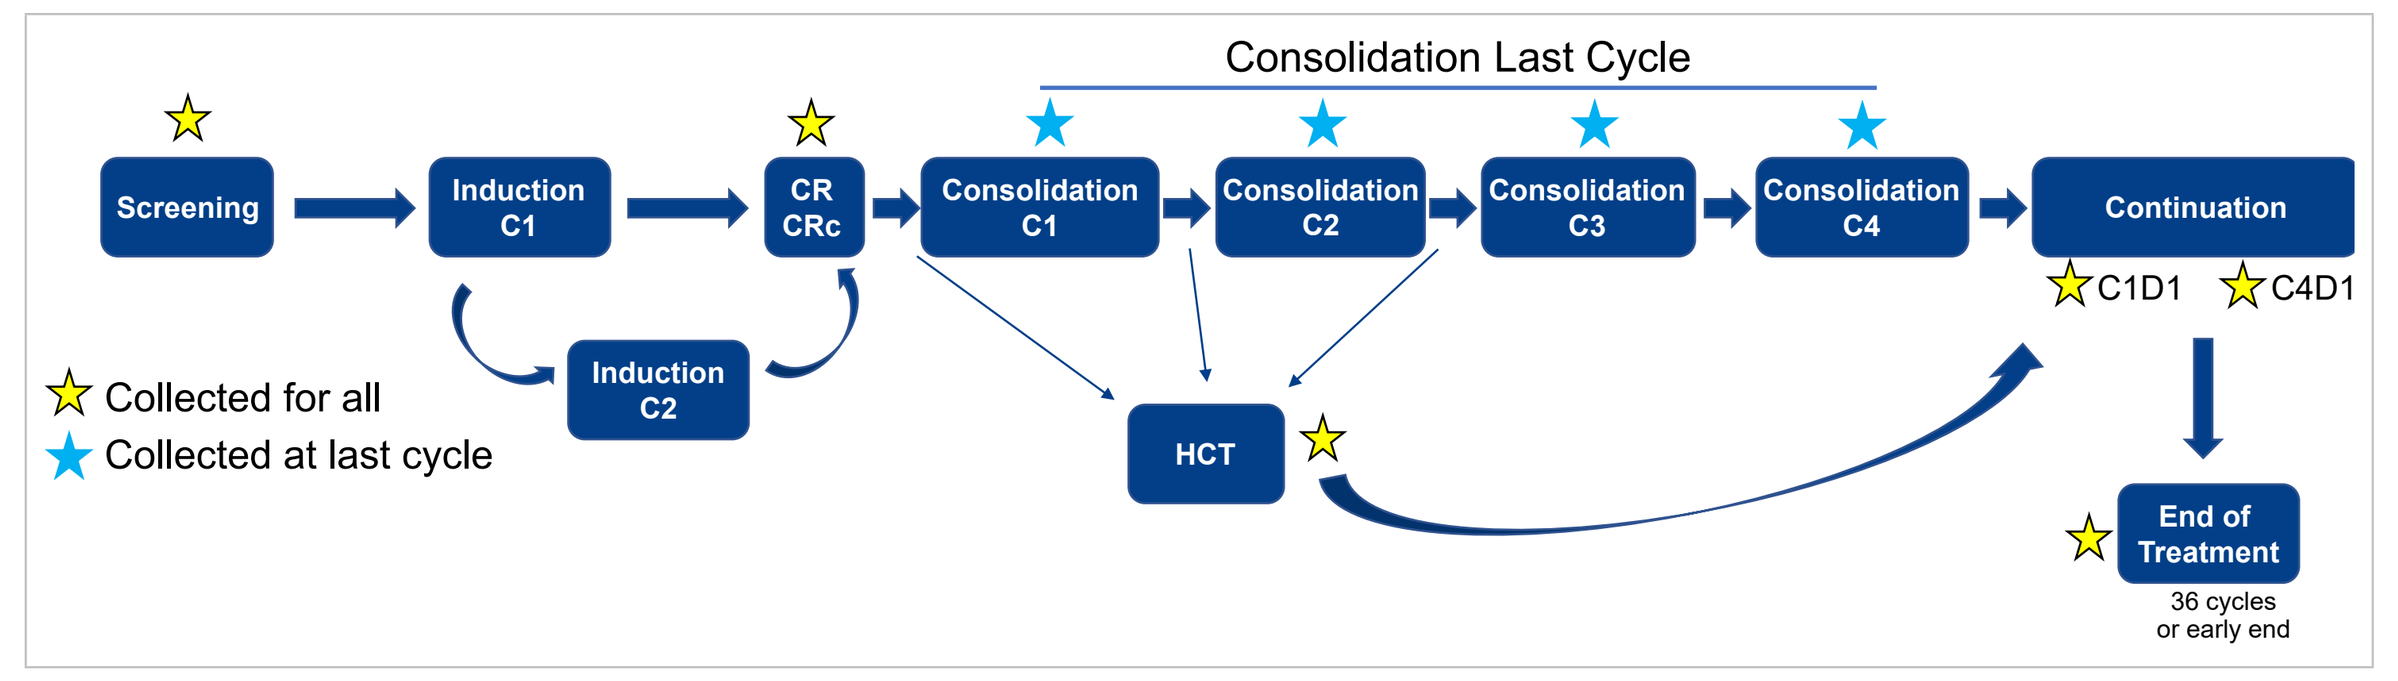
Consolidation Last Cycle
Screening
Induction C1
Induction C2
CR CRc
Consolidation C1
Consolidation C2
Consolidation C3
Consolidation C4
Continuation
C1D1
C4D1
HCT
End of Treatment
36 cycles or early end
Collected for all
Collected at last cycle

C, cycle; CR, complete remission; CRc, composite complete remission; D, day; HCT, hematopoietic cell transplantation.

RESULTS

Table 2. Baseline Characteristics of Patients With CRc by the End of induction

Patient characteristics	Patients who achieved CRc by the end of induction (N=368; 192 with Quiz, 176 with PBO) with available MRD data (N=321)[a]	
	Quizartinib (N=162)	Placebo (N=159)
Age, years		
Median (range)	56 (23-75)	55 (20-75)
<60 years, n (%)	99 (61.1)	96 (60.4)
≥60 years, n (%)	63 (38.9)	63 (39.6)
60-64 years, n (%)	20 (12.3)	25 (15.7)
≥65 years, n (%)	43 (26.5)	38 (23.9)
Sex, n (%)		
Male	73 (45.1)	65 (40.9)
Female	89 (54.9)	94 (59.1)
ECOG PS, n (%)		
0	53 (32.7)	56 (35.2)
1	83 (51.2)	82 (51.6)
2	26 (16.0)	21 (13.2)
Mutated *NPM1*, n (%)	99 (61.1)	102 (64.2)
Mutated *CEBPA*, n (%)	37 (22.8)	39 (24.5)
***FLT3*-ITD/total *FLT3*, n (%)**		
≥3% to ≤25%	57 (35.2)	50 (31.4)
>25% to ≤50%	85 (52.5)	88 (55.3)
>50%	20 (12.3)	21 (13.2)
>25%	105 (64.8)	109 (68.6)
Unknown	0	0
MRD sample collection, n (%)		
Peripheral blood	16 (9.9)	11 (6.9)
Bone marrow aspirate	161 (99.4)	158 (99.4)

[a]MRD data are available for 321 out of 368 patients achieving CRc after 1 or 2 courses of induction (47 patients had no MRD data). *CEBPA*, CCAAT enhancer-binding protein alpha; ECOG PS, Eastern Cooperative Oncology Group performance status; CRc, composite complete remission; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; MRD, measurable residual disease; *NPM1*, nucleophosmin 1.

Figure 4. *FLT3*-ITD MRD Reduction Predicts Overall Survival Across Therapy Time Points

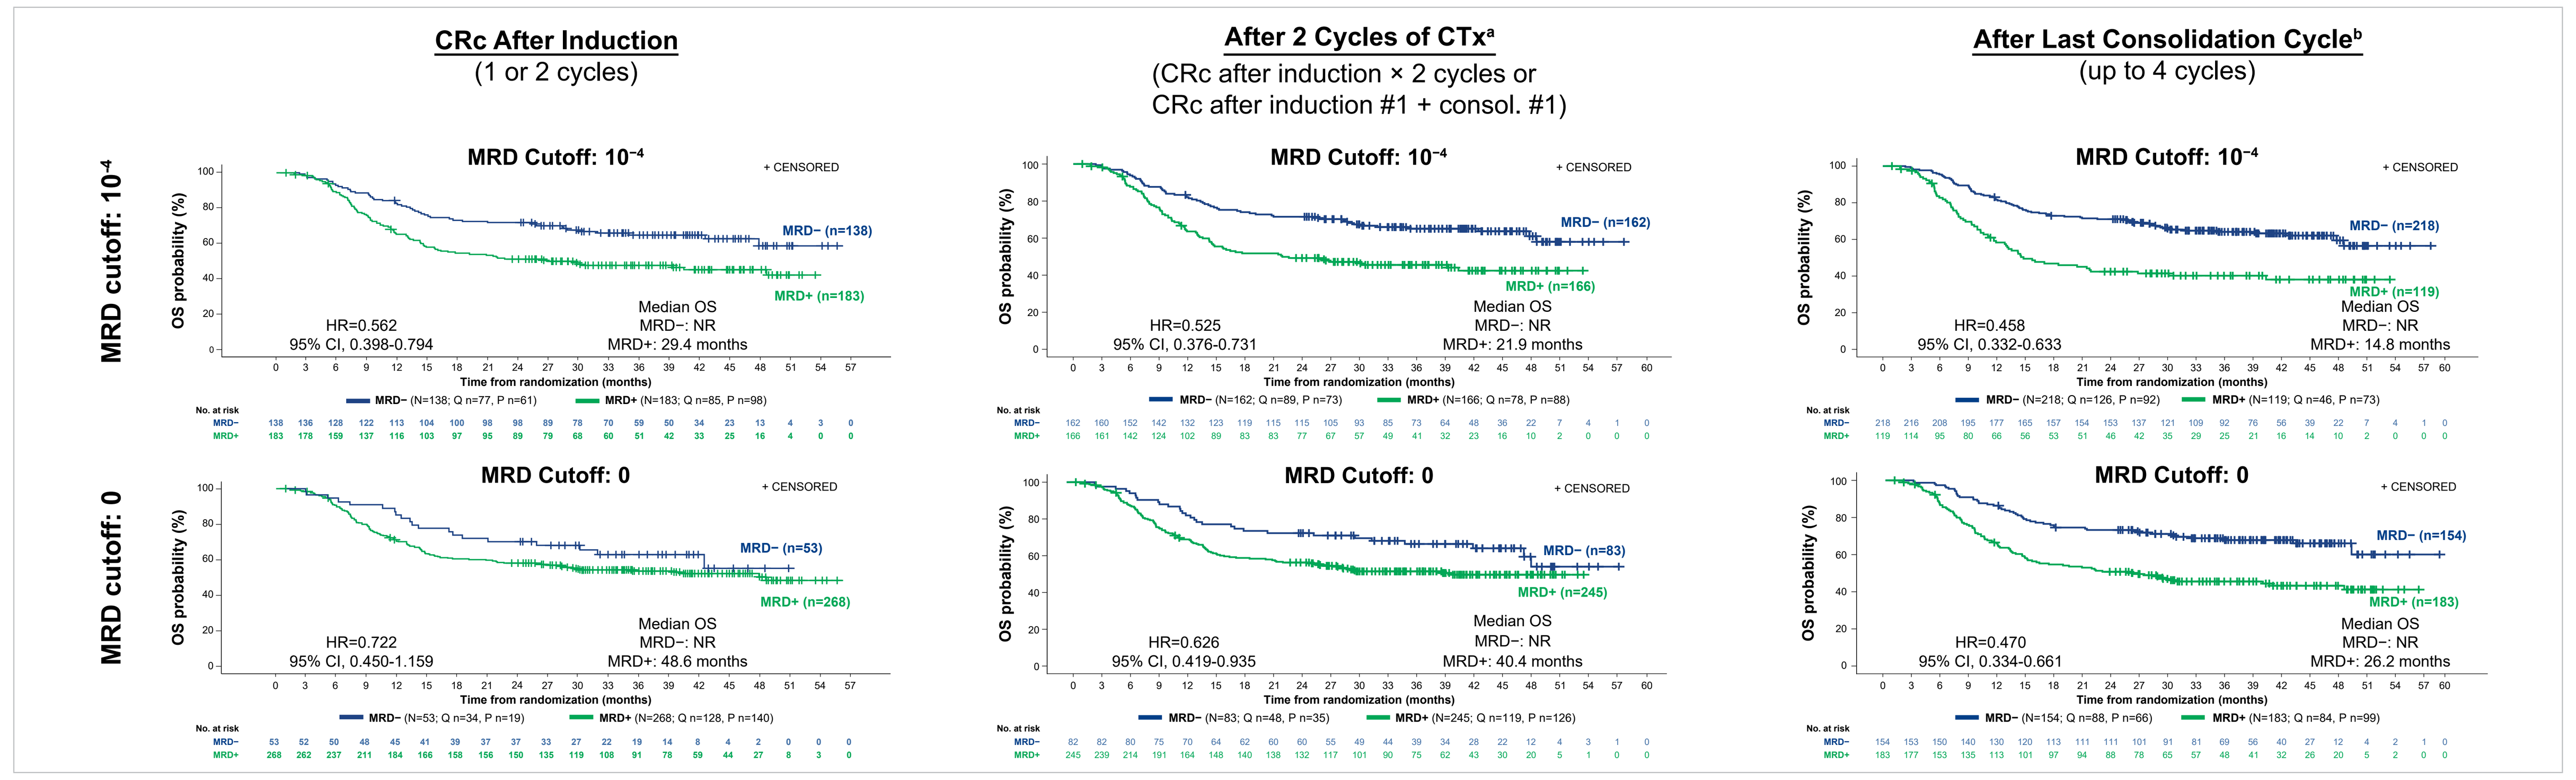
CRc After Induction (1 or 2 cycles)
After 2 Cycles of CTx[a] (CRc after induction × 2 cycles or CRc after induction #1 + consol. #1)
After Last Consolidation Cycle[b] (up to 4 cycles)
MRD cutoff: 10⁻⁴
MRD cutoff: 0
MRD Cutoff: 10⁻⁴
HR=0.562 95% CI, 0.398-0.794 Median OS MRD−: NR MRD+: 29.4 months
HR=0.525 95% CI, 0.376-0.731 Median OS MRD−: NR MRD+: 21.9 months
HR=0.458 95% CI, 0.332-0.633 Median OS MRD−: NR MRD+: 14.8 months
MRD Cutoff: 0
HR=0.722 95% CI, 0.450-1.159 Median OS MRD−: NR MRD+: 48.6 months
HR=0.626 95% CI, 0.419-0.935 Median OS MRD−: NR MRD+: 40.4 months
HR=0.470 95% CI, 0.334-0.661 Median OS MRD−: NR MRD+: 26.2 months
OS probability (%)
Time from randomization (months)

Post hoc analysis. [a]Defined as 2 cycles of induction CTx or 1 cycle of induction CTx + 1 cycle of consolidation cCTx. [b]Include samples up to end of consolidation; if there was no MRD data for the last consolidation cycle, the earlier available MRD status was used, including from induction. #, cycle; CI, confidence interval; CRc, composite complete remission; CTx, chemotherapy; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; HR, hazard ratio; MRD, measurable residual disease; NR, not reached; OS, overall survival; P, placebo; Q, quizartinib.

Figure 5. Across the Treatment Course, Quizartinib Leads to Deeper Responses and More Frequently Eliminates Detectable MRD Than Placebo

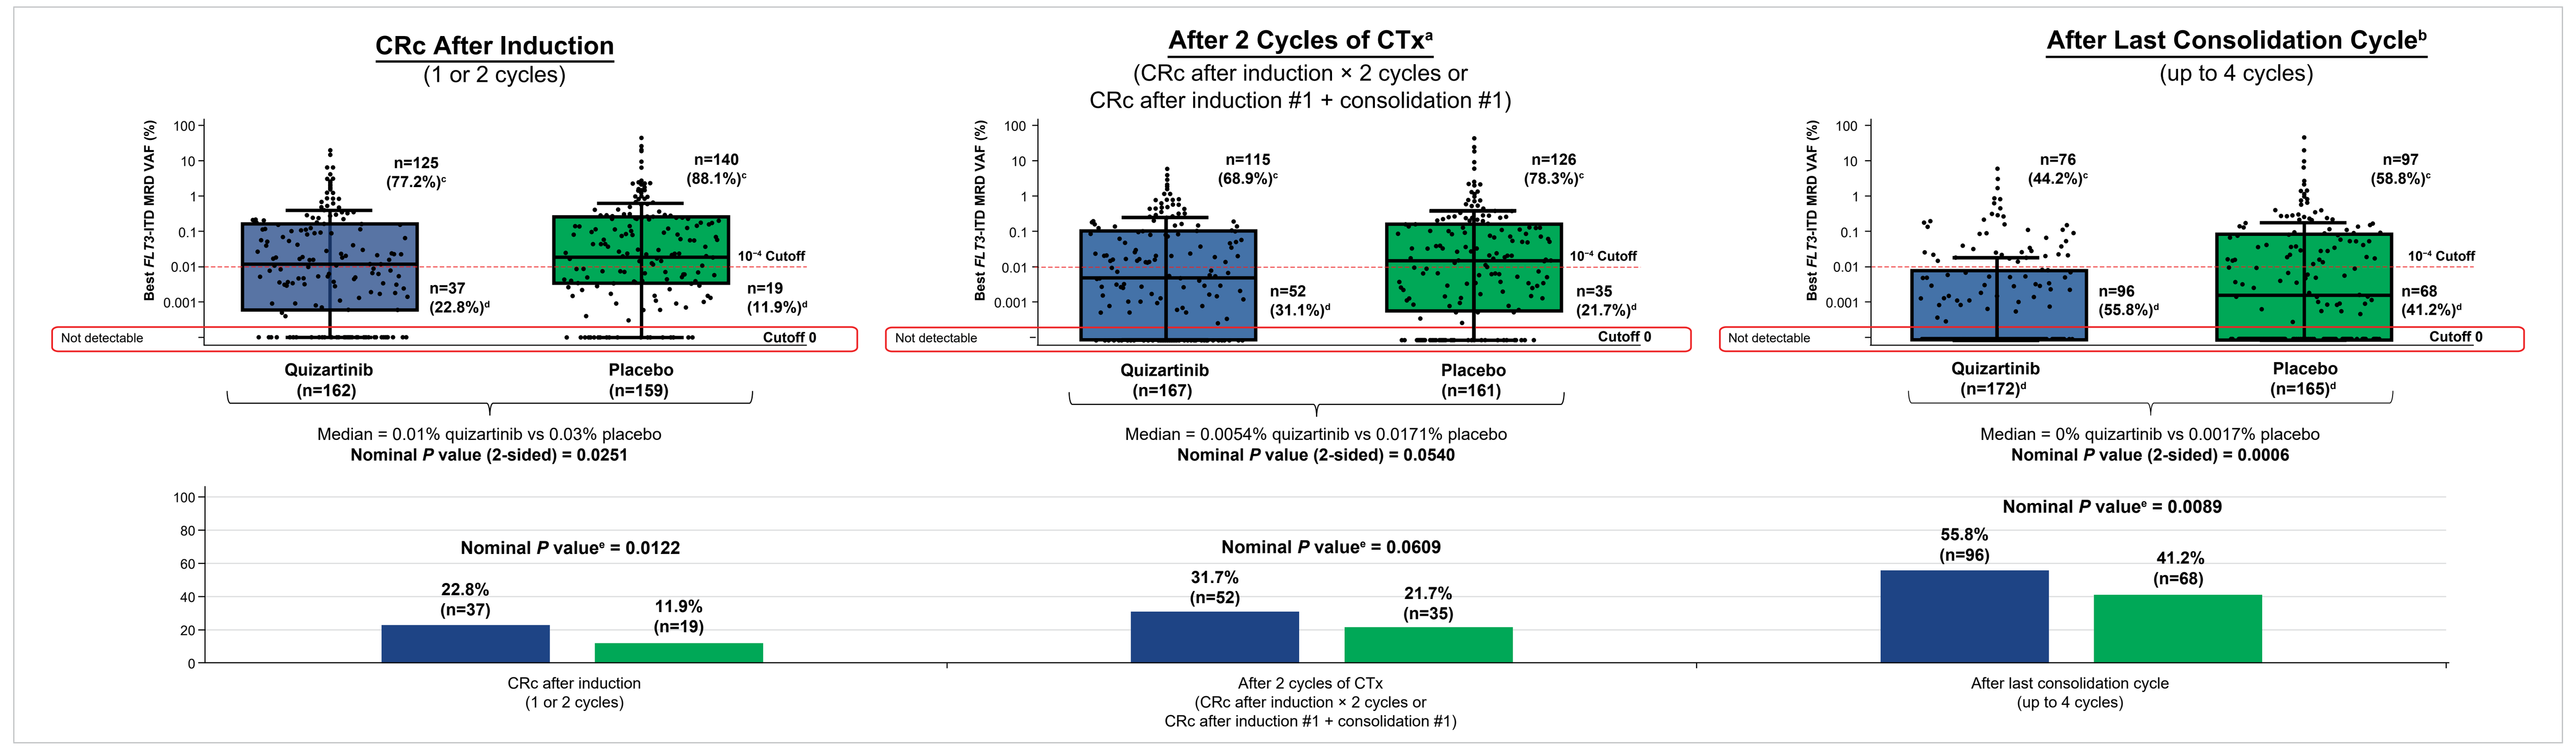
CRc After Induction (1 or 2 cycles)
After 2 Cycles of CTx[a] (CRc after induction × 2 cycles or CRc after induction #1 + consolidation #1)
After Last Consolidation Cycle[b] (up to 4 cycles)
Best FLT3-ITD MRD VAF (%)
Not detectable
10⁻⁴ Cutoff
Cutoff 0
Quizartinib (n=162) Placebo (n=159)
Median = 0.01% quizartinib vs 0.03% placebo
Nominal P value (2-sided) = 0.0251
Quizartinib (n=167) Placebo (n=161)
Median = 0.0054% quizartinib vs 0.0171% placebo
Nominal P value (2-sided) = 0.0540
Quizartinib (n=172)[d] Placebo (n=165)[d]
Median = 0% quizartinib vs 0.0017% placebo
Nominal P value (2-sided) = 0.0006
Nominal P value[e] = 0.0122
Nominal P value[e] = 0.0609
Nominal P value[e] = 0.0089
CRc after induction (1 or 2 cycles)
After 2 cycles of CTx (CRc after induction × 2 cycles or CRc after induction #1 + consolidation #1)
After last consolidation cycle (up to 4 cycles)

Post hoc analysis. [a]Defined as 2 cycles of induction CTx or 1 cycle of induction CTx + 1 cycle of consolidation CTx. [b]Includes samples up to end of consolidation, including from induction. [c]Percentage of patients with *FLT3*-ITD MRD VAF>0 among CRc patients with MRD data. [d]Percentage of patients with *FLT3*-ITD MRD VAF=0 among CRc patients with MRD data. [e]Fisher's exact test. #, cycle; CI, confidence interval; CRc, composite complete remission; CTx, chemotherapy; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; MRD, measurable residual disease; VAF, variant allele frequency.

Figure 6. *FLT3*-ITD MRD Reduction Predicts Survival Across Therapy Time Points (Cutoff 0)

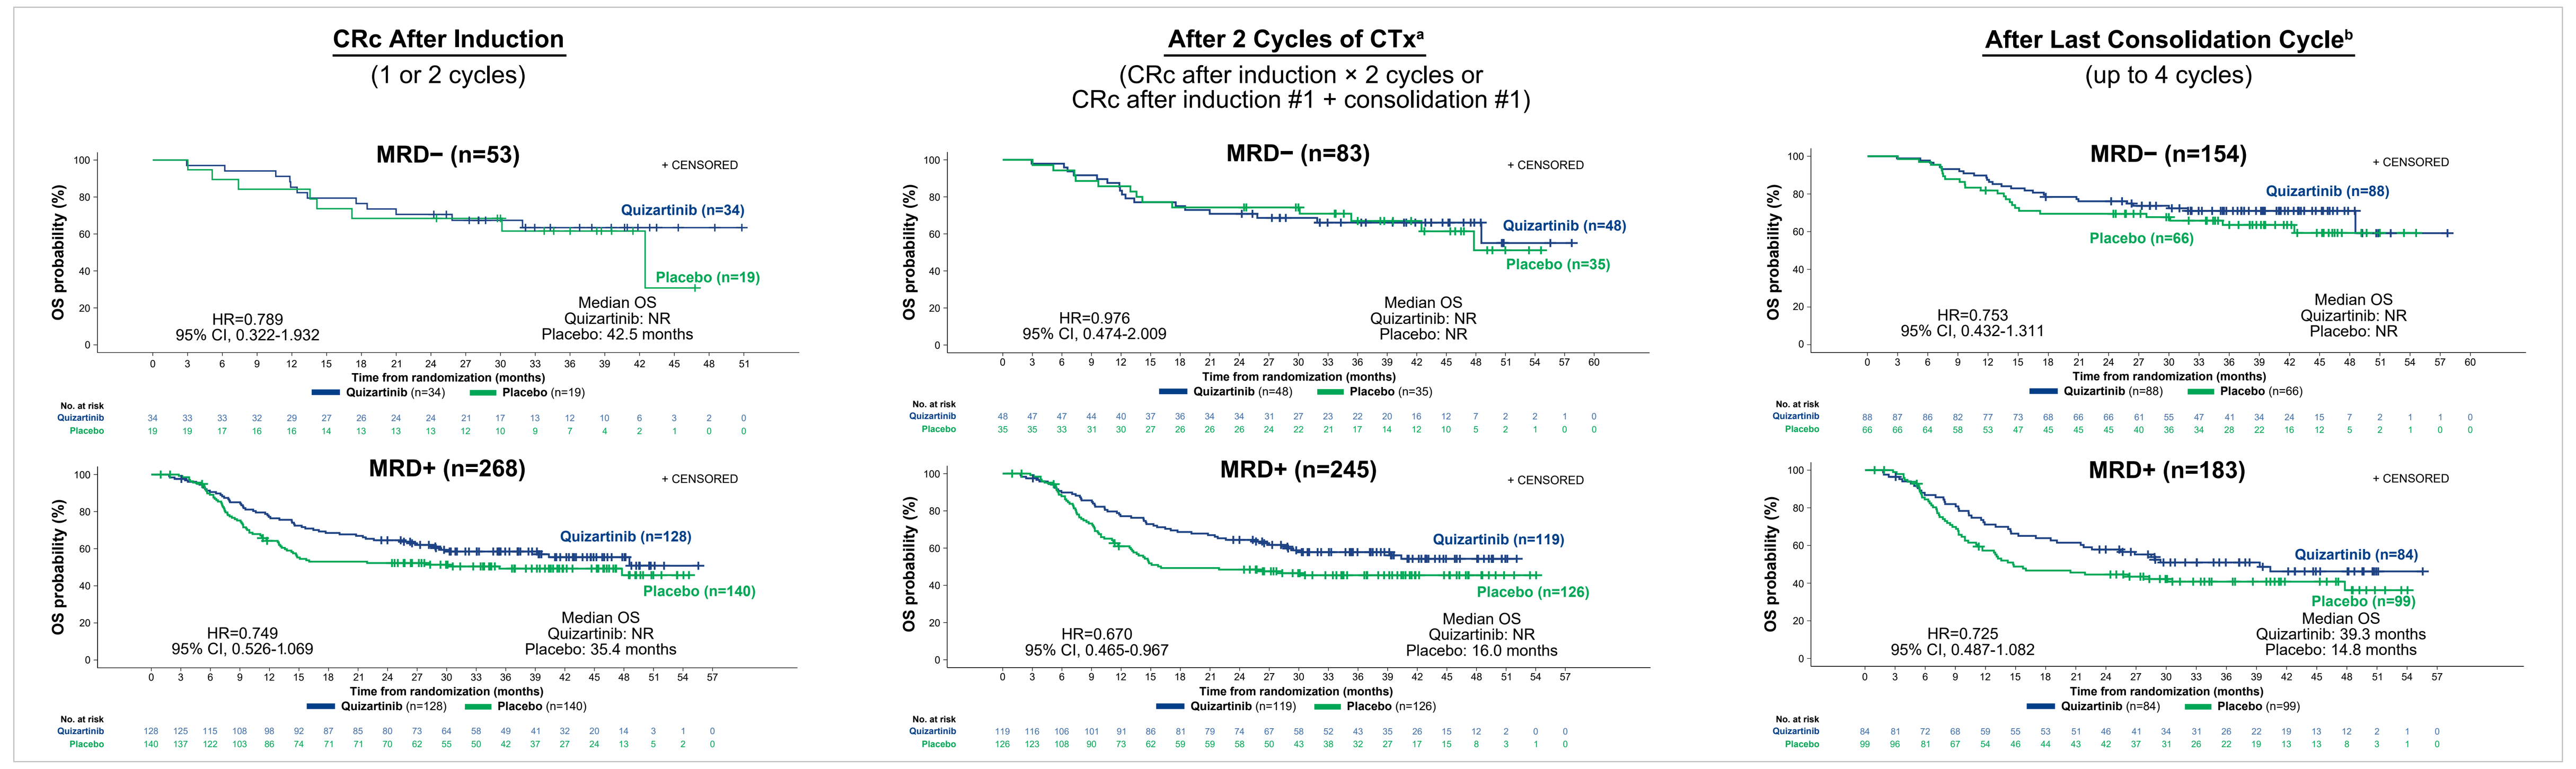
CRc After Induction (1 or 2 cycles)
After 2 Cycles of CTx[a] (CRc after induction × 2 cycles or CRc after induction #1 + consolidation #1)
After Last Consolidation Cycle[b] (up to 4 cycles)
MRD− (n=53)
HR=0.789 95% CI, 0.322-1.932 Median OS Quizartinib: NR Placebo: 42.5 months
MRD− (n=83)
HR=0.976 95% CI, 0.474-2.009 Median OS Quizartinib: NR Placebo: NR
MRD− (n=154)
HR=0.753 95% CI, 0.432-1.311 Median OS Quizartinib: NR Placebo: NR
MRD+ (n=268)
HR=0.749 95% CI, 0.526-1.069 Median OS Quizartinib: NR Placebo: 35.4 months
MRD+ (n=245)
HR=0.670 95% CI, 0.465-0.967 Median OS Quizartinib: NR Placebo: 16.0 months
MRD+ (n=183)
HR=0.725 95% CI, 0.487-1.082 Median OS Quizartinib: 39.3 months Placebo: 14.8 months
OS probability (%)
Time from randomization (months)

Post hoc analysis. [a]Defined as 2 cycles of induction CTx or 1 cycle of induction CTx + 1 cycle of consolidation CTx. [b]Includes samples up to end of consolidation; if there was no MRD data for the last consolidation cycle, the earlier available MRD status was used, including from induction. #, cycle; CI, confidence interval; CRc, composite complete remission; CTx, chemotherapy; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; HR, hazard ratio; MRD, measurable residual disease; NR, not reached; OS, overall survival.

Figure 7. Long ITD Insertions Are Associated With Worse Survival

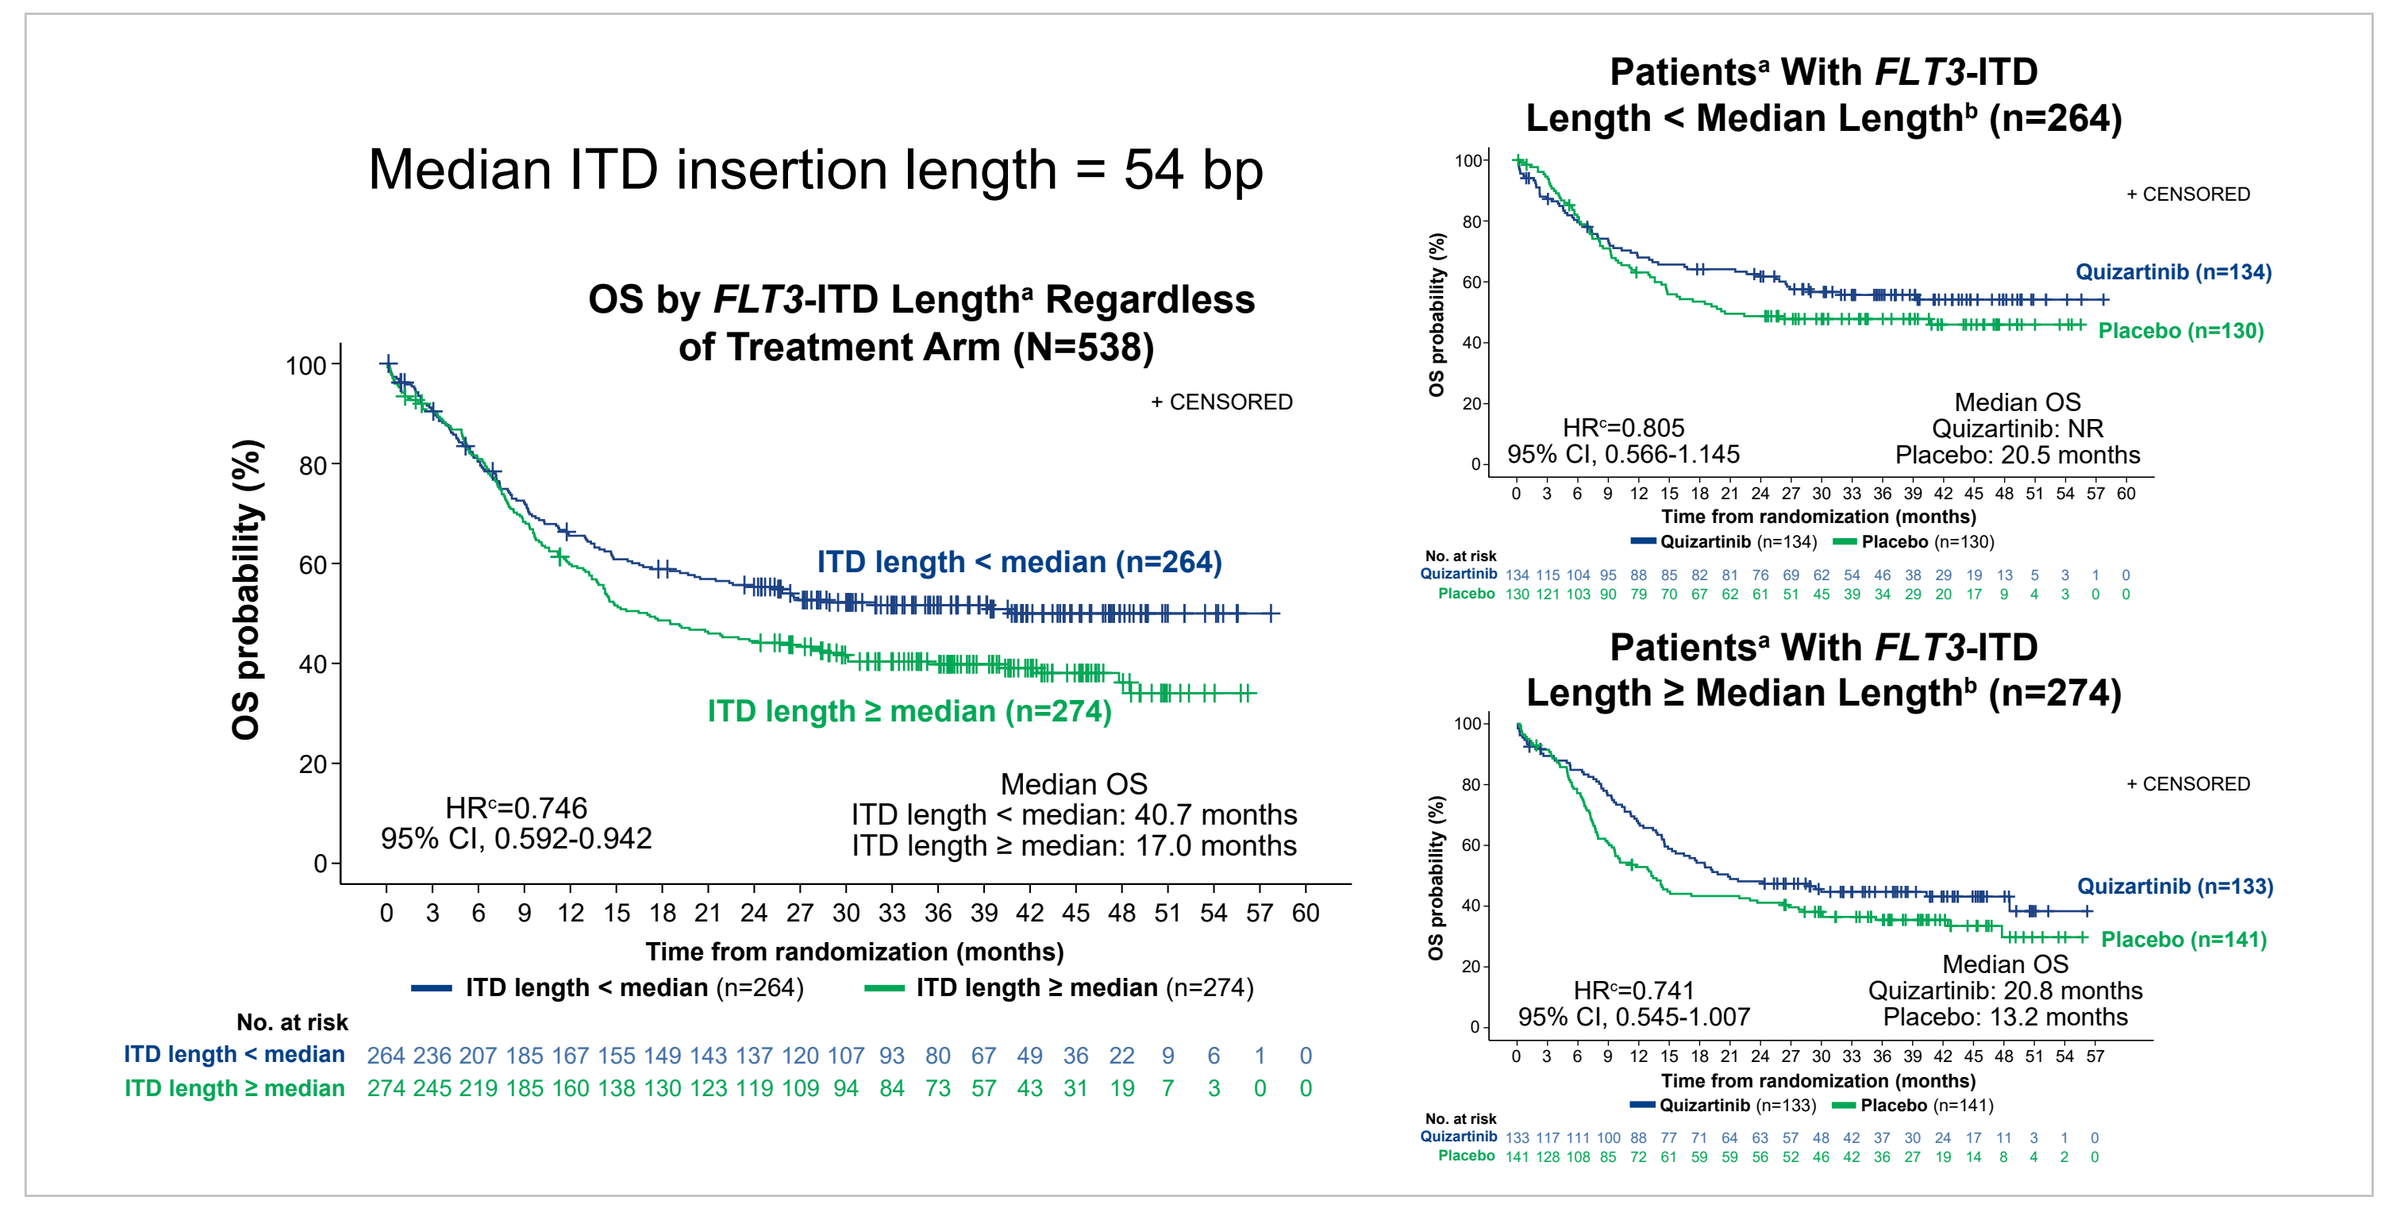
Median ITD insertion length = 54 bp
OS by FLT3-ITD Length[a] Regardless of Treatment Arm (N=538)
ITD length < median (n=264)
ITD length ≥ median (n=274)
HR[c]=0.746 95% CI, 0.592-0.942
Median OS ITD length < median: 40.7 months ITD length ≥ median: 17.0 months
Patients[a] With FLT3-ITD Length < Median Length[b] (n=264)
HR[c]=0.805 95% CI, 0.566-1.145 Median OS Quizartinib: NR Placebo: 20.5 months
Patients[a] With FLT3-ITD Length ≥ Median Length[b] (n=274)
HR[c]=0.741 95% CI, 0.545-1.007 Median OS Quizartinib: 20.8 months Placebo: 13.2 months

Post hoc analysis. [a]Patients may have only 1 ITD length or >1 ITD length. [b]Median ITD length (54 bp) is calculated based on enrollment assay data (Navigate BioPharma *FLT3*-ITD Mutation Assay). [c]Unstratified Cox regression analysis.
CI, confidence interval; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; HR, hazard ratio; ITD, internal tandem duplication; NR, not reached; OS, overall survival.

Figure 8. Multiple ITDs Are Associated With Worse Survival, and Quizartinib Can Improve OS in Patients With Multiple ITDs

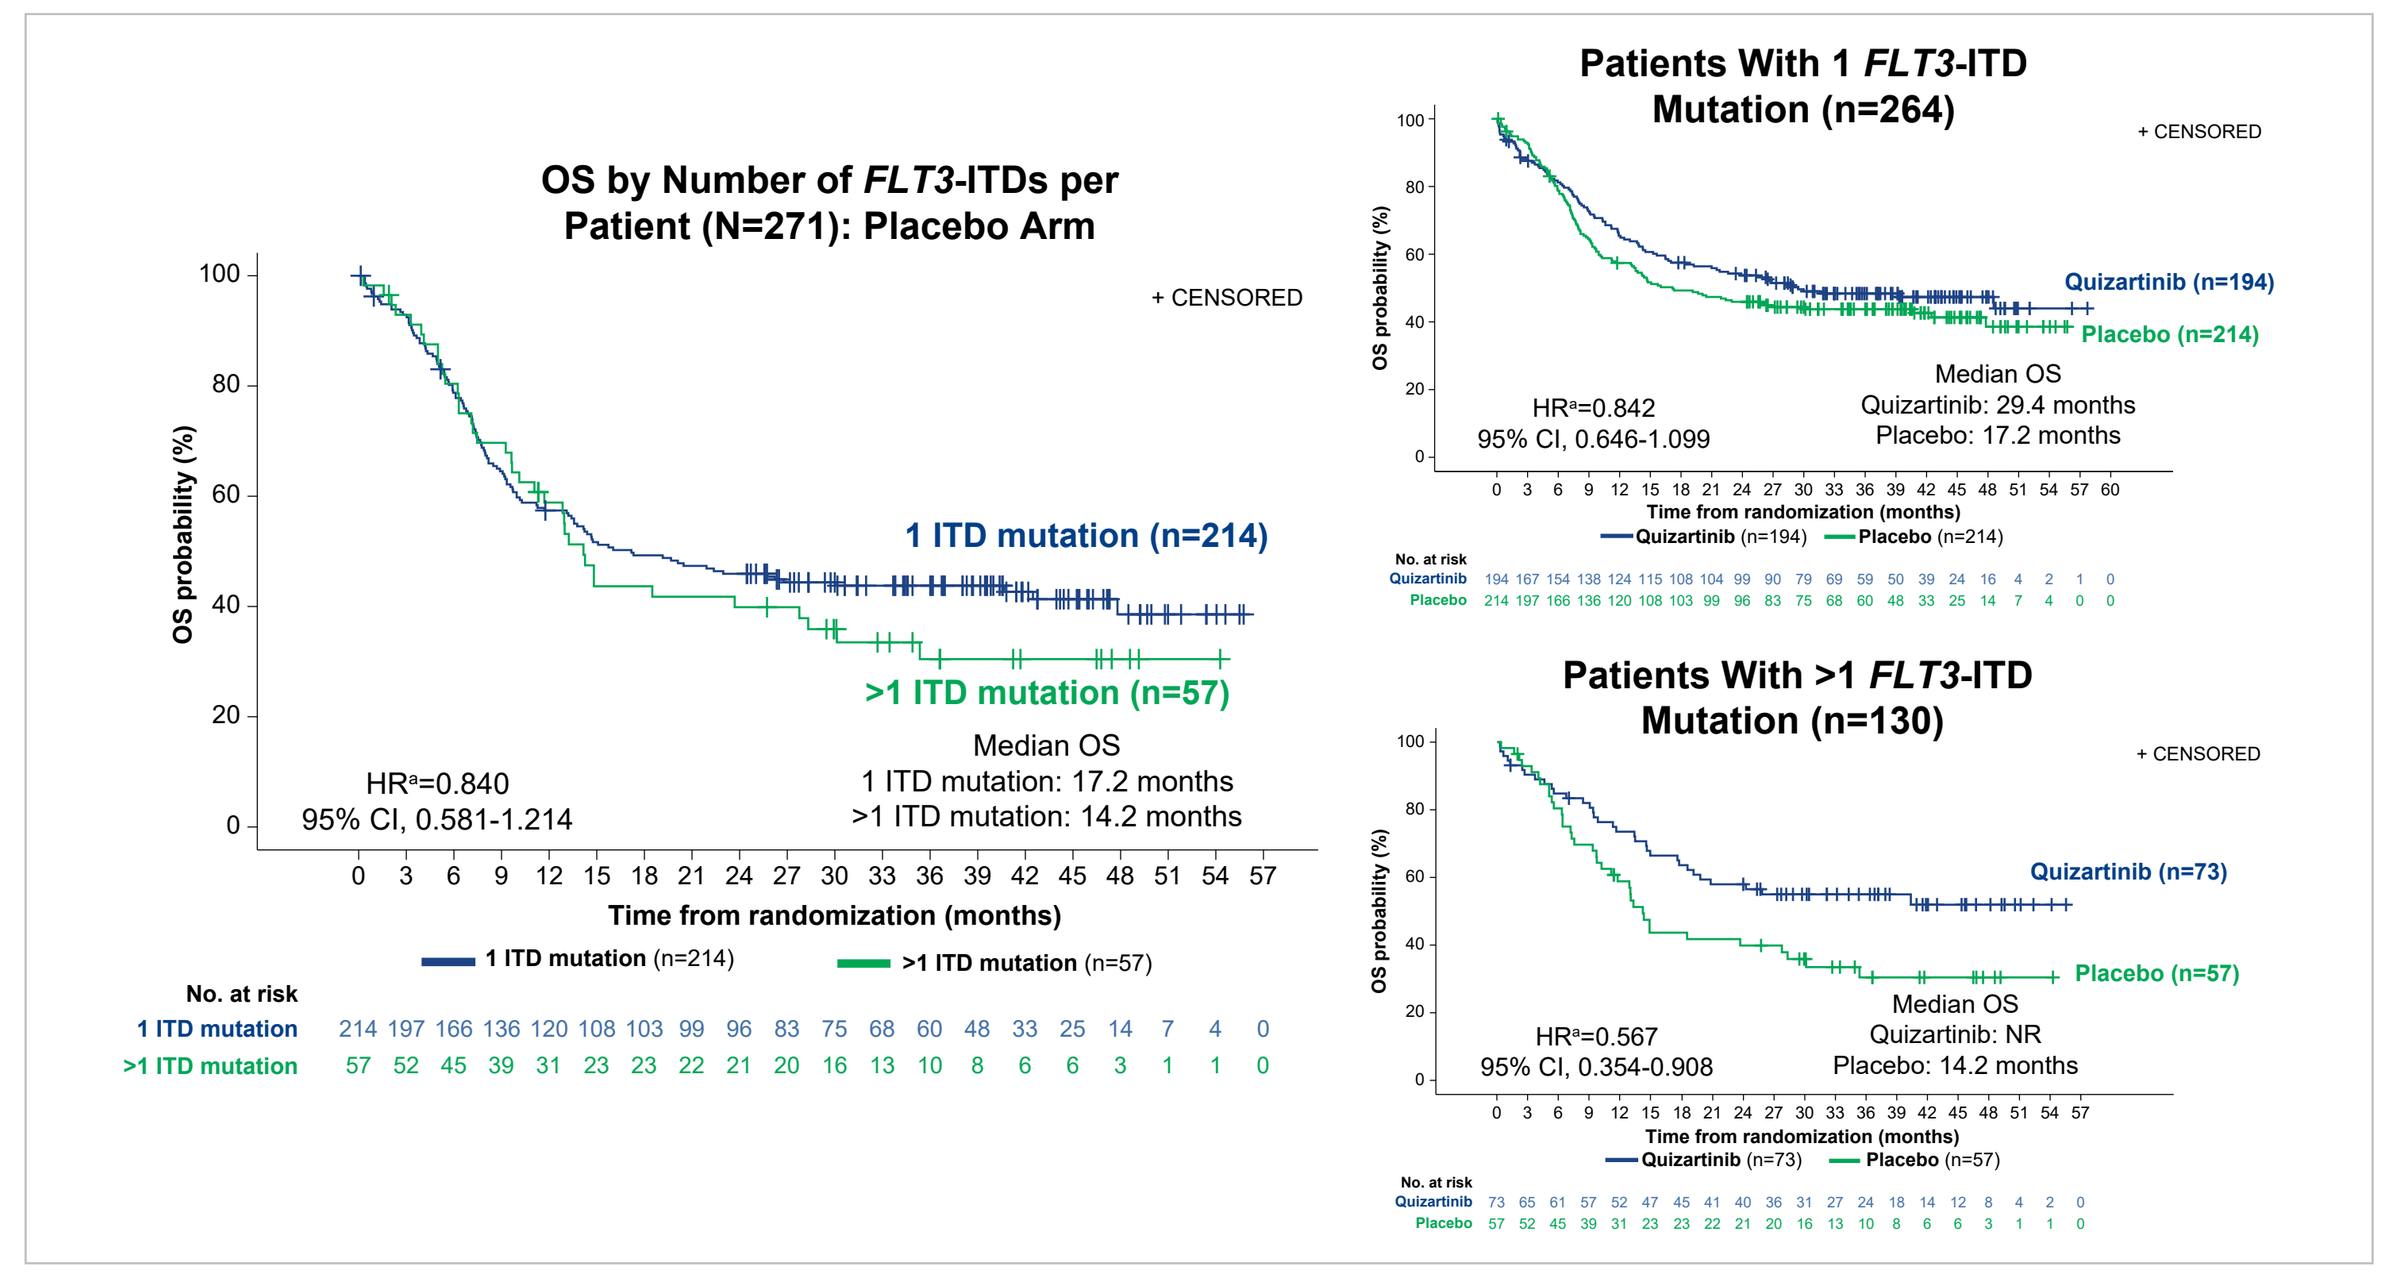
OS by Number of FLT3-ITDs per Patient (N=271): Placebo Arm
1 ITD mutation (n=214)
>1 ITD mutation (n=57)
HR[a]=0.840 95% CI, 0.581-1.214
Median OS 1 ITD mutation: 17.2 months >1 ITD mutation: 14.2 months
Patients With 1 FLT3-ITD Mutation (n=264)
HR[a]=0.842 95% CI, 0.646-1.099 Median OS Quizartinib: 29.4 months Placebo: 17.2 months
Patients With >1 FLT3-ITD Mutation (n=130)
HR[a]=0.567 95% CI, 0.354-0.908 Median OS Quizartinib: NR Placebo: 14.2 months

Post hoc analysis. [a]Unstratified Cox regression analysis.
CI, confidence interval; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; HR, hazard ratio; ITD, internal tandem duplication; NR, not reached; OS, overall survival.

CONCLUSIONS

- These findings demonstrate the potential prognostic utility of *FLT3*-ITD–specific MRD measurements in the clinical management of patients with *FLT3*-ITD+ AML
- Elimination of detectable *FLT3*-ITD MRD is associated with longer OS compared with intensive chemotherapy with or without quizartinib
- Therapy with quizartinib is associated with deeper responses and more frequently eliminates detectable MRD than placebo after induction, after 2 cycles of chemotherapy, and after consolidation
- The presence of multiple ITDs or long ITD inserts at diagnosis did not negatively impact the survival benefits of quizartinib
- Our data suggest that some of the long-term OS benefits conferred by quizartinib derive from an early, deep, and sustained reduction of the *FLT3*-ITD+ leukemia burden

ACKNOWLEDGMENTS

We would like to thank the patients, their families, and caregivers for their participation in the QuANTUM-First study. We would further like to thank the QuANTUM-First steering committee members, the investigators, Donna Hogge, Jack Hsu, the study staff, and independent review committee and data monitoring committee members for their important contributions.

This study is sponsored by Daiichi Sankyo, Inc. Data first presented at the 65th Annual Meeting & Exposition of the American Society of Hematology (ASH) 2023; San Diego, CA, USA; December 9-12, 2023.

Medical writing support was provided by Mohamed Abdelmegeed, MD, PhD, CMPP, Emily Cullinan, PhD, CMPP, and Francesca Balordi, PhD, CMPP, of The Lockwood Group (Stamford, CT, USA), in accordance with Good Publication Practice (GPP 2022) guidelines, with funding by Daiichi Sankyo, Inc.

REFERENCES

1. Aikawa T, et al. *Oncotarget.* 2020;11(11):943-955.
2. Levis M. *Hematology Am Soc Hematol Educ Program.* 2013;2013:220-226.
3. Pratz KW, et al. *Blood.* 2010;115(17):1425-1432.
4. Stone RM, et al. *N Engl J Med.* 2017;377(5):454-464.
5. Perl AE, et al. *N Engl J Med.* 2019;381(18):1728-1740.
6. Erba H, et al. *Lancet.* 2023;401(10388):1571-1583.
7. Jongen-Lavrencic M, et al. *N Engl J Med.* 2018;378(13):1189-1199.
8. Levis M, et al. *Blood Adv.* 2018;2(8):825-831.
9. Döhner H, et al. *Blood.* 2022;140(12):1345-1377.
10. Levis M, et al. *Blood.* 2020;135(1):75-78.
11. Grob T, et. al. *J Clin Oncol.* 2023;41(4):756-765.
12. Kayser S, et al. *Blood.* 2009;114(12):2386-2392.
13. Rucker FG, et al. *Leukemia.* 2022;36(1):90-99.
14. Kottaridis PD, et al. *Blood.* 2001;98(6):1752-1759.
15. Gale RE, et al. *Blood.* 2008;111(5):2776-2784.

Presented at the 12th Annual Meeting of the Society of Hematologic Oncology (SOHO); September 4–7; Houston, TX, US.