Poster 4090

Trastuzumab deruxtecan in patients with HER2-expressing biliary tract cancer and pancreatic cancer: outcomes from DESTINY-PanTumor02

Do-Youn Oh,[1] Iwona Ługowska,[2] Daniil Stroyakovskiy,[3] Kyung Hae Jung,[4] Olivier Dumas,[5] Konstantin Penkov,[6] Arunee Dechaphunkul,[7] Ana Oaknin,[8] Seung Tae Kim,[9] Naureen Starling,[10] Busyamas Chewaskulyong,[11] Chanchai Charonpongsuntorn,[12] Deborah Doroshow,[13] Sheng-Yen Hsiao,[14] Yi-Ping Hung,[15] Lindsey Jung,[16] Nataliya Kuptsova-Clarkson,[17] Flavia Michelini,[18] Soham Puvvada,[17] Funda Meric-Bernstam[19]

[1]Department of Internal Medicine, Seoul National University Hospital, Republic of Korea; [2]Early Phase Clinical Trials Unit, Maria Skłodowska-Curie National Research Institute and Oncology Centre, Warsaw, Poland; [3]Healthcare Department, Moscow City Oncology Hospital No. 62, Russia; [4]Department of Oncology, Asan Medical Center, University of Ulsan College of Medicine, Seoul, Republic of Korea; [5]Hôtel-Dieu de Québec, Canada; [6]Clinical Hospital "RZHD-Medicine", Saint Petersburg, Russia; [7]Medical Oncology Unit, Department of Medicine, Faculty of Medicine, Prince of Songkla University, Songkhla, Thailand; [8]Gynaecologic Cancer Programme, Vall d'Hebron Institute of Oncology, University Hospital Vall d'Hebron, Barcelona, Spain; [9]Sungkyunkwan University School of Medicine, Samsung Medical Center, Seoul, Republic of Korea; [10]Gastrointestinal Unit, The Royal Marsden NHS Foundation Trust and Institute of Cancer Research, London, UK; [11]Medical Oncology Unit, Department of Internal Medicine, Faculty of Medicine, Chiang Mai University, Thailand; [12]Medical Oncology Unit, Department of Internal Medicine, Faculty of Medicine, Srinakharinwirot University, Nakhon Nayok, Thailand; [13]Icahn School of Medicine at Mount Sinai, New York, NY, US; [14]Division of Hematology-Oncology, Department of Internal Medicine, Chi Mei Medical Center, Liouying, Tainan, Taiwan; [15]Taipei Veterans General Hospital, Taiwan; [16]AstraZeneca, Gaithersburg, MD, US; [17]Oncology R&D, AstraZeneca, Gaithersburg, MD, US; [18]Translational Medicine, Oncology R&D, AstraZeneca, Waltham, MA, US; [19]Department of Investigational Cancer Therapeutics, University of Texas MD Anderson Cancer Center, Houston, TX, US

Objectives

- Evaluate subgroup analyses in the biliary tract and pancreatic cancer cohorts from the DESTINY-PanTumor02 study
- Characterize patients who achieved an objective response (OR) in the biliary tract and pancreatic cancer cohorts from the DESTINY-PanTumor02 study

Conclusions

- Trastuzumab deruxtecan (T-DXd) demonstrated clinically meaningful benefit in pretreated patients with human epidermal growth factor receptor 2 (HER2)-expressing biliary tract tumors in DESTINY-PanTumor02
 - The 9 (22.0%) patients who had a confirmed OR by investigator all had a central HER2 status of immunohistochemistry (IHC) 3+
 - Durable responses were observed, with a median duration of response (DOR) of 8.6 months in all patients
- Low patient numbers limit interpretation in the pancreatic cancer cohort; more data are needed to understand which patients with pancreatic cancer may benefit from T-DXd
- The safety findings were consistent with the established profile for T-DXd
 - The most common Grade ≥3 drug-related treatment-emergent adverse event (TEAE) in both cohorts was neutropenia
 - Interstitial lung disease (ILD) / pneumonitis remains an important identified risk; proactive monitoring, early detection, and active management are critical in preventing high-grade ILD/pneumonitis
- These data support T-DXd as a recommended treatment option for pretreated patients with HER2-expressing (IHC 3+) biliary tract cancer

Plain language summary

Why did we perform this research?
Trastuzumab deruxtecan (T-DXd) is an antibody-drug conjugate, which is a chemotherapy with a linker (deruxtecan) joined to an antibody (trastuzumab). Trastuzumab binds to a protein found on cancer cells called human epidermal growth factor receptor 2 (HER2), where it releases the chemotherapy to kill these cells.[1,2] Based partly on results from the DESTINY-PanTumor02 study, T-DXd is a recommended treatment in the USA for people with solid tumors that have higher-than-normal levels of HER2 (HER2-positive, also known as immunohistochemistry [IHC] 3+) and have spread or cannot be completely removed with surgery and who have received prior systemic treatment and have no satisfactory alternative treatment options available.[3] Further evaluation of people with biliary tract and pancreatic cancers (types of solid tumors) who received T-DXd as part of the DESTINY-PanTumor02 study is needed.

How did we perform this research?
This analysis looked at people with HER2-expressing (IHC 3+ and IHC 2+) biliary tract and pancreatic cancers who had received prior systemic treatment or had no satisfactory alternative treatment options available and who received T-DXd as part of the DESTINY-PanTumor02 study.

What were the findings of this research?
Overall, 9 out of 41 people with biliary tract cancer had a response to T-DXd (ie reduction in tumor size); 9 out of 16 people with IHC 3+ tumors had a response. In people with pancreatic cancer, 1 of the 25 people had a response to T-DXd. Side effects were as expected for T-DXd. Severe side effects associated with T-DXd treatment that were observed in >5% of people with biliary tract cancer included a decrease in a type of blood cell called neutrophils (9.8%) leading to neutropenia (9.8%), nausea (7.3%), and fatigue (7.3%). In people with pancreatic cancer, severe side effects associated with T-DXd treatment seen in >5% people included fewer red blood cells leading to anemia (8.0%) and neutropenia (8.0%).

What are the implications of this research?
These results support use of T-DXd as a treatment for people with HER2-positive (IHC 3+) biliary tract cancers that have spread or cannot be completely removed with surgery and who have received prior systemic treatment and have no satisfactory alternative treatment options available. Further research is needed to assess use of T-DXd in people with pancreatic cancers.

Where can I access more information?
For information about DESTINY-PanTumor02, please visit https://clinicaltrials.gov/study/NCT04482309, or see primary data published in the *Journal of Clinical Oncology* here. Please also reach out to Dr. Oh at ohdoyoun@snu.ac.kr.

1. Nakada T, et al. *Chem Pharm Bull (Tokyo)*. 2019;67:173–185; 2. Ogitani Y, et al. *Clin Cancer Res*. 2016;22:5097–5108; 3. US FDA. Resources for Information. Available from: https://www.fda.gov/drugs/resources-information-approved-drugs/fda-grants-accelerated-approval-fam-trastuzumab-deruxtecan-nxki-unresectable-or-metastatic-her2 (Accessed April 12, 2024)

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please click on the link below:

Poster

https://bit.ly/4bhqgya

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from ASCO and the authors of this poster.

This study was supported and sponsored by AstraZeneca in collaboration with Daiichi Sankyo. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201).
Presented at ASCO 2024, May 31–June 4, Chicago, IL, US, by Dr. Do-Youn Oh. Corresponding author email address: ohdoyoun@snu.ac.kr.

Introduction

- HER2 expression is seen in a wide range of solid tumors and is associated with a biologically aggressive phenotype[1–5]
- HER2-directed therapy is standard of care in HER2-expressing and HER2-low unresectable or metastatic breast cancer, HER2-positive unresectable or metastatic gastric and colorectal cancers, and gastroesophageal junction adenocarcinomas, and HER2-mutant non-small cell lung cancer (NSCLC)[6–10]
 - Other HER2-expressing solid tumors are associated with a poor prognosis, with limited treatments and many patients experiencing disease progression on standard therapies[11–14]
- In DESTINY-PanTumor02, T-DXd demonstrated clinically meaningful objective response rates (ORRs), progression-free survival (PFS), and overall survival (OS) in HER2-expressing solid tumors[15]
 - The greatest benefit was observed in patients with HER2 IHC 3+[15]
- In April 2024, T-DXd was granted accelerated approval in the USA for adult patients with unresectable or metastatic HER2-positive (IHC 3+) solid tumors that have progressed after prior treatment and have no alternatives[16]
- This poster reports further subgroup analyses from the DESTINY-PanTumor02 biliary tract and pancreatic cohorts, and characterizes patients with an OR

HER2 prevalence[3,17–20]

	Biliary tract cancer	Pancreatic cancer
IHC 3+	4–13%	1–7%
IHC 2+	12–24%	6–9%

Methods

Patient population

- Aged ≥18 years
- Histologically confirmed locally advanced, unresectable, solid cancers (excluding breast, colorectal, gastric, and NSCLC)
- Progression after ≥1 prior systemic treatment or without treatment options
- Prior HER2-directed therapy allowed
- HER2-expressing tumors with IHC 3+/2+ scored using current ASCO / College of American Pathology guidelines for scoring HER2 in gastric cancer (in situ hybridization testing not required)[21]
 - Patients were enrolled based on local HER2 IHC assessment, where available; otherwise, enrollment was based on central testing
 - HER2 IHC status was assessed centrally using HER2 HercepTest (DAKO) and scored according to gastric-specific criteria[21]
- Eastern Cooperative Oncology Group (ECOG) performance status: 0–1

Study type

Open label, multicenter, multicohort, Phase 2

Treatment

T-DXd 5.4 mg/kg IV Q3W (n≈40 per cohort*)

Trial registration

NCT04482309

Data cutoff

June 8, 2023

Endpoints

Primary:
- Confirmed ORR†

Secondary:
- DOR
- DCR
- PFS
- OS
- Safety and tolerability

Exploratory:
- Subgroup analyses by HER2 status
- Subgroup analyses by biomarkers

Endometrial
Cervical
Ovarian
Bladder
Other tumors‡
Biliary tract
Pancreatic

*Planned recruitment; cohorts with no ORs in the first 15 patients were to be closed †confirmed ORR per RECIST 1.1, as assessed by investigator; ‡patients with tumors that express HER2 (IHC 3+ or 2+), excluding tumors in the tumor-specific cohorts, and breast cancer, NSCLC, gastric cancer, and colorectal cancer. DCR, disease control rate; DOR, duration of response; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; IV, intravenous; NSCLC, non-small cell lung cancer; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; Q3W, every 3 weeks; RECIST, Response Evaluation Criteria in Solid Tumours; T-DXd, trastuzumab deruxtecan

Results and interpretation

- In the biliary tract cancer cohort, all 41 patients assigned to treatment received T-DXd (Table 1)
 - The most common reason for treatment discontinuation was objective disease progression (46.3%)
- In the pancreatic cancer cohort, all 25 patients assigned to treatment received T-DXd (Table 1)
 - Enrollment to the pancreatic cancer cohort was closed early as per study protocol, as no ORs were observed in the first 15 patients enrolled; at this timepoint, 25 patients had been enrolled
 - The most common reason for treatment discontinuation was objective disease progression (52.0%)
- Median (range) follow up in the biliary tract cancer cohort and pancreatic cancer cohort was 6.01 (0.7–29.1) months and 4.99 (1.1–27.2) months, respectively

Table 1. Patient disposition

	Biliary tract cancer	Pancreatic cancer
Assigned to treatment, n	41	25
Treated, n	41	25
T-DXd treatment ongoing at data cutoff,* n (%)	2 (4.9)	0
Discontinued treatment at data cutoff,* n (%)	39 (95.1)	25 (100)
Objective disease progression	19 (46.3)	13 (52.0)
Adverse event	8 (19.5)	3 (12.0)
Patient decision	1 (2.4)	1 (4.0)
Investigator decision	0	1 (4.0)
Subjective disease progression	4 (9.8)	6 (24.0)
Other†	7 (17.1)	1 (4.0)
Median treatment cycles received‡ (range)	5.0 (1–34)	3.0 (1–18)

*Data cutoff was June 8, 2023; †in case of death while on treatment, investigators did not specifically record a reason for discontinuation of T-DXd; ‡a treatment cycle was 21 days
T-DXd, trastuzumab deruxtecan

- Seven patients in the biliary tract cancer cohort and 3 patients in the pancreatic cancer cohort who were enrolled as HER2 IHC 3+/2+ by local testing, as per the eligibility criteria, had a central IHC status of IHC 1+/0/unknown
- The patient population across both the biliary tract and pancreatic cohorts was heavily pretreated (Table 2)
 - In the biliary tract cancer cohort, 7 (17.1%) patients had received prior anti-HER2 therapy, and 8 (19.5%) patients had received prior topoisomerase I (TOP1) inhibitor therapy
 - In the pancreatic cancer cohort, 2 (8.0%) patients had received prior anti-HER2 therapy, and 18 (72.0%) patients had received prior TOP1 inhibitor therapy
- In the pancreatic cancer cohort, 15 (60.0%) patients had a Kirsten rat sarcoma viral oncogene homolog (KRAS) mutation, as detected by circulating tumor DNA (ctDNA; Table 2)

Table 2. Baseline demographics and clinical characteristics

Characteristic			Biliary tract cancer (n=41)	Pancreatic cancer (n=25)
Age, median (range), years			64.0 (31–80)	62.0 (23–80)
Race, n (%)		White	20 (48.8)	17 (68.0)
		Asian	21 (51.2)	6 (24.0)
		Black or African American	0	1 (4.0)
		Other	0	1 (4.0)
ECOG performance status, n (%)		0	13 (31.7)	8 (32.0)
		1	28 (68.3)	17 (68.0)
HER2 status by enrollment test, n (%)		IHC 3+	22 (53.7)	5 (20.0)
		IHC 2+	19 (46.3)	20 (80.0)
HER2 status by central testing, n (%)		IHC 3+	16 (39.0)	2 (8.0)
		IHC 2+	14 (34.1)	19 (76.0)
		IHC 1+	3 (7.3)	1 (4.0)
		IHC 0	7 (17.1)	3 (12.0)
		IHC unknown	1 (2.4)	0
Prior regimens	Median (range)		2 (1–5)	2 (1–4)
	n (%)	1	14 (34.1)	7 (28.0)
		≥2	27 (65.9)	18 (72.0)
Prior anti-HER2 therapy, n (%)			7 (17.1)	2 (8.0)
Prior TOP1 inhibitor therapy, n (%)			8 (19.5)	18 (72.0)
Prior IO therapy, n (%)			5 (12.2)	0
PD-L1 IC prevalence, n (%)		≥1%	21 (51.2)	7 (28.0)
		<1%	14 (34.1)	15 (60.0)
		Unknown*	6 (14.6)	3 (12.0)
KRAS,† n (%)		Mutation detected	4 (9.8)	15 (60.0)
		Mutation not detected	36 (87.8)	8 (32.0)
		Unknown†	1 (2.4)	2 (8.0)

Fifteen patients in the pancreatic cancer cohort had a KRAS mutation detected (central IHC 2+ [n=12], IHC 1+ [n=1], IHC 0 [n=2]). *Data unknown owing to insufficient or no tumor tissue available, or technical problems; †evaluated in a central laboratory and detected by ctDNA
ctDNA, circulating tumor DNA; ECOG, Eastern Cooperative Oncology Group; HER2, human epidermal growth factor receptor 2; IC, immune cell; IHC, immunohistochemistry; IO, immuno-oncology; KRAS, Kirsten rat sarcoma viral oncogene homolog; PD-L1, programmed cell death ligand 1; TOP1, topoisomerase I

- In the biliary tract cohort, the local test for enrollment and the central test had a positive percentage agreement of 66.7% for IHC 3+ and 46.7% for IHC 2+ (Figure 1)
- In the pancreatic cancer cohort, the local test for enrollment and the central test had a positive percent agreement of 25.0% for IHC 3+ and 72.7% for IHC 2+ (Figure 1)

Figure 1. Agreement between enrollment HER2 test and central HER2 test

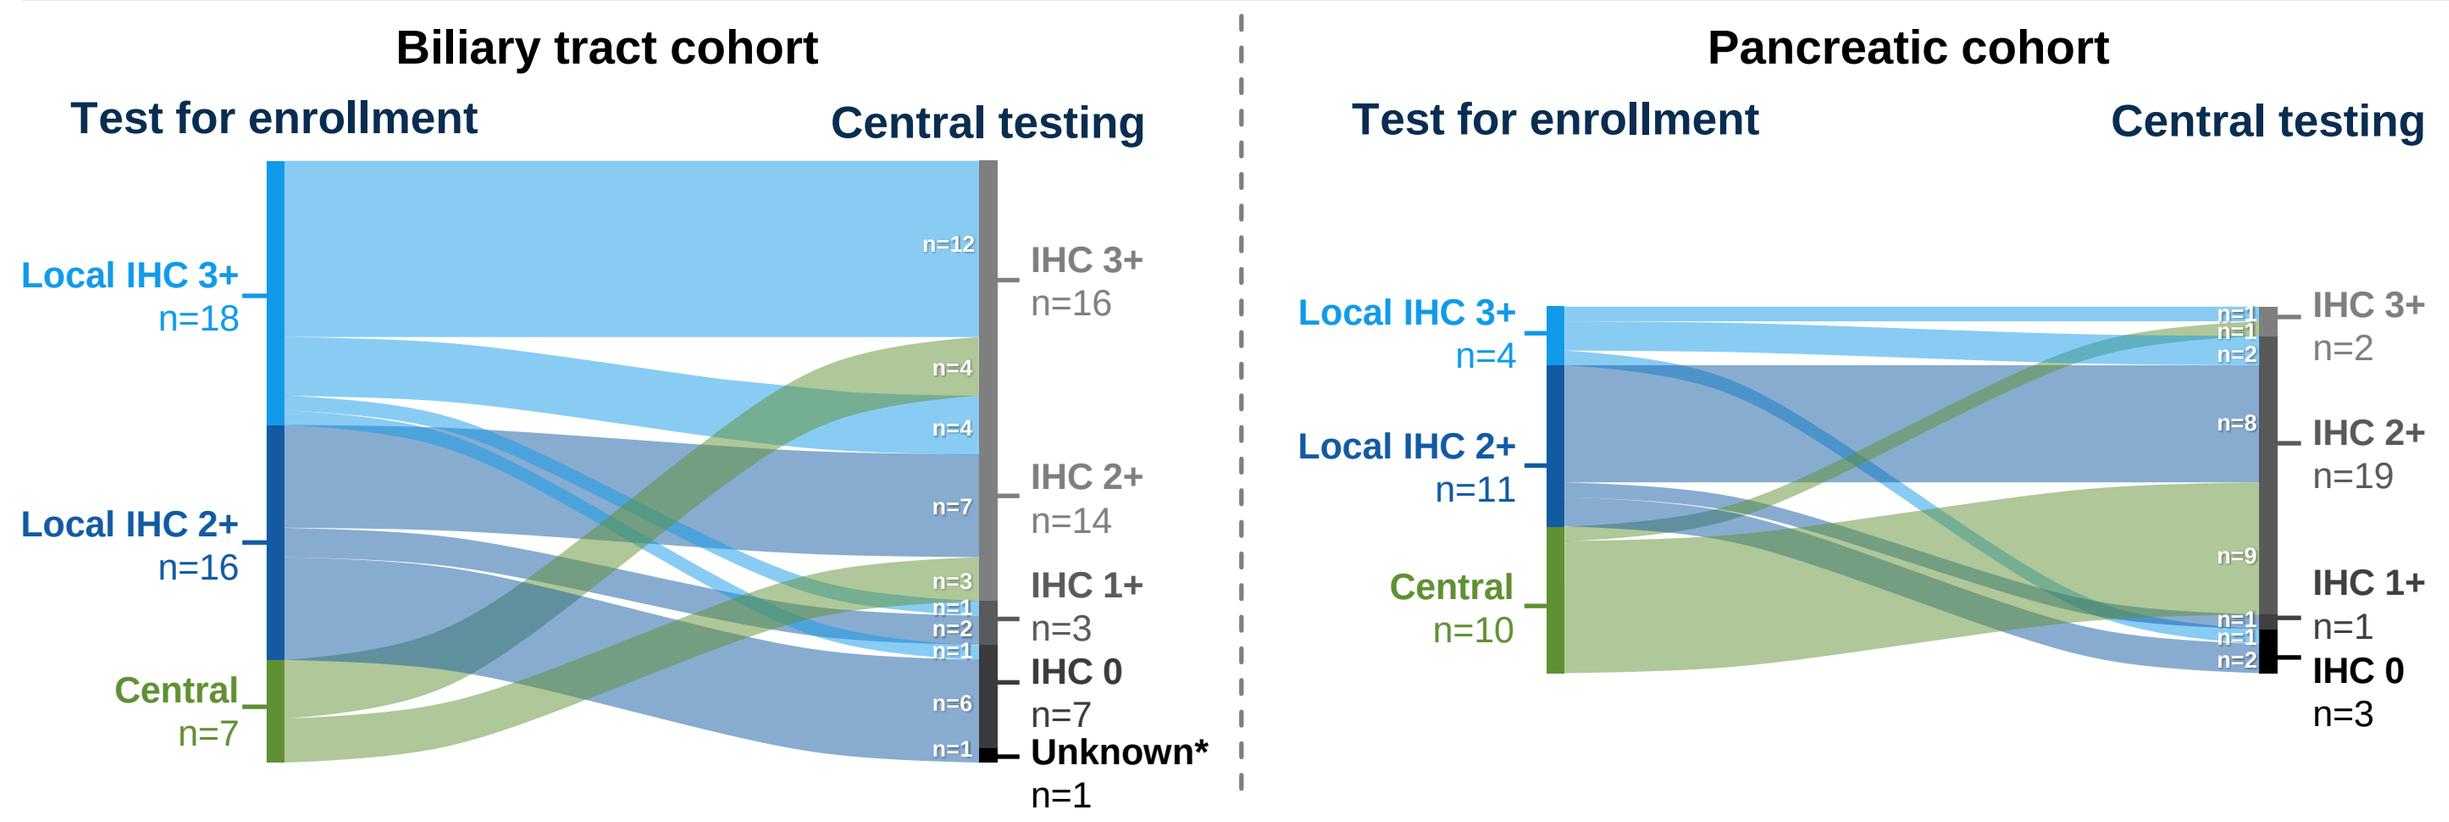

Biliary tract cohort
Test for enrollment
Central testing
Local IHC 3+ n=18
Local IHC 2+ n=16
Central n=7
IHC 3+ n=16
IHC 2+ n=14
IHC 1+ n=3
IHC 0 n=7
Unknown* n=1
Pancreatic cohort
Test for enrollment
Central testing
Local IHC 3+ n=4
Local IHC 2+ n=11
Central n=10
IHC 3+ n=2
IHC 2+ n=19
IHC 1+ n=1
IHC 0 n=3

Patients with a central HER2 IHC status of 1+/0/unknown were enrolled as HER2 IHC 3+/2+ by local testing, as per the eligibility criteria. *Includes patients whose samples were not evaluable and may have included patients who did not provide a sample for central testing
HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry

Efficacy: biliary tract cohort

- The ORR for all patients in the biliary tract cohort by investigator and by independent central review was 22.0% and 26.8%, respectively (Figure 2)
 - All 9 patients (22.0%) who achieved an OR by investigator had a central HER2 status of IHC 3+ (Figure 2; primary tumor sites were ampulla of Vater [n=2/10], extrahepatic [n=2/8], gall bladder [n=4/10], and intrahepatic [n=1/13])
- Median PFS and OS by investigator assessment for all patients in the biliary tract cohort was 4.6 and 7.0 months, respectively (Table 3)

Figure 2. ORR in the biliary tract cohort

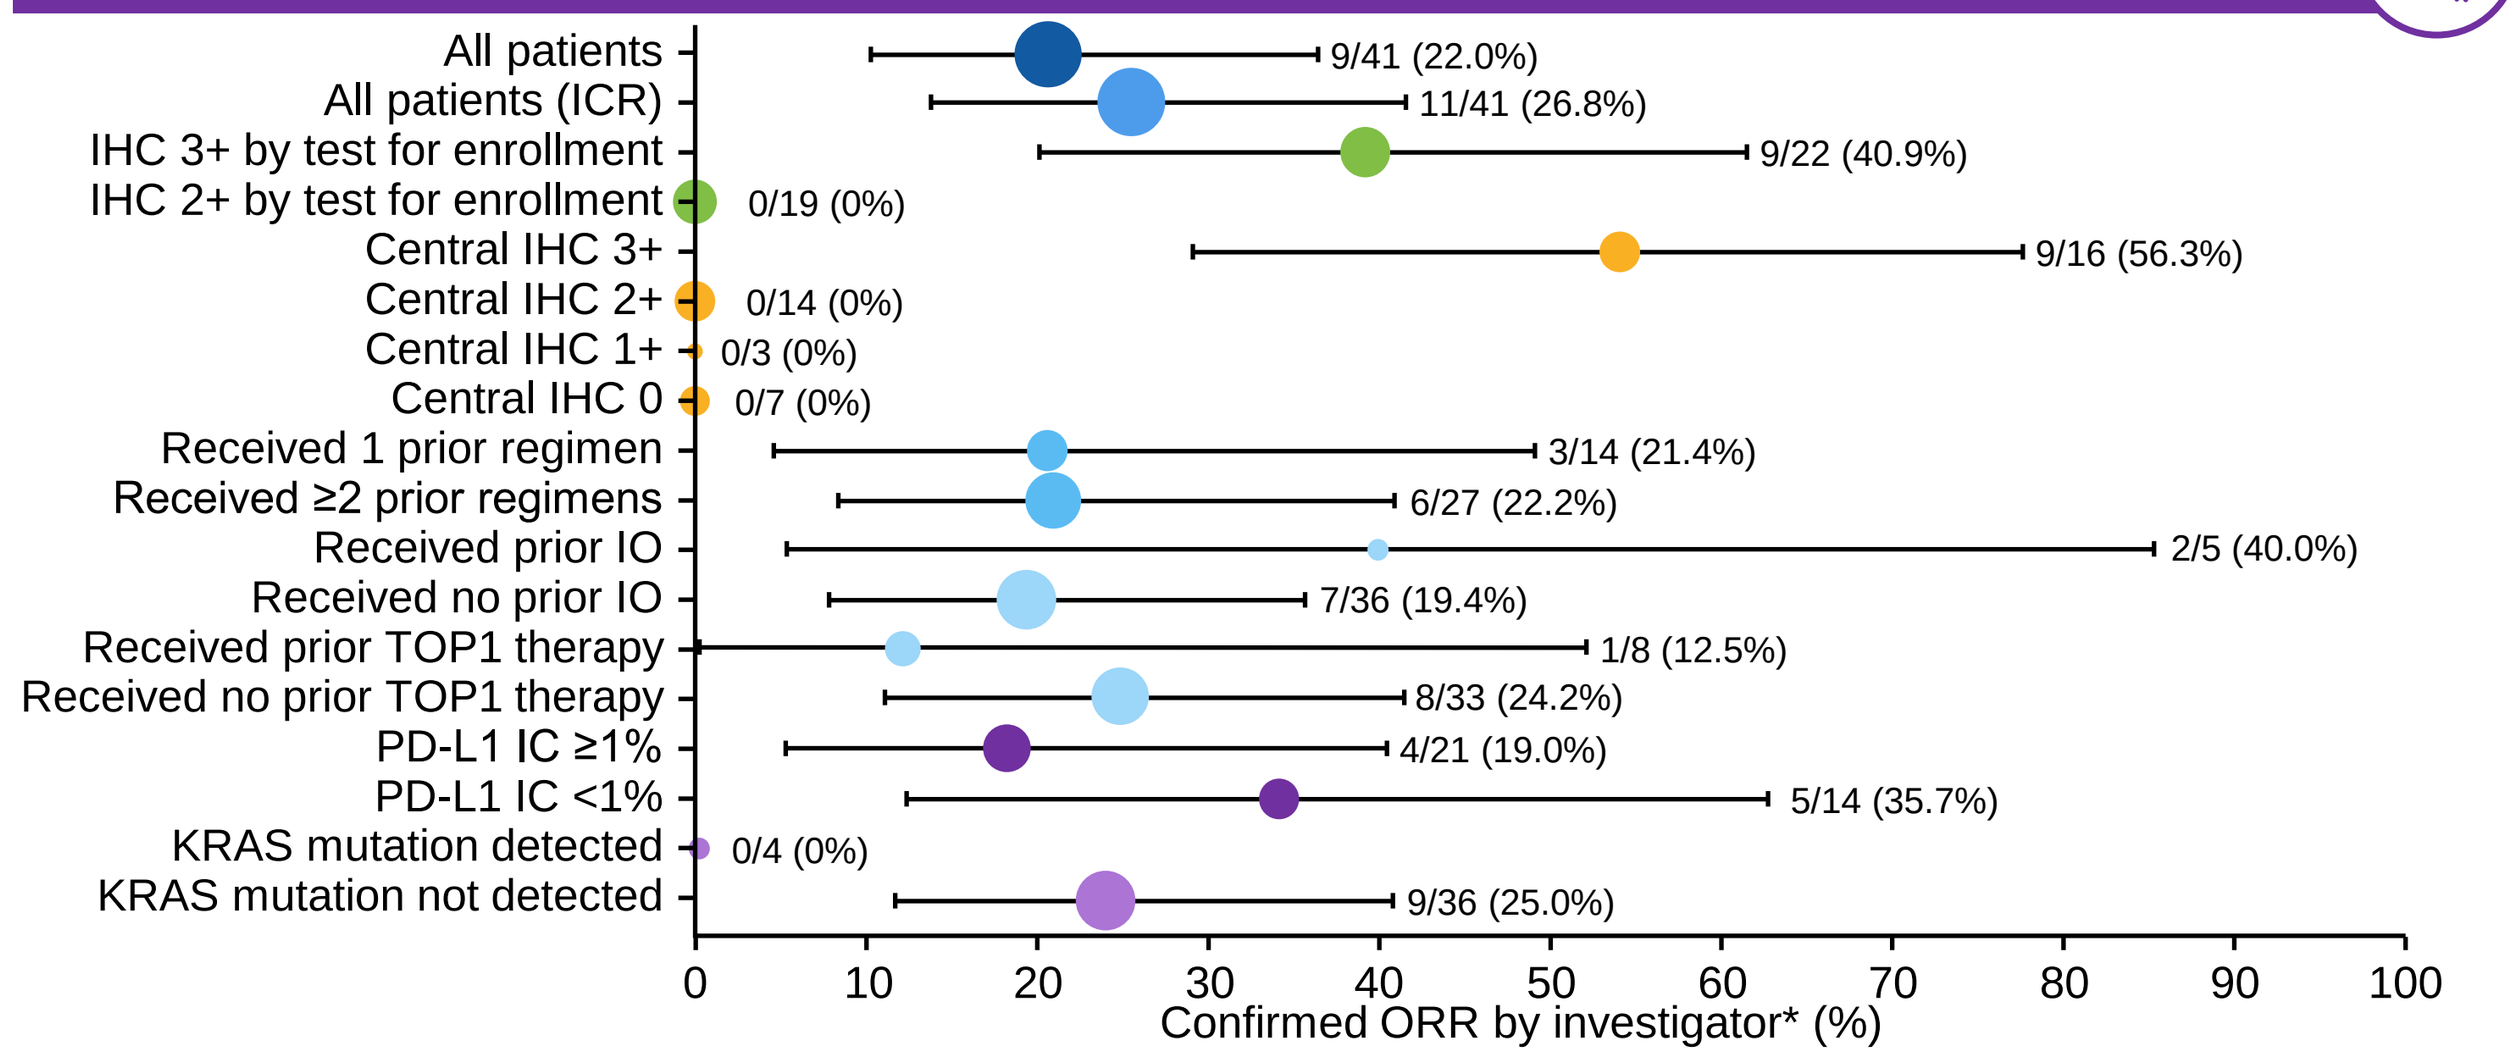

All patients
All patients (ICR)
IHC 3+ by test for enrollment
IHC 2+ by test for enrollment
Central IHC 3+
Central IHC 2+
Central IHC 1+
Central IHC 0
Received 1 prior regimen
Received ≥2 prior regimens
Received prior IO
Received no prior IO
Received prior TOP1 therapy
Received no prior TOP1 therapy
PD-L1 IC ≥1%
PD-L1 IC <1%
KRAS mutation detected
KRAS mutation not detected
9/41 (22.0%)
11/41 (26.8%)
9/22 (40.9%)
0/19 (0%)
9/16 (56.3%)
0/14 (0%)
0/3 (0%)
0/7 (0%)
3/14 (21.4%)
6/27 (22.2%)
2/5 (40.0%)
7/36 (19.4%)
1/8 (12.5%)
8/33 (24.2%)
4/21 (19.0%)
5/14 (35.7%)
0/4 (0%)
9/36 (25.0%)
0 10 20 30 40 50 60 70 80 90 100
Confirmed ORR by investigator* (%)

Response determined by investigator assessment according to RECIST 1.1. Patients with a central HER2 IHC status of 0/unknown were enrolled as HER2 IHC 3+/2+ by local testing. Circle sizes are proportional to the number of patients in each subgroup. Prior therapy and biomarker subgroup analyses do not account for HER2 IHC status. *Unless stated otherwise. IC, immune cell; ICR, independent central review; IHC, immunohistochemistry; IO, immuno-oncology; KRAS, Kirsten rat sarcoma viral oncogene homolog; ORR, objective response rate; PD-L1, programmed cell death ligand 1; RECIST, Response Evaluation Criteria in Solid Tumours; TOP1, topoisomerase I

Table 3. Secondary efficacy outcomes in the biliary tract cohort (central testing)

Characteristic	All patients	All patients (ICR)	HER2 IHC 3+	HER2 IHC 2+	HER2 IHC 1+	HER2 IHC 0
n	41	41	16	14	3	7
Confirmed ORR, n (%) 95% CI	9 (22.0) 10.6, 37.6	11 (26.8) 14.2, 42.9	9 (56.3) 29.9, 80.2	0	0	0
Median DOR, months 95% CI	8.6 2.1, NE	10.9 5.5, NE	8.6 2.1, NE	–	–	–
Median PFS, months 95% CI	4.6 3.1, 6.0	4.1 2.8, 5.3	7.4 2.8, 12.5	4.2 2.8, 6.0	5.1 1.2, NE	3.1 1.2, 5.6
Median OS, months 95% CI	7.0 4.6, 10.2	7.0 4.6, 10.2	12.4 2.8, NE	6.0 3.7, 11.7	5.1 1.6, NE	7.6 3.0, 10.2
DCR at 12 weeks, % 95% CI	65.9 49.4, 79.9	51.2 35.1, 67.1	68.8 41.3, 89.0	71.4 41.9, 91.6	66.7 9.4, 99.2	57.1 18.4, 90.1

OR determined by investigator assessment according to RECIST 1.1; DOR defined as time from date of first documented response (complete or partial), until the date of documented progression or death in the absence of disease progression; PFS defined as time from first dose until date of objective disease progression or death, regardless of withdrawal or receipt of another cancer therapy. OS defined as time from date of first dose until death due to any cause; DCR defined as percentage of patients with a best OR of confirmed complete response or partial response, or with stable disease for at least 5 weeks after first dose. HER2 status reported as confirmed by central testing. Patients with a central HER2 IHC status of 1+/0/unknown were enrolled as HER2 IHC 3+/2+ by local testing. CIs omitted where 0%. CI, confidence interval; DCR, disease control rate; DOR, duration of response; HER2, human epidermal growth factor receptor 2; ICR, independent central review; IHC, immunohistochemistry; NE, not evaluable; OR, objective response; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; RECIST, Response Evaluation Criteria in Solid Tumours

Efficacy: pancreatic cohort

- The ORR for all patients by investigator and by independent central review was 4.0% (1/25 patients) and 12.0% (3/25 patients), respectively
- For patients with central HER2 IHC 2+, ORR by investigator and by independent central review was 5.3% (1/19 patients) and 15.8% (3/19 patients), respectively
- The patient (4.0%) who achieved an OR by investigator had a central HER2 status of IHC 2+ and a non-mutated KRAS status, as detected by ctDNA
- The median DOR and median PFS by investigator assessment for all patients in the pancreatic cohort were 5.7 and 3.2 months, respectively (Table 4)

Table 4. Secondary efficacy outcomes in the pancreatic cohort (central testing)

Characteristic	All patients	All patients (ICR)	HER2 IHC 3+	HER2 IHC 2+	HER2 IHC 1+	HER2 IHC 0
n	25	25	2	19	1	3
Confirmed ORR, n (%) 95% CI	1 (4.0) 0.1, 20.4	3 (12.0) 2.5, 31.2	0	1 (5.3) 0.1, 26.0	0	0
Median DOR, months 95% CI	5.7	NR 5.8, NE	–	5.7	–	–
Median PFS, months 95% CI	3.2 1.8, 7.2	3.2 1.4, 7.0	5.4 2.8, NE	2.8 1.4, 9.1	3.9	3.2 1.2, NE
Median OS, months 95% CI	5.0 3.8, 14.2	5.0 3.8, 14.2	12.4 8.8, NE	4.9 2.4, 15.7	9.2	3.8 3.2, NE
DCR at 12 weeks, % 95% CI	36.0 18.0, 57.5	44.0 24.4, 65.1	50.0 1.3, 98.7	36.8 16.3, 61.6	100	0 0, 70.8

OR determined by investigator assessment according to RECIST 1.1; DOR defined as time from date of first documented response (complete or partial), until the date of documented progression or death in the absence of disease progression; PFS defined as time from first dose until date of objective disease progression or death, regardless of discontinuation of treatment or receipt of another cancer therapy. OS defined as time from date of first dose until death due to any cause; DCR defined as percentage of patients with a best OR of confirmed complete response or partial response, or with stable disease for at least 11 weeks after first dose. HER2 status reported as confirmed by central testing. Patients with a central HER2 IHC status of 1+/0/unknown were enrolled as HER2 IHC 3+/2+ by local testing. CIs omitted where n=1 and where 0%
CI, confidence interval; DCR, disease control rate; DOR, duration of response; HER2, human epidermal growth factor receptor 2; ICR, independent central review; IHC, immunohistochemistry; NE, not evaluable; NR, not reached; OR, objective response; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; RECIST, Response Evaluation Criteria in Solid Tumours

Safety

- Grade ≥3 drug-related TEAEs were experienced by 19 (39.0%) patients with biliary tract cancer (Table 5)
 - The most common (>5%) Grade ≥3 drug-related TEAEs in the biliary tract cohort were neutropenia (9.8%), neutrophil count decreased (9.8%), nausea (7.3%), and fatigue (7.3%)
 - Neutropenia and neutrophil count decrease are listed separately owing to how events were coded based on reported terms (adverse event or laboratory abnormality)
- Adjudicated drug-related ILD/pneumonitis occurred in 7/41 (17.1%) patients with biliary tract cancer (Grade 2: n=5; Grade 3: n=1; Grade 5: n=1)
- Grade ≥3 drug-related TEAEs were experienced by 7 (28.0%) patients with pancreatic cancer (Table 5)
 - The most common (>5%) Grade ≥3 drug-related TEAEs in the pancreatic cohort were anemia (8.0%) and neutropenia (8.0%)
- Adjudicated drug-related ILD/pneumonitis occurred in 1/25 (4.0%) patients with pancreatic cancer (Grade 1)

Table 5. Safety summary

n (%)	Biliary tract cancer (n=41)	Pancreatic cancer (n=25)
Any drug-related TEAEs	33 (80.5)	15 (60.0)
Drug-related TEAEs Grade ≥3	19 (39.0)	7 (28.0)
Serious drug-related TEAEs	5 (12.2)	3 (12.0)
Drug-related TEAEs associated with dose discontinuations	5 (12.2)	1 (4.0)
Drug-related TEAEs associated with dose interruptions	7 (17.1)	0
Drug-related TEAEs associated with dose reductions	9 (22.0)	0
Drug-related TEAEs associated with deaths	0	0

Analyses (by investigator) include patients with biliary tract cancer and pancreatic cancer who received ≥1 dose of T-DXd (n=41 and n=25, respectively); median total treatment duration was 3.45 (range 0.7–23.7) months and 2.07 (range 0.7–12.4) months, respectively
TEAE, treatment-emergent adverse event

Acknowledgments

Thank you to the patients and their caregivers for their participation, and the study site staff for their contributions. The authors would also like to thank members of the interstitial lung disease adjudication committee. This study was supported and sponsored by AstraZeneca in collaboration with Daiichi Sankyo. Under the guidance of the authors and in accordance with Good Publication Practice, medical writing and editorial support was provided by Zoë Hine, BSc, of Helios Medical Communications, part of Helios Global Group, and was funded by AstraZeneca.

Disclosures – Dr. Do-Youn Oh

Consulting or advisory role: Arcus Biosciences, ASLAN Pharmaceuticals, AstraZeneca, Basilea, Bayer, BeiGene, Celgene, Genentech/Roche, Halozyme, IQVIA, Merck Serono, MSD Oncology, Novartis, Taiho Pharmaceutical, Turning Point Therapeutics, Yuhan, Zymeworks. **Research funding:** Array BioPharma, AstraZeneca, BeiGene, Handok, Lilly, MSD, Novartis, SERVIER.

References

1. Yan M, et al. *Cancer Metastasis Rev*. 2015;34:157–164
2. Li Z, et al. *EBioMedicine*. 2020;62:103074
3. Uzunparmak B, et al. *Ann Oncol*. 2023;34:1035–1046
4. Xing F, et al. *Mol Cancer*. 2023;22:6
5. Halle MK, et al. *Br J Cancer*. 2018;118:378–387
6. Owen DH, et al. *J Clin Oncol*. 2023;41:e10–e20
7. Shah MA, et al. *J Clin Oncol*. 2023;41:1470 –1491
8. Giordano SH, et al. *J Clin Oncol*. 2022;40:2612–2635
9. Cervantes A, et al. *Ann Oncol*. 2023;34:10–32
10. Lordick F, et al. *Ann Oncol*. 2022;33:1005–1020
11. Oh D-Y, Bang Y-J. *Nat Rev Clin Oncol*. 2020;17:33–48
12. Diver EJ, et al. *Oncologist*. 2015;20:1058–1068
13. Kurokawa Y, et al. *Gastric Cancer*. 2015;18:691–697
14. Luo H, et al. *PLoS One*. 2018;13:e0191972
15. Meric-Bernstam F, et al. *J Clin Oncol*. 2024;42:47–58
16. Enhertu (fam-trastuzumab deruxtecan-nxki) highlights of prescribing information. 2024. Available from: www.accessdata.fda.gov/drugsatfda_docs/label/2024/761139s028lbl.pdf (Accessed April 19, 2024)
17. Vivaldi C, et al. *Oncologist*. 2020;25:886–893
18. Roa I, et al. *Gastrointest Cancer Res*. 2014;7:42–48
19. Chou A, et al. *Genome Med*. 2013;5:78
20. Han SH, et al. *Diagnostics (Basel)*. 2021;11:653
21. Hofmann M, et al. *Histopathology*. 2008;52:797–805

DESTINY PANTUMOR02