EE417

Healthcare resource use and costs of care in HR+/HER2- metastatic breast cancer: a retrospective US claims data study

Simon M Collin,[1] Jenny Tse,[2] Nazneen Fatima Shaikh,[2] Aimee Near,[2] Erika Hamilton,[3] Adam Brufsky,[4] William Gradishar,[5] Sandhya Mehta,[6] Gráinne Long,[1] Nikki Toms[1]

[1]AstraZeneca Pharmaceuticals Ltd., Cambridge, UK; [2]Real World Solutions, IQVIA, Durham, NC, USA; [3]Sarah Cannon Research Institute, Nashville, TN, USA; [4]Department of Medicine, University of Pittsburgh, Pittsburgh, PA, USA; [5]Robert H. Lurie Comprehensive Cancer Center, Northwestern University Feinberg School of Medicine, Chicago, IL, USA; [6]Daiichi Sankyo Inc., Basking Ridge, NJ, USA

Objective

- This retrospective study quantified real-world healthcare resource use (HCRU) and healthcare costs among patients with hormone receptor (HR)-positive, human epidermal growth factor receptor 2 (HER2) expression-negative metastatic breast cancer (HR+/HER2- mBC) in the United States (US).

Conclusions

- All-cause and BC-related healthcare costs for patients with HR+/HER2- mBC were numerically higher during targeted therapy regimens (mainly CDK4/6 inhibitor with endocrine therapy) compared to endocrine therapy only or chemotherapy regimens.
- However, the costs of clinical events of interest (CEIs) were highest during chemotherapy regimens and may be a consequence of greater toxicity relative to endocrine only or targeted therapy.
- Overall, the high economic and clinical burden of standard of care indicates a need for newer treatments that balance costs of care, including drug costs, with adverse events and clinical benefit, particularly for patients currently treated with chemotherapy.

Plain language summary

Why did we perform this research?

The treatment landscape for metastatic breast cancer has expanded in recent years. This study uses recent healthcare claims data to quantify healthcare resource use and costs among patients treated for the HR+/HER2- subtype of metastatic breast cancer in real-world settings.

How did we perform this research?

This retrospective cohort study used the IQVIA PharMetrics® Plus database to identify patients treated for HR+/HER2- metastatic breast cancer.

What were the findings of this research?

Costs related to all causes and to breast cancer were highest during treatment with targeted therapies (mainly CDK4/6 inhibitor with endocrine therapy), followed by chemotherapy and endocrine therapy only regimens.

Costs due to clinical events varied according to the treatment regimen. During chemotherapy, clinical event-related costs accounted for 66% of average breast cancer-related costs. In contrast, clinical event-related costs were 25% and 11% of average breast cancer-related costs during endocrine therapy only and targeted therapy, respectively.

What are the implications of this research?

The high treatment costs and burden due to clinical events related to targeted agents with endocrine therapy and to chemotherapy regimens indicate a need for newer treatments that balance costs of care with adverse events and clinical benefit.

Corresponding author email address: simon.collin@astrazeneca.com

Poster presented at the ISPOR 2024 Annual Conference, Atlanta, GA, USA; May 5-8

Introduction

- Treatment options for hormone receptor (HR)-positive, human epidermal growth factor receptor 2 (HER2) expression-negative metastatic breast cancer (HR+/HER2- mBC) include endocrine therapy, targeted therapy, and chemotherapy.
- CDK4/6 inhibitors with endocrine therapy are the preferred first-line treatments.[1]
- Drug toxicity is a key consideration for treatment decision-making,[2] especially for chemotherapy.[3]
- This retrospective study used healthcare claims data to quantify real-world healthcare resource use (HCRU) and healthcare costs among patients with HR+/HER2- mBC in the United States (US), including the burden of clinical events of interest (CEIs).

Methods

Study design: Retrospective cohort study.

Data source: IQVIA PharMetrics® Plus database, comprised of fully adjudicated, de-identified medical and pharmacy claims.

Study population:

- Adults (≥18 years) with ≥1 ICD-10 diagnosis code for BC; the date of the earliest diagnosis code for metastasis in the selection window (Mar 1, 2019 to Nov 30, 2022) was the index date (**Fig 1**).
- All patients had 6-month baseline and ≥3 months follow-up period, and met a treatment-based proxy for HR+/HER2- mBC during follow-up:
 - With ≥1 treatment indicated for HR+/HER2- or HR+ mBC, and
 - With no treatments for HER2-positive (HER2+) or triple-negative BC (TNBC).

Outcomes and analysis:

- All-cause, BC-related, and CEI-related HCRU and costs per patient per month (PPPM), including BC treatment costs, were reported during follow-up for the overall cohort and during treated follow-up (up to the 4th line of therapy), by treatment category.
 - BC-related utilization was determined via diagnosis codes or treatments.
 - CEI-related claims, a subset of BC-related utilization, included prophylaxis and treatment of potential adverse effects of BC therapies.
- A hierarchy was applied to assign treatment regimens:
 - Endocrine therapy: regimens with endocrine therapy only (aromatase inhibitor, SERM, GnRH agonist, hormone therapy and others).
 - Chemotherapy: taxane, anthracyclines, cytotoxic, platinum-based and other chemotherapy ± endocrine therapy.
 - Targeted therapy: CDK4/6, mTOR, PIK3CA, PARP, VEGF inhibitors, and ADCs ± endocrine therapy/chemotherapy.
- CEIs were identified using diagnosis codes for hematological, gastrointestinal, hepatic, and ocular events, alopecia, interstitial lung disease, pneumonitis, fatigue, infusion-related reactions, rash, hyperglycemia, and sinus bradycardia.
 - Patients with baseline evidence of chronic conditions (e.g., alopecia) were not considered as at-risk for that CEI during follow-up.
 - For acute conditions, patients were at-risk for another CEI occurrence after 14 days from the initial diagnosis code for the CEI category.

Results

Patient characteristics

- There were 11,419 treated HR+/HER2- mBC patients meeting selection criteria (**Fig 1**).
- The cohort was 98.9% female and the median (Q1, Q3) age was 57 (50, 63) years (**Table 1**).
- The most prevalent comorbidities were obesity (17.2%), diabetes (7.7%), and chronic pulmonary disease (3.2%), based on diagnosis codes.

Figure 1. Patient attrition

With ≥1 diagnosis for breast cancer during the selection window (Mar 1, 2019 – Nov 30, 2022)
N=373,263

↓

With ≥1 diagnosis for metastatic cancer in the selection window and on/after the date of breast cancer diagnosis; the date of earliest metastasis diagnosis was the index date
N=68,600

↓

With 6 months of continuous enrollment before the index date (i.e., 6-month baseline period)
N=50,455

↓

With ≥3 months of continuous enrollment after the index date (i.e., ≥ 3-month follow-up period)
N=45,603

→ Excluded:
- ≥1 diagnosis for any other primary cancer or metastasis up to 15 months before the index date (N=24,484)
- With incomplete data or other data quality issues (N=620)
- Age <18 years in the index year (N=6)
- Evidence of clinical trial enrollment or pregnancy during the study period (N=1,376)

↓

With evidence of HR+/HER2- mBC during follow-up, using a treatment-based proxy*
N=11,429

↓

With ≥1 line of therapy during follow-up
N=11,419

HR+/HER2- mBC Cohort
N=11,419

*Patients with fam-trastuzumab deruxtecan on/after Aug 6, 2022 or sacituzumab govitecan on/after Feb 3, 2023 were allowed to remain in the cohort, due to the respective expanded approvals for HER2- low and HR+/HER2- mBC.

Table 2. Demographic and baseline clinical characteristics

Measures		HR+/HER2- mBC Cohort N=11,419
Age (years)	Median (Q1, Q3)	57 (50, 63)
Sex (n, %)	Female	11,292 (98.9)
Geographic region* (n, %)	South	4,401 (38.5)
	Midwest	3,338 (29.2)
	Northeast	1,937 (17.0)
	West	1,728 (15.1)
Payer type† (n, %)	Commercial or self-insured	10,262 (89.9)
	Medicare Risk	1,108 (9.7)
	Medicaid	29 (0.3)
NCI comorbidity index (n, %)	0	9,782 (85.7)
	>0 to <1	1,495 (13.1)
	≥1	142 (1.2)
NCI comorbidities‡ (n, %)	Diabetes	883 (7.7)
	Chronic pulmonary disease	369 (3.2)
	Congestive heart failure	183 (1.6)
	Renal disease	156 (1.4)
	Liver disease	117 (1.0)
Sites of metastasis (n, %)	Regional	8,984 (78.7)
	Distant visceral (liver, lung, other visceral sites)	730 (6.4)
	Distant CNS (brain and other CNS)	171 (1.5)
	Other (skin, bone and bone marrow, unspecified sites)	2,218 (19.4)

*0.1% of patients had unknown geographic region.
†0.2% of patients had other or unknown payer type.
‡Only comorbidities present in ≥1% of the cohort are shown.

Treatment patterns

- Over median (Q1, Q3) follow-up of 19.0 (10.5, 30.6) months, 79.4% of patients had endocrine therapy only, 30.6% had chemotherapy, and 23.4% had targeted therapy regimens.
- Most patients with targeted therapy had CDK4/6 inhibitors (>96%). Endocrine therapy was observed in 92.1% of targeted therapy regimens, up to the 4th line of targeted therapy.

CEIs during treated follow-up

- The top CEI categories during treatment, among patients at-risk for the events, are as follows:
 - Endocrine therapy only regimens: fatigue (21.8%), ocular events (15.3%), and gastrointestinal events (14.5%).
 - Chemotherapy regimens: gastrointestinal events (42.8%), hematological events (40.9%), and fatigue (24.7%).
 - Targeted therapy regimens: hematological events (37.4%), gastrointestinal events (28.9%), and fatigue (25.7%).
- CEI-related inpatient and emergency department (ED) visits were more frequent during targeted therapy (17.5% and 8.7%) than during endocrine therapy only (4.5% and 1.3%) **(Figure 2).**

Figure 2. CEI-related inpatient and ED visits by treatment category during treated follow-up

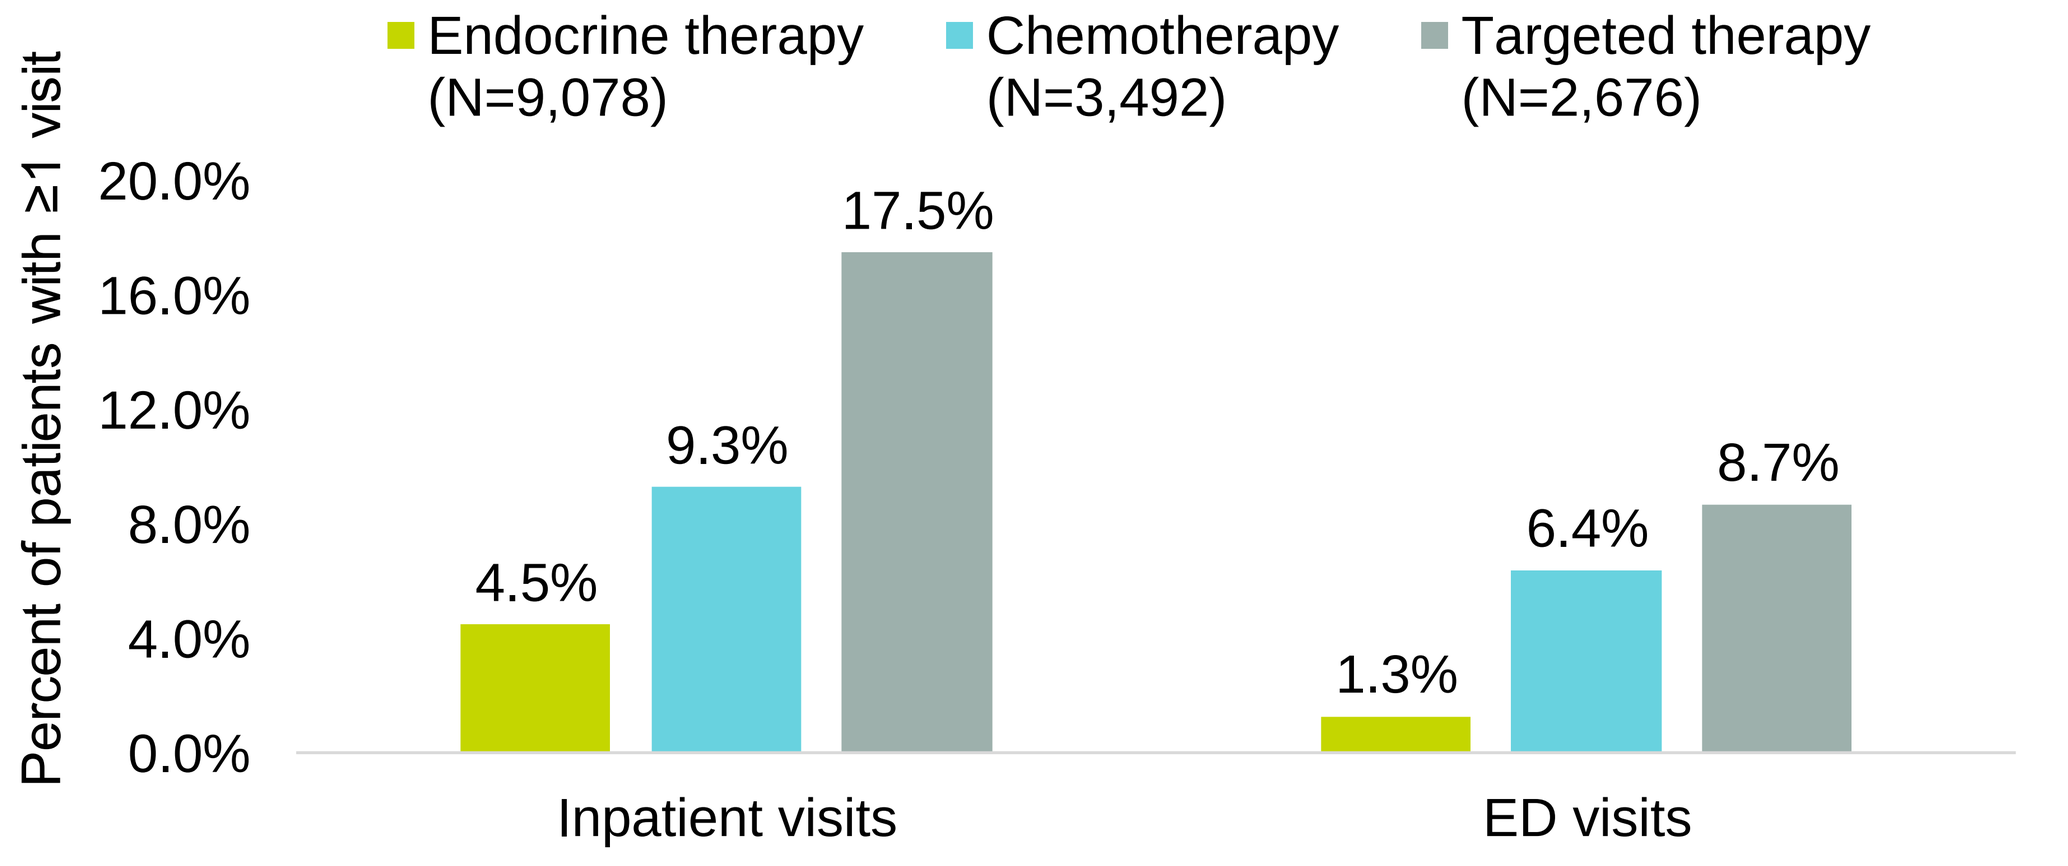
Endocrine therapy (N=9,078)
Chemotherapy (N=3,492)
Targeted therapy (N=2,676)
Percent of patients with ≥1 visit
20.0%
16.0%
12.0%
8.0%
4.0%
0.0%
4.5%
9.3%
17.5%
1.3%
6.4%
8.7%
Inpatient visits
ED visits

Healthcare costs by treatment category

- For the overall cohort, the mean±SD total all-cause costs PPPM were \$8,734±\$9,201 during follow-up, of which 82.3% were BC-related and 17.8% were CEI-related.
- Mean±SD all-cause costs PPPM during treated follow-up were \$4,161±\$21,761 during endocrine therapy only, \$15,902±\$14,301 during chemotherapy, and \$19,384±\$12,318 during targeted therapy **(Figure 3)**.
- The drivers of BC- and CEI-related costs varied by treatment:
 - During targeted therapy, outpatient pharmacy costs were 66.3% of mean BC-related costs (compared to 8.3% during chemotherapy and 9.8% during endocrine therapy only).
 - During chemotherapy, outpatient infused/injectable drug costs were 53.0% of mean BC-related costs (compared to 7.5% during endocrine therapy only and 8.2% during targeted therapy).
 - CEI-related costs were 66.1% of mean BC-related costs during chemotherapy (compared to 24.6% during endocrine therapy only and 10.7% during targeted therapy).

Figure 3. Mean per patient monthly all-cause, BC-related, and CEI-related costs* during treated follow-up

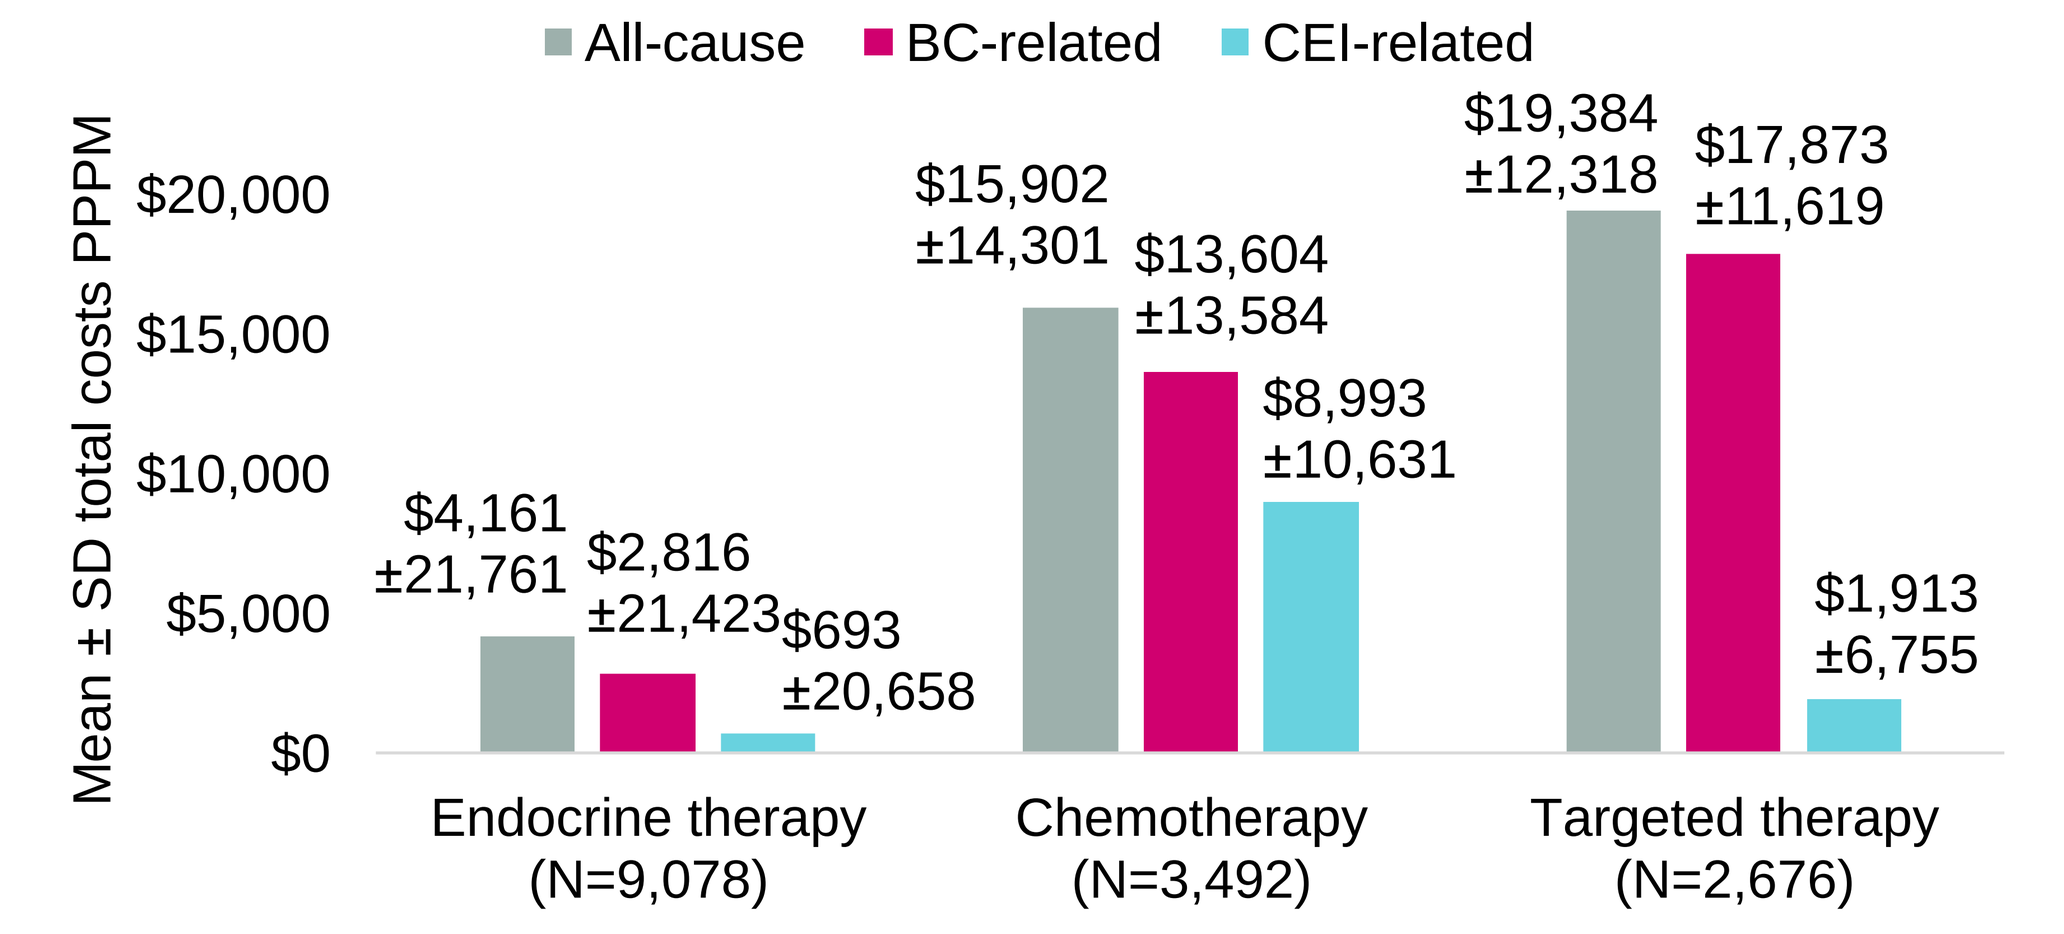
All-cause
BC-related
CEI-related
Mean ± SD total costs PPPM
$20,000
$15,000
$10,000
$5,000
$0
$4,161 ±21,761
$2,816 ±21,423
$693 ±20,658
$15,902 ±14,301
$13,604 ±13,584
$8,993 ±10,631
$19,384 ±12,318
$17,873 ±11,619
$1,913 ±6,755
Endocrine therapy (N=9,078)
Chemotherapy (N=3,492)
Targeted therapy (N=2,676)

*Converted to 2022 USD using the medical component of the Consumer Price Index.

Healthcare costs by treatment category, continued

- During endocrine therapy only and targeted therapy, inpatient visit costs accounted for most CEI-related costs (76.1% and 68.2%, respectively). In contrast, outpatient medical costs contributed to 80.2% of CEI-related costs during chemotherapy **(Figure 4)**.

Figure 4. Subgroups of mean CEI-related costs by treatment category during treated follow-up

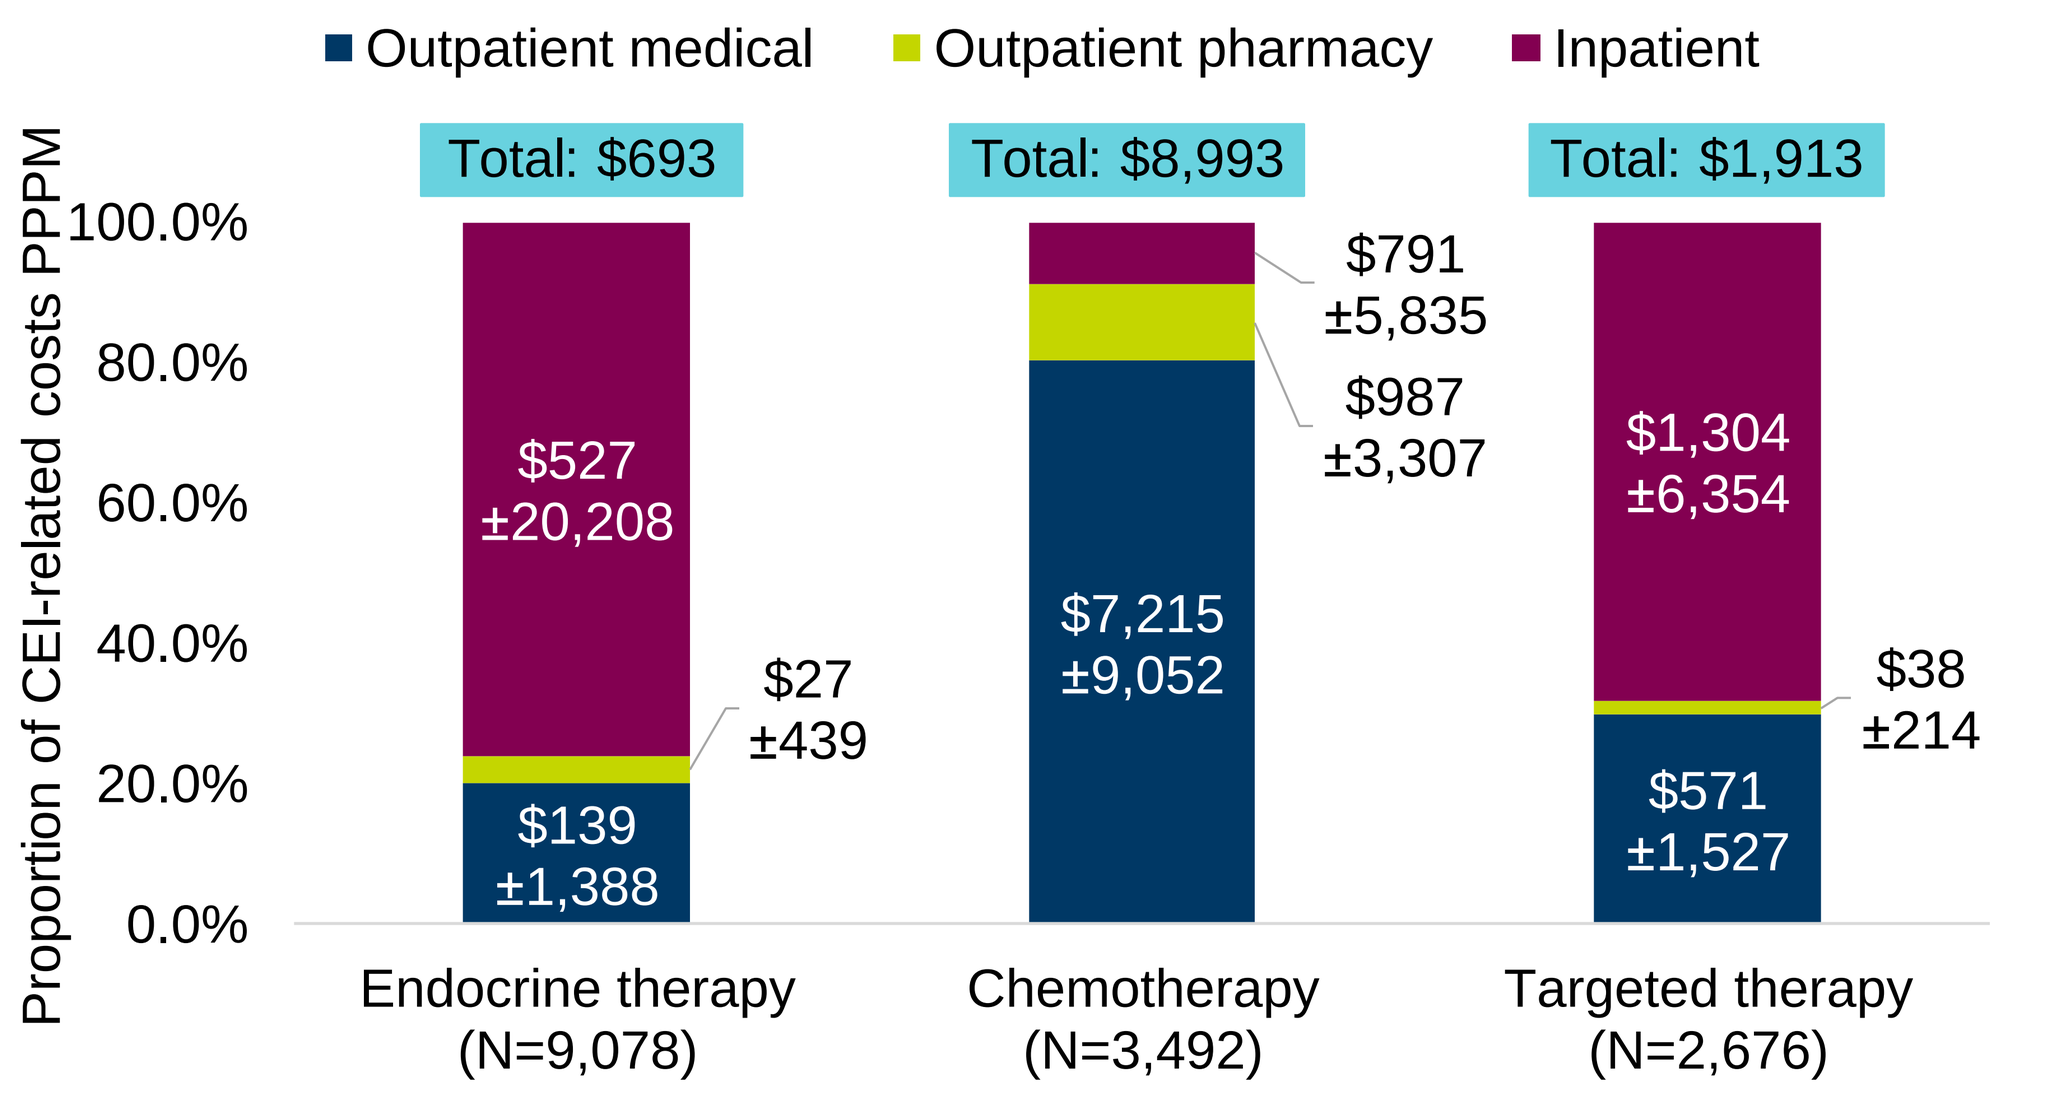
Outpatient medical
Outpatient pharmacy
Inpatient
Total: $693
Total: $8,993
Total: $1,913
Proportion of CEI-related costs PPPM
100.0%
80.0%
60.0%
40.0%
20.0%
0.0%
$527 ±20,208
$27 ±439
$139 ±1,388
$791 ±5,835
$987 ±3,307
$7,215 ±9,052
$1,304 ±6,354
$38 ±214
$571 ±1,527
Endocrine therapy (N=9,078)
Chemotherapy (N=3,492)
Targeted therapy (N=2,676)

Limitations

- There are several limitations inherent to a retrospective study using claims databases, including lack of clinical data (e.g., biomarker testing results) and underreporting of CEIs based on diagnosis codes.
- Patients ≥65 years of age are under-represented in this database of commercially insured individuals. Therefore, these findings may not be generalizable to older patients or those with traditional Medicare coverage.

Conclusions

- All-cause and BC-related healthcare costs for patients with HR+/HER2- mBC were numerically higher during treatment with CDK4/6 inhibitors, which accounted for most targeted therapies in this study, as compared to during endocrine only or chemotherapy regimens. These findings are consistent with existing data.[4]
- However, CEI-related costs were highest during chemotherapy regimens and may be a consequence of greater toxicity relative to endocrine only or targeted therapy.[3]
- Overall, the high economic and clinical burden of standard of care indicates a need for newer treatments that balance costs of care, including drug costs, with adverse events and clinical benefit, particularly for patients currently treated with chemotherapy.

Abbreviations

ADC, Antibody drug conjugate; BC, Breast cancer; CDK4/6, Cyclin-dependent kinase 4/6; CEI, Clinical event of interest; CNS, Central nervous system; ED, Emergency department; GnRH, Gonadotropin-Releasing Hormone; HCRU, Healthcare resource utilization; mTOR, Mammalian target of rapamycin; NCI, National Cancer Institute; PARP, Poly(ADP-ribose) polymerase; PIK3CA, Phosphatidylinositol-4,5-bisphosphate 3-kinase catalytic subunit alpha; PPPM, Per patient per month; Q1, Quartile 1; Q3, Quartile 3; SERM, Selective estrogen receptor modulators; US, United States; USD, US dollar; VEGF, Vascular endothelial growth factor.

Disclosures

Simon Collin, Gráinne Long, and Nikki Toms are employed by AstraZeneca Pharmaceuticals Ltd., which funded this study. Jenny Tse, Nazneen Fatima Shaikh, and Aimee Near are employed by by IQVIA, which received funding from AstraZeneca Pharmaceuticals Ltd. to conduct this research. Adam Brufsky is a consultant for AstraZeneca, Pfizer, Novartis, Lilly, Genentech/Roche, SeaGen, Daiichi Sankyo, Merck, Agendia, Sanofi, Puma, and Myriad and has received research support from Agendia and AstraZeneca. Erika Hamilton and William Gradishar are consultants for AstraZeneca. Sandhya Mehta is employed by Daiichi Sankyo.

References

1. NCCN Clinical Practice Guidelines in Oncology (NCCN Guidelines®) for Breast Cancer V.2.2024. © National Comprehensive Cancer Network, Inc. 2024. All rights reserved. Accessed March 27, 2024.
2. Meisel J, et al. SABCS 2023, Abstract PO5-12-06
3. Elfgen C et al. Cancers (Basel) 2021,13(23):5994.
4. Burne R, et al. J Med Econ 2021, 24(1):806-815.