One-year effectiveness and safety outcomes of edoxaban treatment in Chinese patients with atrial fibrillation: First snapshot of more than 4,800 patients from the ETNA-AF-China study

Xueyuan Guo[1], Juan Du[2], Yuehui Yin[3], Heng Qi[4], Zhisheng Jia[5], Xiaojun Ji[6], Yuqing Zhang[7], Xue Liang[8], Bing Deng[9], Jieyun Liu[10], Yuan Yi[2], Mengqi Liu[2], Cathy Chen[11], Martin Unverdorben[11], Changsheng Ma[1]* On behalf of ETNA-AF-China investigators

[1]Beijing Anzhen Hospital, Capital Medical University, Beijing, China; [2]Daiichi Sankyo (China) Holdings Co., Ltd, Shanghai, China; [3]The Second Affiliated Hospital of Chongqing Medical University, Chongqing, China; [4]The First Hospital of Changsha, Changsha, China; [5]The Fifth People's Hospital of Jinan, Jinan, China; [6]Wenzhou Central Hospital, Wenzhou, China; [7]Nanjing Jiangning Hospital, Nanjing, China; [8]The Fifth Affiliated Hospital of Zhengzhou University, Zhengzhou, China; [9]Longhua Hospital Shanghai University of Traditional Chinese Medicine, Shanghai, China; [10]Kaifeng Central Hospital, Kaifeng, China; [11]Daiichi Sankyo Inc., Basking Ridge, New Jersey, United States of America

BACKGROUND

- Direct oral anticoagulants (DOACs) have become the first-line therapy to prevent ischaemic stroke and systemic embolism (SE) in patients with atrial fibrillation (AF)[1,2,3]
- Edoxaban, a direct factor Xa inhibitor, is globally approved for stroke prevention in AF patients basing on non-inferior efficacy and superior safety over warfarin showed in pivotal randomized clinical trial, ENGAGE AF-TIMI 48[4]
- Real-world data on safety profile of different DOACs are emerging[5]; however, very limited long-term studies report the outcomes of single DOAC in large cohorts of Chinese patients with AF
- ETNA-AF-China (Edoxaban Treatment in routiNe clinical prActice in patients with nonvalvular Atrial Fibrillation) is a multi-centre, prospective, observational study in unselected patients with AF in routine edoxaban treatment in China

OBJECTIVE

- The primary objective of ETNA-AF-China (NCT04747496) was to evaluate safety of real-world edoxaban treatment in patients with AF for up to 2 years, with secondary objective of assessing effectiveness
- Here we firstly present the 1-year outcome data of edoxaban treatment in 5001 Chinese AF patients

METHODS

- The study enrolled patients from 89 centres in four economic regions across Chinese Mainland since 2021
- Patients were followed-up every 3 ± 0.5 months for 1 year, and every 6 ± 1 months until the end of the 2 years
- A total of 4822 patients were included in the full analysis set with dose of either 60 mg OD or 30 mg OD, and at least one follow-up. Patients who discontinued edoxaban were still followed-up for monitoring outcomes
- All key outcomes were adjudicated except for minor bleeding.

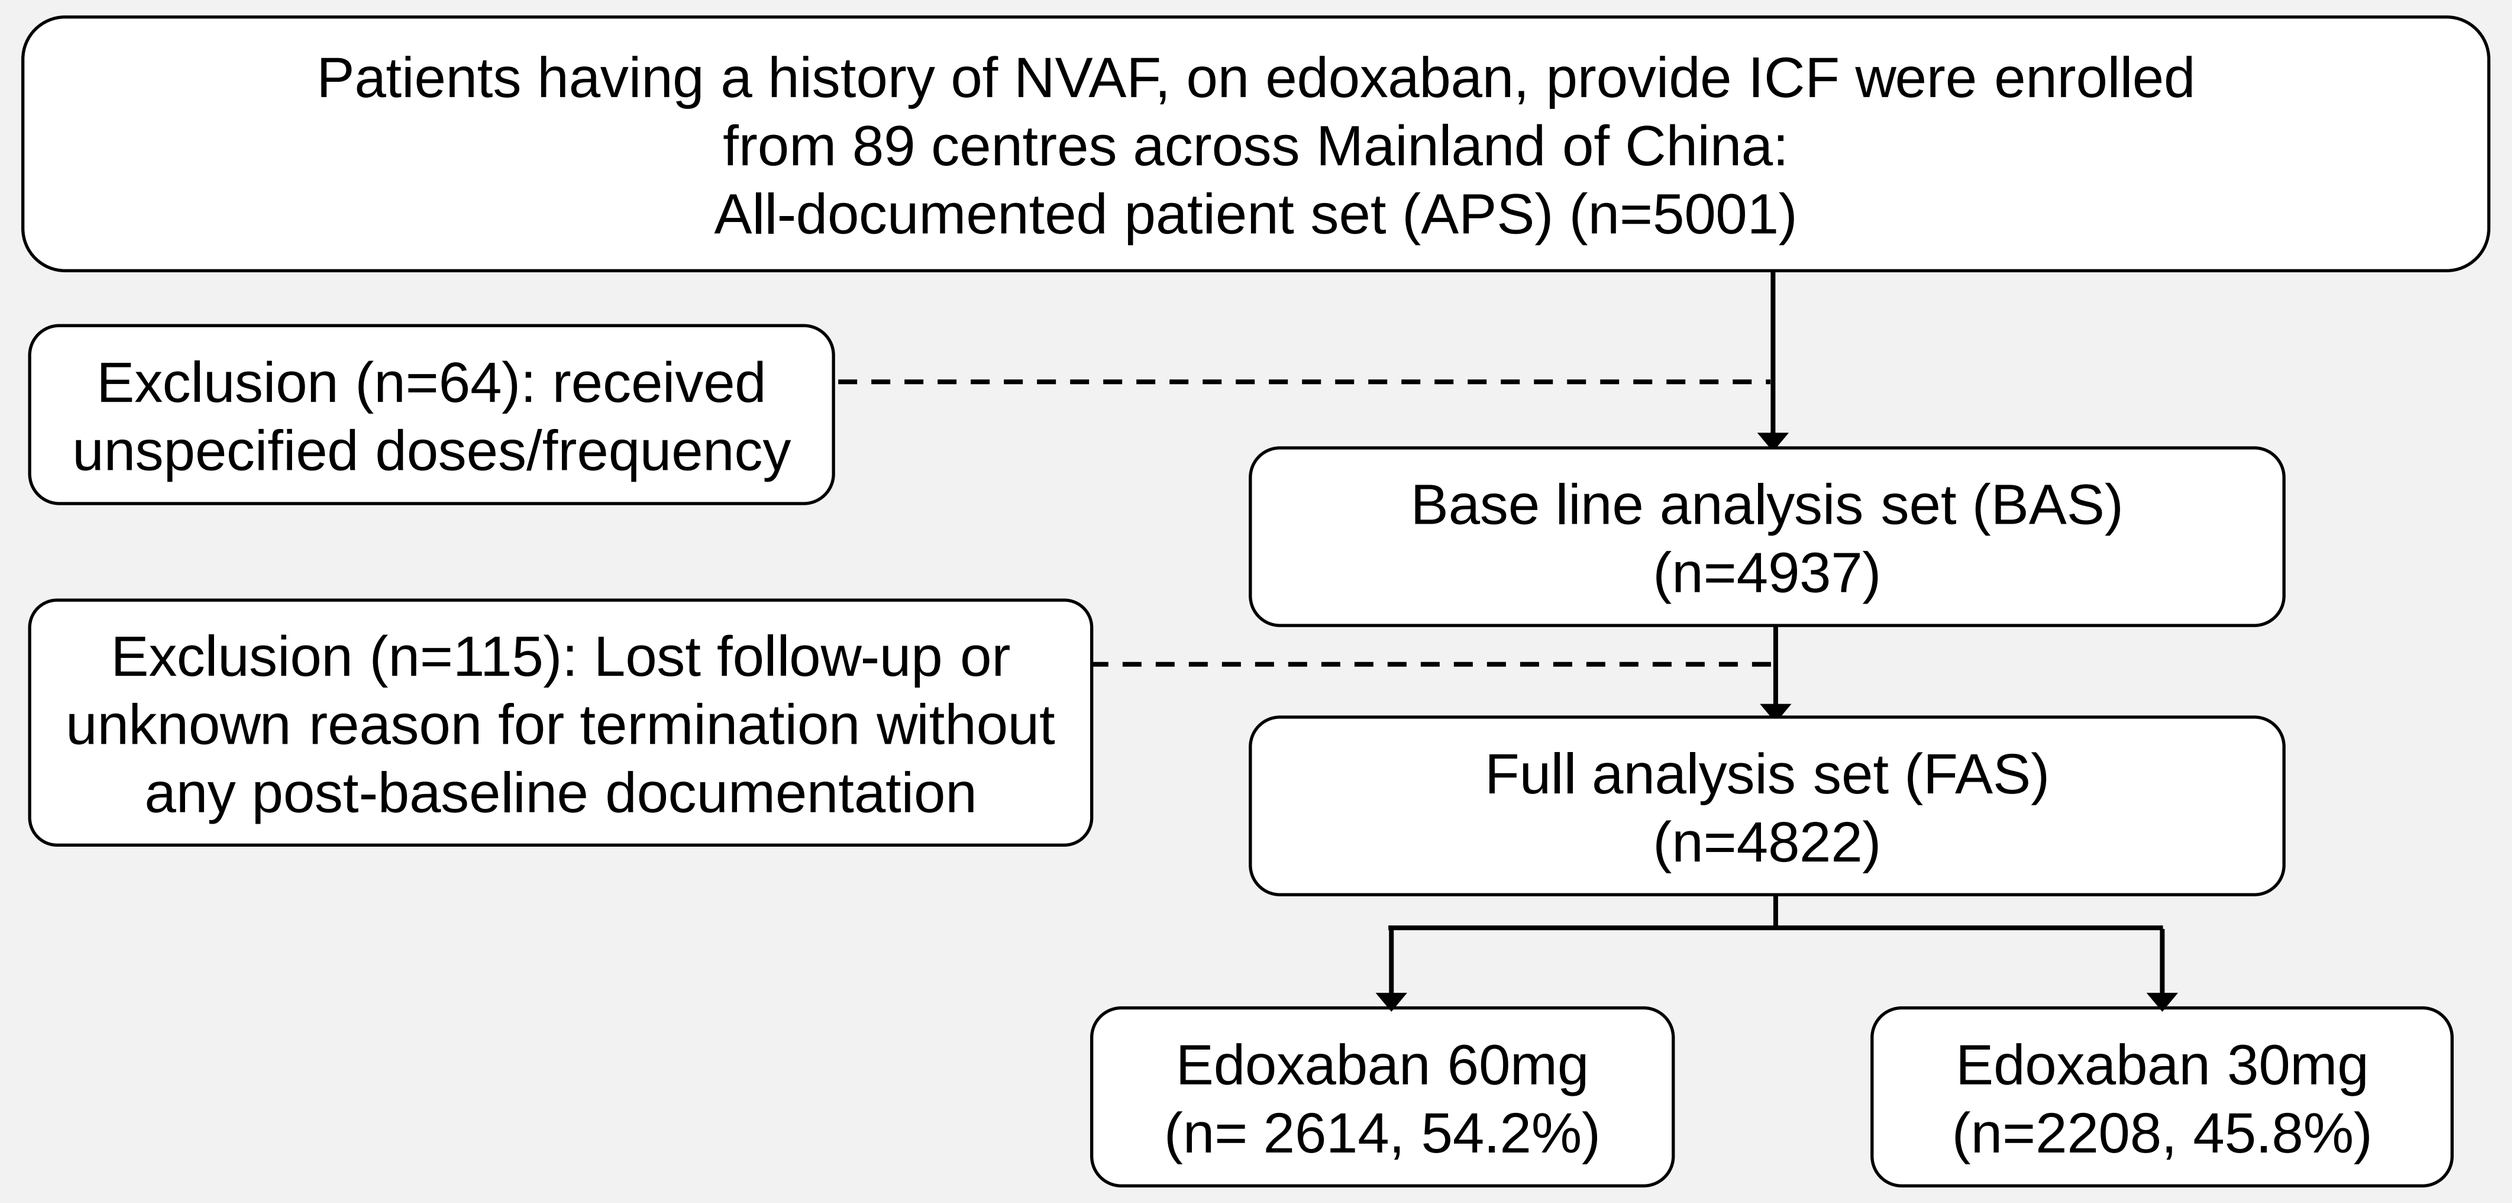
Patients having a history of NVAF, on edoxaban, provide ICF were enrolled
from 89 centres across Mainland of China:
All-documented patient set (APS) (n=5001)
Exclusion (n=64): received unspecified doses/frequency
Base line analysis set (BAS) (n=4937)
Exclusion (n=115): Lost follow-up or unknown reason for termination without any post-baseline documentation
Full analysis set (FAS) (n=4822)
Edoxaban 60mg (n= 2614, 54.2%)
Edoxaban 30mg (n=2208, 45.8%)

Primary endpoints:
- Bleeding events including intracranial hemorrhage (ICH), major bleeding, major gastrointestinal (GI) bleeding
- All cause and CV mortality

Secondary endpoints:
- Stroke (ischaemic and hemorrhagic)
- Systemic embolic events (SEE)
- Transient ischemic attack (TIA)

RESULTS

Baseline demographics (BL) and clinical characteristics by edoxaban dose

- Overall, 4822 patients with AF were treated with edoxaban (60 mg once daily (OD): n=2614; 30 mg OD: n=2208) at BL (**Table 1**)
- The mean (SD) age of the overall patient population was 70.3 (9.5) years, CHA_2DS_2-VASc and HAS-BLED scores were 2.9 (1.4), 1.7 (1.0), with creatinine clearance (CrCl) of 71.2 (27.7) mL/min (**Table 1**)
- Patients receiving 30 mg OD edoxaban at BL were older with higher CHA_2DS_2-VASc score, larger percentage of frail, and higher renal impairment (defined as CrCl ≤80 mL/min) (**Table 1**)

Clinical outcomes

- Annualised rates for all-cause and cardiovascular death in the overall population were very low with 2.22%/year and 0.41%/year, respectively; nearly three times lower in those receiving 60 mg than 30 mg edoxaban (**Figure 1**)
- Annualised event rates for all stroke/SEE (1.03%/year) and ischemic stroke (0.60%/year) were similar between two dose groups (**Figure 1**)
- Overall, annualised rates for major bleeding (1.03%/year), ICH (0.18%/year), major gastrointestinal (GI) bleeding (0.48%/year) events were relatively low, and those on 60 mg dose experienced less above bleeding events (**Figure 1**)
- Cumulative survival analysis demonstrated a slow increase in time-to-first event curve of all-cause death; and from baseline to 1-year follow-up, significant lower events of all-cause death (log-rank p<0.001), major bleeding (p=0.002) in 60 mg vs 30 mg edoxaban groups (**Figure 2**)

Table 1. Baseline demographic and clinical characteristics

Characteristics, n (%) or mean (SD)	Overall (N=4822*)	Edoxaban 60 mg (N=2614)	Edoxaban 30 mg (N=2208)
Age	70.3 (9.5)	67.7 (9.1)	73.4 (9.0)
Male	2752 (57.1)	1789 (68.4)	963 (43.6)
Weight (kg)	68.0 (12.6)	73.3 (11.2)	61.7 (11.3)
CHA_2DS_2-VASc#	2.9 (1.4)	2.6 (1.3)	3.3 (1.4)
Mod. HAS-BLED#	1.7 (1.0)	1.5 (1.0)	2.0 (0.9)
Uncontrolled hypertension episode	737 (15.3)	403 (15.4)	334 (15.1)
Frailty	307 (6.4)	89 (3.4)	218 (9.9)
Serum creatinine (mg/dL)	0.95 (0.3)	0.91 (0.3)	0.99 (0.4)
Creatinine clearance (mL/min)	71.2 (27.7)	81.6 (26.8)	59.5 (23.7)
Renal impairment¶	2674 (55.5)	1122 (42.9)	1552 (70.3)
Renal disease reported by investigator	664 (13.8)	272 (10.4)	392 (17.8)
Recommended doses§	3216 (77.5)	1770 (83.6)	1446 (71.1)

*FAS included patients with baseline data for doses/frequencies of 60 mg OD or 30 mg OD edoxaban, and recorded at least one follow-up record. #Derived variables of CHA_2DS_2-VASc score and HAS-BLED score. HAS-BLED score was calculated without 'labile INR', the renal impairment item was using derived instead of investigator reported. †CrCl calculation was based on CG formula. ¶Renal impairment was derived and renal disease was reported by investigator. §Recommended doses was calculated on percentage of judgeable patients. CHA_2DS_2-VASc: CHF history, hypertension, elderly, diabetes mellitus, stroke (TIA, SEE) history, vascular disease (PAD, MI-STEMI, MI-NSTEMI), female; HAS-BLED: hypertension, abnormal renal function, abnormal liver function, stroke history, bleeding, elderly, drugs, alcohol abuse; CrCl, creatinine clearance; CV, Cardiovascular; GI, gastrointestinal; ICH, intracranial haemorrhage; SEE, systemic embolic event; TIA, transient ischaemic attack; SD, standard deviation.

Figure 1. Annualised event rates (%/year) in the overall population and different dose groups

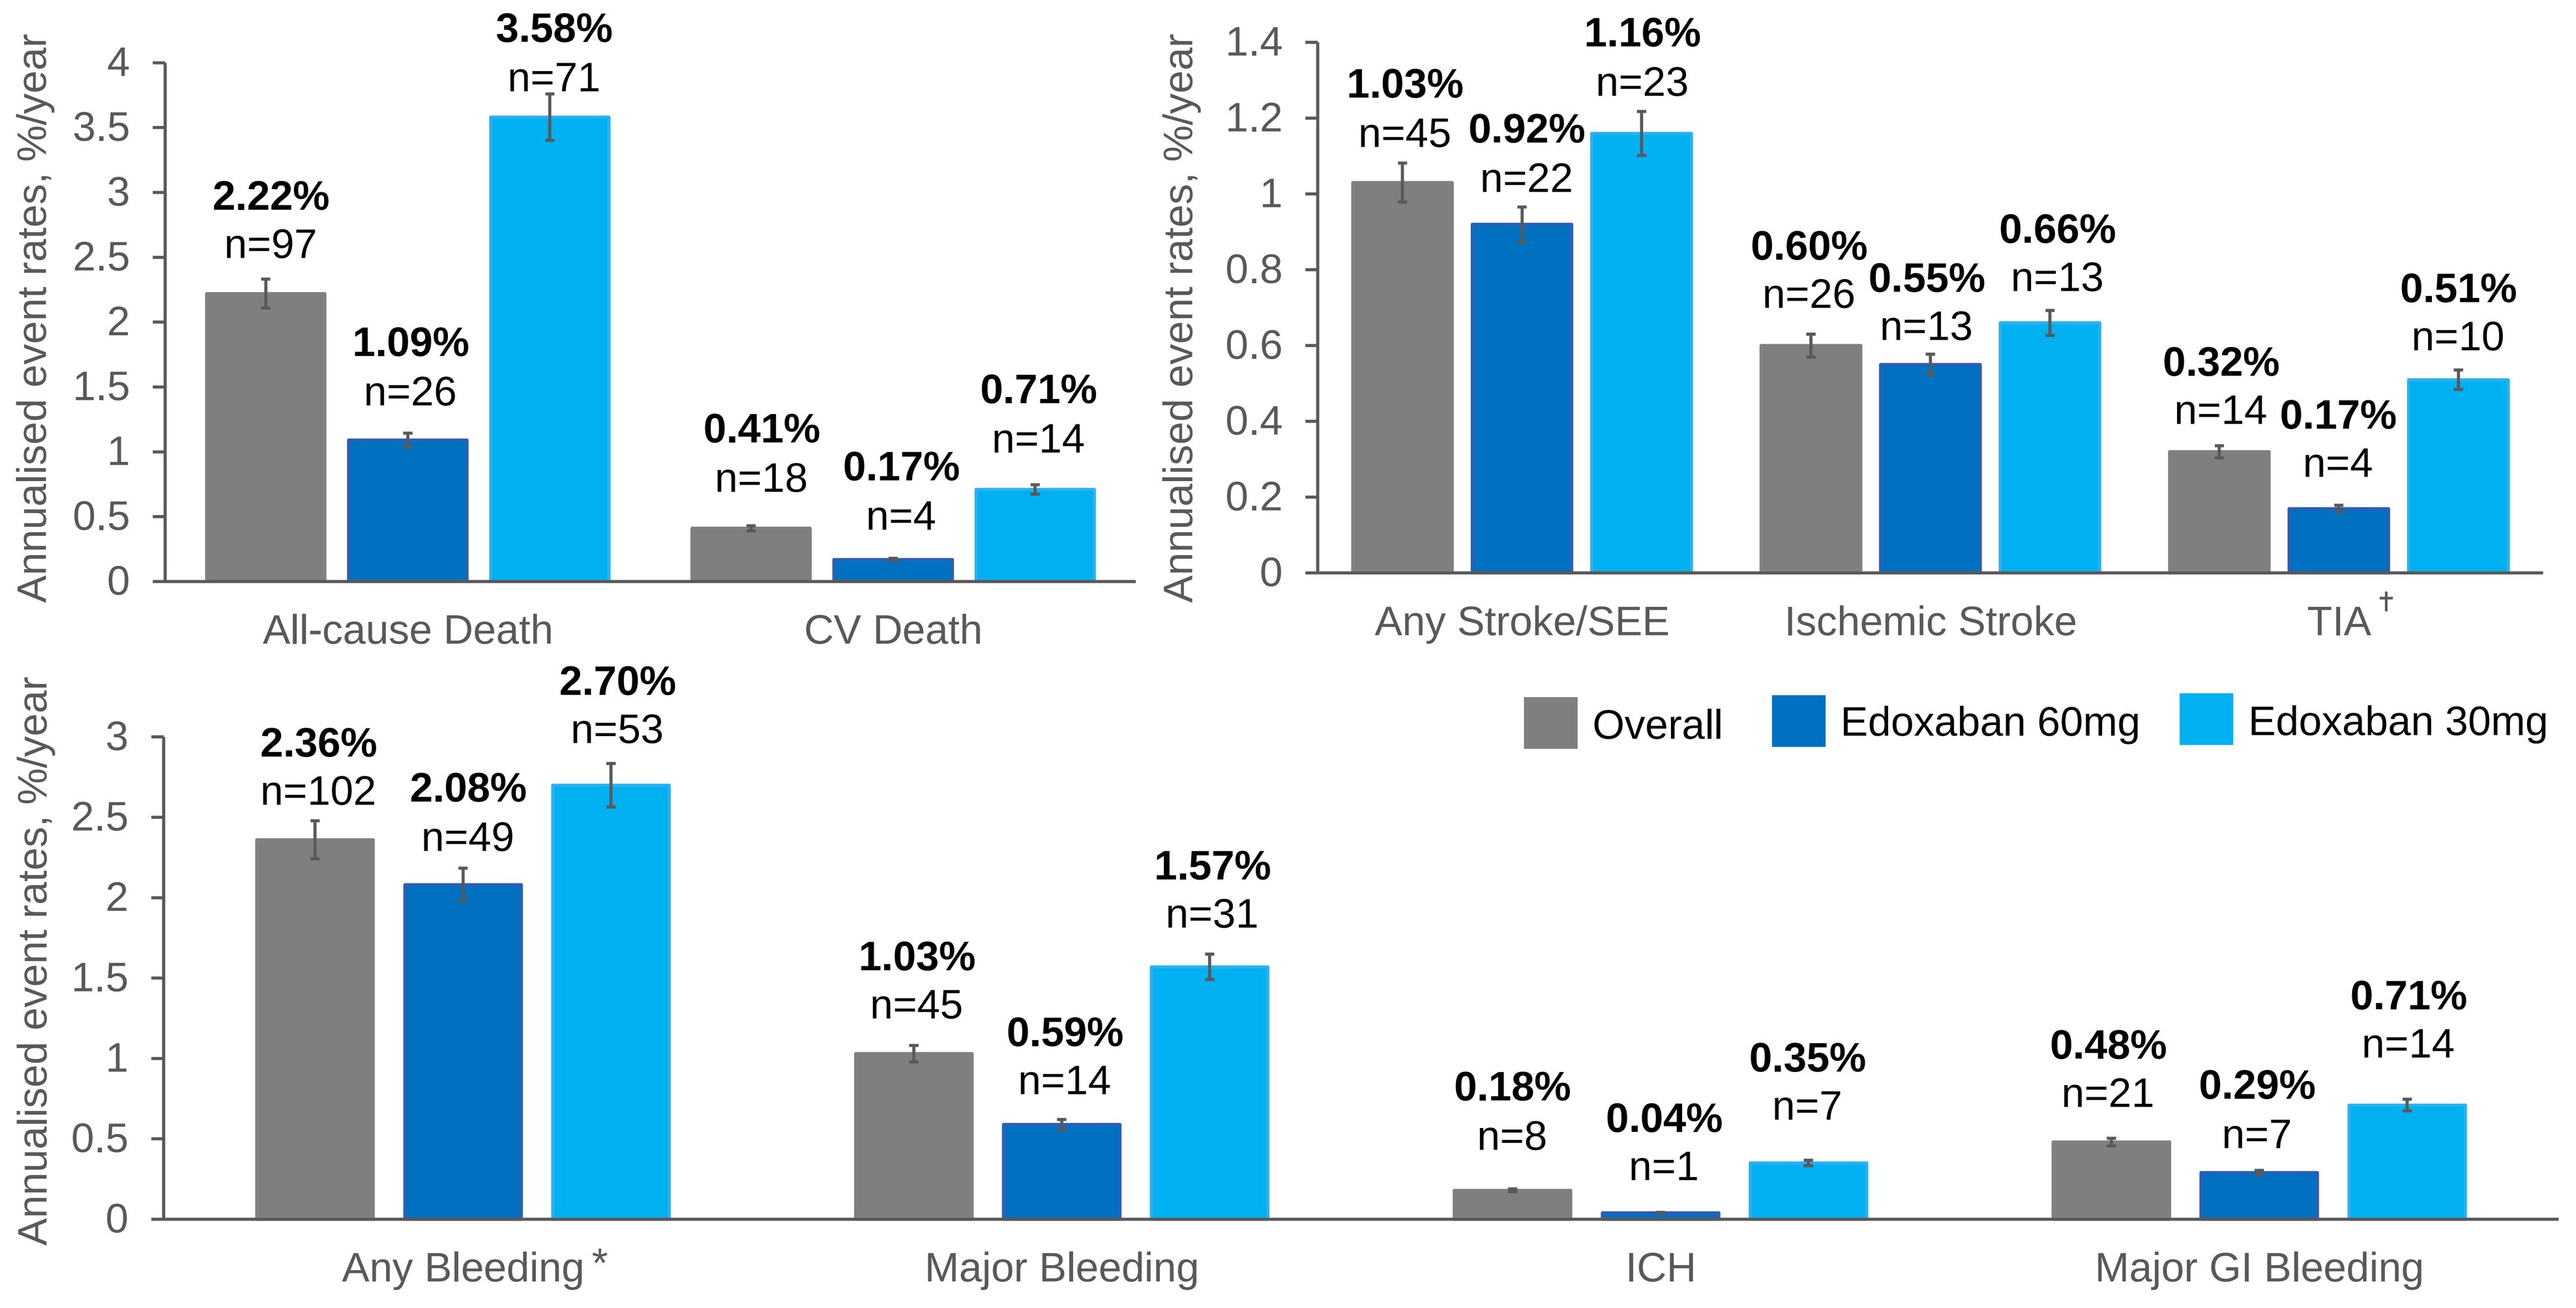
Annualised event rates, %/year
All-cause Death: 2.22% n=97; 1.09% n=26; 3.58% n=71
CV Death: 0.41% n=18; 0.17% n=4; 0.71% n=14
Any Stroke/SEE: 1.03% n=45; 0.92% n=22; 1.16% n=23
Ischemic Stroke: 0.60% n=26; 0.55% n=13; 0.66% n=13
TIA†: 0.32% n=14; 0.17% n=4; 0.51% n=10
Overall
Edoxaban 60mg
Edoxaban 30mg
Any Bleeding*: 2.36% n=102; 2.08% n=49; 2.70% n=53
Major Bleeding: 1.03% n=45; 0.59% n=14; 1.57% n=31
ICH: 0.18% n=8; 0.04% n=1; 0.35% n=7
Major GI Bleeding: 0.48% n=21; 0.29% n=7; 0.71% n=14

*was not adjudicated minor events; †included haemorrhagic stroke or epidural/subdural haematoma.
CV, Cardiovascular; **GI**, gastrointestinal; **ICH**, intracranial haemorrhage; **SEE**, systemic embolic event; **TIA**, transient ischaemic attack.

Figure 2. KM evaluation of 1-year clinical outcomes in the overall population and different dose groups

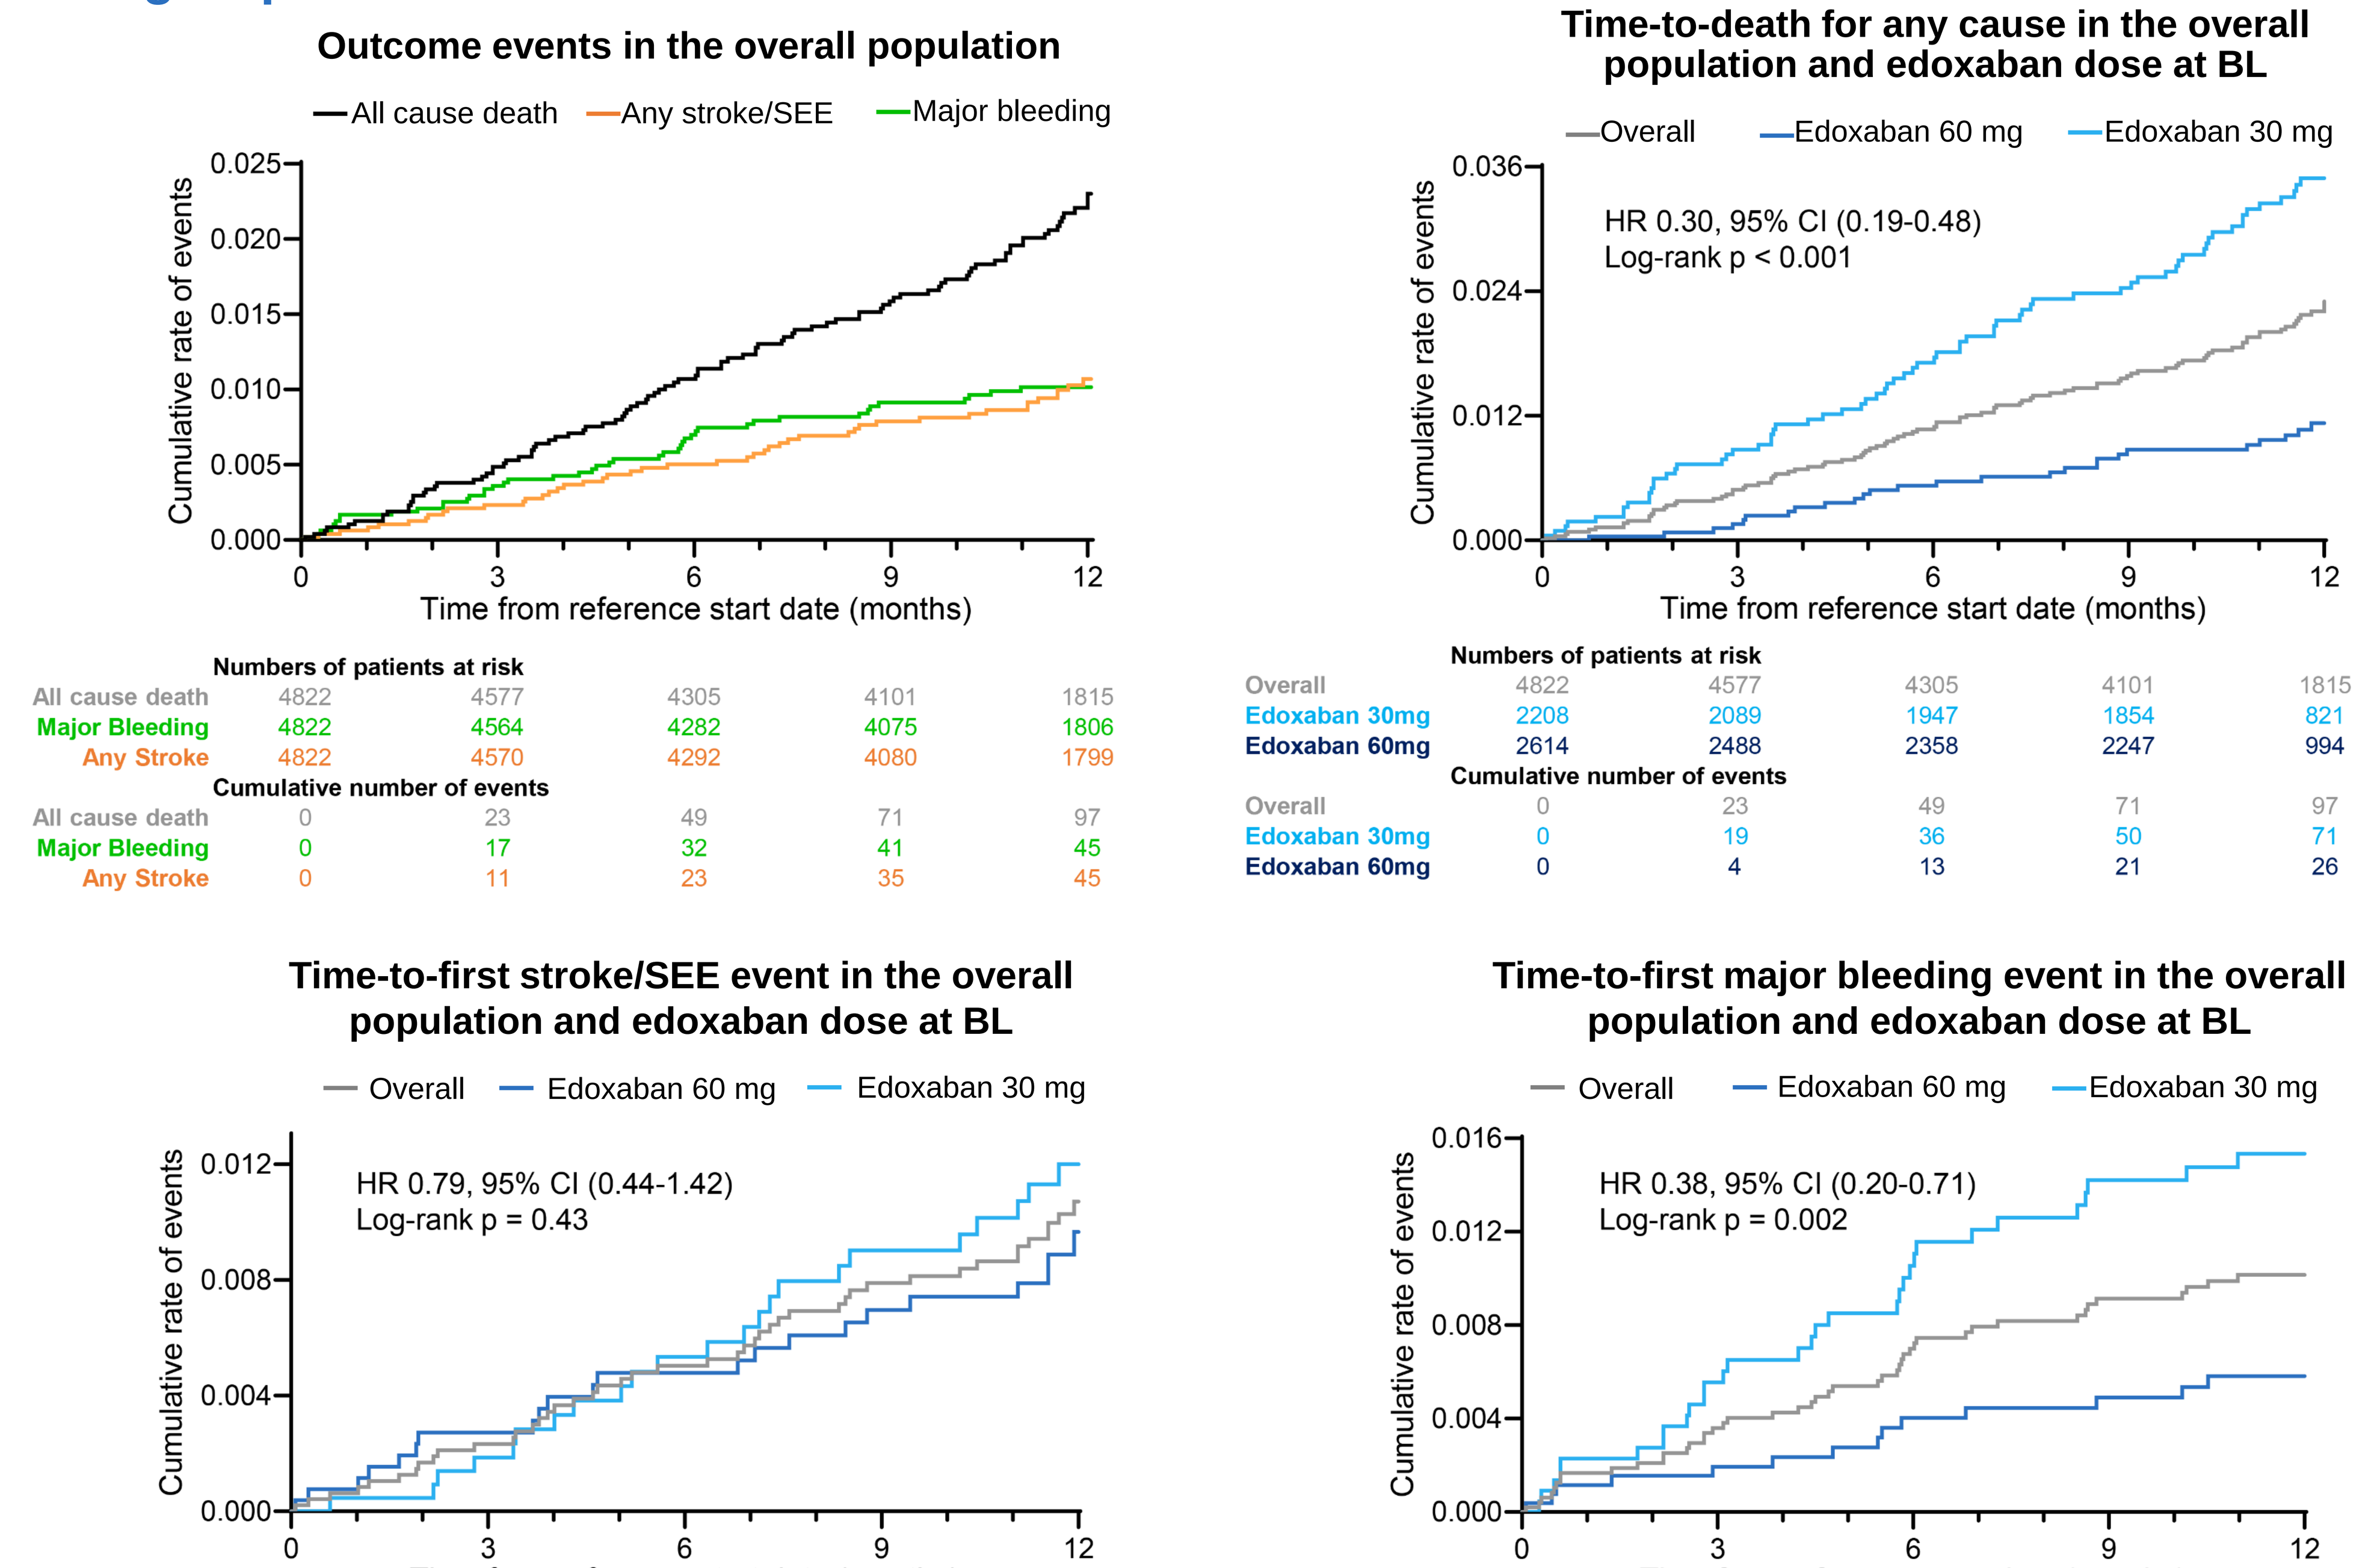
Outcome events in the overall population
All cause death
Any stroke/SEE
Major bleeding
Time-to-death for any cause in the overall population and edoxaban dose at BL
Overall
Edoxaban 60 mg
Edoxaban 30 mg
HR 0.30, 95% CI (0.19-0.48)
Log-rank p < 0.001
Time-to-first stroke/SEE event in the overall population and edoxaban dose at BL
HR 0.79, 95% CI (0.44-1.42)
Log-rank p = 0.43
Time-to-first major bleeding event in the overall population and edoxaban dose at BL
HR 0.38, 95% CI (0.20-0.71)
Log-rank p = 0.002
Cumulative rate of events
Time from reference start date (months)

Outcome events in the overall population

Numbers of patients at risk	0	3	6	9	12
All cause death	4822	4577	4305	4101	1815
Major Bleeding	4822	4564	4282	4075	1806
Any Stroke	4822	4570	4292	4080	1799
Cumulative number of events					
All cause death	0	23	49	71	97
Major Bleeding	0	17	32	41	45
Any Stroke	0	11	23	35	45

Time-to-death for any cause in the overall population and edoxaban dose at BL

Numbers of patients at risk	0	3	6	9	12
Overall	4822	4577	4305	4101	1815
Edoxaban 30mg	2208	2089	1947	1854	821
Edoxaban 60mg	2614	2488	2358	2247	994
Cumulative number of events					
Overall	0	23	49	71	97
Edoxaban 30mg	0	19	36	50	71
Edoxaban 60mg	0	4	13	21	26

Time-to-first stroke/SEE event in the overall population and edoxaban dose at BL

Numbers of patients at risk	0	3	6	9	12
Overall	4822	4570	4292	4080	1799
Edoxaban 30mg	2208	2087	1941	1842	811
Edoxaban 60mg	2614	2483	2351	2238	988
Cumulative number of events					
Overall	0	11	23	35	45
Edoxaban 30mg	0	4	11	18	23
Edoxaban 60mg	0	7	12	17	22

Time-to-first major bleeding event in the overall population and edoxaban dose at BL

Numbers of patients at risk	0	3	6	9	12
Overall	4822	4564	4282	4075	1806
Edoxaban 30mg	2208	2079	1932	1838	816
Edoxaban 60mg	2614	2485	2350	2237	990
Cumulative number of events					
Overall	0	17	32	41	45
Edoxaban 30mg	0	12	22	29	31
Edoxaban 60mg	0	5	10	12	14

CONCLUSIONS

- Low incidences of major bleeding, notably ICH, major GI bleeding, any stroke/SEE and CV death were observed in unselected 4822 patients with AF during 1-year of edoxaban treatment
- In routine care of Chinese AF patients, lower death and major bleeding event rates observed with edoxaban 60 mg were potentially due to the younger population, and lower CHA_2DS_2-VASc, HAS-BLED scores, less frailty
- These findings confirm the effectiveness and safety of edoxaban use in China. More data on the association with dose recommendation and outcomes, predictors of death, bleeding events in this large Chinese AF populations will be investigated further

REFERENCES

1. Steffel J, et al. *Europace* 2021;23(10):1612-1676; **2.** Joglar JA, et al. *Circulation* 2024 Jan 2;149(1):e1-e156; **3.** Chiang CE, et al. *JACC Asia* 2022;2(4):395-411; **4.** Giugliano RP, et al. *N Engl J Med* 2013;369:2093–104; **5.** Chang SS, et al. *Stroke* 2016 Jul;47(7):1803-10;

ACKNOWLEDGEMENTS

The ePoster presentation was sponsored by Daiichi Sankyo (China), Shanghai, China. Authors would like to thank all participated patients and investigators, Ting-ting Sun, Min Tao for patients enrolling and management support, Tracy Zeng, Hui Yang, Yuechan Zhang, Wenyi Ma for support on medication safety monitoring. Poster preparation editorial support was provided by Roopa, Shubham, Ashwini, Ravi, Swathirajan from Indegene Pvt. Ltd., Bengaluru, India and was funded by Daiichi Sankyo (China), Shanghai, China.

ETNA-AF China