Management of edoxaban therapy in patients undergoing emergency/urgent procedures: A subanalysis of the global prospective, observational, multinational EMIT-AF/VTE programme

Paolo Colonna[1], José Souza[2], Manish Saxena[3], Thomas Vanassche[4], Amparo Santamaria[5], James Jin[6], Sabine Köhler[7], Amanda Borrow[6], Martin Unverdorben[6], Christian von Heymann[8]

[1]Department of Cardiology, Polyclinic of Bari – Hospital, Bari, Italy; [2]Daiichi Sankyo Europe GmbH, Munich, Germany; [3]NIHR Barts Cardiovascular Biomedical Research Centre, London, UK; [4]Department of Cardiovascular Sciences, University Hospitals (UZ) Leuven, Leuven, Belgium; [5]Hematology Department, University Hospital Vinalopó, Alicante, Spain; [6]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [7]Serenis GmbH, Munich, Germany; [8]Department of Anaesthesia, Intensive Care Medicine, Emergency Medicine, and Pain Therapy, Vivantes Klinikum im Friedrichshain, Berlin, Germany

PURPOSE

- Interruption of oral anticoagulation is often impossible before emergency/urgent (unplanned) procedures due to the time constraints. This may lead to a higher incidence of adverse outcomes in these patients compared with those who undergo planned procedures[1]
- The Global EMIT-AF/VTE programme (NCT02950168,NCT02951039) found low bleeding and thromboembolic event rates for patients receiving edoxaban who underwent diagnostic or therapeutic procedures[2]
- However, edoxaban management and clinical outcomes may differ among the subgroup of patients with emergency/urgent procedures[3]
- This subanalysis aimed to describe periprocedural edoxaban management and clinical outcomes for patients in the Global EMIT-AF/VTE programme who underwent emergency/urgent procedures

METHODS

- The Global EMIT-AF/VTE programme, conducted in Europe and Asia, prospectively collected data from patients with atrial fibrillation (AF) or venous thromboembolism (VTE) receiving edoxaban and undergoing diagnostic and therapeutic procedures in routine clinical practice[4]
- The current analysis included physician-reported unplanned (emergency/urgent) procedures as defined
 - Emergency surgery: surgery that cannot be delayed, for which there is no alternative therapy or surgeon, and for which a delay could result in death or permanent impairment of health[5]
 - Urgent surgery: surgery required within 48 hours[6]
- Only a patient's first emergency/urgent procedure was included, except for 3 patients who had 2 procedures on the same day; in these cases, the 2 procedures were combined
- Patient baseline characteristics and edoxaban interruption data (categorised as shown in **Figure 1**) were collected and stratified by the European Heart Rhythm Association (EHRA) procedural bleeding risk criteria[7]
- Clinical events were assessed relative to the date of their associated procedure to determine if they were a cause or consequence of the procedure
- Clinical outcomes assessed were major bleeding (MB; as defined by the International Society of Thrombosis and Haemostasis[8]), clinically relevant nonmajor bleeding (CRNMB), minor bleeding, all bleeding events, acute coronary syndrome, acute thromboembolic events (ischaemic stroke, transient ischaemic attack, and systemic embolic events), all-cause mortality, and cardiovascular mortality

RESULTS

Baseline characteristics and procedure types

- Among 144 patients undergoing emergency/urgent procedures, the mean ± standard deviation age at baseline was 74.5 ± 8.7 years, the CHA_2DS_2-VASc score was 3.8 ± 1.6, medical histories of hypertension and dyslipidaemia were reported in 83.3% and 41.0% of patients, and approximately one-quarter of patients had diabetes mellitus (28.5%) or creatinine clearance ≤50 mL/min (25.7%; **Table 1**)
- Most baseline characteristics were similar across procedure bleeding risk groups, except for medical histories of hypertension, dyslipidaemia, valvular heart disease, congestive heart failure, creatinine clearance ≤50 mL/min, and an edoxaban dose of 60 mg/day, which were more frequent among patients who underwent high-risk vs minor- or low-risk procedures
- Vascular access and transcatheter diagnostics and interventions (34.0%) and gastroenterological (27.8%) procedure types were most common (**Figure 2**)
 - The most frequent procedure subtypes were coronary angiography (16.7%; radial access, 10.4%; femoral access, 6.3%), endoscopy (13.9%), insertion of a pacemaker/defibrillator (6.3%), and percutaneous coronary intervention (6.3%)

Periprocedural edoxaban interruption

- The percentages of minor-, low-, and high-risk procedures with pre- and postprocedural edoxaban interruption were 51.1%, 51.2%, and 63.0%, respectively (**Table 2**)
- Preprocedure (days −5 to 0) edoxaban interruption was numerically more common among low-risk (39.1%) vs high- (34.5%) or minor-risk (32.0%) procedures
- Postprocedure edoxaban interruption was numerically more common among high-risk (50.0%) vs low- (31.1%) or minor-risk (38.0%) procedures
- Additionally, 20.3% of procedures had edoxaban interruption that occurred within the 5 days before the procedure (days −5 to −1)
- The median duration of pre- and postprocedural, preprocedural, and postprocedural edoxaban interruption was greater for high- and minor-risk vs low-risk procedures
- By 20 days postprocedure, edoxaban therapy was resumed after approximately 97% and 88% of low- and minor-risk procedures vs 81% of high-risk procedures (**Figure 3**)

Clinical events

- Among 30 recorded clinical events, 18 (60.0%) preceded a procedure that was performed for diagnosis and/or treatment of the event, and thus were not a procedural complication (**Table 3**)
- The only clinical events that occurred after and were caused by a procedure were five bleeding events (3 of which were MB or CRNMB)

CONCLUSIONS

- Bleeding events resulting from an emergency/urgent procedure occurred in <4% of patients and no thromboembolic events occurred, indicating that postprocedure edoxaban therapy can be effectively tailored to optimise clinical outcomes
- Edoxaban resumption was protracted after high-risk vs minor- or low-risk procedures

In this subanalysis of patients in the Global EMIT-AF/VTE programme, the number of clinical events resulting from an emergency/urgent procedure was low, indicating that postprocedural edoxaban therapy was effectively tailored to optimise clinical outcomes.

Edoxaban therapy was resumed later after high bleeding risk procedures compared with those of minor or low risk.

FIGURES AND TABLES

Figure 1. Periprocedural edoxaban interruption periods

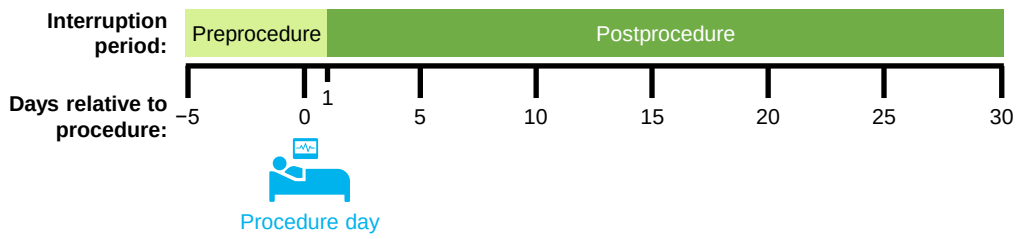
Interruption period:
Preprocedure
Postprocedure
Days relative to procedure:
−5
0
1
5
10
15
20
25
30
Procedure day

Figure 2. Emergency/urgent procedures by type

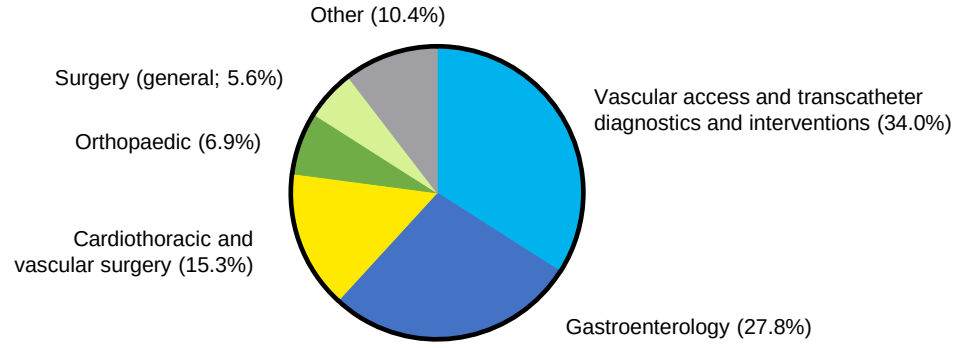
Other (10.4%)
Surgery (general; 5.6%)
Orthopaedic (6.9%)
Cardiothoracic and vascular surgery (15.3%)
Vascular access and transcatheter diagnostics and interventions (34.0%)
Gastroenterology (27.8%)

A total of 144 procedures were assessed.
"Other" includes urology (4.2%); ear, nose, and throat (2.1%); ophthalmology (0.7%); and miscellaneous (3.5%) procedures.

Figure 3. Periprocedural edoxaban use over time

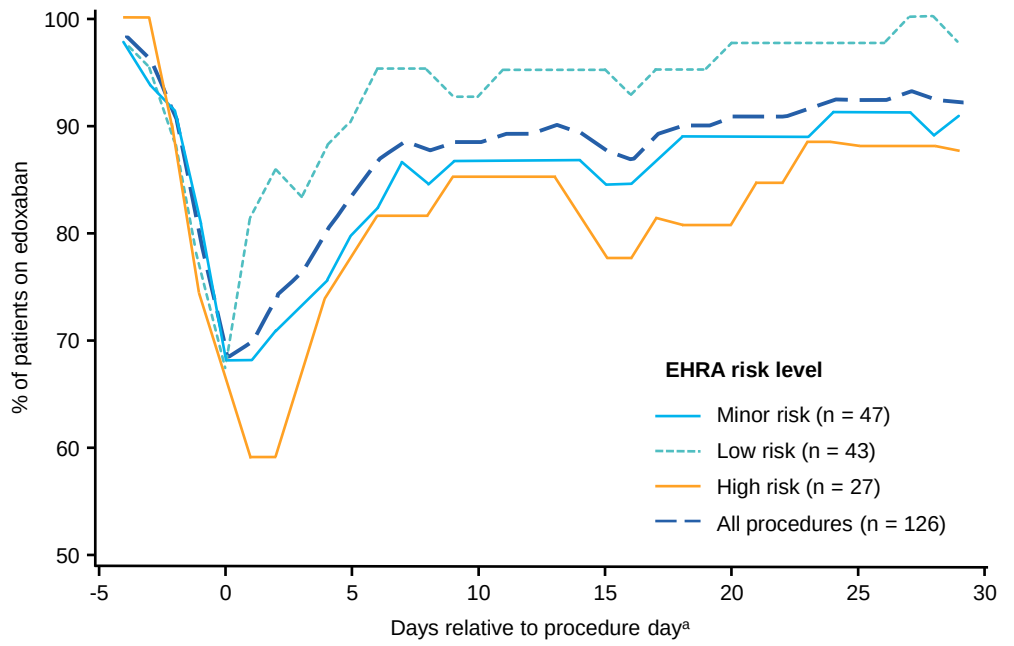
100
90
80
70
60
50
% of patients on edoxaban
-5
0
5
10
15
20
25
30
Days relative to procedure day[a]
EHRA risk level
Minor risk (n = 47)
Low risk (n = 43)
High risk (n = 27)
All procedures (n = 126)

Procedures were excluded if they had no edoxaban data, if edoxaban therapy began less than 5 days before the procedure, or if edoxaban therapy was permanently stopped after the procedure day.
[a]Procedure day is day 0.
EHRA, European Heart Rhythm Association.

Table 1. Overall patient demographics and baseline clinical characteristics by EHRA risk level

Parameter	Minor risk (n = 53)	Low risk (n = 48)	High risk (n = 30)	Unknown risk (n = 13)	All patients (N = 144)
Age, years, mean ± SD	74.0 ± 9.4	74.5 ± 8.7	75.3 ± 6.9	74.7 ± 10.5	74.5 ± 8.7
<65	8 (15.1)	6 (12.5)	1 (3.3)	3 (23.1)	18 (12.5)
65 to <75	20 (37.7)	17 (35.4)	15 (50.0)	3 (23.1)	55 (38.2)
≥75	25 (47.2)	25 (52.1)	14 (46.7)	7 (53.8)	71 (49.3)
Sex, female	20 (37.7)	26 (54.2)	12 (40.0)	5 (38.5)	63 (43.8)
Weight, kg, mean ± SD	66.9 ± 14.7	73.5 ± 14.3	75.7 ± 17.9	61.7 ± 13.9	70.6 ± 15.7
BMI, kg/m², mean ± SD	24.8 ± 4.3	27.1 ± 4.2	26.7 ± 5.1	24.4 ± 3.5	26.0 ± 4.5
Atrial fibrillation	52 (98.1)	44 (91.7)	28 (93.3)	12 (92.3)	136 (94.4)
Venous thromboembolism	1 (1.9)	3 (6.3)	3 (10.0)	1 (7.7)	8 (5.6)
Hypertension	43 (81.1)	38 (79.2)	28 (93.3)	11 (84.6)	120 (83.3)
Dyslipidaemia	19 (35.8)	21 (43.8)	17 (56.7)	2 (15.4)	59 (41.0)
Diabetes mellitus	16 (30.2)	13 (27.1)	8 (26.7)	4 (30.8)	41 (28.5)
Coronary heart disease	12 (22.6)	20 (41.7)	7 (23.3)	0 (0)	39 (27.1)
Valvular heart disease	7 (13.2)	5 (10.4)	6 (20.0)	1 (7.7)	19 (13.2)
Renal disease	13 (24.5)	10 (20.8)	9 (30.0)	2 (15.4)	34 (23.6)
Congestive heart failure	11 (20.8)	10 (20.8)	8 (26.7)	4 (30.8)	33 (22.9)
CrCL, mL/min, mean ± SD	61.6 ± 24.9	69.4 ± 27.9	67.4 ± 25.8	55.6 ± 20.8	65.2 ± 26.0
CrCL ≤50, mL/min	14 (26.4)	10 (20.8)	10 (33.3)	3 (23.1)	37 (25.7)
Heparin (including LMWH)	2 (3.8)	8 (16.7)	3 (10.0)	0 (0)	13 (9.0)
Antiplatelet agents	13 (24.5)	15 (31.3)	4 (13.3)	3 (23.1)	35 (24.3)
HAS-BLED score, mean ± SD	2.1 ± 1.2	2.4 ± 1.4	2.6 ± 0.9	2.2 ± 1.1	2.3 ± 1.2
CHA_2DS_2-VASc score, mean ± SD	3.5 ± 1.6	3.9 ± 1.6	4.1 ± 1.3	4.0 ± 1.9	3.8 ± 1.6
Edoxaban dose					
30 mg/day	24 (45.3)	17 (35.4)	9 (30.0)	8 (61.5)	58 (40.3)
60 mg/day	28 (52.8)	31 (64.6)	21 (70.0)	5 (38.5)	85 (59.0)

Data are shown as n (%) unless otherwise noted.
BMI, body mass index; CHA_2DS_2-VASc, Congestive heart failure, Hypertension, Age ≥75 (doubled), Diabetes, Stroke (doubled), Vascular disease, Age 65 to 74, and Sex category (female); CrCL, creatinine clearance; EHRA, European Heart Rhythm Association; HAS-BLED, Hypertension, Abnormal liver/renal function, Stroke, Bleeding history or predisposition, Labile international normalised ratio, Elderly, Drug/alcohol usage; LMWH, low-molecular-weight heparin; SD, standard deviation.

Table 2. Time and median duration of edoxaban interruption by EHRA risk level relative to emergency/urgent procedures

Interruption period	Minor risk	Low risk	High risk	Unknown risk	All procedures
Pre- and postprocedure, n/N (%)	24/47 (51.1)	22/43 (51.2)	17/27 (63.0)	5/9 (55.6)	68/126[a] (54.0)
Median interruption, days	8.0	3.0	6.0	3.0	5.0
Preprocedure only, n/N (%)	16/50 (32.0)	18/46 (39.1)	10/29 (34.5)	3/13 (23.1)	47/138[b] (34.1)
Median interruption, days	2.0	1.0	2.0	1.0	2.0
Postprocedure only, n/N (%)	19/50 (38.0)	14/45 (31.1)	14/28 (50.0)	5/9 (55.6)	52/132[c] (39.4)
Median interruption, days	6.0	3.0	5.5	2.0	5.0

Procedures were excluded if they had no edoxaban data, if the duration of interruption could not be calculated, if edoxaban therapy began less than 5 days before the procedure, or if edoxaban therapy was permanently stopped after the procedure day.
[a]18 procedures were excluded from either pre- or postprocedural edoxaban interruption analyses.
[b]6 procedures were excluded as the first day of edoxaban was less than 5 days before the procedure.
[c]12 procedures were excluded as edoxaban was permanently stopped after the procedure.
EHRA, European Heart Rhythm Association; N, number of procedures included in the analysis; n, number of procedures with edoxaban interruption.

Table 3. Clinical events stratified by their relation to the emergency/urgent procedure date

Clinical events		Relation to procedure[a]		
	Total events[b]	Preceding events[c]	Events resulting from a procedure	Unrelated events[d]
All events	30 (20.8)	18 (60.0)	5 (16.7)	7 (23.3)
All bleeding[e]	20[f] (13.9)	12 (60.0)	5 (25.0)	3 (15.0)
MB or CRNMB	11 (7.6)	7 (63.6)	3 (27.3)	1 (9.1)
ACS	4 (2.8)	4 (100)	0	0
Stroke	2 (1.4)	2 (100)	0	0
TIA	0	0	0	0
SEE	0	0	0	0
All-cause mortality	4 (2.8)	0	0	4 (100)
CV mortality	1 (0.7)	0	0	1 (100)

[a]Clinical events in relation to procedures are presented as n (% of the clinical event type).
[b]Total events are presented as n (% out of 144 total procedures).
[c]Events occurring prior to a procedure that was performed for diagnosis and/or treatment of the event.
[d]Unrelated events had no connection to the procedure.
[e]Includes MB, CRNMB, and minor bleeding.
[f]One minor bleeding event was excluded as it occurred on the same day as the procedure, and determining its relation to the procedure would require additional information.
ACS, acute coronary syndrome; CRNMB, clinically relevant nonmajor bleeding; CV, cardiovascular; MB, major bleeding; SEE, systemic embolic event; TIA, transient ischaemic attack.

Presented at: European Heart Rhythm Association Congress 2024; 7−9 April 2024; Berlin, Germany

REFERENCES

1. Douketis JD, et al. *Throm Res.* 2016;139:77-81. 2. Colonna P, et al. *Clin Cardiol.* 2020;43:769-80. 3. Siegal DM, et al. *J Throm Thrombolysis.* 2023;55:197-202. 4. Colonna P, et al. *Clin Cardiol.* 2018;41:1123-29. 5. The Free Dictionary. "Emergency surgery." McGraw-Hill Dictionary of Modern Medicine. The McGraw-Hill Companies, Inc.; 2002. Accessed February 26, 2024. https://medical-dictionary.thefreedictionary.com/emergency+surgery 6. The Free Dictionary. "Urgent surgery." McGraw-Hill Dictionary of Modern Medicine. The McGraw-Hill Companies, Inc.; 2002. Accessed February 26, 2024. https://medical-dictionary.thefreedictionary.com/urgent+surgery 7. Steffel J, et al. *Europace.* 2021;00:1-65. 8. Schulman S, et al. *J Thromb Haemost.* 2010;8:202-4.

ACKNOWLEDGEMENTS

Medical writing and editorial support were provided by Molly Yeager, PhD, of AlphaBioCom, a Red Nucleus company, and funded by Daiichi Sankyo, Inc.

DECLARATION OF INTEREST

This study was sponsored by Daiichi Sankyo.
PC reports grants and personal fees from Daiichi Sankyo; personal fees from Bayer, Boehringer Ingelheim, Daiichi Sankyo, and Pfizer/Bristol Myers Squibb; and nonfinancial support from the European Society of Cardiology and the Italian Cardiology Association. **JS** is an employee of Daiichi Sankyo. **MS** reports grants and personal fees from Daiichi Sankyo. **TV** reports grants and personal fees from Daiichi Sankyo, and personal fees from Bayer, Boehringer Ingelheim, Daiichi Sankyo, and LEO Pharma. **AS** reports nothing to disclose. **JJ** is an employee of Daiichi Sankyo. **SK** reports consulting fees from Daiichi Sankyo. **AB** is an employee of Daiichi Sankyo. **MU** is an employee of Daiichi Sankyo. **CvH** reports grants and personal fees from Daiichi Sankyo; personal fees from Artcline GmbH, CSL Behring, Mitsubishi Pharma, Novo Nordisk, Shionogi Pharma, and Sobi Pharma; receipt of a mandate from the German Society of Anaesthesiology and Intensive Care Medicine to write the German Guideline on Preoperative Anaemia; participation in the writing group of the Patient Blood Management Guideline in cardiac surgery on behalf of the European Society of Cardiothoracic Anaesthesiologists in conjunction with the European Society of Cardiothoracic Surgery; and receipt of a mandate to take part in the writing group of the guidelines on the Diagnostics and Treatment of Peripartum Haemorrhage of the Deutsche Gesellschaft für Gynäkologie und Geburtshilfe.

EMIT-AF/VTE