Major bleeding after TAVI in the ENVISAGE-TAVI AF trial: A risk-assessment score

Christian Hengstenberg[1], Martin Unverdorben[2], Johny Nicolas[3], Johanna van Zyl[2], Raúl Moreno[4], Cathy Chen[2], Roxana Mehran[3], Marco Valgimigli[5], Nicolas M Van Mieghem[6], George D Dangas[4,7]

[1]Division of Cardiology, Department of Internal Medicine II, Vienna General Hospital, Medical University, Vienna, Austria; [2]Daiichi Sankyo, Inc., Basking Ridge, New Jersey, United States; [3]Zena and Michael A. Wiener Cardiovascular Institute, Mount Sinai Hospital, New York, New York, United States; [4]Department of Cardiology, University Hospital La Paz, Madrid, Spain; [5]Division of Cardiology, Cardiocentro Ticino Institute, Ente Ospedaliero Cantonale, Università della Svizzera Italiana (USI), Lugano, Switzerland and University of Bern, Bern, Switzerland; [6]Department of Cardiology, Erasmus University Medical Centre, Thoraxcenter, Rotterdam, the Netherlands; [7]School of Medicine, National and Kapodistrian University of Athens, Athens, Greece

BACKGROUND

- In the prospective, randomised ENVISAGE-TAVI AF trial (NCT02943785), there was a higher annualised rate of major bleeding with edoxaban vs VKA in patients with incident or prevalent atrial fibrillation (AF) after successful transcatheter aortic valve implantation (TAVI)[1,2]
- There is no risk assessment tool to predict major bleeding after TAVI in patients on oral anticoagulants for AF
- Estimating the probability of post-TAVI major bleeding in these patients is important to inform decision making regarding anticoagulation strategy

OBJECTIVE

- To create a risk-assessment score to predict the likelihood of major bleeding in patients with incident or prevalent AF after successful TAVI by identifying predictors of major bleeding

METHODS

- This on-treatment analysis of ENVISAGE-TAVI AF included patients who received ≥1 dose of the study drug
- Baseline patient characteristics were summarised for patients with vs without a major bleeding event
- A Cox multivariable regression analysis using a stepwise approach was performed to identify predictors of major bleeding
- A simple point-based additive score was constructed using the variable β estimates from the selected model, with 1 point assigned to the variable with the smallest β estimate and remaining variables scaled relative to the smallest estimate[3]
- Discrimination performance of the risk score was assessed using the area under the receiver operating characteristic (ROC) curve at 1-year and 2-year follow-up

RESULTS

Patient characteristics

- Of the 1377 patients included in this analysis, 139 experienced major bleeding (**Table 1**)
- Patients with vs without major bleeding had a higher prevalence of hypercholesterolemia, were more likely to have undergone percutaneous coronary intervention (PCI) within 30 days before TAVI, and had a higher mean ejection fraction and mean HAS-BLED score (**Table 1**)

HARP score

- Four parameters emerged as predictors of major bleeding (**Table 2**) and formed the HARP (low Haemoglobin, excessive Alcohol use, abnormal Renal function, and PCI performed within 30 days of TAVI) score, with each parameter given a weight of 1 (**Figure 1**)
- There was a significant difference in the risk of major bleeding between all 3 HARP risk score categories (**Figure 1**)
 - Patients with high vs moderate risk had 3.0 times higher risk of major bleeding, whereas patients with high vs low risk had 6.5 times higher risk
 - Patients with moderate vs low risk had 2.1 times higher risk for major bleeding
- The concordance statistic (c-statistic) for the HARP score was 0.6 (**Figure 2**)

CONCLUSIONS

The HARP score components were the identified modifiable (excessive alcohol use and PCI performed within 30 days of TAVI) and nonmodifiable (low haemoglobin, abnormal renal function) clinical risk factors significantly associated with major bleeding among patients with prevalent or incident AF receiving an oral anticoagulant after successful TAVI

A priori identification of these risk factors and the derived HARP score may be useful to perform risk assessments to facilitate discharge planning and follow-up evaluations of patients with AF who undergo TAVI

The HARP (low **H**aemoglobin, excessive **A**lcohol use, abnormal **R**enal function, and **P**CI performed within 30 days of TAVI) score is a novel tool to assess the risk of major bleeding after successful TAVI in patients with AF receiving chronic oral anticoagulation treatment.

TABLES AND FIGURES

Table 1. Baseline patient demographics and clinical characteristics

Parameter	Major bleeding n = 139	No major bleeding n = 1238	*P*-value
Age at enrollment, years, mean ± SD	81.8 ± 5.6	82.1 ± 5.4	0.5
Age ≥75	124 (89.2)	1137 (91.8)	
Sex, female	69 (49.6)	589 (47.6)	0.7
Race, White	119 (85.6)	1027 (83.0)	
BMI, kg/m^2, mean ± SD	27.7 ± 5.7	27.7 ± 5.5	1.0
CrCL, mL/min, mean ± SD	57.0 ± 22.5	58.4 ± 24.3	0.5
HAS-BLED score, mean ± SD	1.8 ± 0.9	1.5 ± 0.7	**<0.0001**
CHA_2DS_2-VASc score, mean ± SD	4.6 ± 1.4	4.5 ± 1.3	0.2
STS score, mean ± SD	5.0 ± 3.3	4.9 ± 3.9	0.8
EuroScore I, mean ± SD	12.8 ± 9.9	12.9 ± 9.9	0.9
EuroScore II, mean ± SD	4.5 ± 4.1	4.6 ± 5.7	0.9
Congestive heart failure	116 (83.5)	1047 (84.6)	0.7
Diabetes mellitus	51 (36.7)	455 (36.8)	1.0
Ejection fraction, mean ± SD	58.4 ± 9.9	55.2 ± 11.5	**0.002**
Hypercholesterolemia	111 (79.9)	853 (68.9)	**0.008**
Hypertension	126 (90.6)	1132 (91.4)	0.8
Ischaemic stroke/TIA	31 (22.3)	202 (16.3)	0.09
Pre-TAVI VKA	70 (50.4)	563 (45.5)	0.3
Pre-TAVI NOAC	34 (24.5)	350 (28.3)	0.4
Predisposition to bleeding	17 (12.2)	102 (8.2)	0.1
Prior PCI	39 (28.1)	315 (25.4)	0.5
PCI performed within 30 days before TAVI	13 (9.4)	49 (4.0)	**0.008**
Valvular heart disease	139 (100.0)	1238 (100.0)	0.7

Data are presented as n (%) unless otherwise specified.

Table 2. Components of the HARP score‡

HARP score component	Hazard ratio (95% CI)	*P*-value	Coefficient	Weight
Haemoglobin: low vs normal*	2.26 (1.53–3.33)	<0.0001	0.814	1
excessive Alcohol use (≥8 drinks/week)	3.17 (1.40–7.18)	0.006	1.155	1
abnormal Renal function†	2.80 (1.19–6.59)	0.02	1.029	1
PCI performed within 30 days before TAVI	2.42 (1.33–4.40)	0.004	0.882	1

Analyses were "Yes" vs "No" unless otherwise indicated.
*Haemoglobin was defined as low (male: <120 g/L; female: <110 g/L) and normal (male: ≥120 g/L; female: ≥110 g/L).
†Abnormal renal function was defined as dialysis, transplant, Cr >2.26 mg/dL or >200 µmol/L.
‡Total number of patients included in the model were those with complete data for excessive alcohol use, haemoglobin, abnormal renal function, and recent PCI (n = 1367).

Figure 1. Occurrence of major bleeding by HARP risk score category

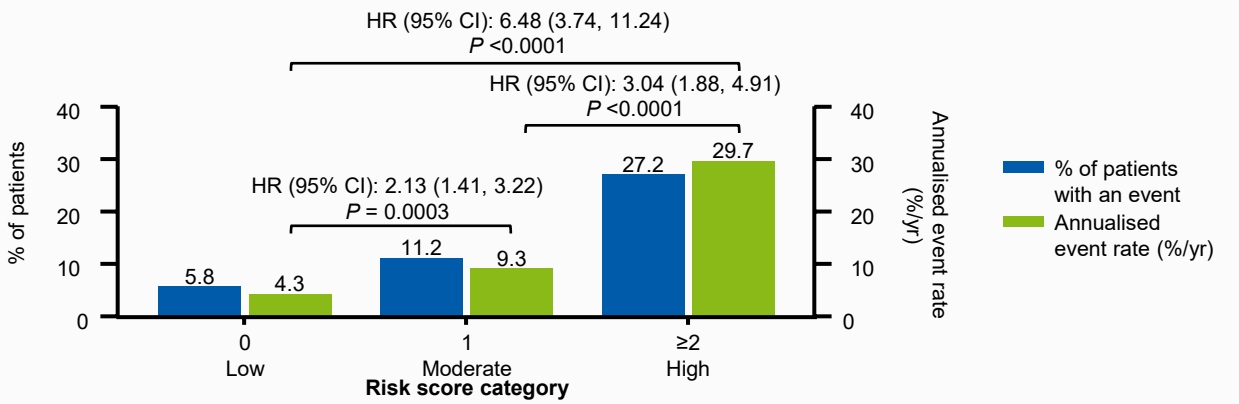
HR (95% CI): 6.48 (3.74, 11.24)
P <0.0001
HR (95% CI): 3.04 (1.88, 4.91)
P <0.0001
HR (95% CI): 2.13 (1.41, 3.22)
P = 0.0003
% of patients
Annualised event rate (%/yr)
0
10
20
30
40
5.8
4.3
11.2
9.3
27.2
29.7
0
Low
1
Moderate
≥2
High
Risk score category
% of patients with an event
Annualised event rate (%/yr)

	Low	Moderate	High
Number of events	32	82	22
Total number of patients*	552	734	81

*Total number of patients included in the model were those with complete data for excessive alcohol use, haemoglobin, abnormal renal function, and recent PCI (n = 1367).

Figure 2. One- (A) and two- (B) year receiver operating characteristic curves for the HARP score

(A)

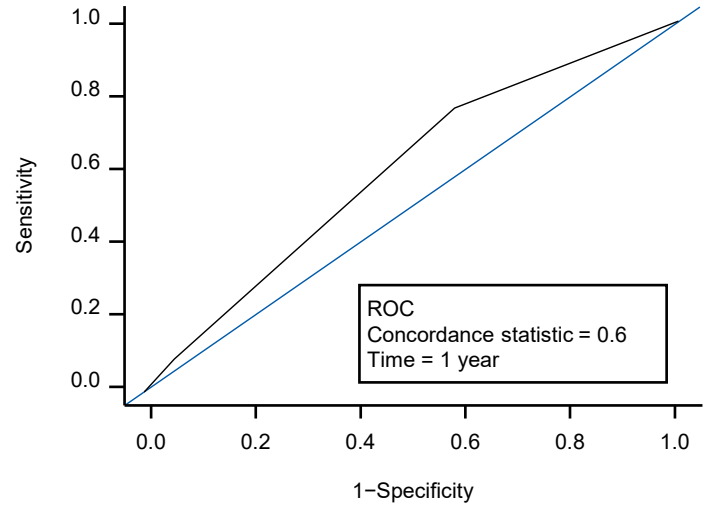
Sensitivity
1−Specificity
0.0
0.2
0.4
0.6
0.8
1.0
ROC
Concordance statistic = 0.6
Time = 1 year

(B)

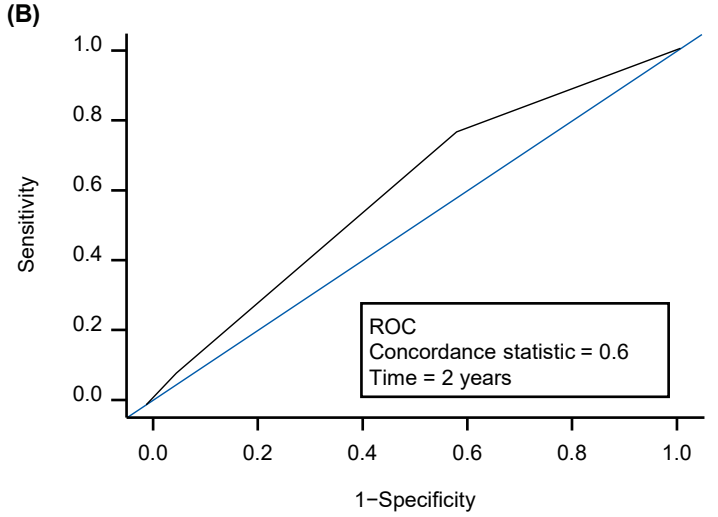
Sensitivity
1−Specificity
0.0
0.2
0.4
0.6
0.8
1.0
ROC
Concordance statistic = 0.6
Time = 2 years

Predictors used in the ROC curve included low haemoglobin, excessive alcohol use, abnormal renal function, and PCI performed within 30 days before TAVI.

Presented at: EuroPCR, May 14–17, 2024, in Paris, France

REFERENCES

1. Van Mieghem NM, et al. *N Engl J Med.* 2021;385(23):2150-60.
2. Van Mieghem NM, et al. *Am Heart J.* 2018;205:63-9.
3. Sullivan LM, et al. *Stat Med.* 2004;23(10):1631-60.

ACKNOWLEDGEMENTS

This study was funded by Daiichi Sankyo. Medical writing and editorial support were provided by Stephanie Justice-Bitner, PhD, of AlphaBioCom, a Red Nucleus company, and funded by Daiichi Sankyo. The authors would like to thank James Jin, PhD for his valuable contributions to the data analysis of this project.

DISCLOSURES

CH: a clinical proctor for Boston Scientific and Edwards Lifesciences; reports payment for speaker bureaus, support for attending meetings, and advisory board participation from Daiichi Sankyo; and received institutional funding from Abbott Vascular, Amgen, Biosensors, Biotronik, Boehringer Ingelheim, Boston Scientific, Daiichi Sankyo, Edwards Lifesciences, Ferrer, Medtronic, Novartis, Philips, Siemens, and Terumo. **MU, JvZ,** and **CC:** employees of Daiichi Sankyo. **JN:** nothing to disclose. **RMo:** received payments for personal fees from Abbott Vascular, Amgen, Biosensors, Boston Scientific, Daiichi Sankyo, Edwards Lifesciences, Ferrer, Medtronic, Philips, and Terumo. **RMe:** received research grants to institution and support for attending meetings from Bayer and Daiichi Sankyo; and consulting fees from Daiichi Sankyo. **MV:** received personal fees from Alvimedica/CID, AstraZeneca, Boston Scientific, Bristol Myers Squibb, Chiesi, Medtronic, Novartis, and Vifor; grants and personal fees from Terumo; and consultancy fees from Abbott Vascular, Bayer, Biotronik, CoreFLOW, Daiichi Sankyo, Idorsia Pharmaceuticals Ltd., PhaseBio, Universität Basel, and Vesalio. **NMVM:** received grants or contracts from Abbott, Abiomed, Boston Scientific, Daiichi Sankyo, Edwards Lifesciences, Medtronic, PulseCath BV, and Siemens. **GDD:** received research grants to institution and support for attending meetings from Bayer and Daiichi Sankyo; and consulting fees from Daiichi Sankyo.

ABBREVIATIONS

AF, atrial fibrillation; BMI, body mass index; CHA_2DS_2-VASc, Congestive heart failure, Hypertension, Age ≥75 (doubled), Diabetes, Stroke (doubled), Vascular disease, Age 65 to 74, and Sex category (female); CI, confidence interval; Cr, creatinine; CrCL, creatinine clearance; HARP, low Haemoglobin, excessive Alcohol use, abnormal Renal function, and PCI performed within 30 days of TAVI; HAS-BLED, Hypertension, Abnormal liver/renal function, Stroke, Bleeding history or predisposition, Labile INR, Elderly, Drugs/alcohol concomitantly; HR, hazard ratio; INR, international normalised ratio; NOAC, non–vitamin K oral anticoagulant; PCI, percutaneous coronary intervention; ROC, receiver operating characteristic; SD, standard deviation; STS, Society of Thoracic Surgeons; TAVI, transcatheter aortic valve implantation; TIA, transient ischaemic attack; VKA, vitamin K antagonist; yr, year.

ENVISAGE-TAVI AF