QuANTUM-First: Efficacy by Age in Newly Diagnosed Patients With FMS-Like Tyrosine Kinase 3-Internal Tandem Duplication–Positive Acute Myeloid Leukemia

P589

Pau Montesinos,[1] Elżbieta Patkowska,[2] Jorge Cortes,[3] Alexander E. Perl,[4] Mikkael A. Sekeres,[5] Hervé Dombret,[6] Sergio Amadori,[7] Jianxiang Wang,[8] Mark J. Levis,[9] Richard F. Schlenk,[10] Li Liu,[11] Yasser Mostafa Kamel,[11] Karima Imadalou,[11] Abderrahmane Laadem,[11] Arnaud Lesegretain,[11] Harry P. Erba[12]

[1]La Fe University and Polytechnic Hospital, Valencia, Spain; [2]Institute of Hematology and Blood Transfusion, Warsaw, Poland; [3]Georgia Cancer Center at Augusta University, Augusta, GA, USA; [4]Division of Hematology/Oncology, University of Pennsylvania, Philadelphia, PA, USA; [5]Sylvester Cancer Center, University of Miami Health System, Miami, FL, USA; [6]Saint Louis Hospital, University of Paris, Paris, France; [7]Tor Vergata Polyclinic Hospital Rome, Rome, Italy; [8]Institute of Hematology and Blood Diseases Hospital, Tianjin, China; [9]Division of Hematologic Malignancies, Johns Hopkins University, Baltimore, MD, USA; [10]National Center of Tumor Diseases Trial Center, German Cancer Research Center, Department of Internal Medicine V, Heidelberg University Hospital, Heidelberg, Germany; [11]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [12]Duke Cancer Institute, Durham, NC, USA

Copies of materials obtained through the QR code or /bit.ly link are for personal use only and may not be reproduced without written permission of the authors.

BACKGROUND

- The phase 3 QuANTUM-First study (NCT02668653) investigated the addition of the oral, highly potent, second-generation, and selective type 2 FMS-like tyrosine kinase 3 (FLT3) inhibitor quizartinib to standard induction and consolidation chemotherapy and/or allogeneic hematopoietic cell transplantation (allo-HCT), followed by continuation monotherapy with quizartinib or placebo for up to 36 cycles[1]
- QuANTUM-First demonstrated a clinically meaningful and statistically significant improvement in overall survival (OS) versus standard therapy (placebo group) in patients 18-75 years of age with newly diagnosed *FLT3*-internal tandem duplication (ITD)–positive acute myeloid leukemia (AML)[1]
 - Quizartinib treatment reduced the relative risk of death by 22.4% versus placebo (hazard ratio [HR], 0.78; 95% CI, 0.62-0.98; *P*=0.032)[1]
 - In a post hoc subgroup analysis of OS by age group (<60 years vs ≥60 years), the HR for OS was 0.68 (95% CI, 0.49-0.95) in patients <60 years of age and 0.91 (95% CI, 0.66-1.26) in patients ≥60 years of age[1]
- Quizartinib has recently been approved in the United States (US),[2,3] European Union (EU),[4,5] UK,[6,7] and Japan[8] in combination with chemotherapy across induction, consolidation, and as maintenance monotherapy (but not after transplantation in the US) for the treatment of adult patients with newly diagnosed *FLT3*-ITD–positive AML

OBJECTIVES

- To further evaluate the impact of age (<60 years vs ≥60 years) on the efficacy of quizartinib in patients with newly diagnosed *FLT3*-ITD–positive AML, by analyzing rates and duration of complete remission (CR), cumulative incidence of relapse (CIR), relapse-free survival (RFS), rates of allo-HCT in the first CR (CR1), and impact of allo-HCT in CR1 on OS and event-free survival (EFS)

METHODS

- A detailed description of the QuANTUM-First study has been previously published[1]
- Eligible adult patients (18-75 years of age) with newly diagnosed *FLT3*-ITD–positive AML were randomized 1:1 to receive standard induction chemotherapy with either quizartinib (40 mg/day) or placebo and stratified by region (EU, North America, or Asia/Australia/South America), age (<60 years vs ≥60 years), and white blood cell (WBC; <40×10^9/L vs ≥40×10^9/L) count at diagnosis[1]
- Patients who achieved CR or CR with incomplete hematologic recovery received ≤4 cycles of standard consolidation chemotherapy (cytarabine 3 g/m^2/dose for patients <60 years of age and 1.5 g/m^2/dose for patients ≥60 years of age) plus quizartinib (40 mg/day) or placebo and/or allo-HCT, followed by 36 cycles (≈3 years) of continuation monotherapy with quizartinib (30-60 mg/day) or placebo[1]
- OS and EFS were calculated in the intent-to-treat population, consisting of all randomized patients
- Duration of CR, CIR, and RFS were calculated in patients who achieved CR at the end of induction, per independent review committee
- The rates of CR were reported along with the 2-sided 95% CIs estimated according to the Clopper-Pearson method
- CIR was calculated from randomization; a competing risk analysis, with death from any cause as a competing risk, was used to compute the curves for CIR
- The medians of duration of CR, RFS, OS, and EFS were calculated using the Kaplan-Meier method, the 2-sided 95% CIs using the method of Brookmeyer–Crowley, and the HRs with the 95% CI using unstratified Cox regression
- All the analyses presented are post hoc analyses
- Analysis of the frequency and severity of adverse events (AE) were conducted on the safety analysis set (defined as all patients who received ≥1 dose of study drug). Grading and management of AEs was as per investigator

RESULTS

Patient and Baseline Characteristics by Age Group

- In QuANTUM-First, 539 patients were randomized to receive either quizartinib (n=268) or placebo (n=271)[1]
- Of 539 randomized patients in QuANTUM-First, 323 (59.9%) were <60 years of age (quizartinib, n=161; placebo, n=162) and 216 (40.1%) were ≥60 years of age (quizartinib, n=107; placebo, n=109; **Table 1**)
- More patients ≥60 years of age had an Eastern Cooperative Oncology Group performance status of 1-2 (72.2%) than patients <60 years of age (61.0%; **Table 1**)
- More patients ≥60 years of age had *NPM1* mutations (61.1%) than patients <60 years of age (46.4%; **Table 1**)
- More patients ≥60 years of age had a lower count of WBCs at diagnosis (55.6%) than patients <60 years of age (47.1%; **Table 1**)

Table 1. Baseline Characteristics by Age Group (ITT Analysis Set)

	<60 years of age		≥60 years of age	
Patient characteristics	Quizartinib (n=161)	Placebo (n=162)	Quizartinib (n=107)	Placebo (n=109)
Age				
Median (range), years	48 (23-59)	49 (20-59)	66 (60-75)	66 (60-75)
ECOG PS, n (%)				
0	57 (35.4)	68 (42.0)	30 (28.0)	30 (27.5)
1	75 (46.6)	73 (45.1)	59 (55.1)	63 (57.8)
2	29 (18.0)	20 (12.3)	18 (16.8)	16 (14.7)
1-2	104 (64.6)	93 (57.4)	77 (72.0)	79 (72.5)
Missing	0	1 (0.6)	0	0
Cytogenetic risk status, n (%)				
Favorable	11 (6.8)	14 (8.6)	3 (2.8)	5 (4.6)
Intermediate	115 (71.4)	114 (70.4)	82 (76.6)	79 (72.5)
Unfavorable	12 (7.5)	17 (10.5)	7 (6.5)	10 (9.2)
Unknown	23 (14.3)	16 (9.9)	15 (14.0)	15 (13.8)
Missing	0	1 (0.6)	0	0
Mutated *NPM1*, n (%)	76 (47.2)	74 (45.7)	66 (61.7)	66 (60.6)
Mutated *CEBPA*, n (%)	21 (13.0)	12 (7.4)	7 (6.5)	12 (11.0)
AML with myelodysplasia-related changes	11 (6.8)	7 (4.3)	12 (11.2)	9 (8.3)
AML type, n (%)				
De novo AML	150 (93.2)	154 (95.1)	93 (86.9)	101 (92.7)
Secondary AML	11 (6.8)	8 (4.9)	14 (13.1)	8 (7.3)
***FLT3*-ITD/total *FLT3*, n (%)**				
≥3% to ≤25%	53 (32.9)	60 (37.0)	41 (38.3)	38 (34.9)
>25% to ≤50%	87 (54.0)	81 (50.0)	56 (52.3)	57 (52.3)
>50%	21 (13.0)	21 (13.0)	9 (8.4)	14 (12.8)
>25%	108 (67.1)	102 (63.0)	65 (60.7)	71 (65.1)
Unknown	0	0	1 (0.9)	0
WBC count at diagnosis of AML, n (%)				
<40×10^9/L	75 (46.6)	77 (47.5)	60 (56.1)	60 (55.0)
≥40×10^9/L	86 (53.4)	85 (52.5)	47 (43.9)	49 (45.0)

AML, acute myeloid leukemia; *CEBPA*, CCAAT enhancer-binding protein alpha; ECOG PS, Eastern Cooperative Oncology Group performance status; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; ITT, intent-to-treat; *NPM1*, nucleophosmin 1; WBC, white blood cell.

Drug Exposure by Age Group

- Drug exposure was similar for quizartinib versus placebo in patients <60 years of age and in patients ≥60 years of age (**Table 2**)

Table 2. Drug Exposure by Age Group (Safety Analysis Set)

	Overall (N=539)		By age group: <60 years		By age group: ≥60 years	
Parameter	Quizartinib (n=265)	Placebo (n=268)	Quizartinib (n=159)	Placebo (n=160)	Quizartinib (n=106)	Placebo (n=108)
Median (range) number of cycles	3 (1-42)	3 (1-41)	3 (1-42)	3 (1-41)	3 (1-40)	3 (1-41)
Median (range) duration of adjusted therapy, weeks[a]	6.00 (0.1-170.1)	6.00 (0.3-155.9)	6.00 (0.1-161.7)	5.93 (0.6-155.6)	6.00 (0.1-170.1)	6.00 (0.3-155.9)
Median (range) dose intensity, mg/day[b]	40.00 (11.74-58.70)	40.00 (16.43-59.17)	40.00 (11.74-58.70)	40.00 (16.43-58.74)	38.12 (20.00-57.91)	40.00 (20.00-59.17)

[a]Adjusted treatment duration (days) = sum of treatment duration (days) minus the planned off-drug days across the 3 treatment phases. [b]Dose Intensity = cumulative dose / adjusted treatment duration in days.

Rates of CR by Age Group

- There were 297 patients who achieved CR at the end of induction (<60 years of age, n=180; ≥60 years of age, n=117; **Table 3**)
- Rates of CR in patients who achieved CR at the end of induction were similar for quizartinib versus placebo in patients <60 years of age (55.9% vs 55.6%) and in patients ≥60 years of age (53.3% vs 55.0%; **Table 3**)

Duration of CR by Age Group

- Median duration of CR was longer with quizartinib versus placebo in patients <60 years of age (not reached vs 16.5 months), with an HR of 0.432 (95% CI, 0.267-0.699; **Table 3**)
- Median duration of CR was numerically longer with quizartinib versus placebo in patients ≥60 years of age (15.9 months vs 9.0 months), with an HR of 0.918 (95% CI, 0.591-1.424; **Table 3**)

Table 3. Rates of CR per IRC at the End of Induction and Duration of CR, by Age Group (ITT Analysis Set)

	Overall (N=539)		By age group: <60 years		By age group: ≥60 years	
Parameter	Quizartinib (n=268)	Placebo (n=271)	Quizartinib (n=161)	Placebo (n=162)	Quizartinib (n=107)	Placebo (n=109)
CR, n (%) [95% CI][a]	147 (54.9) [48.7-60.9]	150 (55.4) [49.2-61.4]	90 (55.9) [47.9-63.7]	90 (55.6) [47.6-63.4]	57 (53.3) [43.4-63.0]	60 (55.0) [45.2-64.6]
Median CR duration,[b] months, (n)[c] [95% CI][d]	38.6 (147) [21.9-NE]	12.4 (150) [8.8-22.7]	NR (90) [NE-NE]	16.5 (90) [9.4-NE]	15.9 (57) [11.1-26.0]	9.0 (60) [5.4-21.6]

[a]Based on the Clopper-Pearson method. [b]Duration of CR defined as the time from the first documented CR, until documented relapse or death from any cause, whichever occurred first. Duration of CR was not censored at the time of allo-HCT. [c]Number of patients who achieved CR by IRC at the end of induction. [d]Based on unstratified Cox regression analysis. CR, complete remission; IRC, independent review committee; ITT, intent-to-treat; NE, not estimable; NR, not reached.

Analysis of CIR by Age Group

- The analysis of CIR in patients who achieved CR at the end of induction showed a lower proportion of patients in the quizartinib arm having relapse at 12 and 24 months versus those in the placebo arm in both age groups (**Figure 1**)
 - In patients <60 years of age, CIR at 24 months was 22.6% in the quizartinib arm versus 37.8% in the placebo arm (**Figure 1A**)
 - In patients ≥60 years of age, CIR at 24 months was 43.9% in the quizartinib arm versus 51.0% in the placebo arm (**Figure 1B**)

Figure 1. CIR for Patients Who Achieved CR per IRC at the End of Induction by Age Group

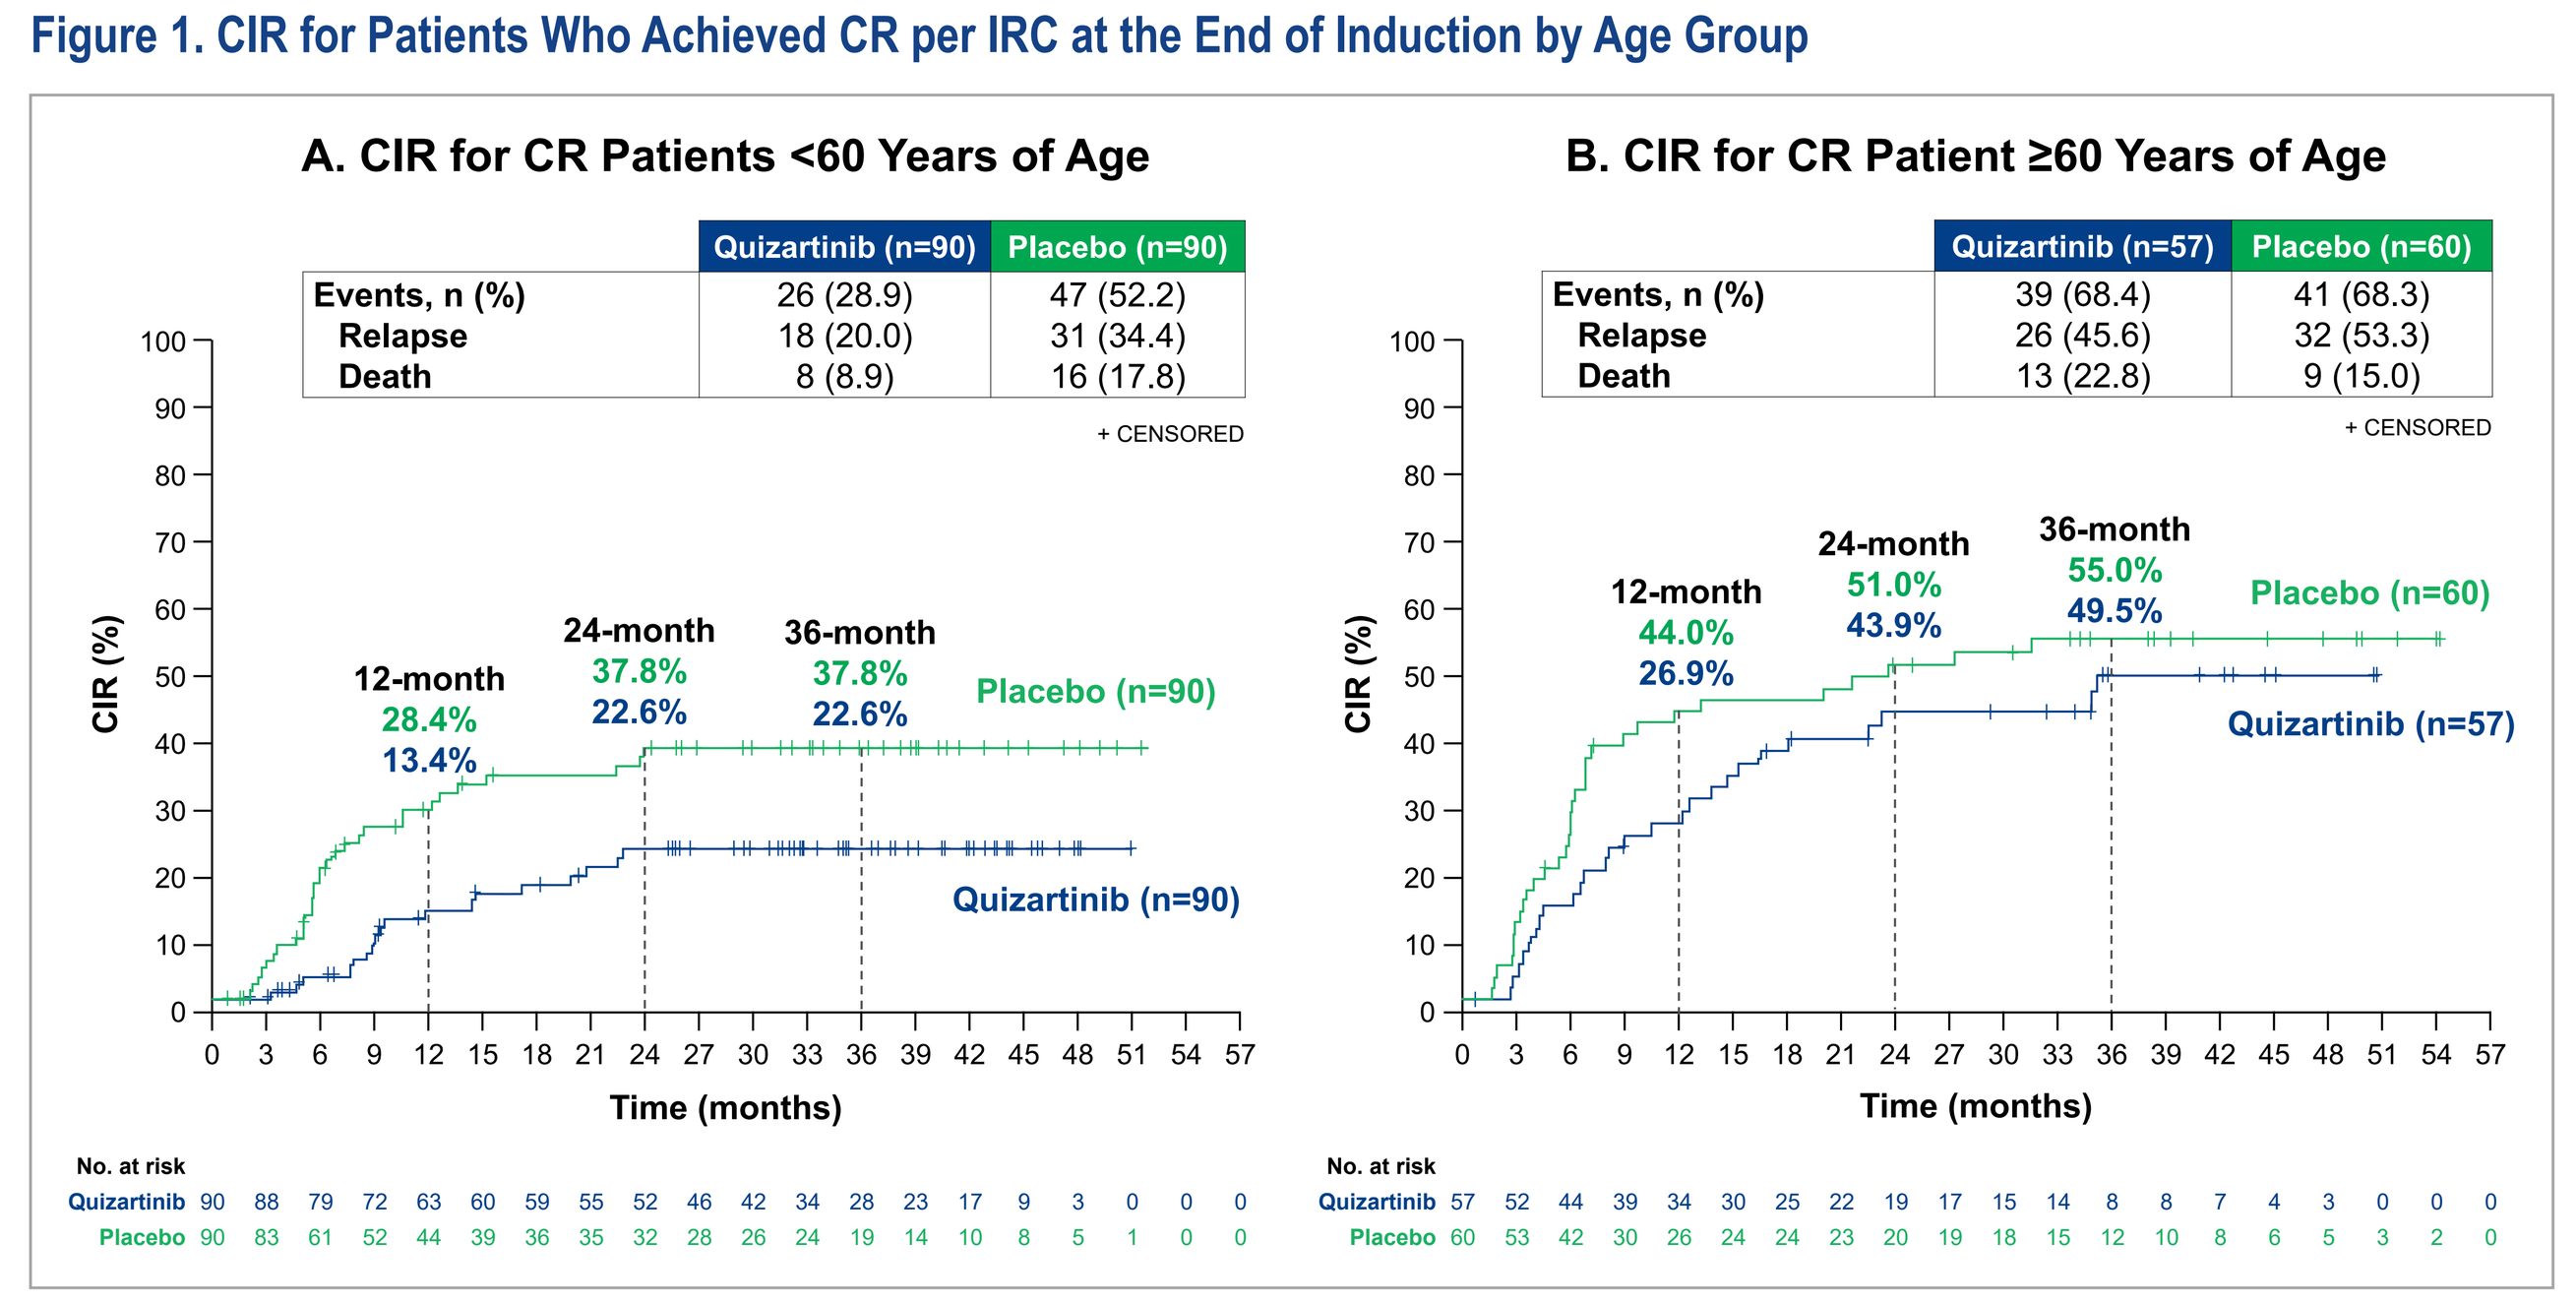
A. CIR for CR Patients <60 Years of Age
Quizartinib (n=90) | Placebo (n=90)
Events, n (%) 26 (28.9) 47 (52.2)
Relapse 18 (20.0) 31 (34.4)
Death 8 (8.9) 16 (17.8)
+ CENSORED
12-month 28.4% 13.4%
24-month 37.8% 22.6%
36-month 37.8% 22.6%
Placebo (n=90)
Quizartinib (n=90)
CIR (%)
Time (months)
No. at risk
Quizartinib 90 88 79 72 63 60 59 55 52 46 42 34 28 23 17 9 3 0 0 0
Placebo 90 83 61 52 44 39 36 35 32 28 26 24 19 14 10 8 5 1 0 0
B. CIR for CR Patient ≥60 Years of Age
Quizartinib (n=57) | Placebo (n=60)
Events, n (%) 39 (68.4) 41 (68.3)
Relapse 26 (45.6) 32 (53.3)
Death 13 (22.8) 9 (15.0)
+ CENSORED
12-month 44.0% 26.9%
24-month 51.0% 43.9%
36-month 55.0% 49.5%
Placebo (n=60)
Quizartinib (n=57)
CIR (%)
Time (months)
No. at risk
Quizartinib 57 52 44 39 34 30 25 22 19 17 15 14 8 8 7 4 3 0 0 0
Placebo 60 53 42 30 26 24 24 23 20 19 18 15 12 10 8 6 5 3 2 0

CI, confidence interval; CIR, cumulative incidence of relapse; CR, complete remission; IRC, independent review committee.

Analysis of RFS by Age Group

- The analysis of RFS in patients <60 years of age who achieved CR at the end of induction showed that the median RFS with quizartinib was longer versus placebo, with HR of 0.425 (95% CI, 0.263-0.687; **Figure 2A**)
- In patients ≥60 years of age, the median RFS was numerically longer with quizartinib versus placebo, with a HR of 0.912 (95% CI, 0.587-1.415; **Figure 2B**)

Figure 2. RFS[a] for Patients Who Achieved CR per IRC at the End of Induction by Age Group

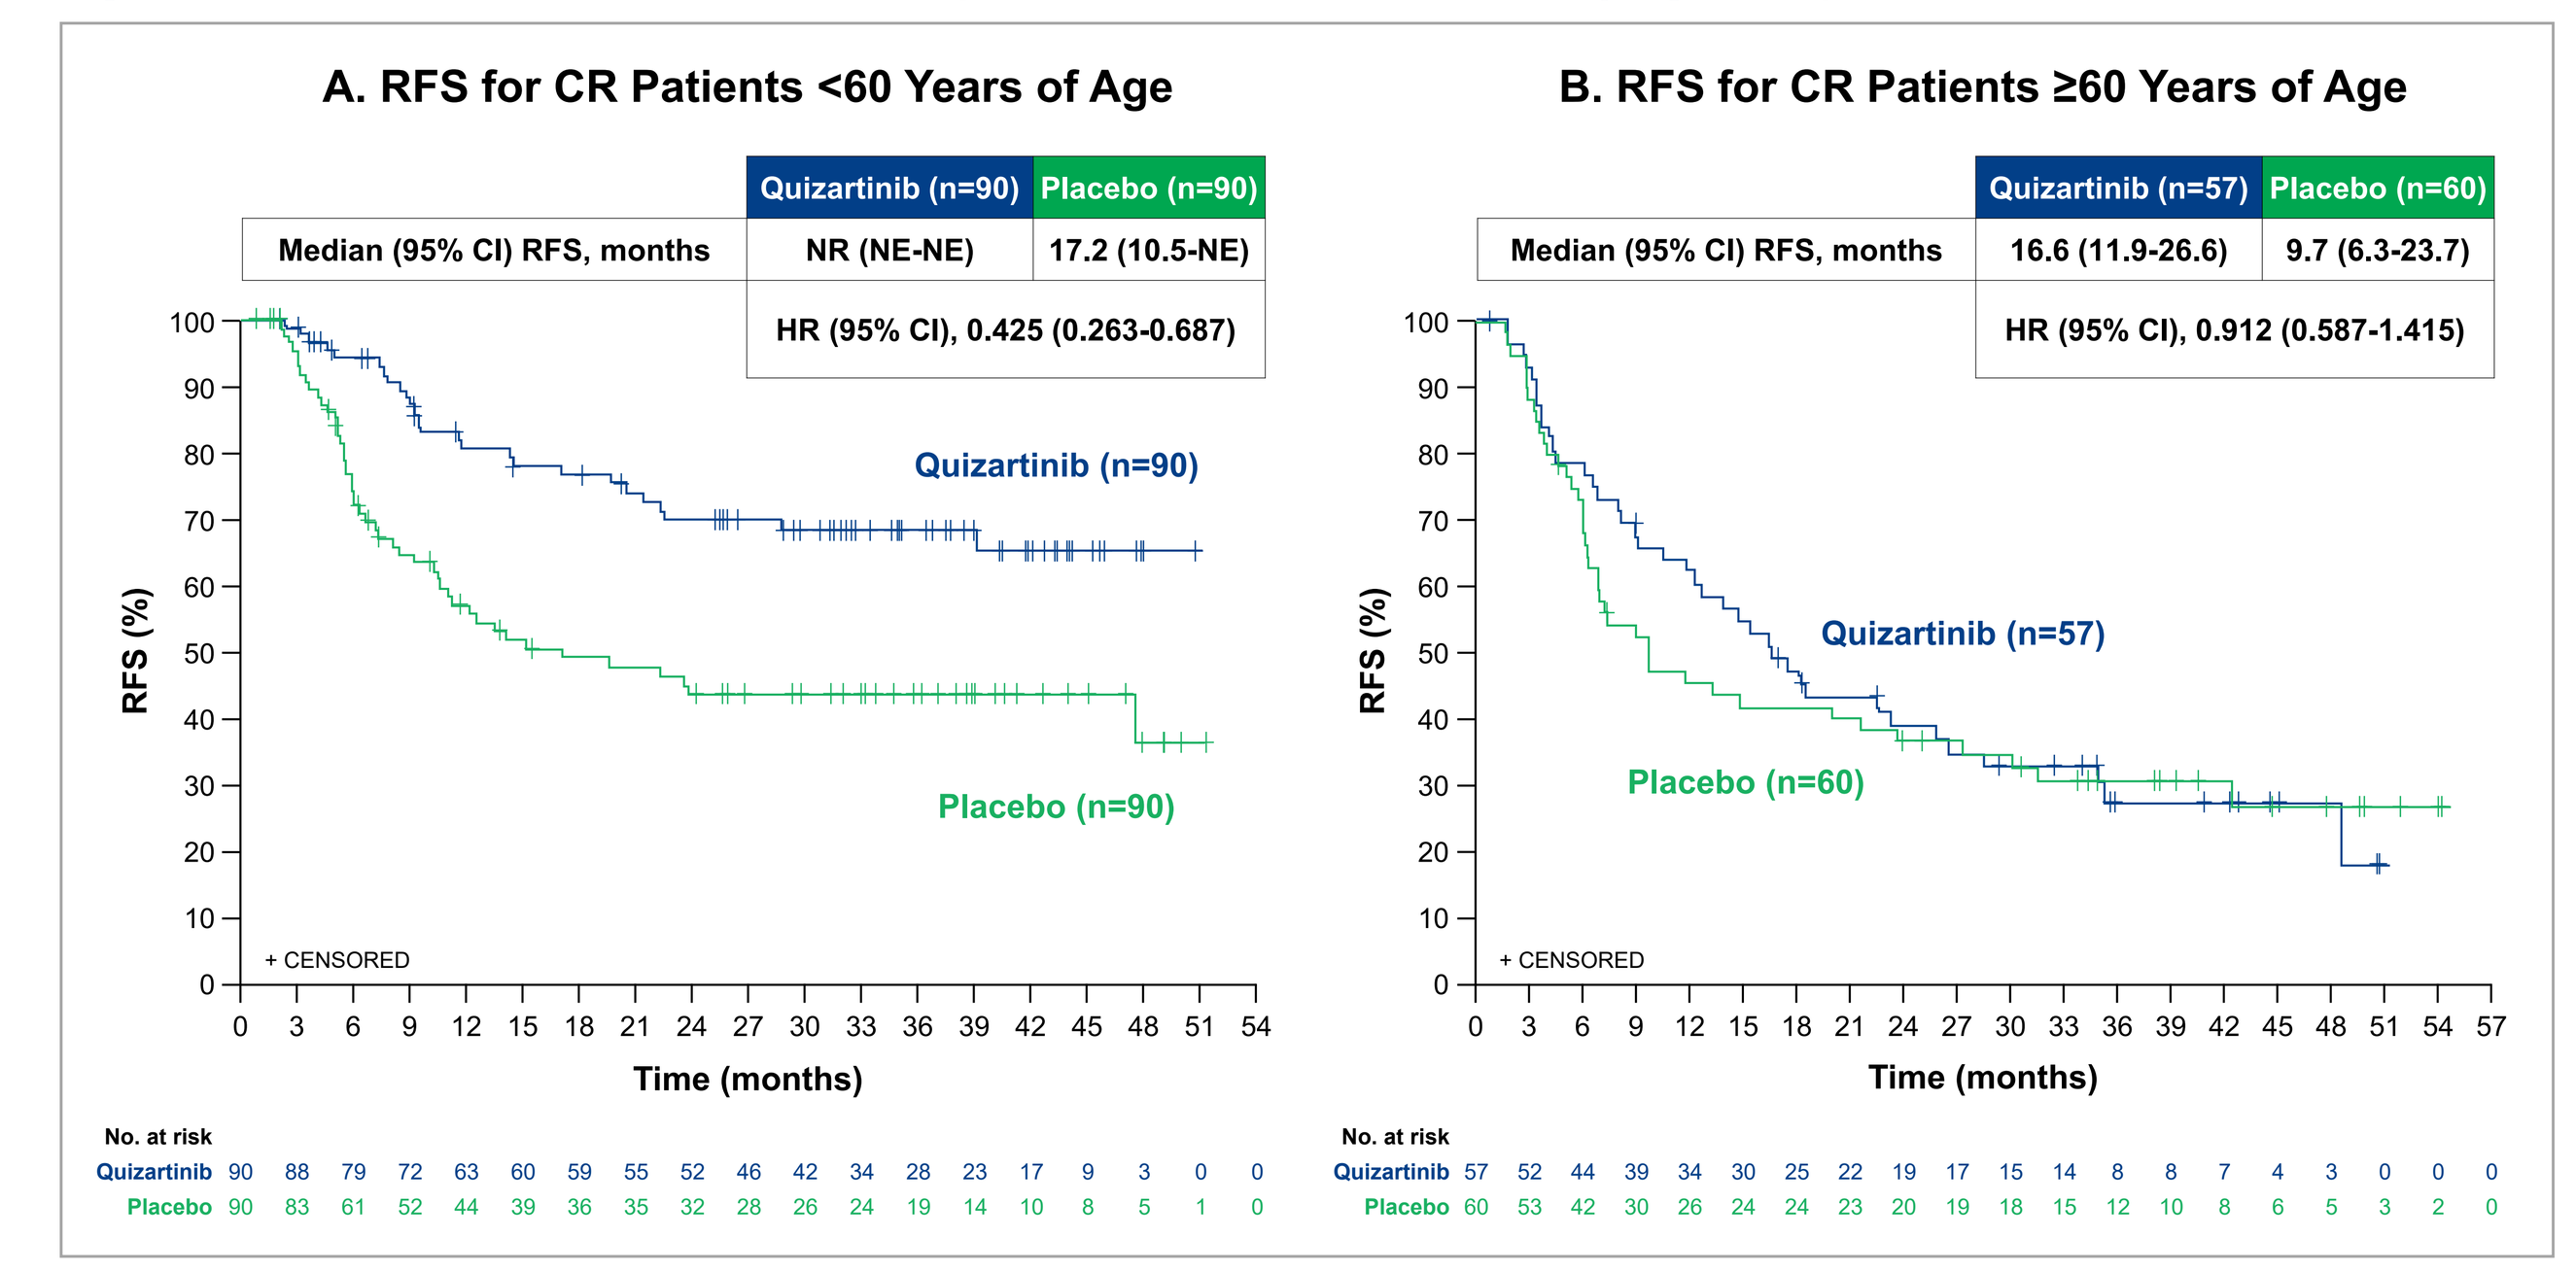
A. RFS for CR Patients <60 Years of Age
Quizartinib (n=90) | Placebo (n=90)
Median (95% CI) RFS, months NR (NE-NE) 17.2 (10.5-NE)
HR (95% CI), 0.425 (0.263-0.687)
Quizartinib (n=90)
Placebo (n=90)
+ CENSORED
RFS (%)
Time (months)
No. at risk
Quizartinib 90 88 79 72 63 60 59 55 52 46 42 34 28 23 17 9 3 0 0
Placebo 90 83 61 52 44 39 36 35 32 28 26 24 19 14 10 8 5 1 0
B. RFS for CR Patients ≥60 Years of Age
Quizartinib (n=57) | Placebo (n=60)
Median (95% CI) RFS, months 16.6 (11.9-26.6) 9.7 (6.3-23.7)
HR (95% CI), 0.912 (0.587-1.415)
Quizartinib (n=57)
Placebo (n=60)
+ CENSORED
RFS (%)
Time (months)
No. at risk
Quizartinib 57 52 44 39 34 30 25 22 19 17 15 14 8 8 7 4 3 0 0 0
Placebo 60 53 42 30 26 24 24 23 20 19 18 15 12 10 8 6 5 3 2 0

[a]RFS defined as the time from randomization until documented relapse or death from any cause, whichever occurred first. CR, complete remission; HR, hazard ratio; IRC, independent review committee; NE, not estimable; NR, not reached; RFS, relapse-free survival.

Rates of Allo-HCT in CR1 by Age Group

- Overall, 157 patients underwent allo-HCTs in CR1 (<60 years of age, n=116; ≥60 years of age, n=41; **Table 4**)
 - In patients <60 years of age, rates of allo-HCT in CR1 were numerically higher with quizartinib versus placebo (39.8% vs 32.1%)
 - In patients ≥60 years of age, rates of allo-HCT in CR1 were similar in the 2 treatment arms (18.7% vs 19.3%)

Table 4. Rates of Allo-HCT in CR1 by Age Group (ITT Analysis Set)

	Overall (N=539)		By age group: <60 years		By age group: ≥60 years	
Parameter	Quizartinib (n=268)	Placebo (n=271)	Quizartinib (n=161)	Placebo (n=162)	Quizartinib (n=107)	Placebo (n=109)
Allo-HCT in CR1, n (%) [95% CI][a]	84 (31.3) [25.8-37.3]	73 (26.9) [21.7-32.6]	64 (39.8) [32.1-47.8]	52 (32.1) [25.0-39.9]	20 (18.7) [11.8-27.4]	21 (19.3) [12.3-27.9]

Includes protocol-specified and nonprotocol-specified allo-HCT. Some patients had both protocol-specified and nonprotocol-specified allo-HCT. [a]Based on the Clopper-Pearson method. Allo-HCT, allogeneic hematopoietic cell transplantation; CR1, first complete remission; ITT, intent-to-treat.

OS by Allo-HCT in CR1 and by Age Group

- Among patients <60 years of age, quizartinib provided a survival benefit, regardless of whether they received allo-HCT in CR1 or not, with HRs of 0.470 and 0.315, respectively (**Figure 3A** and **Figure 3B**)
- Among patients ≥60 years of age, the HRs were 0.919 in those who underwent allo-HCT in CR1, and 0.821 in those who did not undergo allo-HCT in CR1 (**Figure 3C** and **Figure 3D**)

Figure 3. Impact of Allo-HCT in CR1 on OS[a] by Age Group

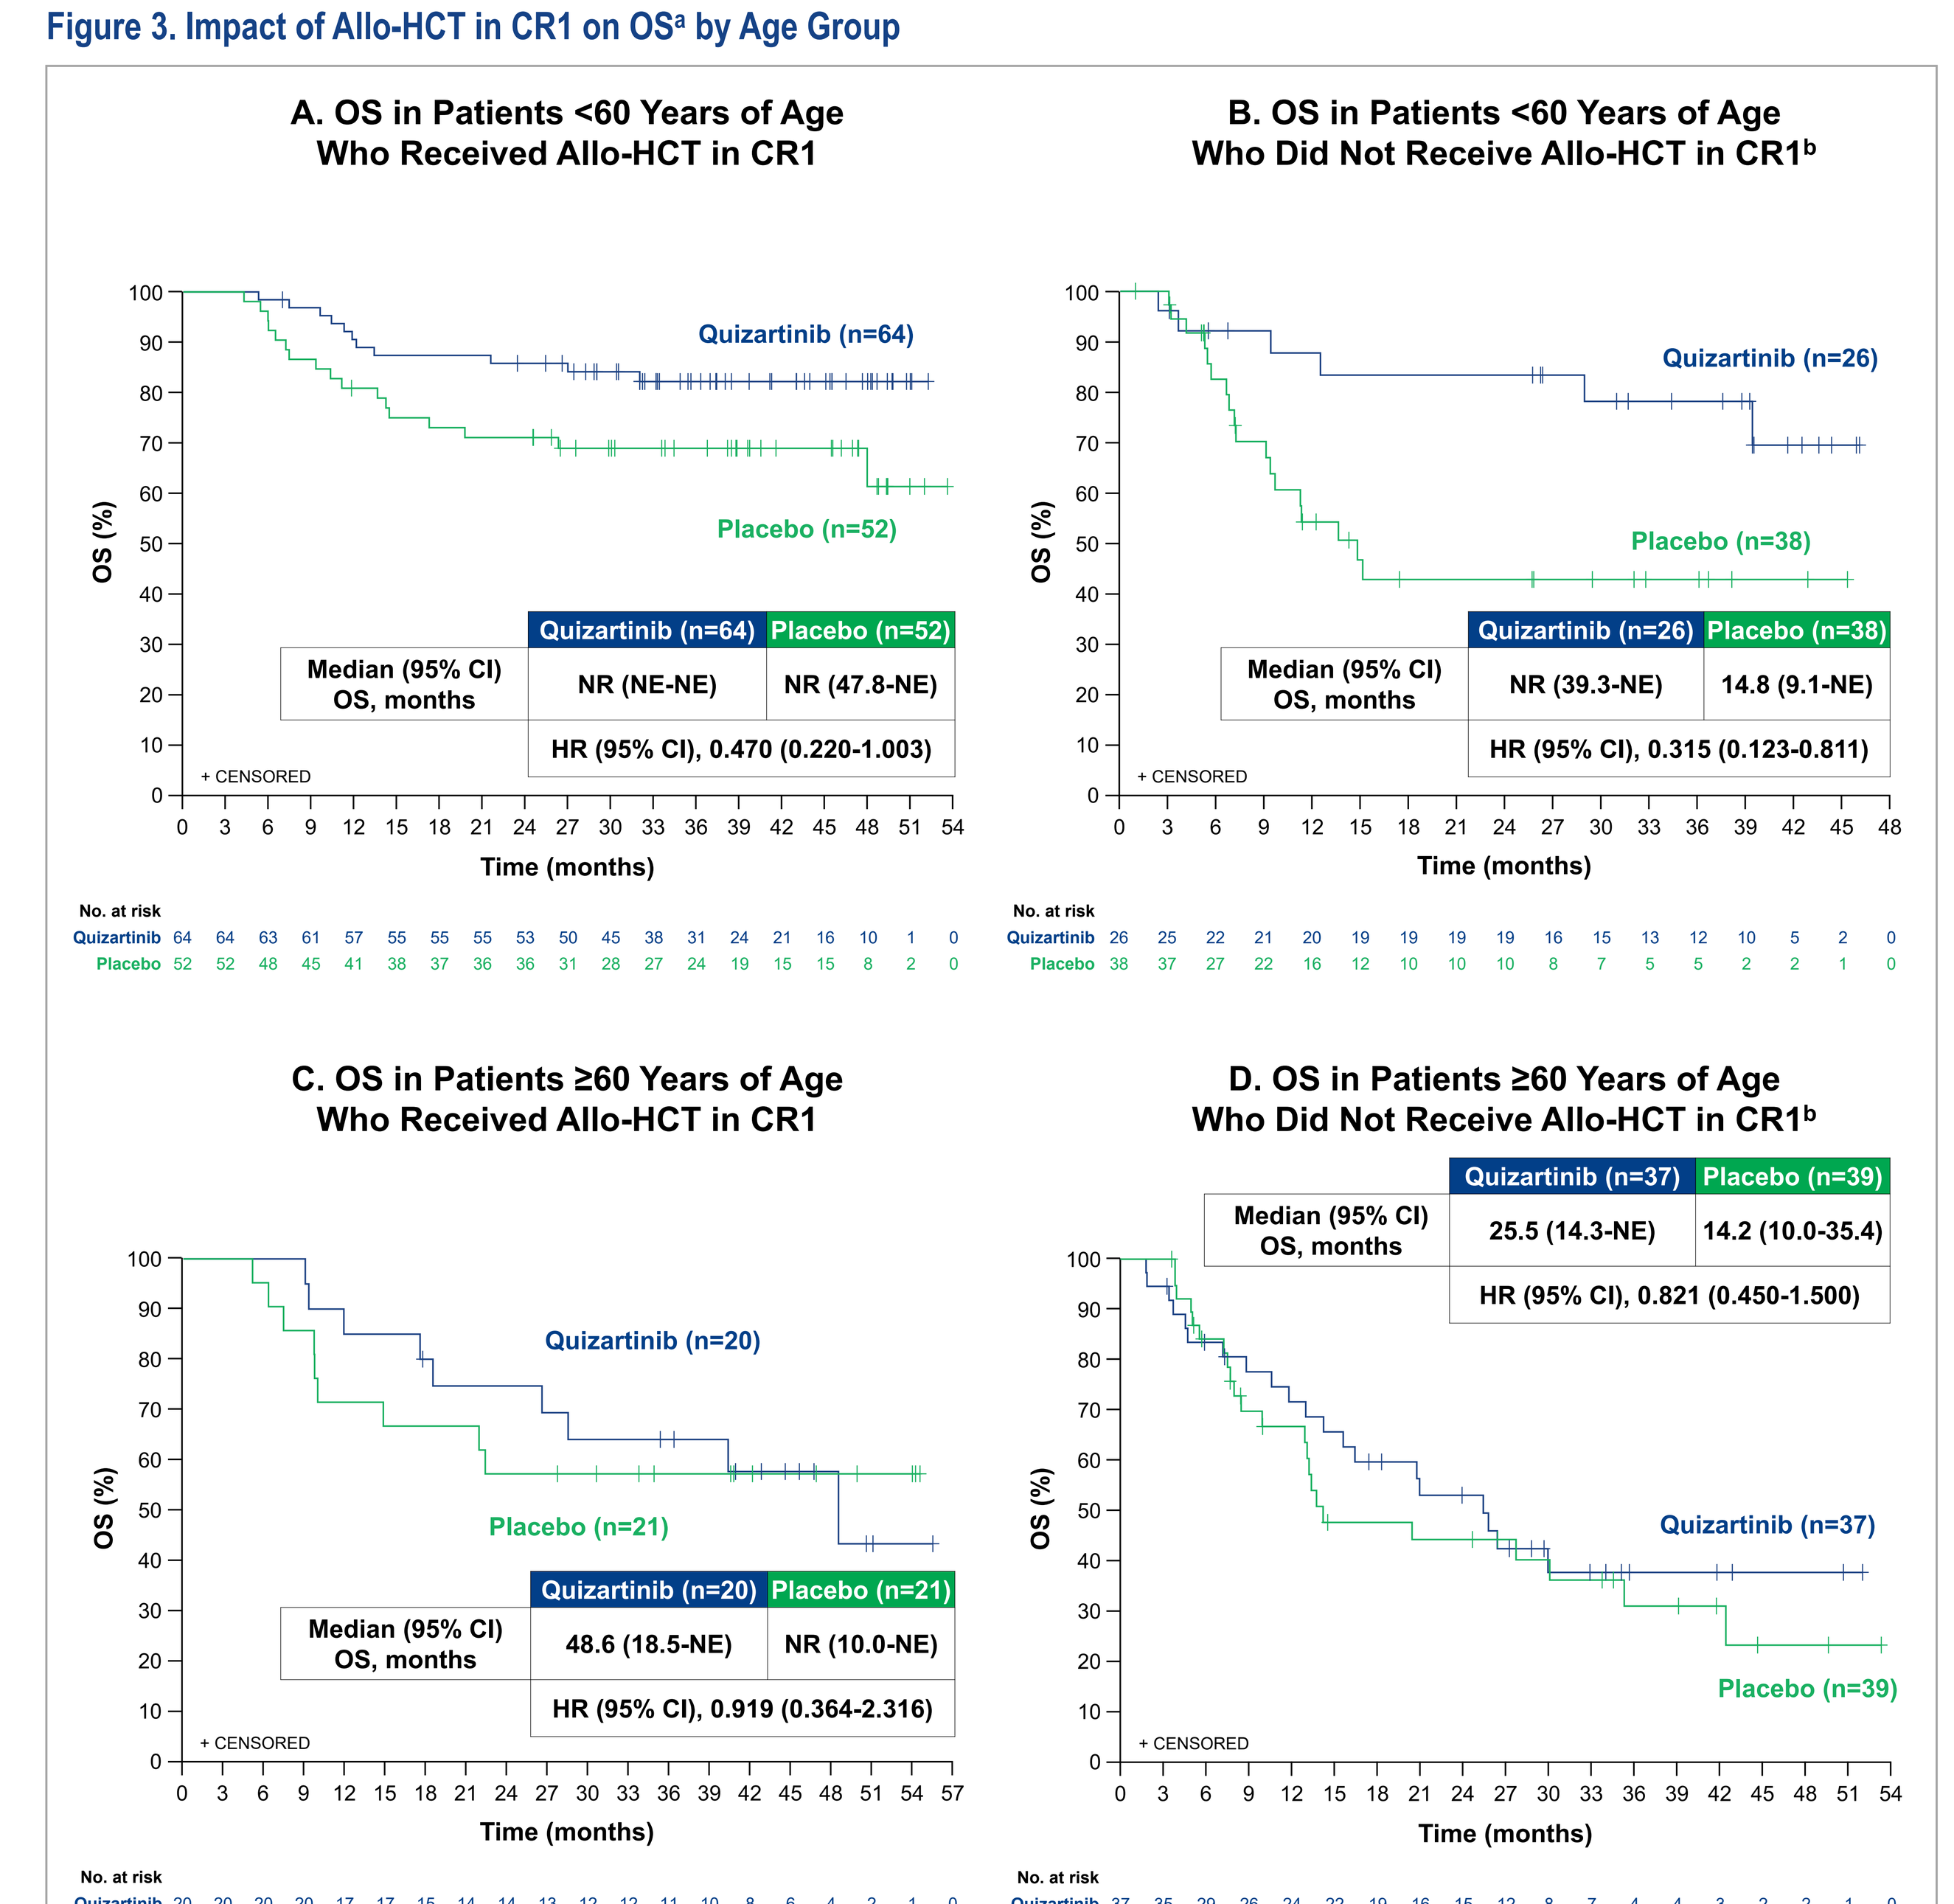
A. OS in Patients <60 Years of Age Who Received Allo-HCT in CR1
Quizartinib (n=64)
Placebo (n=52)
Quizartinib (n=64) | Placebo (n=52)
Median (95% CI) OS, months NR (NE-NE) NR (47.8-NE)
HR (95% CI), 0.470 (0.220-1.003)
+ CENSORED
OS (%)
Time (months)
No. at risk
Quizartinib 64 64 63 61 57 55 55 55 53 50 45 38 31 24 21 16 10 1 0
Placebo 52 52 48 45 41 38 37 36 36 31 28 27 24 19 15 15 8 2 0
B. OS in Patients <60 Years of Age Who Did Not Receive Allo-HCT in CR1[b]
Quizartinib (n=26)
Placebo (n=38)
Quizartinib (n=26) | Placebo (n=38)
Median (95% CI) OS, months NR (39.3-NE) 14.8 (9.1-NE)
HR (95% CI), 0.315 (0.123-0.811)
+ CENSORED
OS (%)
Time (months)
No. at risk
Quizartinib 26 25 22 21 20 19 19 19 19 16 15 13 12 10 5 2 0
Placebo 38 37 27 22 16 12 10 10 10 8 7 5 5 2 2 1 0
C. OS in Patients ≥60 Years of Age Who Received Allo-HCT in CR1
Quizartinib (n=20)
Placebo (n=21)
Quizartinib (n=20) | Placebo (n=21)
Median (95% CI) OS, months 48.6 (18.5-NE) NR (10.0-NE)
HR (95% CI), 0.919 (0.364-2.316)
+ CENSORED
OS (%)
Time (months)
No. at risk
Quizartinib 20 20 20 20 17 17 15 14 14 13 12 12 11 10 8 6 4 2 1 0
Placebo 21 21 20 18 15 14 14 14 12 12 11 10 8 8 6 5 4 3 3 0
D. OS in Patients ≥60 Years of Age Who Did Not Receive Allo-HCT in CR1[b]
Quizartinib (n=37) | Placebo (n=39)
Median (95% CI) OS, months 25.5 (14.3-NE) 14.2 (10.0-35.4)
HR (95% CI), 0.821 (0.450-1.500)
Quizartinib (n=37)
Placebo (n=39)
+ CENSORED
OS (%)
Time (months)
No. at risk
Quizartinib 37 35 29 26 24 22 19 16 15 13 12 8 7 4 4 3 2 2 1 0
Placebo 39 39 30 23 21 14 14 13 13 11 10 9 6 6 4 2 2 1 0

[a]OS was defined as the time from randomization until death from any cause. [b]Includes patients who did not receive allo-HCT at all, as well as patients who received allo-HCT outside of CR1. Allo-HCT outside of CR1 was censored at the start date of the conditioning regimen for allo-HCT. Allo-HCT, allogeneic hematopoietic cell transplantation; CR1, first complete remission; HR, hazard ratio; NE, not estimable; NR, not reached; OS, overall survival.

Analysis of EFS by Age Group

- EFS was similar between treatment arms by age group when 42 days from the last induction cycle was used to define induction treatment failure (ITF; **Table 5**)
- In a sensitivity EFS analysis, in which 56 days from the last induction cycle was used to define ITF:
 - The median EFS was longer with quizartinib versus placebo in patients <60 years of age, with a HR of 0.732 (95% CI, 0.559-0.959; **Table 5**)
 - The median EFS was numerically longer with quizartinib versus placebo in patients ≥60 years of age, with a HR of 0.965 (95% CI, 0.719-1.294; **Table 5**)

Table 5. Analyses of EFS[a] by Age Group (ITT Analysis Set)

	<60 years of age		≥60 years of age	
Parameter	Quizartinib (n=161)	Placebo (n=162)	Quizartinib (n=107)	Placebo (n=109)
Primary analysis[b]				
Median (95% CI) EFS, months	0.0 (0.0-7.6)	0.5 (0.0-5.1)	0.1 (0.0-1.9)	1.0 (0.0-3.8)
HR (95% CI)	0.859 (0.662-1.115)		1.023 (0.766-1.368)	
Sensitivity analysis[c]				
Median (95% CI) EFS, months	9.0 (0.2-19.8)	4.2 (0.0-5.9)	3.2 (0.3-6.8)	2.9 (0.0-6.0)
HR (95% CI)	0.732 (0.559-0.959)		0.965 (0.719-1.294)	

[a]EFS was defined as the time from randomization to lack of complete remission based on independent review committee assessment, relapse, or death from any cause, whichever occurred first. [b]ITF definition: not achieving complete remission by day 42 from the start of the last induction cycle. [c]ITF definition: not achieving complete remission by the end of induction, up to day 56 from the start of the last induction cycle. EFS, event-free survival; HR, hazard ratio; ITF, induction treatment failure; ITT, intent-to-treat.

Safety by Age Group

- Rates of grade 3/4 pancytopenia and grade 3/4 myelosuppression were low in the quizartinib arm in patients <60 years of age (1.9% and 1.3%, respectively) as well as in patients ≥60 years of age (2.8% and 1.9%, respectively), although higher versus the placebo arm across age groups (**Table 6**)
- Among patients with CR, the median time to recovery of either neutropenia or thrombocytopenia was numerically longer in the quizartinib group vs the placebo group across age groups (**Table 6**)
- Early deaths within 30 days and 60 days of the first dose were numerically higher in patients ≥60 years of age in the quizartinib arm versus the placebo arm (8.5% vs 5.6% and 12.3% vs 6.5%, respectively; **Table 6**)
 - Five patients (3 in the quizartinib arm and 2 in the placebo arm) died before receiving treatment
- Infections in patients ≥60 years of age were the main cause of more early deaths (within 30 and 60 days)

Table 6. Summary of Myelosuppression (Safety Analysis Set) and Early Deaths (Safety Analysis Set and ITT Analysis Set)

	Overall: Safety population (N=533)		By age group: <60 years		By age group: ≥60 years	
Parameter	Quizartinib (n=265)	Placebo (n=268)	Quizartinib (n=159)	Placebo (n=160)	Quizartinib (n=106)	Placebo (n=108)
Grade 3 or 4 pancytopenia, n (%)[a]	6 (2.3)	1 (0.4)	3 (1.9)	1 (0.6)	3 (2.8)	0
Grade 3 or 4 myelosuppression, n (%)[a]	4 (1.5)	0	2 (1.3)	0	2 (1.9)	0
Time to hematologic recovery from C1D1 in CR patients with onset of neutropenia or thrombocytopenia during induction, days						
Median (IQR) time to recovery of neutropenia (absolute neutrophil count ≥1000 cells/mm^3)	36 (29-44)	29 (27-38)	37 (29-47)	29.5 (26-39)	34 (29-42)	29 (28-37)
Median (IQR) time to recovery of thrombocytopenia (platelet count ≥100,000 cells/mm^3)	31 (28-40)	29 (26-34)	31 (28-39)	28 (26-34)	31 (28-40)	29 (27-34.5)
Rates of early death, n (%)[b]						
Deaths within 30 days of first dose	15 (5.7)	9 (3.4)	6 (3.8)	3 (1.9)	9 (8.5)	6 (5.6)
Deaths within 60 days of first dose	20 (7.5)	13 (4.9)	7 (4.4)	6 (3.8)	13 (12.3)	7 (6.5)
	ITT population (N=539)		**<60 years**		**≥60 years**	
Parameter	**Quizartinib (n=268)**	**Placebo (n=271)**	**Quizartinib (n=161)**	**Placebo (n=162)**	**Quizartinib (n=107)**	**Placebo (n=109)**
Rates of early death, n (%)[c]						
Deaths within 30 days of first dose	18 (6.7)	11 (4.1)	8 (5.0)	4 (2.5)	10 (9.3)	7 (6.4)
Deaths within 60 days of first dose	23 (8.6)	15 (5.5)	9 (5.6)	7 (4.3)	14 (13.1)	8 (7.3)

The safety-analysis population includes all patients who received ≥1 dose of quizartinib or placebo. Three patients in each group were not treated and were not included in the safety analysis population. If a patient had >1 event, that patient was counted only once. [a]Treatment-emergent adverse events regardless of causality. [b]During induction, 20 patients died in the quizartinib group, and 13 patients died in the placebo group. [c]Includes 5 patients (3 in the quizartinib arm and 2 in the placebo arm) who died before receiving treatment. C, cycle; CR, complete remission; D, day; IQR, interquartile range; ITT, intent-to-treat.

CONCLUSIONS

- QuANTUM-First is the first randomized controlled trial to investigate a targeted therapy for newly diagnosed *FLT3*-ITD–positive AML patients, including those ≥60 years of age (up to 75 years)
- The efficacy subgroup analysis by age was consistent with the primary analysis, demonstrating a benefit provided by quizartinib
- The benefit of quizartinib was evident irrespective of age group in terms of longer duration of CR, lower CIR, longer RFS, EFS (56-day window), and OS
- The benefit-risk profile of quizartinib supports its use in the treatment of newly diagnosed *FLT3*-ITD–positive AML patients

ACKNOWLEDGMENTS

We would like to thank the patients, their families, and caregivers for their participation in the QuANTUM-First study.
We would further like to thank the QuANTUM-First steering committee members, the investigators, the study staff, and independent review committee and data monitoring committee members for their important contributions. This study is sponsored by Daiichi Sankyo, Inc. Medical writing support was provided by Mohamed Abdelmegeed, MD, PhD, CMPP, and Francesca Balordi, PhD, CMPP, of The Lockwood Group (Stamford, CT, USA), in accordance with Good Publication Practice (GPP 2022) guidelines, with funding by Daiichi Sankyo, Inc.

REFERENCES

1. Erba HP, et al. *Lancet*. 2023;401(10388):1571-1583.
2. VANFLYTA® first FLT3 inhibitor approved in the U.S. specifically for patients with newly diagnosed *FLT3*-ITD positive AML. Press release. July 20, 2023. Accessed April 3, 2024. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202307/20230720_E.pdf.
3. VANFLYTA® (quizartinib). Prescribing information. Daiichi Sankyo, Inc.; July 2023. Accessed April 5, 2024. https://daiichisankyo.us/prescribing-information-portlet/getPIContent?productName=Vanflyta&inline=true.
4. VANFLYTA® approved in the EU as the first FLT3 inhibitor specifically for patients with newly diagnosed *FLT3*-ITD positive AML. Press release. November 9, 2023. Accessed April 5, 2024. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202311/20231109_E.pdf.
5. VANFLYTA® (quizartinib). Summary of product characteristics. Daiichi Sankyo, Inc.; November 2023. Accessed April 5, 2024. https://www.ema.europa.eu/en/documents/product-information/vanflyta-epar-product-information_en.pdf.
6. Quizartinib approved to treat adult patients with a type of blood cancer. Press release. March 11, 2024. Accessed May 24, 2024. https://www.gov.uk/government/news/quizartinib-approved-to-treat-adult-patients-with-a-type-of-blood-cancer.
7. VANFLYTA® (quizartinib). Summary of product characteristics. Daiichi Sankyo UK Ltd. March 2024. Accessed May 24, 2024. https://products.mhra.gov.uk/substance/?substance=QUIZARTINIB%20DIHYDROCHLORIDE.
8. VANFLYTA® first FLT3 inhibitor approved in Japan for patients with newly diagnosed *FLT3*-ITD positive AML. Press release. May 25, 2023. Accessed April 5, 2024. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202305/20230525_E.pdf.

Presented at the European Hematology Association (EHA) 2024 Hybrid Congress; June 13-16, 2024; Madrid, Spain

montesinos_pau@gva.es