Poster 936/4

Targeted proteomics and next-generation sequencing (NGS) for biomarker discovery in metastatic colorectal cancer (mCRC)

William J. Kelly[1], Barrett Nuttall[1], Marlon Rebelatto[2], Markus Schick[3], Armin Meier[3], Steve Sweet[4], Beom-Jun Kim[4], David Chain[4], Yeoun Jin Kim[4], Zhongwu Lai[5], Ying Wang[5], Todd H. Creasy[6], Ramya Gurrapu[7], Sakshi Gulati[8], Alice Barkell[9], Brian Dougherty[1], Danielle Carroll[9], Fabiola Cecchi[2]

[1]Translational Medicine, AstraZeneca, Waltham, MA,[2]Translational Medicine, AstraZeneca, Gaithersburg, MD,[3]Computational Pathology, AstraZeneca, Munich, Germany,[4]Diagnostic Proteomics, AstraZeneca, Gaithersburg, MD, [5]Oncology Data Science, AstraZeneca, Waltham, MA,[6]Oncology Data Science, AstraZeneca, Gaithersburg, MD,[7]R&D IT, AstraZeneca, Chennai, India,[8]Oncology Data Science, AstraZeneca, Cambridge, United Kingdom,[9]Translational Medicine, AstraZeneca, Cambridge, United Kingdom

Objectives

- The objective of this retrospective analysis was to characterize the prevalence of HER2 positivity in first-line mCRC and HER2 status with other molecular findings (eg, RAS/RAF mutations)
- We aimed to investigate quantitative protein biomarker profiles of mCRC and integrated the results with genomic data
- We reviewed and compared the expression of some of ADC target antigens including HER2, TROP2 and CEACAM5

Conclusions

- Our analysis provides insights into the molecular characteristics of mCRC. This preliminary data may inform future clinical development and guide patient eligibility to clinical trials and possible treatment with ADCs
- Classification by percentage of HER2-expressing cells using the histoscore algorithm and proteomics identified IHC 3+ as a distinct subpopulation
- HER2 was amplified (ISH and NGS) in all HER2 3+ patients
- HER2 IHC 3+ samples showed the highest protein expression at 30,000 amol/ug (equivalent to ~ 2M receptors/cell)
- ADC target expression is heterogeneous in CRC, however, expression of multiple ADC targets on same cancer raises the possibility of combining ADC therapies
- This analysis has identified protein biomarkers exhibiting variable expression patterns, thereby potential avenues to enhance treatment efficacy and guide options for second-line therapies. Further assessment of proteogenomic characteristics is needed due to small sample size

Plain language summary

Why did we perform this research?

- Colorectal cancer (CRC), a complex disease, requires continued research efforts to optimize treatment strategies
- Biomarker-driven approaches in the management of patients with advanced CRC are leading to improvements in outcomes
- We wanted to look at molecular characteristics of metastatic colorectal cancer (metastatic is when the cancer has spread from its original site to other parts of the body)

How did we perform this research?

- The HORIZON III study is a completed study that included people with metastatic colorectal cancer who had no previous cancer treatment
- Tissue samples from patients who participated in this study were used to perform molecular characteristics of mCRC

What were the findings of this research?

In this patient population we identified protein biomarkers exhibiting variable expression patterns in metastatic colorectal cancer

What are the implications of this research?

These findings may help guide future research on targeted treatments for people with metastatic colorectal cancer.

Where can I access more information?

More information about the HORIZON III study and previous results from this study can be found here: https://doi.org/10.1200/jco.2012.42.5355

This clinical study was sponsored by AstraZeneca in collaboration with Daiichi Sankyo. Poster presented at AACR 2024 by William Kelly.

Poster

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of this poster. Alternatively, please click on the link below.

https://bit.ly/3Pb4Nhr

Copies of this poster obtained through this QR code are for personal use only and may not be reproduced without permission from ASCO® or the authors of this poster.

Poster presented at American Association for Cancer Research AACR; April 5 - 10, 2024 San Diego Convention, California.

Corresponding author email address: fabiola.cecchi@astrazeneca.com

Introduction

- Colorectal cancer (CRC), a complex and heterogeneous disease, requires comprehensive research efforts to optimize treatment strategies
- Chemotherapy has traditionally constituted the fundamental element of systemic therapy for mCRC, although its effectiveness has been constrained by its limited range of mechanisms of action and its adverse toxicity profile
- The most studied classification divides tumors into RAS (KRAS, NRAS, HRAS) and BRAF mutated or wild-type, which guides the use of epidermal growth factor receptor (EGFR) or the B-Raf inhibitors
- Recently identified are the mismatch repair-deficient (dMMR) CRCs (15%), characterized by high microsatellite instability (MSI-H) and response to immunotherapy treatment
- Although HER2 expression occurs in few cases, numerous combinations of HER2 inhibitors have demonstrated efficacy, and antibody-drug conjugates (ADCs) against HER2 represent new strategies in this area[1-4.]
- Numerous ADCs have been evaluated in patients with advanced CRC, although only one to date is supported in guidelines for use. Multiple biomarker-driven approaches in the management of patients with advanced CRC tumors are reviewed in this analysis

Methods

- The HORIZON III trial (NCT00384176) evaluated FOLFOX + bevacizumab or cediranib (AZD2171) as first-line treatment in patients with mCRC[5]
- HER2 expression was assessed by IHC using a monoclonal anti-HER2 antibody (PATHWAY HER2 [4B5] Ventana) on primary tumor sections and scored according to ASCO/CAP guidelines for gastric cancer[6]
- Molecular Characterization: Sample libraries were constructed using KAPA HyperPlus Kit 96 rxn [Roche]
- Target capture was performed and divided among the IDT exome v2 panel, AZ700 panel or both, depending on sample input and tumor content
- Libraries underwent sequencing procedures on the Illumina NovaSeq 6000 platform
- Sample data was uploaded to our internal cBioPortal for genomic analysis[7,8]
- Absolute quantitation of protein targets was performed using parallel reaction monitoring (PRM) mass spectrometry after collecting tumor epithelial cells by laser microdissection and punch biopsies followed by trypsin digestion

Proportion of HER2-expressing cells by IHC status (n = 432)

IHC and ISH status, n (%)	Total N=432
HER2-negative	423 (97.92)[a]
IHC 0	401 (94.80)
IHC 1+	19 (4.40)
IHC 2+/ISH−	3 (0.69)
HER2-positive	8 (1.85)[a]
IHC 2+/ISH+	3 (0.69)
IHC 3+/ISH+	5 (1.16)

Table 1. [a] HER2-negative and HER2-positive categories sum to <100% because 1 sample categorized as IHC 2+ did not have ISH data available.

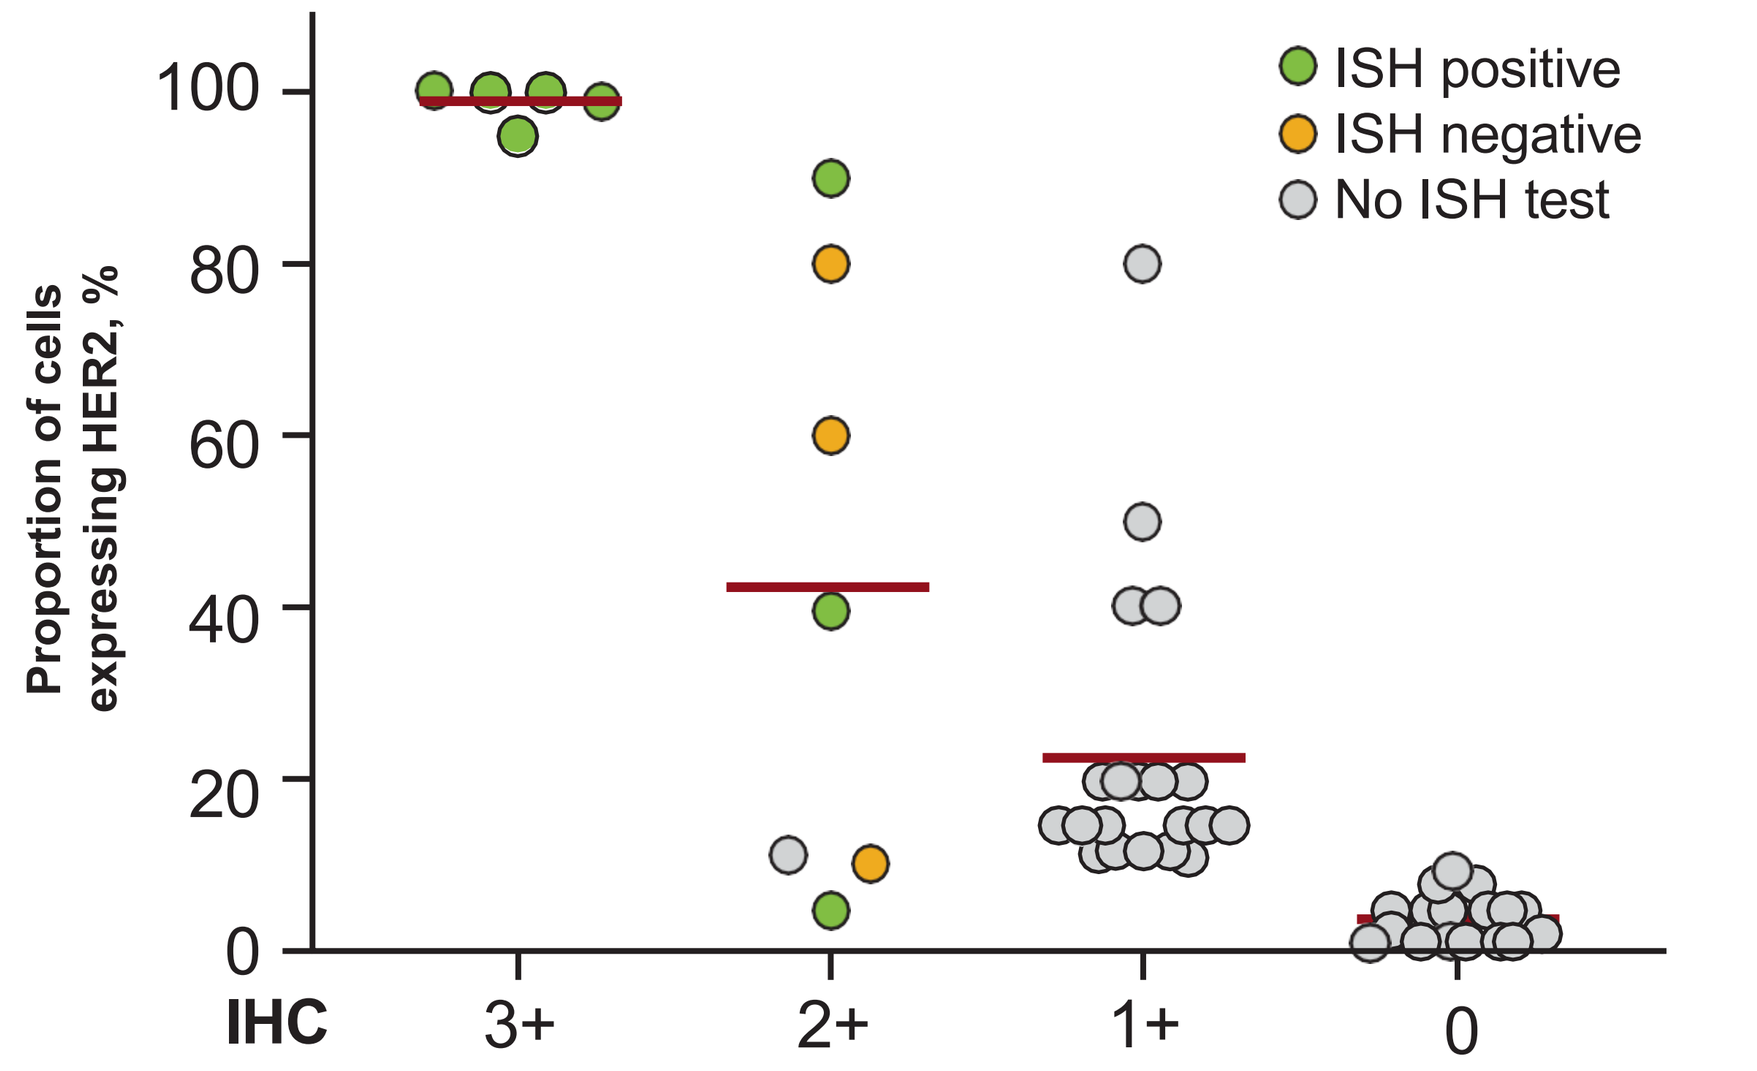
ISH positive
ISH negative
No ISH test
Proportion of cells expressing HER2, %
IHC
3+
2+
1+
0

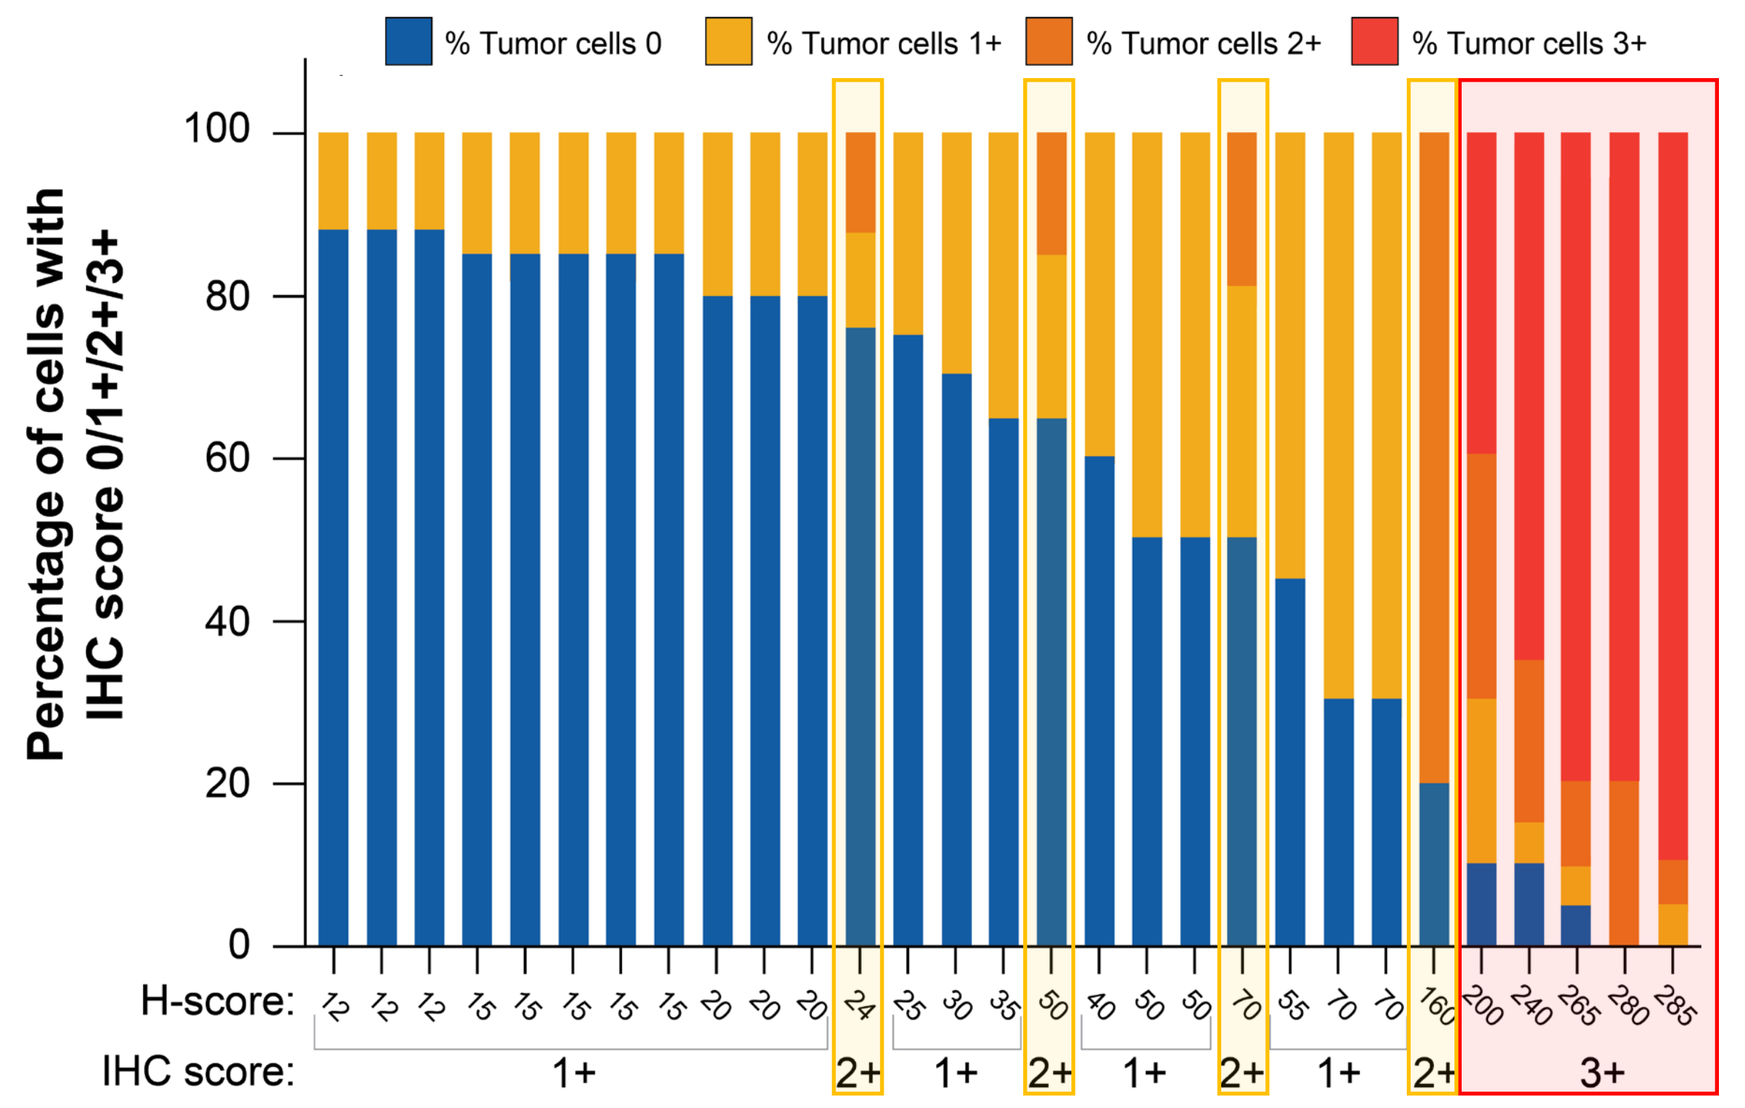
% Tumor cells 0
% Tumor cells 1+
% Tumor cells 2+
% Tumor cells 3+
Percentage of cells with IHC score 0/1+/2+/3+
H-score:
IHC score:

Fig 1A-B. Compared with IHC 3+, IHC 2+ tumors were more heterogeneous, with mixed components of cells weakly expressing or lacking HER2

Oncoprint analysis of NGS mutation data (n = 135)

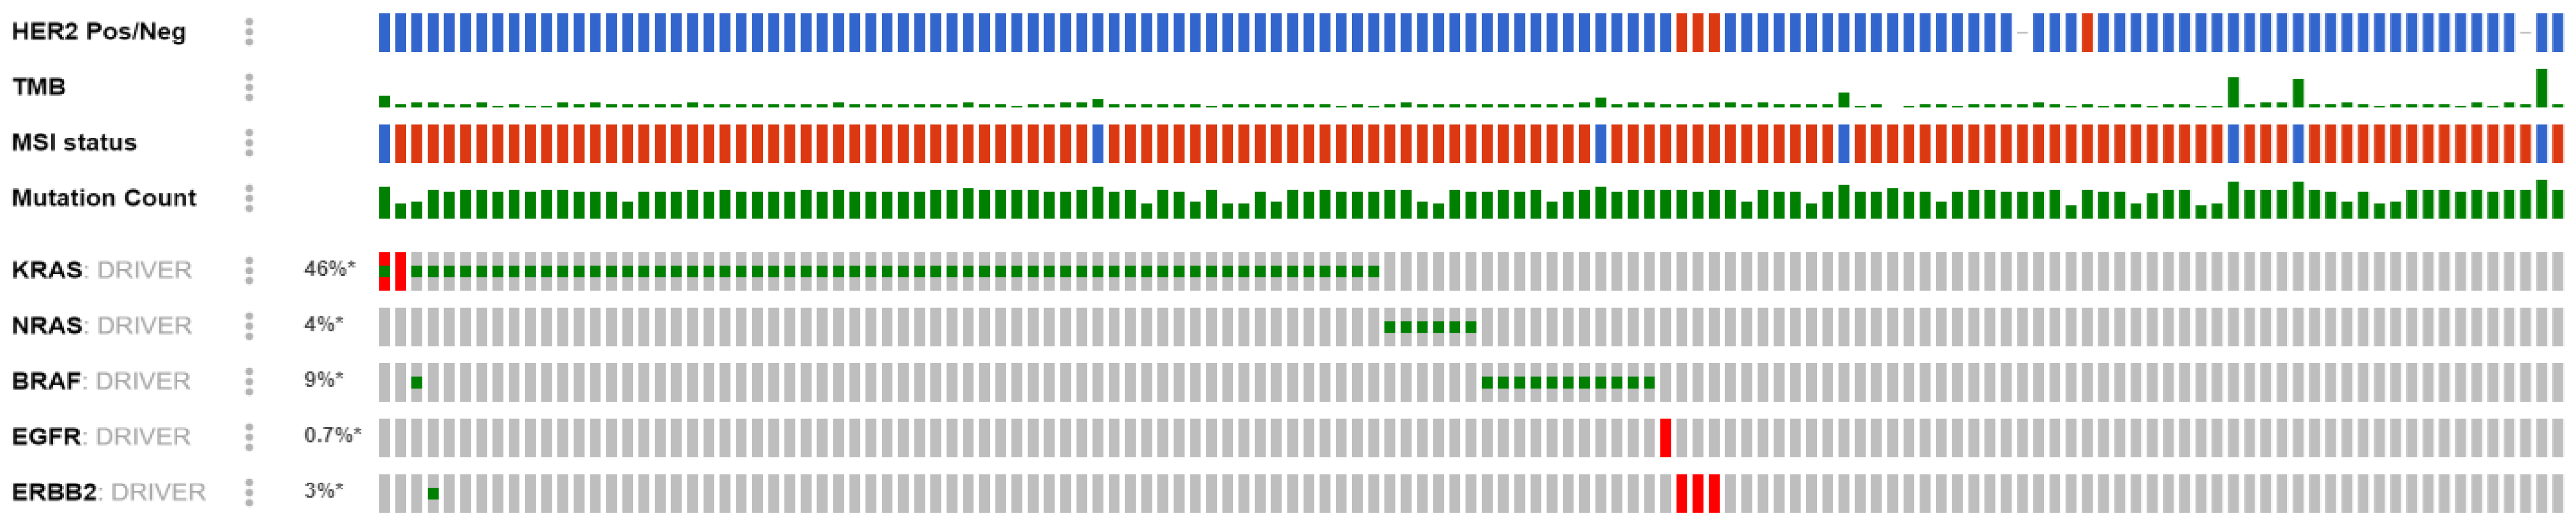
HER2 Pos/Neg
TMB
MSI status
Mutation Count
KRAS: DRIVER 46%*
NRAS: DRIVER 4%*
BRAF: DRIVER 9%*
EGFR: DRIVER 0.7%*
ERBB2: DRIVER 3%*

Fig 2. KRAS, NRAS, BRAF, EGFR, and ERBB2 driver genes were queried and aligned with HER2 IHC status, HER2 ISH positivity, TMB, MSI status, and mutational count clinical tracks[7,8]

Genetic Alteration: Missense Mutation (putative driver) | Amplification | No alterations | Not profiled — HER2 IHC status: 0 | 1+ | 2+ | 3+ — MSI status: MSI-H | MSS | No data

Proteomic analysis of mCRC reveals a wide range of expression for multiple drug targets (n = 60)

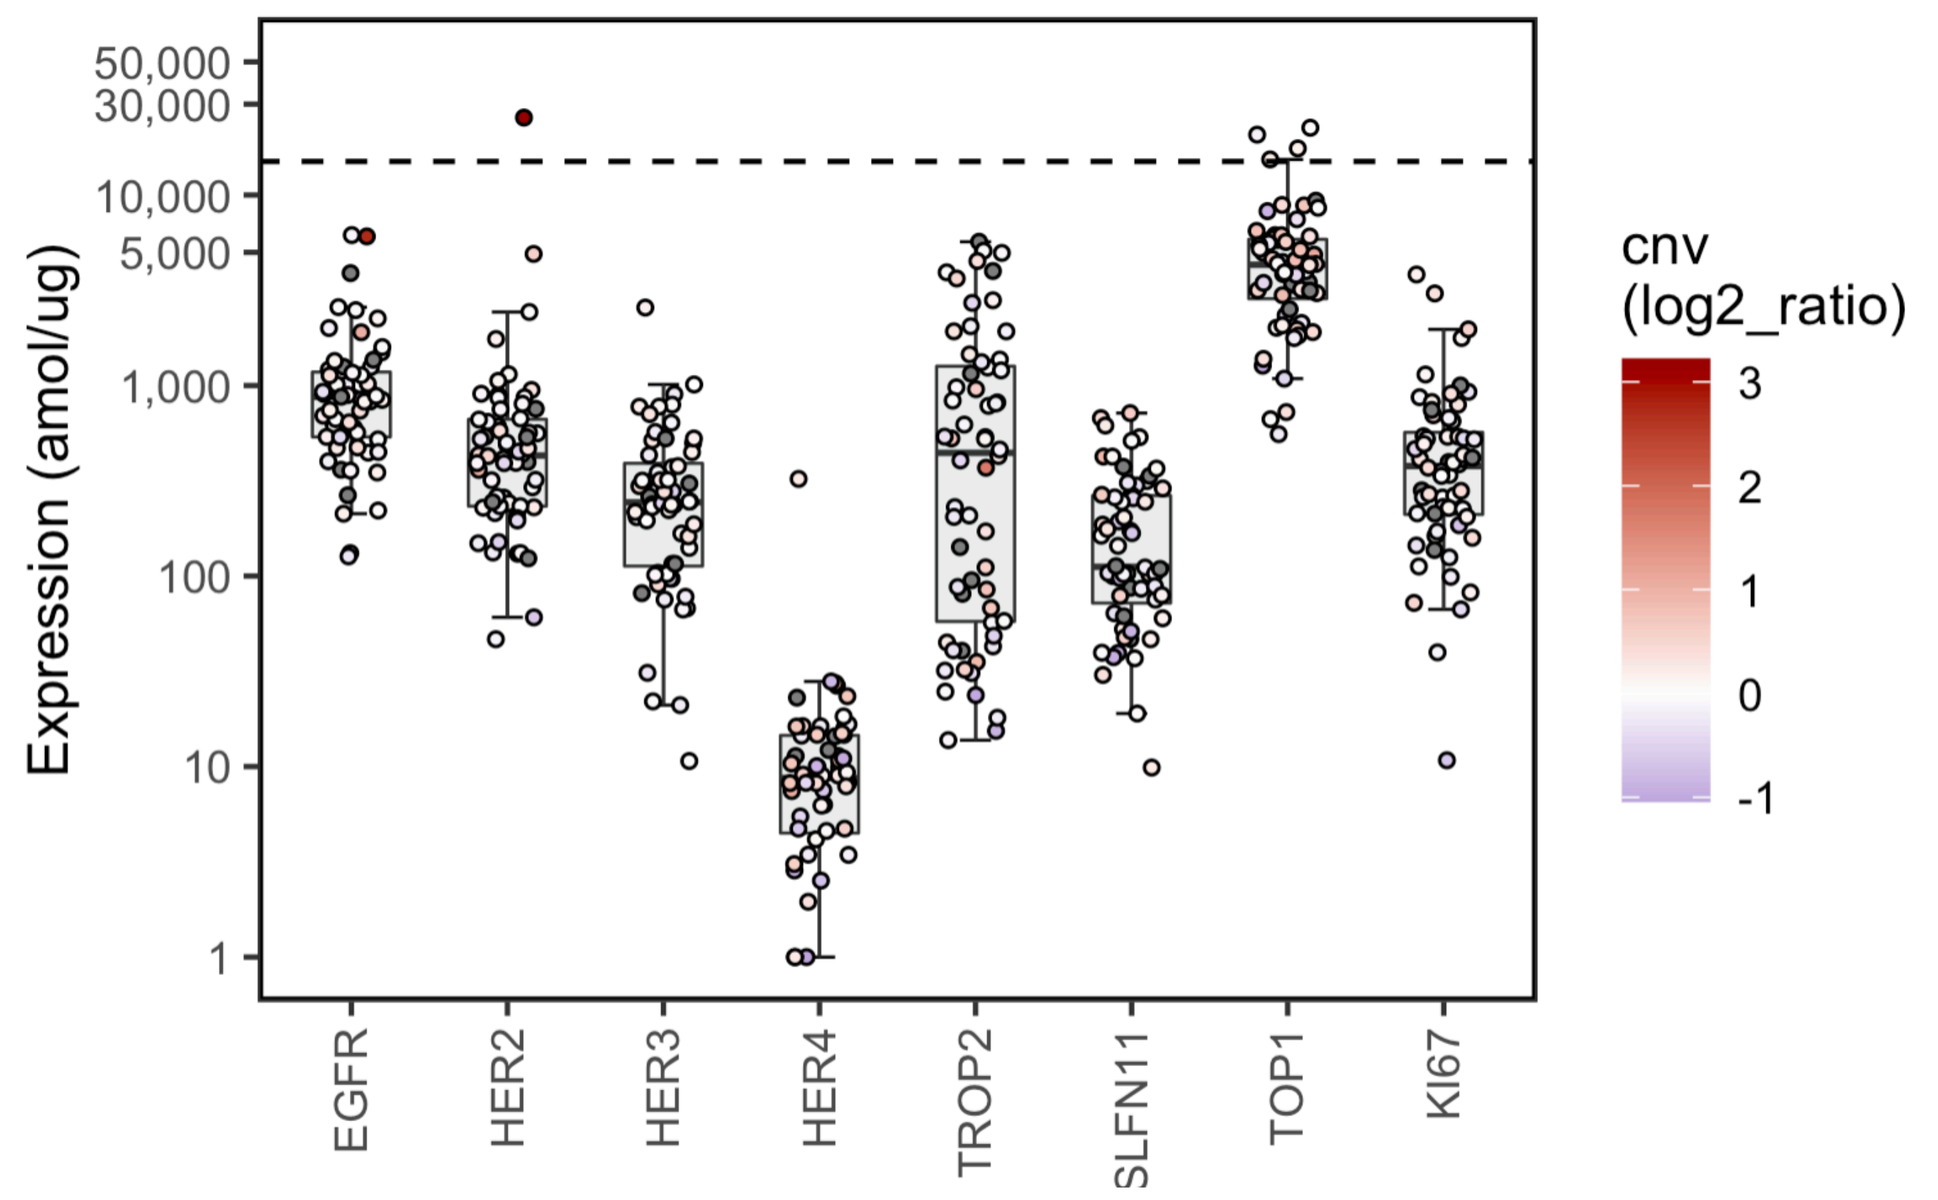
Expression (amol/ug)
EGFR
HER2
HER3
HER4
TROP2
SLFN11
TOP1
KI67
cnv
(log2_ratio)

Fig 3. Multiplexed proteomics allows the quantification of hundreds of proteins among multiple samples

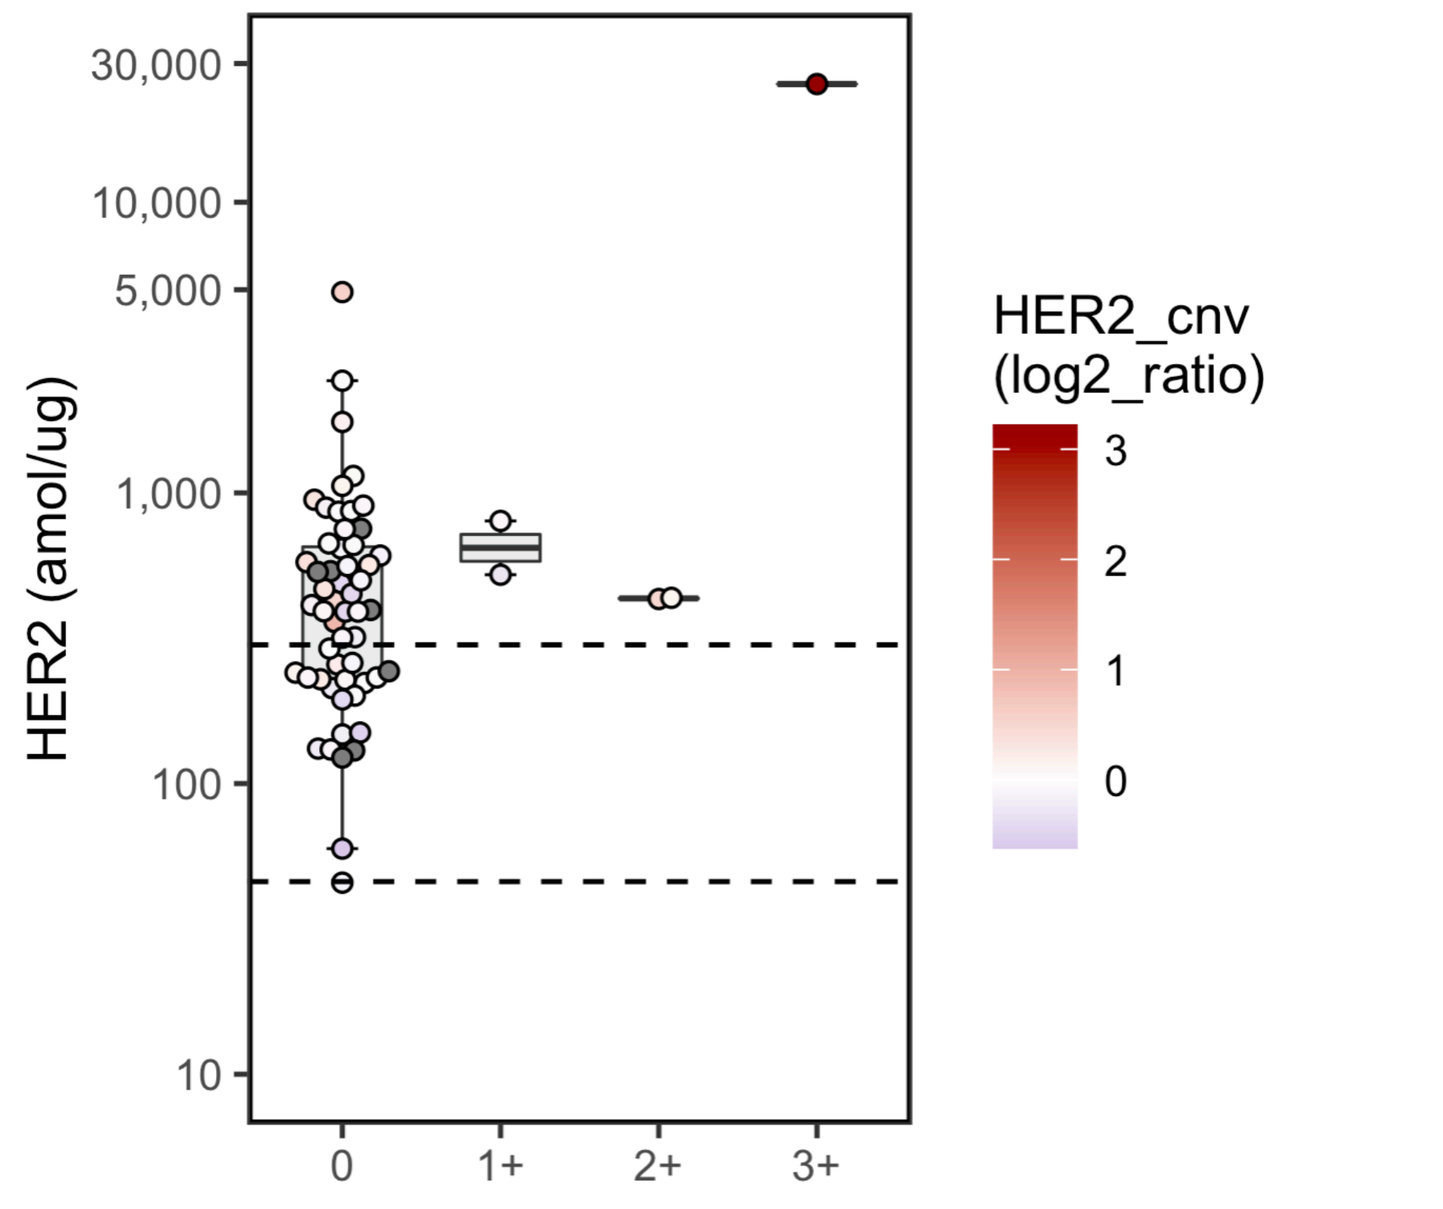
HER2 (amol/ug)
0
1+
2+
3+
HER2_cnv
(log2_ratio)

Fig 4. HER2 protein expression showed broad dynamic range across HER2 IHC scores

Results and interpretation

HER2 IHC analysis was conducted on a population that accurately reflected the overall characteristics of the entire HORIZON III study population. However, due to the absence of archival samples, genomic and proteomic analyses were only feasible in a restricted number of patients.

HER2 immunohistochemistry (IHC) expression (n=432)

- Most samples (~97.9%) showed no HER2 staining, while ~1.8% were HER2-positive (IHC 3+ or IHC 2+/ISH+) (**Table 1** and **Figure 1A**)
- Compared with IHC 3+ tumors, IHC 2+ tumors showed greater heterogeneity, with mixed components of cells weakly expressing HER2 and lacking HER2 expression (**Figure 1B)**
 - When plotted in order of the percentage of tumors cells with an IHC score of 0, tumors classified as IHC 1+ and IHC 2+ demonstrated overlap in histoscore categorization, while the histoscore algorithm identified IHC 3+ tumors as a distinct subpopulation

Molecular Characterization (n=135)

- Data revealed a concurrence between samples displaying IHC scores of 3 and those confirmed to have HER2 amplifications. HER2 was amplified in HER2 IHC 3+ patients; HER2 amplifications don't have mutations in RAS or BRAF, however insufficient sample size leads to inconclusive results (**Figure 2**)
- Most samples demonstrated microsatellite stability, and all HER2-positive patients fell within this category (insufficient sample size leads to inconclusive results)
- KRAS mutations are highly prevalent (46% mutated KRAS). A distinguished pattern of mutual exclusivity among KRAS, NRAS, and BRAF mutations is evident

Quantitative Multiplexed Proteomics (n= 60)

- Multiplexed proteomics allows the quantification of hundreds of proteins among multiple samples (**Figure 3**):
 - Most of the protein expression levels span a range, up to 4 orders of magnitude
 - Nearly all the evaluable samples had detectable HER2. HER2 IHC 3+ sample showed the highest protein expression at 30,000 amol/ug (equivalent to ~ 2M receptors/cell)
 - Response and resistance determinants to cytotoxic payload activity are heterogeneously expressed (TOP1 and SLFN11)
- Within the HER2-negative patients, HER2 protein expression showed broad dynamic range (**Figure 4**).

References

1. Cecchi F. et al. *J Clin Oncol*. 2023;41:194–194
2. Yoshino T. et. al. *Nat Commun*. 2023;14(1):3332
3. Siena S. et al. *Lancet Oncol*. 2021;22(6):779–789
4. Raghav K. et al. *J Clin Oncol*. 2023;41:3501–3501
5. Schmoll H-J. et al. *J Clin Oncol*. 2012;30:3588–3595
6. Bartley AN. et al. *Arch Pathol Lab Med*. 2016;140(12):1345–1363
7. Cerami E. et al. *Cancer Discov*. 2012;2(5):401–4
8. Gao J. et al. *Sci Signal*. 2013;6(269):pl1

Acknowledgments

We thank the patients who participated in this study as well as their families and caregivers.

This study is sponsored by AstraZeneca. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201).

Disclosures

Poster authors are employee of AstraZeneca and owns stock or other interests in that company.

Abbreviations

AACR, American Association for Cancer Research; FFPE, formalin fixed, paraffin embedded; FOLFOX, leucovorin, fluorouracil, and oxaliplatin; EGFR, epidermal growth factor receptor, HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; ISH, in situ hybridization; KRAS, KRAS proto-oncogene; RAF, RAF proto-oncogene; RAS, Ras-like protein. MSS microsatellite stable MSI microsatellite instable, TMB tumor mutational burden