Poster 1366

Dual immune checkpoint blockade enhances the anti-tumor activity of trastuzumab deruxtecan in preclinical models

Liam Jenkins[1], **Laura Kazlauskas**[1], Matt Wilson[2], Scott A. Hammond[3], Theresa Proia[1], Jerome Mettetal[1]

[1]AstraZeneca, Waltham, MA, [2]AstraZeneca, Cambridge, United Kingdom, [3]AstraZeneca, Gaithersburg, MD

Objective

- To evaluate the immunomodulatory effects of trastuzumab deruxtecan (T-DXd) and determine the preclinical anti-tumor activity of T-DXd in combination with dual immune checkpoint blockade (ICB).

Conclusions

- T-DXd induced ATP secretion, NKG2D ligand expression, and immune cell activation in vitro.
- In a hHER2 expressing EMT6 tumor model, the anti-tumor activity of T-DXd was enhanced when combined with inhibitors of PD-L1 plus CTLA-4, or a monovalent bispecific aPD-1/TIGIT antibody (a murine surrogate for rilvegostomig/AZD2936).
- In a humanised Caki-1 model, the anti-tumor activity of T-DXd was enhanced when combined with volrustomig/MEDI5752 – a novel aPD-1/CTLA-4 monovalent bispecific antibody.
- These data provide scientific rationale for combining T-DXd with immuno-oncology (IO) agents targeting CTLA-4 or TIGIT, in addition to the PD-1/PD-L1 axis.

Plain language summary

Why did we perform this research?
To understand whether a drug called trastuzumab deruxtecan can help stimulate the immune system to attack tumor cells, and whether it might work well with other anticancer therapies known to act on the immune system.

How did we perform this research?
Genetically engineered cells and mice were treated with trastuzumab deruxtecan, either alone, or in combination with other therapies. Similar combinations were also tested in mice engrafted with human immune cells. These combinations were then tested for their ability to stimulate the immune system, kill cancer cells and shrink tumors.

What were the findings of this research?
Trastuzumab deruxtecan helped cancer-fighting immune cells to infiltrate tumors, and increased the effectiveness of novel anticancer treatments that stimulate the immune system.

What are the implications of this research?
These laboratory results may inform future clinical studies investigating trastuzumab deruxtecan in combination with next-generation immunotherapies, potentially driving deeper and more durable responses.

Poster presented at AACR Annual Meeting 2024, April 5-10, 2024, San Diego Convention Center, San Diego, CA, USA.

Corresponding author email address: liam.jenkins1@astrazeneca.com and laura.kazlauskas@astrazeneca.com

Introduction

- Trastuzumab deruxtecan (T-DXd) is an antibody-drug conjugate (ADC) composed of an anti-HER2 antibody, a cleavable tetrapeptide-based linker, and a topoisomerase I inhibitor payload[1-4].
- Clinical trials have highlighted the potential benefit in combining T-DXd with immuno-oncology (IO) therapeutics, such as durvalumab (aPD-L1)[5].
- Monovalent bispecific antibodies, such as volrustomig/MEDI5752 (aPD-1/CTLA-4) and rilvegostomig/AZD2936 (aPD-1/TIGIT) are currently in Ph III clinical trials[6,7].

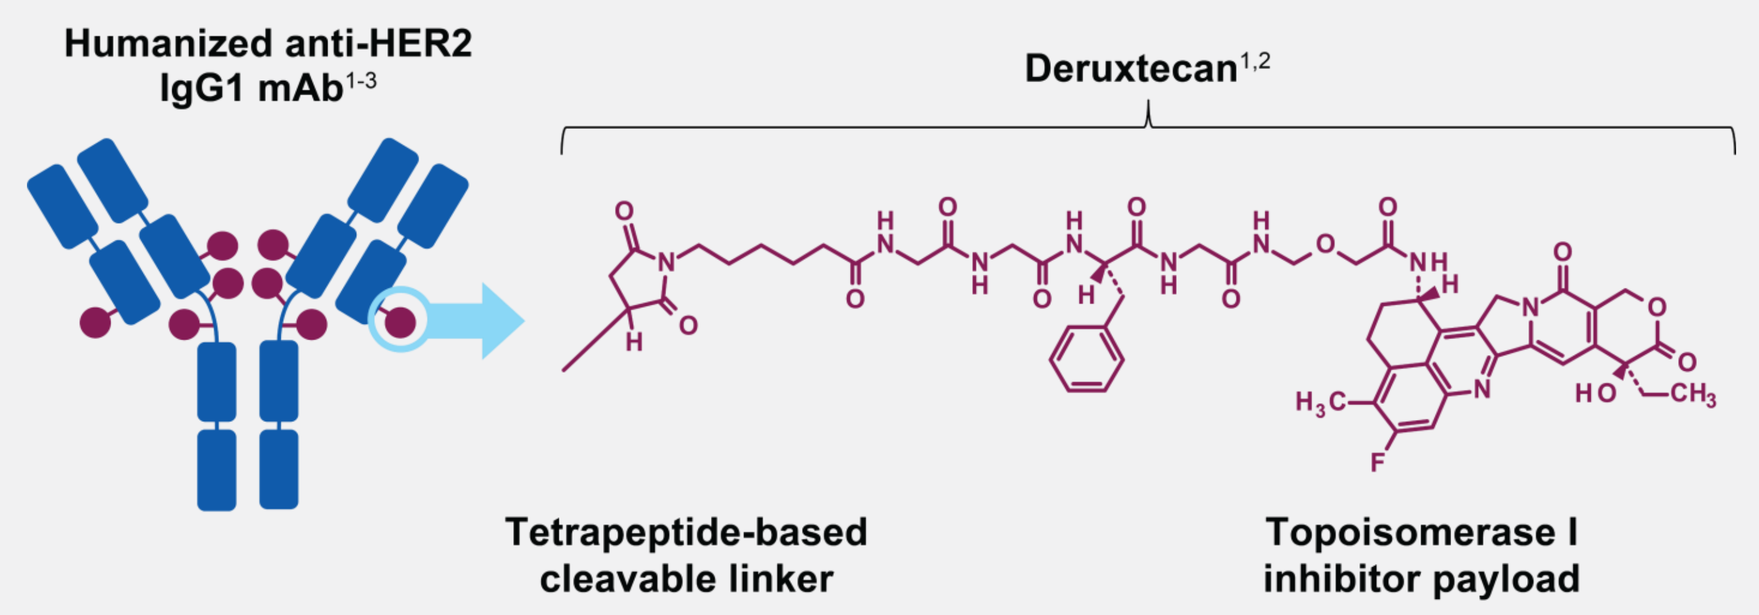
Humanized anti-HER2 IgG1 mAb1-3
Deruxtecan1,2
Tetrapeptide-based cleavable linker
Topoisomerase I inhibitor payload

Methods

In vitro

- 5 human cell lines (NCI-N87, KPL-4, SK-BR-3, HCC-1954, MDA-MB-157) were treated with nab-paclitaxel, doxorubicin, or deruxtecan and assessed for secretion of ATP, and expression of NKG2D ligands.
- Supernatants from treated cells were added to isolated human immune cells, and their activation status assessed via flow cytometry.

In vivo studies in an immunocompetent hHER2-expressing EMT6 tumor model

- BALB/c mice bearing hHER2-expressing murine EMT6 tumors were treated with T-DXd ± aPD-L1, aPD-L1 plus aCTLA-4, or a murine surrogate monovalent bispecific aPD-1/TIGIT antibody. Anti-tumor efficacy or tumoral pharmacodynamic changes were evaluated[8].

In vivo efficacy in a humanized Caki-1 mouse model

- Immunocompromised NSG mice were engrafted with hematopoietic stem cells (HSCs) derived from human cord blood, and implanted with the human renal cell carcinoma line, Caki-1.
- huCaki-1 tumor-bearing mice were treated with T-DXd ± volrustomig (a monovalent bispecific antibody targeting PD-1 and CTLA-4). Anti-tumor efficacy and peripheral blood pharmacodynamic changes were evaluated[8].

Results and interpretation

DXd activates immune cells in vitro

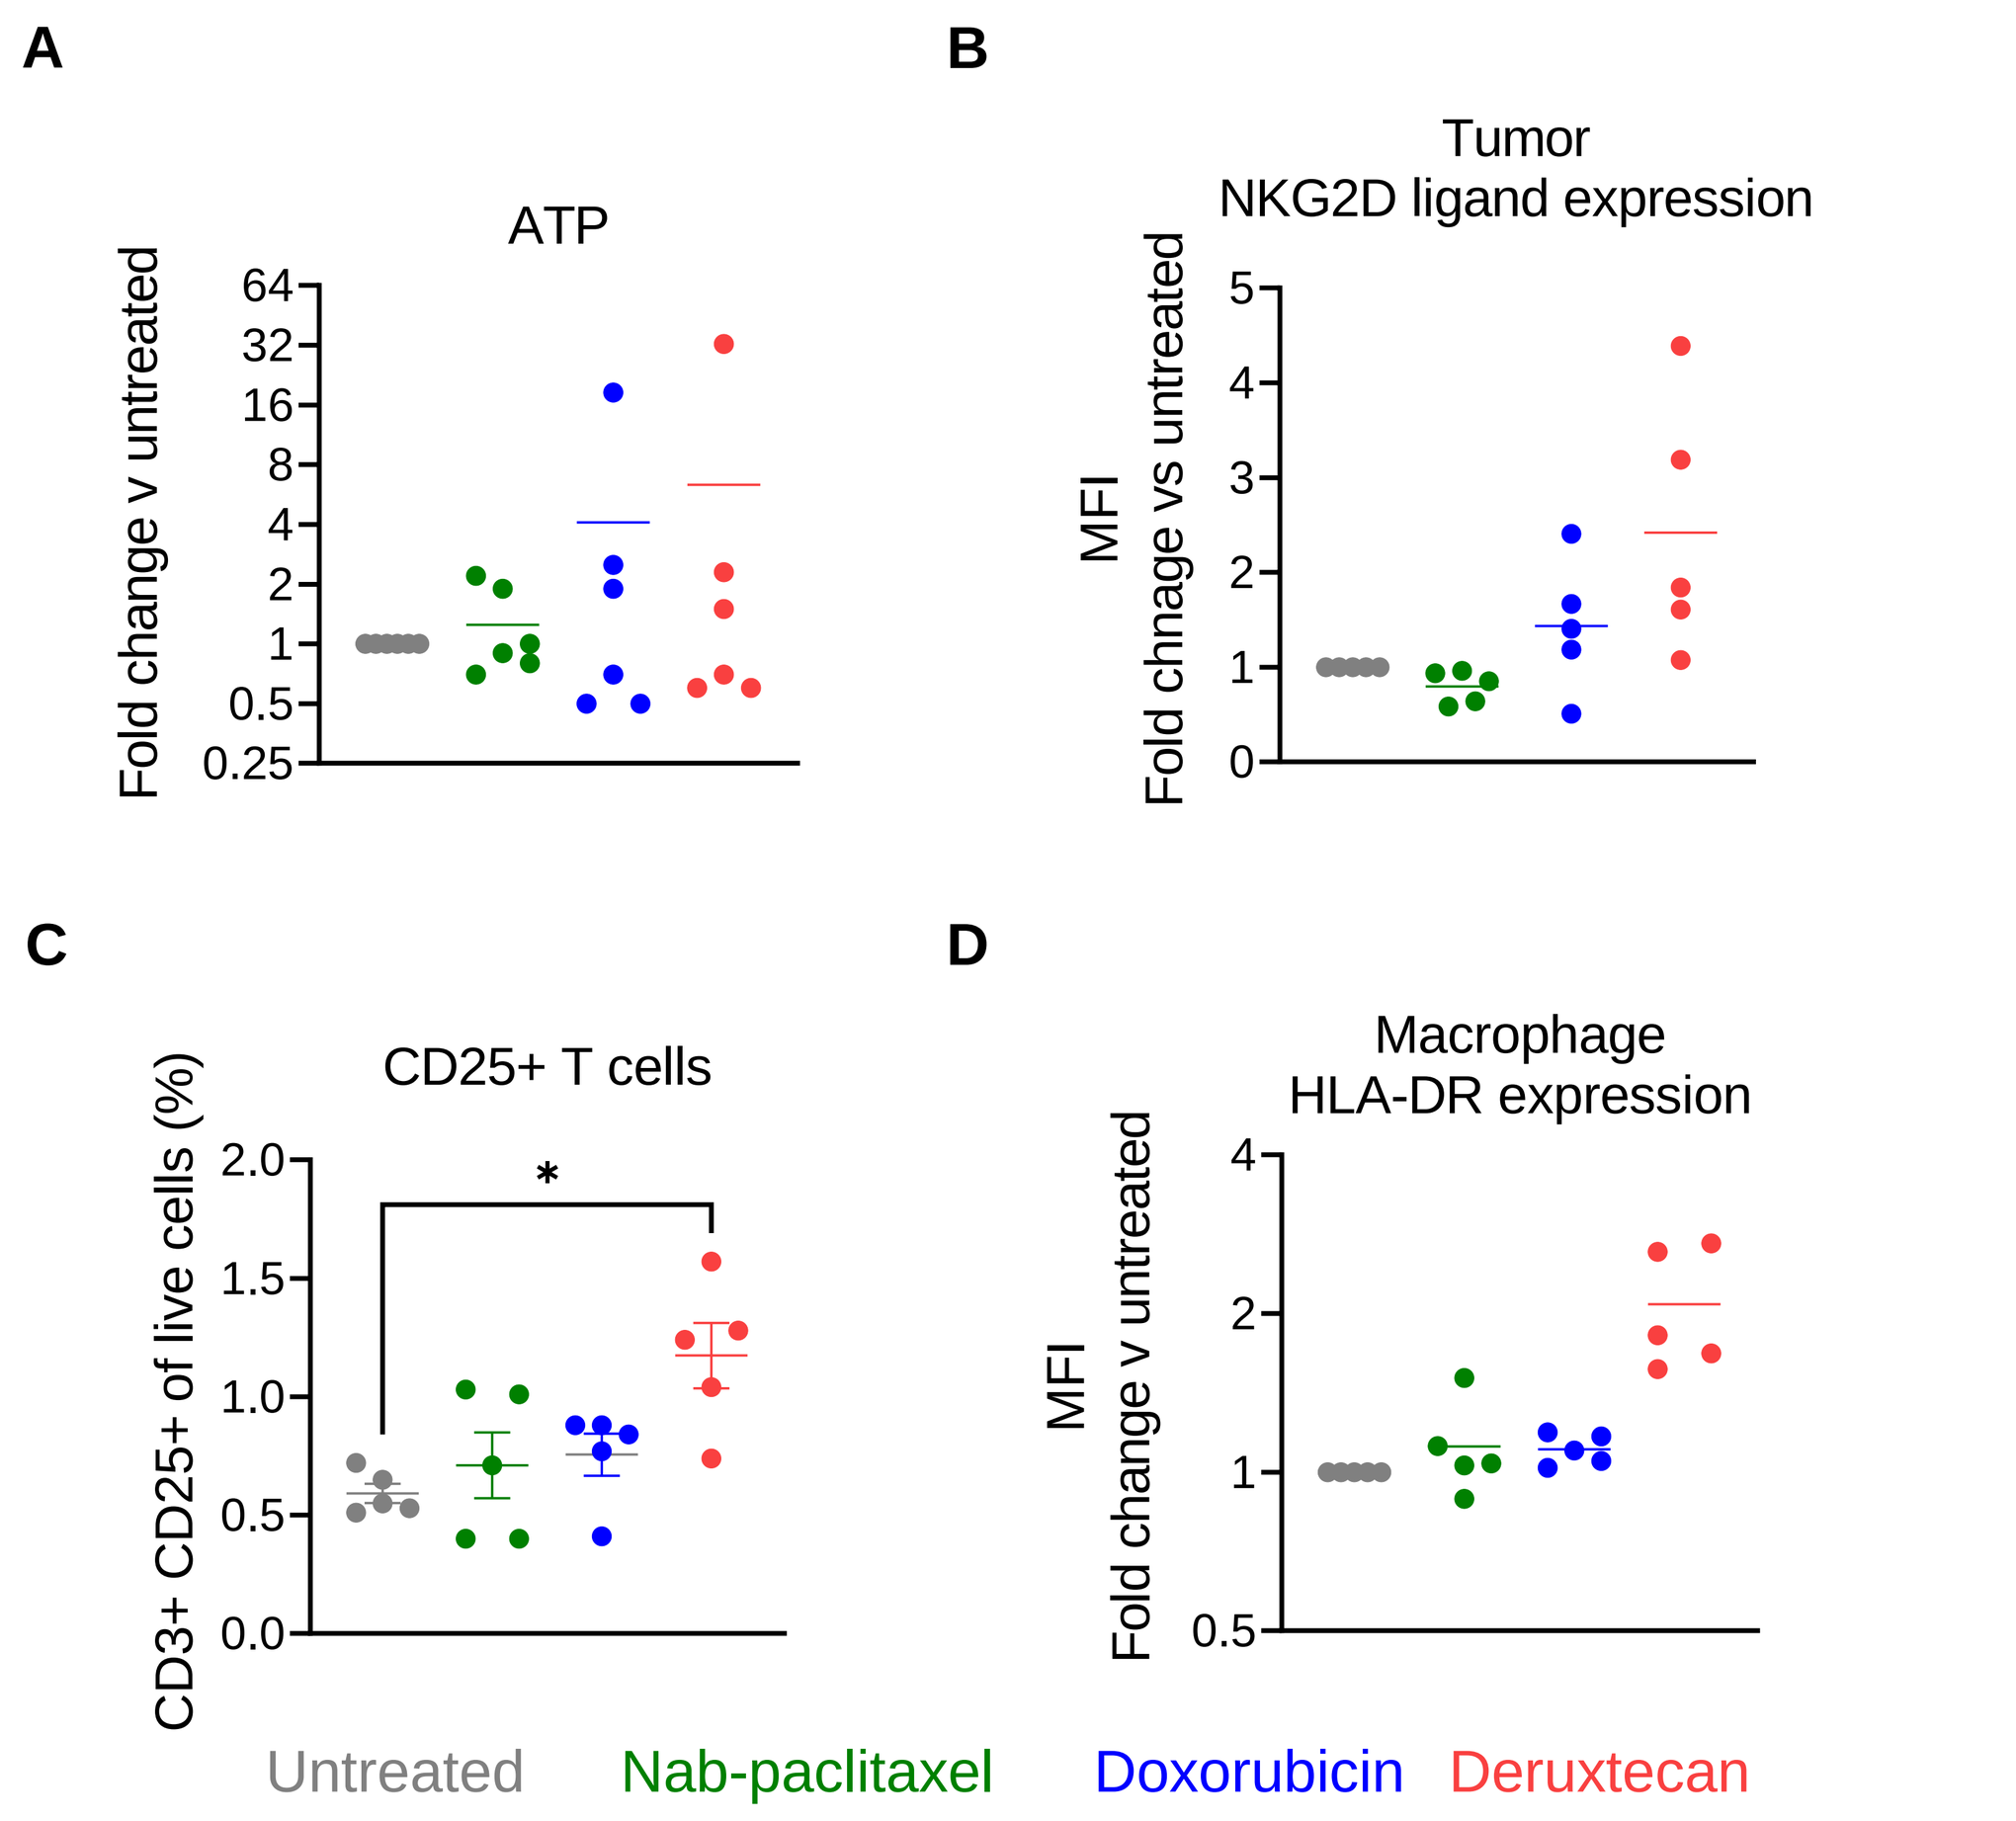
A
ATP
Fold change v untreated
B
Tumor NKG2D ligand expression
MFI Fold chnage vs untreated
C
CD25+ T cells
CD3+ CD25+ of live cells (%)
D
Macrophage HLA-DR expression
MFI Fold change v untreated
Untreated
Nab-paclitaxel
Doxorubicin
Deruxtecan

Figure 1. T-DXd induces ATP secretion, NKG2D ligand expression, and activates immune cells. A panel of 5 human cell lines (NCI-N87, KPL-4, SK-BR-3, HCC-1954, and MDA-MB-157) were treated in vitro with nab-paclitaxel, doxorubicin, or deruxtecan at concentrations driving 100% growth rate inhibition (GRI100) for 3 days. (**A**) Tumor cell secretion of ATP was assessed via luminometry. (**B**) Treated tumor cells were assessed for expression of NKG2D ligands via flow cytometry. (**C**) Supernatants from treated tumor cells were added to human T cells. T cell expression of CD25 was assessed via flow cytometry. (**D**) Supernatants from treated tumor cells were added to CD14+ macrophages isolated from human PBMCs. Macrophage HLA-DR expression was assessed via flow cytometry. *, P <0.05. Paired t-test

T-DXd alters tumor-infiltrating lymphocyte profile

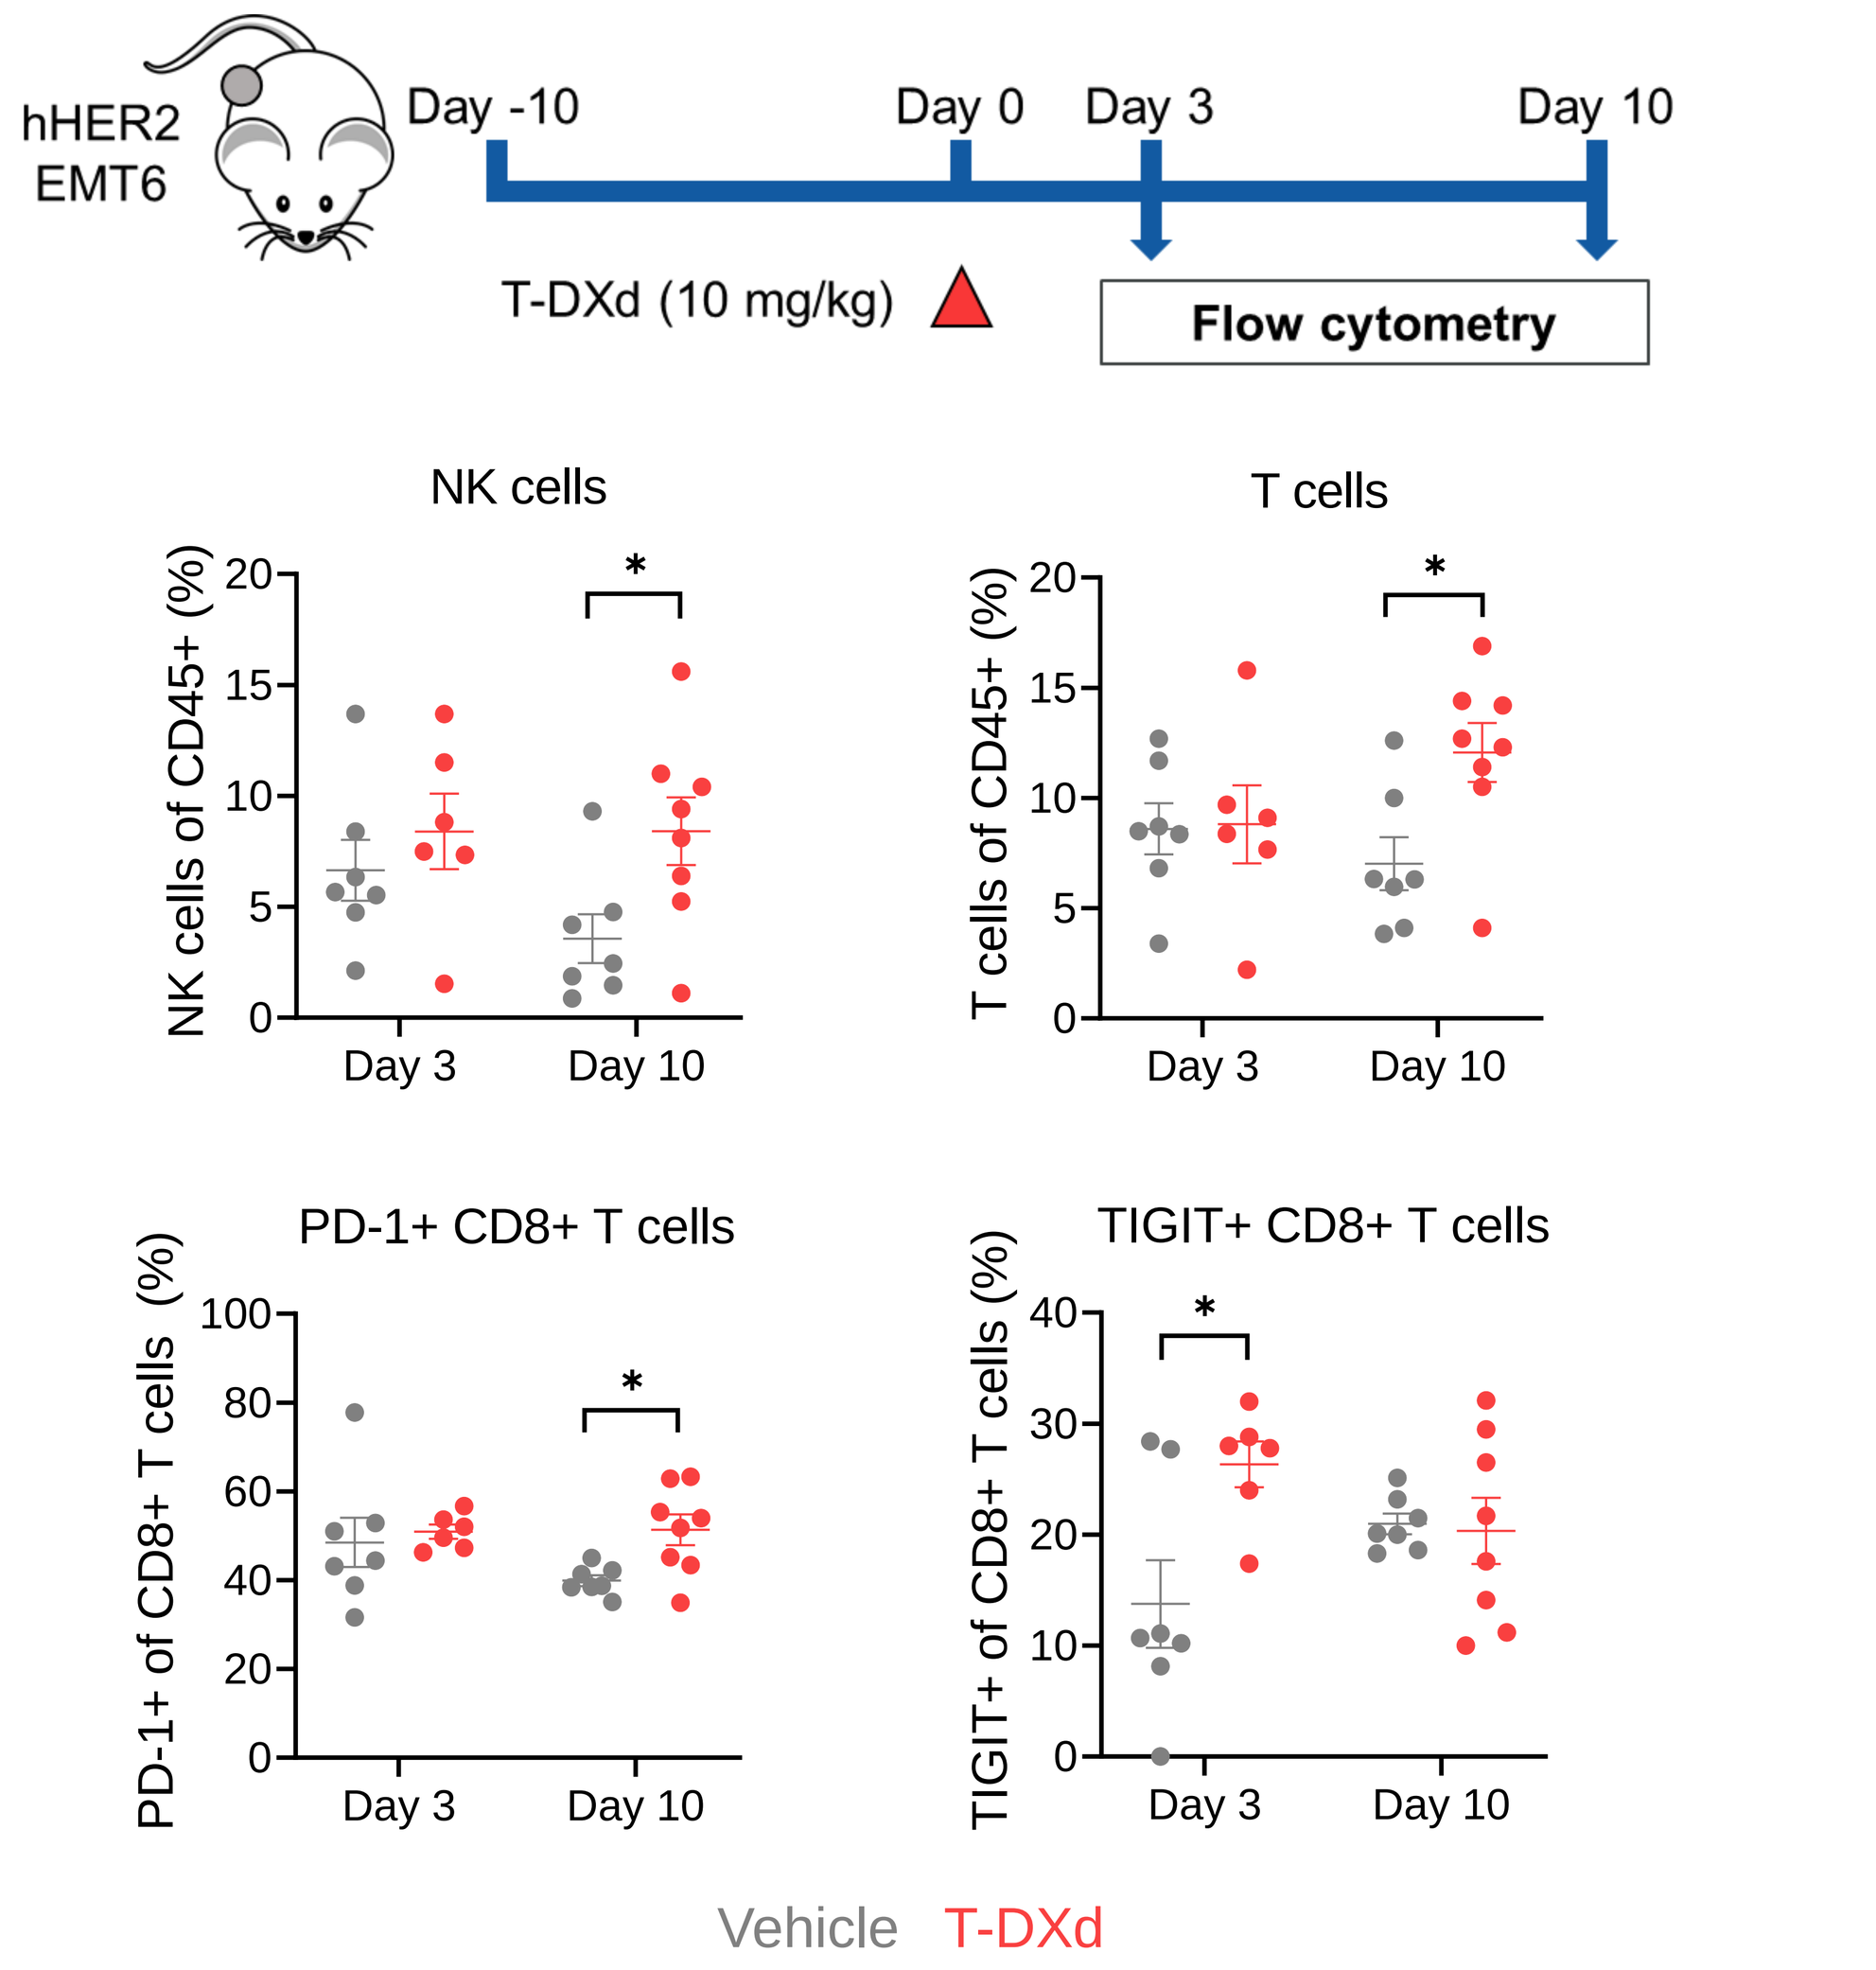
hHER2 EMT6
Day -10
Day 0
Day 3
Day 10
T-DXd (10 mg/kg)
Flow cytometry
NK cells
NK cells of CD45+ (%)
T cells
T cells of CD45+ (%)
PD-1+ CD8+ T cells
PD-1+ of CD8+ T cells (%)
TIGIT+ CD8+ T cells
TIGIT+ of CD8+ T cells (%)
Day 3
Day 10
Vehicle
T-DXd

Figure 2. T-DXd increases tumoral abundance of NK cells (CD3- NKp46+), T cells (CD3+), and PD-1+, and TIGIT+ CD8+ T cells. 5x10^6 EMT6 hHER2 cells were implanted subcutaneously into the flank of BALB/c mice and treated with 10 mg/kg T-DXd. Tumors were collected on days 3 and 10 post treatment and their immune content (CD45+ cells) profiled via flow cytometry (n=6-8).*, P <0.05. Unpaired t-test.

Enhanced anti-tumor activity with aPD-L1 + aCTLA-4 + T-DXd

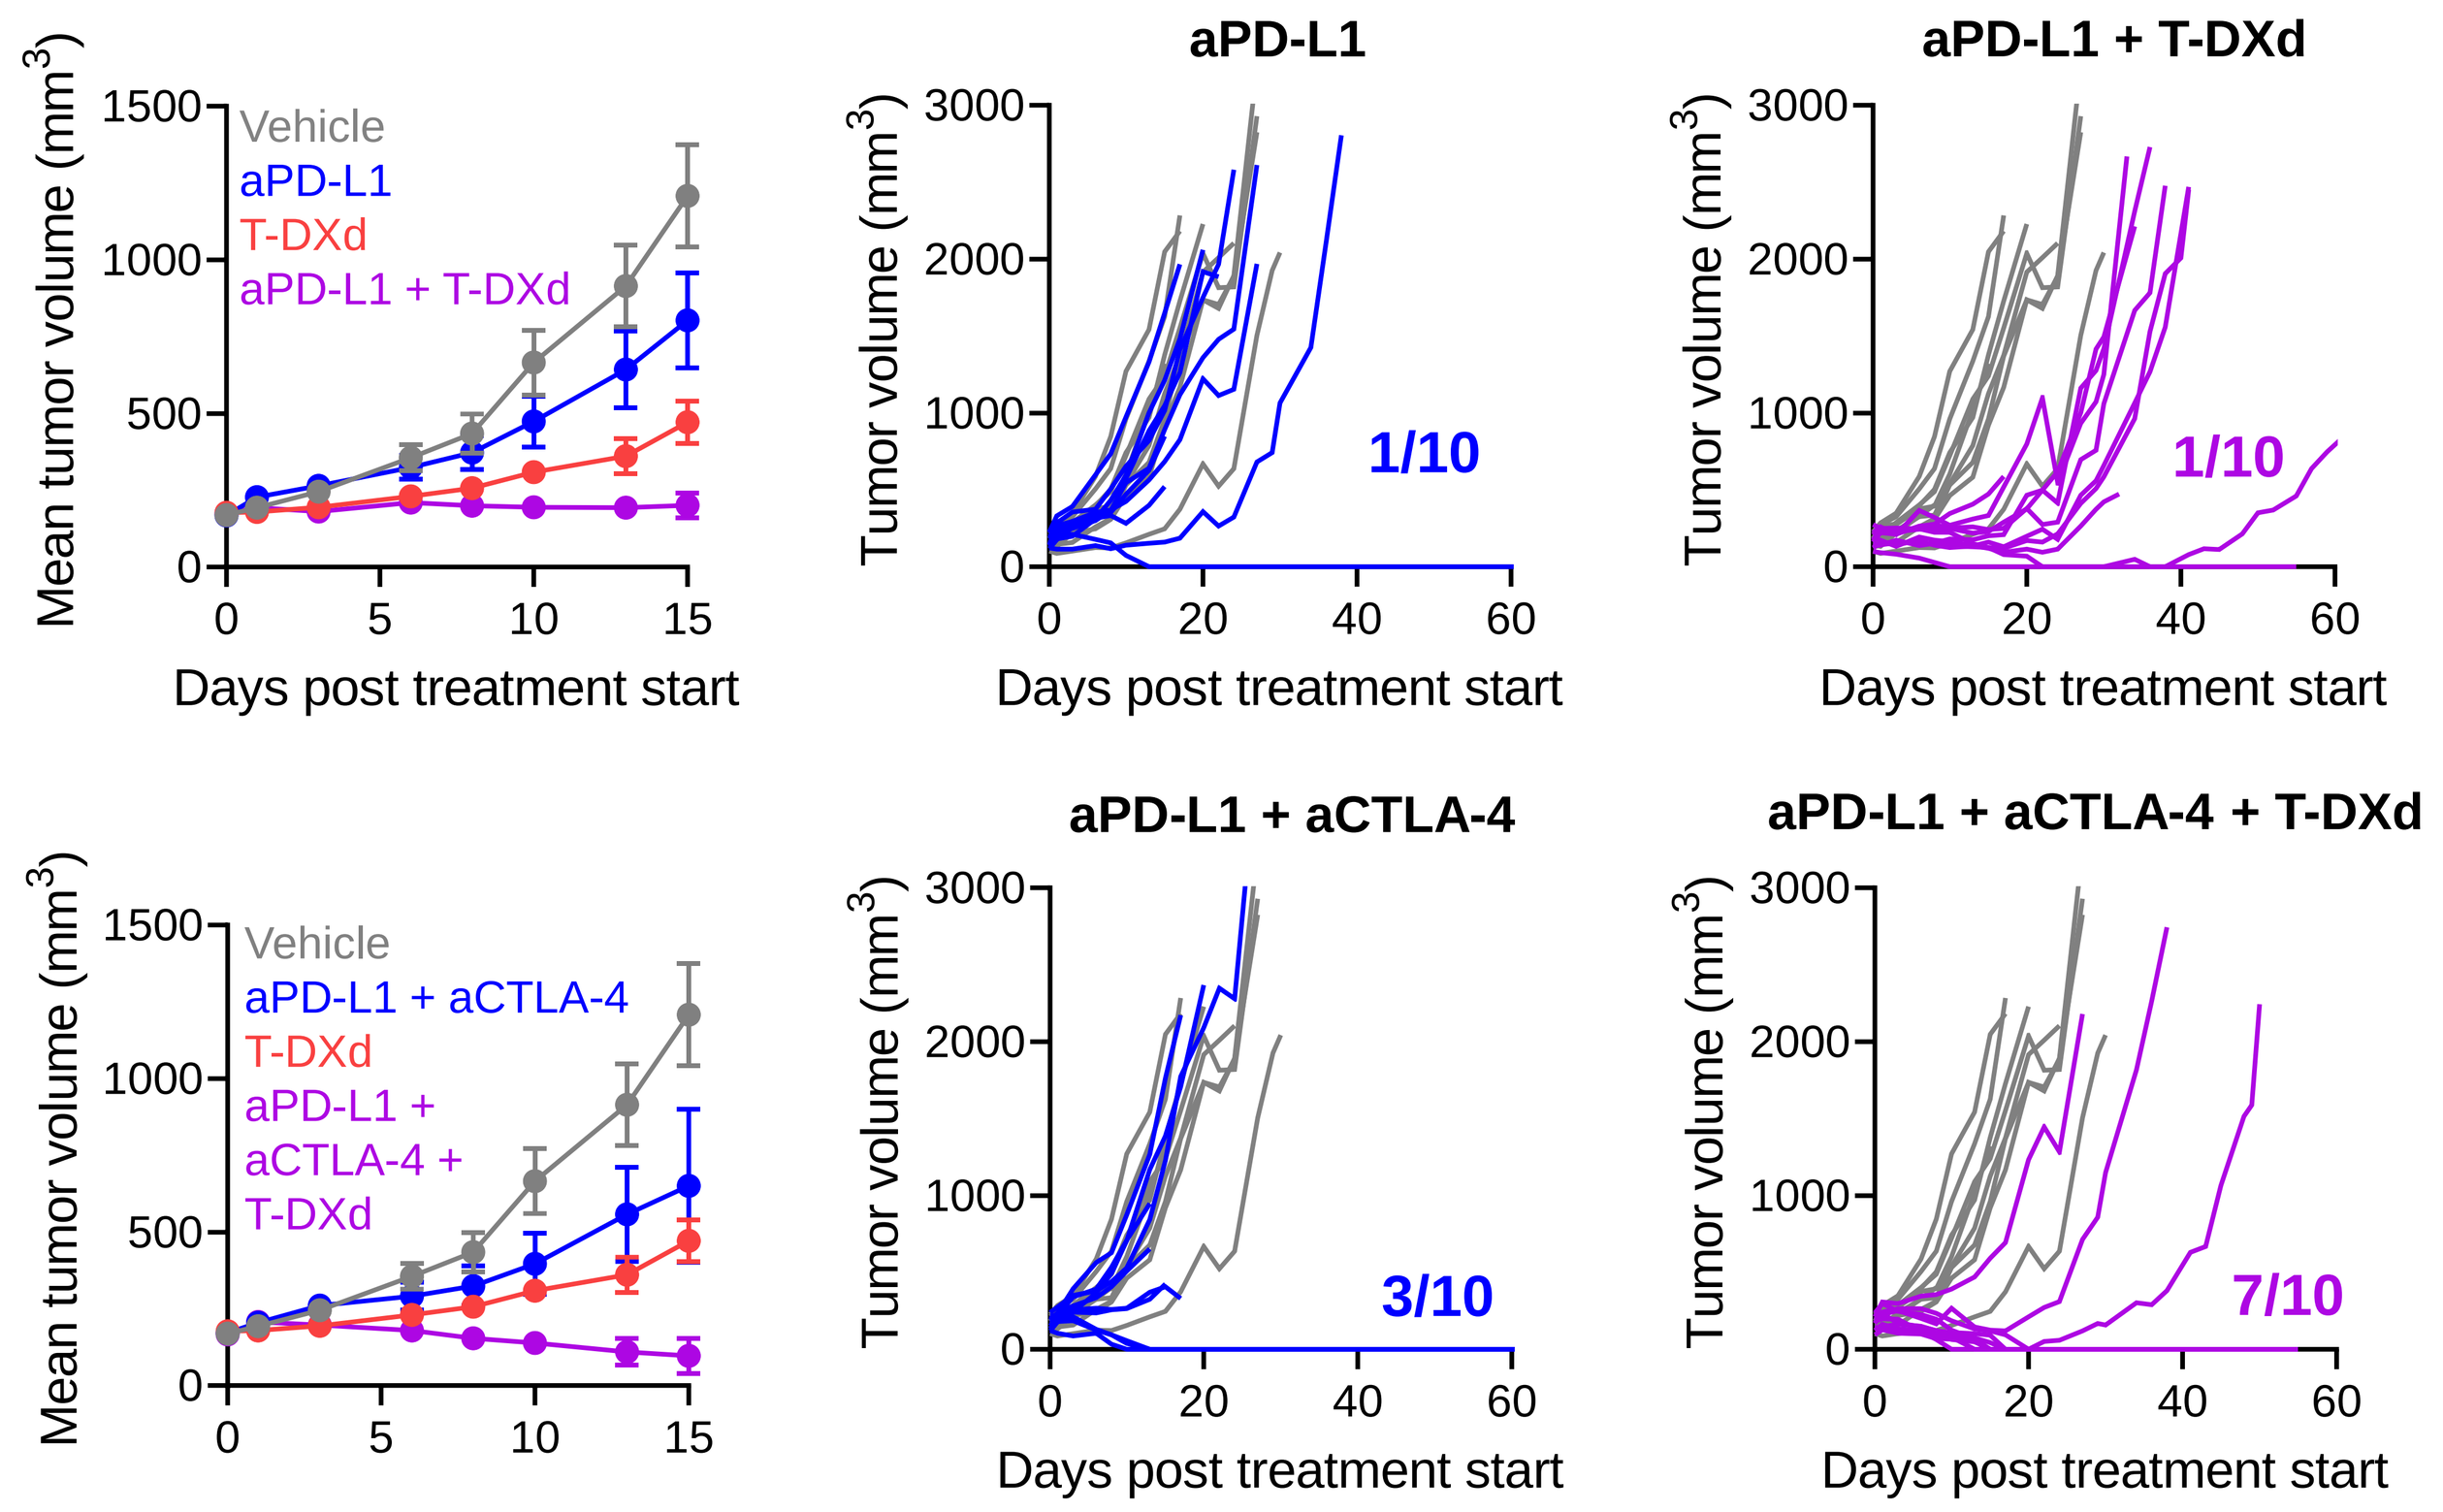
Mean tumor volume (mm3)
Vehicle
aPD-L1
T-DXd
aPD-L1 + T-DXd
aPD-L1
1/10
aPD-L1 + T-DXd
1/10
Vehicle
aPD-L1 + aCTLA-4
T-DXd
aPD-L1 + aCTLA-4 + T-DXd
aPD-L1 + aCTLA-4
3/10
aPD-L1 + aCTLA-4 + T-DXd
7/10
Tumor volume (mm3)
Days post treatment start

Figure 3. T-DXd enhances the anti-tumor activity of combination aPD-L1 + aCTLA-4 antibody treatment in a syngeneic hHER2 EMT6 model. 5x10^6 hHER2 EMT6 cells were implanted subcutaneously into the flank of BALB/c mice (n=10). Treatment was initiated 7 days later. T-DXd (10 mg/kg) was administered IV on treatment days 0 and 7. IO agents (10 mg/kg) were administered IP on treatment days 0, 3, 7 & 10. Complete response rates (CR) are shown.

Table 1. Growth rate inhibition (aPD-L1 + T-DXd)

Treatment	Dose (mg/kg)	GRI (%)	P value	CR
aPD-L1	10	46.2	< 0.05	1/10
T-DXd	10	53.2	< 0.01	0/10
aPD-L1 T-DXd	10 10	108.2	< 0.001	1/10

GRI: growth rate inhibition versus vehicle control[8], day 0-15 post treatment start, Mann-Whitney t-test; CR: complete response rate.

Table 2. Growth rate inhibition (aPD-L1 + aCTLA-4 + T-DXd)

Treatment	Dose (mg/kg)	GRI (%)	P value	CR
aPD-L1 aCTLA-4	10 10	86.2	< 0.05	3/10
T-DXd	10	53.2	< 0.01	0/10
aPD-L1 aCTLA-4 T-DXd	10 10 10	171.9	< 0.001	7/10

GRI: growth rate inhibition versus vehicle control[8], day 0-15 post treatment start, Mann-Whitney t-test; CR: complete response rate.

Enhanced anti-tumor activity with volrustomig + T-DXd in a huCaki-1 model

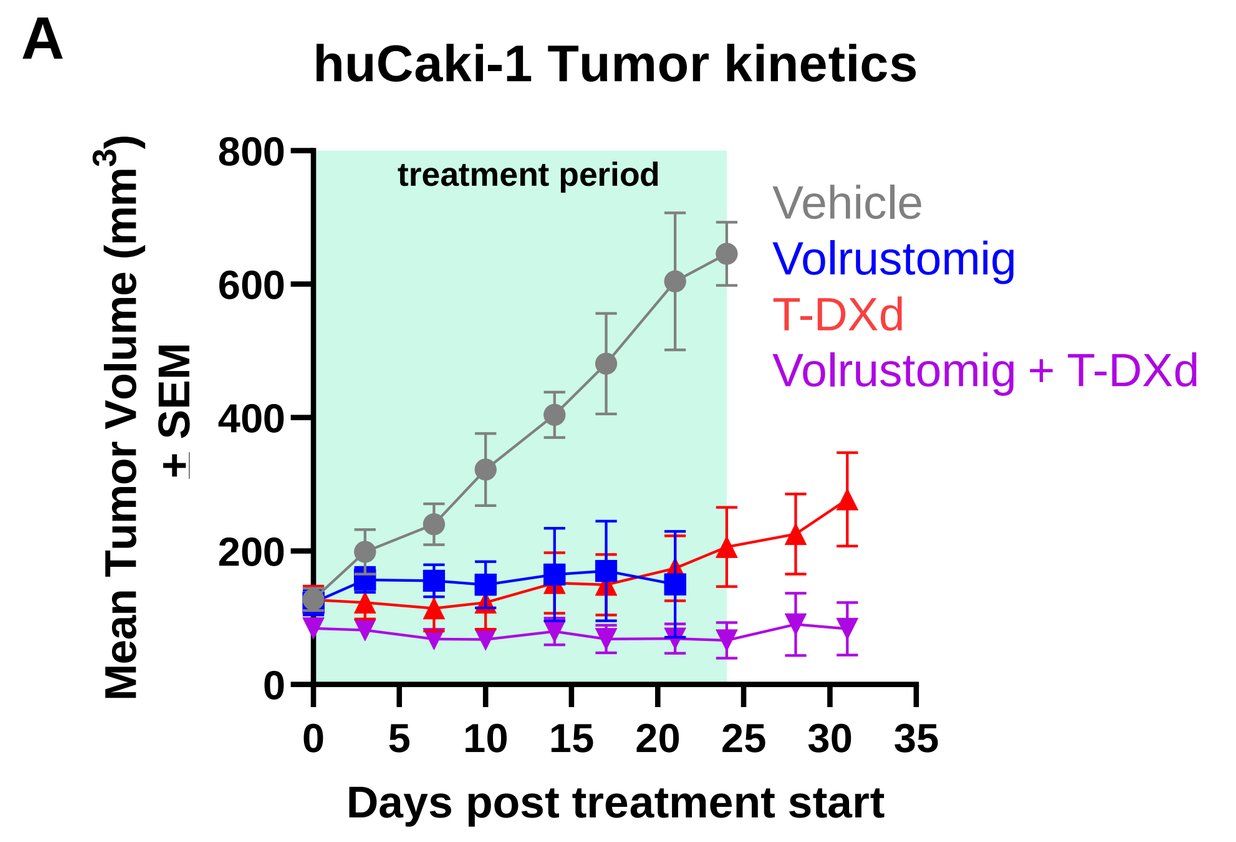
A
huCaki-1 Tumor kinetics
treatment period
Vehicle
Volrustomig
T-DXd
Volrustomig + T-DXd
Mean Tumor Volume (mm3) + SEM
Days post treatment start

Table 3. Growth rate inhibition (volrustomig + T-DXd)

Treatment	Dose (mg/kg)	GRI (%)	P value
Volrustomig	10	92.4	0.082
T-DXd	10	79.2	0.074
Volrustomig T-DXd	10 10	115.5	< 0.05

GRI: growth rate inhibition versus vehicle control[8]. Day 0-31 post treatment start. Mann-Whitney t-test.

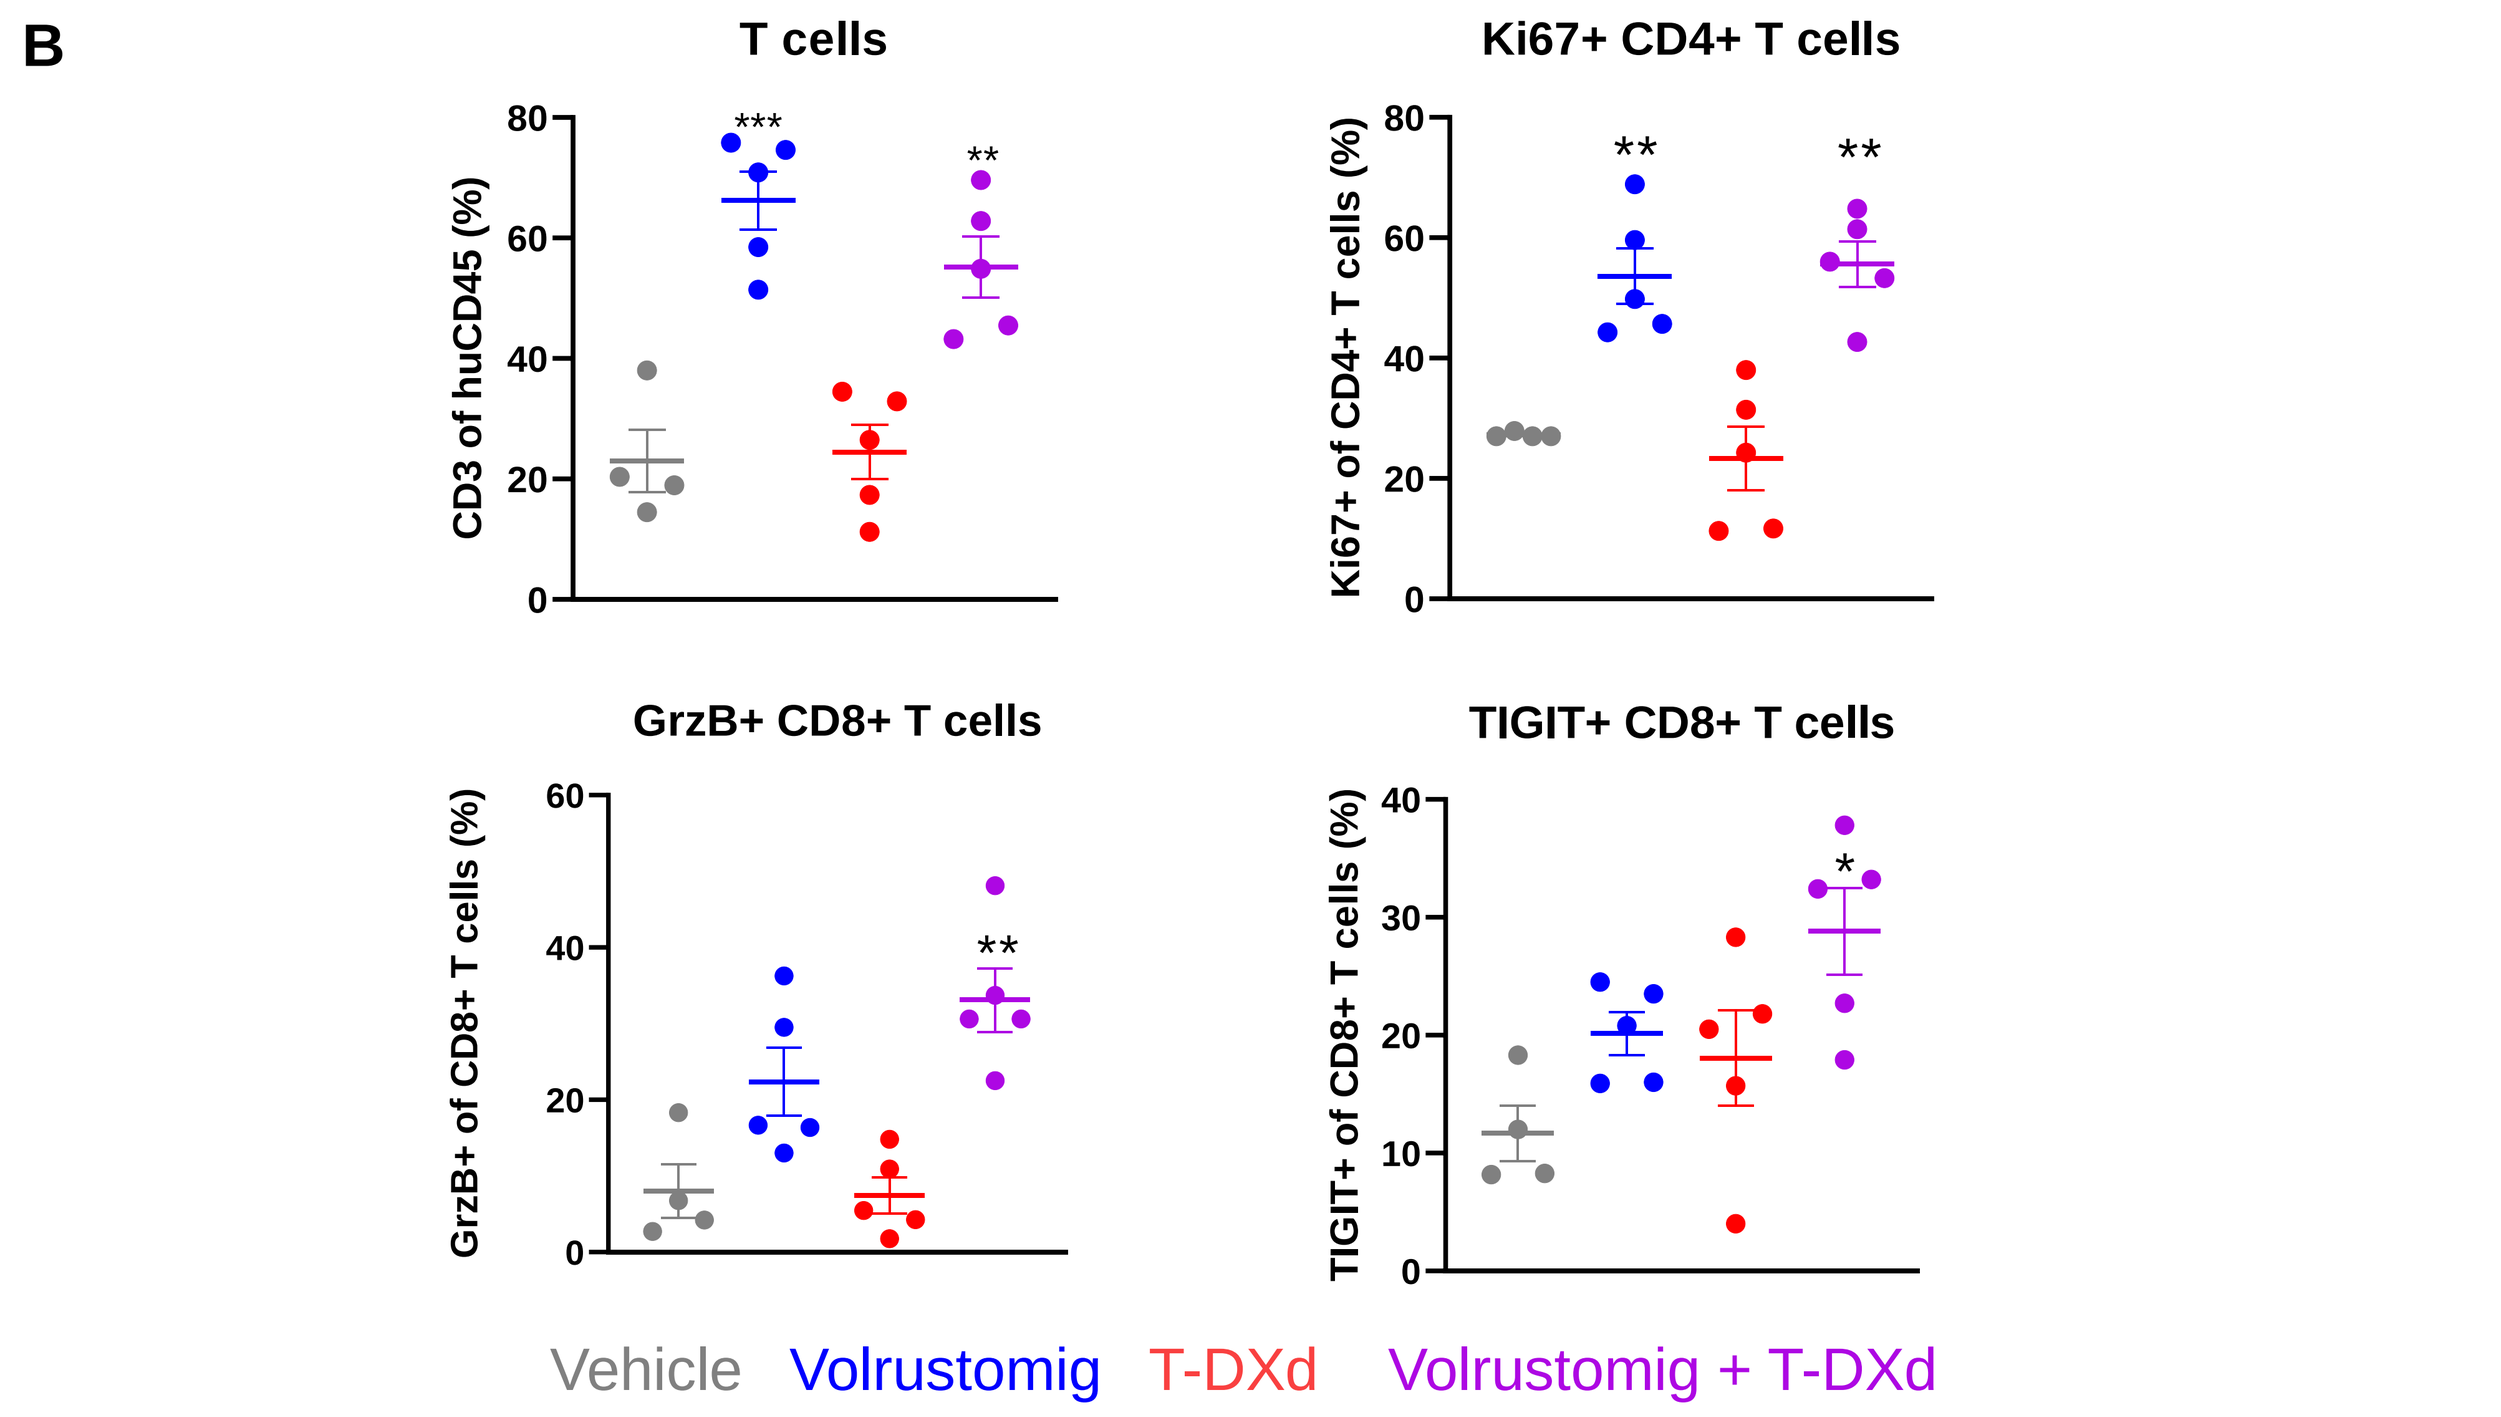
B
T cells
CD3 of huCD45 (%)
Ki67+ CD4+ T cells
Ki67+ of CD4+ T cells (%)
GrzB+ CD8+ T cells
GrzB+ of CD8+ T cells (%)
TIGIT+ CD8+ T cells
TIGIT+ of CD8+ T cells (%)
Vehicle
Volrustomig
T-DXd
Volrustomig + T-DXd

Figure 4. Enhanced durability of response when T-DXd is combined with volrustomig. (**A**) T-DXd administered IV on day 0, volrustomig dosed biweekly IP for 8 doses. (**B**) Increase in T cell abundance, proliferation (Ki67), and effector function (GrzB) versus vehicle control in peripheral blood analyzed via flow cytometry on day 10 (2 days post 3rd volrustomig dose).*, P <0.05; **, P <0.01; ***, P <0.001. One-way ANOVA with Tukey's multiple comparisons test.

Enhanced anti-tumor activity with aPD-L1/TIGIT + T-DXd

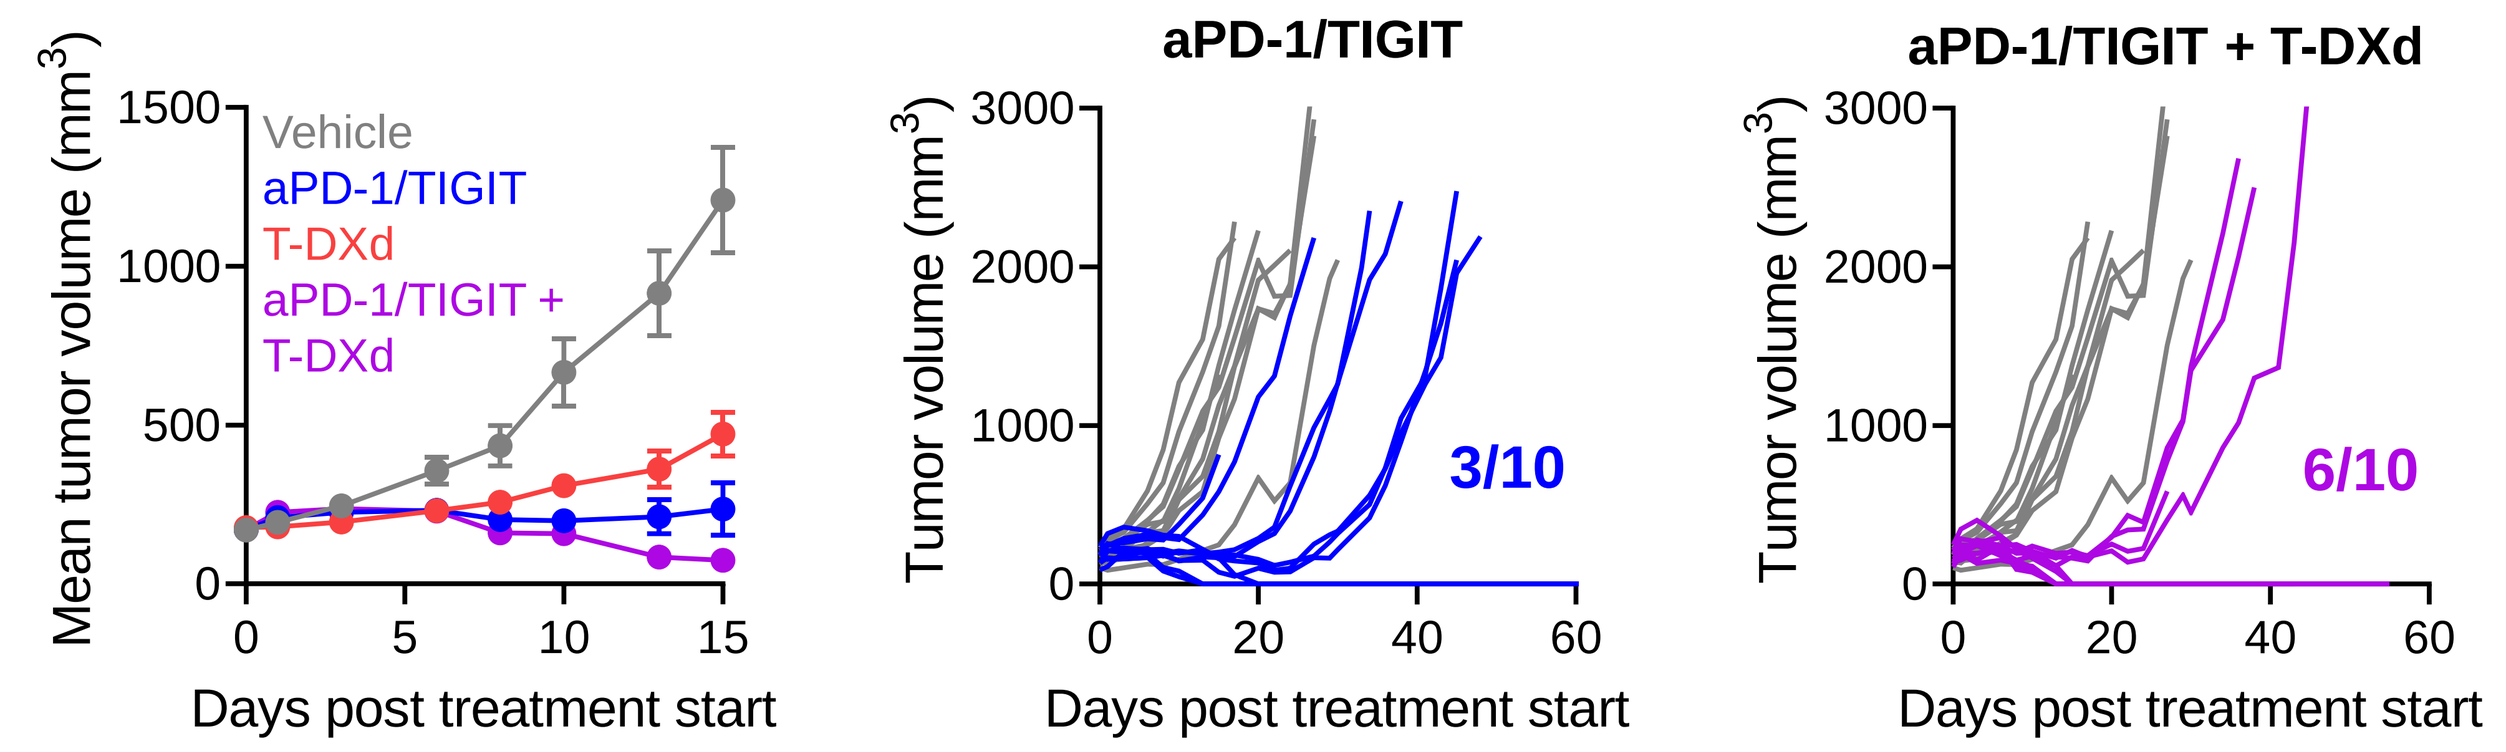
Mean tumor volume (mm3)
Vehicle
aPD-1/TIGIT
T-DXd
aPD-1/TIGIT + T-DXd
aPD-1/TIGIT
3/10
aPD-1/TIGIT + T-DXd
6/10
Tumor volume (mm3)
Days post treatment start

Figure 5. T-DXd enhances the anti-tumor activity of a monovalent bispecific aPD-1/TIGIT antibody in a syngeneic hHER2 EMT6 model. 5x10^6 hHER2 EMT6 cells were implanted subcutaneously into the flank of BALB/c mice (n=10). Treatment was initiated 7 days later. T-DXd (10 mg/kg) was administered IV on treatment days 0 and 7. aPD-1/TIGIT (10 mg/kg) was administered IP on treatment days 0, 3, 7 & 10. Complete response rates (CR) are shown.

Table 4. Growth rate inhibition (aPD-1/TIGIT + T-DXd)

Treatment	Dose (mg/kg)	GRI (%)	P value	CR
aPD-1/TIGIT	10	120.3	< 0.05	3/10
T-DXd	10	53.2	< 0.01	0/10
aPD-1/TIGIT T-DXd	10 10	181.2	< 0.001	6/10

GRI: growth rate inhibition versus vehicle control[8], day 0-15 post treatment start, Mann-Whitney t-test; CR: complete response rate.

aPD-1/TIGIT + T-DXd enhances tumoral NK cell and CD8+ T cell content

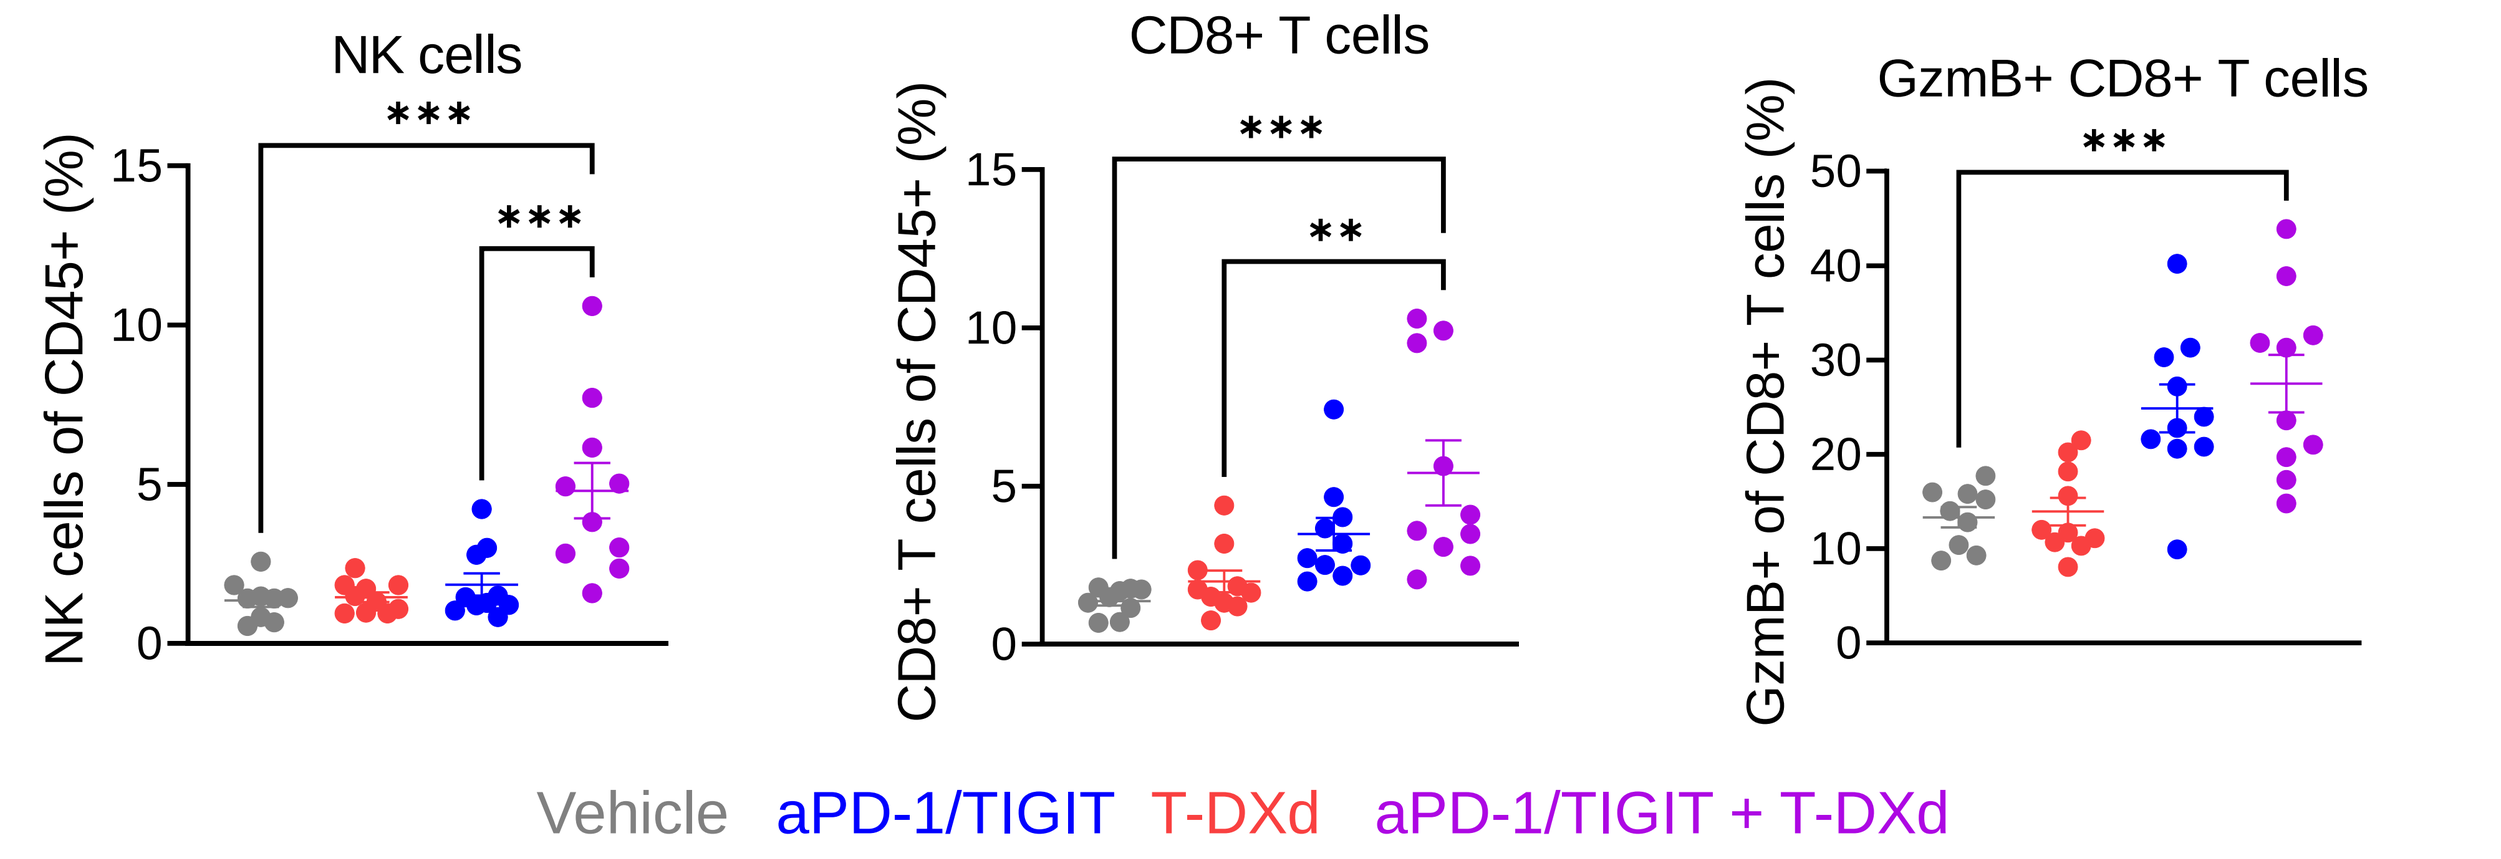
NK cells
NK cells of CD45+ (%)
CD8+ T cells
CD8+ T cells of CD45+ (%)
GzmB+ CD8+ T cells
GzmB+ of CD8+ T cells (%)
Vehicle
aPD-1/TIGIT
T-DXd
aPD-1/TIGIT + T-DXd

Figure 6. Combination aPD-1/TIGIT + T-DXd enhances tumoral NK (CD3- NKp46+) and CD8+ T cell (CD3+ CD8+) abundance in a syngeneic hHER2 EMT6 model. 5x10^6 hHER2 EMT6 cells were implanted subcutaneously into the flank of BALB/c mice (n=10). Treatment was initiated 7 days later. T-DXd (10 mg/kg) was administered IV on treatment day 0. aPD-1/TIGIT (10 mg/kg) was administered IP on treatment days 0, 3 & 7. Tumors were collected on day 8 post treatment and their immune content (CD45+ cells) assessed via flow cytometry.*, P <0.05; **, P <0.01; ***, P <0.001. One-way ANOVA with Tukey's multiple comparisons test.

Dual ICB + T-DXd is superior to aPD-L1 + T-DXd in a hHER2 EMT6 model

Table 5. Growth rate comparison versus aPD-L1 + T-DXd

Treatment	Dose (mg/kg)	Growth rate (median)	Growth rate (SD)	P value	CR
aPD-L1 T-DXd	10 10	0.0022	0.0243	N/A	1/10
aPD-L1 **aCTLA-4** T-DXd	10 10 10	-0.0563	0.0335	< 0.05	7/10
aPD-1/TIGIT T-DXd	10 10	-0.0426	0.0413	< 0.05	6/10

Growth rate data from Figures 3 and 5 used. Mann-Whitney t-test. SD: standard deviation. CR: complete response rate.

Acknowledgements

This study is sponsored by AstraZeneca. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201). We would like to acknowledge our Daiichi-Sankyo colleagues and other members of the T-DXd Joint Daiichi-Sankyo/AstraZeneca Preclinical & Translational Research team who provided support for this project. Rilvegostomig is derived from COM902, developed by Compugen Ltd.

Disclosures

All authors are employees of and hold stock in AstraZeneca.

References

1. Nakada T, et al. Chem Pharm Bull (Tokyo). 2019;67(3):173-185.
2. Ogitani Y, et al. Clin Cancer Res. 2016;22(20):5097-5108.
3. Trail PA, et al. Pharmacol Ther. 2018;181:126-142.
4. Jenkins L, et al. J Immuno Ther Cancer. 2022 (10).
5. Schmid P, et al. J. Clin. Oncol. 2021; 39:1023-3.
6. Dovedi S, et al. Cancer Discovery. 2021; 11(5): 1100-1117
7. Lee K, et al. J Immuno Ther Cancer. 2022: 10.
8. Karp NA, et al. Sci Rep. 2020; 10:6178.