274 Retrospective Cohort Study to Evaluate the Efficacy and Safety of T-DXd in HER2-Positive Unresectable Advanced or Recurrent Gastric or Gastroesophageal Junction Cancer: EN-DEAVOR Study

Koki Nakanishi[1], Yasuhiro Kodera[1], Hisato Kawakami[2], Akitaka Makiyama[3], Hirotaka Konishi[4], Satoshi Morita[5], Yukiya Narita[6], Naotoshi Sugimoto[7], Keiko Minashi[8], Motohiro Imano[9], Rin Inamoto[10], Hiroki Kume[11], Keita Yamaguchi[11], Wataru Hashimoto[11], Kei Muro[6]

[1]Nagoya University Graduate School of Medicine, Aichi, Japan; [2]Kindai University Faculty of Medicine, Osaka-Sayama, Japan; [3]Gifu University Hospital, Gifu, Japan; [4]Kyoto Prefectural University of Medicine, Kyoto, Japan; [5]Kyoto University Graduate School of Medicine, Kyoto, Japan; [6]Aichi Cancer Center Hospital, Aichi, Japan; [7]Osaka International Cancer Institute, Osaka, Japan; [8]Chiba Cancer Center, Chiba, Japan; [9]Kindai University, Osaka, Japan; [10]Saitama Cancer Center, Saitama, Japan; [11]Daiichi Sankyo Co. Ltd., Tokyo, Japan

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

BACKGROUND & OBJECTIVES

- Human epidermal growth factor receptor 2 (HER2) is overexpressed in approximately 20% of patients with gastric or gastroesophageal junction (G/GEJ) cancer, and this leads to poor patient prognosis[1,2]
- Trastuzumab deruxtecan (T-DXd) is a HER2-targeting antibody-drug conjugate (ADC) that has demonstrated efficacy in patients with HER2-positive advanced G/GEJ cancer (Eastern Cooperative Oncology Group performance status [ECOG PS] 0/1) in third- or later-line (3L+) (DESTINY-Gastric01 trial) and second-line (2L) (DESTINY-Gastric02 trial) settings with acceptable safety profiles[3,4]
 - T-DXd has also shown efficacy in patients with HER2-low G/GEJ cancer in 3L+ settings with an acceptable safety profile in exploratory cohorts of the DESTINY-Gastric01 trial[5]
- However, real-world data on treatment with T-DXd remain limited[6]
 - Moreover, evidence in patients not eligible for clinical trial participation (e.g., elderly patients and those with ECOG PS ≥2) is lacking
- This retrospective cohort study (UMIN000049032) assessed the real-world effectiveness and safety of T-DXd in patients with HER2-positive, unresectable, advanced/recurrent G/GEJ cancer

CONCLUSIONS

- This retrospective cohort study demonstrated the real-world effectiveness and safety of T-DXd in patients with HER2-positive, unresectable, advanced/recurrent G/GEJ cancer in Japan
- No new safety signals were identified

METHODS

Study Outline

- A non-interventional, observational, retrospective cohort study conducted at 63 sites in Japan
 - Enrollment period: September 25, 2020, to September 30, 2021, for the first dose of T-DXd (**Fig. 1**)
 - Observation period: September 25, 2020, to September 30, 2022 (**Fig. 1**)

Fig. 1. Study Outline

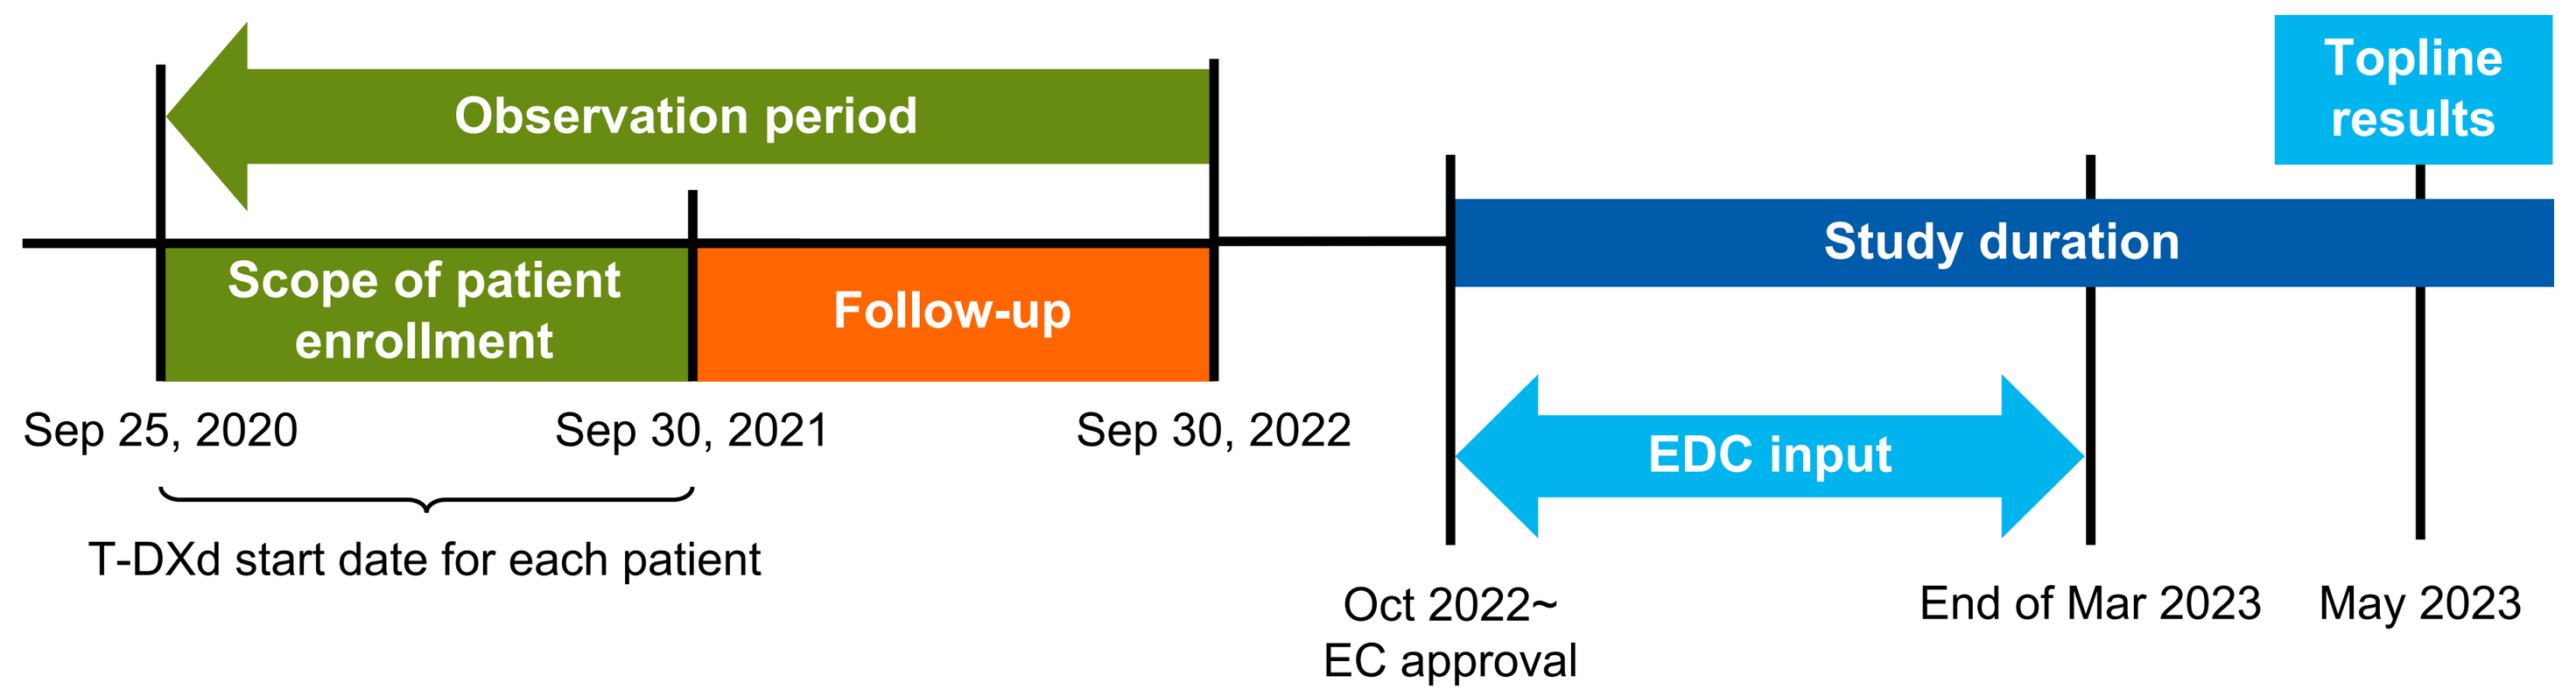

Observation period
Scope of patient enrollment
Follow-up
Study duration
Topline results
EDC input
Sep 25, 2020
Sep 30, 2021
Sep 30, 2022
T-DXd start date for each patient
Oct 2022~ EC approval
End of Mar 2023
May 2023

EC, ethics committee; EDC, electronic data capture; T-DXd, trastuzumab deruxtecan.

Eligibility Criteria

- Inclusion criteria:
 - Age ≥20 years
 - Histopathologically confirmed, HER2-positive (immunohistochemistry [IHC]3+ or IHC2+ with in situ hybridization [ISH]+) G/GEJ cancer
 - Unresectable, advanced/recurrent G/GEJ cancer that has progressed after cancer chemotherapy
 - T-DXd initiation date: September 25, 2020, to September 30, 2021
- Key exclusion criteria:
 - Patients with active, multiple, primary malignancies that may affect the evaluation of T-DXd treatment
 - Patients who had received T-DXd in other interventional studies or at other sites

Treatment Outcomes

- Effectiveness:
 - Overall survival (OS), real-world progression-free survival (rwPFS), and time to treatment failure (TTF)
 - Response rate: objective response rate (ORR) and best overall response
 - Best percent change in the sum of diameters for all target lesions
- Safety:
 - Common Terminology Criteria for Adverse Events (CTCAE) grade ≥3 treatment-emergent adverse events (TEAEs)
 - TEAEs leading to dose reduction, interruption, or discontinuation

RESULTS

Patient Disposition

- Among 318 patients enrolled, 312 were eligible for the analysis (6 were excluded because of incorrect enrollment)
- A total of 226 patients had target lesions

Baseline Characteristics

- Patient age ranged from 27 to 89 years (median: 70.0 years) (**Table 1**)
- A total of 38 (12.2%) patients had ECOG PS ≥2, and 135 (43.3%) had ascites (**Table 1**)

Table 1. Baseline Characteristics (All Eligible Patients)

		All eligible patients (n=312)
Male sex, n (%)		235 (75.3)
Age (years), median (range)		70.0 (27–89)
ECOG PS ≥2, n (%)		38 (12.2)
HER2 status at initial diagnosis–IHC3+, n (%)		217 (69.6)
Site of primary lesions–stomach, n (%)		264 (84.6)
Any surgeries for primary lesions, n (%)		107 (34.3)
Histological type of primary lesions, n (%)	Diffuse	79 (25.3)
	Intestinal	170 (54.5)
	Others/unknown	63 (20.2)
≥2 metastatic organs, n (%)		192 (61.5)
Ascites–yes, n (%)		135 (43.3)
Number of previous lines, n (%)	≤2	161 (51.6)
	3	73 (23.4)
	≥4	78 (25.0)
Previous therapies, n (%)	Taxane	290 (92.9)
	Trastuzumab	288 (92.3)
	Immune checkpoint inhibitor	131 (42.0)
	Ramucirumab	256 (82.1)
	Platinum	280 (89.7)
	Irinotecan	49 (15.7)
	Pyrimidine fluoride	296 (94.9)
	Others	42 (13.5)
Trastuzumab-free interval (months, n=286), median (range)		6.8 (0.1–70.6)

ECOG PS, Eastern Cooperative Oncology Group performance status; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry.

T-DXd Dose in Each Treatment Cycle

- In total, 34.9% of patients received <4 cycles of T-DXd
- T-DXd dose in Cycle 1 was >5.4 to ≤6.4 mg/kg in 244 (78.2%) patients, >4.4 to ≤5.4 mg/kg in 50 (16.0%) patients, and >3.2 to ≤4.4 mg/kg in 17 (5.4%) patients (**Fig. 2**)

Fig. 2. T-DXd Dose in Each Treatment Cycle (All Eligible Patients)

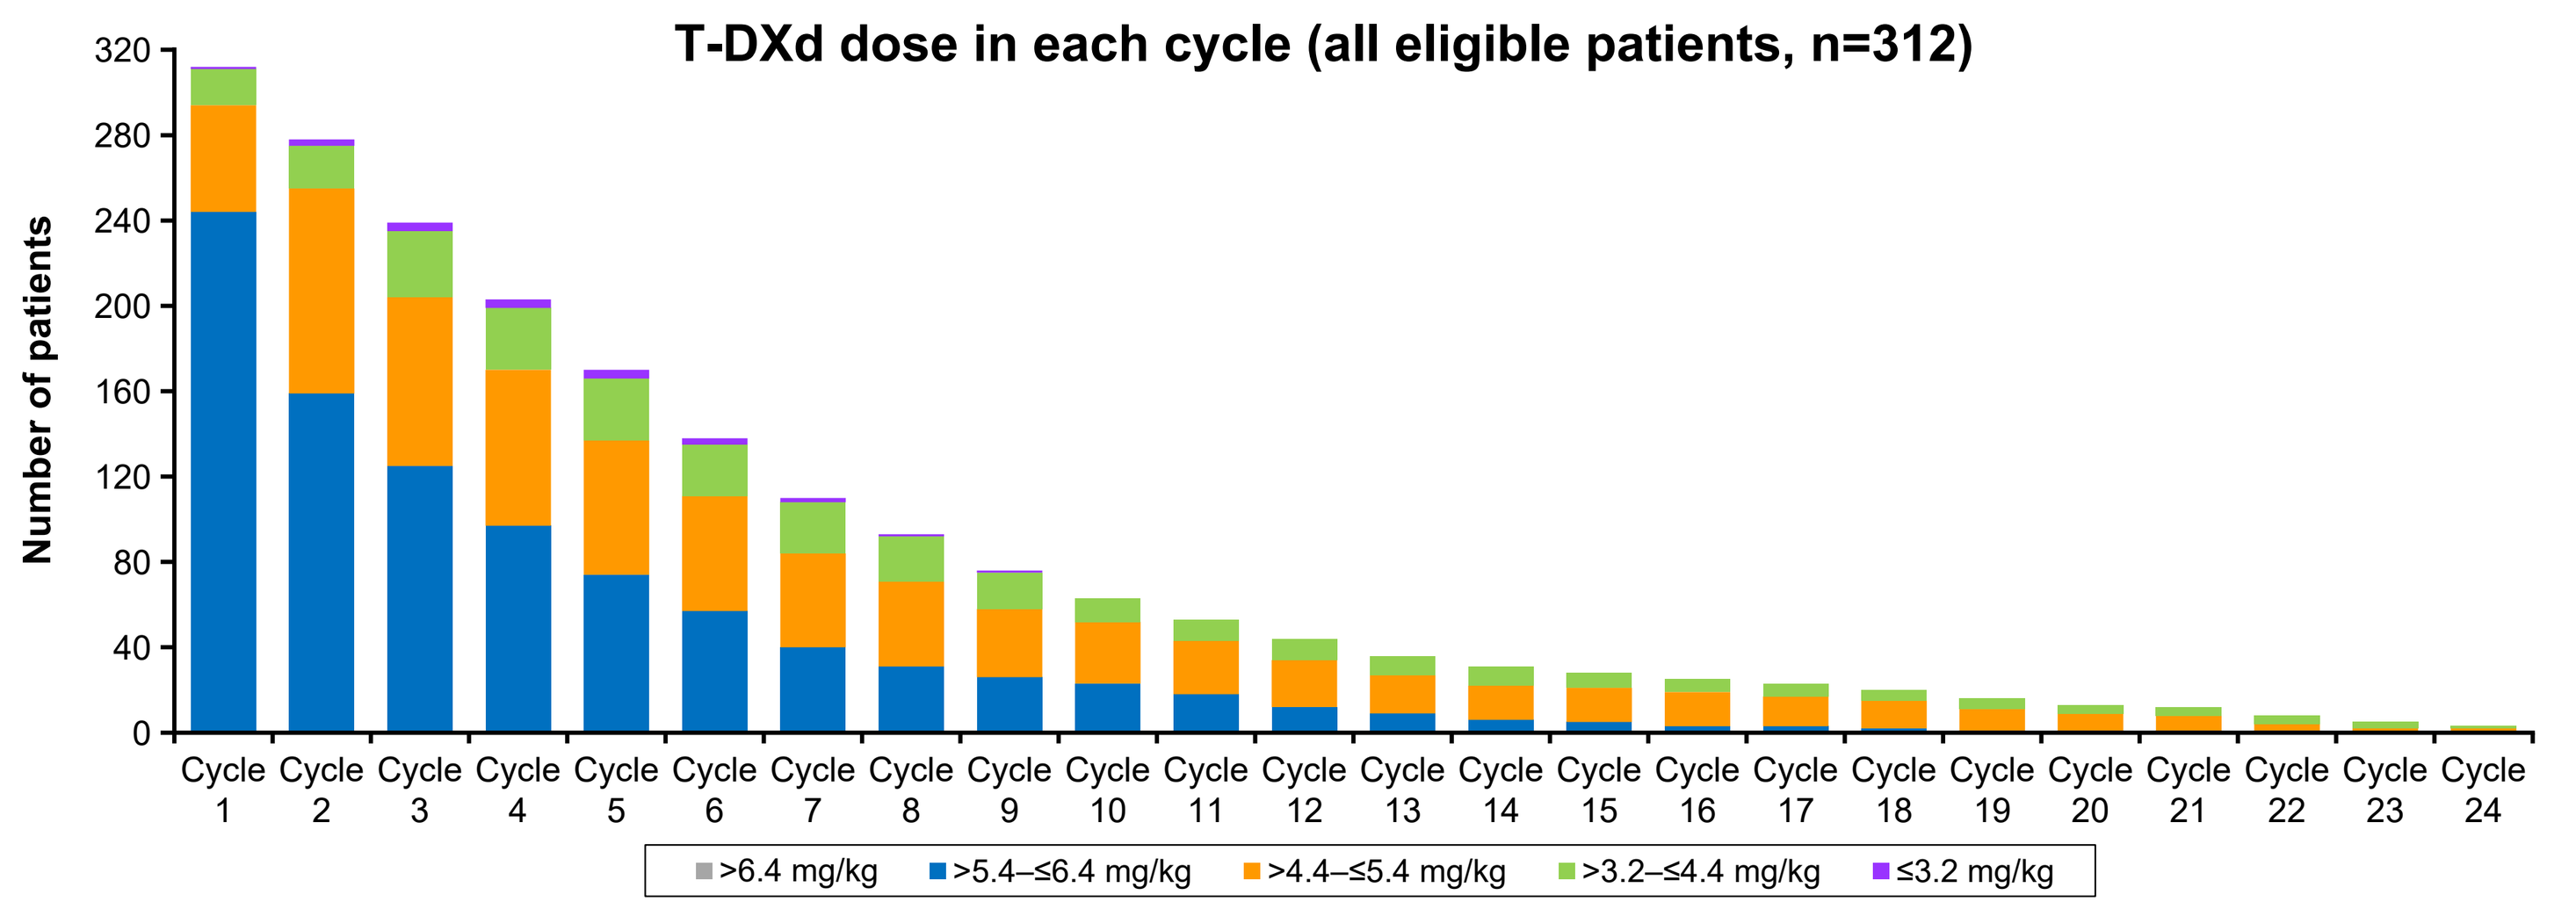

T-DXd dose in each cycle (all eligible patients, n=312)
Number of patients
Cycle 1 … Cycle 24
>6.4 mg/kg
>5.4–≤6.4 mg/kg
>4.4–≤5.4 mg/kg
>3.2–≤4.4 mg/kg
≤3.2 mg/kg

T-DXd, trastuzumab deruxtecan.

OS, rwPFS, and TTF

- The median OS, median rwPFS, and median TTF were 8.90, 4.57, and 3.94 months, respectively (**Fig. 3**)

Fig. 3. OS, rwPFS, and TTF (All Eligible Patients)

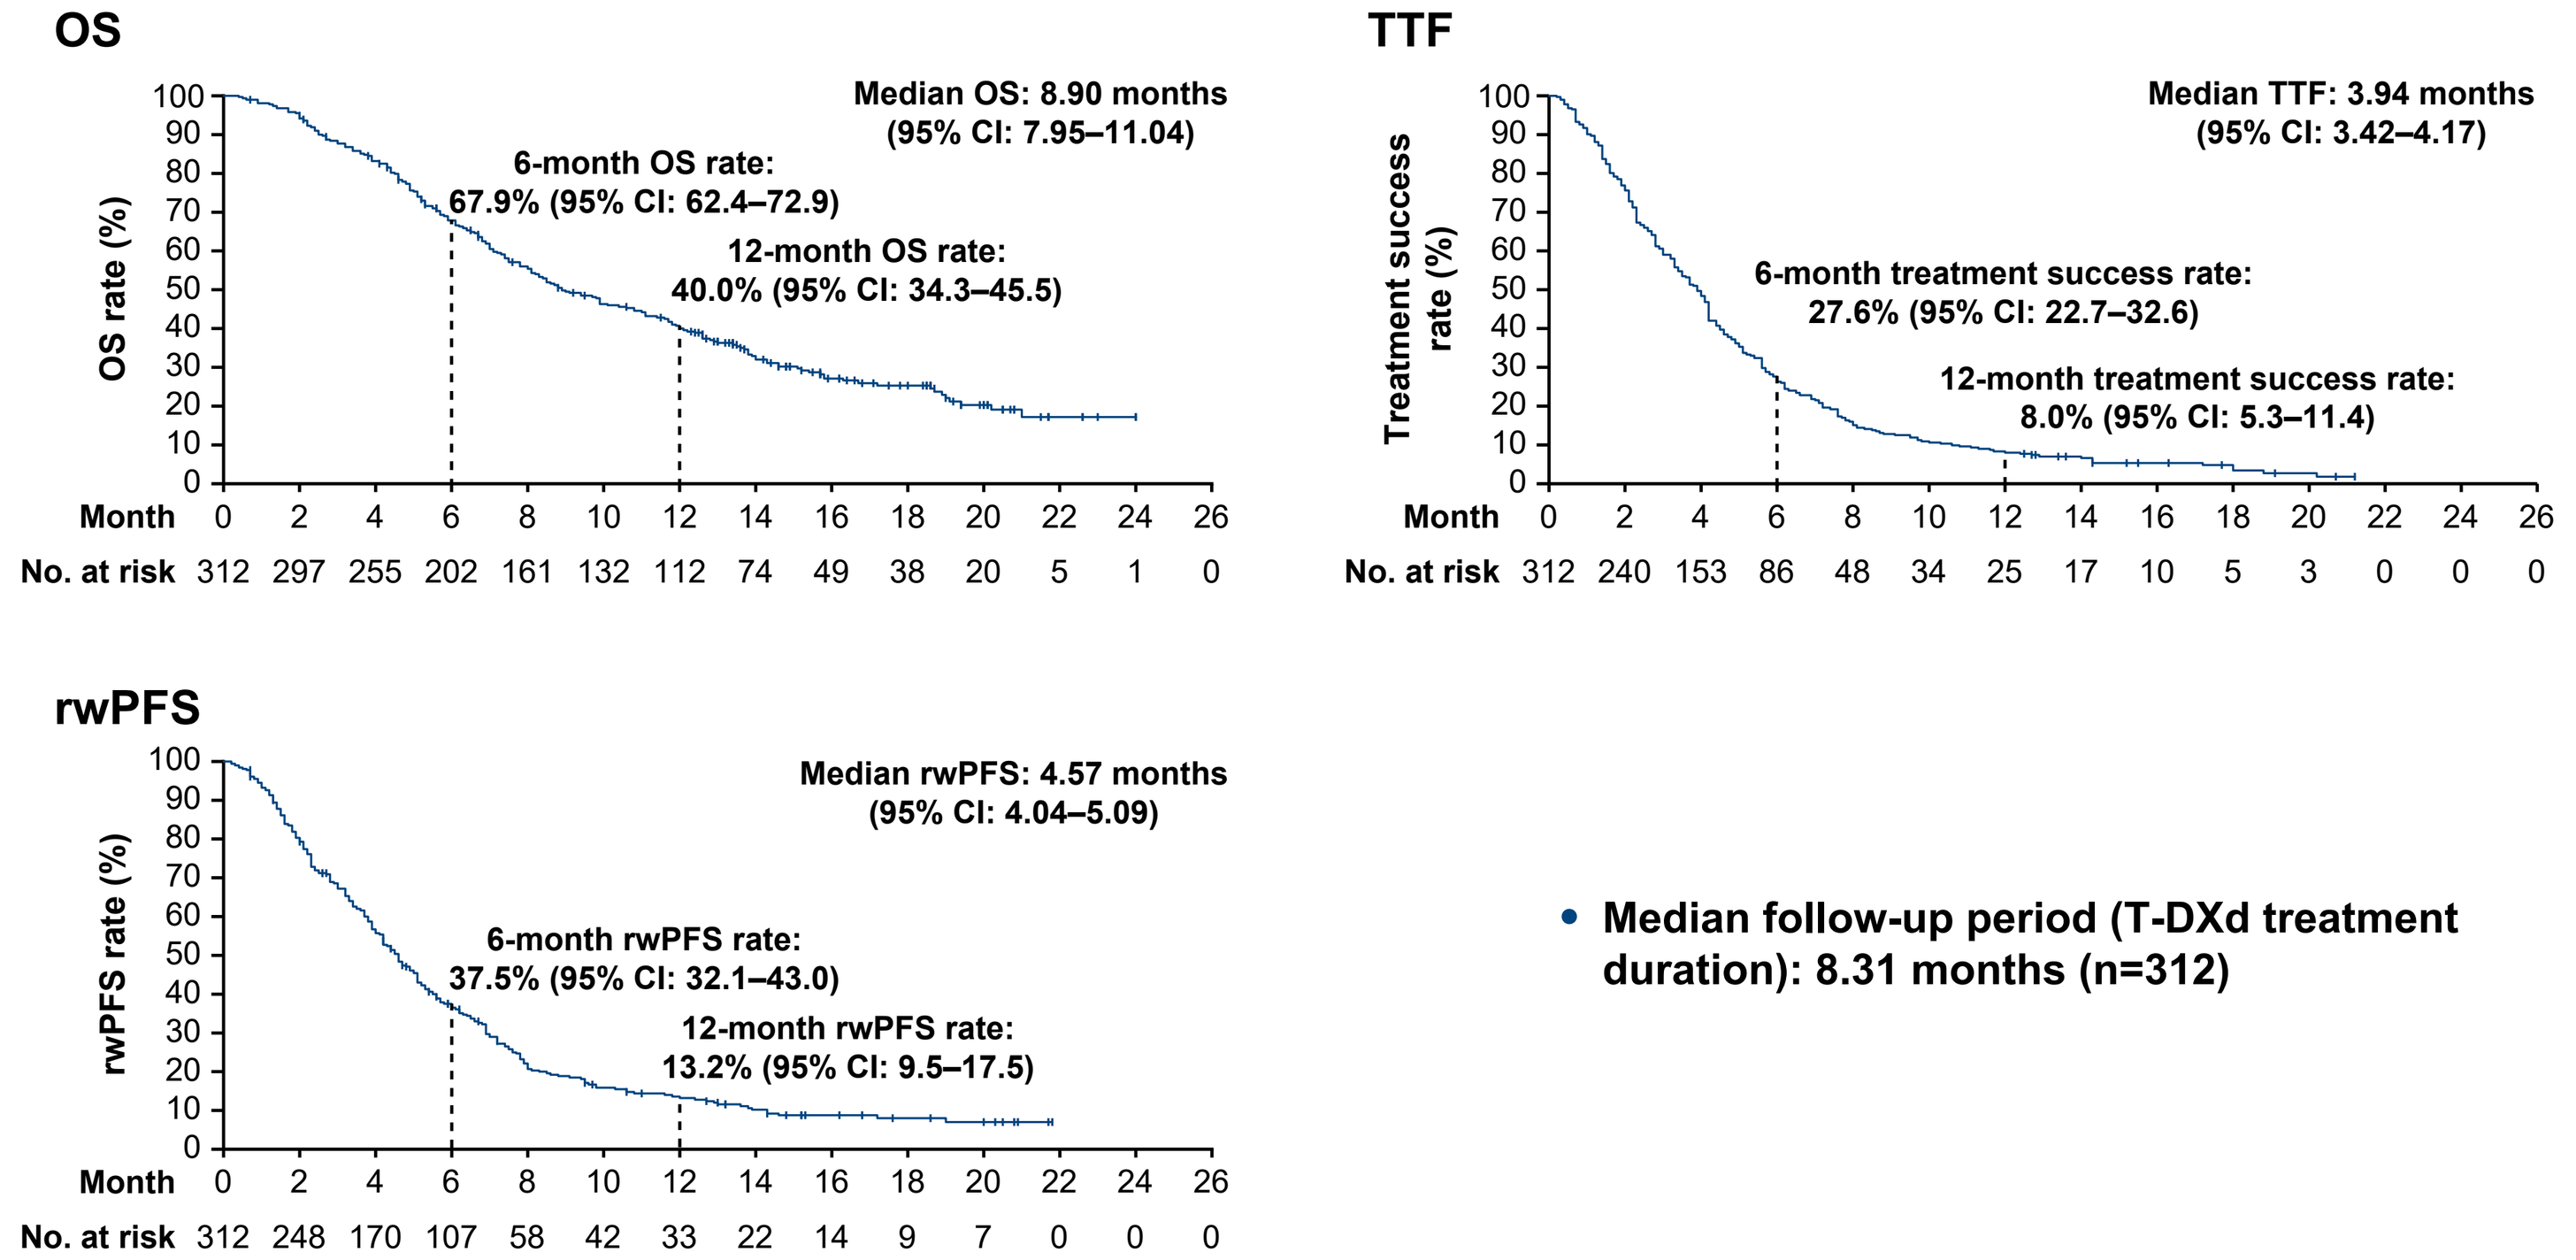

OS
Median OS: 8.90 months (95% CI: 7.95–11.04)
6-month OS rate: 67.9% (95% CI: 62.4–72.9)
12-month OS rate: 40.0% (95% CI: 34.3–45.5)
OS rate (%)
Month 0 2 4 6 8 10 12 14 16 18 20 22 24 26
No. at risk 312 297 255 202 161 132 112 74 49 38 20 5 1 0
TTF
Median TTF: 3.94 months (95% CI: 3.42–4.17)
6-month treatment success rate: 27.6% (95% CI: 22.7–32.6)
12-month treatment success rate: 8.0% (95% CI: 5.3–11.4)
Treatment success rate (%)
Month 0 2 4 6 8 10 12 14 16 18 20 22 24 26
No. at risk 312 240 153 86 48 34 25 17 10 5 3 0 0 0
rwPFS
Median rwPFS: 4.57 months (95% CI: 4.04–5.09)
6-month rwPFS rate: 37.5% (95% CI: 32.1–43.0)
12-month rwPFS rate: 13.2% (95% CI: 9.5–17.5)
rwPFS rate (%)
Month 0 2 4 6 8 10 12 14 16 18 20 22 24 26
No. at risk 312 248 170 107 58 42 33 22 14 9 7 0 0 0

- **Median follow-up period (T-DXd treatment duration): 8.31 months (n=312)**

The median OS, rwPFS, and TTF were estimated using the Brookmeyer and Crowley method.
CI, confidence interval; OS, overall survival; rwPFS, real-world progression-free survival; T-DXd, trastuzumab deruxtecan; TTF, time to treatment failure.

Response Rate

- The ORR was 42.9% (95% confidence interval [CI], 36.4–49.7) and disease control rate (DCR) was 81.4% (95% CI, 75.7–86.3) in patients with target lesions (n=226) (**Table 2**)
- The majority of patients with target lesions recorded partial response (PR) (40.7%) or stable disease (SD) (38.5%) as best overall response (**Table 2**)

Table 2. Response Rate (Patients with Target Lesions)

Response rate	Patients with target lesions (n=226)
ORR, n (%)	97 (42.9) [95% CI, 36.4–49.7]
Best overall response, n (%)	
CR	5 (2.2)
PR	92 (40.7)
SD	87 (38.5)
PD	31 (13.7)
NE	11 (4.9)

The 95% CI was estimated using the Clopper-Pearson method.
CI, confidence interval; CR, complete response; NE, not evaluable; ORR, objective response rate; PD, progressive disease; PR, partial response; SD, stable disease.

Best Percent Change from Baseline in the Sum of Diameters for All Target Lesions

- The median best percentage change in the sum of diameters for all target lesions was -24.7% (range: -100.0 to 127.7) (**Fig. 4**)

Fig. 4. Best Percent Change from Baseline in the Sum of Diameters for All Target Lesions (Patients with Target Lesions)

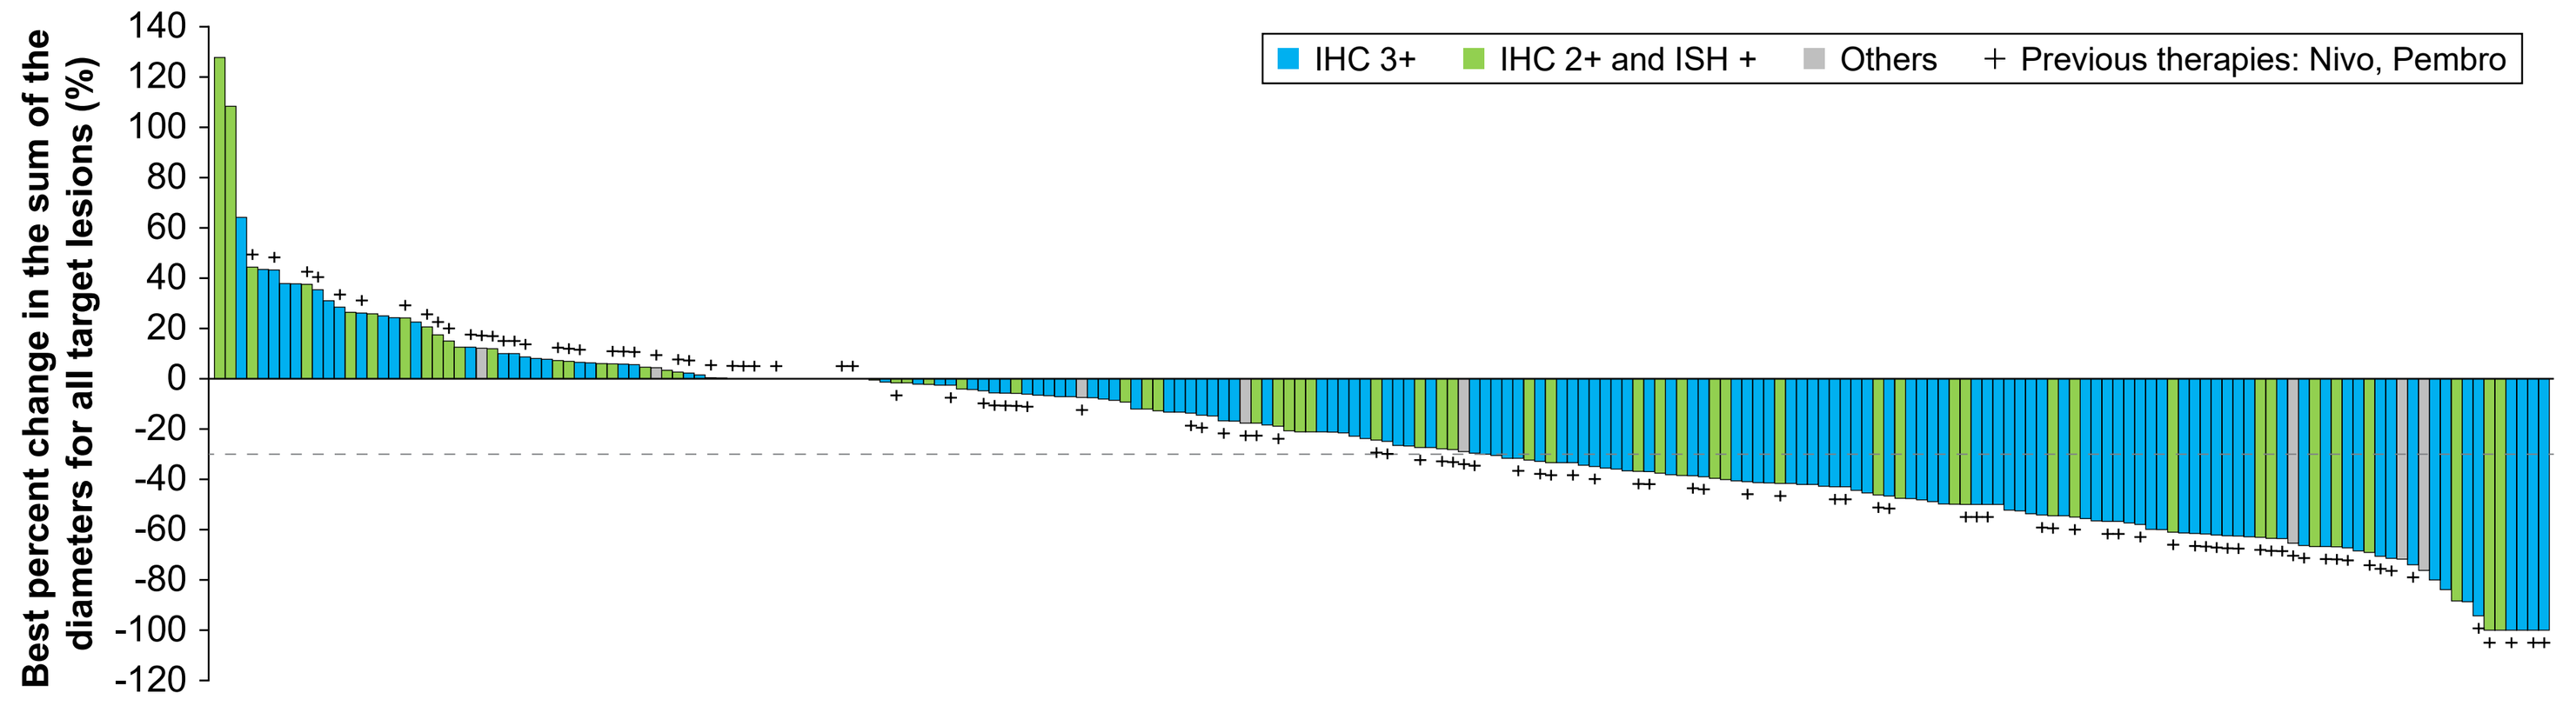

IHC 3+
IHC 2+ and ISH +
Others
+ Previous therapies: Nivo, Pembro
Best percent change in the sum of the diameters for all target lesions (%)

IHC, immunohistochemistry; ISH, in situ hybridization; Nivo, nivolumab; Pembro, pembrolizumab

Grade ≥3 TEAEs

- The most common grade ≥3 TEAEs are summarized in **Table 3**
- Grade 5 TEAEs were observed in 8 patients (interstitial pneumonia [n=5], febrile neutropenia, pneumonia, and pneumonia cytomegaloviral [n=1 each])

Table 3. Most Common (>4%) Grade ≥3 TEAEs (All Eligible Patients)

Patients with CTCAE grade ≥3 TEAEs, n (%)	All eligible patients (n=312)[a]
Any grade ≥3 TEAEs	151 (48.4)
Hematotoxicity TEAEs	89 (28.5)
Neutrophil count decreased	61 (19.6)
Anemia	29 (9.3)
Platelet count decreased	13 (4.2)
Non-hematotoxicity TEAEs	88 (28.2)
Anorexia	29 (9.3)
Malaise	14 (4.5)
Interstitial pneumonia	14 (4.5)
Nausea	13 (4.2)

TEAEs were coded using MedDRA version 26.0. [a]Multiple selections were possible when reporting TEAEs.
CTCAE, Common Terminology Criteria for Adverse Events; MedDRA, Medical Dictionary for Regulatory Activities; TEAE, treatment-emergent adverse event.

TEAEs Leading to Dose Reduction, Interruption, or Discontinuation

- The most common hematotoxicity TEAE that led to dose reduction, interruption, or discontinuation was neutrophil count decreased (24.4%) (**Table 4**)
- The most common non-hematotoxicity TEAE that led to dose reduction, interruption, or discontinuation was anorexia (17.6%) (**Table 4**)

Table 4. Most Common (>5%) TEAEs Leading to Dose Reduction, Interruption, or Discontinuation (All Eligible Patients)

Patients with TEAEs, n (%)	All eligible patients (n=312)			
	Dose reduction, interruption, or discontinuation	Dose reduction[a]	Dose interruption[a]	Discontinuation[a]
Any TEAEs	190 (60.9)	115 (36.9)	106 (34.0)	74 (23.7)
Hematotoxicity TEAEs	97 (31.1)	61 (19.6)	72 (23.1)	4 (1.3)
Neutrophil count decreased	76 (24.4)	49 (15.7)	60 (19.2)	1 (0.3)
Anemia	17 (5.4)	9 (2.9)	9 (2.9)	3 (1.0)
Non-hematotoxicity TEAEs	139 (44.6)	67 (21.5)	48 (15.4)	74 (23.7)
Anorexia	55 (17.6)	39 (12.5)	16 (5.1)	19 (6.1)
Malaise	47 (15.1)	32 (10.3)	15 (4.8)	11 (3.5)
Nausea	29 (9.3)	19 (6.1)	8 (2.6)	10 (3.2)
Interstitial pneumonia	29 (9.3)	0 (0.0)	0 (0.0)	29 (9.3)

TEAEs were coded using MedDRA version 26.0. AEs leading to dose reduction, interruption, or discontinuation with an incidence of >5% are listed. [a]Multiple selections were possible when reporting TEAEs.
MedDRA, Medical Dictionary for Regulatory Activities; TEAE, treatment-emergent adverse event.

ACKNOWLEDGMENTS

We thank all patients, their families, and the facility staff involved in this study.
Medical writing support was provided by Sarayu Pai, PhD, CMPP, and Mami Hirano, MSc, of Cactus Life Sciences (part of Cactus Communications) and funded by Daiichi Sankyo Co., Ltd.
This study was sponsored by Daiichi Sankyo Co., Ltd. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for T-DXd.

REFERENCES

1. Ariga S. *J Clin Med*. 2023;12:3391.
2. Ma C, et al. *Front Oncol*. 2023;13:1080990.
3. Shitara K, et al. *N Engl J Med*. 2020;382:2419-2430.
4. Van Cutsem E, et al. *Lancet Oncol*. 2023;24:744-756.
5. Yamaguchi K, et al. *J Clin Oncol*. 2023;41:816-825.
6. Matsumoto T, et al. *J Clin Med*. 2022;11:2247.

Contact: kume.hiroki.he@daiichisankyo.co.jp

EN-DEAVOR