Presented at ASCO GI 2024; January 18–20, San Francisco, US

Phase 1b/2, open-label dose-escalation and -expansion study evaluating trastuzumab deruxtecan (T-DXd) monotherapy and combinations in patients with HER2+ and HER2-low gastric cancer (DESTINY-Gastric03)

Yelena Y Janjigian,[1] Saeed Raoufmoghaddam,[2] Magdalena Sztachelska,[3] Satya Das[4]

[1]Gastrointestinal Oncology Service, Memorial Sloan Kettering Cancer Center, NY, US; [2]Late-Stage Development, Oncology R&D, AstraZeneca, Cambridge, UK; [3]Late-Stage Development, Oncology R&D, AstraZeneca, Warsaw, Poland; [4]Late-Stage Development, Oncology R&D, AstraZeneca, Gaithersburg, MD, US

TPS424

Plain Language Summary

Why are we performing this research?
Some people with gastric cancers have higher than normal levels of a protein called human epidermal growth factor receptor 2 (HER2), which is found on the surface of cancer cells and promotes tumor growth; these cancers are called HER2-positive.[1] Trastuzumab deruxtecan (T-DXd) is a type of drug called an antibody-drug conjugate, which is a chemotherapy with a linker (together called deruxtecan) joined to an antibody (trastuzumab). Trastuzumab binds HER2 found on the surface of cancer cells, where it releases the chemotherapy to kill these cells.[2,3] T-DXd is approved in the US, EU, and Japan for people with HER2-positive gastric cancers that have spread to nearby tissues or elsewhere in the body and that cannot be removed by surgery (advanced cancer), and who have received prior chemotherapy. One of the aims of this clinical study is to investigate whether T-DXd in combination with other cancer drugs/treatments will be beneficial as a first-line therapy.

How are we performing this research?
DESTINY-Gastric03 (DG-03) is an ongoing clinical study that is assessing how well T-DXd works in combination with other cancer drugs/treatments such as pembrolizumab (a cancer drug that targets a protein called programmed cell death protein 1),[4] volrustomig (MEDI5752, a cancer drug that targets two proteins called programmed cell death protein 1 and cytotoxic T-lymphocyte-associated protein 4),[5] and chemotherapy, in participants with HER2-expressing advanced gastric cancers. The primary outcome of interest is the percentage of participants who have a decrease in the size or number of tumors after treatment.

Who will participate in this study?
Participants who have HER2-expressing advanced gastric cancer will be included in this study. In Part 1, we will assess T-DXd in participants who have received previous treatment for their cancer after it has spread, and Parts 2 and 3 we will assess in participants who have not received any prior treatment.

Where can I access more information?
For more information about DG-03, please visit https://clinicaltrials.gov/study/NCT04379596. You can also speak to your doctor about this and other clinical studies.

1. Gravalos C, Jimeno A. *Ann Oncol.* 2008;19:1523–1529; 2. Nakada T, et al. *Chem Pharm Bull (Tokyo).* 2019;67:173–185; 3. Ogitani Y, et al. *Clin Cancer Res.* 2016;22:5097–5108; 4. Keytruda (pembrolizumab) highlights of prescribing information. 2023. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/125514s148lbl.pdf (Accessed December 1, 2023); 5. AstraZeneca. Press release. October 11, 2023. Available from: https://www.astrazeneca.com/media-centre/press-releases/2023/astrazeneca-advances-scientific-leadership-across-multiple-cancers-with-first-data-from-four-pivotal-phase-iii-trials-at-esmo.html (Accessed November 30, 2023)

Background

- An estimated 18% of gastric or gastroesophageal cancers are human epidermal growth factor receptor 2-positive (HER2+) (immunohistochemistry [IHC] 3+, IHC 2+/in situ hybridization [ISH]+)[1]
- T-DXd is an antibody-drug conjugate composed of an anti-HER2 monoclonal antibody, a tetrapeptide-based cleavable linker, and a topoisomerase I inhibitor payload[2,3]
- T-DXd has demonstrated durable response rates and meaningful clinical activity in previously treated patients with HER2+ gastric cancer in the DESTINY-Gastric01,[4] DESTINY-Gastric02,[5] and DESTINY-Gastric06[6] studies
 - Antitumor activity was also seen in patients with HER2-low gastric cancer; in DESTINY-Gastric01, objective response rates (ORRs) of 26.3% and 9.5% were seen in patients with IHC 2+/ISH− and IHC 1+, respectively[4]

T-DXd is approved in several countries including the US, EU, and Japan for the treatment of adult patients with locally advanced or metastatic HER2+ gastric or gastroesophageal junction adenocarcinoma who have received a prior trastuzumab-based regimen[7–9]

- The FDA granted accelerated approval for first-line pembrolizumab (an anti-programmed cell death protein 1 [PD-1] antibody) and trastuzumab plus chemotherapy in patients with metastatic HER2+ gastric cancer whose tumors express programmed cell death ligand 1 (PD-L1), as determined by an FDA-approved test, after the results of a Phase 3 clinical trial showed clinical benefit in HER2+ gastric cancer[10,11]
- A preclinical study in a fibrosarcoma cell line showed that volrustomig (MEDI5752, an anti-PD-1/cytotoxic T-lymphocyte-associated protein 4 [CTLA-4] bispecific antibody) leads to increased antitumor activity compared with anti-PD-1 or anti-CTLA-4 antibodies alone[12,13]
- In DESTINY-Gastric03 (DG-03), we are evaluating the safety and efficacy of T-DXd in combination with chemotherapy and/or immunotherapies in HER2-expressing gastric cancers
 - The results of Parts 1A and 1B of this study were previously published, showing an ORR of 50% and 43% with T-DXd plus 5-fluorouracil or T-DXd plus capecitabine, respectively[14]

- **Following publication of results from Parts 1A and 1B, the DG-03 study has been updated:**
 - **In Part 2, Arm 2F has been added to investigate T-DXd + fluoropyrimidine + pembrolizumab**
 - **Part 3 has been added to investigate volrustomig in combination with T-DXd + fluoropyrimidine**

Study Design and Population

Part 1 ENROLLMENT COMPLETE (1A and 1B published)[12]
Dose escalation (3 + 3)*

- **1A**: T-DXd IV q3w + 5-FU IV on Days 1–5, q3w
 - **1D(a)**: T-DXd IV q3w + 5-FU IV on Days 1–5 q3w + oxaliplatin IV q3w
- **1B**: T-DXd IV q3w + cap (oral) BID on Days 1–14
 - **1D(b)**: T-DXd IV q3w + cap (oral) BID on Days 1–14 + oxaliplatin IV q3w
- **1C**: T-DXd IV q3w + 5-FU IV on Days 1–5, q3w
 - **1E(a)**: T-DXd IV q3w + 5-FU IV on Days 1–5 + durvalumab IV q3w
 - **1E(b)**: T-DXd IV q3w + cap (oral) BID on Days 1–14 + durvalumab IV q3w

Parts 2A–2E Dose expansion ENROLLMENT COMPLETE
N≈40 participants per arm;
2F RECRUITING: N≈30 participants†‡

- 2A: Trastuzumab + fluoropyrimidine + platinum-based chemotherapy
- 2B: T-DXd monotherapy IV q3w
- 2C:§ T-DXd IV q3w + fluoropyrimidine ± oxaliplatin IV q3w
- 2D:¶‖ T-DXd IV q3w + fluoropyrimidine + pembrolizumab IV q3w
- 2E:‖ T-DXd IV q3w + pembrolizumab IV q3w
- **2F:‖**†† T-DXd IV q3w + fluoropyrimidine + pembrolizumab IV q3w**

Part 3 RECRUITING
Volrustomig safety cohort
N=6 per dose‡‡

- T-DXd IV q3w + fluoropyrimidine + volrustomig IV q3w**

Part 3 RECRUITING
Volrustomig main cohort
N≈18–30†

- 3A: HER2+ T-DXd IV q3w + fluoropyrimidine + volrustomig IV q3w**§§
- 3B: HER2-low T-DXd IV q3w + fluoropyrimidine + volrustomig IV q3w**§§

During Part 1 of the study, the FDA granted accelerated approval for first-line pembrolizumab plus trastuzumab and chemotherapy for HER2+ gastric cancer in patients whose tumors express PD-L1.[11] Therefore, the study sponsor amended the study design to include Arm 2F, and terminated Arm 2A early as this was a control arm using the standard of care

*At the start of Part 1 of the study, ≥three participants will be enrolled into each arm at the starting dose; subsequent enrollments will follow a '3 + 3' dosing design to inform dose-escalation or de-escalation decisions or to confirm a RP2D; †N numbers provided are target cohort numbers as patient enrollment is ongoing; ‡participants will be randomized into each arm and stratified by HER2 status (IHC 3+ or IHC 2+/ISH+) based on local tissue test results, except for Arm 2F where participants will be enrolled; §T-DXd and fluoropyrimidine (Investigator choice of 5-FU [IV, Days 1–5, q3w] or cap [oral, Days 1–14, BID]) starting dose will be the RP2D of the combination from Part 1 (Arms 1A and 1B) with or without oxaliplatin at the dose established in Part 1; ¶T-DXd and fluoropyrimidine (Investigator choice of 5-FU [IV, Days 1–5, q3w] or cap [oral, Days 1–14, BID]) starting dose will be the RP2D of the combination from Part 1 (Arms 1A and 1B); ‖the safety review committee will perform a safety assessment after the first six participants randomized (Arms 2D and 2E) or enrolled (Arm 2F) to each arm complete ≥21 days of treatment; **Investigator choice of 5-FU (IV, Days 1–5, q3w) or cap (oral, Days 1–14, BID); ††a limit will be applied to ensure <20% of participants have IHC 2+/ISH+ status; ‡‡HER2+ or HER2-low participants will be enrolled into the safety cohort; ≥six DLT-evaluable participants will be recruited to receive volrustomig at the SD and ≥six participants will be enrolled to receive volrustomig at the E1 if the SD of volrustomig meets the prespecified DLT rules; §§RP2D of volrustomig based on the safety review committee's review of the Part 3 safety cohort data and safety data from other trials
5-FU, 5-fluorouracil; BID, twice daily; cap, capecitabine; DLT, dose-limiting toxicity; E1, escalation dose; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; ISH, in situ hybridization; IV, intravenous; q3w, every 3 weeks; RP2D, recommended Phase 2 dose; SD, starting dose; T-DXd, trastuzumab deruxtecan

Enrollment start: 3 June 2020
Currently recruiting participants for Parts 2F and 3

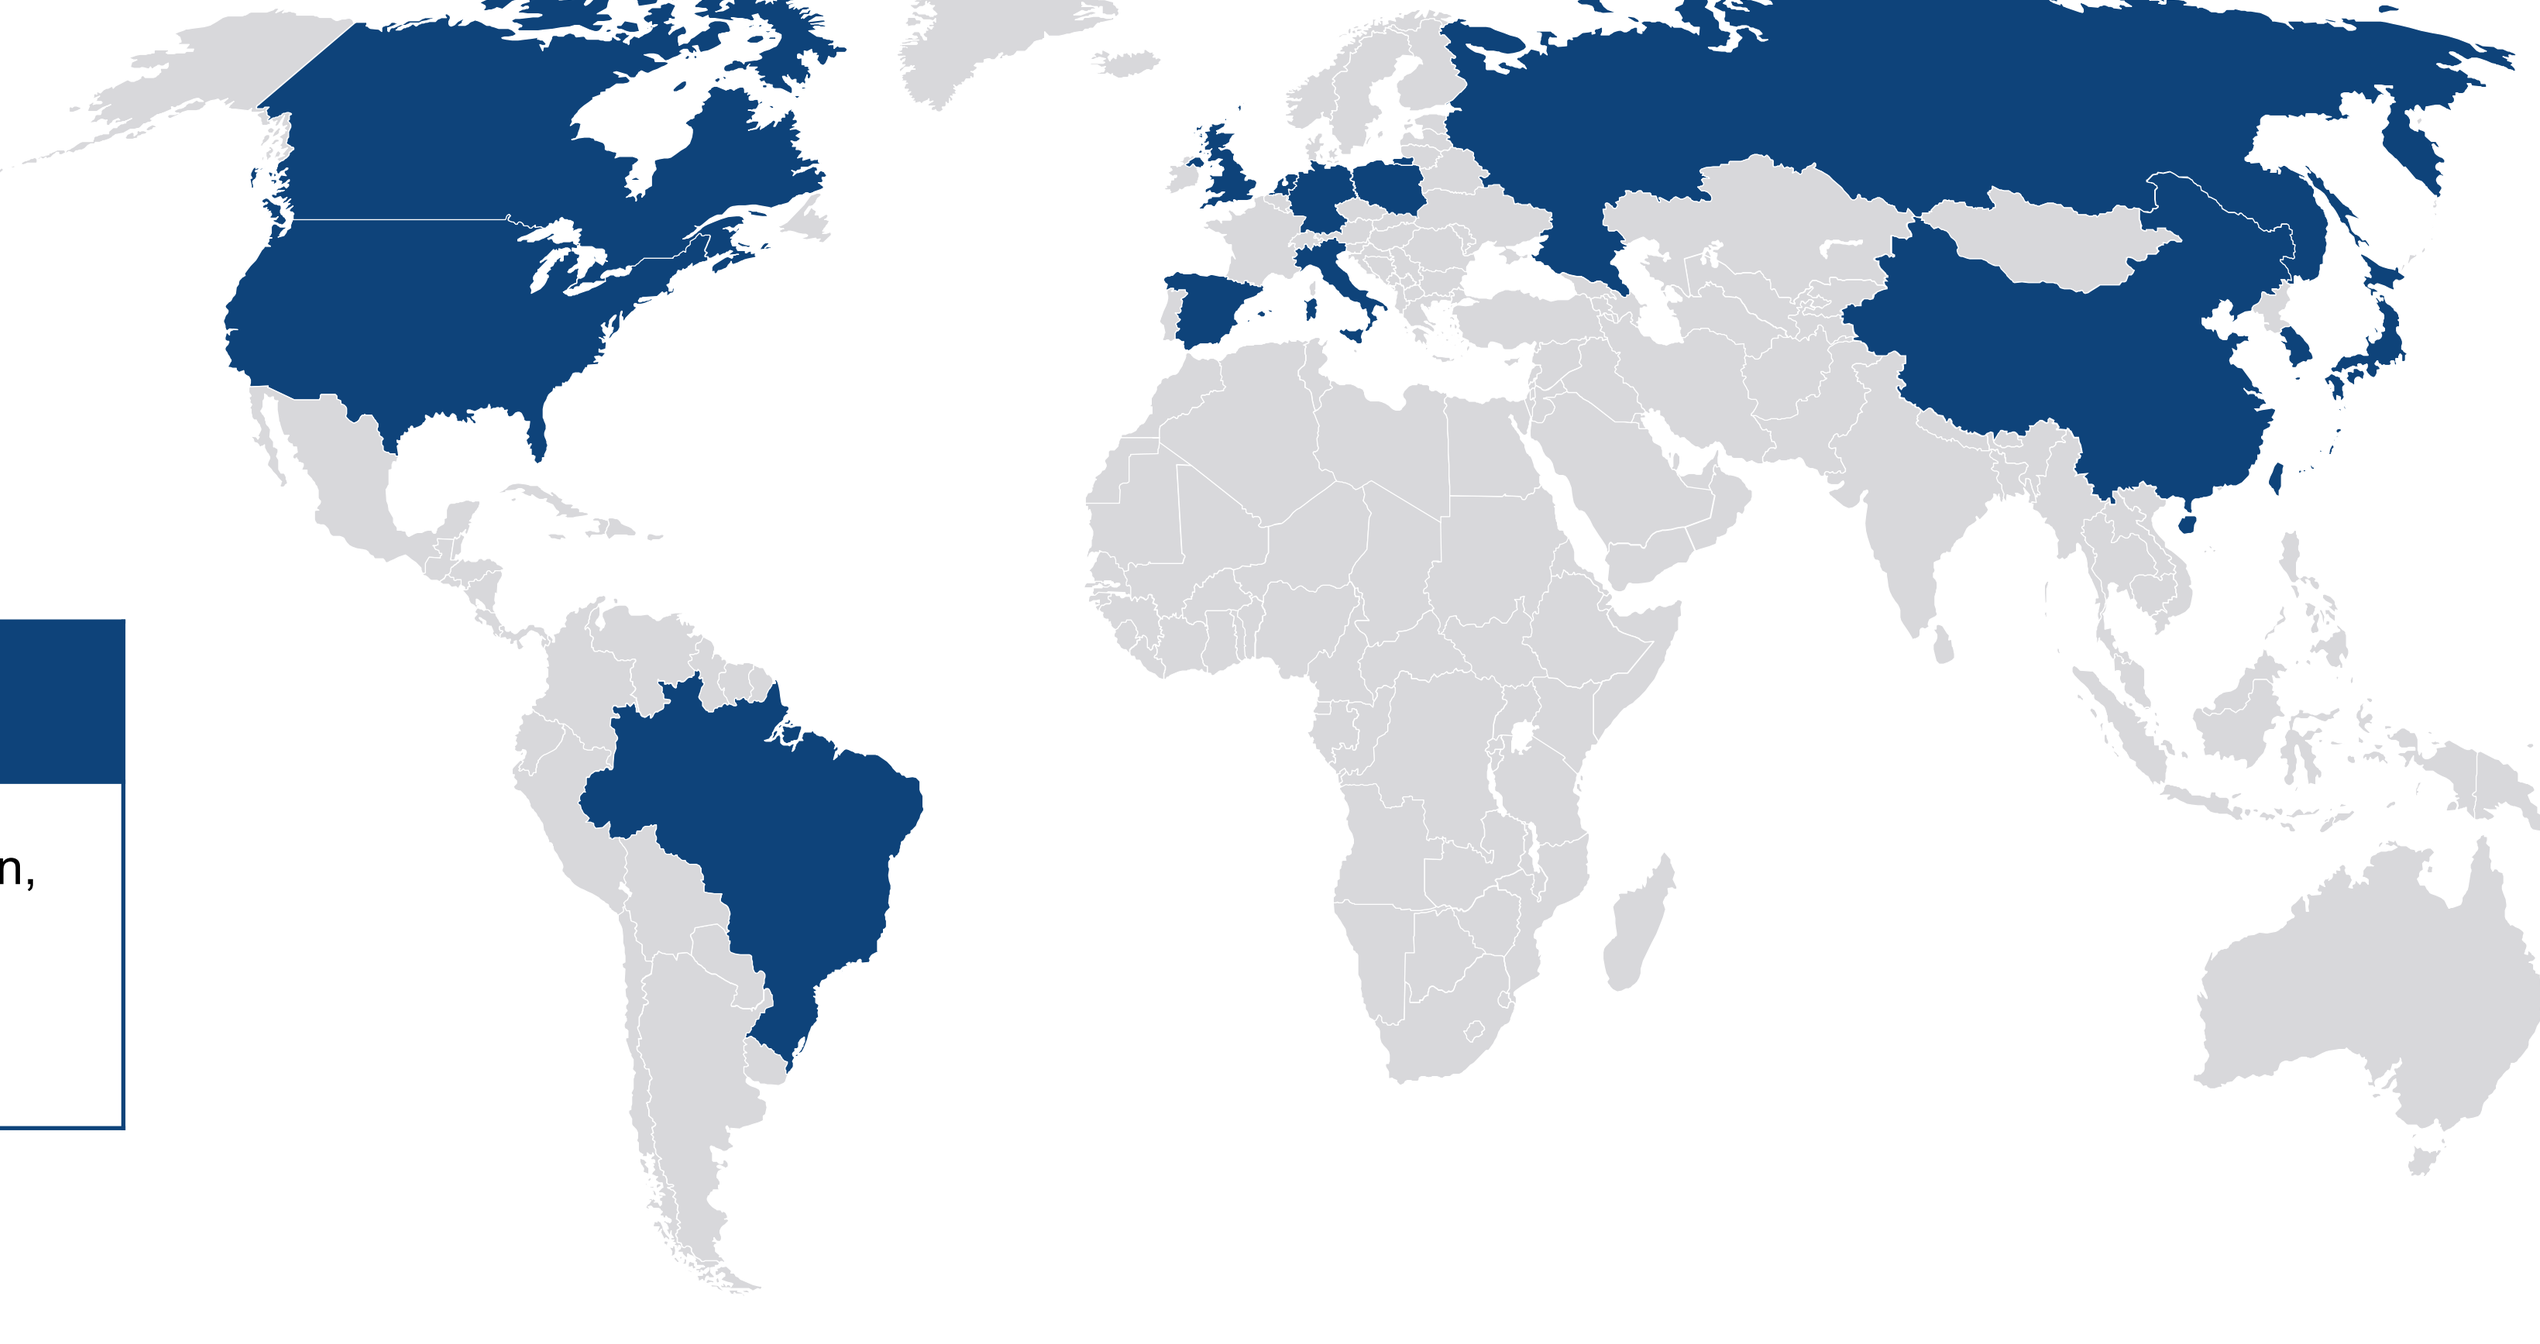

Countries with participating study sites

Brazil, Canada, China, Germany, Italy, Japan, Poland, Republic of Korea, Russia, Spain, Taiwan, the Netherlands, United States, United Kingdom

Key Inclusion Criteria

- Adults ≥18 years
- Pathologically documented unresectable, locally advanced or metastatic gastric, gastroesophageal junction, or esophageal adenocarcinoma
- **Parts 1, 2, and 3A only:** HER2+ status (IHC 3+ or IHC 2+/ISH+) as determined by local tissue assessment
- **Part 3B only:** HER2-low status (IHC 2+/ISH− or IHC 1+) as determined by local tissue assessment
- Previously treated (at least second line following trastuzumab-containing therapy) (**Part 1 only**) or treatment-naïve (**Parts 2 and 3 only**) unresectable or metastatic gastric, gastroesophageal junction, or esophageal adenocarcinoma
- Adjuvant therapies are permitted if all systemic therapy was completed and no progression occurred ≥6 months prior to unresectable or metastatic diagnosis
- Measurable target disease as assessed by the Investigator based on Response Evaluation Criteria In Solid Tumors version 1.1 (RECIST v1.1)
- Eastern Cooperative Oncology Group performance status of 0 or 1
 - **Part 3 only:** life expectancy ≥12 weeks
- Left ventricular ejection fraction ≥50% within 28 days of randomization (Part 2) or enrollment (Part 3)
- Adequate organ and bone marrow function within 14 days before randomization (Part 2) or enrollment (Part 3)

Key Exclusion Criteria

- Disease progression ≤6 months post completion of prior neoadjuvant/adjuvant therapy and prior to the diagnosis of unresectable or metastatic disease
- **Part 2 only:** multiple primary malignancies within 3 years
- **Part 3 only:**
 - History of another primary malignancy except malignancy treated with curative intent and with no known active disease ≥2 years before treatment assignment
 - Disease progression within 6 months of completing neoadjuvant/adjuvant anti-PD-L1, anti-PD-1, or anti-CTLA-4 treatment
- Lack of physiological integrity of the upper gastrointestinal tract, or malabsorption syndrome
- Known dihydropyrimidine dehydrogenase enzyme deficiency based on local or central laboratory testing
- Grade ≥2 peripheral neuropathy or hearing loss
- Uncontrolled intercurrent illness that would limit compliance with study requirements or substantially increase risk of incurring adverse events (AEs)
- Spinal cord compression, any leptomeningeal disease, or active central nervous system metastases
- Any autoimmune, connective tissue, or inflammatory disorders
- Medical history of myocardial infarction (MI) ≤6 months before treatment assignment (Part 2F) or randomization (Parts 2A to 2E), symptomatic congestive heart failure (New York Heart Association Class II–IV), unstable angina pectoris, clinically important cardiac arrhythmias, or recent (<6 months) cardiovascular event including stroke
 - **Part 3 only:** cardiomyopathy of any etiology, history of MI ≤12 months before treatment assignment
- Lung-specific, intercurrent, clinically significant illnesses, including any underlying pulmonary disorder and prior pneumonectomy
- History of non-infectious interstitial lung disease (ILD)/pneumonitis or current/suspected ILD/pneumonitis that cannot be ruled out by imaging at screening
- Active primary immunodeficiency, known human immunodeficiency virus, or active hepatitis B or C infection
- Pleural effusion, ascites, or pericardial effusion that requires drainage, peritoneal shunt, or cell-free and concentrated ascites reinfusion therapy

Key Study Endpoints for Parts 2 and 3

1° Primary endpoints

- Confirmed ORR as determined by the Investigator per RECIST v1.1

2° Secondary endpoints

- Investigator-assessed per RECIST v1.1
 - Disease control rate (DCR)
 - Duration of response (DOR)
 - Progression-free survival (PFS)
 - Overall survival (OS)
- Frequency of AEs and serious AEs, dose-limiting toxicities, and changes in laboratory parameters, vital signs, body weight, and electrocardiogram results
- Serum concentration of T-DXd, volrustomig (Part 3), total anti-HER2 antibody, and MAAA-1181
- Presence of anti-drug antibodies (ADAs) for T-DXd and volrustomig (Part 3)
- Comparison of ORR, DCR, DOR, PFS, and OS between participants based on local HER2 results and central HER2 results

Exploratory endpoints

- Serum concentration and presence of ADAs for pembrolizumab (Part 2)
- Candidate biomarker identification by mutation, protein expression analysis, and morphological assessment of tissue and blood

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials. Alternatively, please click on the link: https://bit.ly/47skhVv

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from ASCO GI® and the author of this poster.

Poster

References

1. Van Cutsem E, et al. *Gastric Cancer.* 2015;18:476–484
2. Nakada T, et al. *Chem Pharm Bull (Tokyo).* 2019;67:173–185
3. Ogitani Y, et al. *Clin Cancer Res.* 2016;22:5097–5108
4. Yamaguchi K, et al. *J Clin Oncol.* 2023;41:816–825
5. Van Cutsem E, et al. *Lancet Oncol.* 2023;24:744–756
6. Shen L, et al. *Ann Oncol.* 2023;34:S1542–S1543
7. Enhertu (fam-trastuzumab deruxtecan-nxki) highlights of prescribing information. 2022. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2022/761139s021lbl.pdf (Accessed November 20, 2023)
8. Enhertu (trastuzumab deruxtecan) summary of product characteristics. 2023. Available from: https://www.ema.europa.eu/en/documents/product-information/enhertu-epar-product-information_en.pdf (Accessed November 20, 2023)
9. Pharmaceuticals and Medical Devices Agency. Review report. August 21, 2020. Available from: https://www.pmda.go.jp/files/000238707.pdf (Accessed December 1, 2023)
10. Janjigian YY, et al. *Lancet.* 2023;402:2197–2208
11. Keytruda (pembrolizumab) highlights of prescribing information. 2023. Available from: https://www.accessdata.fda.gov/drugsatfda_docs/label/2023/125514s148lbl.pdf (Accessed December 1, 2023)
12. AstraZeneca. Press release. October 11, 2023. Available from: https://www.astrazeneca.com/media-centre/press-releases/2023/astrazeneca-advances-scientific-leadership-across-multiple-cancers-with-first-data-from-four-pivotal-phase-iii-trials-at-esmo.html (Accessed November 30, 2023)
13. Dovedi SJ, et al. *Cancer Discov.* 2021;11:1100–1117
14. Janjigian YY, et al. *J Clin Oncol.* 2022;40(4 Suppl.):295 (Abstract)

Acknowledgments

We thank the patients who are participating in this study, as well as their families and caregivers. This study is sponsored by AstraZeneca in collaboration with Daiichi Sankyo. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for trastuzumab deruxtecan (T-DXd; DS-8201). The authors would like to acknowledge Megan Winter from AstraZeneca for her contributions to the DG-03 study design.

Under the guidance of authors and in accordance with Good Publication Practice, medical writing and editorial support was provided by Abbie Dodd, BSc of Helios Medical Communications, and was funded by AstraZeneca.

Disclosures

Yelena Y Janjigian reports stock options with Inspirna; honoraria from AbbVie, AmerisourceBergen, AskGene Pharma, Inc., Arcus Biosciences, Astellas, AstraZeneca, Basilea Pharmaceutica, Bayer, Bristol Myers Squibb, Clinical Care Options, Daiichi Sankyo, Eli Lilly, Geneos Therapeutics, GlaxoSmithKline, Guardant Health, Inc., Imedex, Imugene, Inspirna, Lynx Health, Merck, Merck Serono, Mersana Therapeutics, Michael J. Hennessy Associates, Paradigm Medical Communications, PeerView Institute, Pfizer, Research To Practice, Seagen, Silverback Therapeutics, Zymeworks Inc.; research funding from AstraZeneca, Arcus Biosciences, Bayer, Bristol Myers Squibb, Cycle for Survival, Department of Defense, Eli Lilly, Fred's Team, Genentech/Roche, Inspirna, Merck, NCI, Transcenta.

This presentation is the intellectual property of the author/presenter. Contact them at janjigiy@mskcc.org for permission to reprint and/or distribute