4254

Kinetics of Complete Remission and Complete Remission Duration and Its Impact on Overall Survival and Event-Free Survival in QuANTUM-First

Pau Montesinos,[1] Alexander E. Perl,[2] Mikkael A. Sekeres,[3] Li Liu,[4] Yasser Mostafa Kamel,[4] Karima Imadalou,[4] Abderrahmane Laadem,[4] Arnaud Lesegretain,[4] Richard F. Schlenk,[5] Harry P. Erba,[6] Hervé Dombret[7]

[1]Hospital Universitari i Politècnic La Fe, Valencia, Spain; [2]Division of Hematology/Oncology, University of Pennsylvania, Philadelphia, PA, USA; [3]Sylvester Cancer Center, University of Miami Health System, Miami, FL, USA; [4]Daiichi Sankyo, Basking Ridge, NJ, USA; [5]National Center of Tumor Diseases Trial Center, German Cancer Research Center, Department of Internal Medicine V, Heidelberg University Hospital, Heidelberg, Germany; [6]Duke Cancer Institute, Durham, NC, USA; [7]Saint Louis Hospital, University of Paris, Paris, France

BACKGROUND

- The phase 3 study QuANTUM-First (NCT02668653) was a randomized placebo-controlled study of quizartinib, a highly potent and selective type 2 FMS-like tyrosine kinase 3–internal tandem duplication (*FLT3*-ITD) inhibitor, in adult patients with newly diagnosed *FLT3*-ITD–positive acute myeloid leukemia (AML) up to age 75 years in combination with standard chemotherapy and/or allogeneic hematopoietic cell transplantation (allo-HCT), followed by continuation monotherapy with quizartinib or placebo for up to 36 cycles (~3 years)[1]
 - Quizartinib treatment reduced the relative risk of death by 22.4% versus placebo (hazard ratio [HR], 0.776; 95% CI, 0.615-0.979; *P*=0.0324)[1] (**Figure 1**)
 - Although rates of complete remission (CR) were similar between quizartinib and placebo (54.9% [95% CI, 48.7-60.9] and 55.4% [95% CI, 49.2-61.4], respectively), median duration of CR for quizartinib versus placebo was longer (38.6 months [95% CI, 21.9-not estimable (NE)] versus 12.4 months [95% CI, 8.8-22.7]; HR, 0.621 [95% CI, 0.451-0.857])[1] (**Figure 2**)
- Quizartinib has recently been approved by the United States Food and Drug Administration (FDA),[2,3] the Japanese health agency,[4] and the European Medicines Agency.[5] Based on QuANTUM-First data,[1] quizartinib is approved in combination with chemotherapy across induction, consolidation, and as maintenance monotherapy (but not after allo-HCT in the United States) for the treatment of adult patients with newly diagnosed *FLT3*-ITD–positive AML

Figure 1. OS (Primary Endpoint) in QuANTUM-First[1]

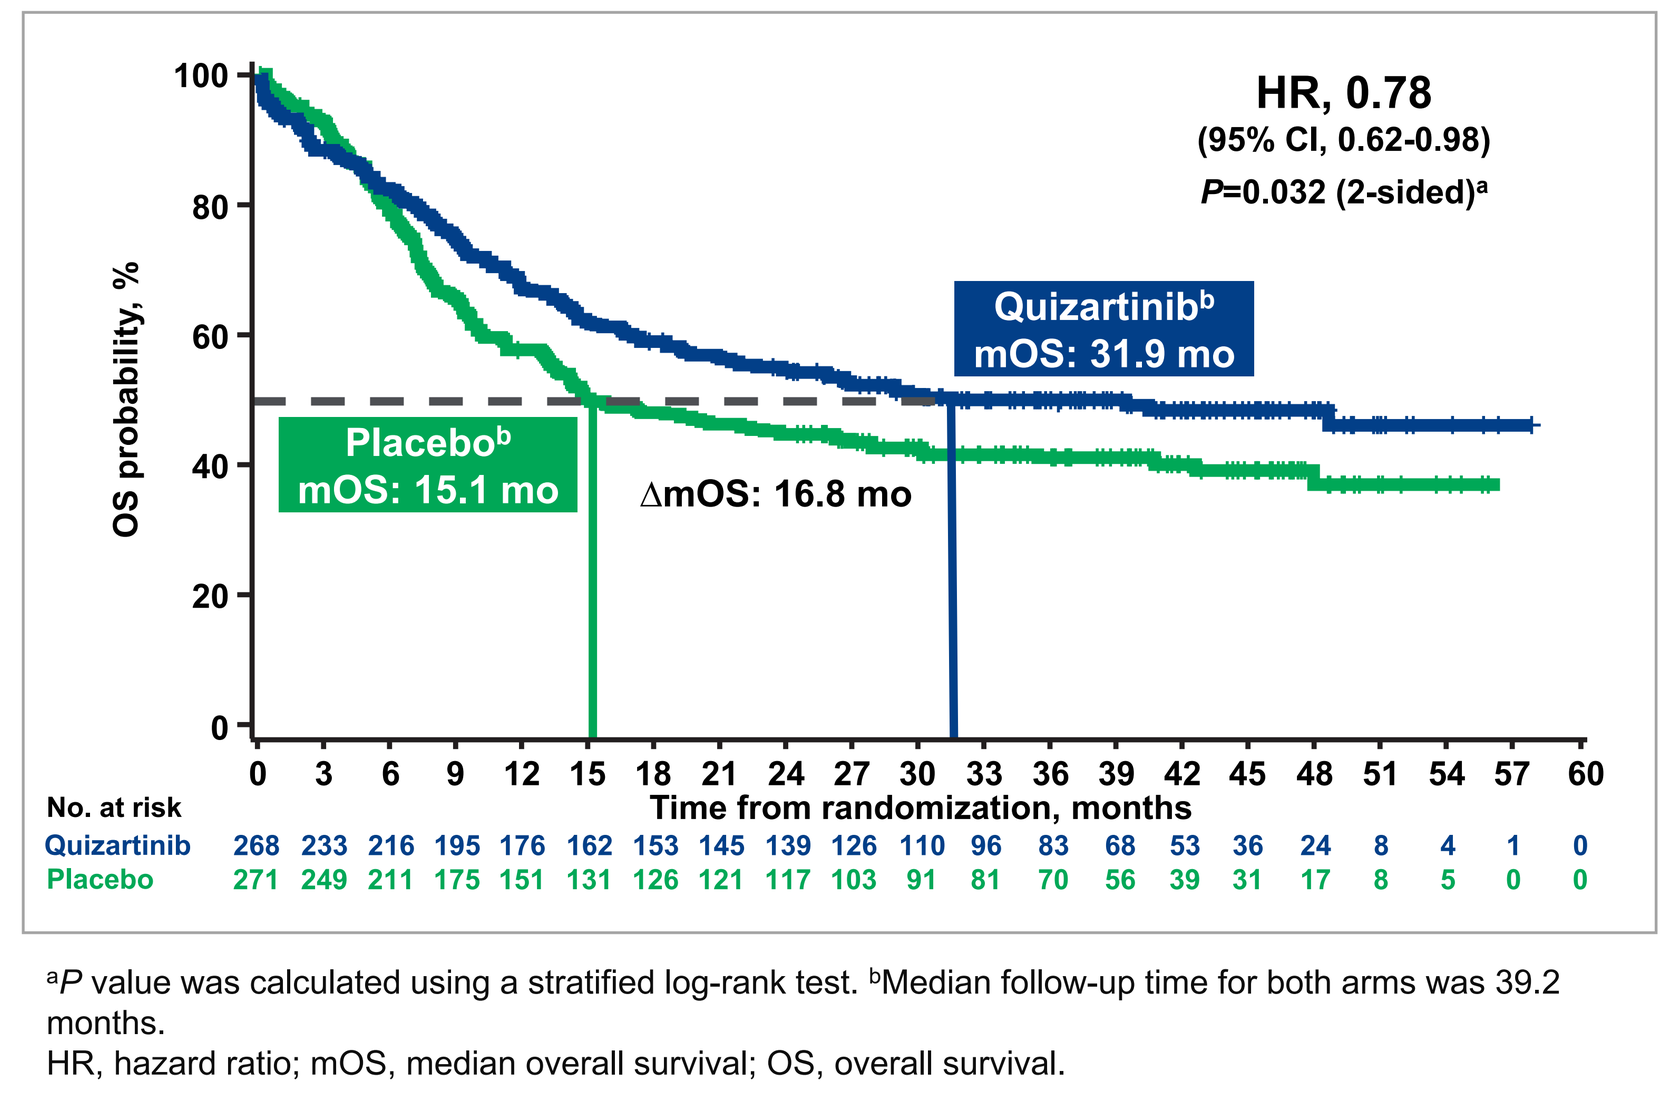
HR, 0.78
(95% CI, 0.62-0.98)
P=0.032 (2-sided)a
Quizartinibb
mOS: 31.9 mo
Placebob
mOS: 15.1 mo
ΔmOS: 16.8 mo
OS probability, %
Time from randomization, months
No. at risk
Quizartinib 268 233 216 195 176 162 153 145 139 126 110 96 83 68 53 36 24 8 4 1 0
Placebo 271 249 211 175 151 131 126 121 117 103 91 81 70 56 39 31 17 8 5 0 0

[a]*P* value was calculated using a stratified log-rank test. [b]Median follow-up time for both arms was 39.2 months.
HR, hazard ratio; mOS, median overall survival; OS, overall survival.

Figure 2. Duration of CR in Patients Who Achieved CR During Induction[1]

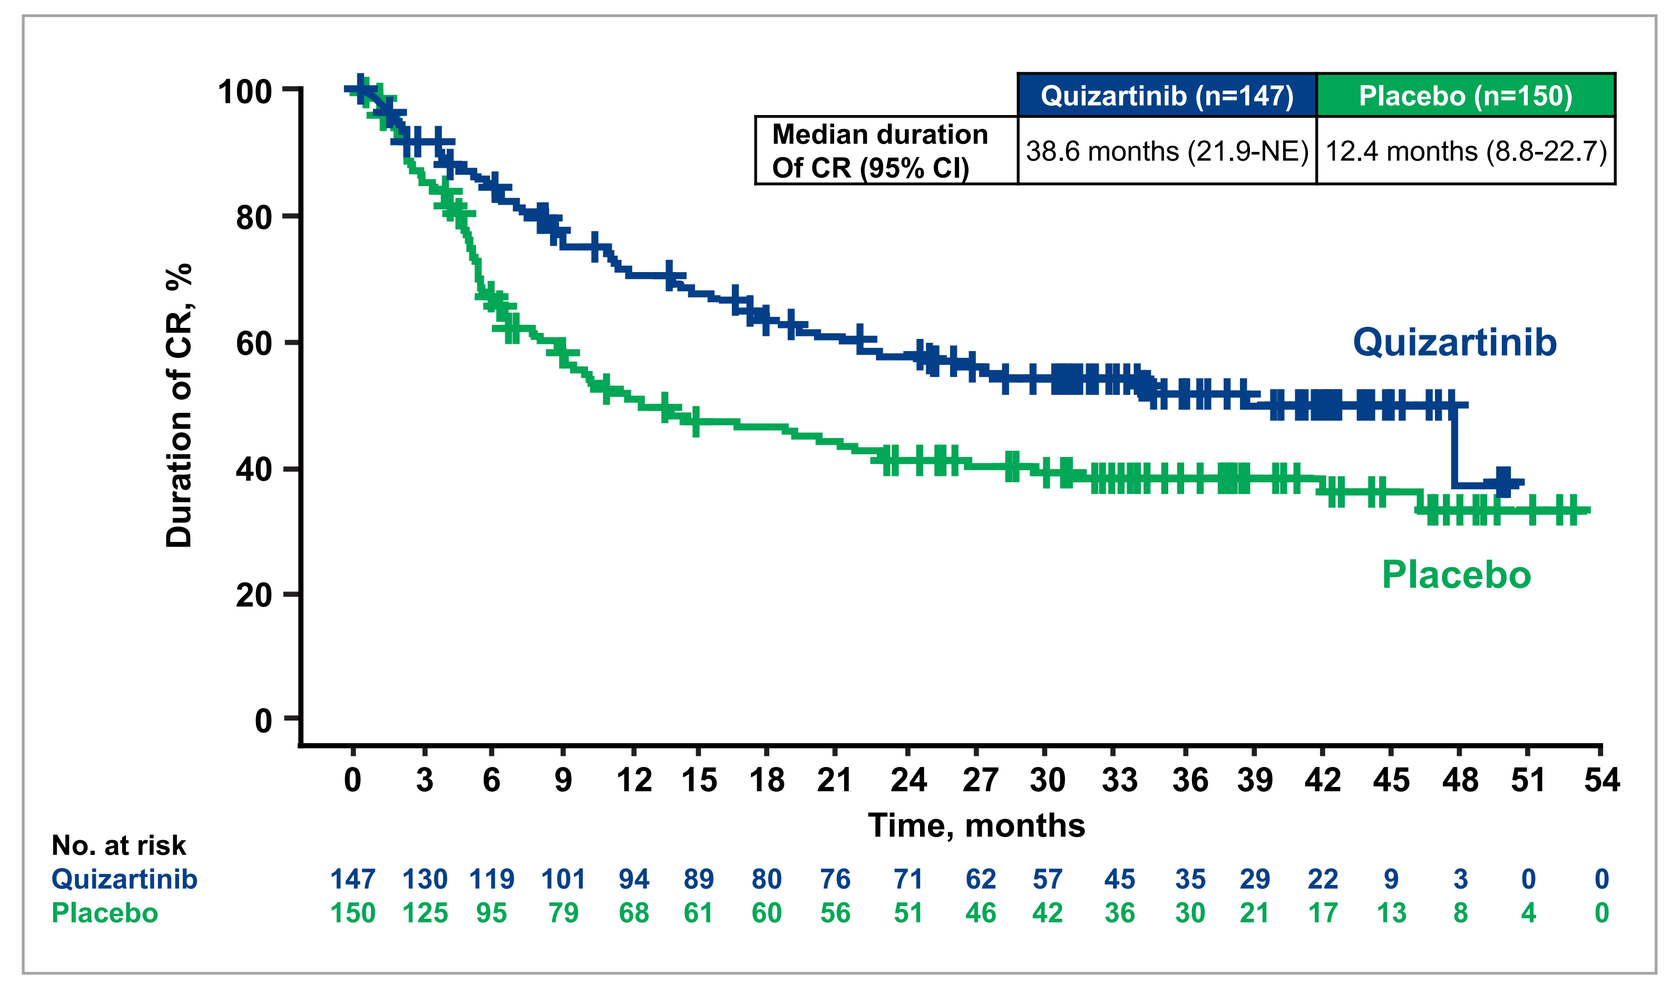
Quizartinib (n=147)
Placebo (n=150)
Median duration Of CR (95% CI)
38.6 months (21.9-NE)
12.4 months (8.8-22.7)
Quizartinib
Placebo
Duration of CR, %
Time, months
No. at risk
Quizartinib 147 130 119 101 94 89 80 76 71 62 57 45 35 29 22 9 3 0 0
Placebo 150 125 95 79 68 61 60 56 51 46 42 36 30 21 17 13 8 4 0

CR, complete remission; NE, not estimable.

- In these post hoc analyses, we asked whether the duration of CR may be a key driver for the overall survival (OS) benefit provided by quizartinib in the QuANTUM-First study

OBJECTIVES

- To assess the impact of duration of CR on OS
- To assess the kinetics of CR achievement over time after induction

METHODS

- A detailed description of the QuANTUM-First study has been previously published (**Figure 3**)[1]
- Eligible adult patients (aged 18-75 years) with newly diagnosed *FLT3*-ITD–positive AML were randomized 1:1 to receive standard induction chemotherapy with either quizartinib (40 mg/day) or placebo and stratified by region (EU, North America, or Asia/Australia/South America), age (<60 years, ≥60 years), and white blood cell (WBC; <40×10⁹/L, ≥40×10⁹/L) count at diagnosis
- Quizartinib was administered from days 8 to 21 (40 mg) in each cycle
- The induction period could include 1 or 2 cycles
- Patients who achieved CR or CR with incomplete hematologic recovery (CRi) received ≤4 cycles of high-dose cytarabine plus quizartinib (40 mg/day) or placebo and/or allo-HCT, followed by 36 cycles (~3 years) of continuation monotherapy with quizartinib (30-60 mg/day) or placebo
- OS was the primary endpoint
- EFS and rates of CR and composite complete remission (CRc) were secondary endpoints
- Duration of CR was an exploratory endpoint
- Median OS was estimated based on the Kaplan-Meier method; HR with 95% CI was estimated using a stratified Cox proportional hazards model
- The impact of duration of CR and CRc on OS was analyzed by extended Cox regression model stratified by region, age, and WBC count at diagnosis and included treatment group and CR or CRc duration (time-dependent covariate)
 - CR (or CRc) duration was 0 for patients who did not achieve CR (or CRc), 1 for patients with CR (or CRc) throughout the duration of sustained CR (or CRc), and 0 for patients with CR (or CRc) after they relapsed
- The impact of duration of CR on OS was analyzed by a multistate model[6]
 - All patients started in the state "Randomization" at time 0. All possible transitions a patient is at risk for were assessed
 - A patient remains in the current state until the next event occurs or censoring (at the end of follow-up) occurs
- For both the extended Cox regression model and multistate model, the relative differences between the treatment arms are expressed in terms of HR (95% CI, range)

Figure 3. QuANTUM-First Phase 3 Study: Quizartinib Plus Standard Induction Chemotherapy and Consolidation, Followed by Single-Agent Quizartinib or Placebo

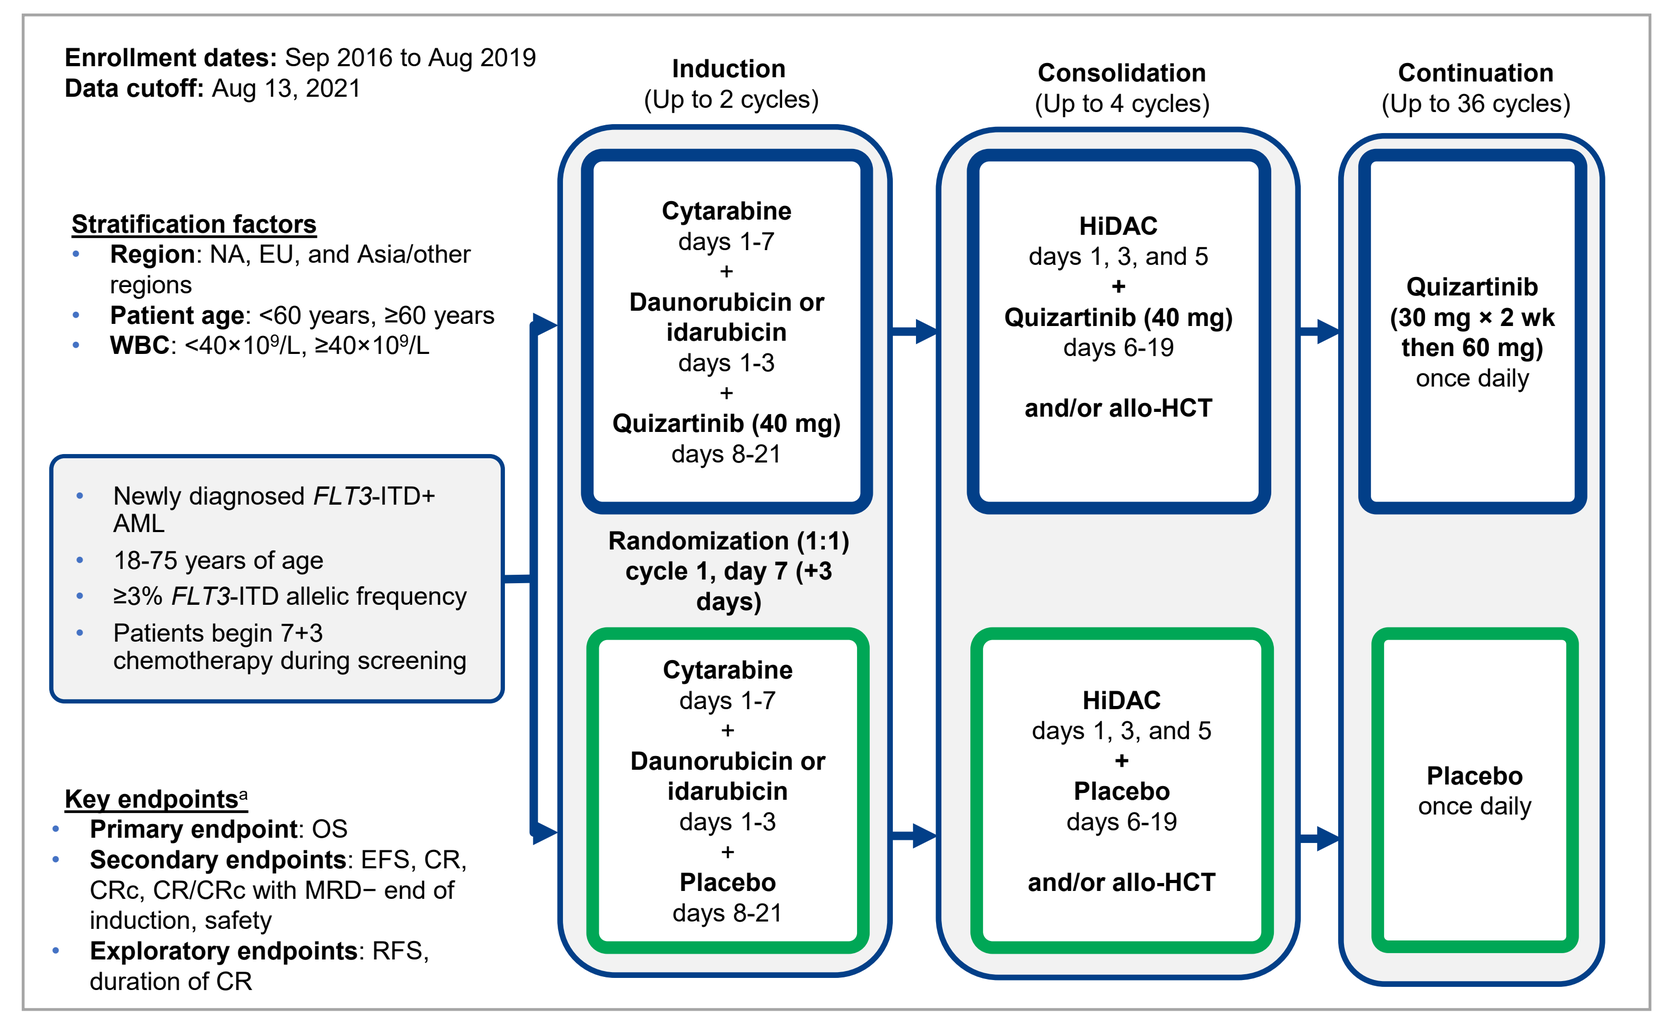
Enrollment dates: Sep 2016 to Aug 2019
Data cutoff: Aug 13, 2021
Stratification factors
Region: NA, EU, and Asia/other regions
Patient age: <60 years, ≥60 years
WBC: <40×10⁹/L, ≥40×10⁹/L
Newly diagnosed FLT3-ITD+ AML
18-75 years of age
≥3% FLT3-ITD allelic frequency
Patients begin 7+3 chemotherapy during screening
Key endpointsa
Primary endpoint: OS
Secondary endpoints: EFS, CR, CRc, CR/CRc with MRD− end of induction, safety
Exploratory endpoints: RFS, duration of CR
Induction (Up to 2 cycles)
Cytarabine days 1-7 + Daunorubicin or idarubicin days 1-3 + Quizartinib (40 mg) days 8-21
Randomization (1:1) cycle 1, day 7 (+3 days)
Cytarabine days 1-7 + Daunorubicin or idarubicin days 1-3 + Placebo days 8-21
Consolidation (Up to 4 cycles)
HiDAC days 1, 3, and 5 + Quizartinib (40 mg) days 6-19 and/or allo-HCT
HiDAC days 1, 3, and 5 + Placebo days 6-19 and/or allo-HCT
Continuation (Up to 36 cycles)
Quizartinib (30 mg × 2 wk then 60 mg) once daily
Placebo once daily

ClinicalTrials.gov identifier: NCT02668653.
[a]A hierarchical testing procedure was used to test the primary endpoint of OS, followed by EFS, CR, CRc, CR with *FLT3*-ITD MRD negativity, and CRc with *FLT3*-ITD MRD negativity.
Allo-HCT, allogeneic hematopoietic cell transplantation; AML, acute myeloid leukemia; CR, complete remission; CRc, composite complete remission; DoCR, duration of complete remission; EFS, event-free survival; EU, European Union; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; HiDAC, high-dose cytarabine; MRD, measurable residual disease; NA, North America; OS, overall survival; RFS, relapse-free survival; WBC, white blood cell.

RESULTS

Patient disposition

- In QuANTUM-First, 539 patients were randomized to receive either quizartinib (n=268) or placebo (n=271)
- A total of 297 patients achieved CR based on independent review committee assessment after 1-2 courses of induction as follows: 147 (54.9%) patients on quizartinib and 150 (55.4%) on placebo
- Among patients who achieved CR after induction, 130 (88.4%) on quizartinib and 140 (93.3%) on placebo entered consolidation, while 94 (63.9%) on quizartinib and 72 (48%) on placebo entered continuation (**Figure 4**)

Figure 4. Diagram of CR Patient Flow

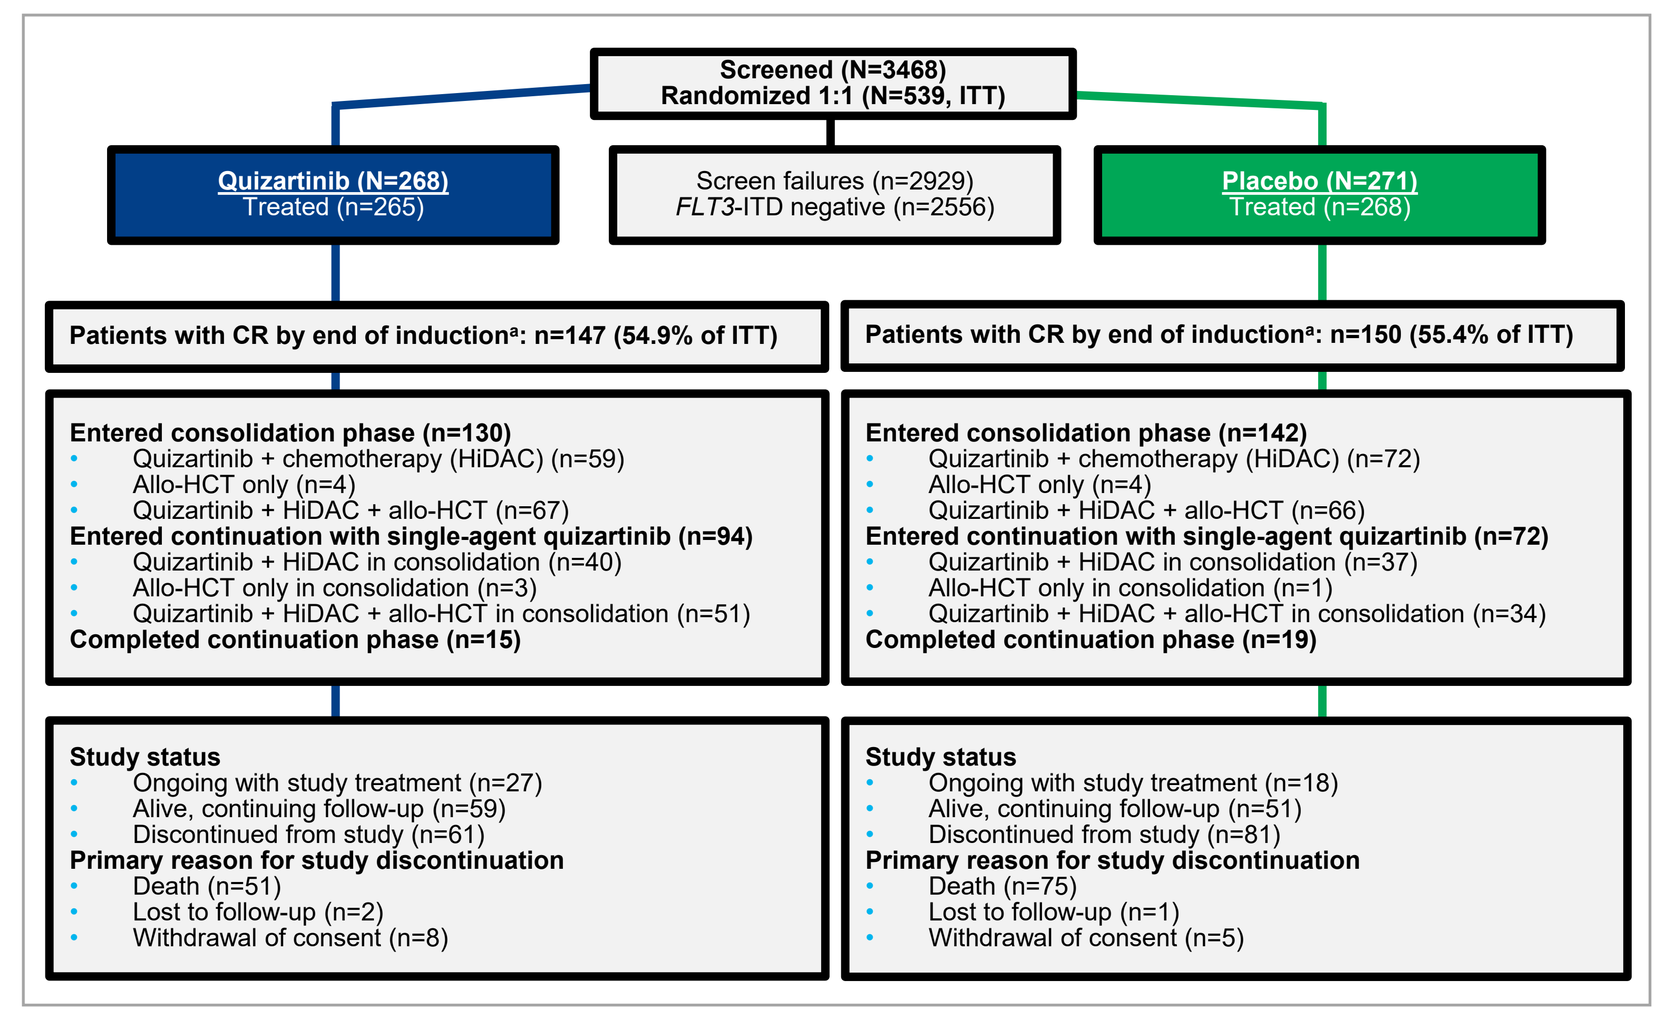
Screened (N=3468)
Randomized 1:1 (N=539, ITT)
Quizartinib (N=268)
Treated (n=265)
Screen failures (n=2929)
FLT3-ITD negative (n=2556)
Placebo (N=271)
Treated (n=268)
Patients with CR by end of inductiona: n=147 (54.9% of ITT)
Patients with CR by end of inductiona: n=150 (55.4% of ITT)
Entered consolidation phase (n=130)
Quizartinib + chemotherapy (HiDAC) (n=59)
Allo-HCT only (n=4)
Quizartinib + HiDAC + allo-HCT (n=67)
Entered continuation with single-agent quizartinib (n=94)
Quizartinib + HiDAC in consolidation (n=40)
Allo-HCT only in consolidation (n=3)
Quizartinib + HiDAC + allo-HCT in consolidation (n=51)
Completed continuation phase (n=15)
Entered consolidation phase (n=142)
Quizartinib + chemotherapy (HiDAC) (n=72)
Allo-HCT only (n=4)
Quizartinib + HiDAC + allo-HCT (n=66)
Entered continuation with single-agent quizartinib (n=72)
Quizartinib + HiDAC in consolidation (n=37)
Allo-HCT only in consolidation (n=1)
Quizartinib + HiDAC + allo-HCT in consolidation (n=34)
Completed continuation phase (n=19)
Study status
Ongoing with study treatment (n=27)
Alive, continuing follow-up (n=59)
Discontinued from study (n=61)
Primary reason for study discontinuation
Death (n=51)
Lost to follow-up (n=2)
Withdrawal of consent (n=8)
Study status
Ongoing with study treatment (n=18)
Alive, continuing follow-up (n=51)
Discontinued from study (n=81)
Primary reason for study discontinuation
Death (n=75)
Lost to follow-up (n=1)
Withdrawal of consent (n=5)

[a]After 1-2 courses of induction.
Allo-HCT, allogeneic hematopoietic cell transplantation; CR, complete remission; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; HiDAC, high-dose cytarabine; ITT, intent-to-treat.

Baseline demographic and disease characteristics

- Patient baseline characteristics were well balanced between quizartinib versus placebo (**Table 1**)
- Disease characteristics were as expected for a population with newly diagnosed *FLT3*-ITD AML who have a high burden of aggressive disease

Table 1. Baseline Demographics and Disease Characteristics of Patients With CR by End of Induction

Patient characteristics	Quizartinib (N=147)	Placebo (N=150)
Age		
Median (range), years	56.0 (23-75)	55.5 (20-74)
<60 years, n (%)	90 (61.2)	90 (60.0)
≥60 years, n (%)	57 (38.8)	60 (40.0)
60-64 years, n (%)	22 (15.0)	25 (16.7)
≥65 years, n (%)	35 (23.8)	35 (23.3)
Sex, n (%)		
Male	65 (44.2)	60 (40.0)
Female	82 (55.8)	90 (60.0)
ECOG PS, n (%)[a]		
0	52 (35.4)	55 (36.7)
1	68 (46.3)	75 (50.0)
2	27 (18.4)	20 (13.3)
Mutated *NPM1*, n (%)	98 (66.7)	97 (64.7)
Mutated *CEBPA*, n (%)	38 (25.9)	35 (23.3)
***FLT3*-ITD/total *FLT3*, n (%)[b,c]**		
≥3% to ≤25%	46 (31.3)	50 (33.3)
>25% to ≤50%	82 (55.8)	83 (55.3)
>50%	19 (12.9)	17 (11.3)
>25%	101 (68.7)	100 (66.7)
WBC count at diagnosis of AML, n (%)		
<40×10⁹/L	65 (44.2)	76 (50.7)
≥40×10⁹/L	82 (55.8)	74 (49.3)

[a]One patient in the placebo group was missing an ECOG PS. [b]VAF was assessed using central laboratory testing. [c]One patient with unknown *FLT3*-ITD/total *FLT3* was positive per local laboratory testing.
AML, acute myeloid leukemia; *CEBPA*, CCAAT enhancer binding protein alpha; ECOG PS, Eastern Cooperative Oncology Group performance status; *FLT3*-ITD, FMS-like tyrosine kinase 3–internal tandem duplication; *NPM1*, nucleophosmin 1; VAF, variant allele frequency.

OS in patients who achieved CR (A) and patients who did not achieve CR

- Median OS in patients who achieved CR was not reached (95% CI, 48.6 months-NE) in the quizartinib arm versus 35.4 months (95% CI, 15.1-NE) in the placebo arm (HR, 0.572 [95% CI, 0.400-0.816]; **Figure 5A**)
- Median OS in patients who did not achieve CR was 10.6 months (95% CI, 8.0-14.6) in the quizartinib arm versus 9.6 months (95% CI, 7.4-14.7) in the placebo arm (HR, 1.013 [95% CI, 0.746-1.374]; **Figure 5B**)
- Achievement of CR in the quizartinib arm drives longer survival

Figure 5. Kaplan-Meier Plot of OS in Patients Who Achieved CR (A) and in Those Who Did Not Achieve CR (B) by IRC Assessment

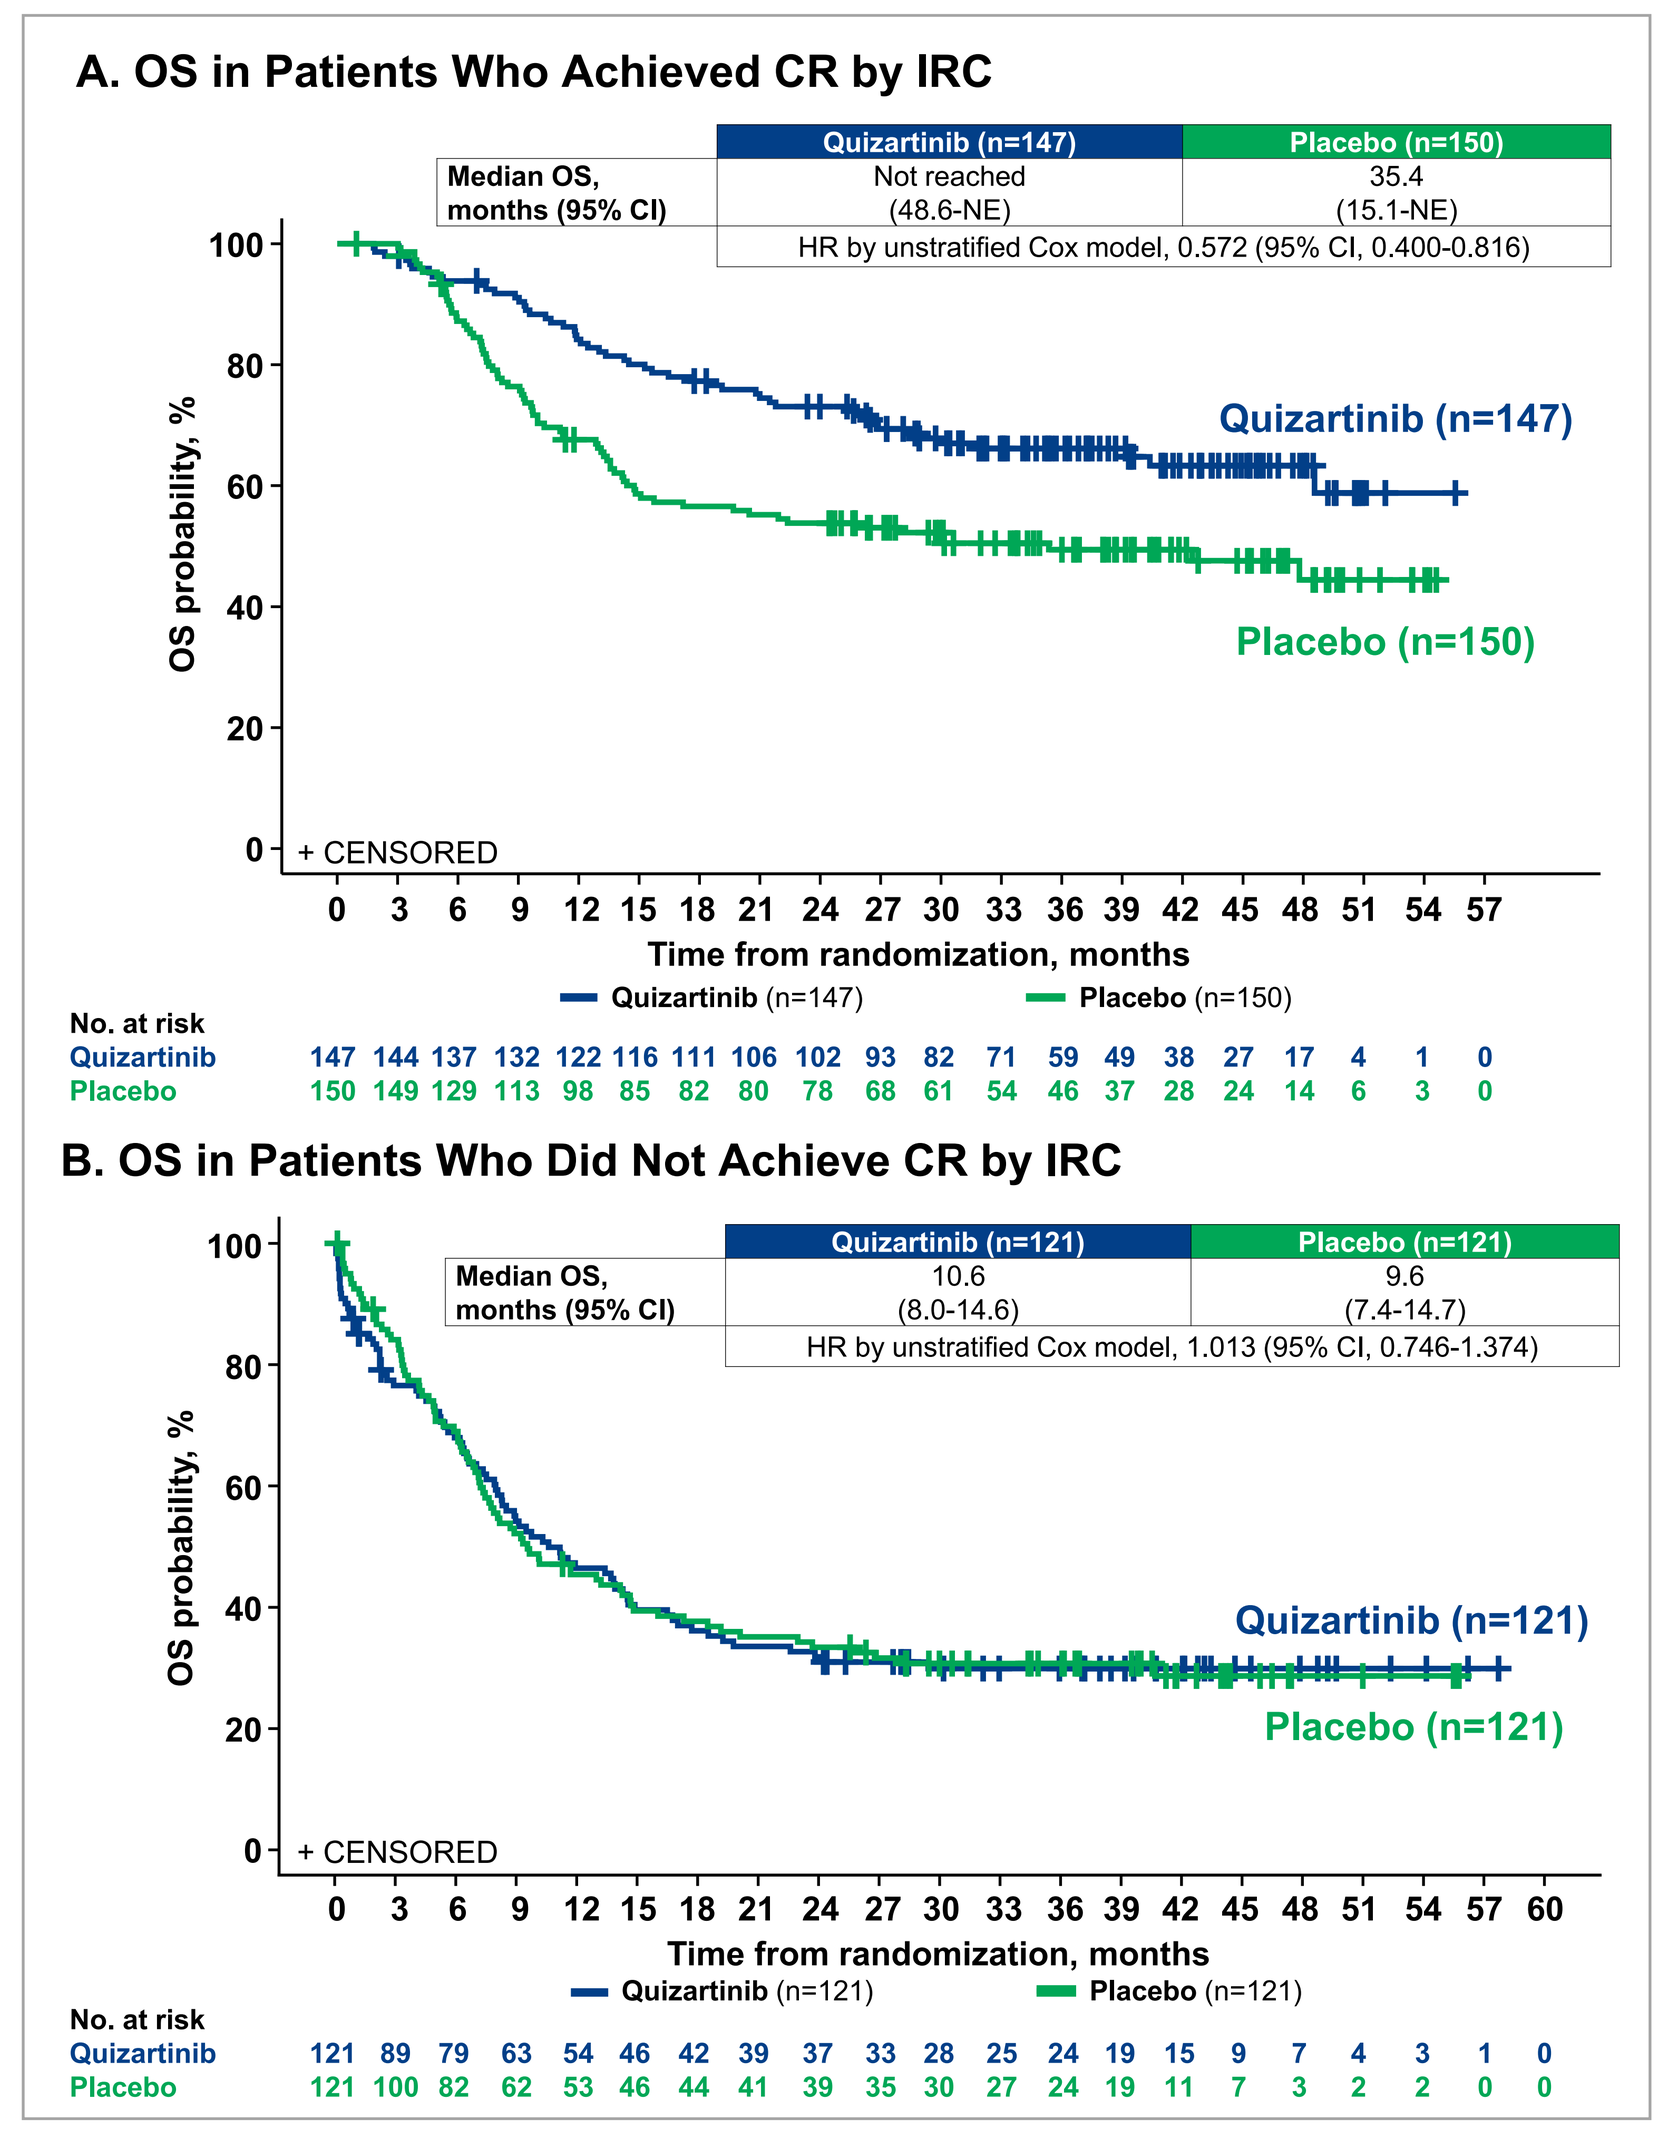
A. OS in Patients Who Achieved CR by IRC
Quizartinib (n=147)
Placebo (n=150)
Median OS, months (95% CI)
Not reached (48.6-NE)
35.4 (15.1-NE)
HR by unstratified Cox model, 0.572 (95% CI, 0.400-0.816)
+ CENSORED
OS probability, %
Time from randomization, months
No. at risk
Quizartinib 147 144 137 132 122 116 111 106 102 93 82 71 59 49 38 27 17 4 1 0
Placebo 150 149 129 113 98 85 82 80 78 68 61 54 46 37 28 24 14 6 3 0
B. OS in Patients Who Did Not Achieve CR by IRC
Quizartinib (n=121)
Placebo (n=121)
Median OS, months (95% CI)
10.6 (8.0-14.6)
9.6 (7.4-14.7)
HR by unstratified Cox model, 1.013 (95% CI, 0.746-1.374)
No. at risk
Quizartinib 121 89 79 63 54 46 42 39 37 33 28 25 24 19 15 9 7 4 3 1 0
Placebo 121 100 82 62 53 46 44 41 39 35 30 27 24 19 11 7 3 2 2 0 0

IRC, independent review committee; HR, hazard ratio; NE, not estimable; OS, overall survival.

Predictive factors for OS

- A multivariable extended Cox regression was conducted, stratified by region, age, and WBC count at diagnosis and included treatment group and CR duration status as a time-dependent covariate (**Figure 6**)
- Based on the extended Cox regression model, CR duration status covariate was strongly predictive for OS (HR, 0.156 [95% CI, 0.113-0.216]; nominal *P*<0.0001; **Figure 6A**)
 - Similar results were found when CRc duration was used as time-dependent variable (**Figure 6B**)
- The HR for quizartinib versus placebo effect on OS was higher in the model with the time-dependent covariate CR duration status (0.851 [95% CI, 0.672-1.076]; **Figure 6A**), as well as in the model with the time-dependent covariate CRc duration status (1.004 [95% CI, 0.790-1.276]; **Figure 6B**), compared with the HR reported in the primary OS analysis of 0.776 (95% CI, 0.615-0.979)[1]
- These data imply that a substantial fraction of OS benefit provided by quizartinib in the QuANTUM-First study was mediated through its effects on achieving a durable CR or CRc

Figure 6. Extended Cox Regression Analysis of OS Stratified by Region, Age, and WBC With CR (A) or CRc (B) Duration Status as Time-Dependent Variables

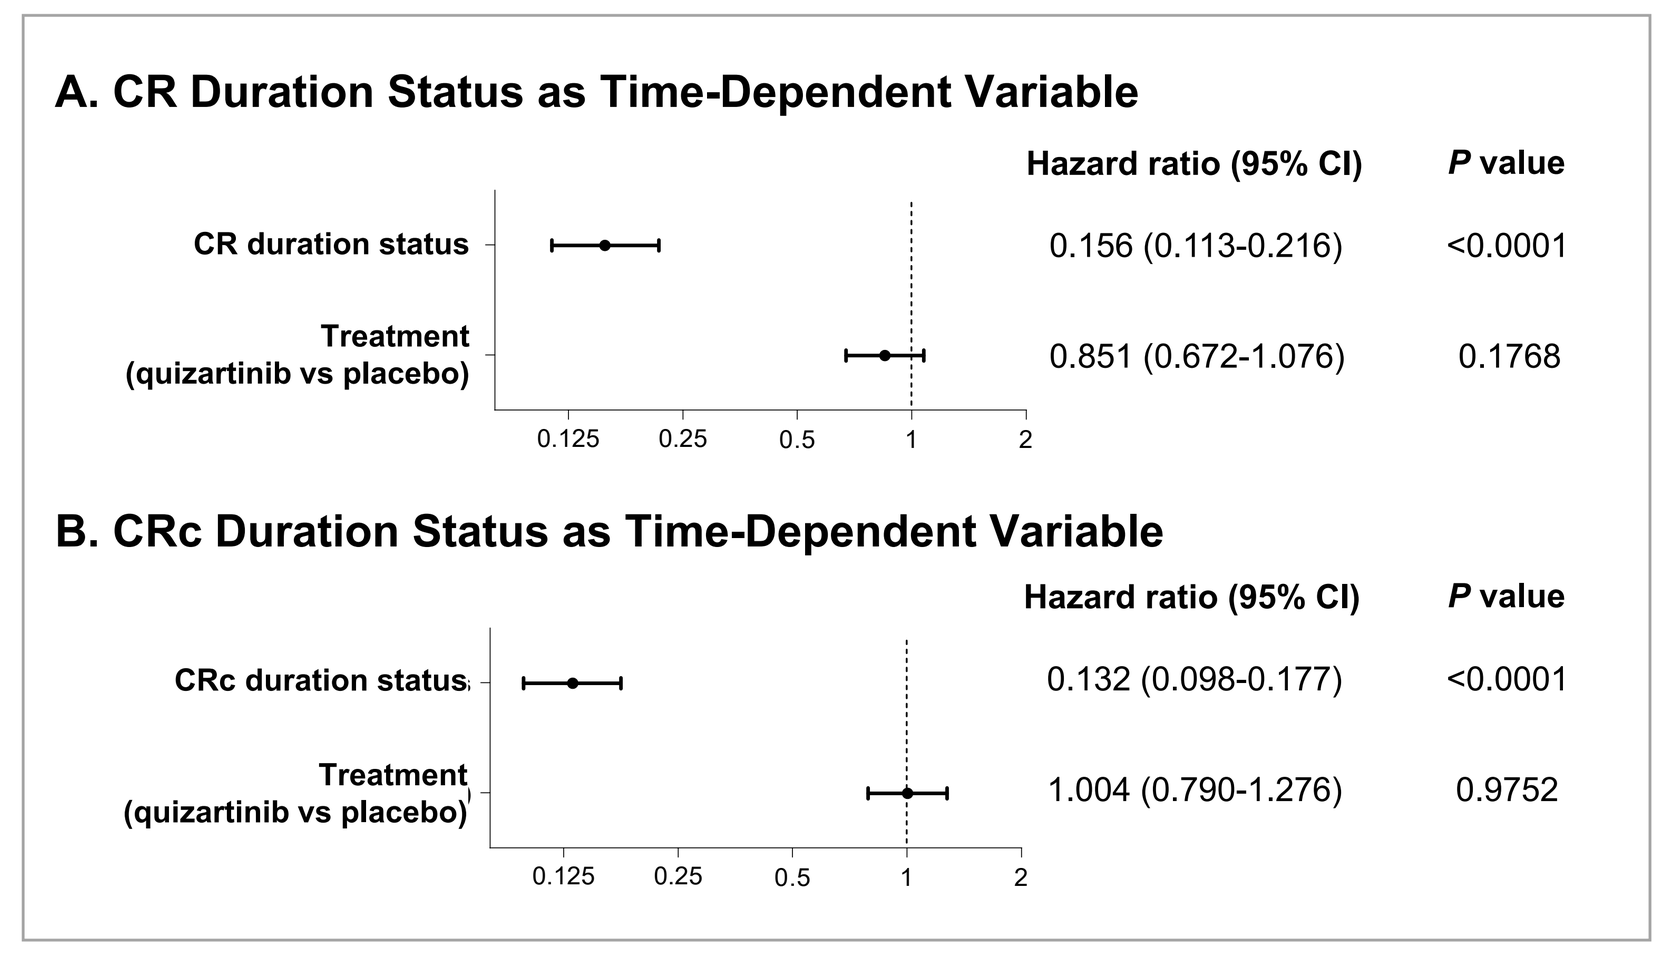
A. CR Duration Status as Time-Dependent Variable
Hazard ratio (95% CI)
P value
CR duration status
0.156 (0.113-0.216)
<0.0001
Treatment (quizartinib vs placebo)
0.851 (0.672-1.076)
0.1768
B. CRc Duration Status as Time-Dependent Variable
CRc duration status
0.132 (0.098-0.177)
<0.0001
Treatment (quizartinib vs placebo)
1.004 (0.790-1.276)
0.9752

Post hoc analysis.
CR, complete remission; CRc, composite complete remission; OS, overall survival; WBC, white blood cell.

Transition probabilities

- A multistate model was used to estimate the transition probabilities starting from CR (**Figure 7A**) and from randomization (**Figure 7B**)
- Based on this model, a higher percentage of patients stayed in CR and a lower percentage of deaths occurred in the quizartinib arm versus the placebo arm (**Figure 7**)
- Quizartinib was associated with lower risk of relapse after achievement of CR versus placebo (HR, 0.517; 95% CI, 0.331-0.807)
- These data further support the notion that a substantial fraction of OS benefit provided by quizartinib in the QuANTUM-First study was mediated through its effects on achieving a durable CR

Figure 7. Multistate Model: Transition Probabilities to All States Starting From CR (A)[a] and From Randomization (B)[b]

At each point in time, the distance between 2 adjacent curves represents the probability of being in the corresponding state, conditional on being in the state "CR" (A) or in the state "Randomization" (B) at time 0. The probability of being in an intermediate state can both increase and decrease over time, while the probability of absorbing (death) states can only increase over time.

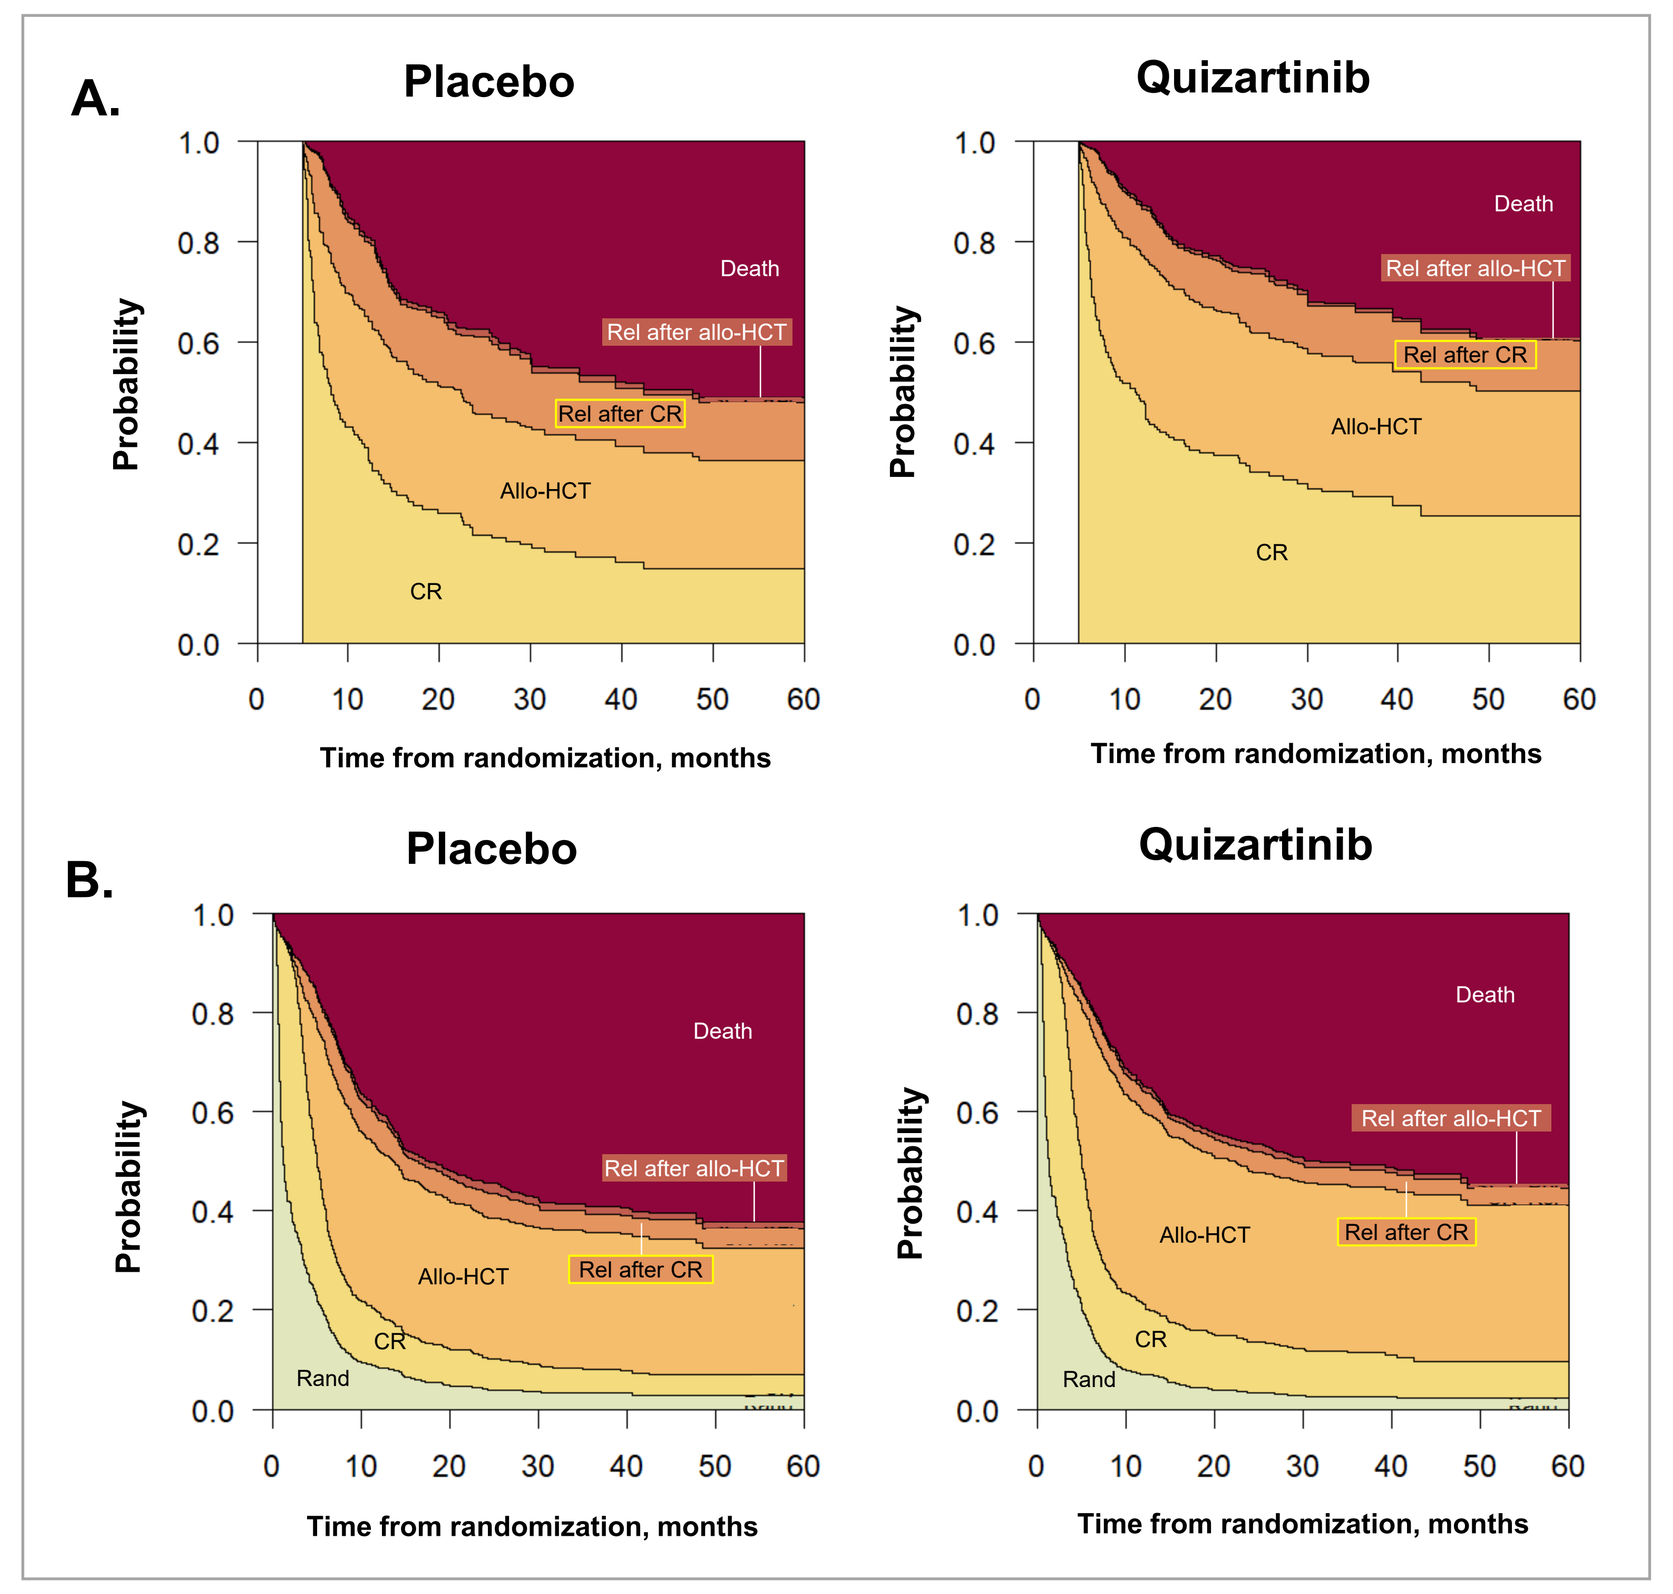
A.
Placebo
Quizartinib
Death
Rel after allo-HCT
Rel after CR
Allo-HCT
CR
Probability
Time from randomization, months
B.
Placebo
Quizartinib
Rand

[a]All patients start in the state "CR" at time 0. [b]All patients start in the state "Randomization" at time 0. All possible transitions a patient is at risk for are color-coded. A patient remains in the current state until the next event occurs or censoring (at the end of follow-up) occurs.
Allo-HCT, allogeneic hematopoietic cell transplantation; CR, complete remission; Rand, randomization; Rel, relapse.

EFS and induction treatment failure (ITF) in QuANTUM-First

- EFS was similar between arms when ITF was defined (**Table 2**), according to FDA guidance, as not achieving CR by day 42 from the start of the last induction cycle (HR, 0.916 [95% CI, 0.754-1.114]; *P*=0.24; primary EFS analysis)[1]
- EFS favored quizartinib over placebo when ITF was defined as not achieving CRc or CR, by the end of induction up to day 56 from the last induction cycle (HR, 0.729 [95% CI, 0.592-0.897]; nominal *P*=0.0031; and 0.818 [95% CI, 0.669-0.999; nominal *P*=0.032], respectively; **Table 2**)[1]

Table 2. Analyses of EFS per IRC Assessment (ITT Population)[1]

Definition	Median (95% CI), months		HR (95% CI)[a]	*P* value[a]
	Quizartinib (N=268)	Placebo (N=271)		
Primary analysis of EFS ITF: no CR by day 42 from the start of the last induction cycle	0.03 (0.03-0.95)	0.71 (0.03-3.42)	0.916 (0.754-1.114)	0.2371
Sensitivity/supplementary analysis of EFS ITF: no CR by end of induction up to day 56 from the start of the last induction cycle	5.0 (1.8-9.0)	3.4 (1.7-5.5)	0.818 (0.669-0.999)	0.0323[b]
Sensitivity/supplementary analysis of EFS ITF: no CRc (CR + CRi) by the end of induction, up to day 56 from the start of the last induction cycle (original protocol-defined primary analysis)	11.9 (8.1-16.5)	5.7 (4.0-6.9)	0.729 (0.592-0.897)	0.0031[b]

[a]HR and 2-sided *P* value were calculated using a stratified log-rank test. [b]Nominal *P* values.
CR, complete remission; CRc, composite complete remission; CRi, complete remission with incomplete hematologic recovery; EFS, event-free survival; HR, hazard ratio; IRC, independent review committee; ITF, induction treatment failure; ITT, intent-to-treat.

Time to hematologic recovery from induction cycle 1 day 1[1]

- Among patients with CR, the median time to recovery of neutropenia was 36 days in the quizartinib arm and 29 in the placebo arm; the median time to recovery of thrombocytopenia was 31 days in the quizartinib arm and 29 in the placebo arm (**Table 3**)

Table 3. Time to Hematologic Recovery From Induction Cycle 1 Day 1[1]

Time to hematologic recovery from C1D1 in patients with CR with onset of neutropenia or thrombocytopenia during induction	Patient with CR per IRC assessment	
	Quizartinib (N=147)	Placebo (N=150)
Time to recovery of neutropenia (ANC ≥1000 cells/mm³), median (IQR), days	36 (29-44)	29 (27-38)
Time to recovery of thrombocytopenia (PLT ≥100,000 cells/mm³), median (IQR), days	31 (28-40)	29 (26-34)

ANC, absolute neutrophil count; C1D1, cycle 1 day 1; CR, complete remission; IQR, interquartile range; IRC, independent review committee; ITF, induction treatment failure; PLT, platelet.

Relevance of a 56-Day Window on EFS

- Between day 42 and the end of the induction phase, there were 51 patients who achieved CR (quizartinib, n=33; placebo, n=18), and they were considered as ITF with EFS event on day 1 in the primary analysis of EFS[1] (**Figure 8**)
 - Among these 51 patients, 9 (quizartinib, n=5; placebo, n=4) had CRi by day 42 and achieved CR after day 42 in the induction phase (**Figure 8**)
- These data indicate the relevance of a 56-day window on EFS and that FLT3 therapy has different mechanism of action than cytotoxic therapy

Figure 8. Kinetics of Achieving CR After Day 42 of Last Induction Cycle

Patient characteristics	Quizartinib	Placebo	Total
Number of patients who achieved CR after day 42 of last induction cycle[a]	33	18	51
Number of patients who achieved CRi by day 42 of last induction cycle and achieved CR after day 42 of last induction cycle[a]	5	4	9

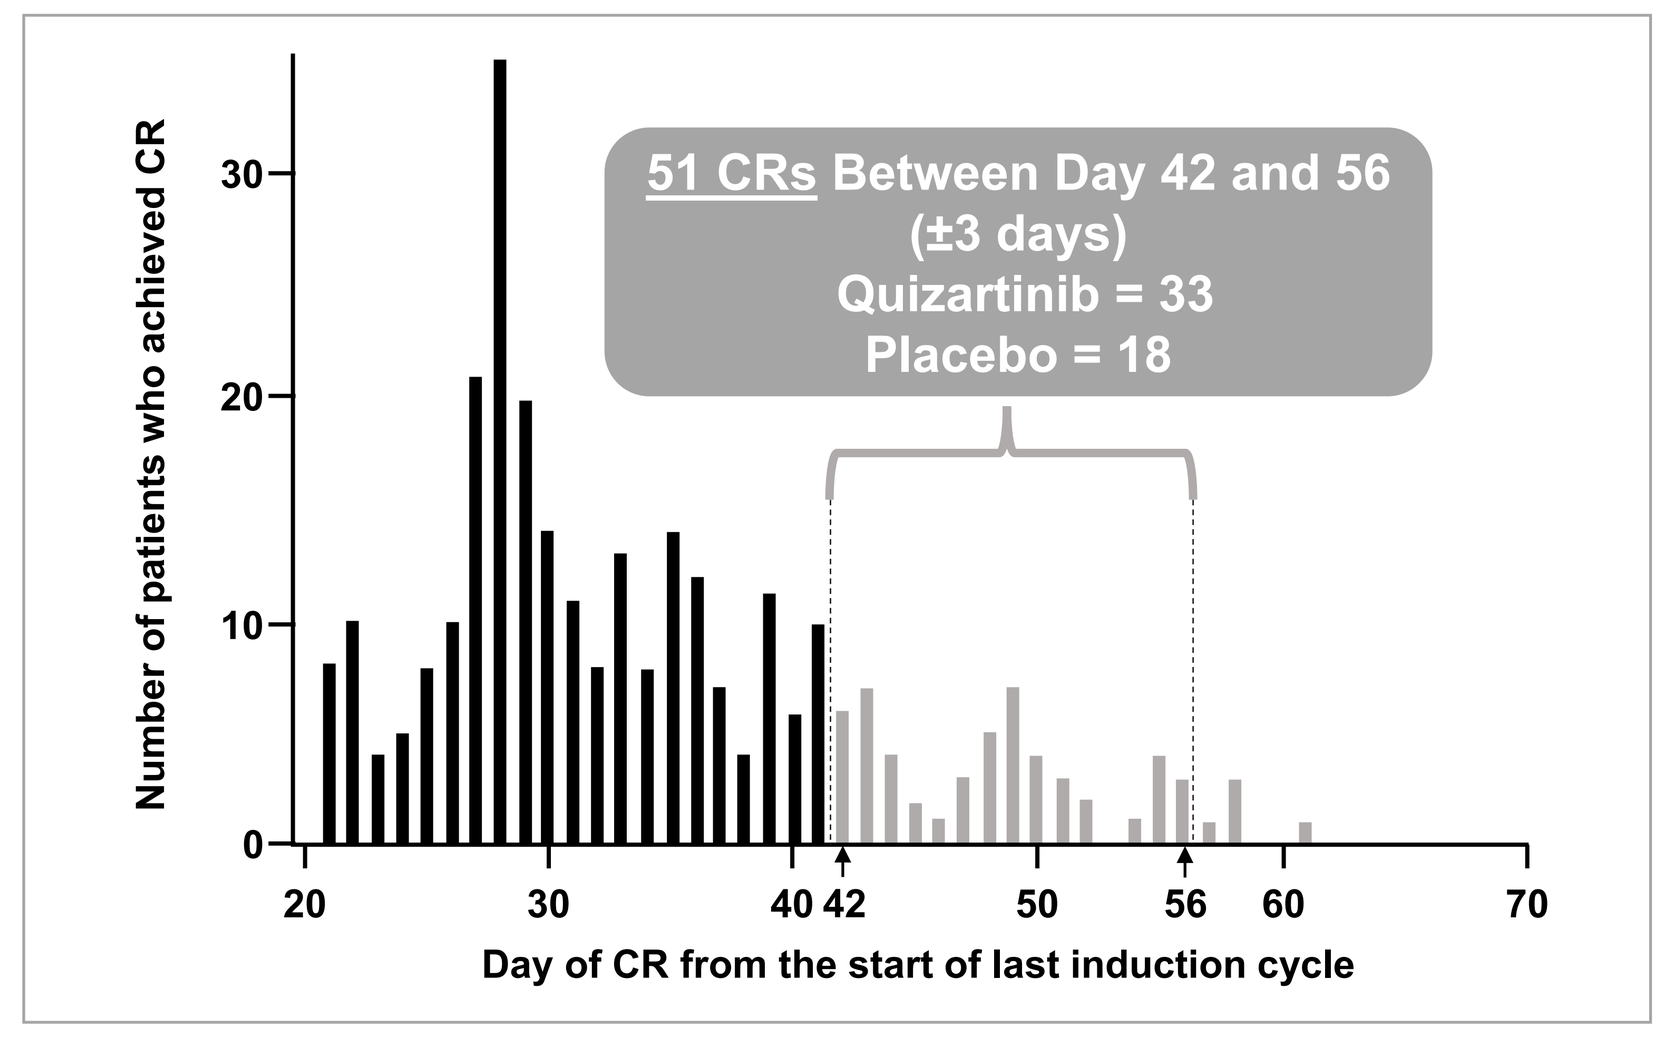
51 CRs Between Day 42 and 56
(±3 days)
Quizartinib = 33
Placebo = 18
Number of patients who achieved CR
Day of CR from the start of last induction cycle

[a]Between day 42 and the end of induction phase.
CR, complete remission; CRi, complete remission with incomplete hematologic recovery.

CONCLUSIONS

In QuANTUM-First:
- A substantial fraction of the estimated effect of quizartinib on OS was mediated through its effect on achieving a durable CR or CRc
- More patients in the quizartinib arm versus the placebo arm achieved CR after day 42 of the last induction cycle, suggesting that there was a delay in CR achievement with quizartinib
 - Longer EFS was observed in the quizartinib arm over placebo when ITF was defined as not achieving CR or CRc at the end of induction, over a 56-day window (without 42-day window)
 - This may be driven by late and durable responders in the quizartinib arm

REFERENCES

1. Erba HP, et al. *Lancet*. 2023;401(10388):1571-1583.
2. Daiichi Sankyo press release. VANFLYTA® first FLT3 inhibitor approved in the U.S. specifically for patients with newly diagnosed *FLT3*-ITD positive AML. Accessed October 13, 2023. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202307/20230720_E.pdf.
3. VANFLYTA® (quizartinib). Package insert. Daiichi Sankyo, Inc.; July 2023.
4. Daiichi Sankyo press release. VANFLYTA® first FLT3 inhibitor approved in Japan for patients with newly diagnosed *FLT3*-ITD positive AML. Accessed October 13, 2023. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202305/20230525_E.pdf.
5. Daiichi Sankyo press release. VANFLYTA® approved in the EU as the first FLT3 inhibitor specifically for patients with newly diagnosed *FLT3*-ITD positive AML. Accessed November 9, 2023. https://www.daiichisankyo.com/files/news/pressrelease/pdf/202311/20231109_E.pdf.
6. Bakunina K, et al. *Cancer Med*. 2022;11(3):630-640.

ACKNOWLEDGMENTS

We would like to thank the patients, their families, and caregivers for their participation in the QuANTUM-First study. We would further like to thank the QuANTUM-First steering committee members, the investigators, the study staff, and independent review committee and data monitoring committee members for their important contributions. This study is sponsored by Daiichi Sankyo, Inc. Medical writing support was provided by Mohamed Abdelmegeed, MD, PhD, CMPP, and Francesca Balordi, PhD, CMPP, of The Lockwood Group (Stamford, CT, USA), in accordance with Good Publication Practice (GPP 2022) guidelines, with funding by Daiichi Sankyo, Inc.

Copies of this poster or presentation obtained through Quick Response (QR) Code or https://bit.ly/DSIASH2023 are for personal use only and may not be reproduced without permission from the author of this material.

Presented at the 65th American Society of Hematology (ASH) Annual Meeting and Exposition; December 9-12, 2023; San Diego, CA, USA