San Antonio Breast Cancer Symposium® - December 5-9, 2023

PO2-04-04

Trastuzumab deruxtecan for the treatment of patients with HER2-positive breast cancer with brain and/or leptomeningeal metastases: an updated overall survival analysis using data from a multicenter retrospective study (ROSET-BM study)

Takashi Yamanaka[1], Naoki Niikura[2], Takahiro Nakayama[3], Mitsugu Yamamoto[4], Kazuo Matsuura[5], Kenichi Inoue[6], Sachiko Takahara[7], Hironori Nomura[8], Shosuke Kita[9], Miki Yamaguchi[10], Tomoyuki Aruga[11], Nobuhiro Shibata[12], Akihiko Shimomura[13], Yuri Ozaki[14], Shuji Sakai[15], Daisuke Takiguchi[16], Takehiko Takata[16], Armin Bastanfard[16], Kazuhito Shiosakai[16], Junji Tsurutani[17]

[1]Kanagawa Cancer Center, [2]Tokai University, [3]Osaka International Cancer Institute, [4]Hokkaido Cancer Center, [5]Saitama Medical University International Medical Center, [6]Saitama Prefectural Cancer Center, [7]Kitano Hospital, [8]University of the Ryukyus Hospital, [9]National Cancer Center Hospital, [10]Kurume General Hospital, [11]Tokyo Metropolitan Cancer and Infectious Diseases Center Komagome Hospital, [12]Kansai Medical University Hospital, [13]National Center for Global Health and Medicine, [14]Aichi Cancer Center, [15]Tokyo Women's Medical University, [16]Daiichi Sankyo Co., Ltd., [17]Showa University.

Purpose

- At SABCS2022, we presented results of ROSET-BM (UMIN000044995) study showing the real-world effectiveness of T-DXd in HER2-positive BC patients with BM and/or LMC.
- To confirm the long-term effectiveness of T-DXd in these patients, we conducted an updated analysis using additional 1-year follow-up data.

Conclusions

- The updated results of this retrospective chart review study show that T-DXd has promising effectiveness in HER2-positive MBC patients with long-term, heavily pretreated BM and LMC.
- The multivariate analysis results showed no difference even in factors that were generally considered to have a poor prognosis.
- Subgroup (BM classification) analyses showed that median OS in patients with active BM was 27.0 months (95% CI, 16.4 to NR).
- The incidence, frequency, and timing of onset of ILD were all similar to those of Japanese subgroup in previous studies.

Plain language summary

Why did we perform this research?
Treatment options for HER2-positive breast cancer (in which the cancer cells have higher than normal levels of HER2 protein) that has spread from its original site to the brain (brain metastases [BM]) and/or membranes and fluid surrounding the brain and spinal cord (leptomeningeal carcinomatosis [LMC]) are limited. We wanted to investigate whether trastuzumab deruxtecan (T-DXd) can be used to treat HER2-positive breast cancer in patients with BM and/or LMC in the real-world.

How did we perform this research?
We looked at the medical records of people being treated with T-DXd at multiple centers. We investigated how effective and safe T-DXd was when used to treat people with HER2-positive breast cancer with BM and/or LMC, and where pictures of the brain were available, whether T-DXd affected BM and/or LMC tumors.

What were the findings of this research and what are the implications?
Overall survival (OS) rate, what percentage of patients were still alive, at 2 years were determined by extending the follow-up period. For patients with BM that have not been treated or have progressed after treatment, the median OS was 27 months. Progression-free survival (PFS) and time to treatment failure (TTF) results were consistent with previously reported 1-year results.
This study revealed that the incidence of ILD in patients with BM who received T-DXd was similar to that reported in previous studies targeted at Japanese people.

Where can I access more information?
https://center6.umin.ac.jp/cgi-open-bin/ctr_e/ctr_view.cgi?recptno=R000051405

This presentation is the intellectual property of the author/presenter. Contact them at mtintksh@msn.com for permission to reprint and/or distribute.

Introduction

- ROSET-BM (UMIN000044995) was a retrospective chart review study to evaluate the effectiveness of trastuzumab deruxtecan (T-DXd) in HER2-positive breast cancer (BC) patients with brain metastases (BM) and/or leptomeningeal carcinomatosis (LMC) in Japan.
- At SABCS2022, we presented results showing the real-world effectiveness of T-DXd in HER2-positive BC patients with BM and/or LMC (data cutoff date: October 31, 2021).
- Here, we present the latest results, updated using an additional 1 year of follow-up data, for the HER2-positive BC patients with BM and/or LMC who were alive on October 31, 2021 (data cutoff date: October 31, 2022).

Methods

Study design

- Patients who started T-DXd treatment between May 25, 2020, and April 30, 2021, were registered for ROSET-BM.
- Overall survival (OS), progression-free survival (PFS), and time to treatment failure (TTF) were evaluated.
- Median survival times and 95% confidence intervals (CIs) were calculated using the Kaplan–Meier method.
- In addition, prespecified and exploratory subgroup analyses were performed based on patient background characteristics.
- Univariate analysis and multivariate analysis with step-wise method were peroformed by Cox proportional hazards model to evaluate the relation between baseline prognostic factors and OS.

Patients

- ***Inclusion criteria***
 Age ≥20 years.
 HER2-positive breast cancer with BM (including stable BM after local treatment, BM before local treatment, and symptomatic BM).
- ***Exclusion criteria***
 Patient's desire not to participate in the study prior to database lock.
 Previous treatment with T-DXd during participation in a clinical trial (DESTINY-Breast series).

Definition of analytical BM and LMC subgroups

- ***Analytical active BM subgroup***
 - Patients with enlarging brain tumors, as confirmed by comparison of imaging data obtained at two time points before T-DXd administration.
 - Patients who had undergone Whole brain radiation therapy (WBRT) within 30 days before T-DXd administration, or who had LMC, were excluded.
- ***Analytical stable BM subgroup***
 - Patients with BM who were not classified by ICR as having active BM and/or LMC.
 - Patients with active BM who had undergone WBRT within 30 days before T-DXd administration.
- ***LMC subgroup***
 - Patients whose findings indicated LMC with or without BM.

References

1. Modi S, et al.: *N Engl J Med* 382: 610-621, 2020
2. Hurvitz SA: San Antonio Breast Cancer Symposium 2021, GS3-01
3. Jacobson A: *Oncologist* 27: S3-S4, 2022

Acknowledgement

We thank the patients who are participating in this study as well as their families and caregivers. Editorial assistance was provided by Infront Medical Publications Inc., and funded by Daiichi Sankyo Co., Ltd.

Disclosure

This study is sponsored by Daiichi Sankyo Co., Ltd. In March 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for Trastuzumab Deruxtecan (T-DXd; DS-8201).
For queries, please contact Takashi Yamanaka (mtintksh@msn.com).

Results

Patient disposition

344 sites that used T-DXd purchased from wholesalers in Japan in 2020
→ **124 sites: no response; 135 sites: no eligible patients; 23 sites: declined to participate in the study**
↓
113 patients (62 sites) were enrolled
→ **9 patients did not meet inclusion criteria**
↓
104 patients were eligible (total analysis population)

Baseline characteristics

	N = 104
Sex	
Male / female	1 (1.0) / 103 (99.0)
Age, years	
<65 / ≥65	75 (72.1) / 29 (27.9)
HER2 status (IHC)	
0, 1+ / 2+ / 3+ Unknown	0 (0.0) / 18 (17.3) / 84 (80.8) 2 (1.9)
HER2 status (ISH)	
Positive / negative	29 (27.9) / 1 (1.0)
Unknown	74 (71.2)
No. of prior therapies for MBC	
0–2 chemotherapies	25 (24.0)
≥3	79 (76.0)
Median (Q1, Q3)	4.0 (3.0, 7.0)
ECOG PS	
0 / 1 / 2 / 3 or 4 Unknown	27 (26.0) / 54 (51.9) / 12 (11.5) / 4 (3.8) / 7 (6.7)

	N = 104
Visceral metastasis (excluding BM)	79 (76.0)
Clinical presentation of BM	
Symptomatic	32 (30.8)
Asymptomatic	72 (69.2)
Drug used for symptoms of BM	
Steroids	15 (14.4)
Anti-epileptics	11 (10.6)
Classification of BM by ICR	
Active BM	90 (86.5)
Without LMC	73 (70.2)
With LMC	17 (16.3)
Stable BM	6 (5.8)
LMC only	2 (1.9)
Image not classified	6 (5.8)
Time from MBC diagnosis to first administration of T-DXd, months	37.5 (1.7, 256)
Time from BM diagnosis to first administration of T-DXd, months	18.9 (0.1, 129)

Data are n (%) or median (min, max), unless otherwise stated.
IHC, immunohistochemistry; ISH, *in situ* hybridization; MBC, metastatic breast cancer; Q, quartile; ECOG PS, European Clinical Oncology Group performance status; ICR, independent central review.

Summary of results

Variable	Primary results October 2021 data cutoff (N = 104)	Updated results October 2022 data cutoff (N = 104)
Median follow-up duration, months (95% CI)	11.2 (10.2–12.5)	20.4 (16.4–22.5)
Median OS, months (95% CI)	Not reached (16.1 to not reached)	Not reached (20.6 to not reached)
1-year OS rate, %	74.9	74.8
2-year OS rate, %	Not applicable	56.0
Median PFS, months (95% CI)	16.1 (12.0 to not reached)	14.6 (10.6–20.8)
Median TTF, months (95% CI)	9.7 (6.3–13.0)	9.3 (6.3–11.8)
Discontinued due to ILD, %	18.3	23.1

ILD, interstitial lung disease.

OS

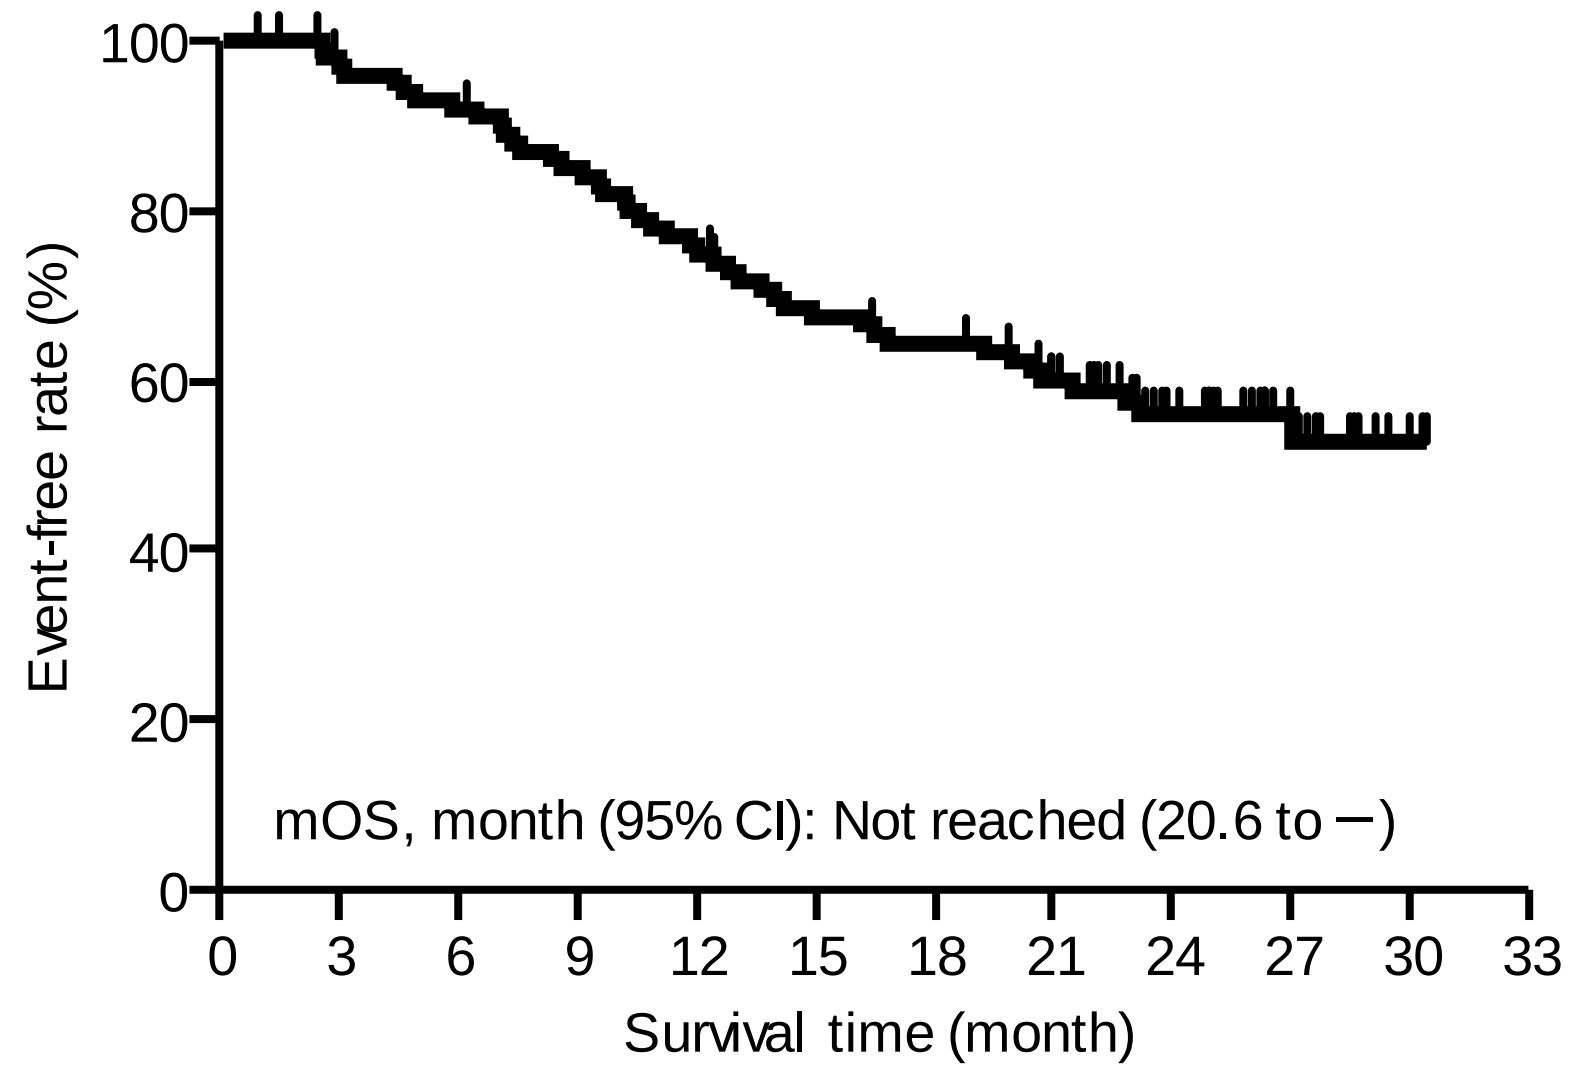
100
80
60
40
20
0
Event-free rate (%)
mOS, month (95% CI): Not reached (20.6 to –)
0 3 6 9 12 15 18 21 24 27 30 33
Survival time (month)

At risk 104 97 92 84 74 65 61 51 35 17 2 0

mOS: median overall survival.

PFS

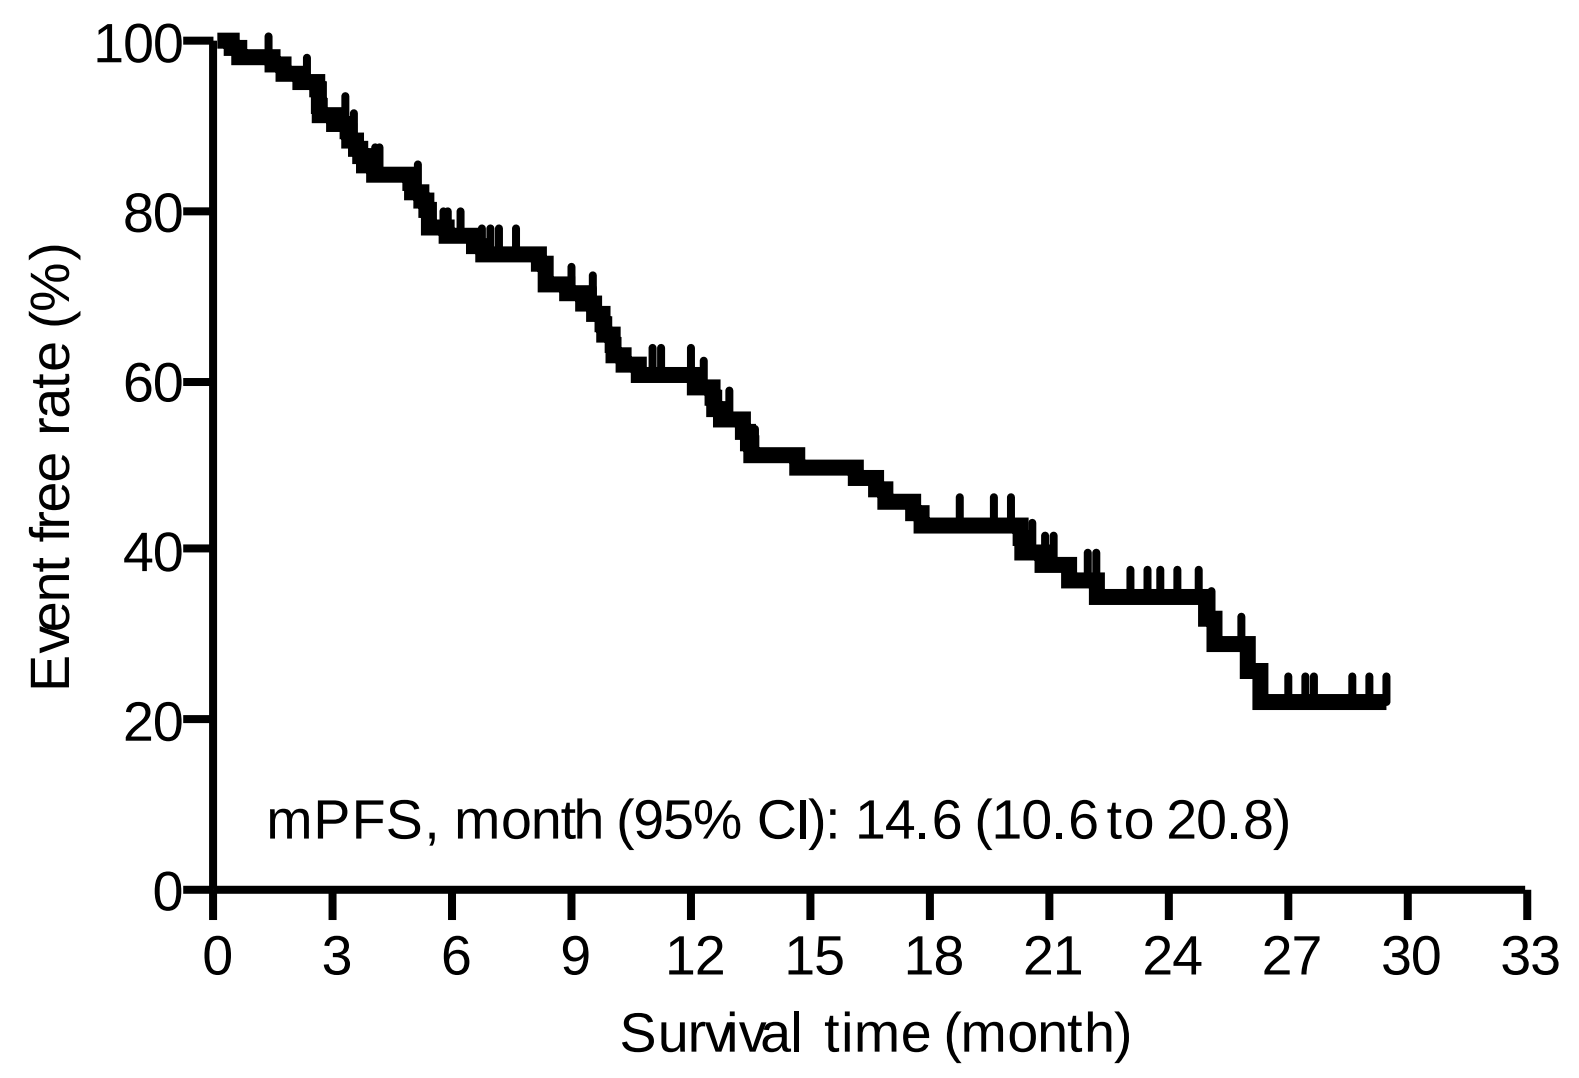
100
80
60
40
20
0
Event free rate (%)
mPFS, month (95% CI): 14.6 (10.6 to 20.8)
0 3 6 9 12 15 18 21 24 27 30 33
Survival time (month)

At risk 104 92 72 60 47 36 31 23 15 5 0 0

mPFS: median progression-free survival.

TTF

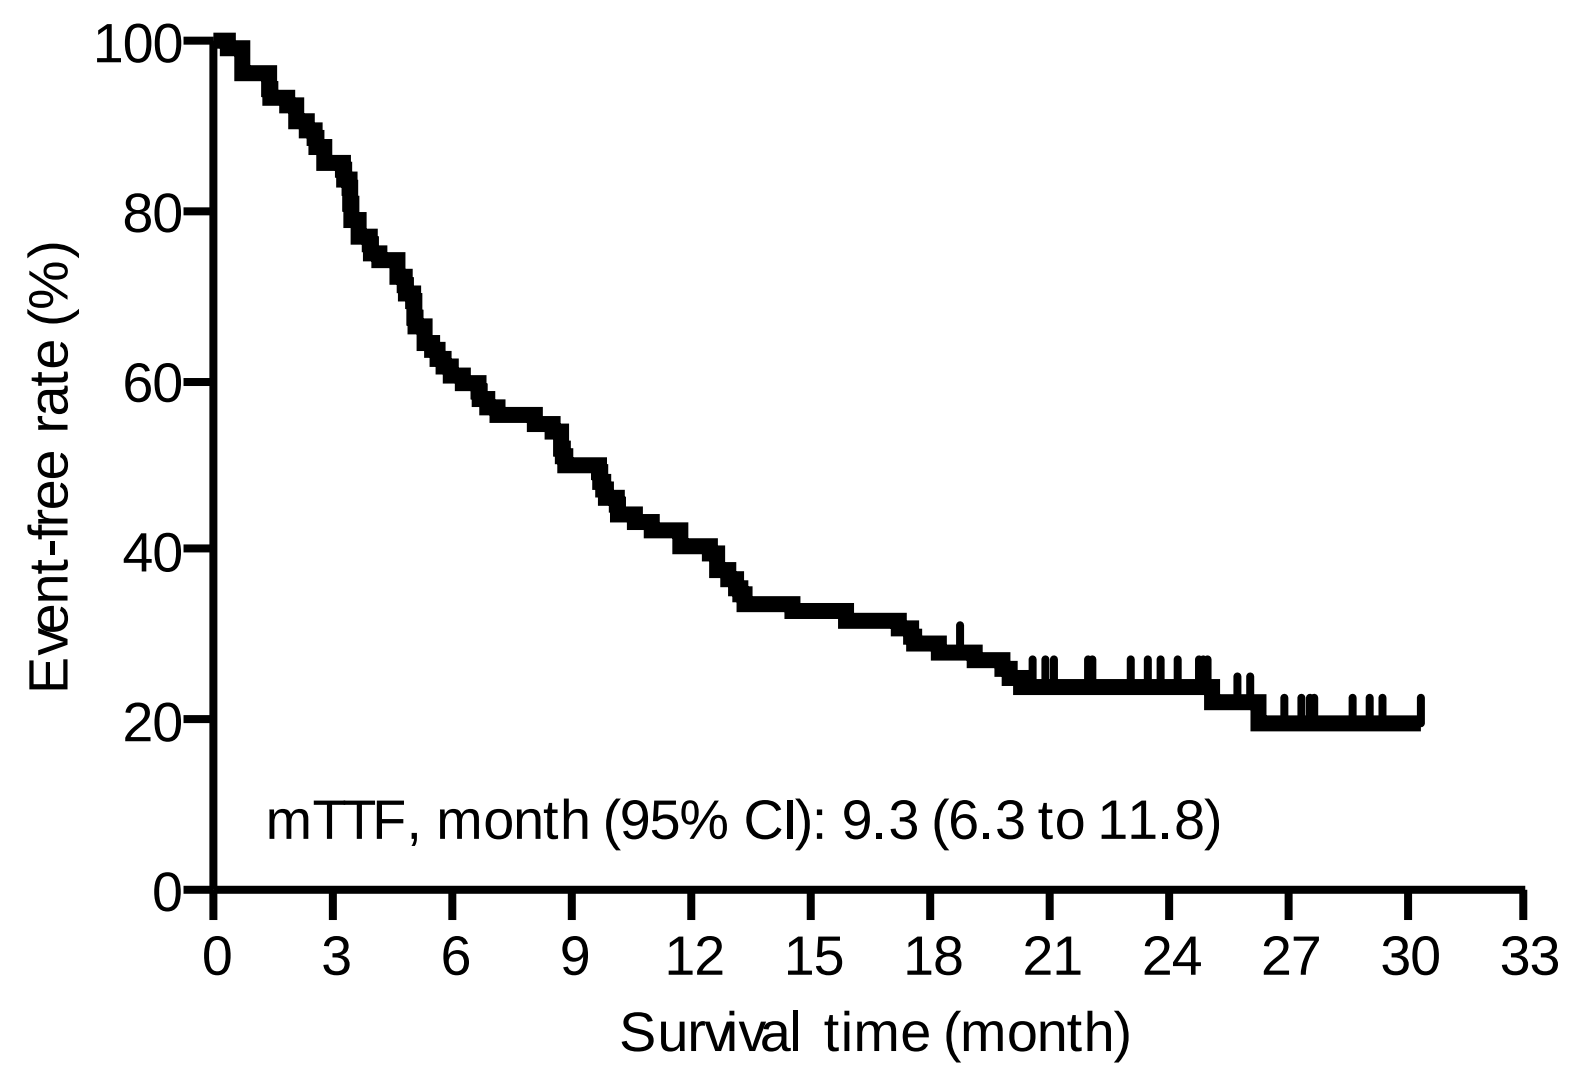
100
80
60
40
20
0
Event-free rate (%)
mTTF, month (95% CI): 9.3 (6.3 to 11.8)
0 3 6 9 12 15 18 21 24 27 30 33
Survival time (month)

At risk 104 89 63 52 42 34 30 22 16 7 1 0

AE, adverse event; ILD, interstitial lung disease; mTTF, median time to treatment failure; PD, progressive disease. * Ulcerative keratitis, malaise, pneumonia, platelet count decreased (n = 1 for each).

Reason for discontinuation

	N = 104
Event	81 (77.9)
Discontinuation of T-DXd treatment	74 (71.2)
PD	37 (35.6)
AE	28 (26.9)
ILD/ lung disorder	24 (23.1)
Grade1	14 (13.5)
Grade2	3 (2.9)
Grade3	5 (4.8)
Grade4	2 (1.9)
Grade5	0 (0.0)
AE excluding ILD*	4 (3.8)
Other	9 (8.7)
Death due to any reason	1 (1.0)
Unknown (Prior Date for the First Post-Medication)	6 (5.8)

OS by BM classification

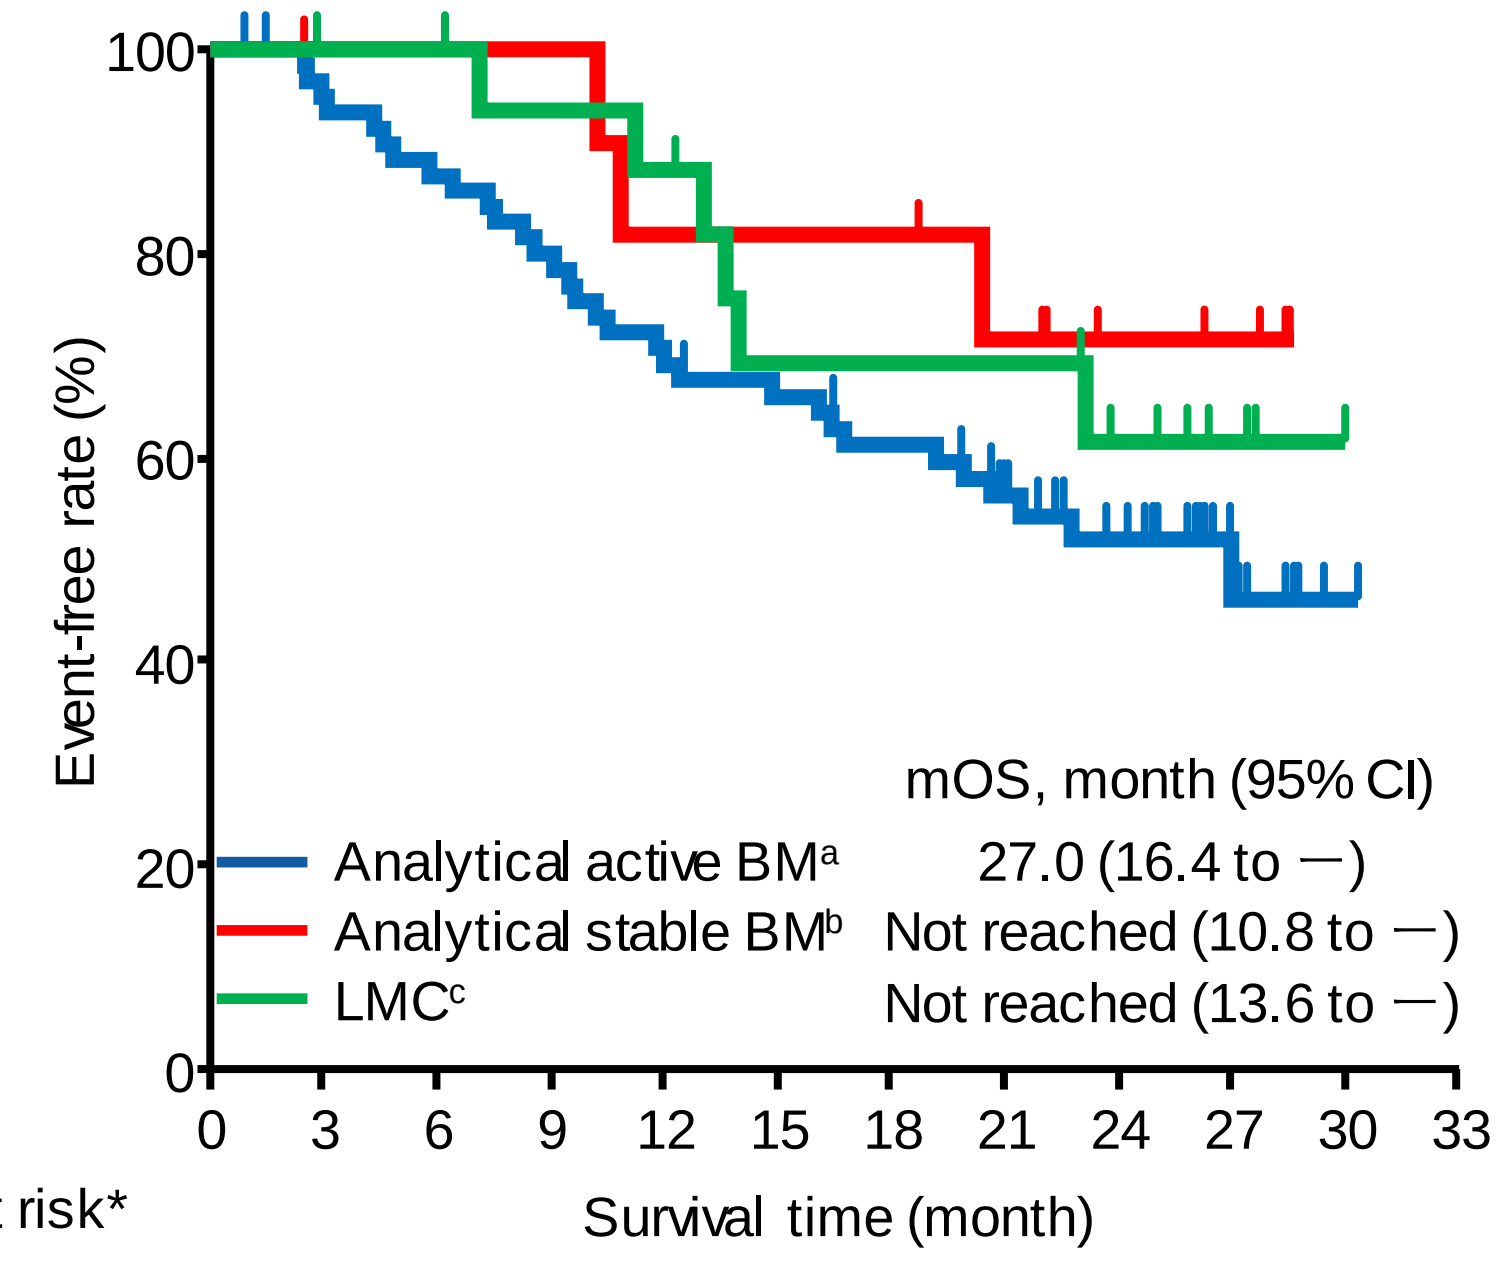
100
80
60
40
20
0
Event-free rate (%)
mOS, month (95% CI)
Analytical active BM[a] 27.0 (16.4 to –)
Analytical stable BM[b] Not reached (10.8 to –)
LMC[c] Not reached (13.6 to –)
0 3 6 9 12 15 18 21 24 27 30 33
Survival time (month)

At risk*												
Analytical active BM[a]	67	62	57	52	45	42	38	30	22	8	1	0
Analytical stable BM[b]	12	11	11	11	9	9	9	7	4	3	0	0
LMC[c]	19	18	18	16	15	11	11	11	7	4	0	0

* Of the 104 patients, 6 patients had a brain imaging data at baseline which were not classified by ICR.
[a] Active (excluding patients who had undergone WBRT within 30 days of T-DXd administration), [b] Stable + active (including patients who had undergone WBRT within 30 days of T-DXd administration), [c] Active with LMC or LMC only.

Time to discontinuation of T-DXd due to ILD

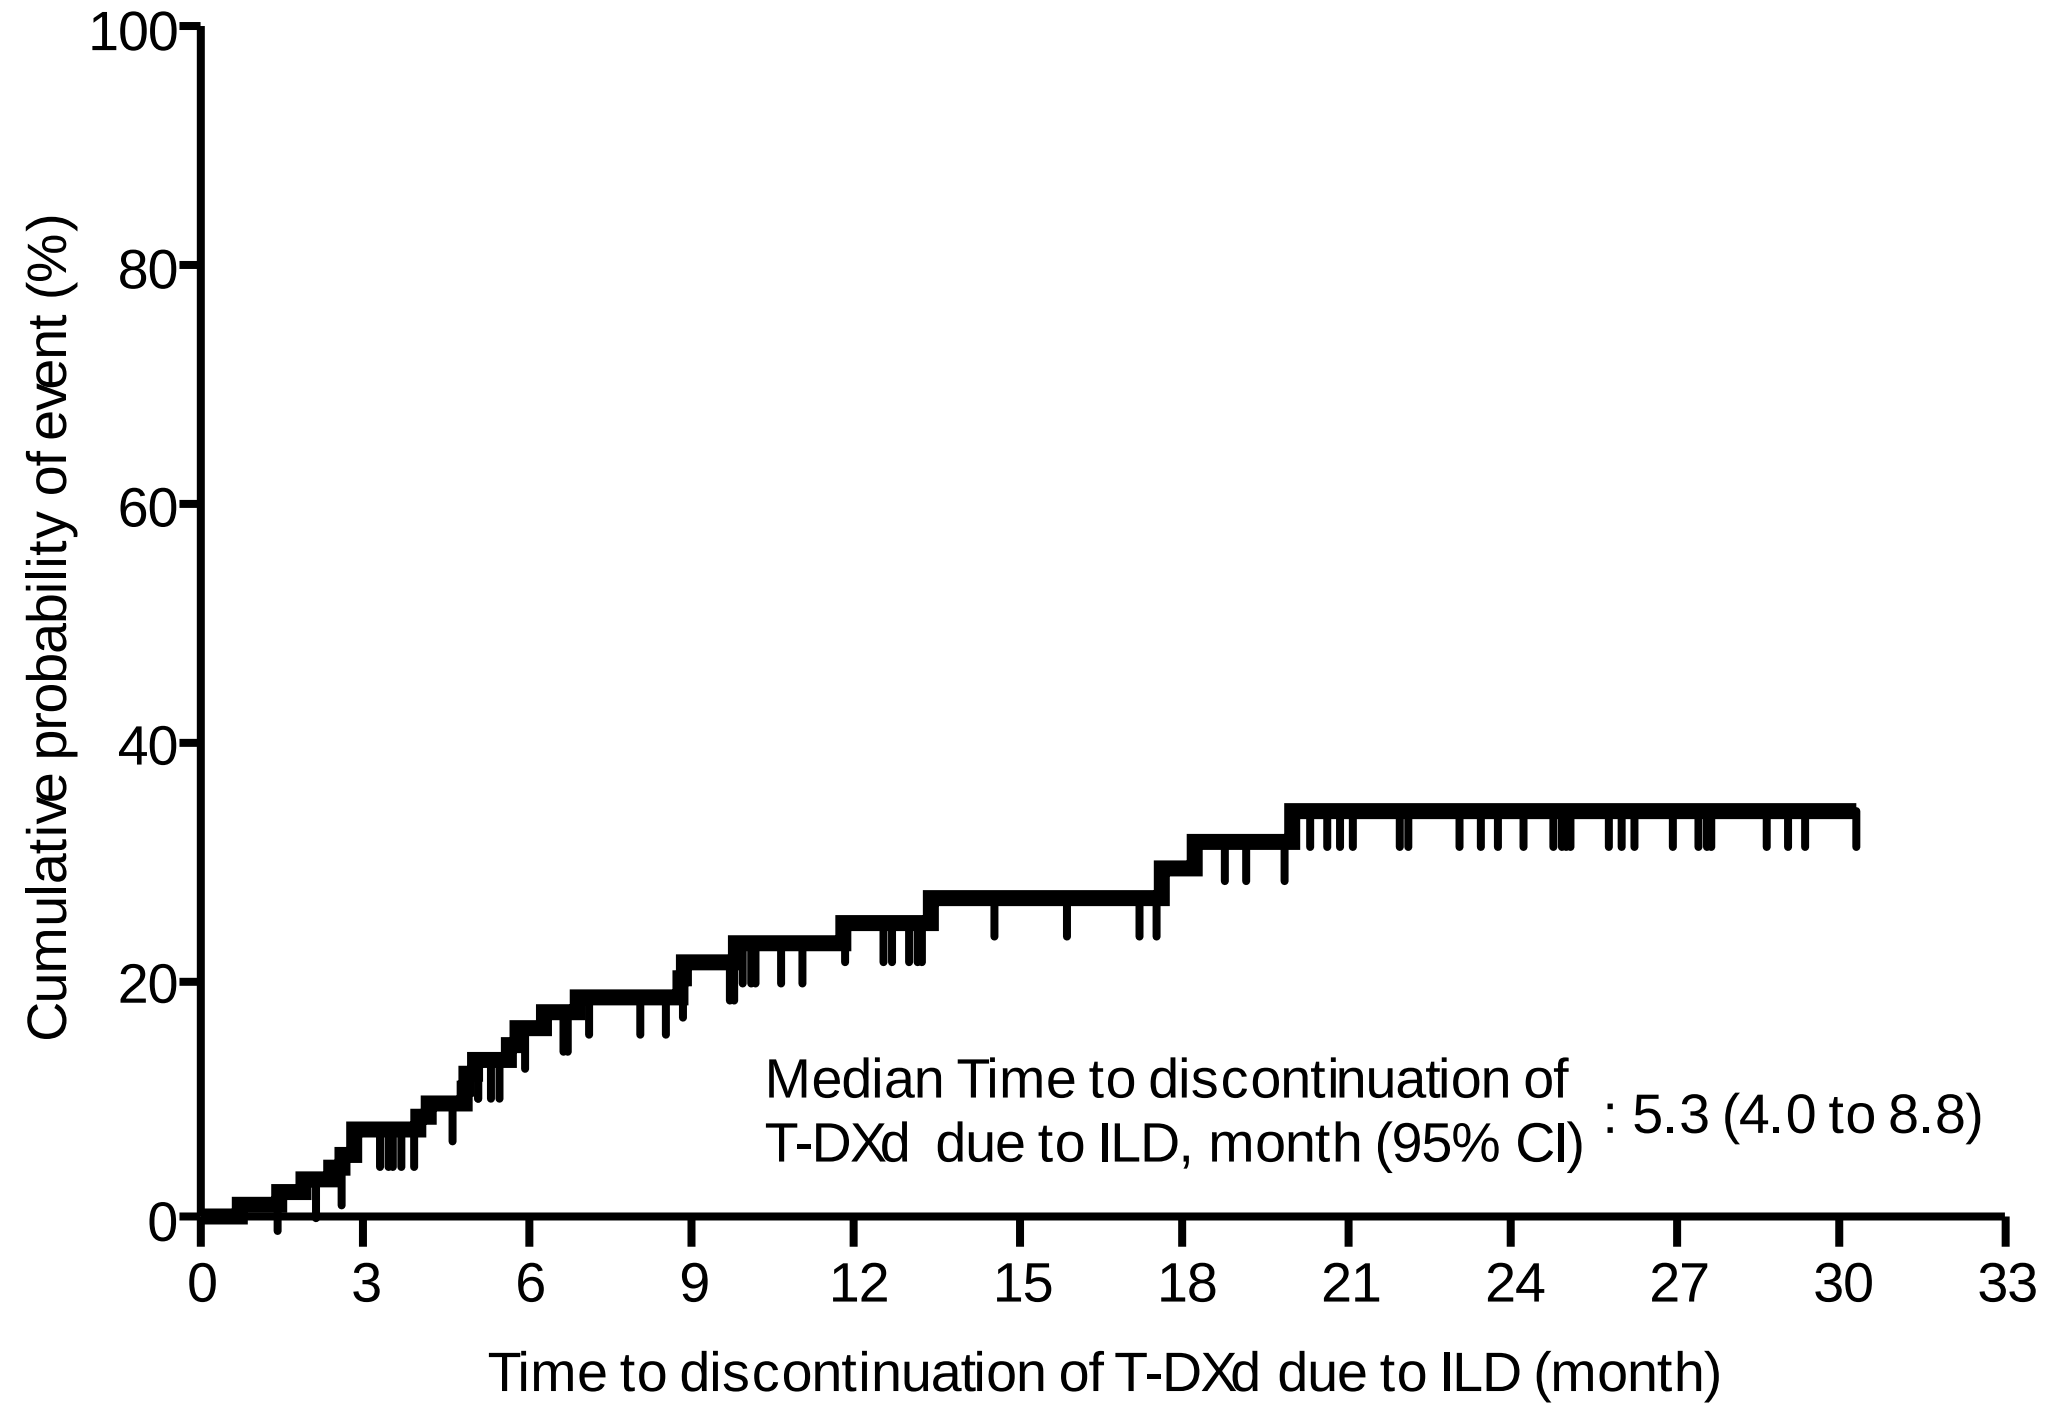
100
80
60
40
20
0
Cumulative probability of event (%)
Median Time to discontinuation of T-DXd due to ILD, month (95% CI) : 5.3 (4.0 to 8.8)
0 3 6 9 12 15 18 21 24 27 30 33
Time to discontinuation of T-DXd due to ILD (month)

At risk 104 89 63 52 42 34 30 22 16 7 1 0

・This graph shows the Kaplan-Meier curve for 24 cases.
・5.3 months is the median time among those who had ILD/lung disorder.

Cox regression analysis for OS from first administration of T-DXd

Variable	Univariate (N = 104) Adjusted HR (95% CI), *p* value
Age (years): ≥60 vs <60	0.64 (0.34-1.20), *p* = 0.164
HER2 IHC: 3+ vs 2+/ISH+	0.58 (0.29-1.19), *p* = 0.136
ER: negative vs positive	1.19 (0.66-2.17), *p* = 0.562
Steroid use at the time of T-DXd administration: absence vs presence	0.72 (0.32-1.62), *p* = 0.429
Surgery: de novo vs recurrence	0.69 (0.35-1.36), *p* = 0.280
ECOG PS: 0 or 1 vs ≥2	0.88 (0.37-2.11), *p* = 0.776
Number of BMs: 1 vs ≥2	1.45 (0.70-3.00), *p* = 0.318
LMC: without LMC vs LMC	1.48 (0.62-3.52), *p* = 0.379
Line number: 0-2 vs ≥3	1.01 (0.51-2.01), *p* = 0.976
Metastasis (excluding BM): negative vs positive	0.00 (0.00-NA), *p* = 0.990

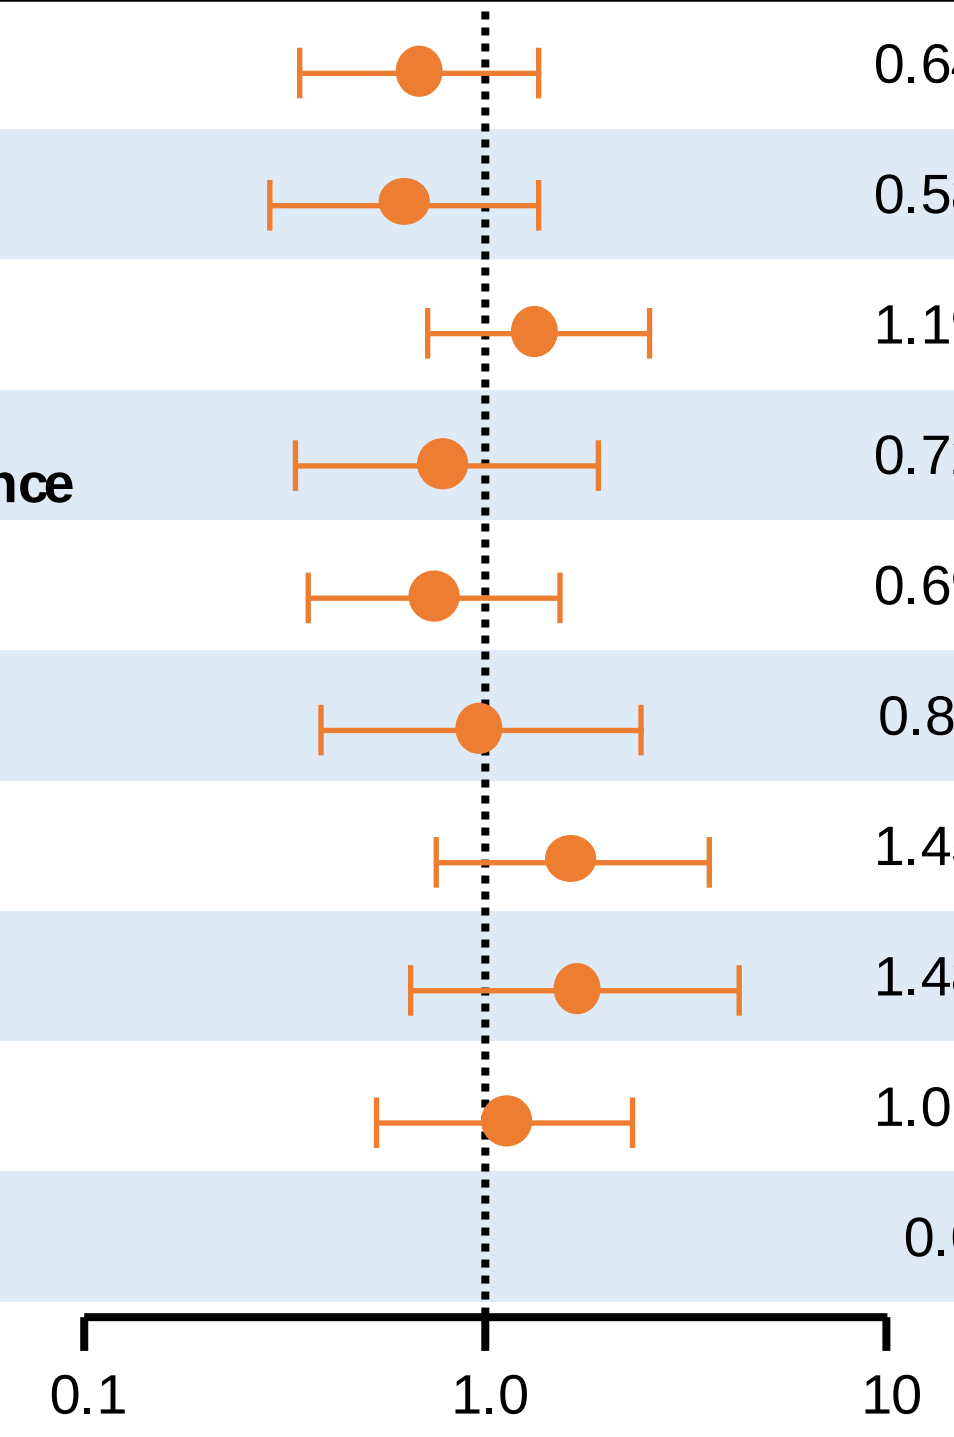
0.1
1.0
10

Results by the multivariate analysis with step-wise method were not shown. This is because significant factors were not selected.

Factors involving fewer than 10 patients (metastasis [excluding BM]: negative vs positive) are not shown.
Stepwise, analysis set: full population.