Poster W-001

Exposure-Response Analysis of Stomatitis Associated with Datopotamab Deruxtecan, a TROP2-Directed Antibody-Drug Conjugate, in Patients with Advanced Non-Small Cell Lung Cancer

Yasong Lu[1*], Kashyap Patel[2*], Hanbin Li[3], David R. Wada[3], Naoyuki Tajima[1], Ying Hong[1], Hong Zebger-Gong[4], Tushar Garimella[1], Pavan Vaddady[1#]

1. Quantitative Clinical Pharmacology, Daiichi Sankyo, Inc, Basking Ridge, NJ; 2. Certara Strategic Consulting; 3. QuanTx Consulting; 4. Oncology Clinical Development, Daiichi Sankyo Europe GmbH, Munich, Germany * Affiliation at time of the study # Presenting Author

BACKGROUND

- Datopotamab deruxtecan (Dato-DXd) demonstrated encouraging antitumor activity and a manageable safety profile in patients with heavily treated NSCLC in the Phase 1 trial TROPION-PanTumor01 (TP01; NCT03401385)[1] and is being investigated in NSCLC and other tumors.
- A clinical toxicity associated with Dato-DXd treatment is stomatitis, its mechanism yet to be elucidated.

OBJECTIVES

- To characterize the exposure response (ER) relationship of the treatment-emergent stomatitis of any Grade and Grade ≥ 2 associated with Dato-DXd

CONCLUSIONS

- Treatment-emergent adverse event stomatitis is associated with Dato-DXd and with Dato-DXd exposure metric Cavg.
- The analysis supports Dato-DXd monotherapy dose of 6 mg/kg Q3W as the starting dose[10].

INTRODUCTION

- Dato-DXd is a novel, investigational TROP2-directed antibody-drug conjugate composed of a humanized anti-TROP2 immunoglobulin G1 monoclonal antibody covalently linked to an exatecan derivative (DXd)—a highly potent topoisomerase I inhibitor payload—via a stable, tumor-selective, tetrapeptide-based cleavable linker[2]
- TROPION-PanTumor01 (NSCLC cohort shown in supplemental **Figure S1**) is an ongoing phase 1 dose-escalation and -expansion study of Dato-DXd in advanced solid tumors[3]
 - Currently available data from the advanced/metastatic NSCLC and metastatic triple-negative breast cancer (TNBC) cohorts demonstrate encouraging antitumor activity and a manageable safety profile in patients with these diseases[4,5]
- A dose-limiting toxicity associated with Dato-DXd treatment is stomatitis, the mechanism of which is yet to be elucidated.
- Here we present the ER relationship of the treatment-emergent stomatitis of any Grade and Grade ≥ 2 from TROPION-PanTumor01

METHODS

- **Part I: Exploratory Cross Drug Analysis**
 - An exploratory cross drug analysis was conducted to investigate the relevance of PK exposure to stomatitis rates across internal and external compounds: Dato-DXd, T-DXd, HER3-DXd, DX-8951f (analog of DXd), and Trodelvy (payload SN-38) (Supplemental **Table S1**)
 - PK data were from DS database, publications, or derived from reported PopPK models. Stomatitis rates were from DS database or publications
- **Part II: Dato-DXd focused ER analysis**
 - Data: PK and stomatitis (any Grade and Grade ≥ 2) data from NSCLC patients in TP01 (cutoff: 30 Jul 2021) (N = 210)
 - Exposure metrics were derived from PopPK model. For subjects with events, exposures were calculated from the 1st dose to the end of event cycle. For subjects without events, exposures were calculated from the 1st dose to the end of last treatment cycle or data cut, whichever occurred earlier
 - Logistic Regression without or with at least 1 event (didn't consider recurrence) and Time-to-event analysis using Cox proportional hazards modeling were performed.
 - Covariates of clinical interest were evaluated using a stepwise forward selection and backward elimination approach. Following covariates were sequentially tested on the slope and intercept of the univariate logistic models: categorical age, sex, baseline ECOG performance score, country, number of prior lines of therapy, smoker status, and whether last prior line of therapy was IO. Race and region were not investigated due to strong associations with the country variable.

RESULTS

Overview of stomatitis events associated with Dato-DXd

Table 1. Distribution of Grades of Stomatitis

Dose (mg/kg)	N	Grade 1 N (%)	Grade 2 N (%)	Grade 3 N (%)	Grade 4 N (%)	Grade 5 N (%)	Any Grade N (%)	Grade ≥3 N (%)
4	50	17 (34%)	4 (8%)	0 (0%)	0 (0%)	0 (0%)	21 (42%)	0 (0%)
6	50	18 (36%)	11 (22%)	1 (2%)	0 (0%)	0 (0%)	30 (60%)	1 (2%)
8	80	16 (20%)	24 (30%)	3 (3.8%)	1 (1.3%)	0 (0%)	44 (55%)	4 (5%)
10	8	1 (12.5%)	2 (25%)	1 (12.5%)	0 (0%)	0 (0%)	4 (50%)	1 (12.5%)

Majority of stomatitis events were Grade 1-2 and most events occurred in the initial treatment cycles.

Part I: Exploratory Cross Drug Analysis

- The mechanism of action and the payload differences between the compounds makes this analysis exploratory in nature and should not be taken as conclusive evidence. This was conducted to showcase the differences between compounds and to evaluate whether stomatitis rate correlates with the relevant exposure metric of the relevant entity.
- Compared with other drugs, Dato-DXd yields lowest exposure of DXd but is associated with highest stomatitis rate. A potential pharmacological hypothesis is that TROP2 expression on epithelia enables high uptake of Dato-DXd into epithelia (**Table 2**).

Table 2. Cross Drug Parameter Estimates of the PopPK Model

	Dato-DXd	T-DXd[6]	HER3-DXd	DX-8951f	Trodelvy[7]
Approved/ relevant clinical dose regimen	6 mg/kg Q3W	5.4 mg/kg Q3W	5.4 mg/kg Q3W	0.2-0.5 mg/m^2/day	10 mg/kg QW on D1 and D8 of 21-day cycle
Payload	DXd	DXd	DXd	Exatecan	SN-38
Stomatitis any Grade	60%	14%	13%	30% [8]	17%
Unconjugated payload Cmax (ng/mL)	2.9	4.4	19	39 – 77 [9]	91
Unconjugated payload AUC_{21day} (ng*h/mL)	420	672	888	713 - 1783 [8]	5460
Intact Cmax (ug/mL)	137	122	115	-	240
Intact AUC_{21day} (ug*h/mL)	16728	17640	12312	-	10680

- Based on exposure comparison, the considerable difference in the stomatitis rates may not be explained by Cmax. Trodelvy has higher and more frequent peaks than Dato-DXd. AUC_{21day} may be relevant but probably can not fully explain the differences in the event rates.

Part II: Dato-DXd focused ER analysis - Logistic Regression

- Logistic regression was performed with binary endpoint (no event or at least one event) of stomatitis. Cycle 1 Cmax, Ctrough, and AUC; Cavg to the end of cycle in which the event occurred were used as exposure metrics. Statistically significant E-S relationships (p<0.05; slopes of exposure effects different from 0) were identified for stomatitis any Grade and Grade ≥2.
- A positive relationship was observed between all Dato-DXd and DXd exposure metrics versus any Grade (**Figure 1A)** and Grade ≥2 stomatitis. **Figure 1B** shows the relationship between probability of any Grade and Grade ≥2 stomatitis with the selected exposure metric (Dato-DXd Cavg to time of event). Multivariate analyses identified statistically significant covariate effects on the slope (exposure) parameter for Japan country on any Grade stomatitis and sex on Grade ≥2 stomatitis (**Eqs. 1 and 2**).

$$logit\,(pr.stomatitis\ any\ Grade) = -6.38 + 1.77 \times log(Cavg,_{Dato\ DXd}) + 0.425 \times log(Cavg,_{Dato\ DXd}) \times (if\ from\ Japan) \quad \textbf{Eq. 1}$$

$$logit\,(pr.stomatitis\ Grade \geq 2) = -8.02 + 1.87 \times log(Cavg,_{Dato\ DXd}) + 0.221 \times log(Cavg,_{Dato\ DXd}) \times (if\ female) \quad \textbf{Eq. 2}$$

Part II: Dato-DXd focused ER analysis - Logistic Regression (contd.)

Figure 1. Univariate Logistic Fits for Dato-DXd and DXd Exposures vs. Probability of Stomatitis

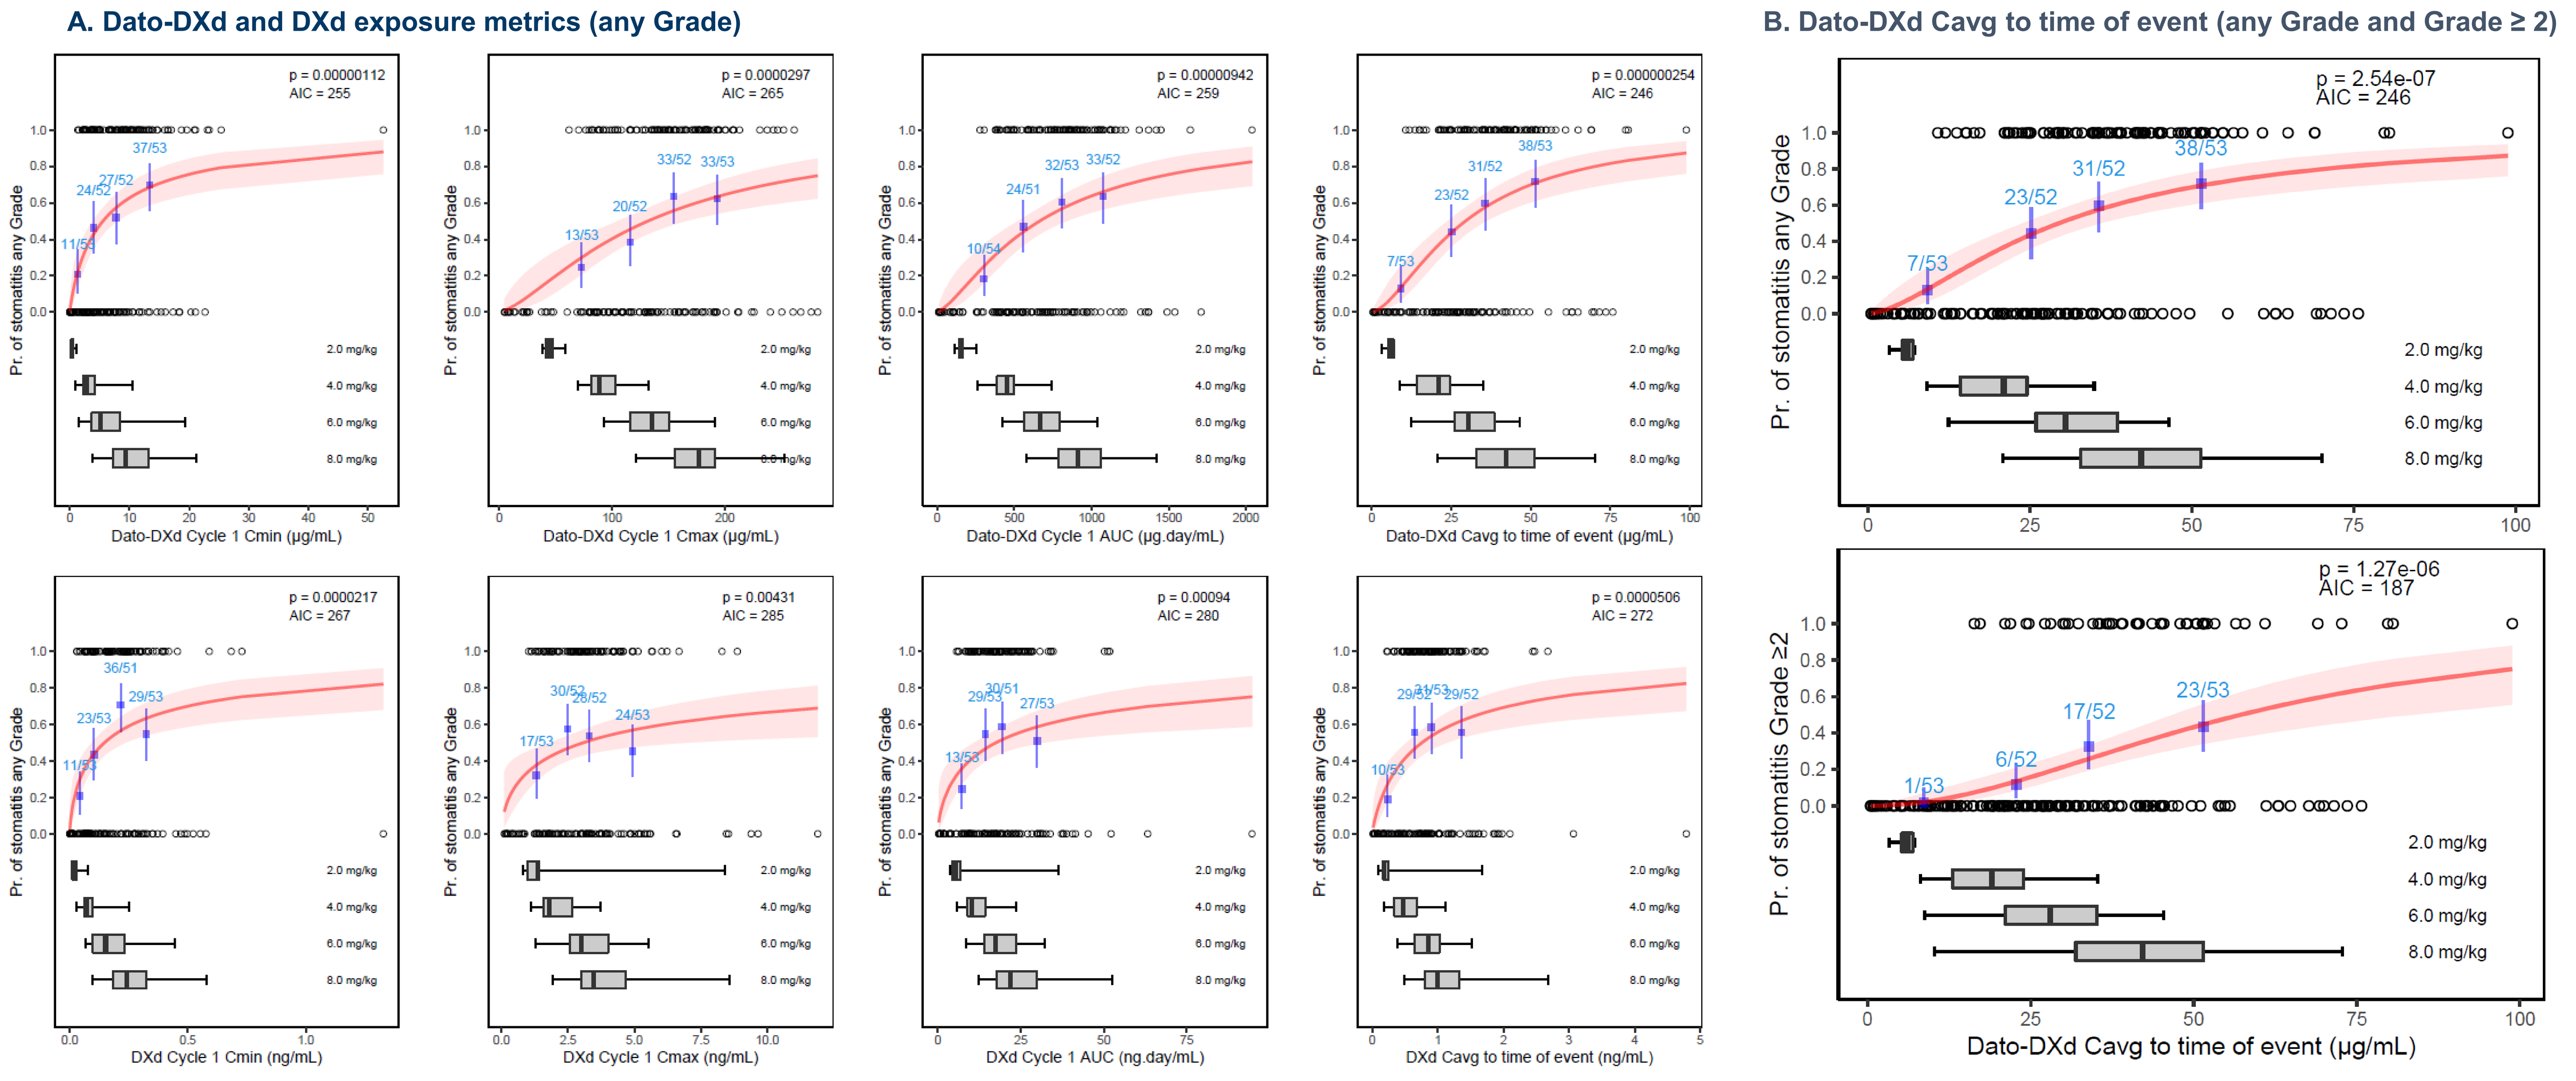

A. Dato-DXd and DXd exposure metrics (any Grade)
B. Dato-DXd Cavg to time of event (any Grade and Grade ≥ 2)
p = 0.00000112
AIC = 255
Dato-DXd Cycle 1 Cmin (µg/mL)
p = 0.0000297
AIC = 265
Dato-DXd Cycle 1 Cmax (µg/mL)
p = 0.00000942
AIC = 259
Dato-DXd Cycle 1 AUC (µg.day/mL)
p = 0.000000254
AIC = 246
Dato-DXd Cavg to time of event (µg/mL)
p = 0.0000217
AIC = 267
DXd Cycle 1 Cmin (ng/mL)
p = 0.00431
AIC = 285
DXd Cycle 1 Cmax (ng/mL)
p = 0.00094
AIC = 280
DXd Cycle 1 AUC (ng.day/mL)
p = 0.0000506
AIC = 272
DXd Cavg to time of event (ng/mL)
Pr. of stomatitis any Grade
2.0 mg/kg
4.0 mg/kg
6.0 mg/kg
8.0 mg/kg
p = 2.54e-07
AIC = 246
7/53
23/52
31/52
38/53
p = 1.27e-06
AIC = 187
1/53
6/52
17/52
23/53
Pr. of stomatitis Grade ≥2
Dato-DXd Cavg to time of event (µg/mL)

Subjects were stratified into exposure quartiles. The circles at y=0 represent subjects that did not experience the AE, and the circles at y= l show subjects that did. The blue squares are observed proportions of AE per exposure quartile, also shown as a numerical value. The vertical blue bars are the 90% CIs of observed proportions. The red line is the linear or log-linear logistic regression fit, and the pink-shaded region represents the corresponding 95% CI. The horizontal boxplots show the exposure distribution (where whiskers represent 2.5th to the 97.5th percentiles of the data) for the 2, 4, 6, and 8 mg/kg Q3W dose groups. The p-value is for the slope of the logistic regression fit.

Part II: Dato-DXd focused ER analysis - Time-to-event Analysis

With the objective of exploring the relationship between exposure and stomatitis free survival, a time-to-event analysis was performed. Kaplan Meier curves by exposure quartiles is shown in **Figure 2**. 72% of 1st event (onset) occurred within Cycle 1. Based on multivariate Cox proportional hazard modeling, hazard of stomatitis was positively correlated with log-transformed Cavg of Dato-DXd with country (Japan) as a significant covariate for any Grade stomatitis; and with sex (female) for Grade ≥ 2 stomatitis respectively (similar to the logistic regression results).

Based on the time course of AE probability predicted from multivariate Cox proportional hazard models (not shown here), the maximum difference at the plateaus of the time course between 4 and 6 mg/kg was 21.3% for Any Grade stomatitis and 18.8% for Grade ≥ 2 stomatitis. Using totality of evidence based on both safety and efficacy results, 6 mg/kg was chosen as the starting clinical dose[10] and to manage stomatitis events, dose reduction to 4 mg/kg is possible based on current toxicity management guidelines (TMG).

Current TMG were not in place for NSCLC cohort in TP01. An ongoing TP01 sub study is aimed at providing further insight into potential mechanisms of stomatitis and appropriate prevention strategies and clinical management.

Figure 2. Kaplan Meier Curves by Exposure Quartiles

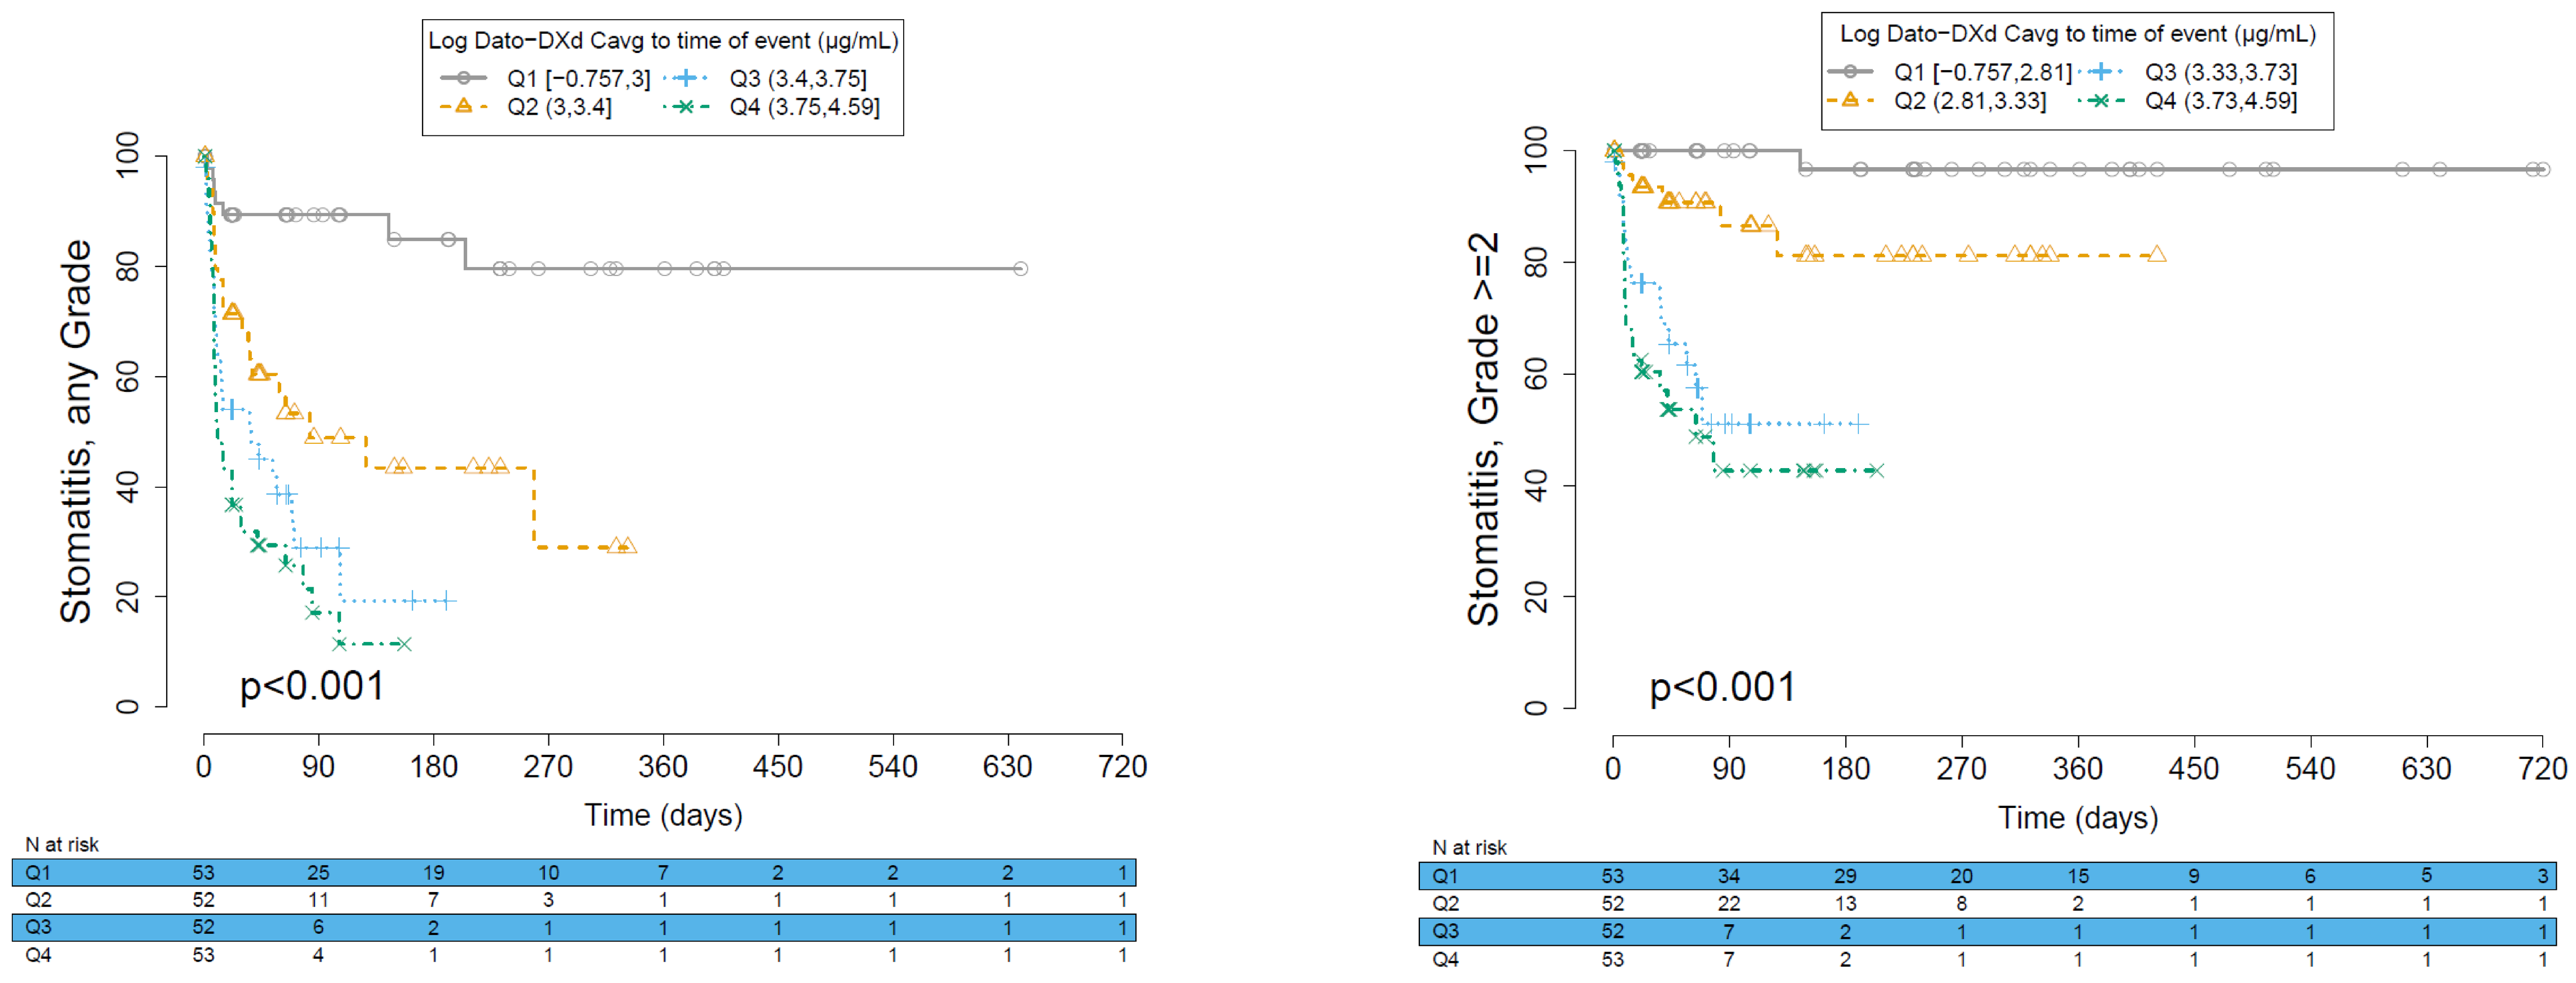

Log Dato−DXd Cavg to time of event (µg/mL)
Q1 [−0.757,3]
Q2 (3,3.4]
Q3 (3.4,3.75]
Q4 (3.75,4.59]
Stomatitis, any Grade
p<0.001
Time (days)
N at risk
Q1 53 25 19 10 7 2 2 2 1
Q2 52 11 7 3 1 1 1 1 1
Q3 52 6 2 1 1 1 1 1 1
Q4 53 4 1 1 1 1 1 1 1
Log Dato−DXd Cavg to time of event (µg/mL)
Q1 [−0.757,2.81]
Q2 (2.81,3.33]
Q3 (3.33,3.73]
Q4 (3.73,4.59]
Stomatitis, Grade >=2
p<0.001
Time (days)
N at risk
Q1 53 34 29 20 15 9 6 5 3
Q2 52 22 13 8 2 1 1 1 1
Q3 52 7 2 1 1 1 1 1 1
Q4 53 7 2 1 1 1 1 1 1

P-value is calculated using the log-rank test. The square bracket signifies inclusion of value and parenthesis signifies exclusion of value.

References

1. ClinicalTrials.gov. Accessed October 9, 2023. https://clinicaltrials.gov/ct2/show/NCT04656652
2. Okajima D, *et. al. Mol Cancer Ther*. 2021;20(12):2329-2340.
3. ClinicalTrials.gov. Accessed August 10, 2022. https://clinicaltrials.gov/ct2/show/NCT03401385
4. Garon EB, *et. al.* Presented at: IASLC 2021 World Conference on Lung Cancer; September 8-14, 2021. Abstract MA03.02.
5. Krop I, *et. al.* Presented at: San Antonio Breast Cancer Symposium; December 7-10, 2021. Abstract GS1-05.
6. Enhertu prescribing information accessed May 2021
7. Trodelvy prescribing information accessed Jun 2021
8. Esteva et al. 2003
9. Rowinsky et al. 2000
10. Lu Y, *et. al. CPT:PSP. https://doi.org/10.1002/psp4.13058*

Disclosures

At the time of the study: **YL**, **NT**, **YH**, **HZ**, **TG**, **and PV** are paid employees of Daiichi Sankyo and own stock in Daiichi Sankyo. **KP** an employee of Certara, **HL** and **DW** employees of QuanTx Consulting provided services to Daiichi Sankyo for this study.

Acknowledgements

We thank the patients, their families, and their caregivers for their participation and study staff for their contributions. This study is sponsored by Daiichi Sankyo, Inc. In July 2020, Daiichi Sankyo entered into a global development and commercialization collaboration agreement with AstraZeneca for datopotamab deruxtecan (Dato-DXd).

Poster

Supplementary material

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials.

Copies of this poster and supplemental material obtained through QR Code are for personal use only and may not be reproduced without permission from the author of this poster.

Poster presented at American Conference on Pharmacometrics (ACoP14), Abstract PMX-884. November 5 - November 8, 2023; National Harbor, MD, USA.

Corresponding author email address: pvaddady@dsi.com