151P

Baseline Circulating Tumor DNA Biomarker Analysis of Patients With Human Epidermal Growth Factor Receptor 2 Overexpressing Metastatic Non–Small Cell Lung Cancer Treated With Trastuzumab Deruxtecan

Egbert F. Smit,[1] Enriqueta Felip,[2] Dipesh Uprety,[3] Misako Nagasaka,[4] Kazuhiko Nakagawa,[5] Luis Paz-Ares,[6] Jose M. Pacheco,[7] Bob T. Li,[8] David Planchard,[9] Christina Baik,[10] Yasushi Goto,[11] Haruyasu Murakami,[12] Kaline Pereira,[13] Ayumi Taguchi,[14] Makito Koga,[14] Yusuke Kuwahara,[13] Mike Zou,[13] Wenqin Feng,[13] Zenta Tsuchihashi,[13] Pasi A. Jänne[15]

[1]Leiden University Medical Center, Leiden, Netherlands; [2]Vall d'Hebron University Hospital and Vall d'Hebron Institute of Oncology, Barcelona, Spain; [3]Barbara Ann Karmanos Cancer Institute, Detroit, MI, USA; [4]University of California Irvine School of Medicine, Orange, CA, USA; [5]Kindai University Faculty of Medicine, Osaka, Japan; [6]Clinical Universidad de Navarra, Navarra, Spain; [7]University of Colorado Anschutz Medical Campus, Aurora, CO, USA; [8]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [9]Gustave Roussy, Villejuif, France; [10]University of Washington, Seattle, WA, USA; [11]National Cancer Center Hospital, Tokyo, Japan; [12]Shizuoka Cancer Center, Shizuoka, Japan; [13]Daiichi Sankyo, Inc, Basking Ridge, NJ, USA; [14]Daiichi Sankyo, Ltd., Tokyo, Japan; [15]Lowe Center for Thoracic Oncology, Dana-Farber Cancer Institute, Boston, MA, USA

Background

- Overexpression of human epidermal growth factor receptor 2 (HER2) has been reported to occur in approximately 10-15% of patients with non–small cell lung cancer (NSCLC)[1]; therefore, HER2 represents a potential molecular target for therapy[2]
- Trastuzumab deruxtecan (T-DXd), a HER2-targeting antibody-drug conjugate, was granted accelerated approval status in 2022 by the FDA for the treatment of adult patients with unresectable or metastatic NSCLC whose tumors have activating *HER2* (*ERBB2*) mutations as detected by an FDA-approved test and who have received prior systemic therapy[3]
- There are no HER2-targeted therapies approved or recommended for the treatment of patients with HER2-overexpressing NSCLC[4-5]
- DESTINY-Lung01 (NCT03505710), a multicenter, open-label, phase 2 trial of adult patients with previously treated unresectable or metastatic HER2-overexpressing (cohorts 1 and 1a) and *HER2*-mutant (cohort 2) NSCLC, demonstrated promising antitumor activity of T-DXd in all cohorts[6,7]
 - The objective response rate (ORR) by independent central review in patients with HER2-overexpressing NSCLC was 26.5% (95% CI, 15.0-41.1) in cohort 1 and 34.1% (95% CI, 20.1-50.6) in cohort 1a, and efficacy was observed in patients with HER2 immunohistochemistry (IHC) 3+ and HER2 IHC2+[7]
- We investigated associations between baseline gene mutations in circulating tumor DNA (ctDNA) and clinical outcomes in patients with HER2-overexpressing NSCLC (cohorts 1 and 1a) from the DESTINY-Lung01 trial (data cutoff December 3, 2021)
 - Potential biomarkers, including activating mutations in key oncogenes (*EGFR, RAS, BRAF*), and other genes of interest were investigated

Conclusions

- T-DXd showed clinical efficacy in patients with HER2-overexpressing NSCLC, regardless of the presence of activating mutations in *EGFR, KRAS,* or *NRAS* at baseline
- Mutations in *STK11* and *KEAP1* were associated with shorter mPFS
- Limitations of this analysis are that it did not adjust for any differences in baseline characteristics or association between baseline biomarkers

Plain Language Summary

Why did we perform this research?

- Approximately 10-15% of patients with non–small cell lung cancer (NSCLC) have tumors that overexpress the protein human epidermal growth factor receptor 2 (HER2)[1]. The antibody-drug conjugate, trastuzumab deruxtecan (T-DXd), is approved for the treatment of patients with NSCLC who have activating mutations in the *HER2* (*ERBB2*) gene as determined by an FDA-approved test. However, there are no HER2-targeted therapies approved or recommended for the treatment of HER2-overexpressing NSCLC. The DESTINY-Lung01 trial showed antitumor activity of T-DXd in cohorts of patients with *HER2*-mutant and HER2-overexpressing NSCLC[2,3]. This study investigated associations between gene mutations and clinical outcomes in the HER2-overexpressing cohorts of DESTINY-Lung01 to identify potential biomarkers and genes of interest for T-DXd treatment in this patient population.

How did we perform this research?

- Activating mutations in key cancer genes (oncogenes; *EGFR, KRAS, NRAS,* and *BRAF*) and other genes of interest at baseline were identified by circulating tumor DNA (ctDNA) analysis, and the effects of mutations on clinical efficacy of T-DXd were assessed.

What were the findings of this research and what are the implications?

- Among 90 patients from the DESTINY-Lung01 HER2-overexpressing cohorts, 86 had samples available for ctDNA analysis. Activating mutations were detected at baseline in the key oncogenes *EGFR, KRAS, NRAS,* and *BRAF*. Although mutations in the *HER2* gene were detected at baseline, none were activating/oncogenic. Mutations were detected in other genes of interest, including *STK11* and *KEAP1*. Clinical efficacy of T-DXd was observed even in the presence of activating mutations in *EGFR, KRAS,* or *NRAS* at baseline. However, mutations in *STK11* and *KEAP1* were associated with shorter progression-free survival (the median time until a patient's cancer got worse or caused death).

Where can I access more information?

- To learn more about the DESTINY-Lung01 study, you can visit https://www.clinicaltrials.gov/study/NCT03505710

References

1. Heinmöller P et al. *Clin Cancer Res*. 2003;9:5238-43.
2. Li BT et al. *N Engl J Med*. 2022;386(3):241-51.
3. Smit EF et al. Proceedings of the European Society of Medical Oncology Annual Meeting 2022. Poster #975P.

Copies of this e-poster obtained through QR, AR, and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

This information is the intellectual property of the authors/presenters.

Please contact Prof Egbert F. Smit at e.f.smit@lumc.nl for permission to reprint and/or distribute.

Presented at the European Society for Medical Oncology (ESMO) Annual Meeting; Madrid, Spain, October 20-24, 2023

Methods

- Among 90 patients in DESTINY-Lung01 with HER2-overexpressing NSCLC, 49 received T-DXd 6.4 mg/kg Q3W (cohort 1) and 41 received T-DXd 5.4 mg/kg Q3W (cohort 1a) (**Figure 1**)

Figure 1. DESTINY-Lung01 Study Design

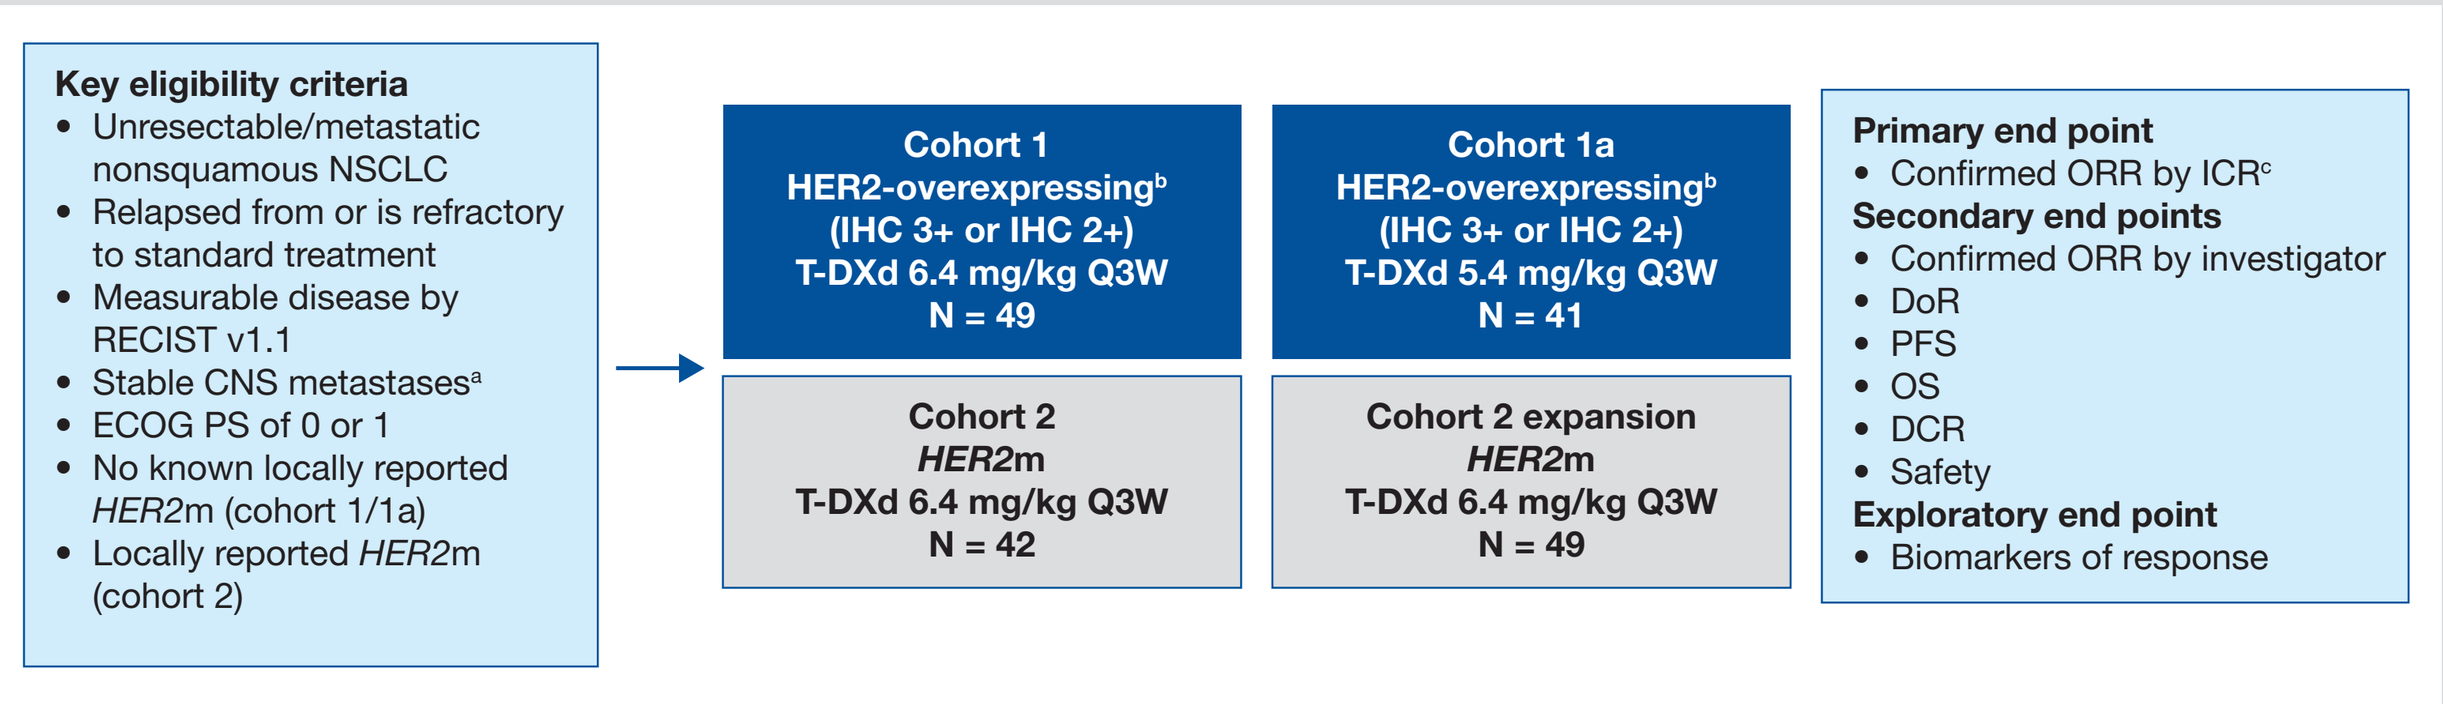

Key eligibility criteria
• Unresectable/metastatic nonsquamous NSCLC
• Relapsed from or is refractory to standard treatment
• Measurable disease by RECIST v1.1
• Stable CNS metastases[a]
• ECOG PS of 0 or 1
• No known locally reported HER2m (cohort 1/1a)
• Locally reported HER2m (cohort 2)
Cohort 1 HER2-overexpressing[b] (IHC 3+ or IHC 2+) T-DXd 6.4 mg/kg Q3W N = 49
Cohort 1a HER2-overexpressing[b] (IHC 3+ or IHC 2+) T-DXd 5.4 mg/kg Q3W N = 41
Cohort 2 HER2m T-DXd 6.4 mg/kg Q3W N = 42
Cohort 2 expansion HER2m T-DXd 6.4 mg/kg Q3W N = 49
Primary end point
• Confirmed ORR by ICR[c]
Secondary end points
• Confirmed ORR by investigator
• DoR
• PFS
• OS
• DCR
• Safety
Exploratory end point
• Biomarkers of response

No stratification factors were used to assign patients.
[a]Patients with clinically inactive, treated or untreated brain metastases that were asymptomatic and did not require ongoing steroid or anticonvulsant therapy were allowed to enroll.
[b]HER2 overexpression was assessed by central testing using archival tissue. [c]Per RECIST v1.1.

- Among these 90 patients, 86 had baseline ctDNA samples that were successfully analyzed with Guardant Health OMNI and were included in the current analysis
- Activating mutations in key oncogenes (*EGFR, KRAS, NRAS,* and *BRAF*) and other genes of interest at baseline were identified by ctDNA analysis, and the effects of T-DXd on clinical efficacy (ORR and PFS) were assessed
 - For analysis of key oncogenes, *NRAS* and *KRAS* variants were considered mutant if there was mutation at codons G12, G13, A59, Q61, K117, or A146
 - *EGFR* variants were considered mutant if variants were determined as "oncogenic" or "likely oncogenic" based on the OncoKB precision oncology database
 - Possible germline mutations, synonymous mutations, mutations that are not oncogenic (ie, not considered "likely oncogenic," "oncogenic," or "predicted oncogenic" based on OncoKB) with variant allele fractions less than 0.2, and clonal hematopoiesis of indeterminate potential mutations reported by Guardant Health were excluded
 - Analyses were not adjusted for baseline characteristics and did not account for T-DXd dose
- Statistical analysis
 - Point estimates and 2-sided 95% exact binomial CIs were calculated for ORR in each biomarker subgroup. The Kaplan-Meier method was used to estimate median event times with 2-sided 95% CIs calculated using Brookmeyer and Crowley methods

Results

Activating Mutations at Baseline

- The ctDNA gene alteration landscape at baseline (including gene alterations with single nucleotide variants [SNVs], deletions, insertions, and amplifications in 8 or more cases) is shown in **Figure 2**

Figure 2. ctDNA Gene Alteration Landscape[a,b] at Baseline

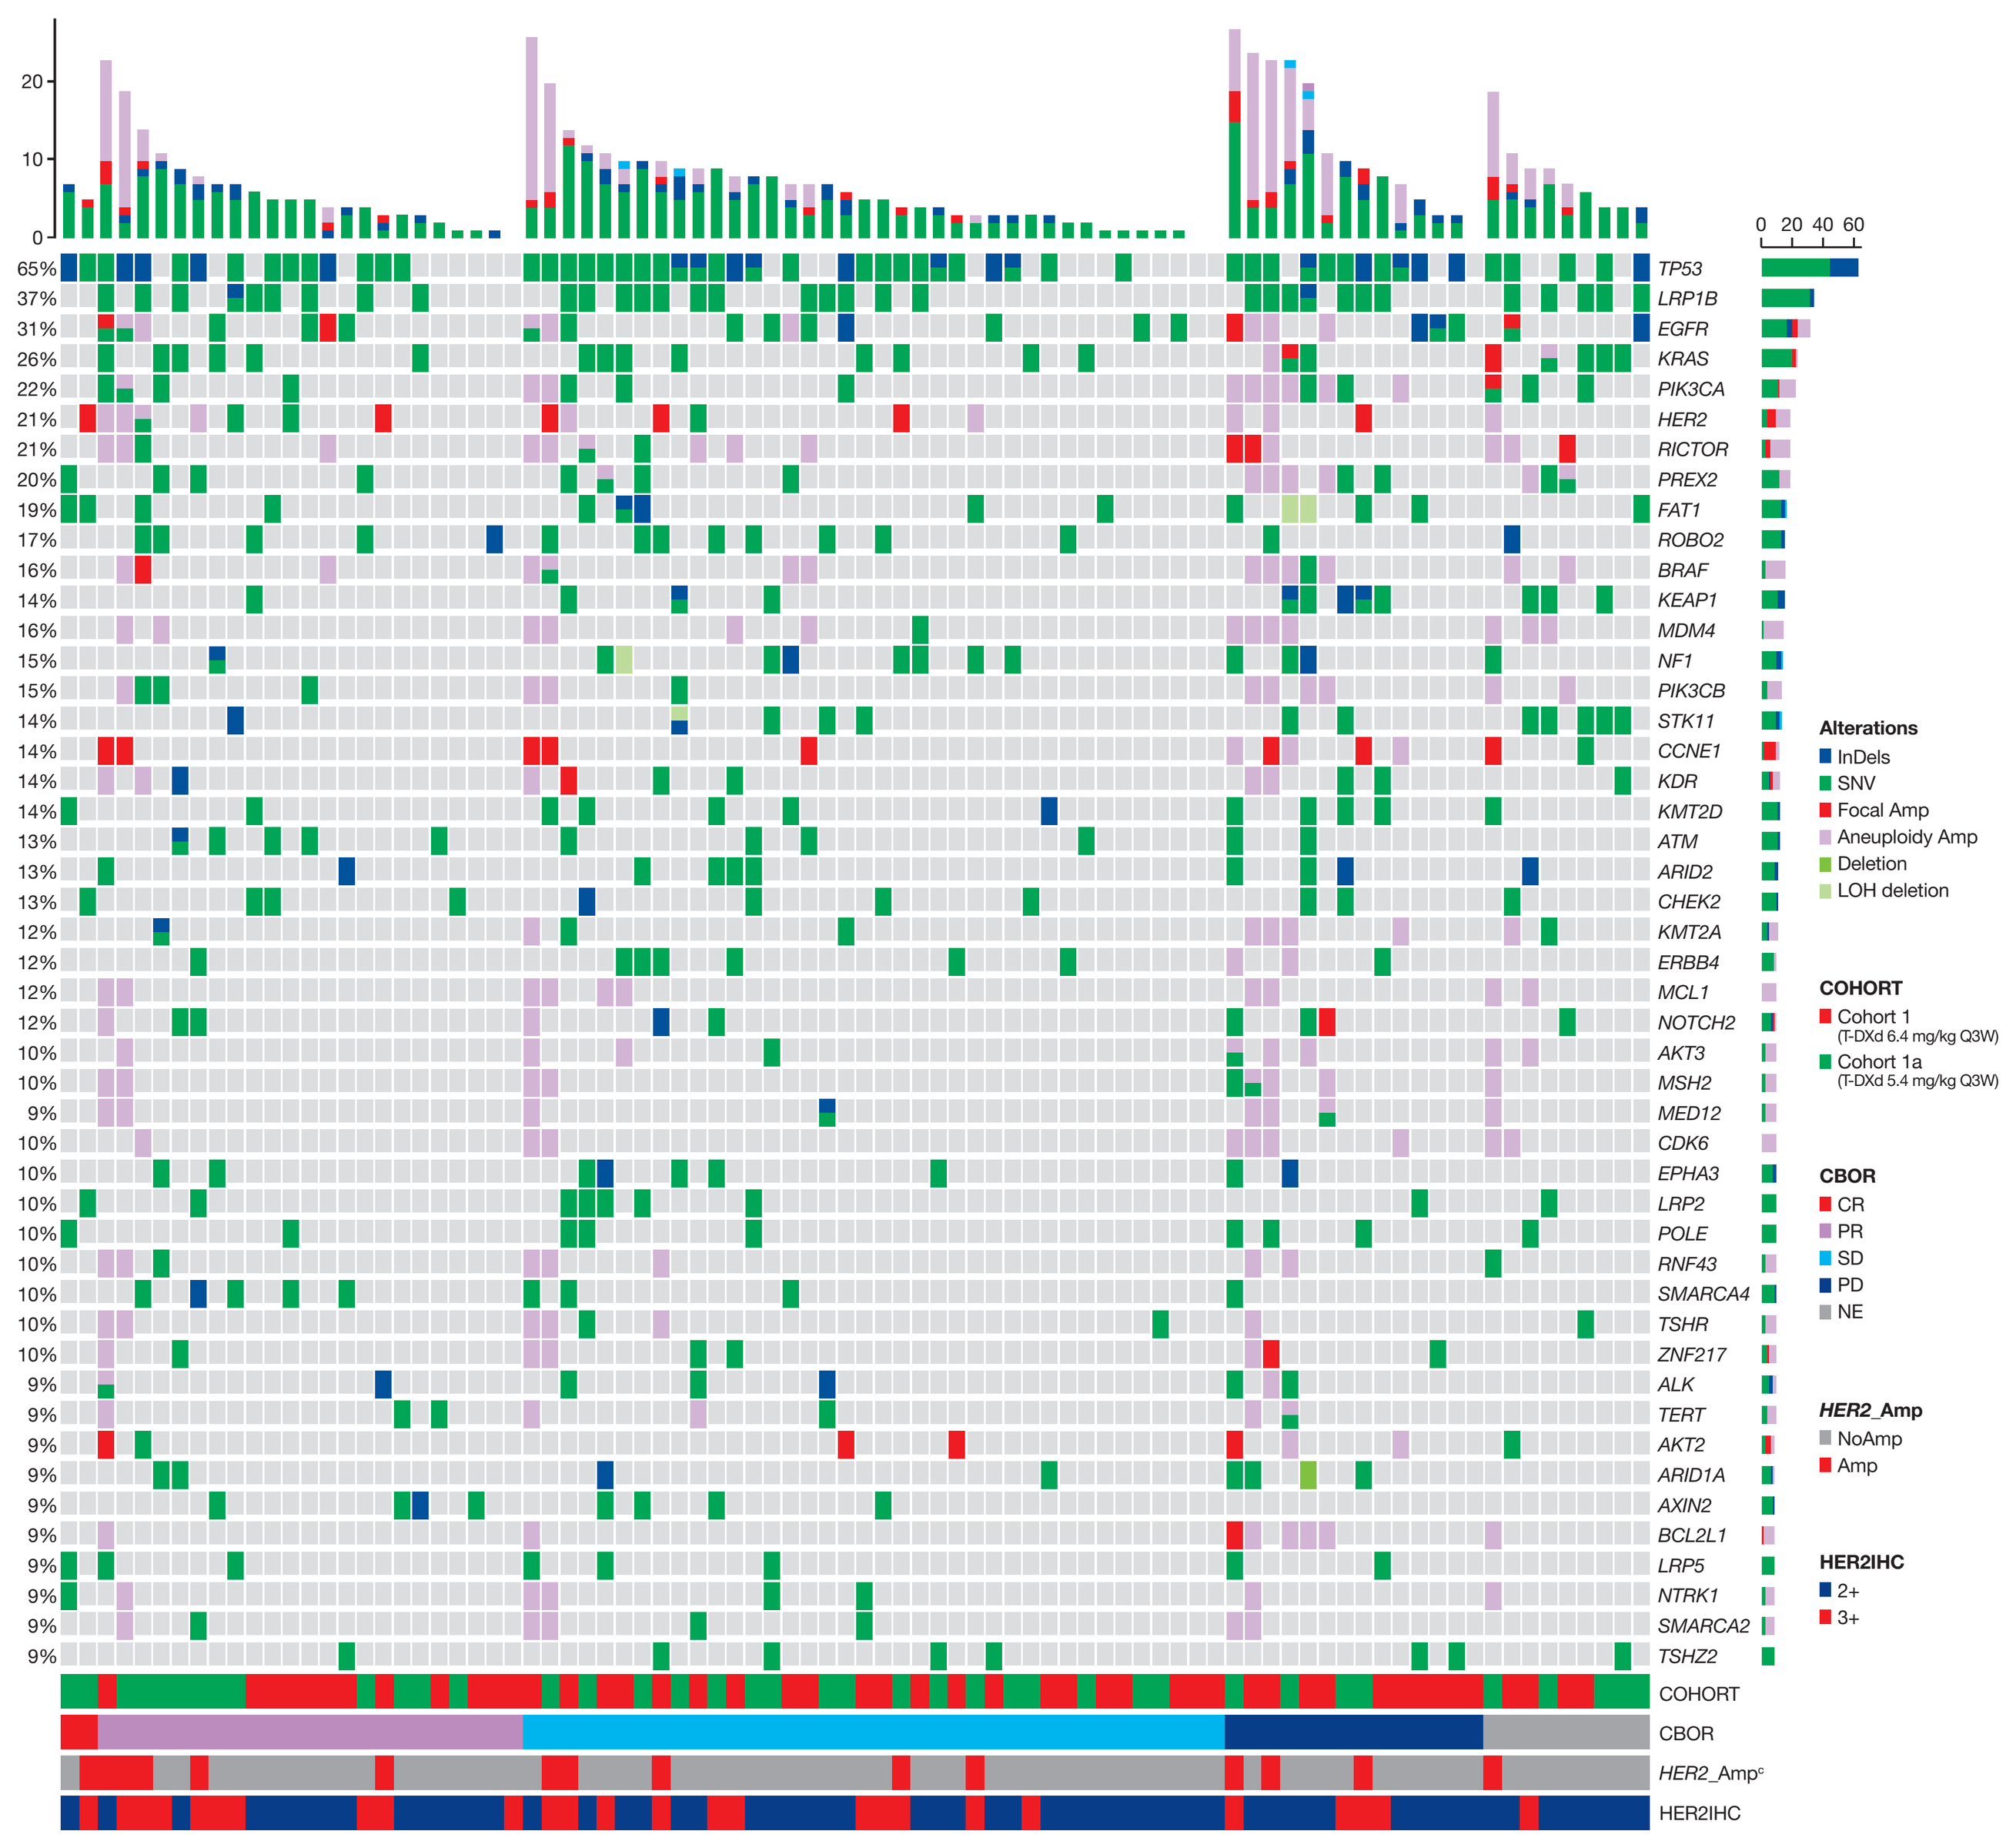

TP53
LRP1B
EGFR
KRAS
PIK3CA
HER2
RICTOR
PREX2
FAT1
ROBO2
BRAF
KEAP1
MDM4
NF1
PIK3CB
STK11
CCNE1
KDR
KMT2D
ATM
ARID2
CHEK2
KMT2A
ERBB4
MCL1
NOTCH2
AKT3
MSH2
MED12
CDK6
EPHA3
LRP2
POLE
RNF43
SMARCA4
TSHR
ZNF217
ALK
TERT
AKT2
ARID1A
AXIN2
BCL2L1
LRP5
NTRK1
SMARCA2
TSHZ2
COHORT
CBOR
HER2_Amp[c]
HER2IHC
Alterations
InDels
SNV
Focal Amp
Aneuploidy Amp
Deletion
LOH deletion
COHORT
Cohort 1 (T-DXd 6.4 mg/kg Q3W)
Cohort 1a (T-DXd 5.4 mg/kg Q3W)
CBOR
CR
PR
SD
PD
NE
HER2_Amp
NoAmp
Amp
HER2IHC
2+
3+

[a]Gene alterations with SNVs, deletions, insertions, and amplifications in 8 or more patients; [b]The *ERBB2* (*HER2*) SNVs that were detected in 4 patients (L1167V, H318Q, G518R, V1128F) were not oncogenic activating mutations based on OncoKB. [c]Based on fluorescence in-situ hybridization analysis in tissue.

- At baseline, activating mutations were identified in *EGFR, KRAS, NRAS,* and *BRAF* (**Table 1**)
- 4 patients had SNVs in *ERBB2* (*HER2*), but none of these were activating mutations
- The mutation status for each patient is available via the QR code on this poster

Table 1. Activating Mutations in Key Oncogenes and Their Prevalence at Baseline

Gene	Criteria for Activating Mutation	Patients, n (%)
EGFR[a]	Oncogenic/likely oncogenic mutation in OncoKB database	16 (18.6)
KRAS	Mutations at G12/G13/A59/Q61/K117/A146	20 (23.3)
NRAS		1 (1.2)
BRAF	Mutations at V600	1 (1.2)

[a]*EGFR* mutations included 7 T790M, 7 L858R, 1 S768I, 4 E746_A750del, 1 P596L, and 2 G719A. No activating mutations in *ERBB2* (*HER2*) were detected.

Efficacy According to Mutation Status

- Among the 16 patients with *EGFR*-activating mutations, 13 had received prior EGFR TKI treatment and among the 10 patients with *EGFR*-activating mutations in cohort 1, 6 patients harbored the T790M mutation, whereas only 1 of 6 patients in cohort 1a had this mutation (**Figure 3**)

Figure 3. Best Percentage Change in Tumor Size From Baseline By HER2 Status, Mutation Status, and Prior Therapy

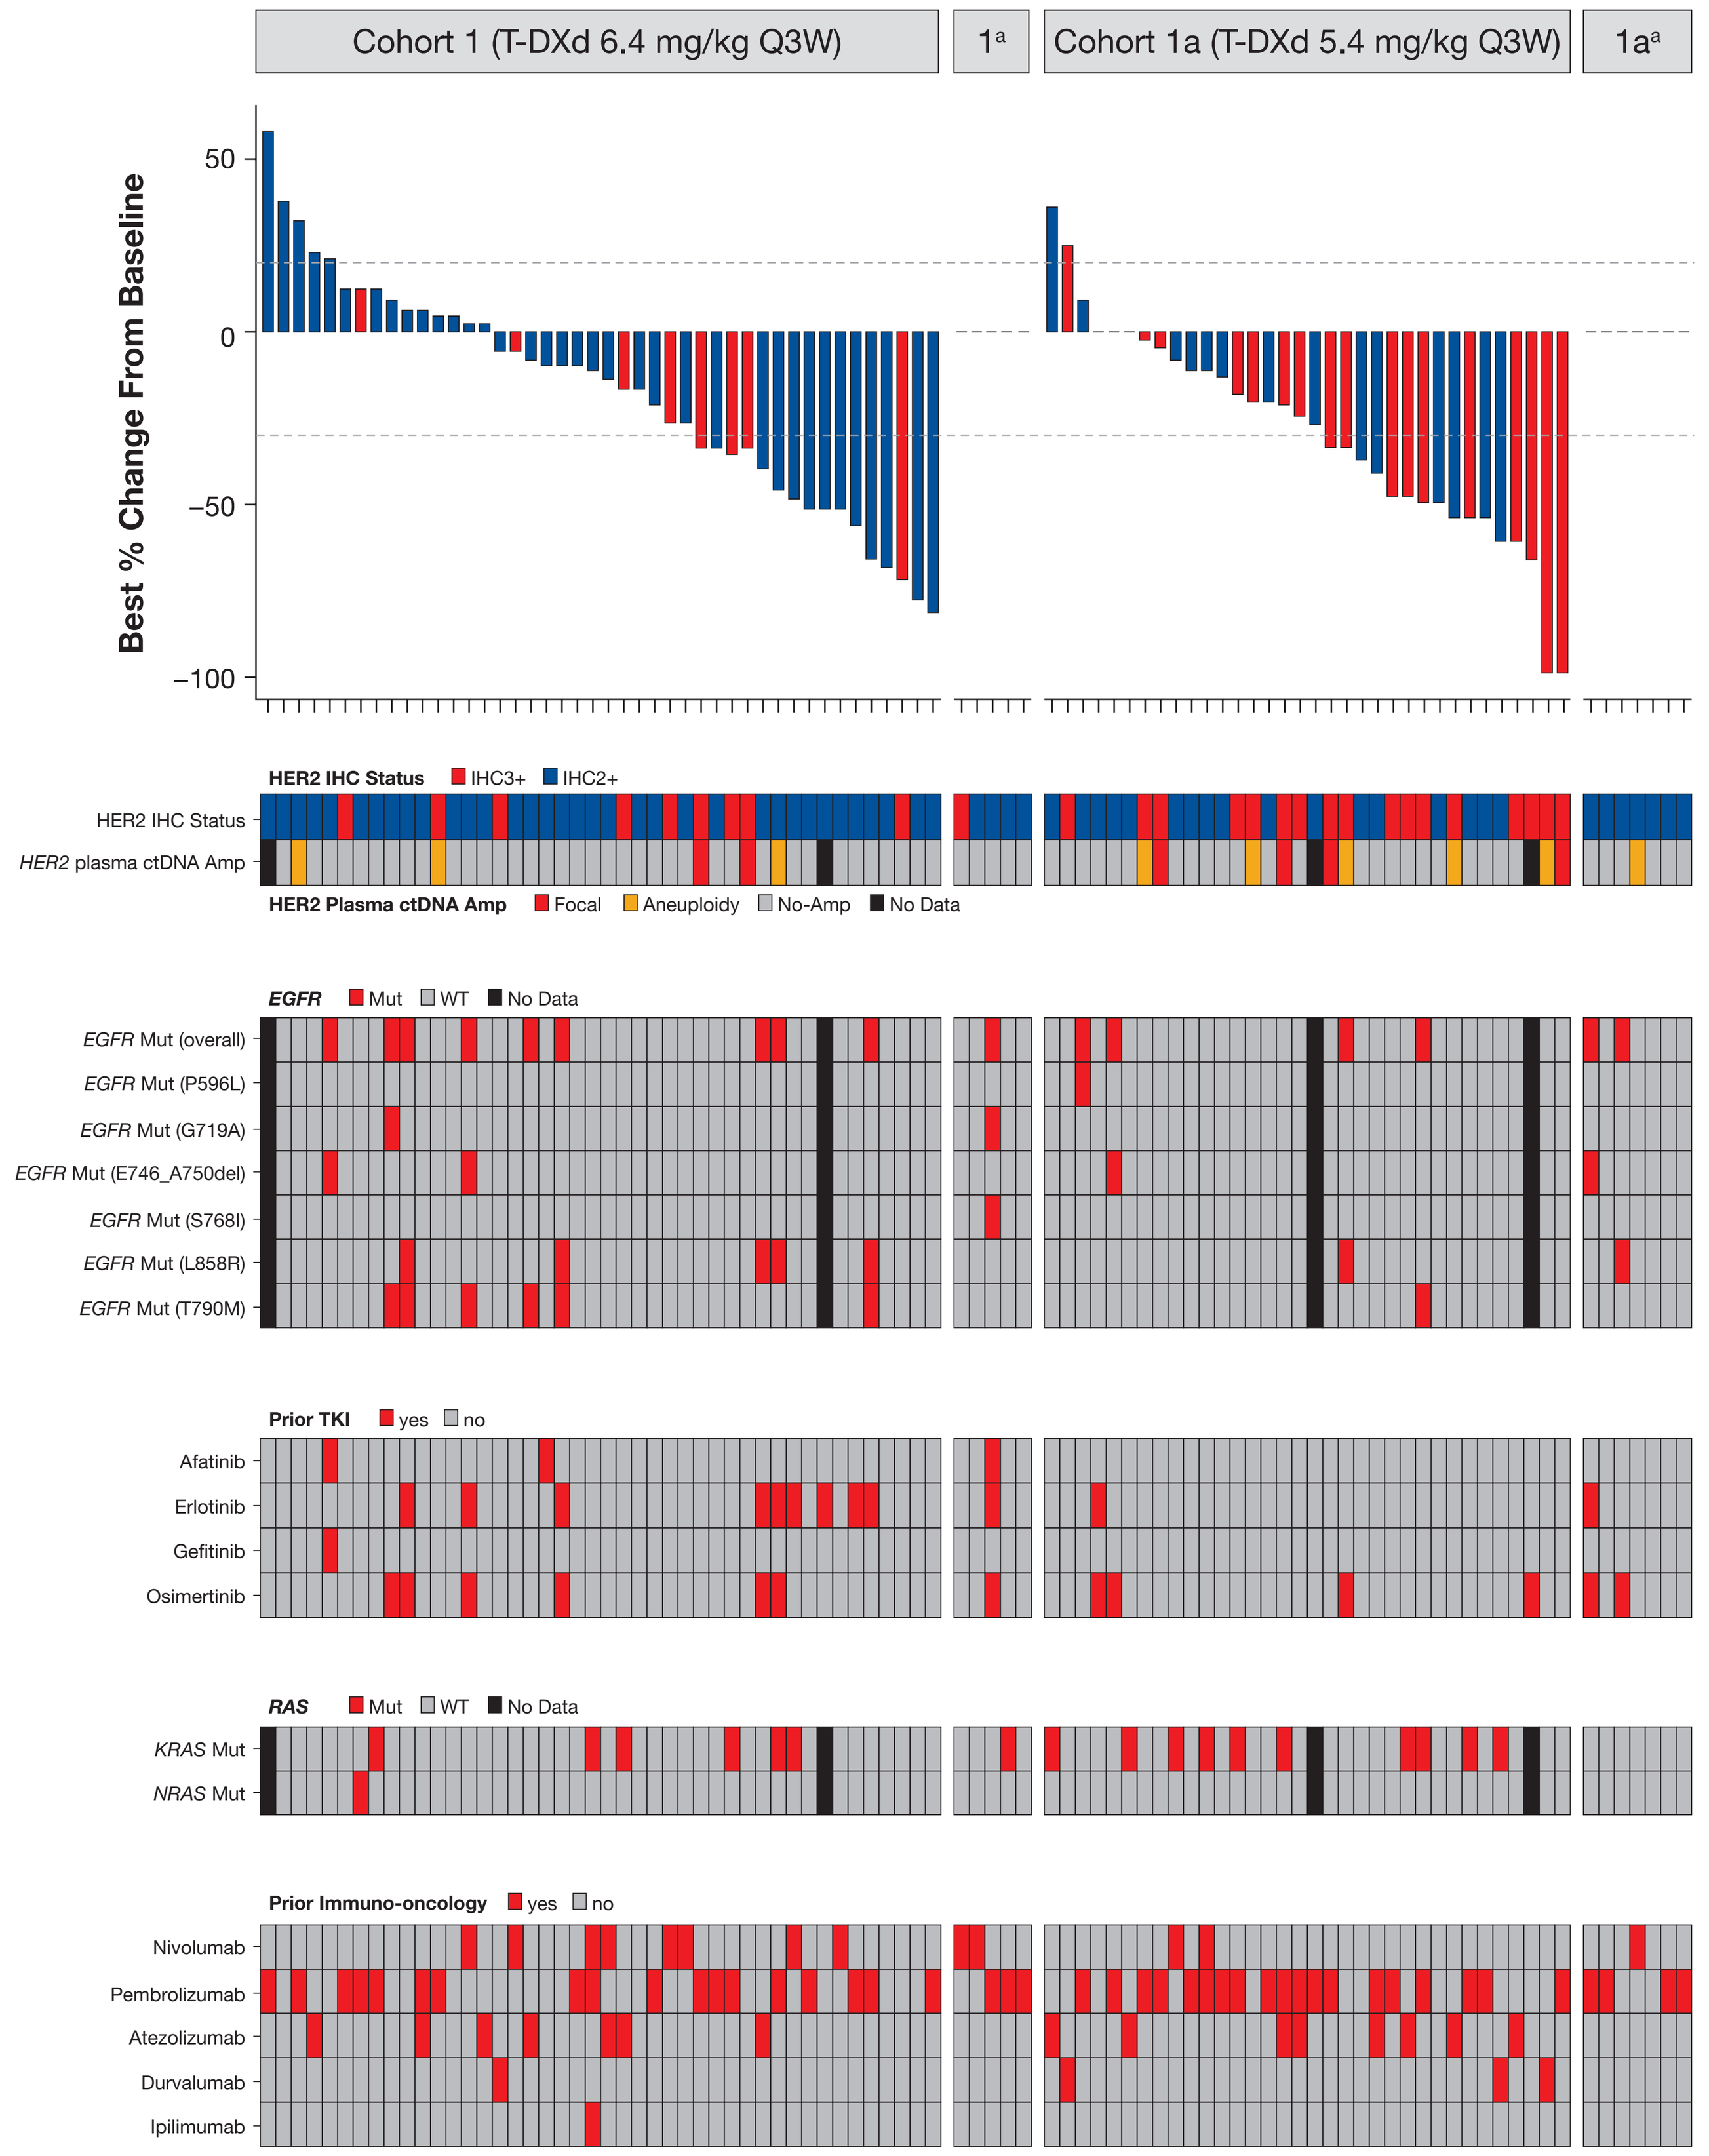

Cohort 1 (T-DXd 6.4 mg/kg Q3W)
1[a]
Cohort 1a (T-DXd 5.4 mg/kg Q3W)
1a[a]
Best % Change From Baseline
50
0
−50
−100
HER2 IHC Status
IHC3+
IHC2+
HER2 IHC Status
HER2 plasma ctDNA Amp
HER2 Plasma ctDNA Amp
Focal
Aneuploidy
No-Amp
No Data
EGFR
Mut
WT
No Data
EGFR Mut (overall)
EGFR Mut (P596L)
EGFR Mut (G719A)
EGFR Mut (E746_A750del)
EGFR Mut (S768I)
EGFR Mut (L858R)
EGFR Mut (T790M)
Prior TKI
yes
no
Afatinib
Erlotinib
Gefitinib
Osimertinib
RAS
Mut
WT
No Data
KRAS Mut
NRAS Mut
Prior Immuno-oncology
yes
no
Nivolumab
Pembrolizumab
Atezolizumab
Durvalumab
Ipilimumab

[a]Patients from cohorts 1 and 1a who were not evaluable for tumor size.
Upper horizontal dashed line indicates a 20% increase in tumor size (disease progression) and lower horizontal dashed line indicates a 30% decrease in tumor size (partial response).

Results (continued)

- The overall ORR among the 86 evaluable patients included in the biomarker analysis was 29.1% (95% CI, 19.8-39.9%), and the overall median PFS was 5.7 months (95% CI, 4.2-7.4 months)
- There were no clear effects of *EGFR*- and *RAS*-activating mutations on ORR and median PFS (**Figures 4, 5**). Mutations in *STK11* and *KEAP1* were associated with shorter median PFS; however, the number of patients with mutations was small (n = 12) (**Figures 4, 6**)

Figure 4. Efficacy ([A] ORR and [B] mPFS) According to Activating Mutation Status

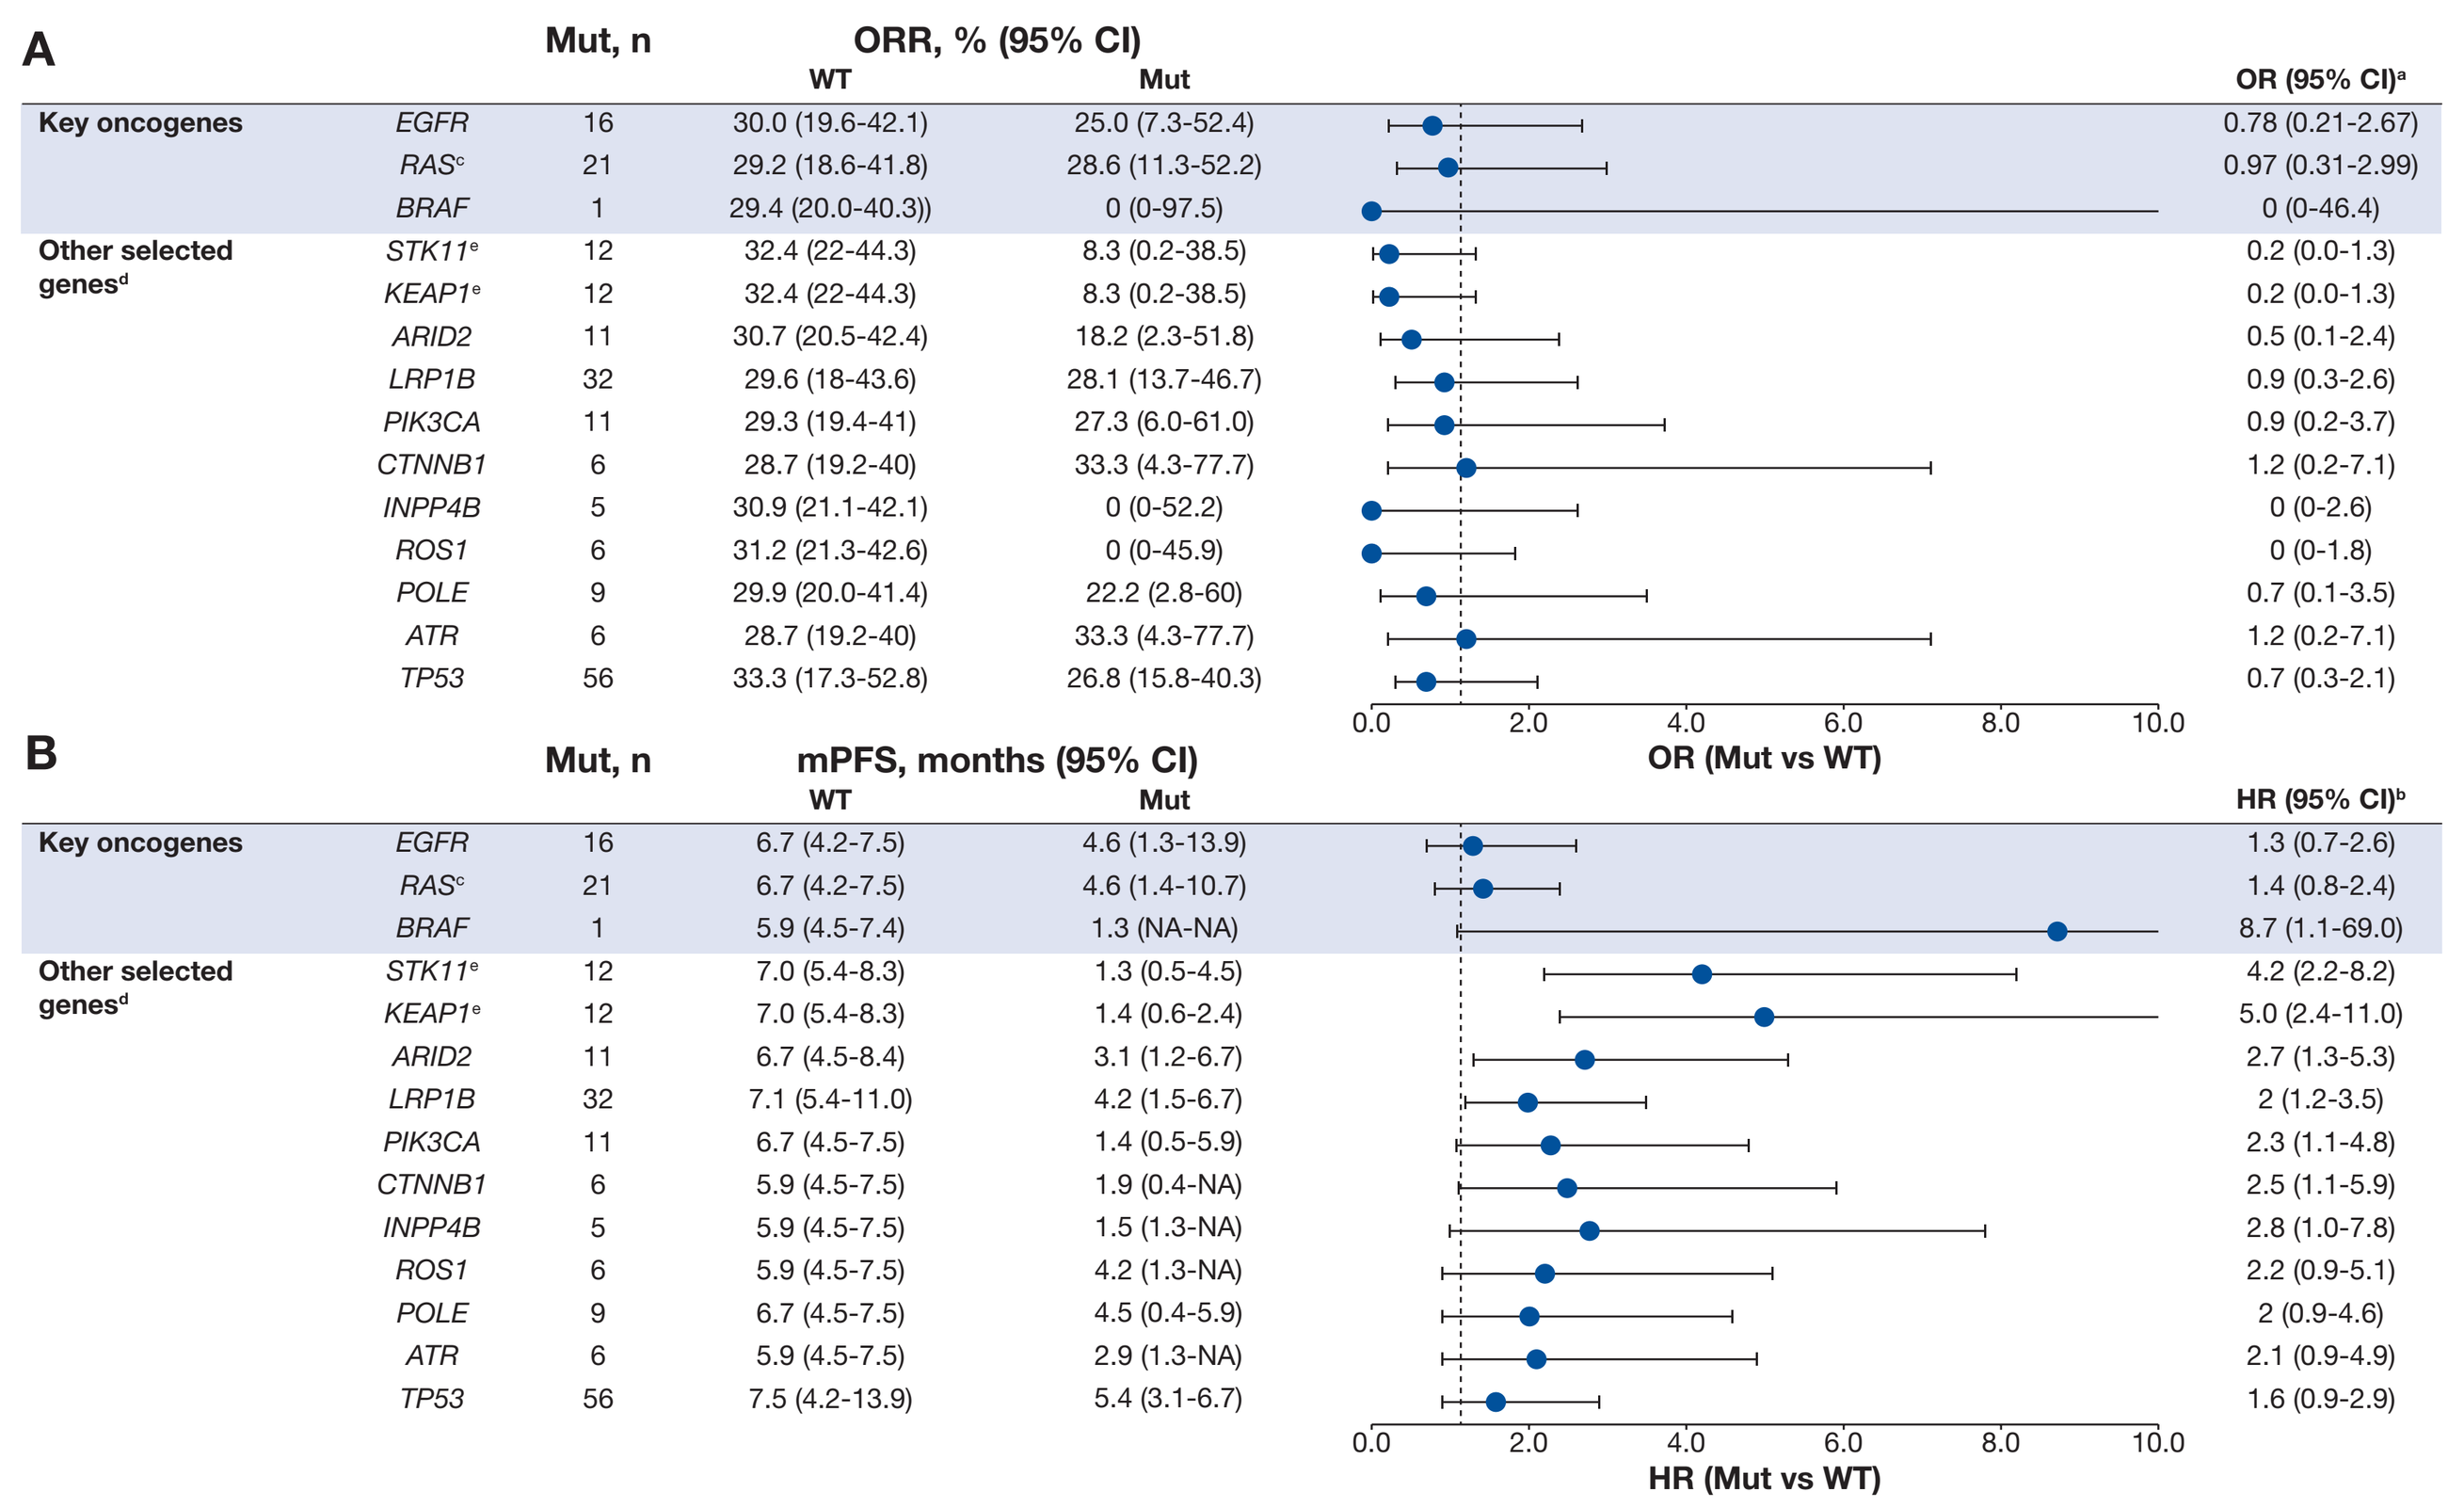

A
Mut, n
ORR, % (95% CI)
WT
Mut
OR (95% CI)[a]
Key oncogenes
EGFR 16 30.0 (19.6-42.1) 25.0 (7.3-52.4) 0.78 (0.21-2.67)
RAS[c] 21 29.2 (18.6-41.8) 28.6 (11.3-52.2) 0.97 (0.31-2.99)
BRAF 1 29.4 (20.0-40.3)) 0 (0-97.5) 0 (0-46.4)
Other selected genes[d]
STK11[e] 12 32.4 (22-44.3) 8.3 (0.2-38.5) 0.2 (0.0-1.3)
KEAP1[e] 12 32.4 (22-44.3) 8.3 (0.2-38.5) 0.2 (0.0-1.3)
ARID2 11 30.7 (20.5-42.4) 18.2 (2.3-51.8) 0.5 (0.1-2.4)
LRP1B 32 29.6 (18-43.6) 28.1 (13.7-46.7) 0.9 (0.3-2.6)
PIK3CA 11 29.3 (19.4-41) 27.3 (6.0-61.0) 0.9 (0.2-3.7)
CTNNB1 6 28.7 (19.2-40) 33.3 (4.3-77.7) 1.2 (0.2-7.1)
INPP4B 5 30.9 (21.1-42.1) 0 (0-52.2) 0 (0-2.6)
ROS1 6 31.2 (21.3-42.6) 0 (0-45.9) 0 (0-1.8)
POLE 9 29.9 (20.0-41.4) 22.2 (2.8-60) 0.7 (0.1-3.5)
ATR 6 28.7 (19.2-40) 33.3 (4.3-77.7) 1.2 (0.2-7.1)
TP53 56 33.3 (17.3-52.8) 26.8 (15.8-40.3) 0.7 (0.3-2.1)
0.0 2.0 4.0 6.0 8.0 10.0
OR (Mut vs WT)
B
Mut, n
mPFS, months (95% CI)
WT
Mut
HR (95% CI)[b]
Key oncogenes
EGFR 16 6.7 (4.2-7.5) 4.6 (1.3-13.9) 1.3 (0.7-2.6)
RAS[c] 21 6.7 (4.2-7.5) 4.6 (1.4-10.7) 1.4 (0.8-2.4)
BRAF 1 5.9 (4.5-7.4) 1.3 (NA-NA) 8.7 (1.1-69.0)
Other selected genes[d]
STK11[e] 12 7.0 (5.4-8.3) 1.3 (0.5-4.5) 4.2 (2.2-8.2)
KEAP1[e] 12 7.0 (5.4-8.3) 1.4 (0.6-2.4) 5.0 (2.4-11.0)
ARID2 11 6.7 (4.5-8.4) 3.1 (1.2-6.7) 2.7 (1.3-5.3)
LRP1B 32 7.1 (5.4-11.0) 4.2 (1.5-6.7) 2 (1.2-3.5)
PIK3CA 11 6.7 (4.5-7.5) 1.4 (0.5-5.9) 2.3 (1.1-4.8)
CTNNB1 6 5.9 (4.5-7.5) 1.9 (0.4-NA) 2.5 (1.1-5.9)
INPP4B 5 5.9 (4.5-7.5) 1.5 (1.3-NA) 2.8 (1.0-7.8)
ROS1 6 5.9 (4.5-7.5) 4.2 (1.3-NA) 2.2 (0.9-5.1)
POLE 9 6.7 (4.5-7.5) 4.5 (0.4-5.9) 2 (0.9-4.6)
ATR 6 5.9 (4.5-7.5) 2.9 (1.3-NA) 2.1 (0.9-4.9)
TP53 56 7.5 (4.2-13.9) 5.4 (3.1-6.7) 1.6 (0.9-2.9)
0.0 2.0 4.0 6.0 8.0 10.0
HR (Mut vs WT)

[a]OR based on Fisher exact model. [b]HR based on Cox proportional hazard model. [c]*KRAS/NRAS*. [d]Selected based on *P* value ≤0.1 according to log rank test for significance for PFS (not shown; a total of 52 genes were analyzed with a frequency of gene mutation ≥5). [e]*STK11* and *KEAP1* mutations occurred concurrently in 7 patients.

Figure 5. Kaplan-Meier Analyses for Effects of (A) *EGFR* and (B) *RAS* Activating Mutations on PFS

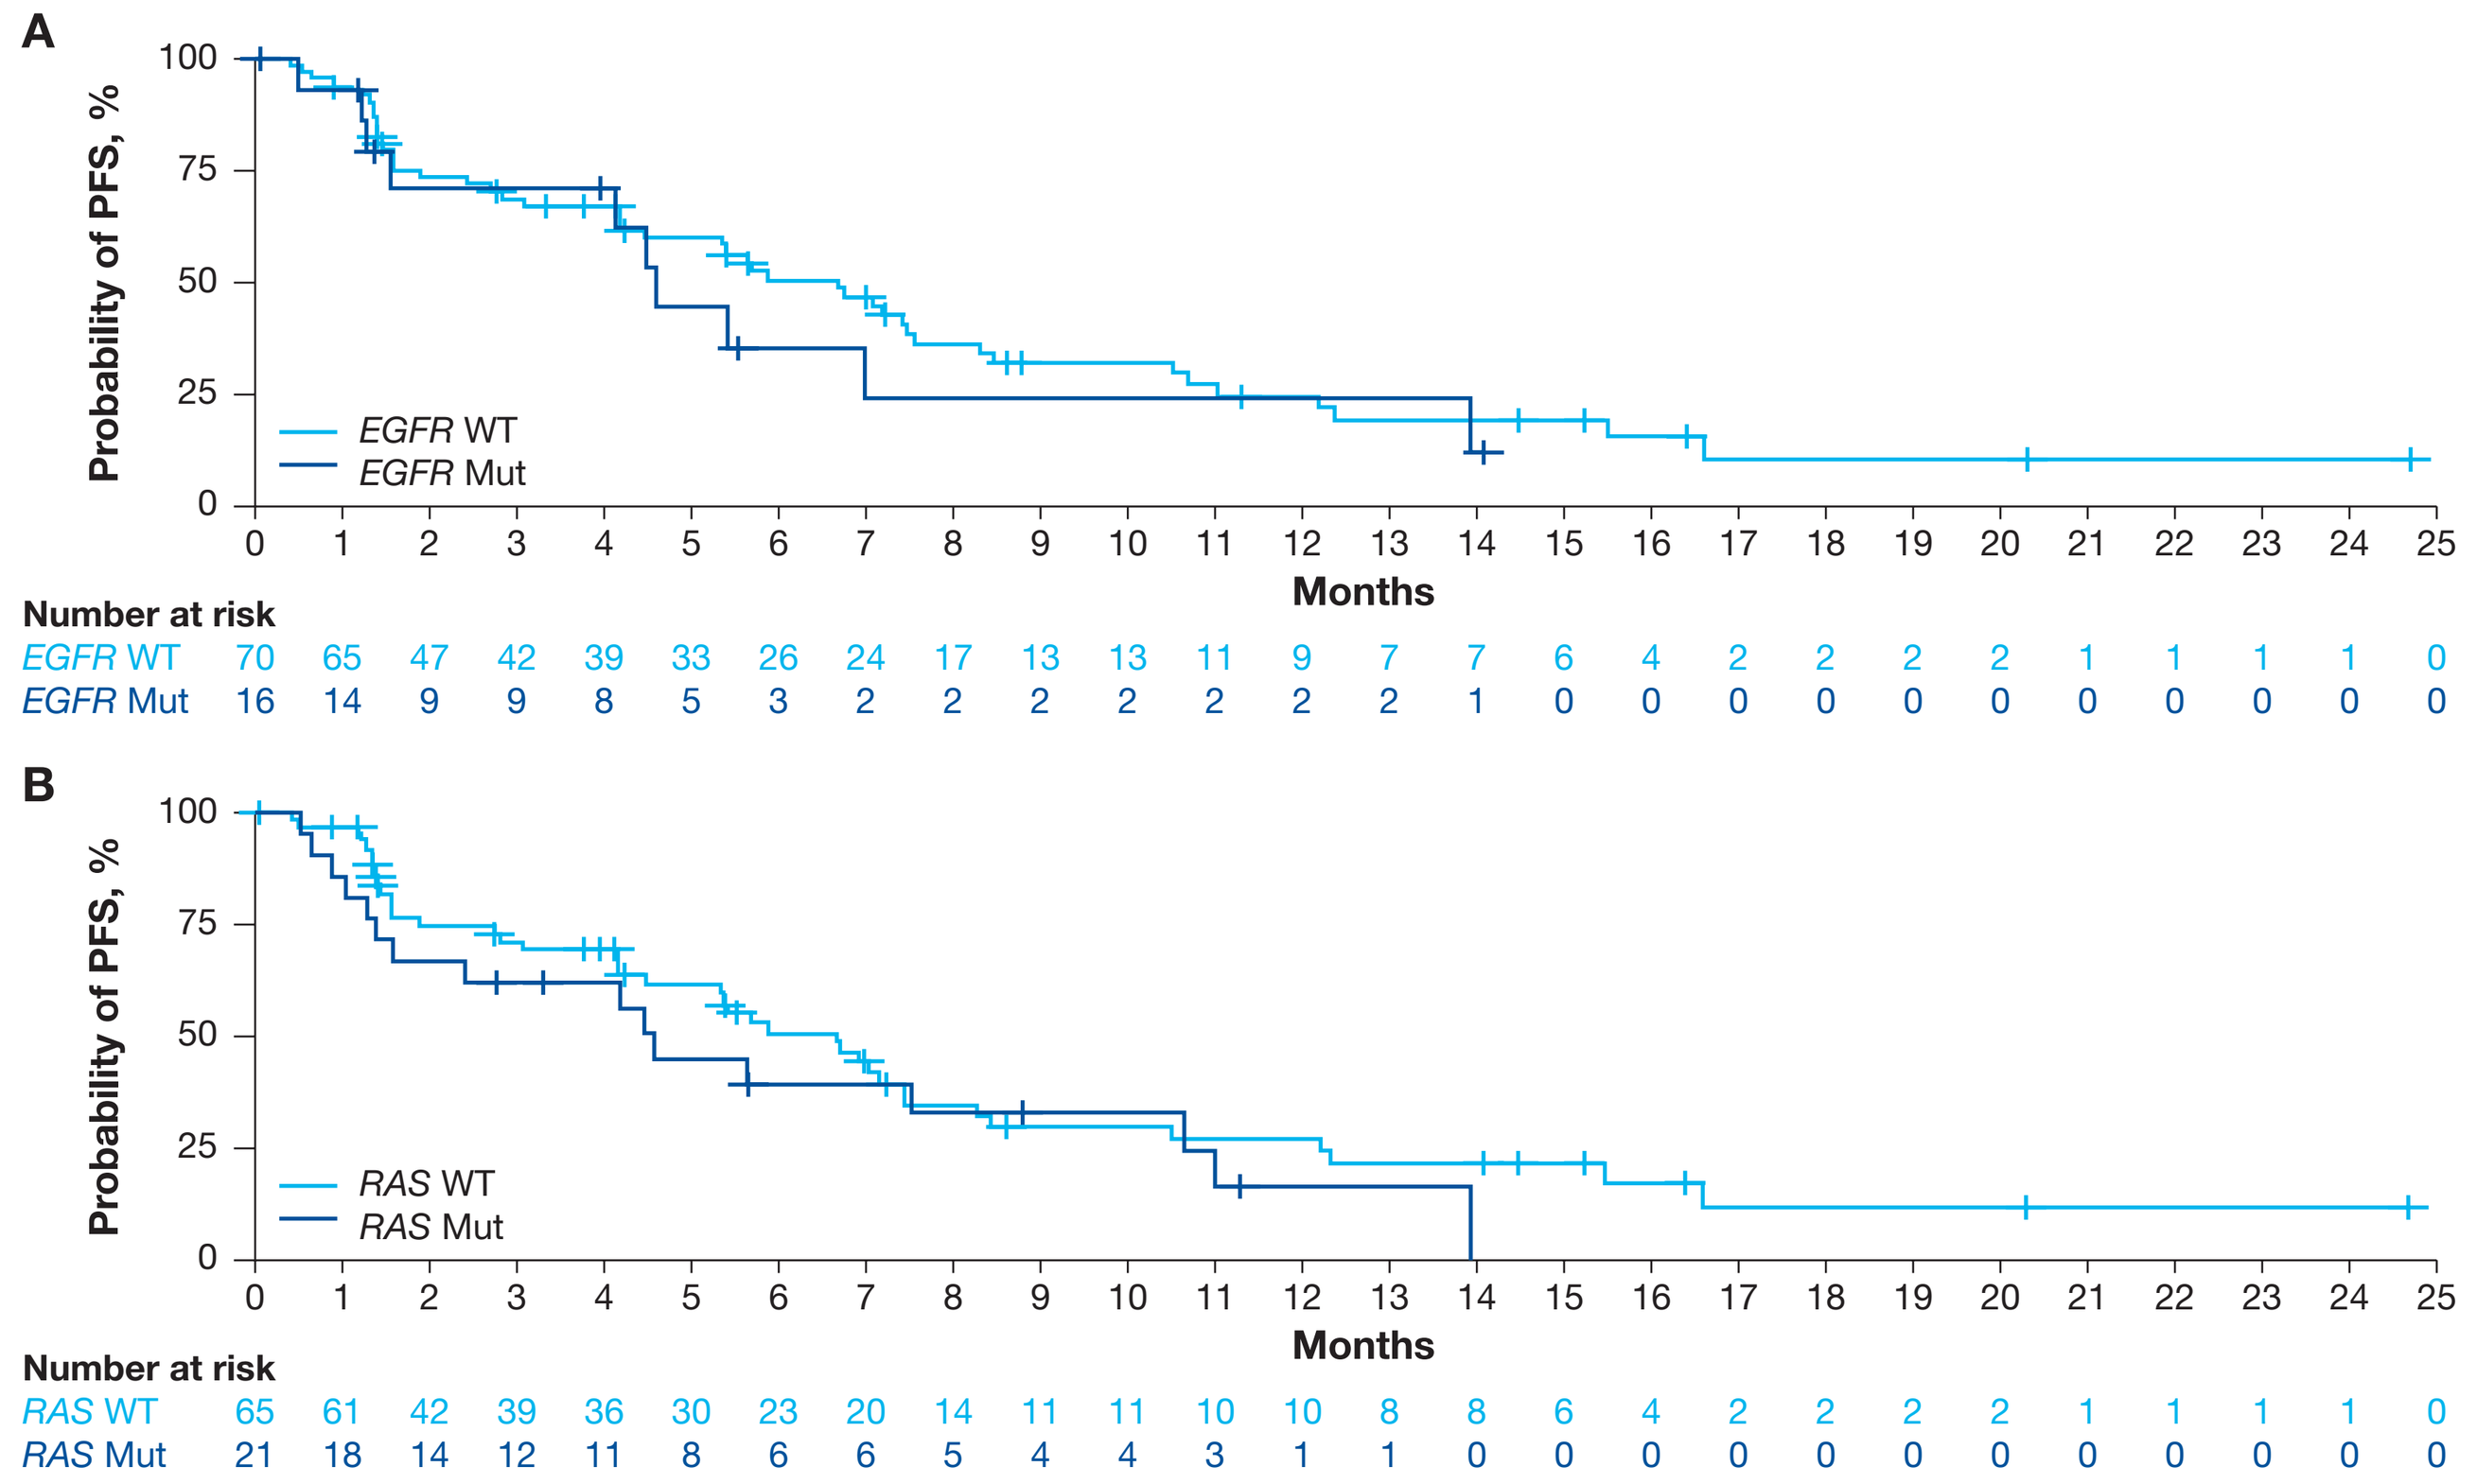

A
Probability of PFS, %
100
75
50
25
0
EGFR WT
EGFR Mut
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25
Months
Number at risk
EGFR WT 70 65 47 42 39 33 26 24 17 13 13 11 9 7 7 6 4 2 2 2 2 1 1 1 1 0
EGFR Mut 16 14 9 9 8 5 3 2 2 2 2 2 2 2 1 0 0 0 0 0 0 0 0 0 0 0
B
Probability of PFS, %
100
75
50
25
0
RAS WT
RAS Mut
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25
Months
Number at risk
RAS WT 65 61 42 39 36 30 23 20 14 11 11 10 10 8 8 6 4 2 2 2 2 1 1 1 1 0
RAS Mut 21 18 14 12 11 8 6 6 5 4 4 3 1 1 0 0 0 0 0 0 0 0 0 0 0 0

Figure 6. Kaplan-Meier Analyses for Effects of (A) *STK11* and (B) *KEAP1* Activating Mutations on PFS

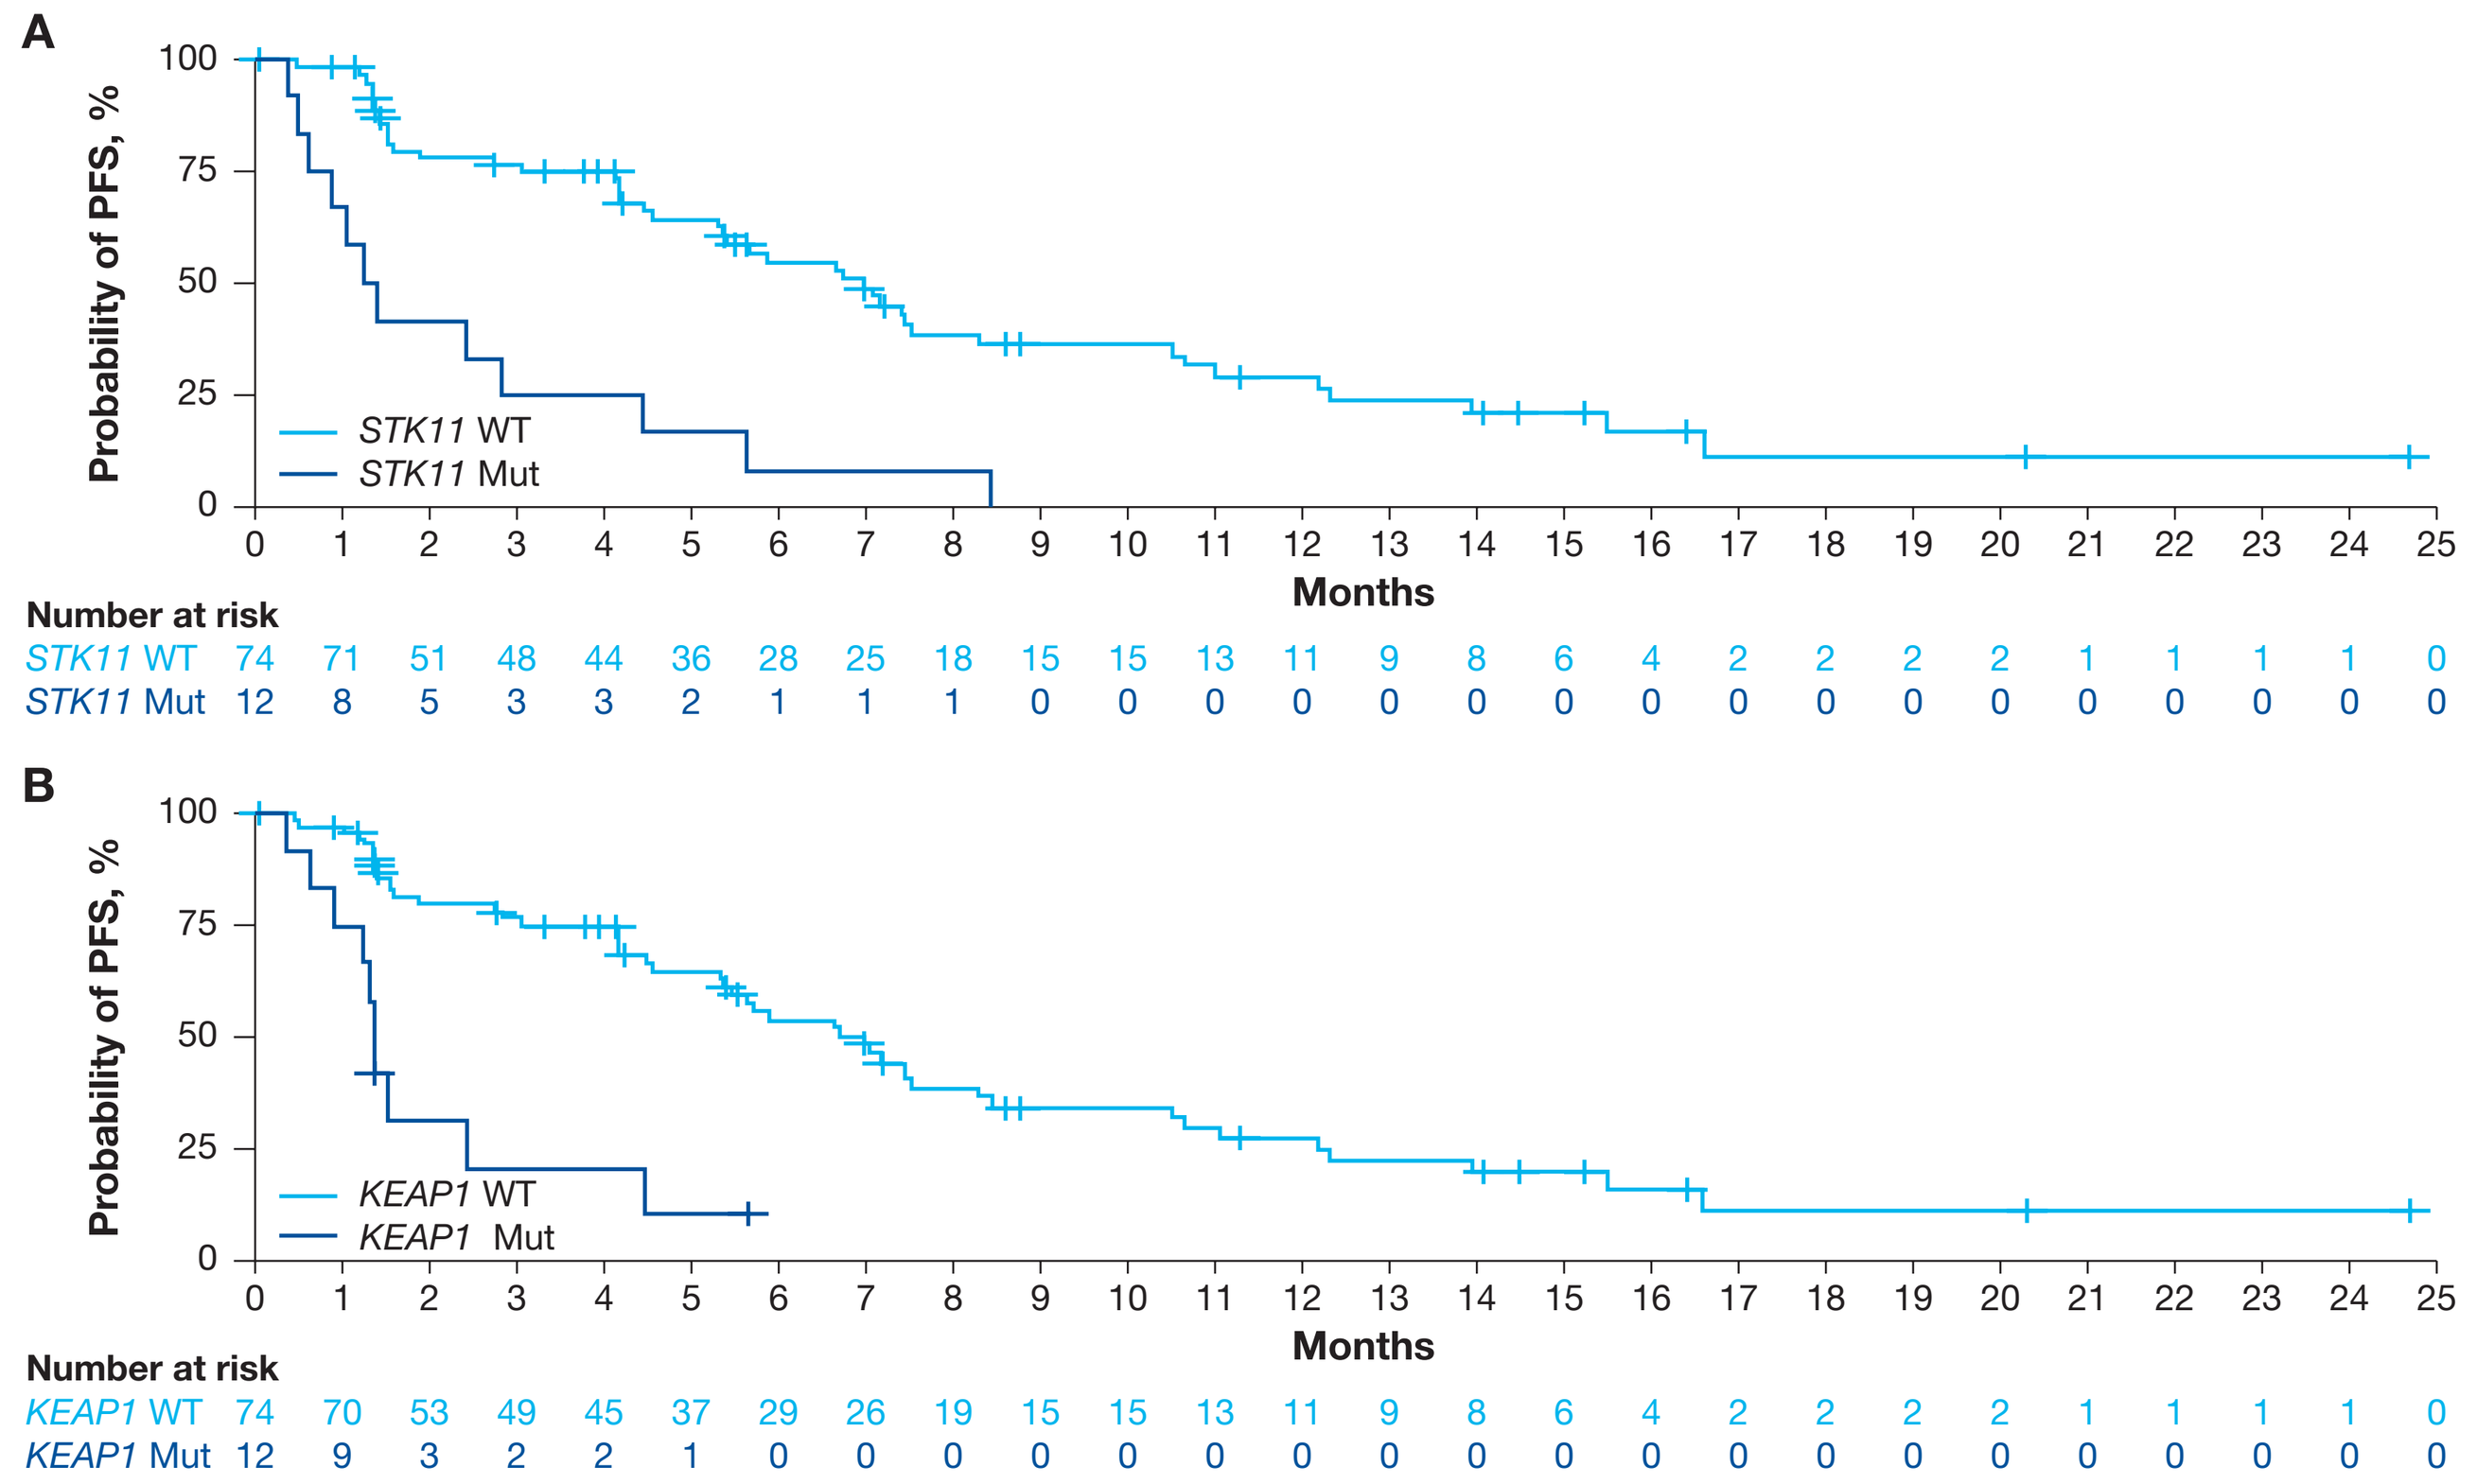

A
Probability of PFS, %
100
75
50
25
0
STK11 WT
STK11 Mut
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25
Months
Number at risk
STK11 WT 74 71 51 48 44 36 28 25 18 15 15 13 11 9 8 6 4 2 2 2 2 1 1 1 1 0
STK11 Mut 12 8 5 3 3 2 1 1 1 0 0 0 0 0 0 0 0 0 0 0 0 0 0 0 0 0
B
Probability of PFS, %
100
75
50
25
0
KEAP1 WT
KEAP1 Mut
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25
Months
Number at risk
KEAP1 WT 74 70 53 49 45 37 29 26 19 15 15 13 11 9 8 6 4 2 2 2 2 1 1 1 1 0
KEAP1 Mut 12 9 3 2 2 1 0 0 0 0 0 0 0 0 0 0 0 0 0 0 0 0 0 0 0 0

Abbreviations

Amp, amplification; CBOR, confirmed best overall response; CR, complete response; ctDNA, circulating tumor DNA; DCR, disease control rate; DoR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; HER2, human epidermal growth factor receptor 2; *HER2*m, human epidermal growth factor receptor 2 mutated; HR, hazard ratio; ICR, independent central review; IHC, immunohistochemistry; InDel, insertion/deletion; INV, investigator assessed; LOH, loss of heterozygosity; mPFS, median progression-free survival; mut, mutant; NA, not available; NE, not evaluable; NSCLC, non–small cell lung cancer; OR, odds ratio; ORR, objective response rate; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PR, partial response; Q3W, every 3 weeks; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; SNV, single nucleotide variant; T-DXd, trastuzumab deruxtecan; TKI, tyrosine kinase inhibitor; WT, wild type.

References

1. Heinmöller P et al. *Clin Cancer Res*. 2003;9:5238-43.
2. Liu S et al. *Clin Cancer Res*. 2018;24:2594-604.
3. Enhertu. Prescribing information. Daiichi Sankyo; 2022.
4. Planchard D et al. *Ann Oncol*. 2018;29(Suppl 4):iv192-iv237.
5. Wu Y-L et al. *Ann Oncol*. 2019;30:171-210.
6. Li BT et al. *N Engl J Med*. 2022;386(3):241-251.
7. Smit EF et al. Proceedings of the European Society of Medical Oncology Annual Meeting 2022. Poster #975P.

Acknowledgments

Under the guidance of authors, medical writing and editorial support was provided by Greg Town, BSc, and Sara Duggan, PhD, of ApotheCom and was funded by Daiichi Sankyo, Inc.

Disclosures

Prof Egbert F. Smit declares no conflicts of interest.

DESTINY-Lung01