A phase 1, 2-part, multicenter, first-in-human, dose-escalation and dose-expansion study of DS-1103a with trastuzumab deruxtecan (T-DXd) in patients with advanced solid tumors

375TiP

Ludimila Cavalcante,[1,*] Lillian L. Siu,[2] John Toso,[3] Derek Mires,[3] Ryota Shiga,[3] Jessica Maruca,[3,4] Girma Ayele,[3] Pradeep Kumar Vuppala,[3] Yougan Cheng,[3] Leanne Huang,[3] Jaime E. Connolly Rohrbach,[3] Yoko Ishimoto,[5] Mayumi Sue,[5] Philippe L. Bedard[2]

[1]Novant Health Cancer Institute, Charlotte, NC, USA; [2]Princess Margaret Cancer Centre, Toronto, ON, Canada; [3]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [4]Rutgers University, New Brunswick, NJ, USA; [5]Daiichi Sankyo Co., Ltd., Tokyo, Japan.

*Presenting author.

INTRODUCTION

- Therapeutic advances with immune checkpoint inhibitors have rapidly emerged and improved outcomes for patients with various types of cancer; however, many challenges and unmet needs remain[1]
- Growing evidence demonstrates that tumor-associated macrophages in the tumor microenvironment promote cancer cell proliferation and immunosuppression[2]
 - Prohibiting the interaction between cancer cells and these macrophages may be the key to tumor suppression and cancer treatment[2]
- Blockade of the signal regulatory protein α (SIRPα)-CD47 interaction is a promising pathway to restore the antitumor immune functions of macrophages and reduce tumor growth[3,4]
- DS-1103a, a recombinant humanized immunoglobulin G4 anti-SIRPα antibody, blocks the SIRPα-CD47 pathway by binding to the major human variants of SIRPα
- Trastuzumab deruxtecan (T-DXd), an antibody-drug conjugate consisting of an anti–human epidermal growth receptor 2 (HER2) antibody and topoisomerase I inhibitor payload, is approved for the treatment of various cancers, including breast cancer, non-small cell lung cancer, and gastric cancer[5]
- In nonclinical studies, DS-1103a enhanced trastuzumab- or T-DXd–induced antibody-dependent cellular phagocytosis (ADCP), suggesting a potential benefit of the combination
 - Antimouse SIRPα antibody (a surrogate of DS-1103a) enhanced T-DXd antitumor efficacy in a syngeneic mouse model[6]
 - In a quantitative systems pharmacology model, the starting dose of DS-1103a for use in future first-in-human studies (100 mg intravenously [IV] every 3 weeks) in combination with T-DXd was determined both in vitro and in vivo based on the achievement of intratumoral occupancy and induced ADCP activity[7]
- This phase 1 study will assess the preliminary safety and efficacy of DS-1103a in combination with T-DXd in advanced solid tumors (ClinicalTrials.gov Identifier: NCT05765851)

OBJECTIVES

- To investigate the safety and tolerability and determine the recommended dose for expansion (RDE) of DS-1103a in combination with T-DXd
- To further evaluate the safety and the efficacy of DS-1103a at the RDE in combination with T-DXd in patients with HER2-low metastatic breast cancer

METHODS

- This is a phase 1, global, multicenter, first-in-human, 2-part, dose-escalation and dose-expansion study designed to evaluate DS-1103a in combination with T-DXd in patients with advanced (unresectable or metastatic) HER2-expressing/HER2-mutated solid tumors
- The study design is displayed in **Figure 1**
- Dose-escalation (Part 1) will enroll approximately 30 patients with pathologically documented HER2-expressing (eg, immunohistochemistry [IHC] 1+ or greater) or HER2-mutated (with activating mutation determined by next-generation sequencing or other appropriate analysis techniques) advanced solid tumors not amenable to standard of care
- Dose-expansion (Part 2) will enroll approximately 48 patients with HER2-low (IHC 2+/in situ hybridization [ISH]– or IHC 1+ [ISH– or untested]) breast cancer who received 1 to 2 prior lines of chemotherapy in the recurrent or metastatic setting
- Key inclusion criteria
 - Adults ≥18 years of age at the time of informed consent
 - The presence of ≥1 measurable lesion based on computed tomography or magnetic resonance imaging per Response Evaluation Criteria in Solid Tumors (RECIST) v1.1 as assessed by the investigator
 - Eastern Cooperative Oncology Group performance status of 0 or 1
- Key exclusion criteria
 - Prior treatment with an anti-CD47 or anti-SIRPα therapy
 - An inadequate treatment washout period prior to the start of study treatment, defined as follows:
 - Major surgery: ≤4 weeks (or ≤2 weeks for low-invasive cases)
 - Radiation therapy, including palliative stereotactic radiation therapy to the chest: ≤4 weeks
 - Palliative stereotactic radiation therapy to other anatomic areas: ≤2 weeks
 - Received any systemic agent from a previous treatment regimen or clinical study within the specified time frame prior to administration of study treatment
- Study drug(s) administration
 - In Part 1, DS-1103a monotherapy will be administered via IV infusion starting on Cycle 1 Day 1, with a proposed starting dose of 100 mg
 - Starting on Cycle 2 Day 1, T-DXd (5.4 mg/kg) will also be administered via IV infusion
 - In Part 2, patients will receive an RDE of DS-1103a, as established by Part 1, in combination with T-DXd starting on Cycle 1 Day 1
- The anticipated total duration of the study is expected to be approximately 44 months

Figure 1. Study design

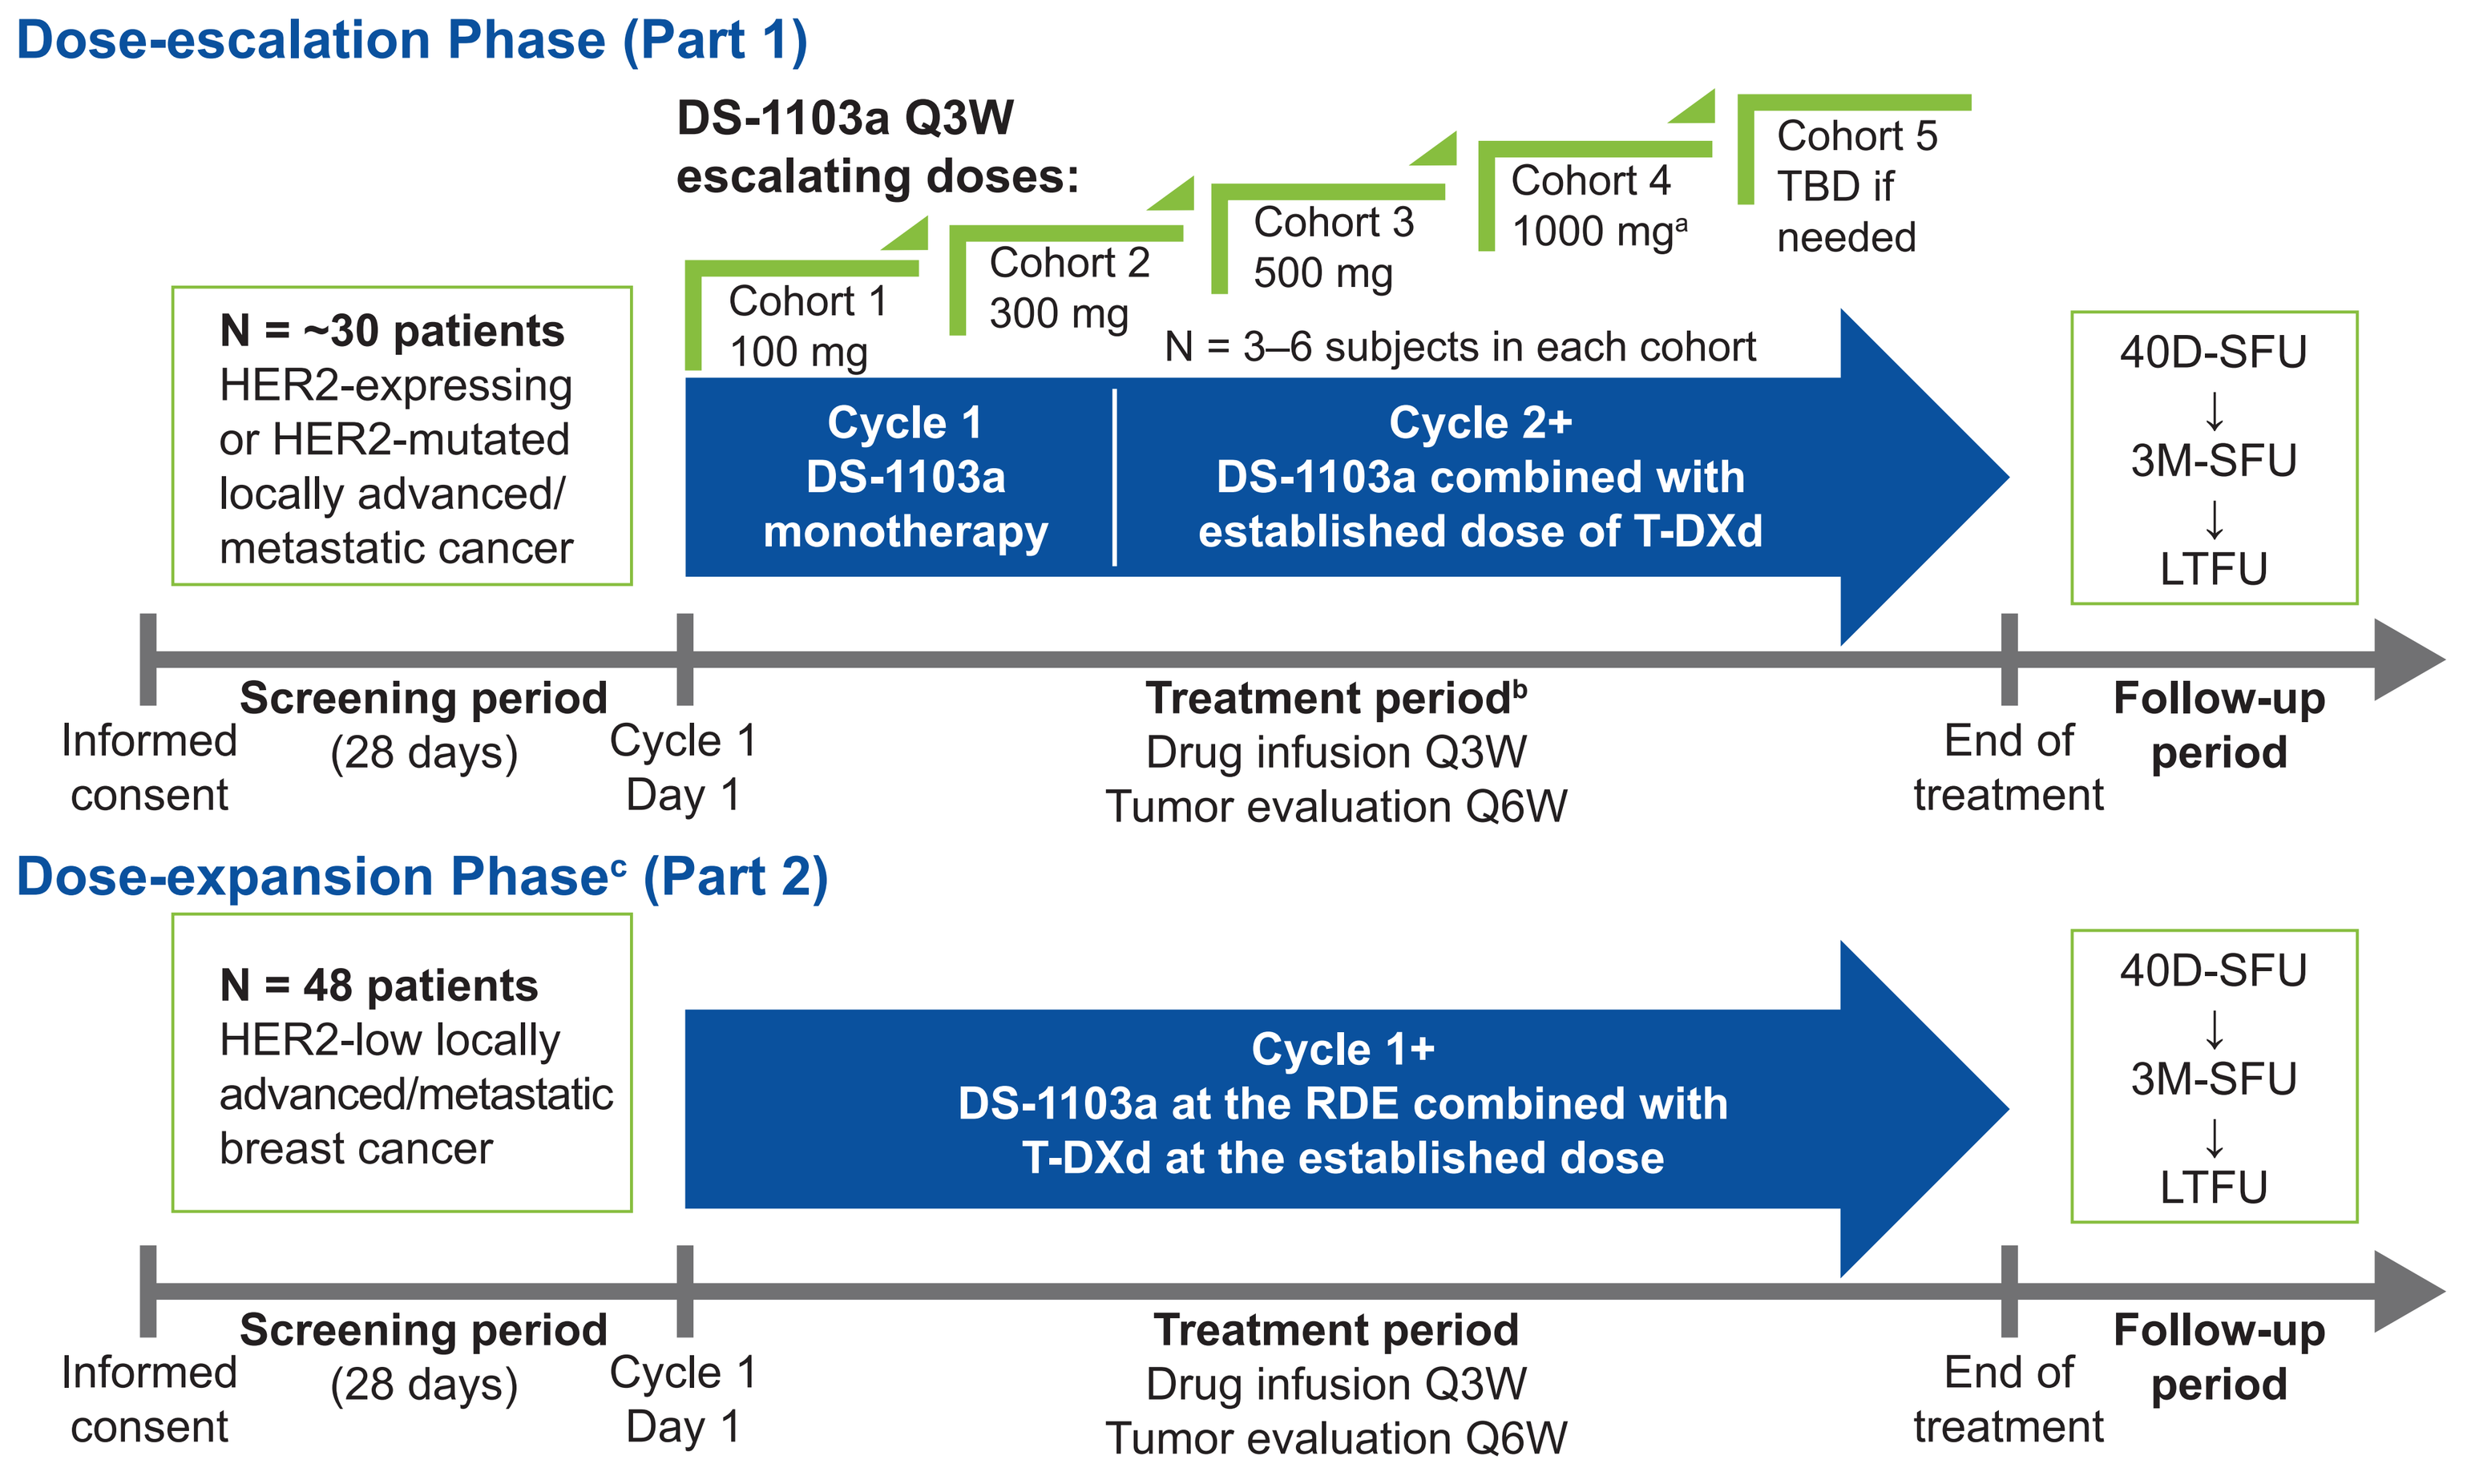
Dose-escalation Phase (Part 1)
DS-1103a Q3W escalating doses:
Cohort 1 100 mg
Cohort 2 300 mg
Cohort 3 500 mg
Cohort 4 1000 mg[a]
Cohort 5 TBD if needed
N = 3–6 subjects in each cohort
N = ~30 patients HER2-expressing or HER2-mutated locally advanced/ metastatic cancer
Cycle 1 DS-1103a monotherapy
Cycle 2+ DS-1103a combined with established dose of T-DXd
40D-SFU ↓ 3M-SFU ↓ LTFU
Informed consent
Screening period (28 days)
Cycle 1 Day 1
Treatment period[b] Drug infusion Q3W Tumor evaluation Q6W
End of treatment
Follow-up period
Dose-expansion Phase[c] (Part 2)
N = 48 patients HER2-low locally advanced/metastatic breast cancer
Cycle 1+ DS-1103a at the RDE combined with T-DXd at the established dose
40D-SFU ↓ 3M-SFU ↓ LTFU
Informed consent
Screening period (28 days)
Cycle 1 Day 1
Treatment period Drug infusion Q3W Tumor evaluation Q6W
End of treatment
Follow-up period

HER2, human epidermal growth receptor 2; Q3W, every 3 weeks; Q6W, every 6 weeks; T-DXd, trastuzumab deruxtecan; TBD, to be determined; D, day; M, month; SFU, safety follow-up; LTFU, long-term follow-up; RDE, recommended dose for expansion.
[a]Use of a DS-1103a dose >1000 mg may be allowed depending on the observed safety, pharmacokinetic and pharmacodynamic data and the recommendation of the Bayesian optimal interval design.
[b]The dose-limiting toxicity evaluation period includes Cycles 1 and 2 in Part 1.
[c]Further discussion with health authorities on the study design of Part 2 is planned prior to commencing dose expansion.

- Endpoints and evaluations
 - In Part 1, the primary endpoint will be to investigate the safety and tolerability of DS-1103a and to determine the RDE of DS-1103a in combination with T-DXd
 - In Part 2, the primary endpoint will be to further evaluate the safety and tolerability of DS-1103a at the RDE in combination with T-DXd and determine the objective response rate (ORR)
 - The ORR will be assessed by blinded independent central review per RECIST v1.1
 - Secondary endpoints include the antitumor activity of DS-1103a in combination with T-DXd, the pharmacokinetic profile of DS-1103a, and the incidence of antidrug antibodies against DS-1103a
 - Safety evaluations will include endpoints such as dose-limiting toxicities, treatment-emergent adverse events (TEAEs), serious adverse events, adverse events of special interest, and TEAEs leading to study drug discontinuation

ENROLLMENT STATUS

- First patient dosed: June 2023
- Estimated primary completion: June 2026
- Enrolling and planned recruitment sites (**Figure 2**)

Figure 2. Global map of enrolling and planned recruitment sites

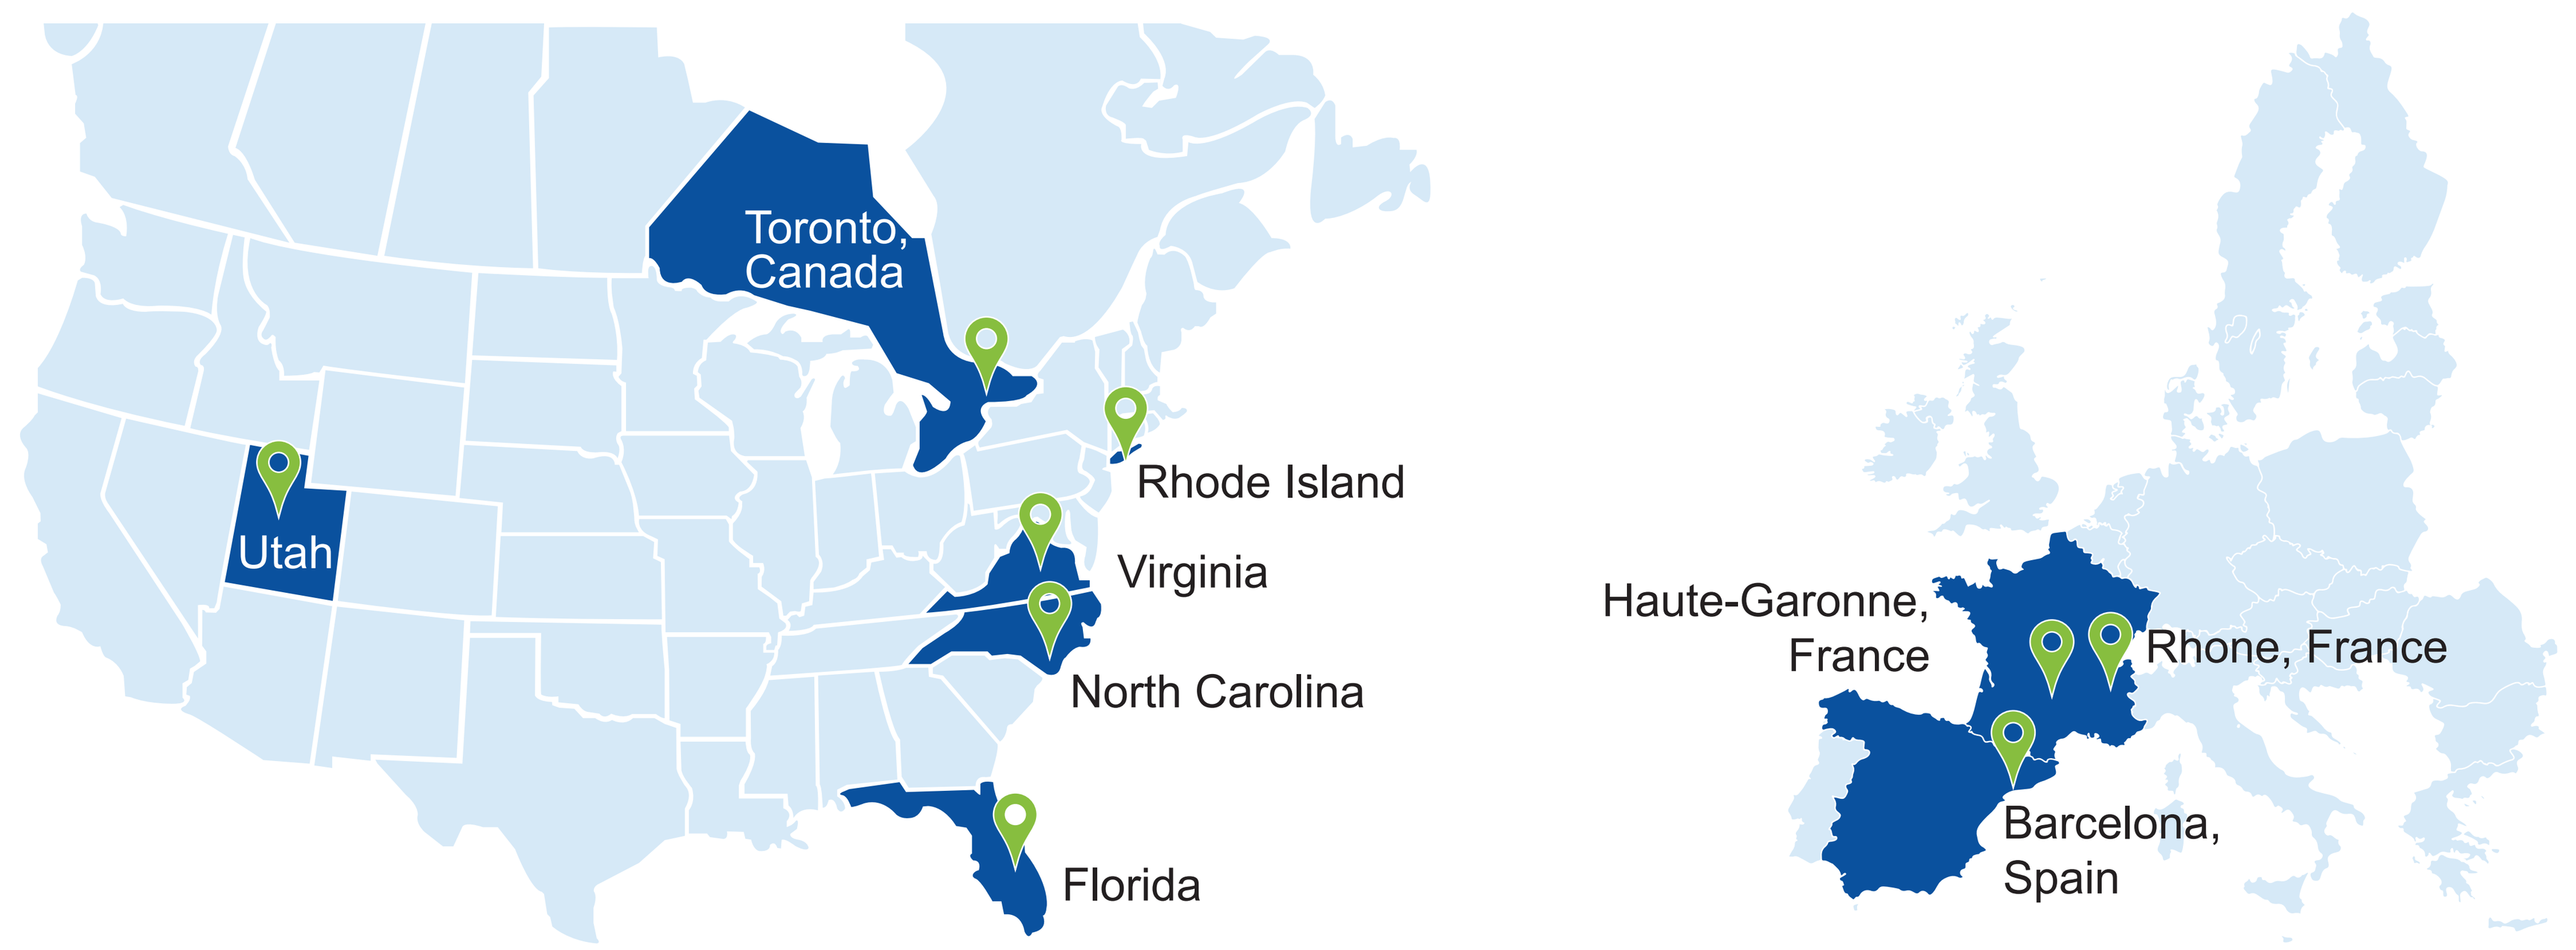
Toronto, Canada
Rhode Island
Utah
Virginia
North Carolina
Florida
Haute-Garonne, France
Rhone, France
Barcelona, Spain

REFERENCES

1. Hegde PS, Chen DS. *Immunity*. 2020;52(1):17-35.
2. Ngambenjawong C, et al. *Adv Drug Deliv Rev*. 2017;114:206-221.
3. Matozaki T, et al. *Trends Cell Biol*. 2009;19(2):72-80.
4. McCracken MN, et al. *Clin Cancer Res*. 2015;21(16):3597-3601.
5. ENHERTU [package insert]. Daiichi Sankyo, Inc.; 2019.
6. Sue M, et al. *J Immunother Cancer*. 2022;10(suppl 2):A843. Abstract A808.
7. Cheng Y, et al. *J Clin Oncol*. 2023;41(16 suppl):e14509.

ACKNOWLEDGMENTS

This study is funded by Daiichi Sankyo, Inc. Medical writing and editorial assistance were provided by Lumanity Communications Inc. and were financially supported by Daiichi Sankyo, Inc.

DISCLOSURES

LC, JM, and PKV have nothing to disclose. LLS serves as a consultant for Merck, Pfizer, AstraZeneca, Roche, GSK, Voronoi, Arvinas, Tessa, Navire, Relay, Daiichi Sankyo, Amgen, Marengo, Medicenna, Tubulis, and LTZ Therapeutics; receives grant/research support from Novartis, Bristol Myers Squibb, Pfizer, Boehringer Ingelheim, GSK, Roche/Genentech, AstraZeneca, Merck, Bayer, AbbVie, Amgen, Symphogen, Intensity Therapeutics, Mirati, Shattuck Labs, BioNTech, 23andMe, and EMD Serono; holds stock in Treadwell Therapeutics (spouse); and reports a relationship with the cofounder of Treadwell Therapeutics (spouse). JT, DM, RS, GA, YC, LH, JECR, YI, and MS are employees of Daiichi Sankyo and may have equity with Daiichi Sankyo. PLB serves as a consultant or advisor for Seattle Genetics, Lilly, Amgen, Merck, Gilead Sciences, Zymeworks, Repare Therapeutics, Bristol Myers Squibb, and Pfizer; and receives research funding from Bristol Myers Squibb, Sanofi, AstraZeneca, Genentech/Roche, GSK, Novartis, Merck, Seattle Genetics, Lilly, Amgen, Bicara, Zymeworks, Medicenna, Bayer, and Takeda.

Presented at the European Society for Medical Oncology (ESMO) Congress; October 20-24, 2023; Madrid, Spain.