Poster # P2.08-01

TROPION-Lung08: Datopotamab Deruxtecan Plus Pembrolizumab in Untreated Advanced/Metastatic Non-Small Cell Lung Cancer (NSCLC)

Caicun Zhou,[1] Benjamin P. Levy,[2] Jacob Sands,[3] Sagun Parakh,[4] Shunichi Sugawara,[5] Konstantinos Syrigos,[6] Jin Seok Ahn,[7] Mingfang Zhao,[8] Chien-Chung Lin,[9] Enriqueta Felip,[10] Federico Cappuzzo,[11] Martin Reck,[12] James C. H. Yang,[13] Rich Joseph,[14] Siddhartha Rawat,[14] Jingdong Xie,[14] Isamu Okamoto[15]

[1]Department of Oncology, Shanghai Pulmonary Hospital and Thoracic Cancer Institute, School of Medicine, Tongji University, Shanghai, China; [2]The Sidney Kimmel Comprehensive Cancer Center at Johns Hopkins, Washington, DC, USA; [3]Dana-Farber Cancer Institute, Boston, MA, USA; [4]Austin Hospital, Heidelberg, VIC, Australia; [5]Sendai Kousei Hospital, Sendai, Japan; [6]Sotiria General Hospital of Chest Diseases, Athens, Greece; [7]Samsung Medical Center, Seoul, Republic of Korea; [8]The First Affiliated Hospital of China Medical University, Shenyang, China; [9]National Cheng Kung University Hospital, College of Medicine, National Cheng Kung University, Tainan, Taiwan; [10]Vall d'Hebron University Hospital, Vall d'Hebron Institute of Oncology, Barcelona, Spain; [11]Istituto Nazionale Tumori Regina Elena, Rome, Italy; [12]Lung Clinic Grosshansdorf, Airway Research Center North (ARCN), Grosshansdorf, Germany; [13]Department of Oncology, National Taiwan University Hospital, Taipei, Taiwan; [14]Daiichi Sankyo, Inc, Basking Ridge, NJ, USA; [15]Kyushu University Hospital, Fukuoka, Japan

Background

- While first-line treatment with immunotherapy (with or without chemotherapy) has improved outcomes in patients with programmed cell death 1 ligand 1 (PD-L1)–expressing advanced/metastatic (adv/met) NSCLC, almost all patients inevitably experience disease progression[1-4]
- Pembrolizumab as monotherapy has shown superior efficacy compared with chemotherapy in treatment-naive patients with advanced NSCLC and PD-L1 expression ≥50%. However, with initial pembrolizumab monotherapy, only approximately one-third of patients are expected to be alive after 5 years[4,5]
- Datopotamab deruxtecan (Dato-DXd) is an antibody-drug conjugate (ADC) composed of a humanized anti–trophoblast cell-surface antigen 2 (TROP2) immunoglobulin G1 (IgG1) monoclonal antibody covalently linked to a topoisomerase I inhibitor payload via a plasma stable tetrapeptide-based cleavable linker[6]
- In the phase 1 TROPION-PanTumor01 trial (NCT03401385), Dato-DXd 6-mg/kg monotherapy demonstrated an objective response rate (ORR) of 26% with a median duration of response (DOR) of 10.5 months and a manageable safety profile in pretreated patients with adv/met NSCLC[7]
 - In addition, preclinical studies showed that DXd ADCs combined with an anti–programmed cell death 1 protein (PD-1) antibody demonstrated superior anti-tumor activity compared with monotherapy with either agent alone[8]
- In the phase 3 TROPION-Lung01 trial (NCT04656652), Dato-DXd 6-mg/kg monotherapy met the dual primary endpoint of progression free survival (PFS), demonstrating a significant improvement in PFS compared with docetaxel in patients with adv/met NSCLC who received ≥1 prior therapy[9]
- In the phase 1b TROPION-Lung02 trial (NCT04526691), first-line Dato-DXd + pembrolizumab doublet treatment and Dato-DXd + pembrolizumab + platinum chemotherapy triplet treatment demonstrated an ORR of 60% and 55%, respectively; the median DOR was not reached in either group[10]
- TROPION-Lung08 (NCT05215340) is a global, randomized, open-label, phase 3 trial of Dato-DXd plus pembrolizumab vs pembrolizumab alone in treatment-naive patients with adv/met NSCLC without actionable genomic alterations and with PD-L1 ≥50%[11,12]

Structure and 7 Key Attributes of Dato-DXd

Dato-DXd is an ADC composed of 3 components[6]

- A humanized anti-TROP2 IgG1 mAb, covalently linked to:
- A topoisomerase I inhibitor payload, an exatecan derivative, via
- A tetrapeptide-based cleavable linker

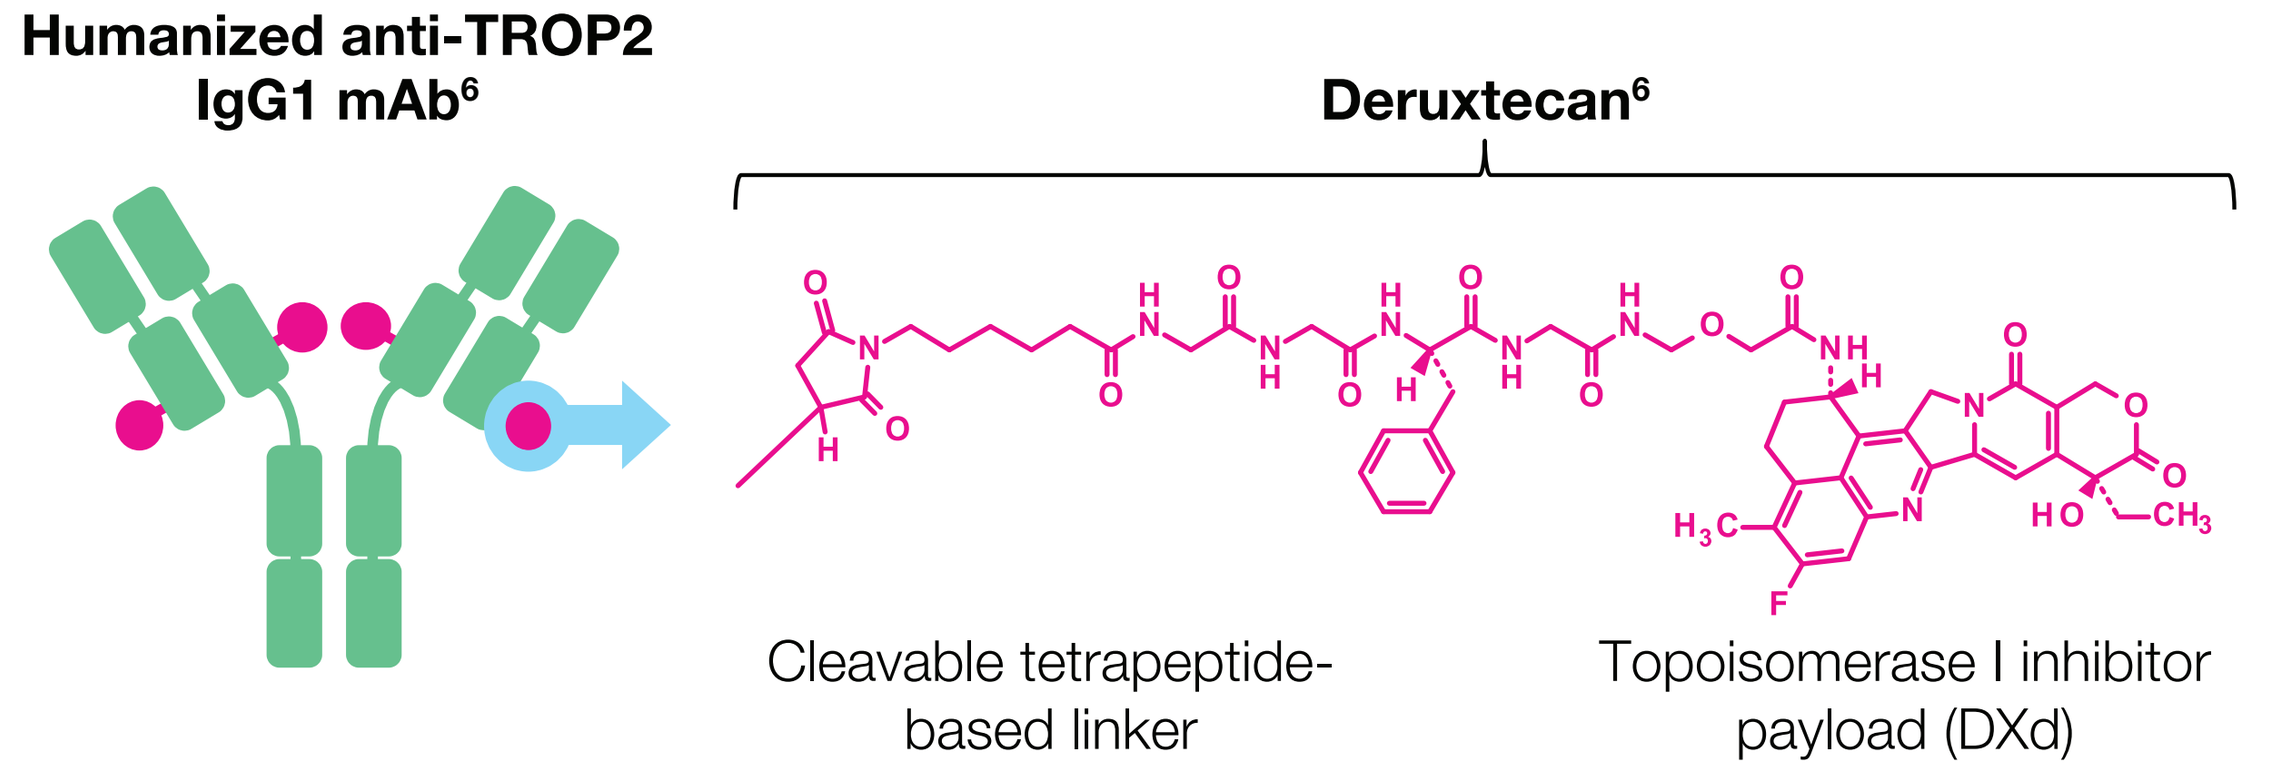

Humanized anti-TROP2 IgG1 mAb[6]
Deruxtecan[6]
Cleavable tetrapeptide-based linker
Topoisomerase I inhibitor payload (DXd)

Payload mechanism of action: topoisomerase I inhibitor[6,13,14,a]
High potency of payload[6,13,14,a]
Optimized drug-to-antibody ratio: ≈4[6,a,b]
Payload with short systemic half-life[6,12,13,a,b]
Stable linker-payload[6,13,14,a]
Tumor-selective cleavable linker[6,12,13,a]
Bystander antitumor effect[13,15,a]

DXd, topoisomerase I inhibitor payload (an exatecan derivative); IgG1, immunoglobulin G1; mAb, monoclonal antibody; TROP2, trophoblast cell-surface antigen 2.
[a] The clinical relevance of these features is under investigation. [b] Based on animal data.

Presented at the IASLC 2023 World Conference on Lung Cancer; September 9-12, 2023; Singapore and virtual.

Methods

Study Design and Population

Patient population (N≈740)

- Stage IIIB, IIIC, or IV NSCLC
- No actionable genomic alterations (documented *EGFR*/*ALK*/*ROS1* negative)
- No previous systemic therapy for adv/met NSCLC[a]
- ECOG PS 0 or 1
- Central PD-L1 TPS ≥50%

Stratified by:

- Histology (squamous vs nonsquamous)
- Geography (East Asia vs rest of world)
- Smoking status (former/current vs never)
- ECOG PS (0 vs 1)

R 1:1

- Dato-DXd 6 mg/kg IV Q3W + pembrolizumab 200 mg IV Q3W
- Pembrolizumab 200 mg IV Q3W

Treatment until:

- Occurrence of progressive disease by BICR, unacceptable toxicity, or other discontinuation criteria
- Completion of 35 cycles of pembrolizumab (no maximum limit for Dato-DXd)

Key Inclusion Criteria

- Age ≥18 years (if the legal age of consent is >18 years old, then follow local regulatory requirements)
- Histologically confirmed NSCLC
 - Stage IIIB or IIIC (and not a candidate for surgical resection or definitive chemoradiation) or stage IV
 - Documented negative test results for *EGFR*, *ALK*, and *ROS1* actionable genomic alterations
 - No known genomic alterations in *NTRK*, *BRAF*, *RET*, *MET*, or other driver kinases with approved therapies
- Tumor tissue sample for the measurement of TROP2 protein expression and exploratory biomarker analyses
- Tumor with high PD-L1 expression (TPS ≥50%) as determined by PD-L1 IHC 22C3 pharmDx
- Measurable disease using RECIST version 1.1
- ECOG PS of 0 or 1

Key Exclusion Criteria

- Prior systemic treatment for advanced or metastatic NSCLC
- Prior treatment with any of the following, including in the adjuvant/neoadjuvant setting[a]:
 - Any agent, including an ADC, containing a chemotherapeutic agent targeting topoisomerase I
 - TROP2-targeted therapy
 - Any anti–PD-1, anti–PD-L1, or anti–PD-L2 agent or any agent directed to another stimulatory or co-inhibitory T-cell receptor
 - Any other immune checkpoint inhibitors
- Active or prior documented ILD/pneumonitis or suspected ILD/pneumonitis that cannot be ruled out by imaging at screening
- Prior radiotherapy ≤4 weeks before start of study intervention or >30 Gy to the lung within 6 months
- Clinically severe pulmonary compromise resulting from intercurrent pulmonary illnesses
- Clinically significant corneal disease
- History of severe hypersensitivity reactions to either the drug substances or inactive ingredients (including but not limited to polysorbate 80) of Dato-DXd or pembrolizumab

Endpoints

Primary endpoints

- PFS by BICR[b]
- OS

Key secondary endpoint

- ORR by BICR[b]

Other secondary endpoints

- PFS by investigator[b]
- ORR by investigator[b]
- DOR by BICR and investigator[b]
- TTR by BICR and investigator[b]
- DCR by BICR and investigator[b]
- PFS2
- PRO
- Safety
- Immunogenicity

Exploratory endpoints

- Biomarkers, pharmacogenetics, and pharmacokinetics exposures will be evaluated for potential association with efficacy and safety

TROPION-Lung08 Study Sites by Region

Enrollment is ongoing

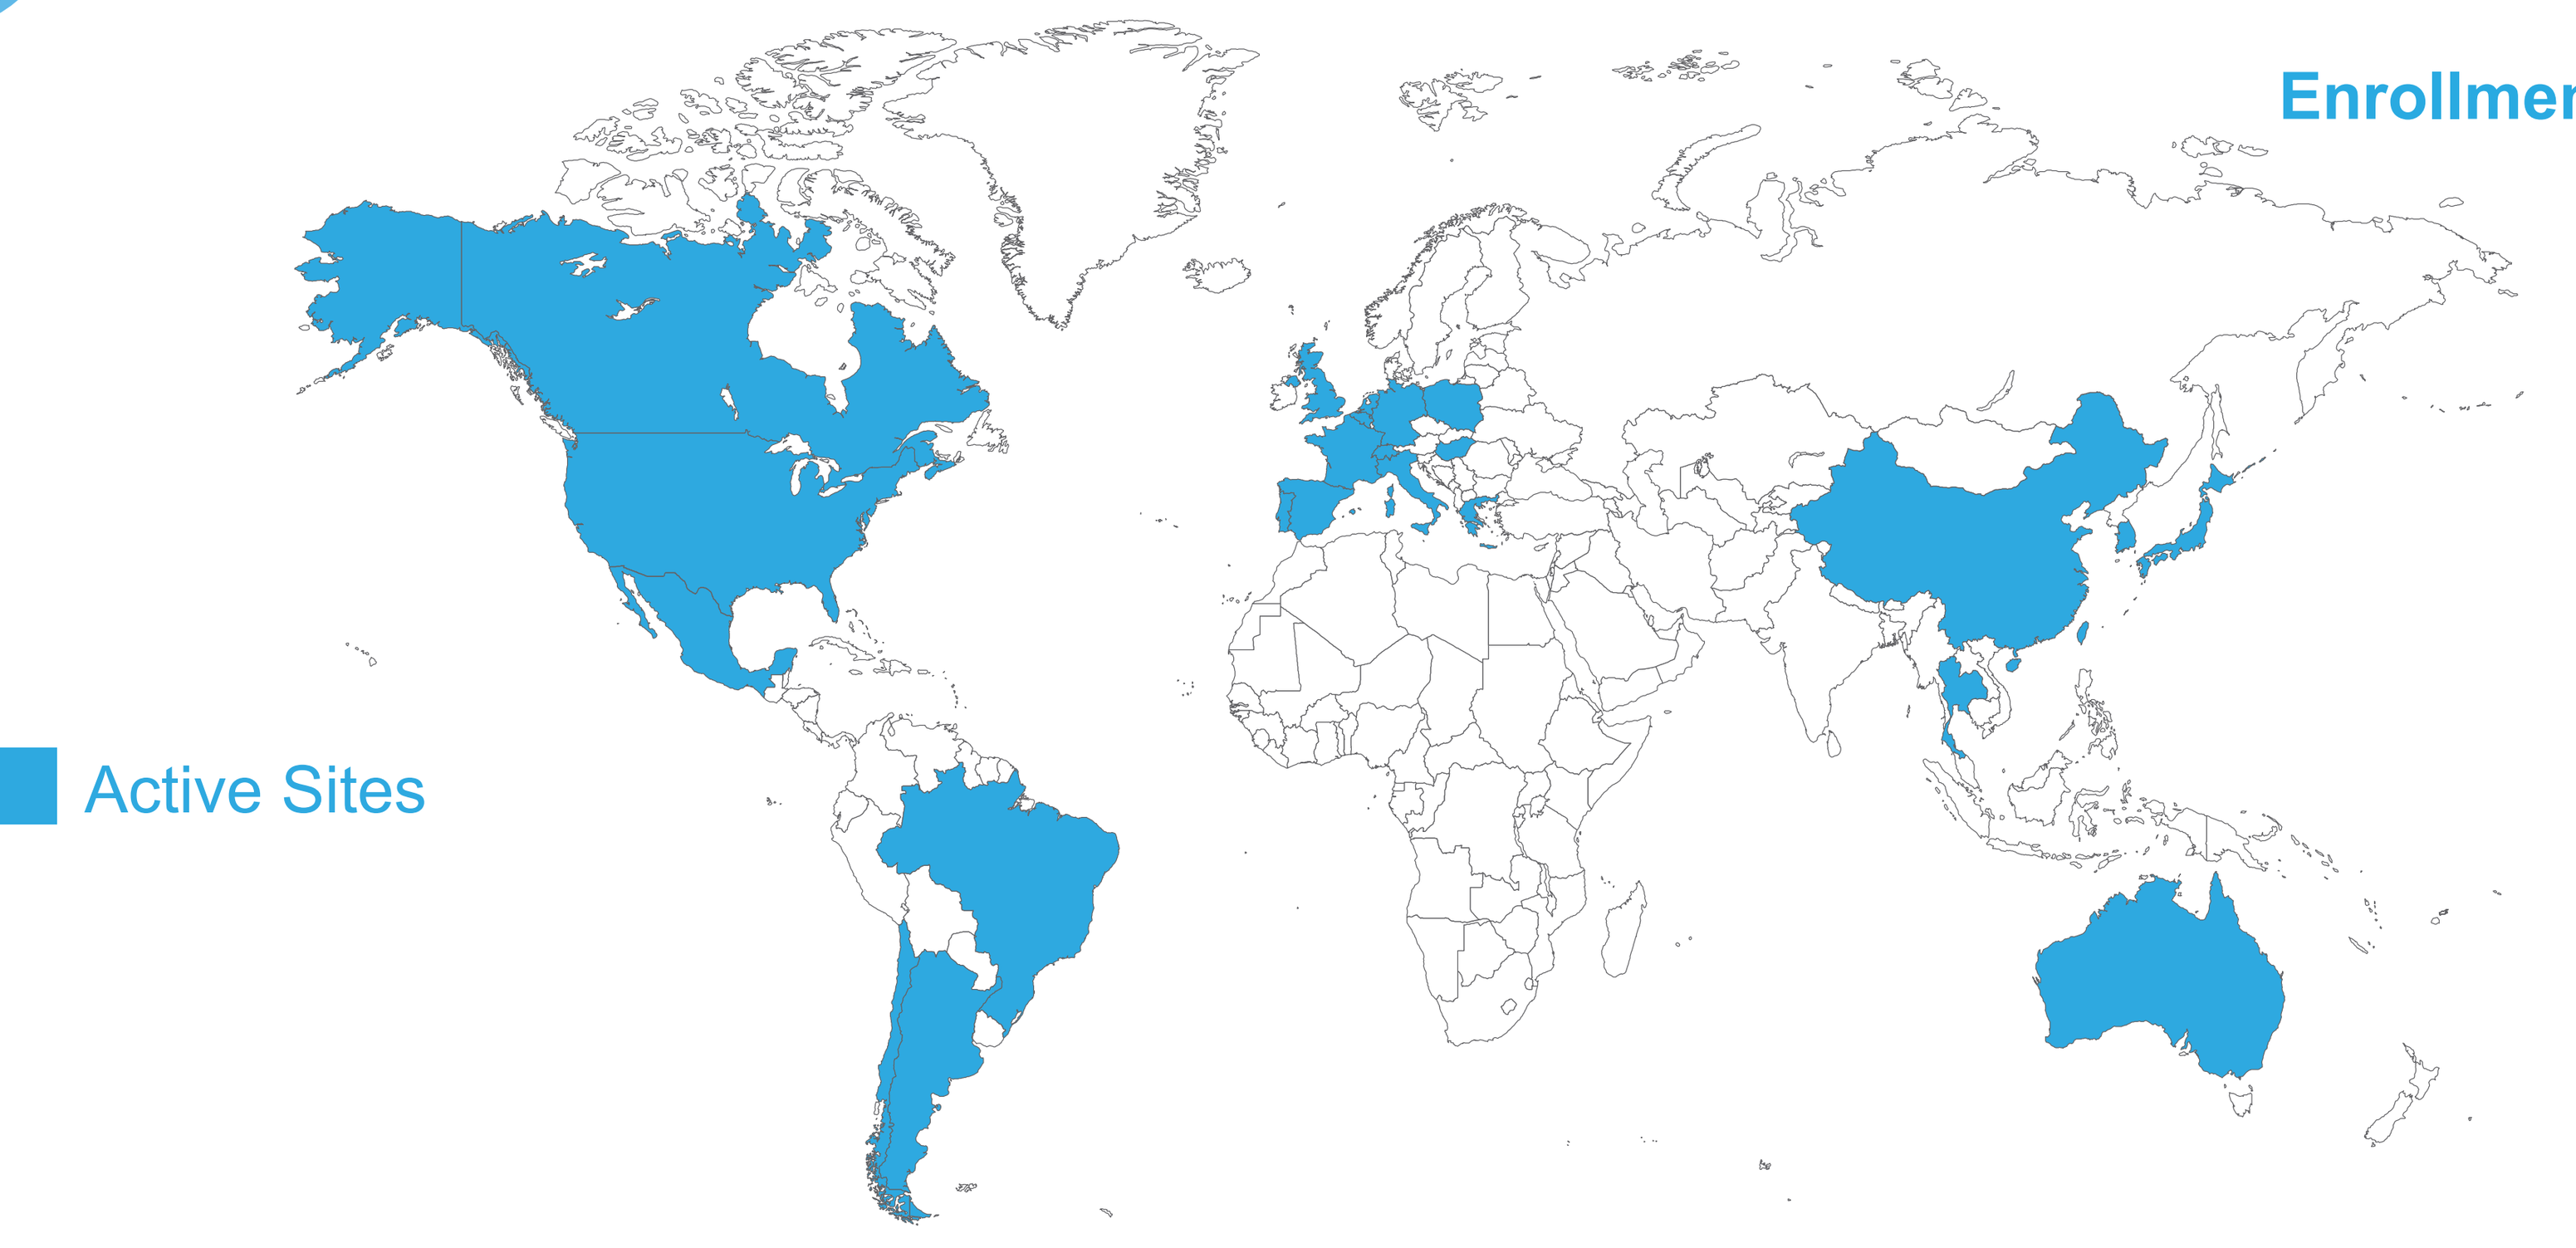

Active Sites

ADC, antibody-drug conjugate; Adv/met, advanced/metastatic; ALK, anaplastic lymphoma kinase; BICR, blinded independent central review; BRAF, B-Raf proto-oncogene, serine/threonine kinase; Dato-DXd, datopotamab deruxtecan; DCR, disease control rate; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; EGFR, epidermal growth factor receptor; ILD, interstitial lung disease; IV, intravenous; MET, MET proto-oncogene, receptor tyrosine kinase; NTRK, neurotrophic receptor tyrosine kinase; ORR, objective response rate; PD-L1, programmed cell death 1 ligand 1; PFS, progression free survival; PFS2, time to second progression; PRO, patient reported outcomes; Q3W, every 3 weeks; R, randomization; RECIST, Response Evaluation Criteria in Solid Tumors; RET, ret proto-oncogene; ROS1, ROS proto-oncogene 1, receptor tyrosine kinase; TPS, tumor proportion score; TTR, time to response.
[a] Patients who previously received neoadjuvant/adjuvant therapy without immune checkpoint inhibitors may be considered for enrollment if therapy was completed ≥6 months prior to the diagnosis of adv/met disease. [b] Assessed per RECIST 1.1.

References

1. Paz-Ares L, et al. *J Thorac Oncol.* 2020;15:1657-1669.
2. Mok TSK, et al. *Lancet.* 2019;393:1819-1830.
3. Rodríguez-Abreu D, et al. *Ann Oncol.* 2021;32:881-895.
4. Reck M, et al. *J Clin Oncol.* 2021;39:2339-2349.
5. Garon EB, et al. *J Clin Oncol.* 2019;37:2518-2527.
6. Okajima D, et al. *Mol Cancer Ther.* 2021;20:2329-2340.
7. Shimizu T, et al. *J Clin Oncol.* Published online June 16, 2023.
8. Iwata T, et al. *Mol Cancer Ther.* 2018;17:1494-1503.
9. News Release. 2023. Available at https://www.businesswire.com/news/home/20230702048365/en/Datopotamab-Deruxtecan-Met-Dual-Primary-Endpoint-of-Progression-Free-Survival-in-Patients-with-Advanced-Non-Small-Cell-Lung-Cancer-in-TROPION-Lung01-Phase-3-Trial
10. Goto Y, et al. Presented at: 2023 ASCO Annual Meeting; June 2-6, 2023; Chicago, IL, and virtual. Abstract 9004.
11. ClinicalTrials.gov. https://clinicaltrials.gov/ct2/show/NCT05215340
12. Levy BP, et al. *Future Oncol.* 2023;19:1461-1472.
13. Nakada T, et al. *Chem Pharm Bull (Tokyo).* 2019;67:173-185.
14. Ogitani Y, et al. *Clin Cancer Res.* 2016;22:5097-5108.
15. Ogitani Y, et al. *Cancer Sci.* 2016;107:1039-1046.

Acknowledgments

We thank the patients, their families, and their caregivers for their participation and study staff for their contributions.

This study is sponsored by Daiichi Sankyo, Inc. In July 2020, Daiichi Sankyo entered into a global development and commercialization collaboration with AstraZeneca for datopotamab deruxtecan (Dato-DXd).

Pembrolizumab is being provided under agreement by Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Rahway, NJ, USA. The authors acknowledge Sung Jin (Laura) Kim of Merck & Co., Inc., Rahway, NJ, USA for her contributions during protocol development.

Medical writing support was provided by Ari Simenauer, PhD, of SciMentum, Inc., The Nucleus Group Holdings, Inc., and was funded by Daiichi Sankyo, Inc. Editorial support was provided in accordance with Good Publication Practice guidelines (ismpp.org/gpp3).

Funding

This study is sponsored by Daiichi Sankyo, Inc.

Please scan this quick response (QR) code with your smartphone camera or app to obtain additional materials. Alternatively, please use the link below.

https://bit.ly/3S2ilea

Copies of this poster/presentation and other materials obtained through the QR or text keycodes are for personal use only and may not be reproduced without written permission of the authors.